AXL as a therapeutic target in adenoid cystic carcinoma: preclinical evaluation of AXL targeting antibody-drug conjugate (ADCT-601)

UCSF Helen Diller Family Comprehensive Cancer Center

Adenoid Cystic Carcinoma Research Foundation

Joseph O. Humtsoe[1]; Leilani Jones[1]; Shorook Naara[1]; Francesca Zammarchi[2], Nicole Spardy Burr[3], Patrick H. Van Berkel[2], Patrick K. Ha[1], Hyunseok Kang[1].

University of California San Francisco, San Francisco, CA[1], ADC Therapeutics UK (Ltd), London, UK[2], Adenoid Cystic Carcinoma Research Foundation, Boston, MA, USA[3].

Abstract # LB084

Background

- Adenoid cystic carcinoma (ACC) is a rare cancer of salivary gland cancer; with a significant unmet clinical need.
- Conventional systemic therapy has limited efficacy; no FDA approved drugs for patients with metastatic disease.
- Previous studies with proteomic analysis identified AXL as a potential therapeutic target in ACC[1,2].
- AXL, a member of the TAM tyrosine kinase receptor family has been generally associated with poor prognosis; overexpression and activation is implicated in conferring resistance to conventional systemic therapies in solid tumors.
- Our recent work showed that AXL cross-talks with splice variants of FGFR1 and confers resistance to selected FGFR inhibitors in ACC[3].

Objective

- ADCT-601 (mipasetamab uzoptirine) is an antibody-drug conjugate targeting human AXL carrying **a** novel pyrrolobenzodiazepine dimer cytotoxin and cleavable linker as the payload[4].
- The goal of this study is to evaluate AXL as a therapeutic target in ACC using ADCT-601 in pre-clinical models.

Methods and Materials

- ACC-01 and HACC-2A cell lines were obtained from the University of Texas MD-Anderson Cancer Center, and University of Michigan, respectively. ACCX were from University of Virginia, and Adenoid Cystic Carcinoma Research Foundation (ACCRF).
- AXL expression in human ACC patient tumor microarray (TMA) was evaluated by immunohistochemistry (IHC) using monoclonal AXL antibody (C89E7, Cell Signaling Technology).
- In vitro, cells treated with saline vehicle, isotype control IgG-ADC or ADCT-601 were analyzed by MTT-based cytotoxic assay and immunoblotting. AXL was detected using human AXL antibody (AF154; R&D Systems).
- In vivo, anti-tumorigenicity was assessed in two ACC patient derived xenograft (PDX) models. A single dose of vehicle, control IgG-ADC or ADCT-601 was administered intravenously.

