Pre-clinical Activity of hLL2-PBD, a Novel Anti-CD22 Antibody-Pyrrolobenzodiazepine (PBD) Conjugate in Models of Non-Hodgkin Lymphoma

Francesca Zammarchi [1], David G. Williams [2], Karin Havenith [1], Lauren Adams [2], Francois D'Hooge [2], Phil Howard [1, 2], John A. Hartley [1,2,3], Patrick van Berkel [1]

[1] ADC Therapeutics Sarl, Epalinges, Switzerland; [2] Spirogen, London, United Kingdom; [3] University College London, London, United Kingdom

Introduction

- **CD22 is a type-I transmembrane sialoglycoprotein, whose expression is restricted to the B-cell lineage [1]. CD22 is also found highly expressed on most malignant mature B cells, including follicular lymphoma (FL), marginal zone lymphoma (MZL), mantle cell lymphoma (MCL), diffuse large B-cell lymphoma (DLBCL), small lymphocytic lymphoma (SLL) and chronic lymphocytic leukemia (CLL) [2, 3]. Moreover, CD22 is expressed in > 90% of cases of B-precursor acute lymphoblastic leukemia (ALL) [4].**

Figure 1- CD22

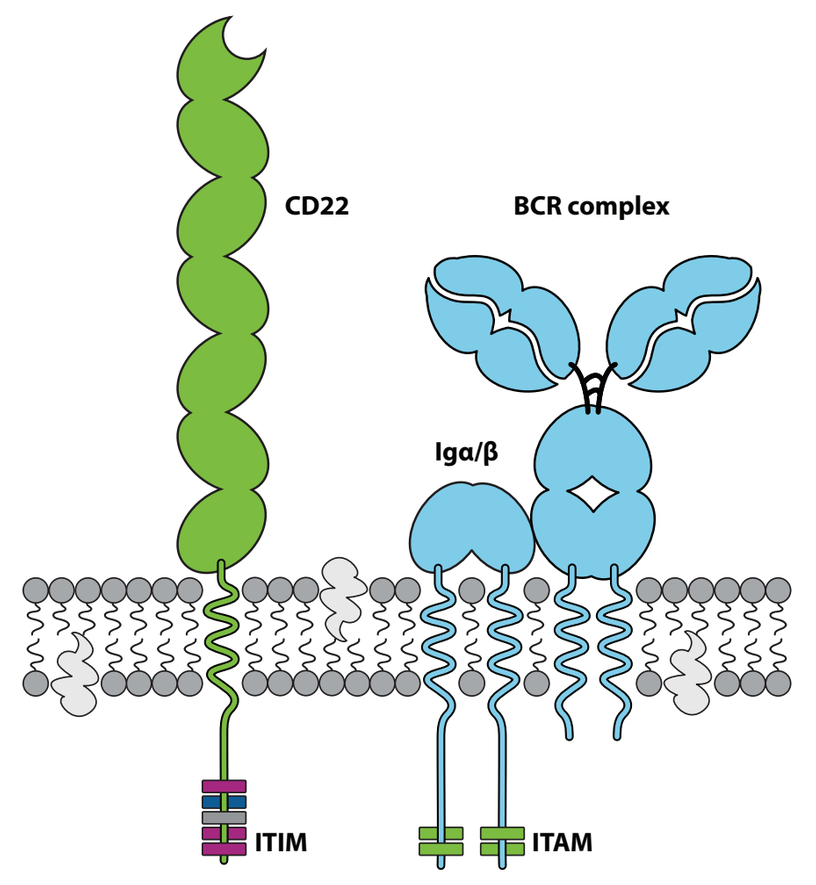
CD22
BCR complex
Igα/β
ITIM
ITAM

- **The differential and favourable expression profile of CD22 in tumour versus normal tissue, together with its rapid internalization upon binding ligand or antibody [5], make CD22 an attractive target for antibody drug conjugate (ADC)-mediated treatment of B-cell malignancies.**

- **hLL2-PBD is an ADC composed of the humanized anti-human CD22 monoclonal antibody epratuzumab (hLL2), stochastically conjugated via a cathepsin-cleavable valine-alanine (val-ala) peptide linker to a PBD dimer toxin. The drug to antibody ratio (DAR) is 2.5. PBD dimers represent a novel class of toxin, which bind DNA in the minor groove and form highly cytotoxic interstrand cross-links.**

Figure 2- hLL2-PBD

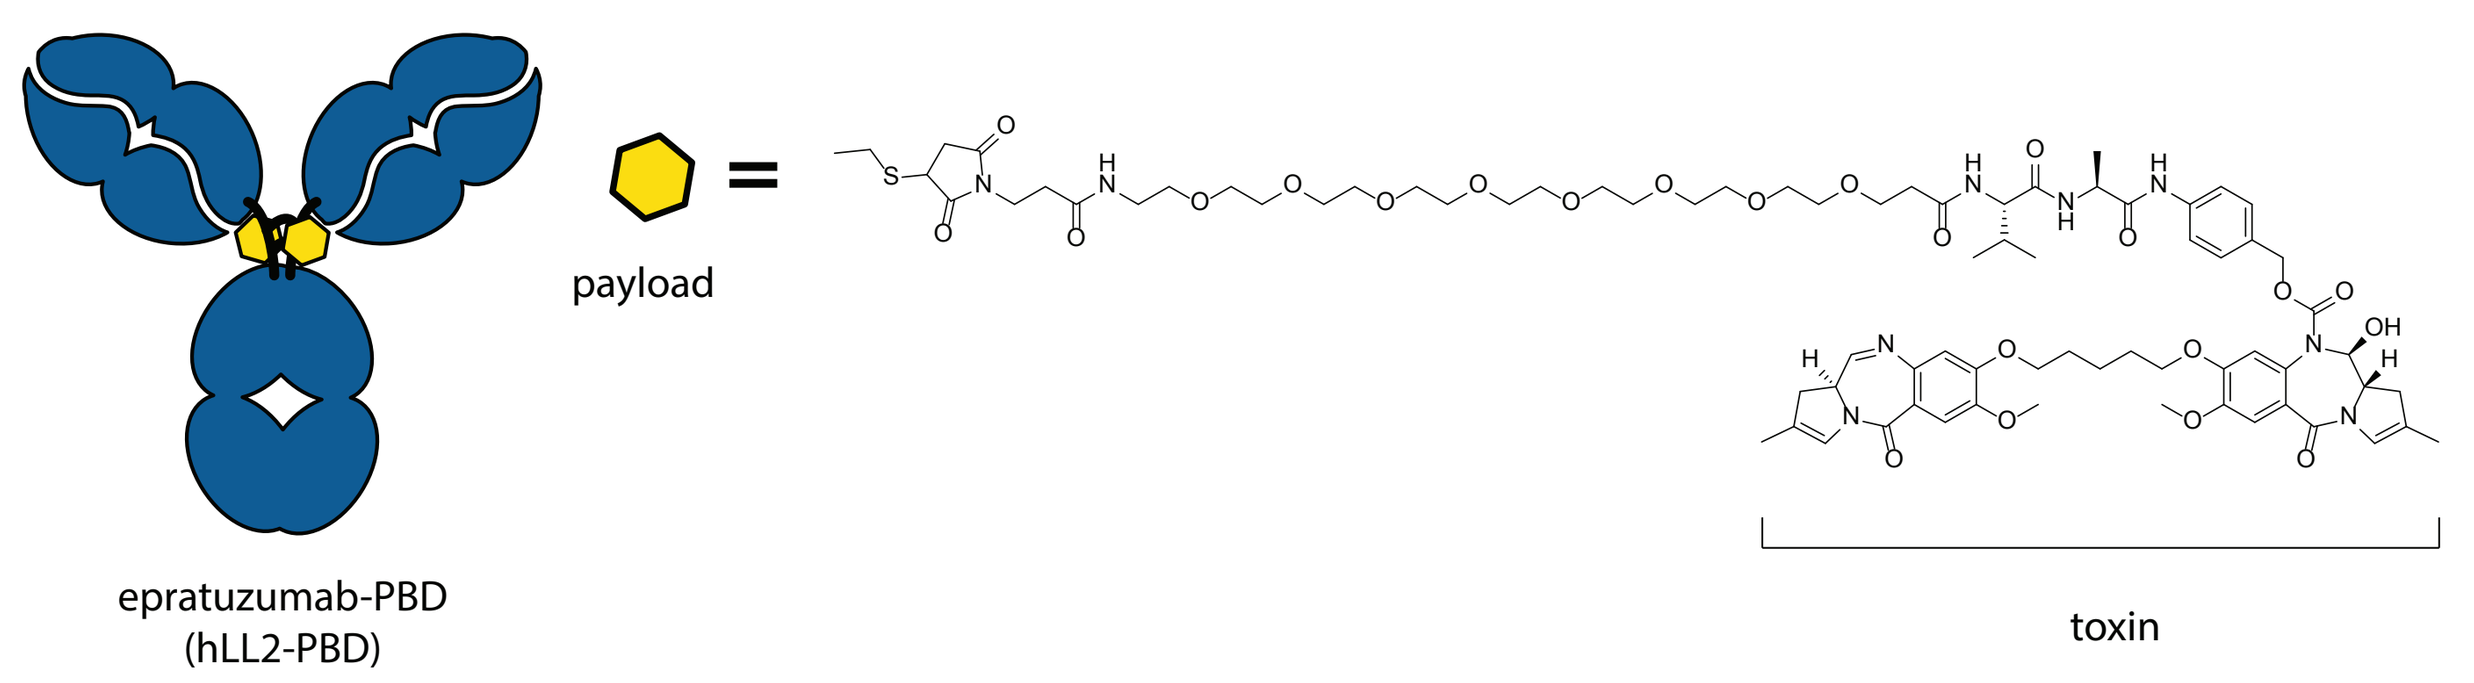
payload
epratuzumab-PBD
(hLL2-PBD)
toxin

