The anti-CD25 antibody-drug conjugate camidanlumab tesirine (ADCT-301) presents a strong preclinical activity both as single agent and in combination in lymphoma cell lines

Poster #270

Filippo Spriano [1], Chiara Tarantelli [1], Gaetanina Golino [1], Eugenio Gaudio [1], Lorenzo Scalise [1], Luciano Cascione [1], Emanuele Zucca [1,2], Patrick H. Van Berkel [3], Anastasios Stathis [2], Francesca Zammarchi [3], Francesco Bertoni [1]
[1] Università della Svizzera italiana, Institute of Oncology Research, Bellinzona, Switzerland; [2] Oncology Institute of Southern Switzerland, Bellinzona, Switzerland; [3] ADC Therapeutics (UK) Ltd., London, UK

BACKGROUND

Camidanlumab tesirine (ADCT-301) is an anti-CD25 antibody-drug conjugate (ADC) conjugated via a protease cleavable linker to SG3199, a highly cytotoxic DNA minor groove cross-linking pyrrolobenzodiazepine dimer (Flynn et al. Mol Cancer Ther 2016). ADCT-301 is currently in phase I as single agent in relapsed/refractory lymphomas (NCT02432235), advanced solid tumors (NCT03621982) and just concluded in acute myeloid leukemia (NCT02588092). We assessed its preclinical activity as single agent in 57 lymphoma cell lines and in combination with selected drugs in T cell lymphoma-derived cell lines.

Material and Methods

Cell lines were exposed to increasing concentrations of ADCT-301 for 96h followed by MTT proliferation assay. CD25 expression was measured both at cell surface level via fluorescence quantitation (Quantum Simply Cellular microspheres) and at RNA level (Illumina HT-12 arrays and HTG EdgeSeq Oncology Biomarker Panel). Combination studies of increasing doses of ADCT-301 and increasing doses of additional drugs were assessed by MTT proliferation assay over 96h in FE-PD, Karpas-299, KI-JK and MAC1 cell lines. Chou-Talalay method was used to calculate median combination index (CI) (synergism CI<0.9, additive CI=0.9-1.1, antagonism/no benefit CI> 1.1).

CONCLUSIONS

The strong single agent anti-lymphoma activity and the observed in vitro synergisms with targeted agents support the current ADCT-301 clinical development and identify potential combination partners for future clinical studies.

RESULTS

ADCT-301 presented much stronger activity in T (n=9, median IC50=4 pM; 95% C.I., 1.6pM-0.9nM) than B cell lymphomas (n=48, median IC50 = 0.7 nM; 95% C.I., 0.4-2.6 nM) (P=0.047)(Figure 1). In vitro activity was highly correlated with CD25 expression both at cell surface level (n=53, Pearson r = -0.50, P=0.0001) and RNA level (n=53, Pearson r = -0.52, P<0.0001). CD25 was also more highly expressed in T than B cell lymphoma (P<0.0001), in agreement with the IC50s differences, and the correlation was still maintained within the subgroups (T cell lymphomas, Pearson r = -0.90, P=0.0021; B cell lymphomas; Pearson r = -0.3, P=0.05) (Figure 2).
Based on the higher activity in T-cell lymphomas, ADCT-301-containing combinations were evaluated in 4 cell lines derived from peripheral T cell lymphoma not otherwise specified (n=1) (FE-PD), ALK-pos (n=2) (Karpas-299, KI-JK) or ALK-neg (n=1) (MAC1) anaplastic large cell lymphoma (ALCL) (Figure 3). ADCT-301 plus the mTOR inhibitor everolimus showed synergism in 4/4 cell lines. Combinations with the PI3K inhibitor copanlisib, the BCL2 inhibitor venetoclax and the HDAC inhibitor vorinostat were synergistic 3/4 cell lines (FE-PD, Karpas-299, MAC1 for copanlisib; Karpas-299, KI-JK, MAC1 for venetoclax; FE-PD, KI-JK, MAC1 for vorinostat). The combination with pralatrexate was synergistic in 2/2 ALK-pos ALCL cell lines. The addition of bortezomib or romidepsin led to synergism in 2/4 cell lines (FE-PD, MAC1 for bortezomib and Karpas-299, KI-JK for romidepsin). Finally, the combinations with bendamustine and with 5-azacytidine achieved synergism in 1 out 4 cell lines (ALK+ALCL Karpas-299).

