Analysis of ADCT-602 pre-clinical activity in B-cell lymphoma models and identification of potential biomarkers for its activity

Eugenio Gaudio [1], Chiara Tarantelli [1], Luciano Cascione [1], Filippo Spriano [1], Gaetanina Golino [1], Lorenzo Scalise [1], Emanuele Zucca [1,2], Anastasios Stathis [2], Patrick H. Van Berkel [3], Francesca Zammarchi [3], Francesco Bertoni [1]

[1] Institute of Oncology Research, Faculty of Biomedical Sciences, USI, Bellinzona, Switzerland ; [2] Oncology Institute of Southern Switzerland, Bellinzona, Switzerland; [3] ADC Therapeutics, London, United Kingdom.

Introduction

CD22 is a cell surface marker expressed by the vast majority of normal and neoplastic B-cells. ADCT-602 is an antibody drug conjugate (ADC) composed of Emab-C220, an engineered version of the anti-CD22 humanized IgG1 antibody epratuzumab, site-specifically conjugated to SG3249, which includes the DNA minor groove crosslinking pyrrolobenzodiazepine (PBD) dimer SG3199 linked to the antibody via a protease-cleavable linker (Zammarchi et al, ASH 2016). ADCT-602 is currently being tested in a phase I/II clinical trial (NCT03698552) in recurrent or refractory B-cell acute lymphoblastic leukemia (B-ALL) patients. Here, we assessed its *in vitro* anti-lymphoma activity, also exploring for potential biomarkers and mechanisms of resistance.

Material and Methods

Fifty-seven human lymphoma cell lines were exposed to ADCT-602, an isotype-control ADC and the PBD dimer SG3199 as single agents for 96h, followed by MTT proliferation assay and IC50 calculation. Quantum Simply Cellular (QSC) microspheres were used for the quantitative determination of cellular CD22 antigen expression (Bangs Laboratories. Inc). Differences in IC50 values among lymphoma subtypes were calculated using the Wilcoxon rank-sum test. Statistical significance was defined by P values of 0.05 or less. Sensitivity analysis to ADCT-602 was performed by integrating different omics data, such as Illumina HT-12 microarray data (GSE94669), HTG EdgeSeq Oncology Biomarker Panel data (GSE103934) and DNA copy number variations.

Conclusions

ADCT-602 showed *in vitro* anti-tumor activity across a large panel of B-cell lymphoma models of various histology. Expression signatures and other features (MZL or DHT DLBCL histology), but not the expression levels of its target, were associated with different sensitivity to the ADC. Our data supports the clinical evaluation of ADCT-602 in patients with B-cell lymphoma in addition to B-ALL.

Contact *Francesco Bertoni, MD, Institute of Oncology Research, via Vela 6, 6500 Bellinzona, Switzerland; phone: +41 91 8200 367; e-mail: frbertoni@mac.com*

Results

The median IC50 for ADCT-602 was 200 pM (95%C.I, 90-400 pM) in 48 B-cell lymphoma lines (including three Hodgkin lymphoma cell lines), and 1850 pM in nine T-cell lymphoma lines (95%C.I, 700-15000 pM) (**Figure 1**). ADCT-602 was more active in B- than in T-cell lymphomas, as expected based on the pattern of CD22 expression ($P < 0.005$). Focusing on B-cell lymphomas, ADCT-602 *in vitro* activity was not correlated with its target expression measured both at the cell surface protein level (absolute quantitation, n=48, r=0.06 P=ns) and at the RNA level (Illumina HT-12 arrays, n=42, r=0.28, P=ns; HTG EdgeSeq Oncology Biomarker Panel, n=36, r=0.24, P=ns) (**Figures 2-3**). *In vitro* activity was stronger in marginal zone lymphoma (MZL) cell lines than other B-cell lymphoma models (median IC50s 62.5 vs 312.5 pM; P = 0.03) as well as in diffuse large B-cell lymphoma (DLBCL) cell lines with BCL2 and MYC translocations (DHT DLBCL) versus DLBCL with none or a single translocation (median IC50s 25 vs 400 pM, P = 0.03). No associations were seen with TP53 status or other histology (mantle cell lymphoma, DLBCL, DLBCL cell of origin).