Aim of this study

Characterization of the *in vitro* cytotoxicity, *in vivo* efficacy and pharmacokinetics (PK) of hLL2-PBD.

Materials & Methods

Cytotoxicity of hLL2-PBD, Hu10F4-vcMMAE [2] and non-binding PBD-ADC on human Ramos, Daudi and WSU-DLCL2 cells was determined by the CellTiter 96® AQueous One Solution Cell Proliferation Assay (MTS assay) (Promega).

In vivo, hLL2-PBD was administered as a single dose to CB.17 Severe Combined ImmunoDeficient (SCID) mice containing Daudi or Ramos xenografts (both as subcutaneous models) and compared to a single dose of Hu10F4-vcMMAE.

PK analysis of hLL2-PBD was performed in female CB.17 SCID mice (8 weeks old). Serum samples were collected from three mice per group for each time point after a single dose administration (1 mg/kg). Time points were 15 minutes (min), 30 min, 1 hour (hr), 8 hr, 24 hr, 3 days, 7, 10, 14, 18, 21, and 28 days after the dose.

Quantitation of total hLL2 antibody and of PBD-conjugated hLL2 was done by ELISA. For total hLL2 quantitation, a biotinylated anti-human IgG-Fc antibody was used for capture. For quantitation of PBD-conjugated antibody, an anti-PBD mouse antibody was used for capture. For both assays, anti-human IgG-Fc-HRP conjugated antibody was used for detection.

Results

Figure 3- Scale-up production of hLL2-PBD is a straightforward and high yielding process

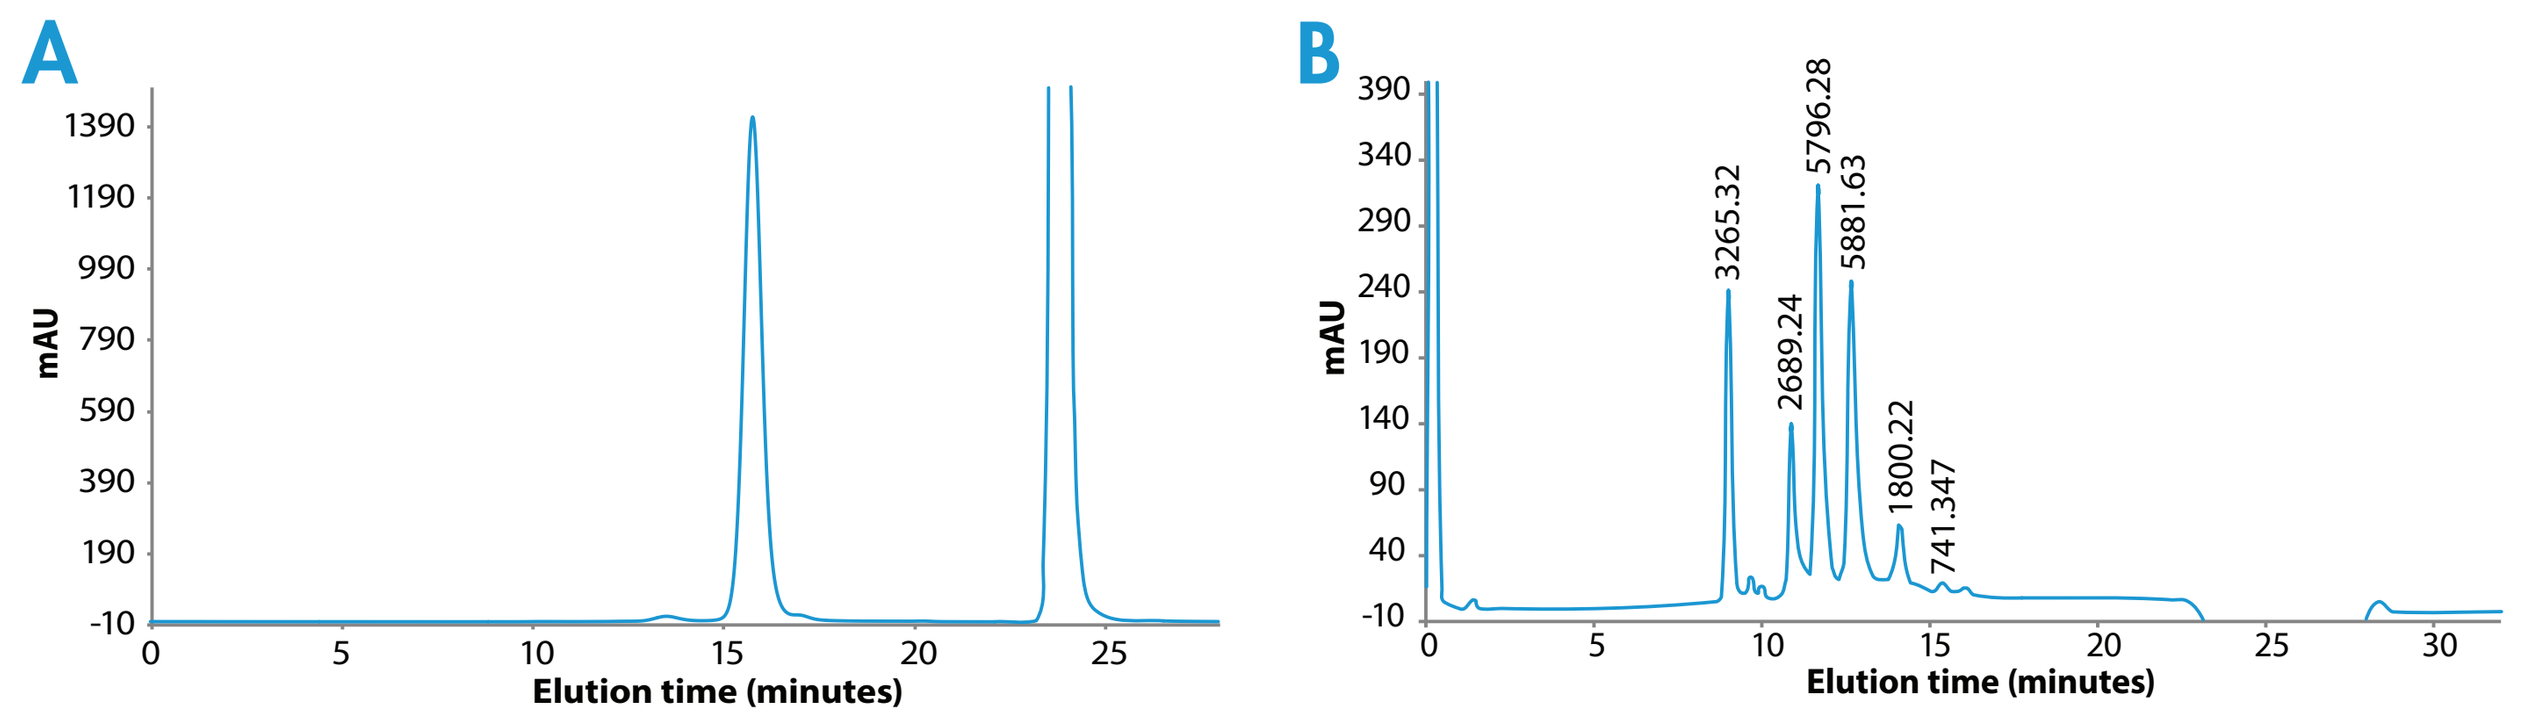
A
B
mAU
Elution time (minutes)
3265.32
2689.24
5796.28
5881.63
1800.22
741.347