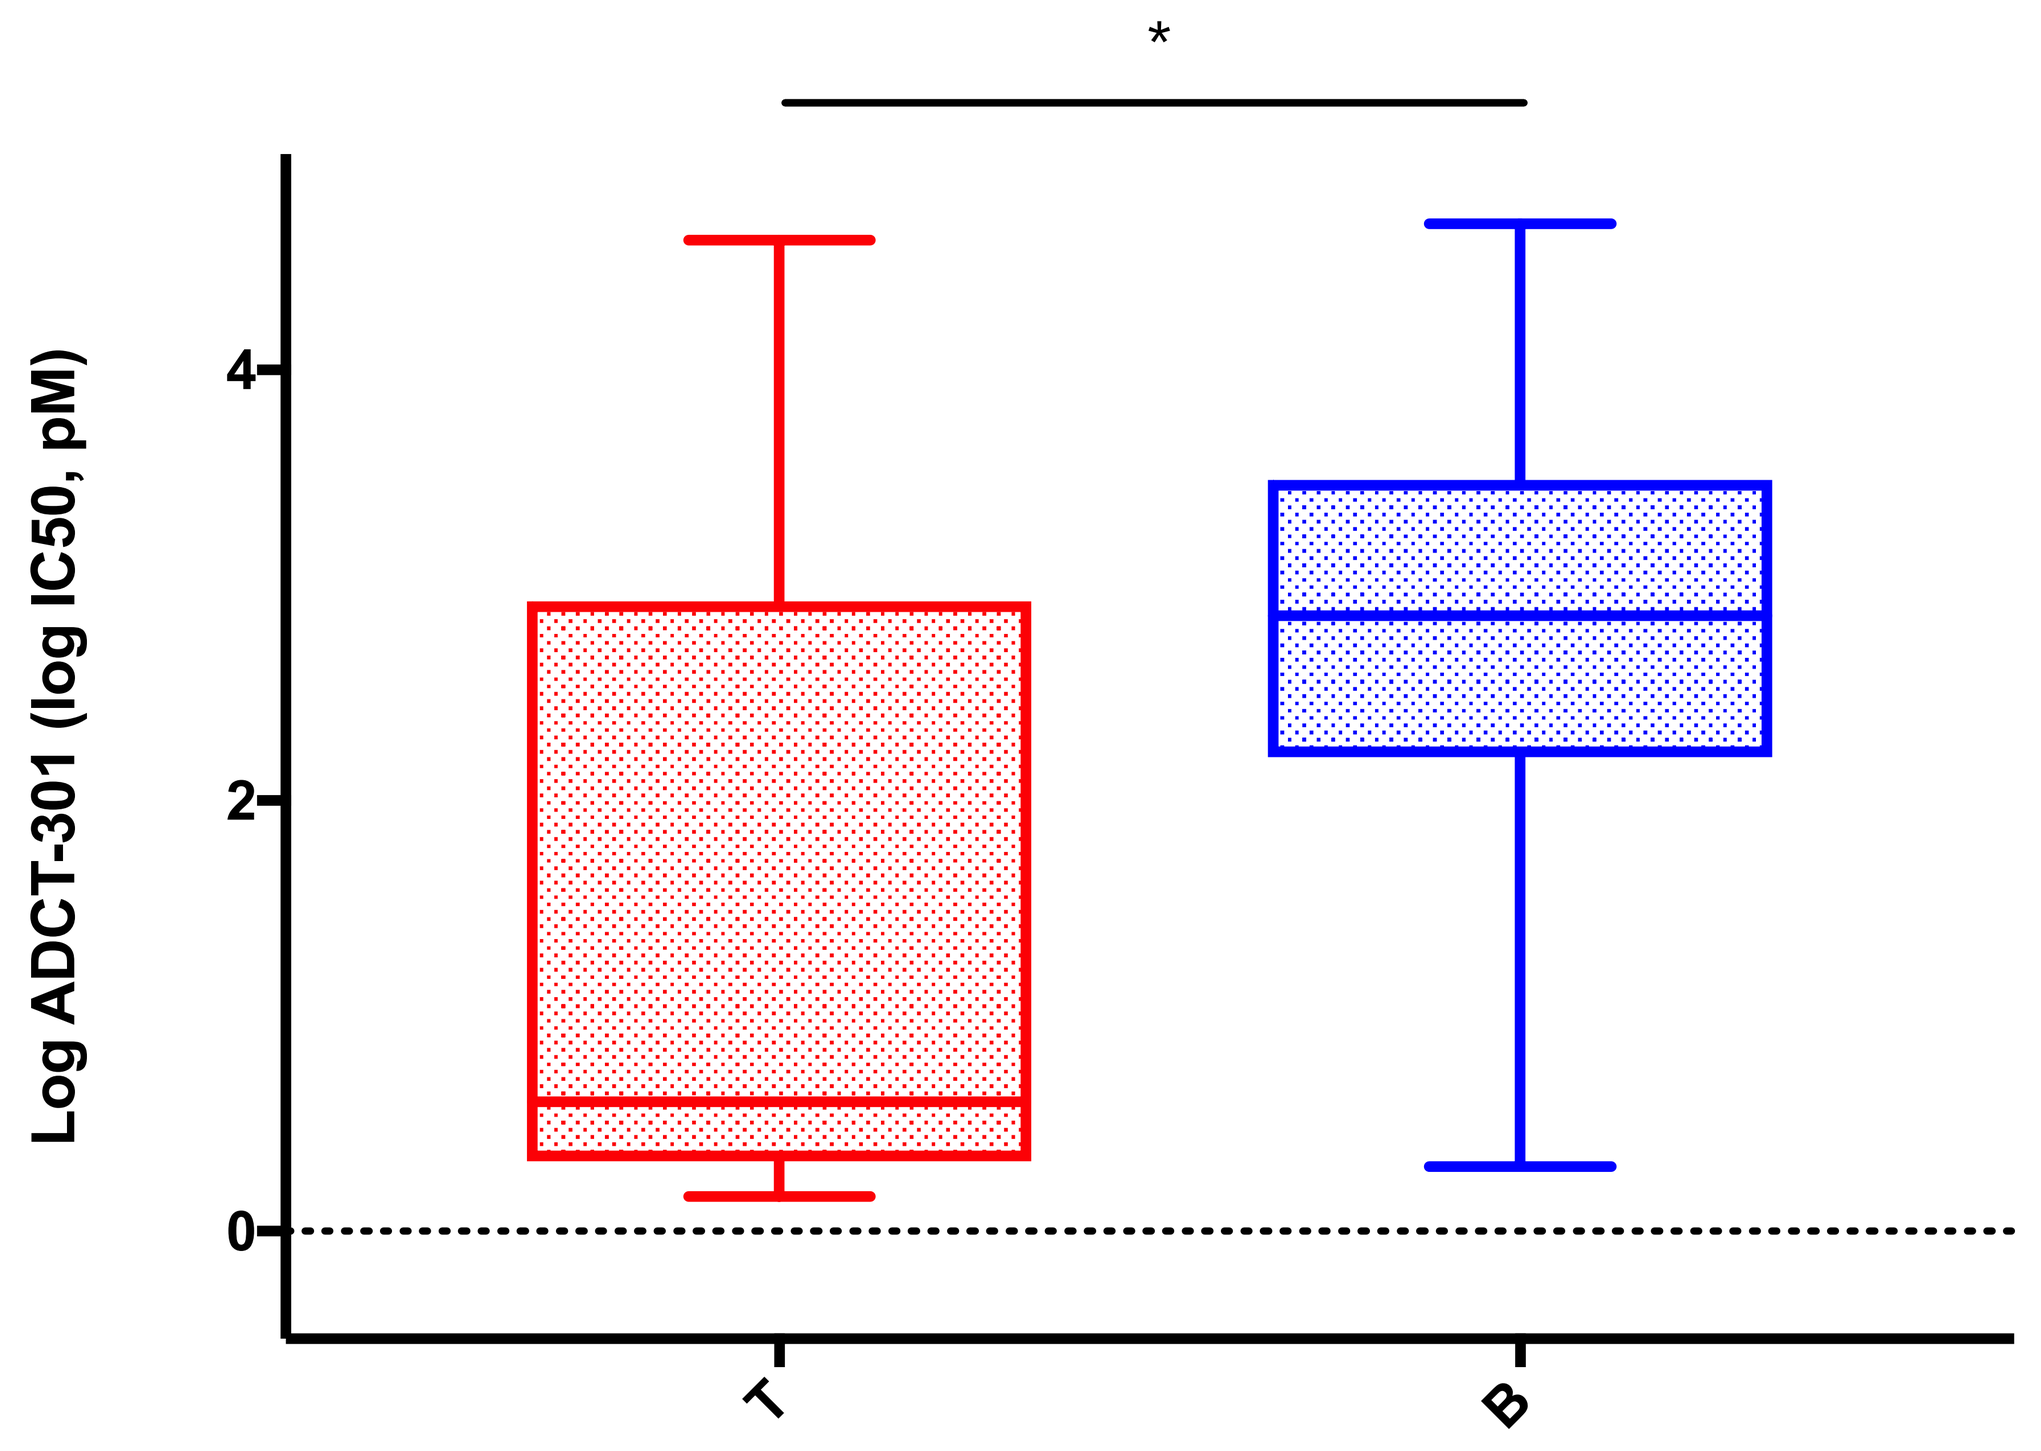
*
Log ADCT-301 (log IC50, pM)
T
B

FIGURE 1. ADCT-301 IC50 comparison between T cells and B cells. Line in the middle of the boxes represents the median and the boxes extend from 25th to 75th percentile (interquartile range, IQ); whiskers extend to the upper and lower adjacent values (± 1.5 IQ).