We then exploited the gene expression profiling data using the Illumina HT-12 microarray gene expression technology. Within all the B-cell lymphoma cell lines (sensitive, n= 25; resistant, n= 23) we identified 1.207 genes down-regulated (FC-) and 1,104 genes up-regulated (FC+) in resistant cell lines. To delineate the pathways associated with the different degrees of sensitivity to ADCT-602, we performed a gene set enrichment analysis (GSEA; GSEA hallmarks and c2.common pathways) on the pre-ranked limma data. Transcripts up-regulated in resistant cell lines were enriched of genes coding for proteins involved in respiratory electron transport, oxidative phosphorylation and proteasome (**Figure 4**). Conversely, transcripts up-regulated in the sensitive cell lines were enriched of genes coding for proteins involved in inflammation, chemokine signaling, p53 response, IL2/STAT5 signaling, hypoxia, TGF-beta and interferon response (**Figure 5**). Similar gene signatures were picked up using the HTG platform, which can be applied to formalin-fixed paraffin-embedded clinical specimens, despite the smaller number of investigated genes.

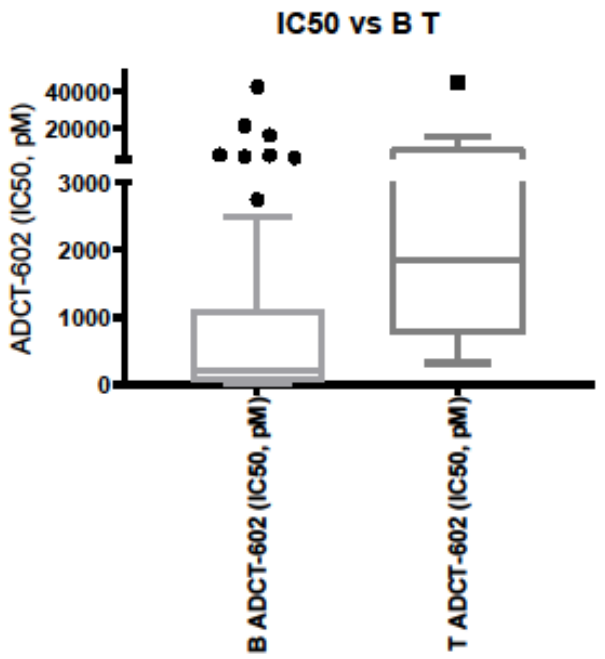
IC50 vs B T
ADCT-602 (IC50, pM)
40000
20000
3000
2000
1000
0
B ADCT-602 (IC50, pM)
T ADCT-602 (IC50, pM)

Figure 1. Distribution of ADCT-602 IC50 values in lymphoma cell lines based on their B- or T-cell origin. MTT proliferation assay and IC50 calculation on cell lines exposed (96h) to increasing ADCT-602 concentrations. The line in the middle of the box represents the median and the box extends from the 25th to the 75th percentile (interquartile range, IQ); the whiskers extend to the upper and lower adjacent values (i.e., 15 IQ). P<0.005.

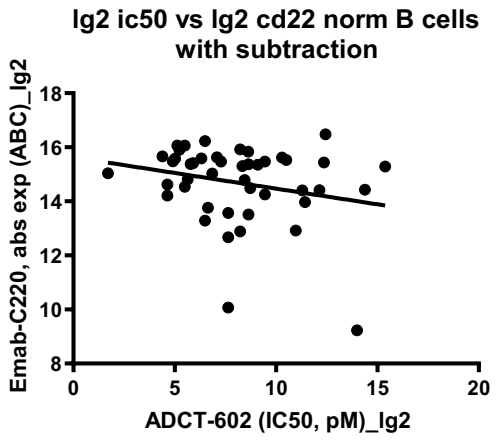
lg2 ic50 vs lg2 cd22 norm B cells with subtraction
Emab-C220, abs exp (ABC)_lg2
ADCT-602 (IC50, pM)_lg2

Figure 2. ADCT-602 *in vitro* activity does not correlate with CD22 surface protein expression in B-cell lymphoma cell lines. Graph shows IC50 values and absolute fluorescence quantitation with Quantum Simply Cellular microspheres. P > 0.05, r = -0.24.