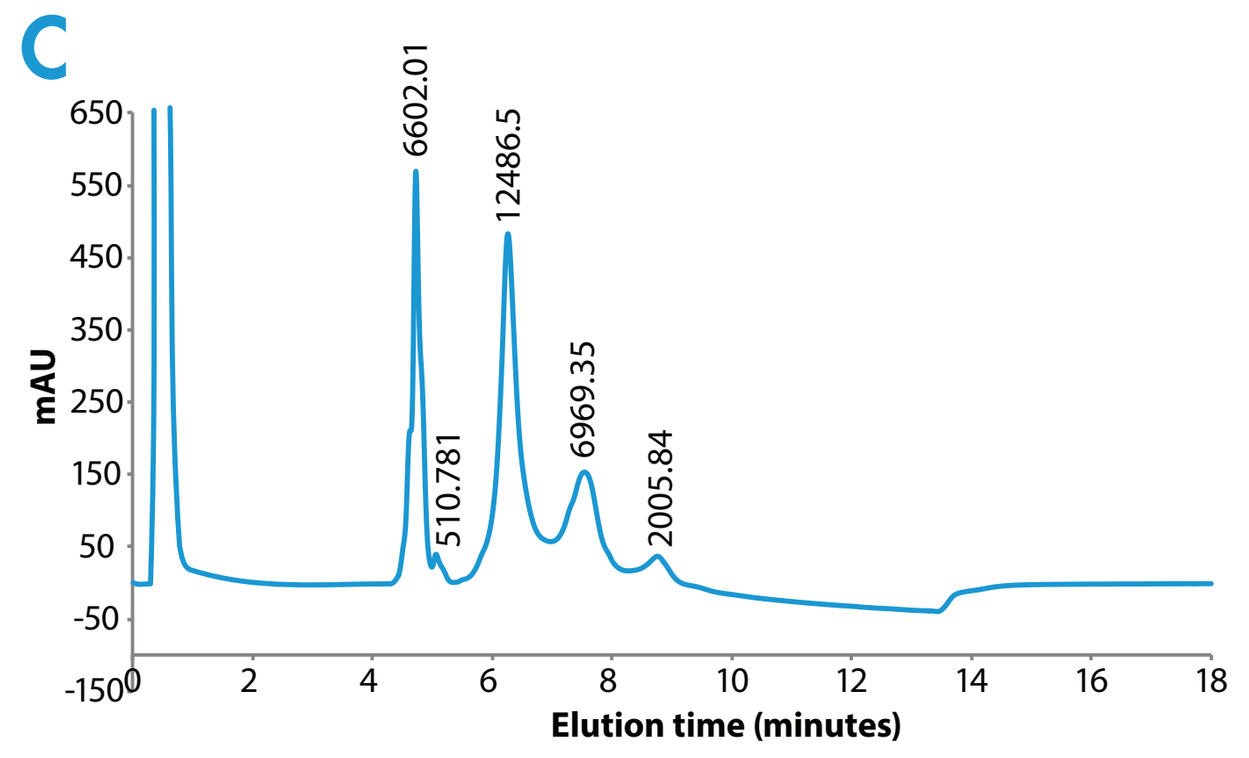
C
mAU
Elution time (minutes)
6602.01
510.781
12486.5
6969.35
2005.84

D

	hLL2-PBD
concentration (mg/mL)	1.97
monomer	98.7% (1.3% dimers)
DAR by PLRP	2.5
% DAR 0 by HIC	23.1
% DAR 2 by HIC	43.7
% DAR 4 by HIC	24.4

hLL2-PBD with a DAR of 2.5 was produced at the 100 mg scale with high yield (97%) without using an aggregate removal step.

A. hLL2-PBD size exclusion chromatography (SEC), **B.** polymer reversed phase (PLRP), and **C.** hydrophobic interaction chromatography (HIC) analysis. **D.** Summary table of manufactured hLL2-PBD analysis.