Results

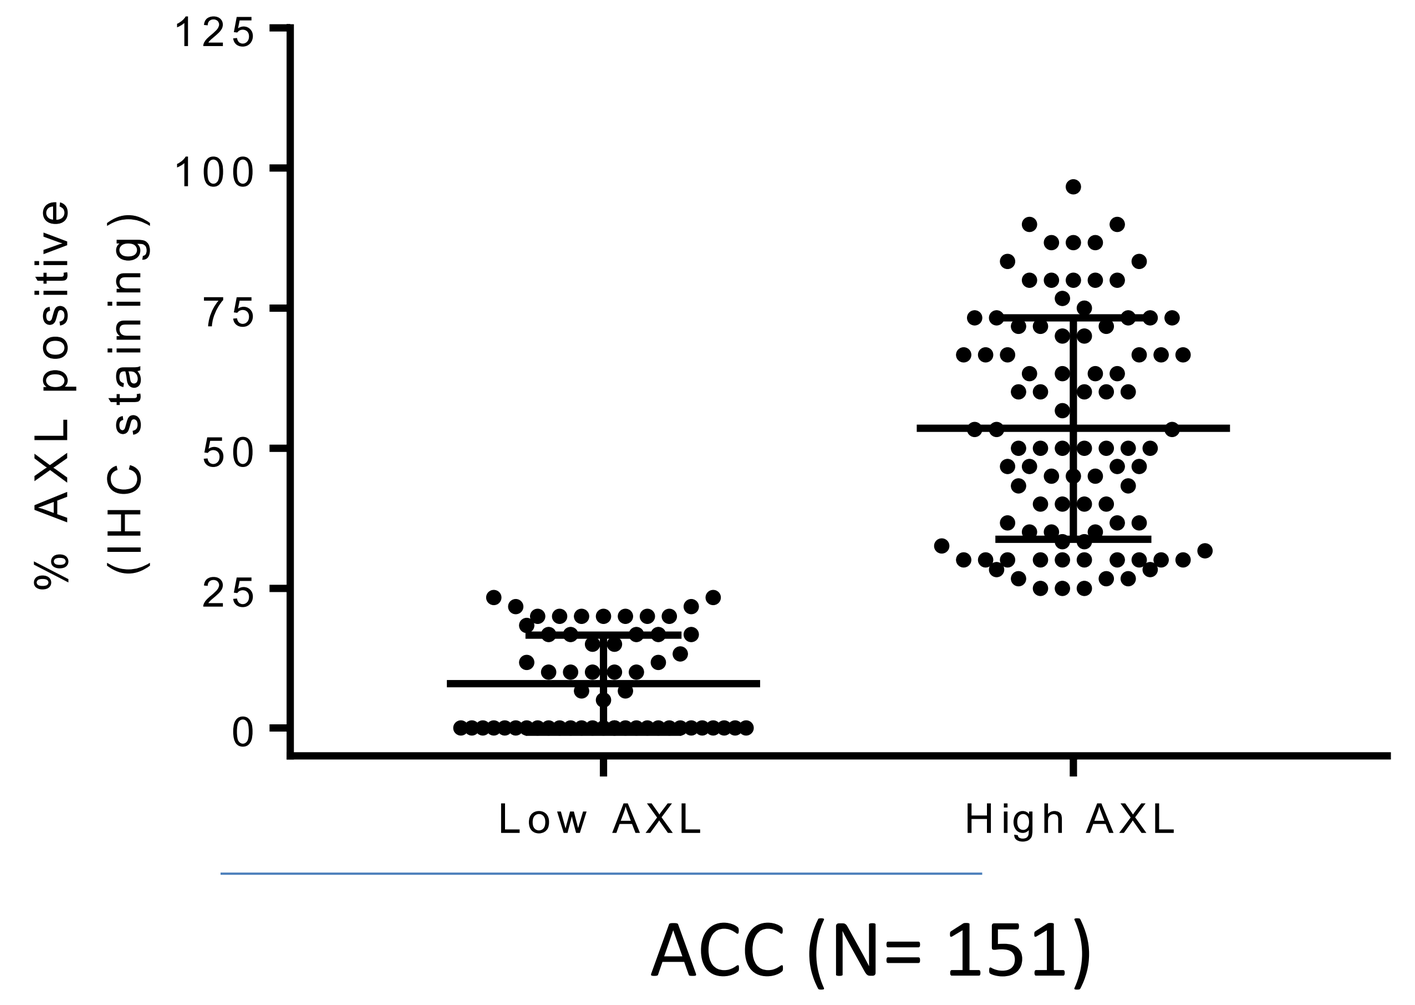
% AXL positive (IHC staining)
Low AXL
High AXL
ACC (N= 151)

Figure 1. AXL expression in tumors from ACC patients. High AXL expression defined by >25% cells expressing AXL was observed in 60 % of patient samples in TMA.