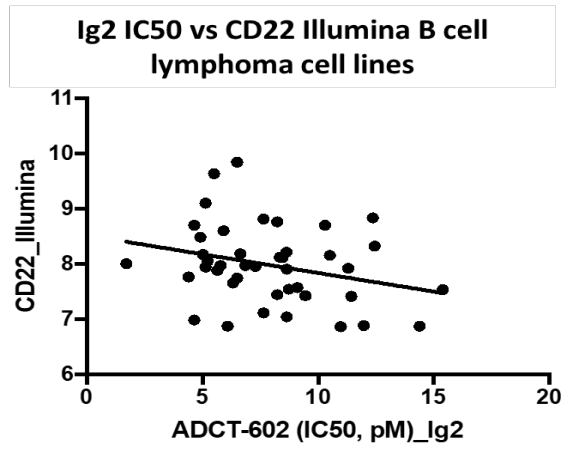
Ig2 IC50 vs CD22 Illumina B cell lymphoma cell lines
CD22_Illumina
ADCT-602 (IC50, pM)_lg2

Figure 3. ADCT-602 *in vitro* activity does not correlate with CD22 RNA expression as measured using microarrays in B-cell lymphoma cell lines. Graph shows IC50 values and CD22 RNA expression levels measured via microarray (Illumina). P > 0.05, r = -0.28.

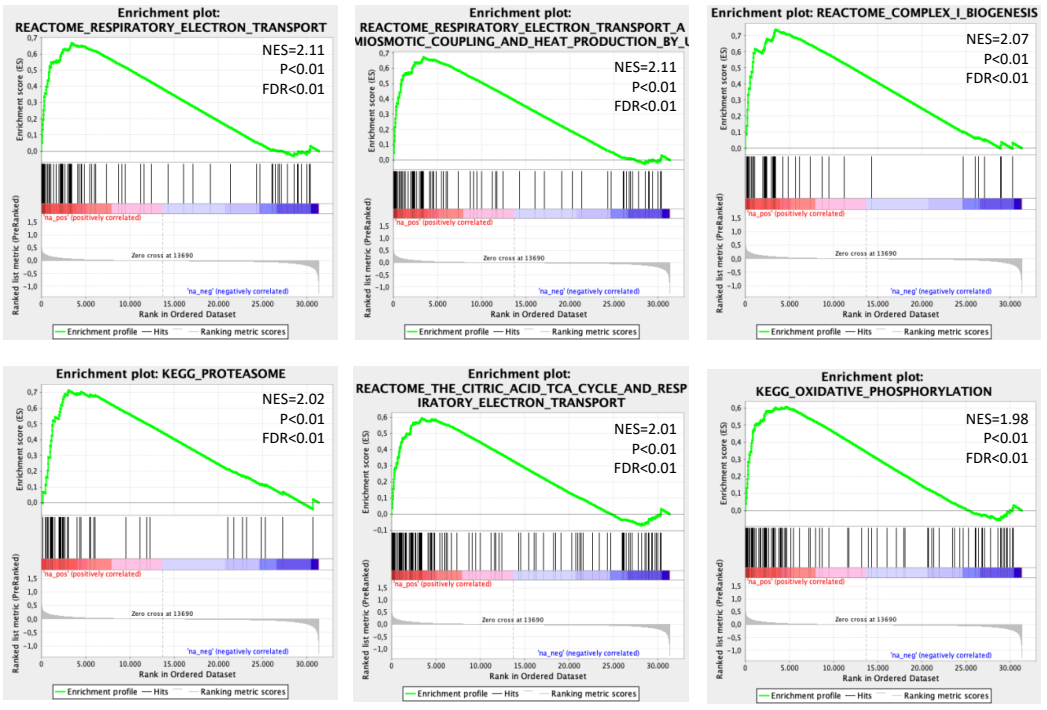
Enrichment plot: REACTOME_RESPIRATORY_ELECTRON_TRANSPORT
NES=2.11
P<0.01
FDR<0.01
Enrichment plot: REACTOME_RESPIRATORY_ELECTRON_TRANSPORT_A MIOSMOTIC_COUPLING_AND_HEAT_PRODUCTION_BY_U
NES=2.11
P<0.01
FDR<0.01
Enrichment plot: REACTOME_COMPLEX_I_BIOGENESIS
NES=2.07
P<0.01
FDR<0.01
Enrichment plot: KEGG_PROTEASOME
NES=2.02
P<0.01
FDR<0.01
Enrichment plot: REACTOME_THE_CITRIC_ACID_TCA_CYCLE_AND_RESPIRATORY_ELECTRON_TRANSPORT
NES=2.01
P<0.01
FDR<0.01
Enrichment plot: KEGG_OXIDATIVE_PHOSPHORYLATION
NES=1.98
P<0.01
FDR<0.01