Figure 4- Targeted cytotoxicity

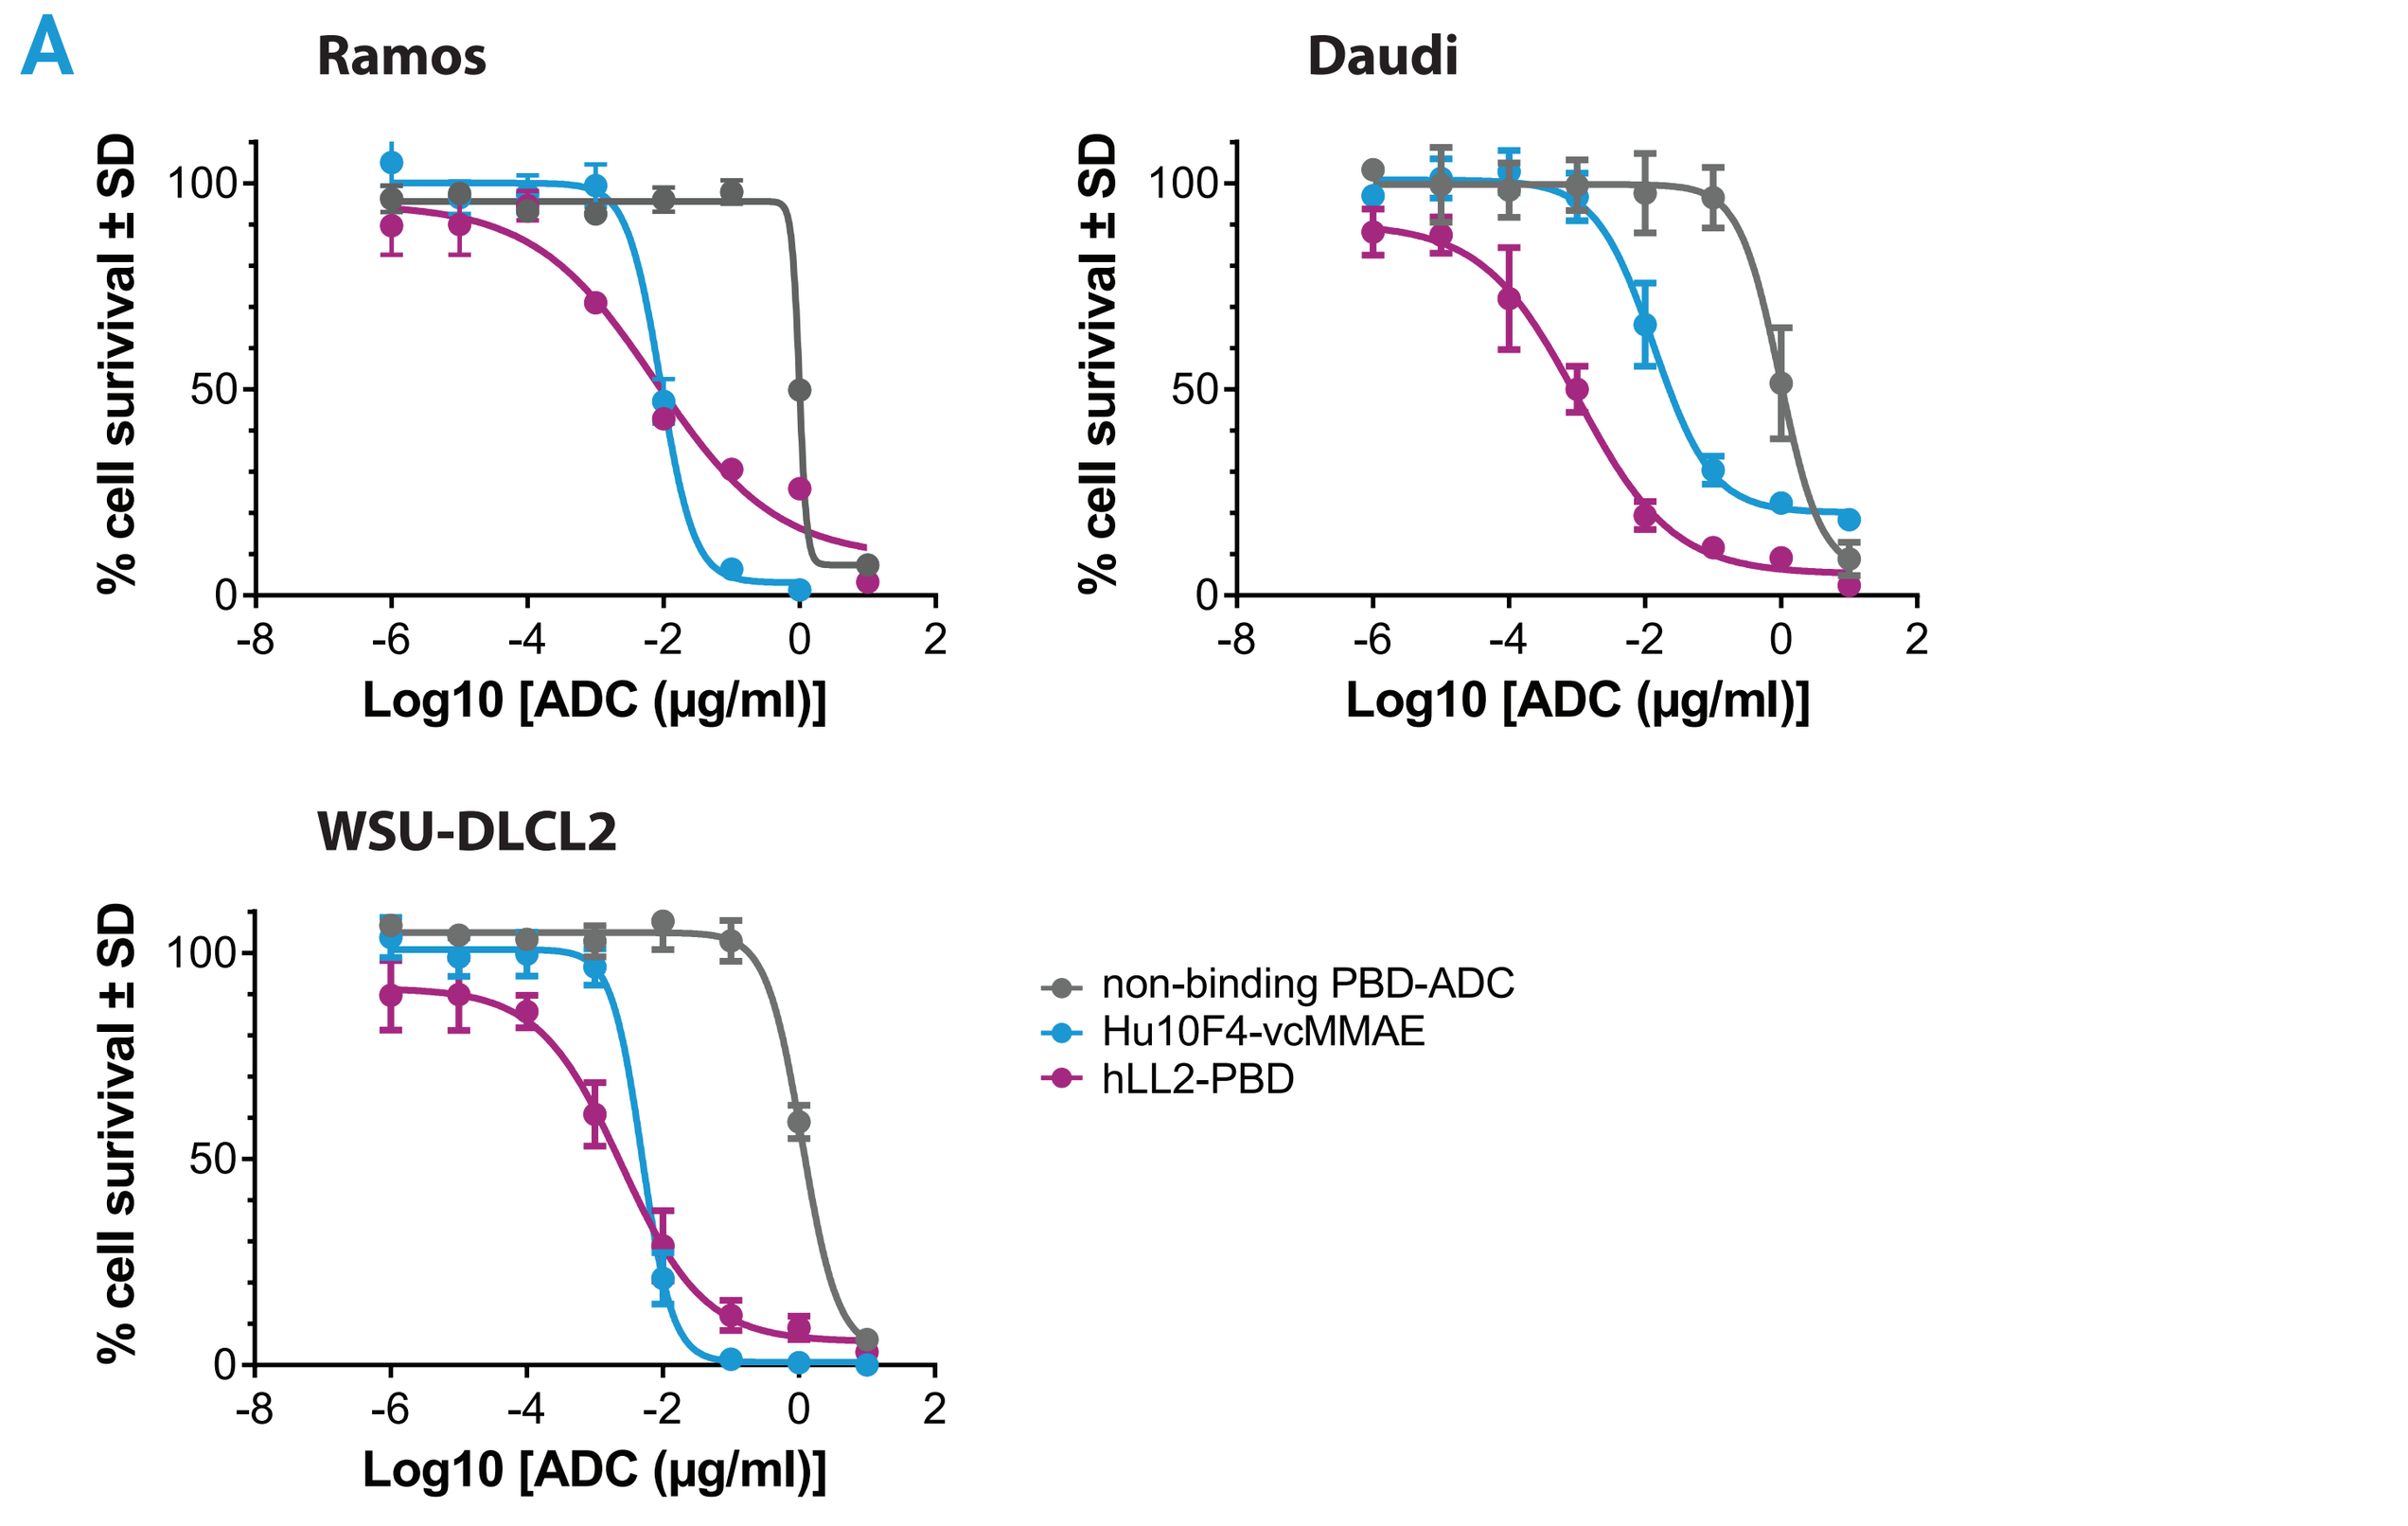
A
Ramos
Daudi
WSU-DLCL2
% cell survival ± SD
Log10 [ADC (µg/ml)]
non-binding PBD-ADC
Hu10F4-vcMMAE
hLL2-PBD

B

cell line (tumor type)		Ramos (BL)	Daudi (BL)	WSU-DLCL2 (DLCBL)
mean GI_{50} µg/mL (pM)	Non-binding PBD-ADC (DAR 2.7)	0.99 (6,600)	0.96 (6,400)	1.12 (7,500)
	hLL2-PBD (DAR 2.5)	0.008 (53)	0.001 (6.7)	0.002 (13)
	Hu10F4-vcMMAE (DAR 4.6)	0.009 (60)	0.013 (87)	0.005 (33)

A. *In vitro* cytotoxicity of hLL2-PBD, Hu10F4-vcMMAE and non-binding PBD-ADC after 96-hour exposure on three lymphoma cell lines, as measured by the MTS cell viability assay.

B. Mean GI_{50} values of the three ADCs (hLL2-PBD, Hu10F4-vcMMAE and non-binding PBD-ADC) in Ramos, Daudi and WSU-DLCL2 cell lines. BL: Burkitt's lymphoma, DLBCL: diffuse large B cell lymphoma.