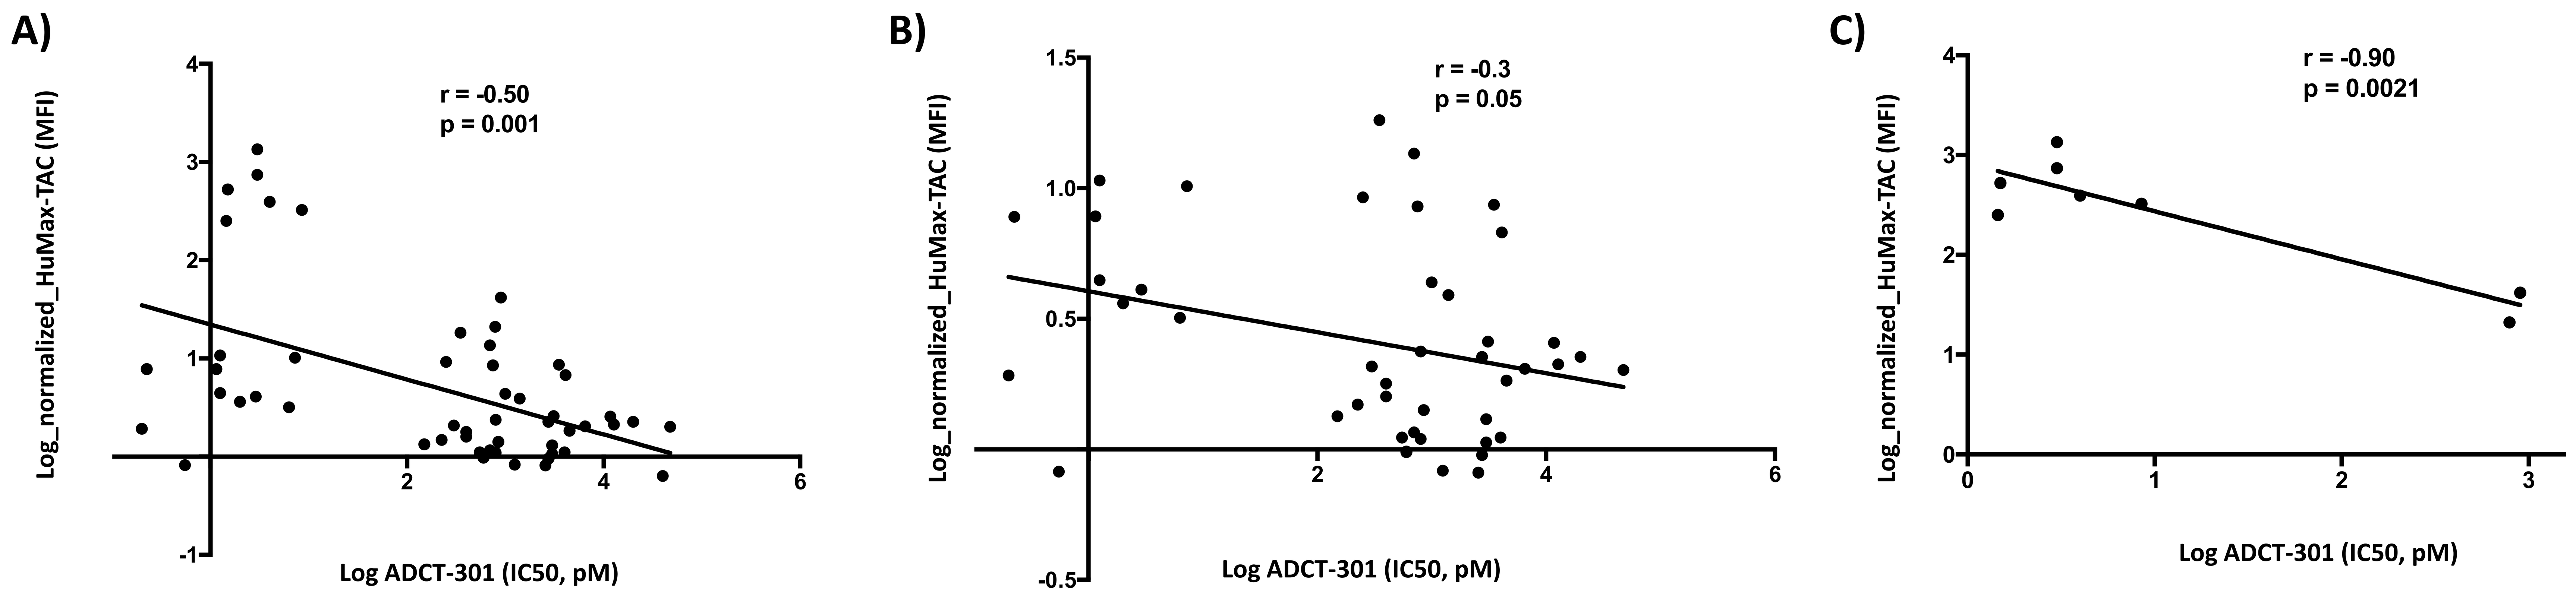
A)
r = -0.50
p = 0.001
B)
r = -0.3
p = 0.05
C)
r = -0.90
p = 0.0021
Log_normalized_HuMax-TAC (MFI)
Log ADCT-301 (IC50, pM)

FIGURE 2. Pearson correlation between CD25 surface expression and ADCT-301 IC50. A) correlation among all cell lines; B) correlation in B-cell lymphomas only; C) correlation in T-cell lymphomas. HuMax-TAC, recombinant Human monoclonal antibody binding to Human CD25; MFI, mean fluorescence intensity.