Figure 4. Gene expression signatures associated with resistance to ADCT-602 in B-cell lymphomas. Representative gene set enrichment analysis (GSEA) plots.

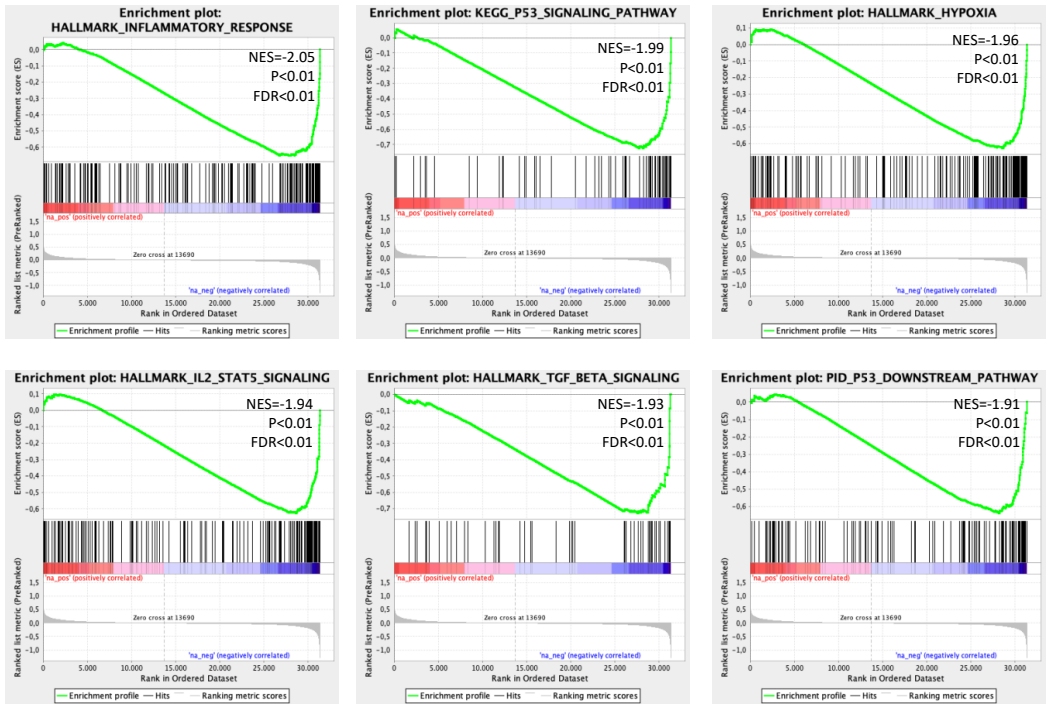
Enrichment plot: HALLMARK_INFLAMMATORY_RESPONSE
NES=-2.05
P<0.01
FDR<0.01
Enrichment plot: KEGG_P53_SIGNALING_PATHWAY
NES=-1.99
P<0.01
FDR<0.01
Enrichment plot: HALLMARK_HYPOXIA
NES=-1.96
P<0.01
FDR<0.01
Enrichment plot: HALLMARK_IL2_STAT5_SIGNALING
NES=-1.94
P<0.01
FDR<0.01
Enrichment plot: HALLMARK_TGF_BETA_SIGNALING
NES=-1.93
P<0.01
FDR<0.01
Enrichment plot: PID_P53_DOWNSTREAM_PATHWAY
NES=-1.91
P<0.01
FDR<0.01

Figure 5. Gene expression signatures associated with sensitivity to ADCT-602 in B-cell lymphomas. Representative gene set enrichment analysis (GSEA) plots.