Figure 5- *In vivo* antitumor efficacy in subcutaneously (s.c.) implanted Daudi model

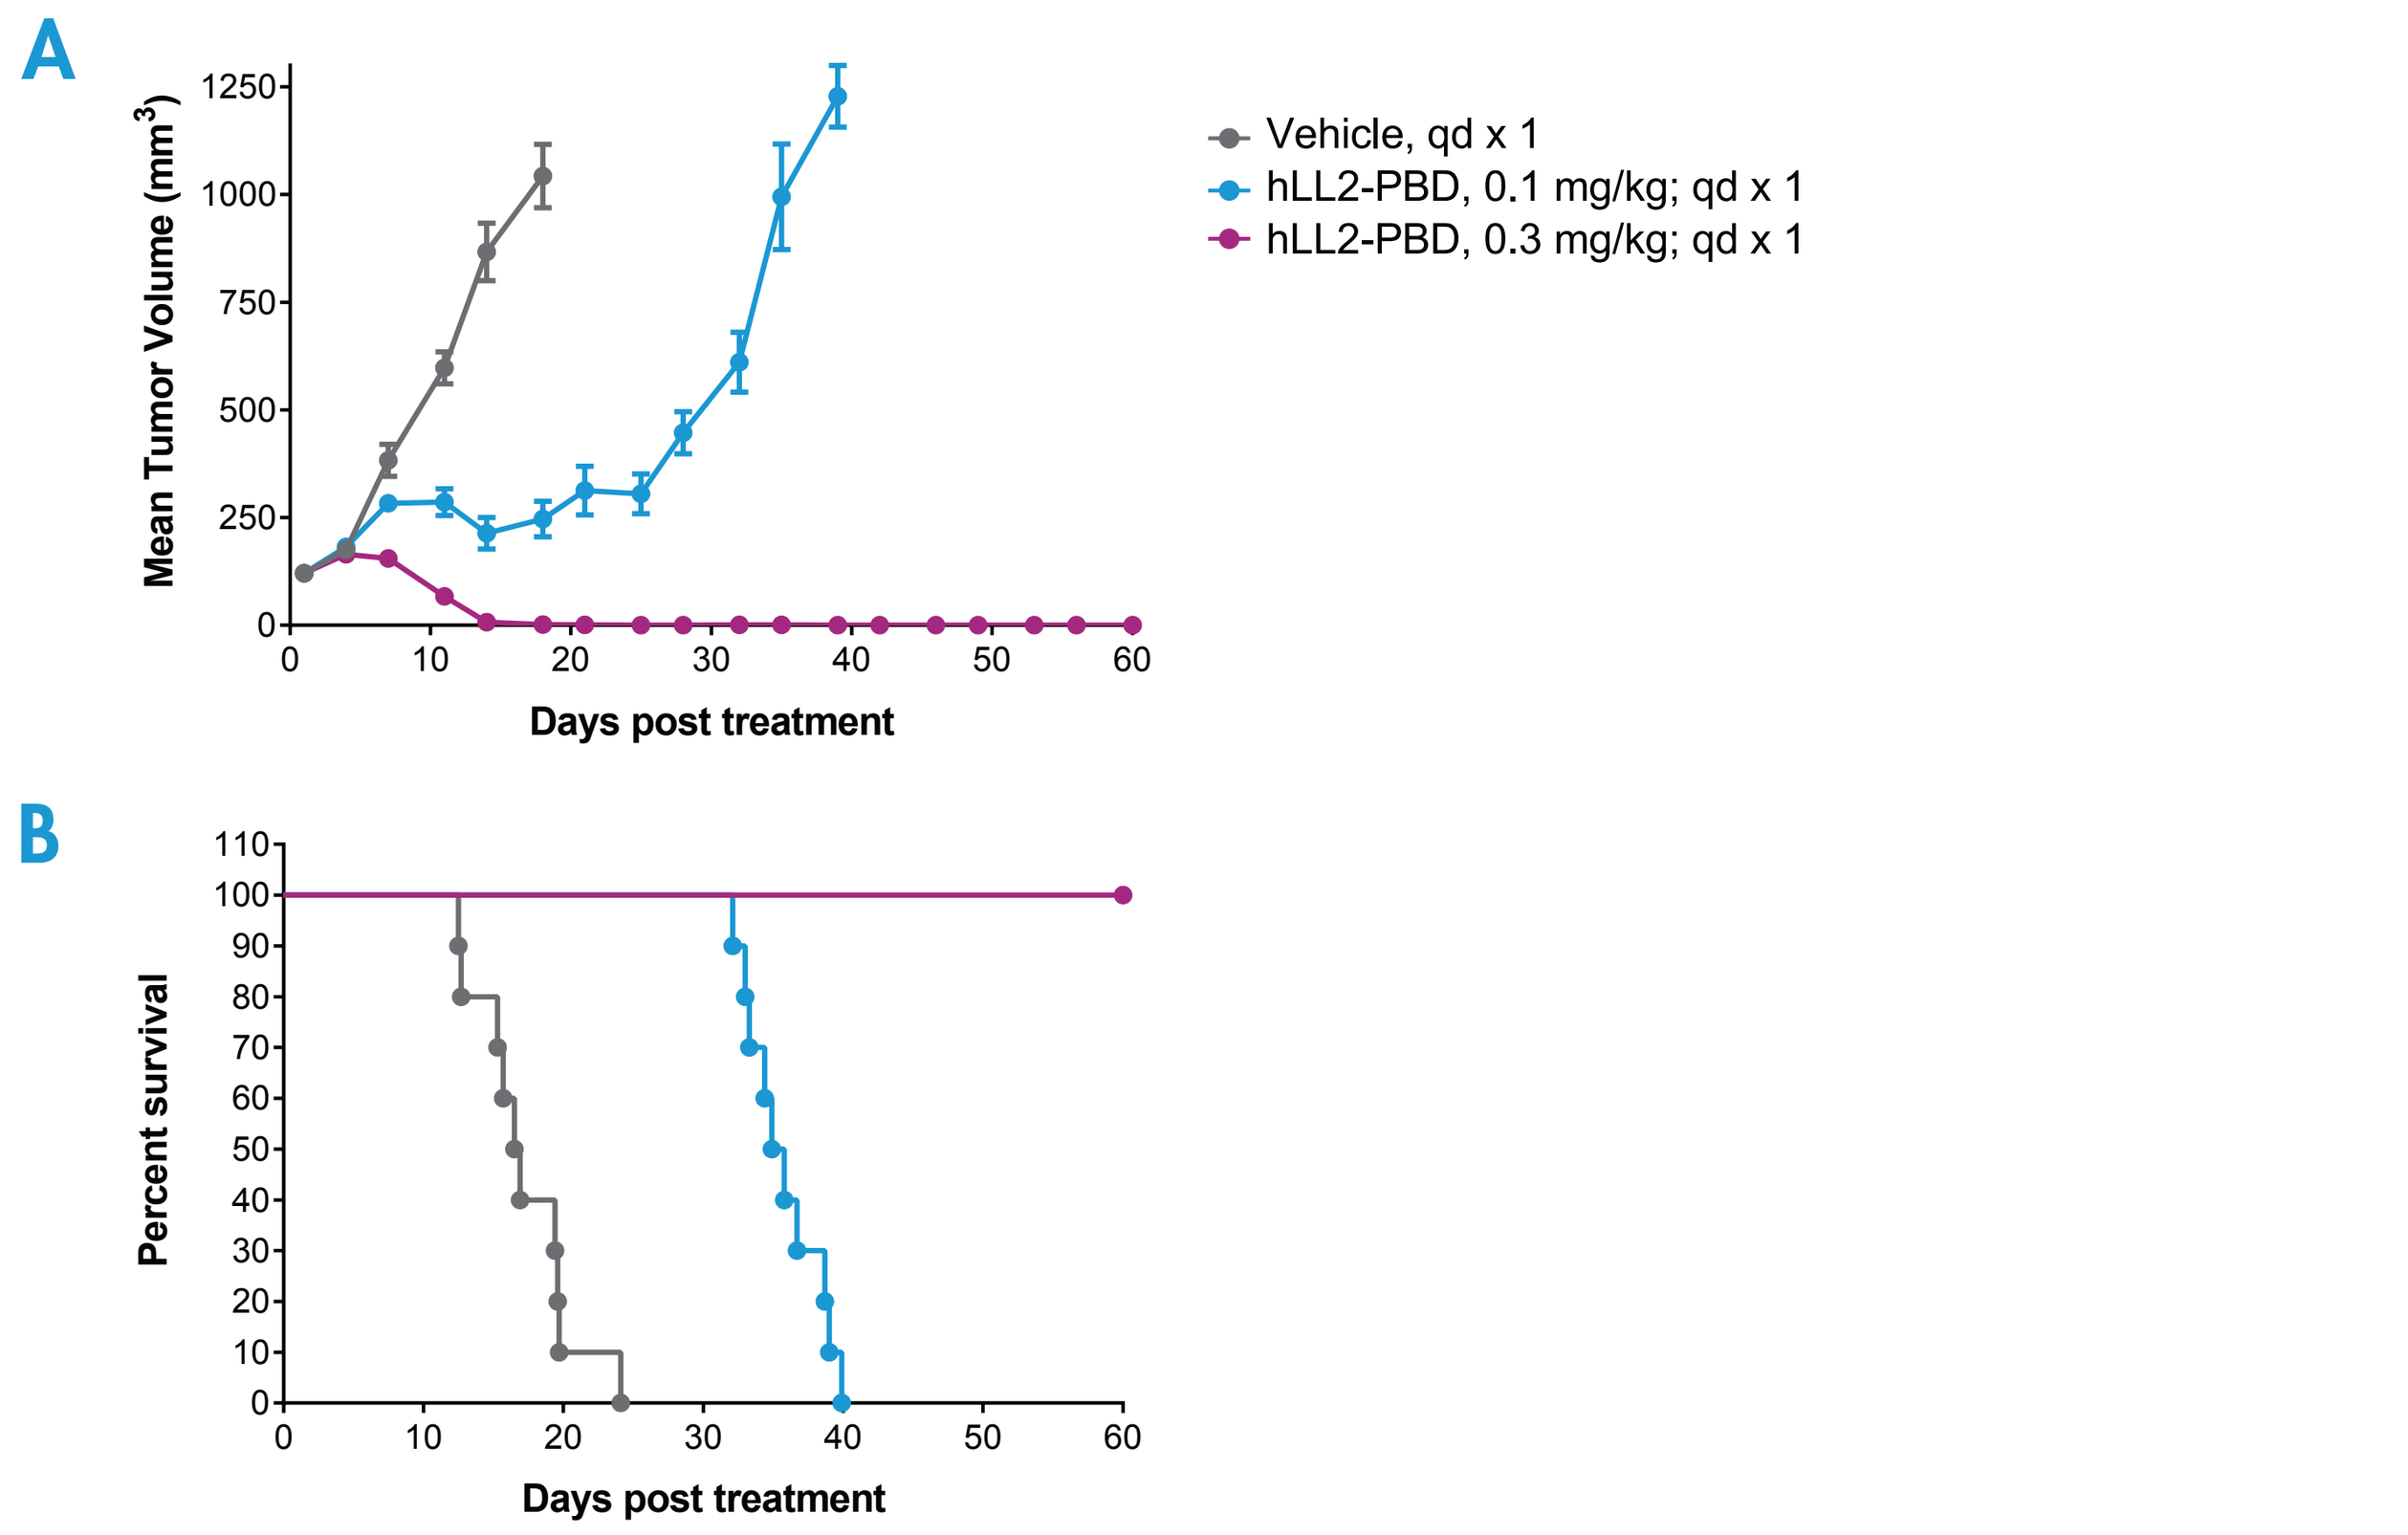
A
Mean Tumor Volume (mm³)
Days post treatment
Vehicle, qd x 1
hLL2-PBD, 0.1 mg/kg; qd x 1
hLL2-PBD, 0.3 mg/kg; qd x 1
B
Percent survival
Days post treatment