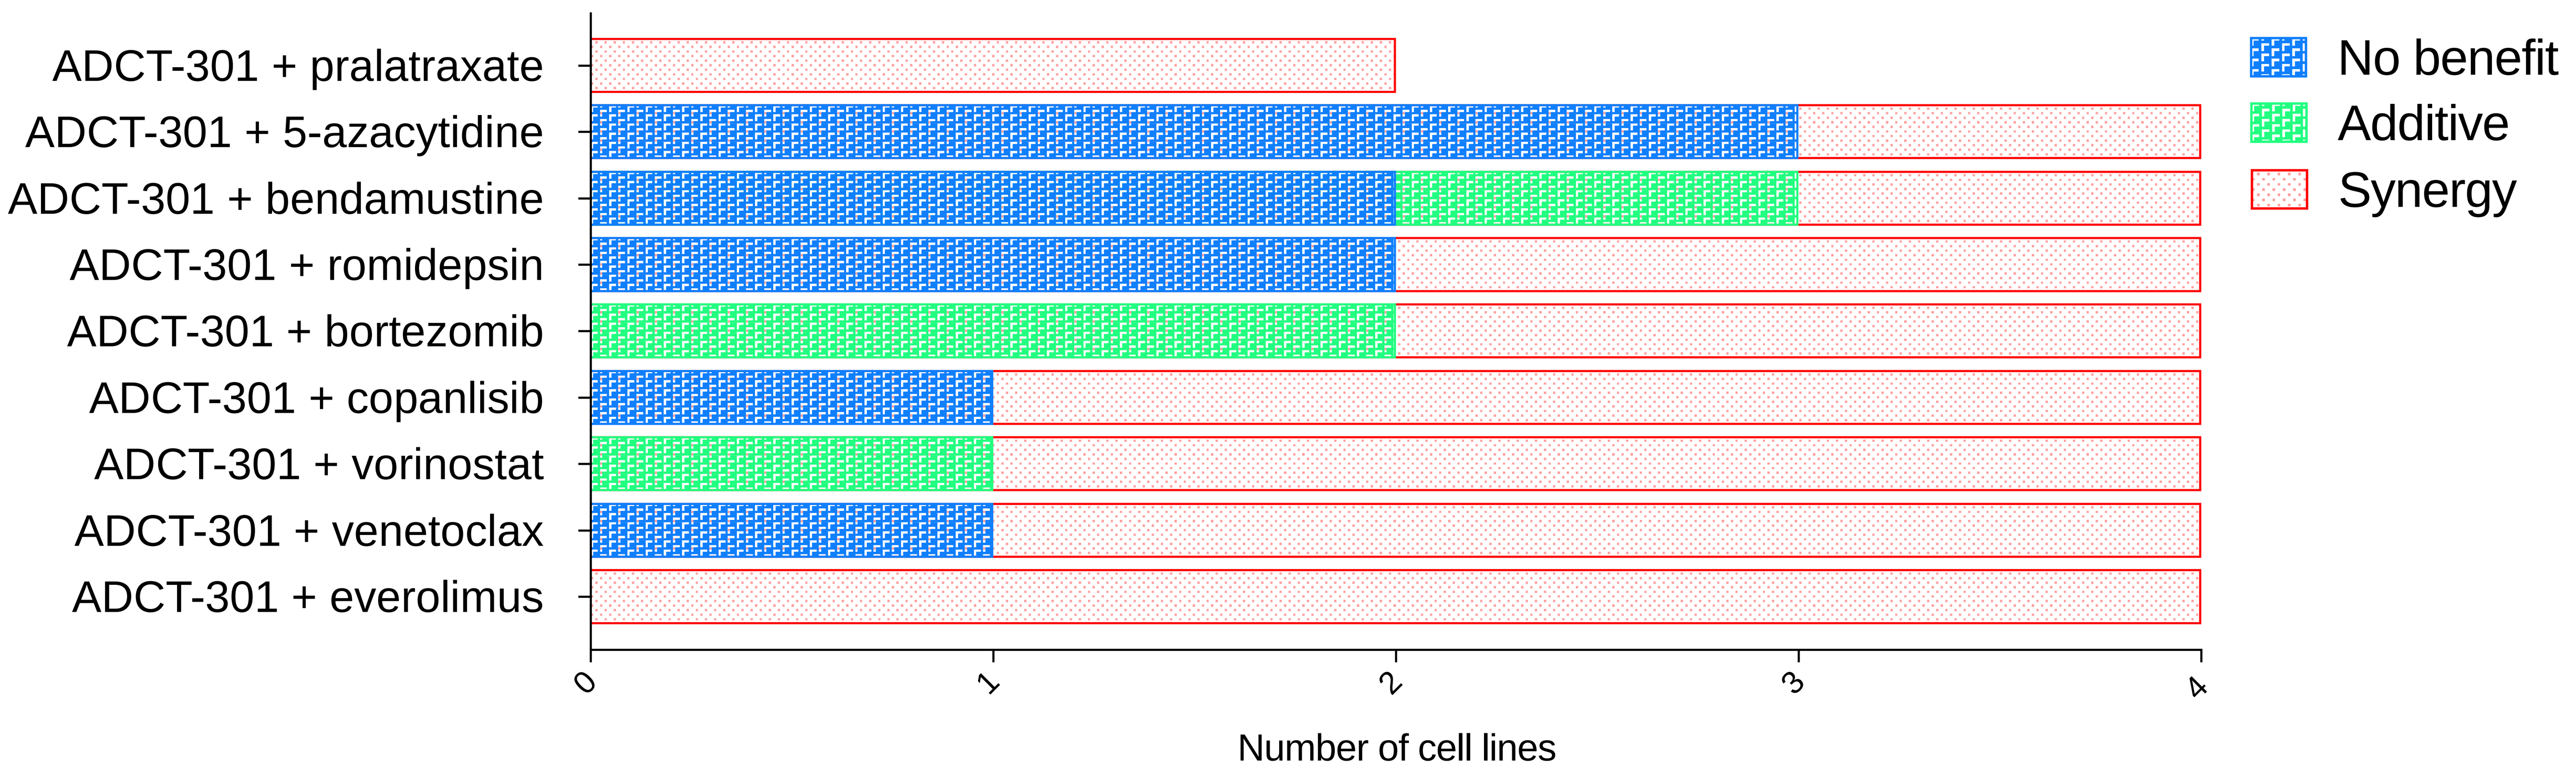
ADCT-301 + pralatraxate
ADCT-301 + 5-azacytidine
ADCT-301 + bendamustine
ADCT-301 + romidepsin
ADCT-301 + bortezomib
ADCT-301 + copanlisib
ADCT-301 + vorinostat
ADCT-301 + venetoclax
ADCT-301 + everolimus
Number of cell lines
No benefit
Additive
Synergy

FIGURE 3. ADCT-301 containing combinations in 4 T cell lymphomas. Synergism, additive effect and no benefit were defined using the Chou-Talalay Combination Index (CI). Synergism, CI < 0.9; additive effect, 0.9 < CI < 1.1; no benefit, CI >1.1.

Contact: Francesco Bertoni, e-mail: francesco.bertoni@ior.usi.ch