A. Eight-week-old SCID mice were s.c. implanted with 1 x 10^7 Daudi cells. When tumors reached mean volumes of 120 mm^3, mice were sorted into groups of 10 mice each and doses were administered. hLL2-PBD, administered i.v., as a single dose, at either 0.1 mg/kg or 0.3 mg/kg, induced a dose-dependent anti-tumor response. At the highest dose tested (0.3 mg/kg) all the animals were classified as tumor-free survivors at the end of the study (day 60).

B. Kaplan-Meier survival curves showing the dose-dependent extension of survival (log-rank test, p ≤ 0.001 for each comparison).

Figure 6- *In vivo* antitumor efficacy in s.c. implanted Ramos model

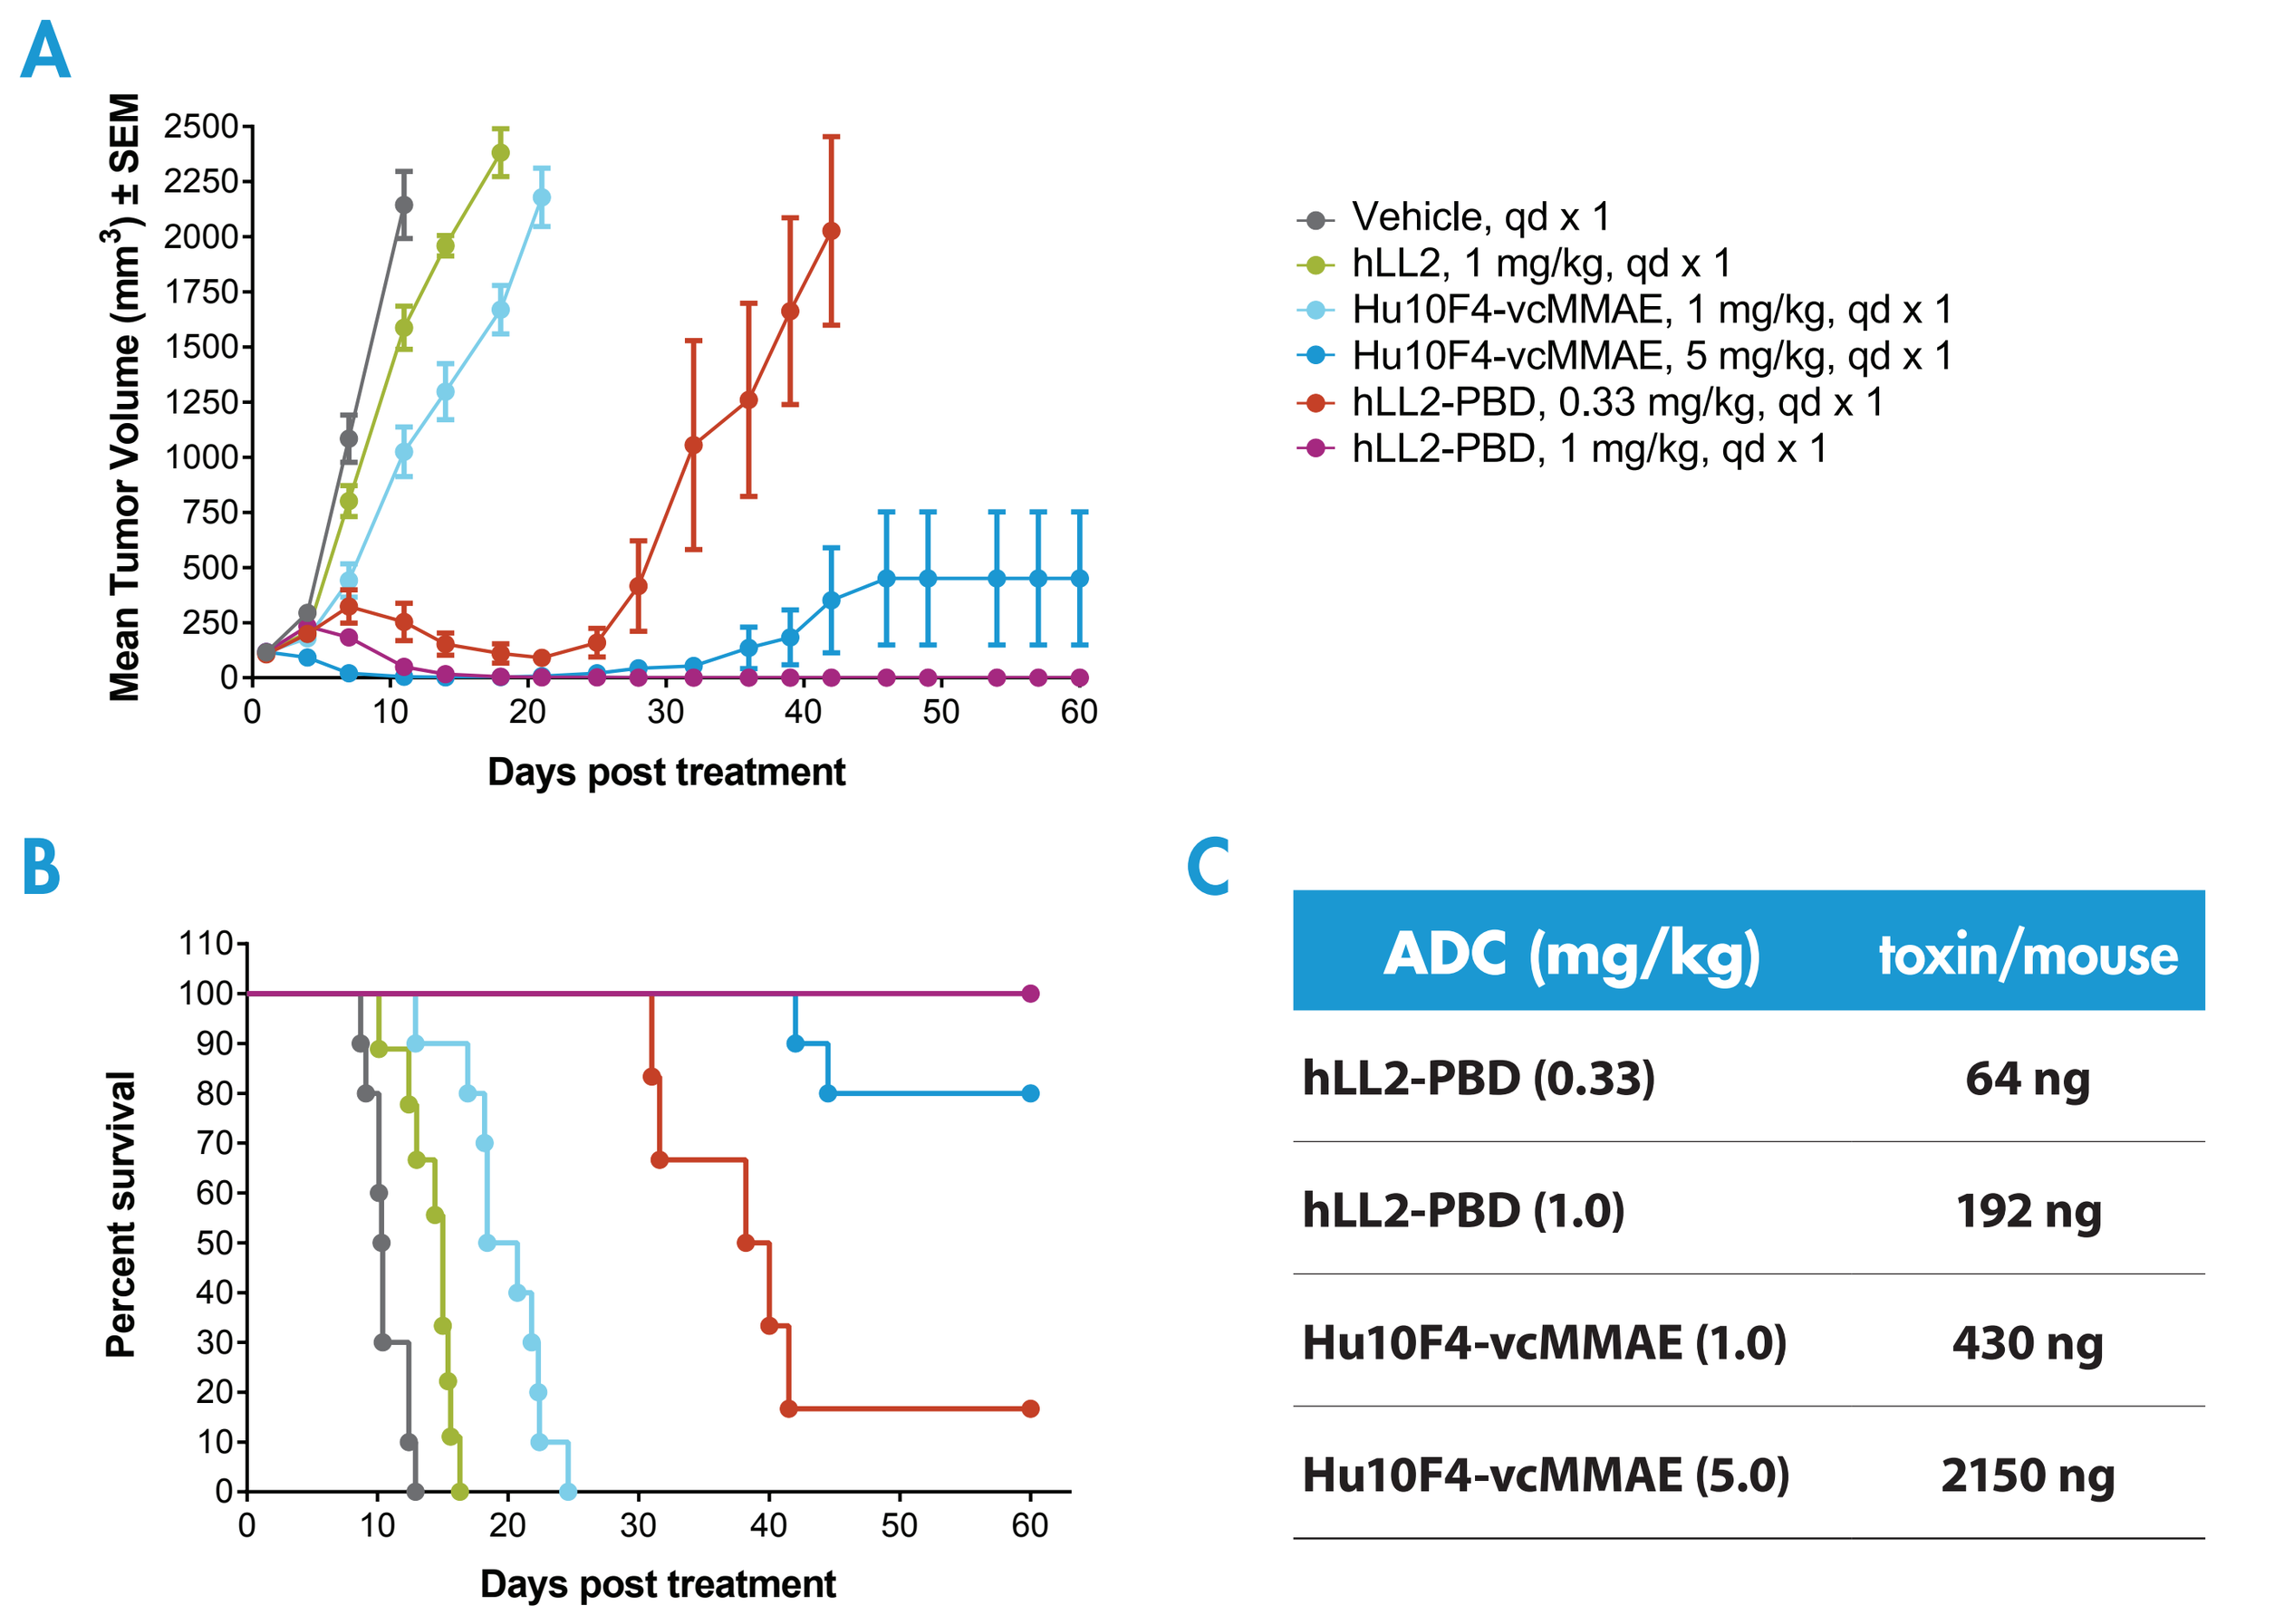
A
Mean Tumor Volume (mm³) ± SEM
Days post treatment
Vehicle, qd x 1
hLL2, 1 mg/kg, qd x 1
Hu10F4-vcMMAE, 1 mg/kg, qd x 1
Hu10F4-vcMMAE, 5 mg/kg, qd x 1
hLL2-PBD, 0.33 mg/kg, qd x 1
hLL2-PBD, 1 mg/kg, qd x 1
B
Percent survival
Days post treatment

C

ADC (mg/kg)	toxin/mouse
hLL2-PBD (0.33)	64 ng
hLL2-PBD (1.0)	192 ng
Hu10F4-vcMMAE (1.0)	430 ng
Hu10F4-vcMMAE (5.0)	2150 ng

A. Eight- to nine-week-old SCID mice were s.c. implanted with 1 x 10^7 Ramos cells. When tumors reached mean volumes of 116 mm^3, mice were sorted into groups of 10 mice each and doses were administered. hLL2-PBD (DAR 2.5), HuB10F4-vcMMAE (DAR 4.6) and unconjugated hLL2 were administered i.v., as a single dose. hLL2-PBD activity was remarkably superior to Hu10F4-vcMMAE when tested at the same dose level (1 mg/kg).

B. Kaplan-Meier survival curves. hLL2-PBD administered at 1 mg/kg induced a significant increase of survival compared to Hu10F4-vcMMAE when tested at the same dose level (log-rank test, p ≤ 0.001).

C. Table indicates equivalent toxin amount (ng) administered to every mouse.

Figure 7- PK analysis

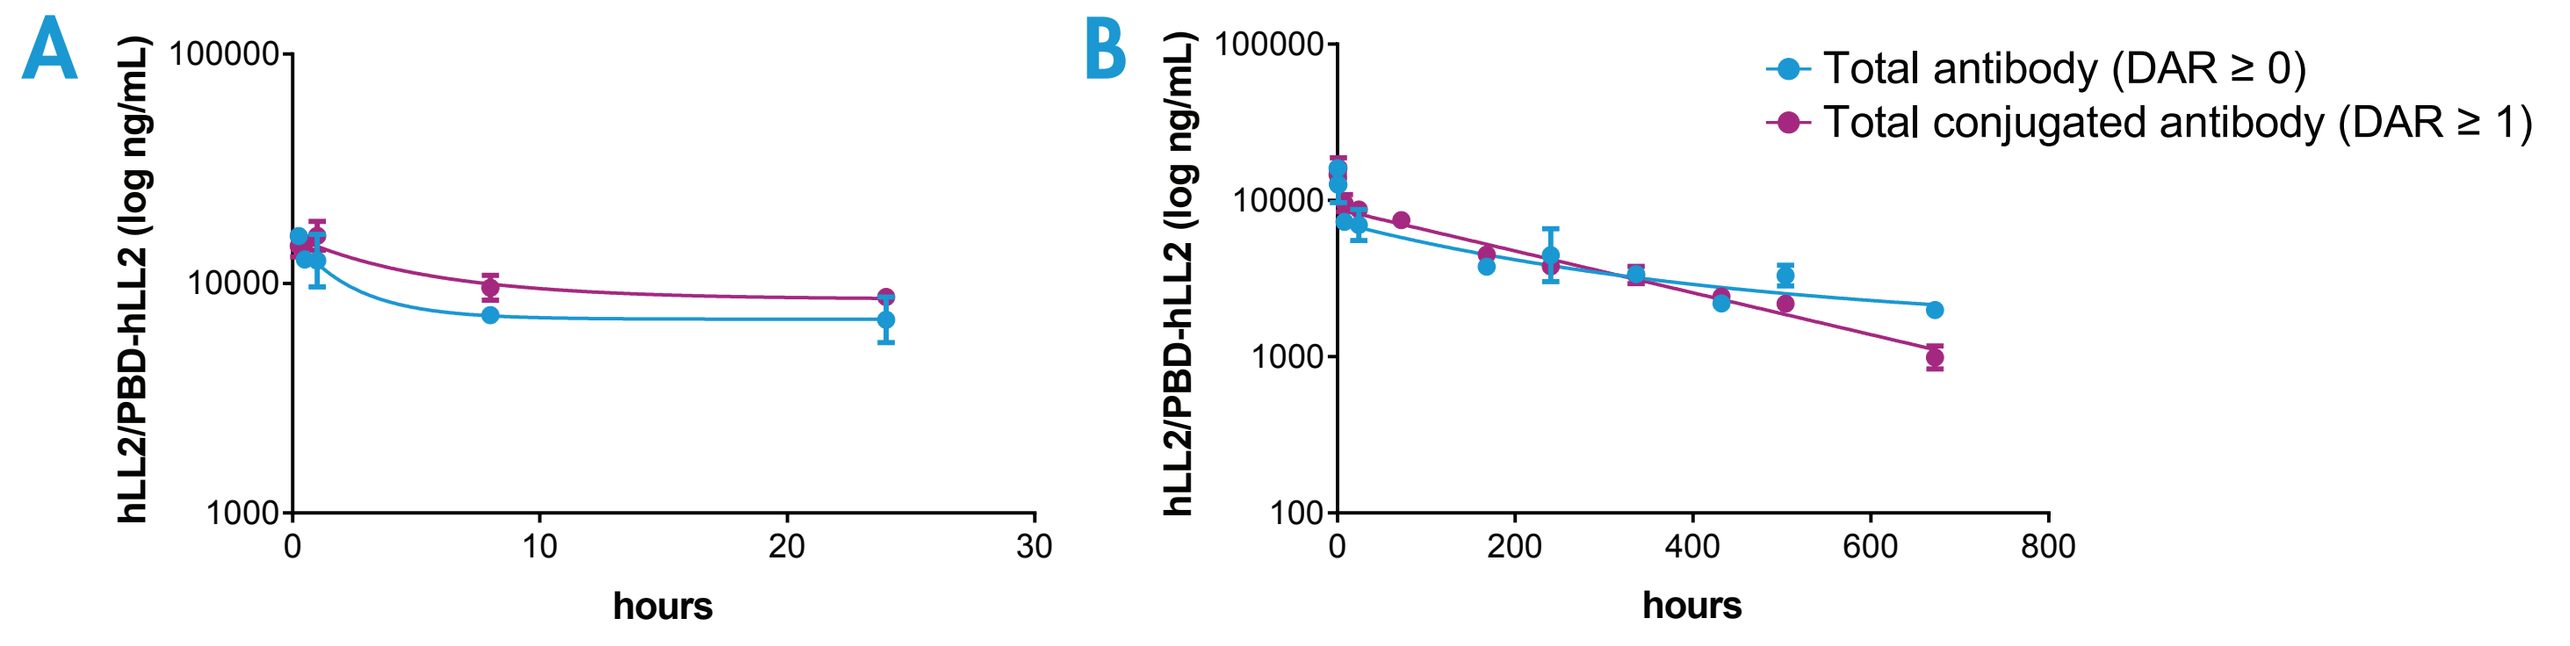
A
B
hLL2/PBD-hLL2 (log ng/mL)
hours
Total antibody (DAR ≥ 0)
Total conjugated antibody (DAR ≥ 1)

C

Total antibody PK (DAR ≥ 0) (NCA parameters)					half life	
	C_{max} (ng/mL)	T_{max} (hrs)	C_{last} (ng/mL)	T_{last} (hrs)	half life	
mean	16134.1	0.25	1996.0	672	368.1 hrs	15.3 days
Total conjugated antibody PK (DAR ≥ 1) (NCA parameters)						
	C_{max} (ng/mL)	T_{max} (hrs)	C_{last} (ng/mL)	T_{last} (hrs)	half life	
mean	16284.1	1.0	997.1	672	219.2 hrs	9.1 days

Quantification of unconjugated and PBD-conjugated mAb hLL2 in mouse serum, obtained via an anti-human IgG-Fc antibody or an anti-PBD antibody.

A. Analysis up to 24 hours. **B.** Analysis for the whole duration of the study, 672 hours. **C.** Table with PK parameters according to a non-compartmental PK analysis (NCA).

Conclusions

- **Generation of hLL2-PBD with DAR 2.5 was achieved using a simple, robust and high yielding process.**

- **hLL2-PBD and Hu10F4-vcMMAE showed comparable, potent and specific *in vitro* cytotoxicity in CD22-expressing human Burkitt's lymphoma-derived cell lines Ramos and Daudi and in human diffuse large B-cell lymphoma-derived cell line WSU-DLCL2. An isotype control ADC showed a strongly reduced *in vitro* activity against the three cell lines.**

- ***In vivo*, single-dose hLL2-PBD administration demonstrated remarkable anti-tumour efficacy in Ramos and Daudi s.c. xenografts. At equivalent doses hLL2-PBD was markedly superior to Hu10F4-vcMMAE (an anti-CD22 ADC developed by Genentech) in the Ramos model.**

- **Analysis of hLL2-PBD in non-tumour bearing mice showed a favourable PK profile, with a blood half-life of approximately 9 days.**

- **Overall, these data demonstrate the potent *in vitro* and *in vivo* anti-tumour activity of hLL2-PBD against CD22-positive hematological tumours and warrant further development of this ADC into the clinic.**

Acknowledgments

***In vivo* experiments:** Charles River Discovery Research Services.
Manufacture of hLL2-PBD: ADC Biotechnology.

References

1. Tedder, T.F., J.C. Poe, and K.M. Haas, *CD22: a multifunctional receptor that regulates B lymphocyte survival and signal transduction.* Adv Immunol, 2005. 88: p. 1-50.
2. Li, D., et al., *DCDT2980S, an anti-CD22-monomethyl auristatin E antibody-drug conjugate, is a potential treatment for non-Hodgkin lymphoma.* Mol Cancer Ther, 2013. 12(7): p. 1255-65.
3. Polson, A.G., et al., *Anti-CD22-MCC-DM1: an antibody-drug conjugate with a stable linker for the treatment of non-Hodgkin's lymphoma.* Leukemia, 2010. 24(9): p. 1566-73.
4. Raponi, S., et al., *Flow cytometric study of potential target antigens (CD19, CD20, CD22, CD33) for antibody-based immunotherapy in acute lymphoblastic leukemia: analysis of 552 cases.* Leuk Lymphoma, 2011. 52(6): p. 1098-107.
5. Du, X., et al., *Differential cellular internalization of anti-CD19 and -CD22 immunotoxins results in different cytotoxic activity.* Cancer Res, 2008. 68(15): p. 6300-5.