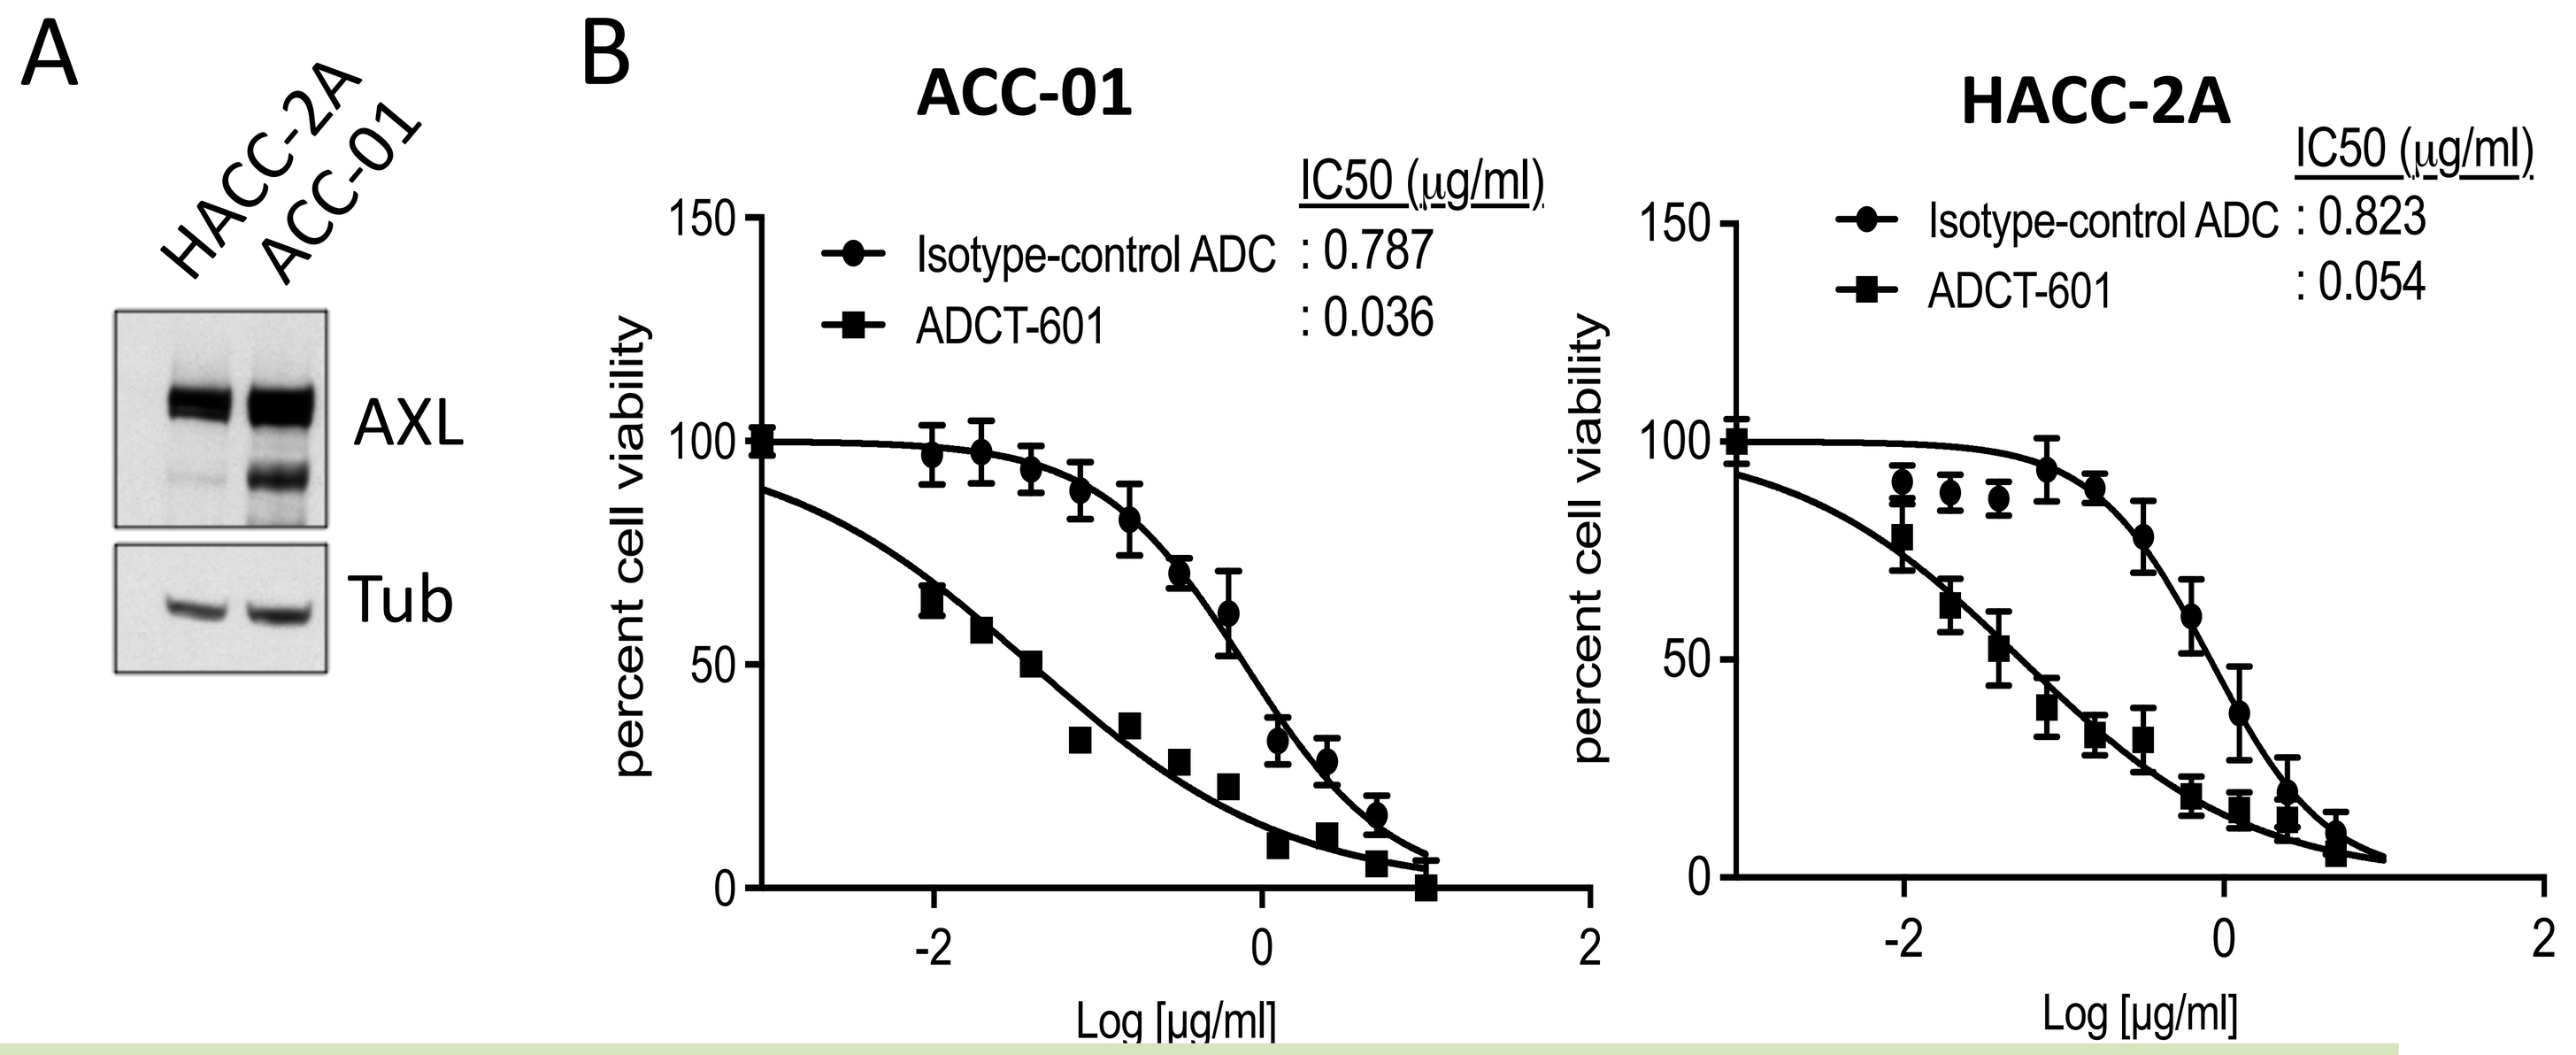
A
HACC-2A
ACC-01
AXL
Tub
B
ACC-01
IC50 (µg/ml)
Isotype-control ADC : 0.787
ADCT-601 : 0.036
HACC-2A
IC50 (µg/ml)
Isotype-control ADC : 0.823
ADCT-601 : 0.054
percent cell viability
Log [µg/ml]

Figure 2. Cytotoxic effect of ADCT-601 in ACC cells. (**A**). Western blot analysis showing AXL expression in the two ACC cell lines. (**B**). Cells were treated with control IgG-ADC or ADCT-601. Cell viability was then measured after 5 days. Data is representative of three independent experiments.

Figure 3. ADCT-601 induces DNA damage and apoptosis in ACC cell lines. Cells were treated with increasing doses of ADCT-601 for 48 hr. Cell lysates were prepared and induction of key apoptotic markers were analyzed by Western blot as indicated.

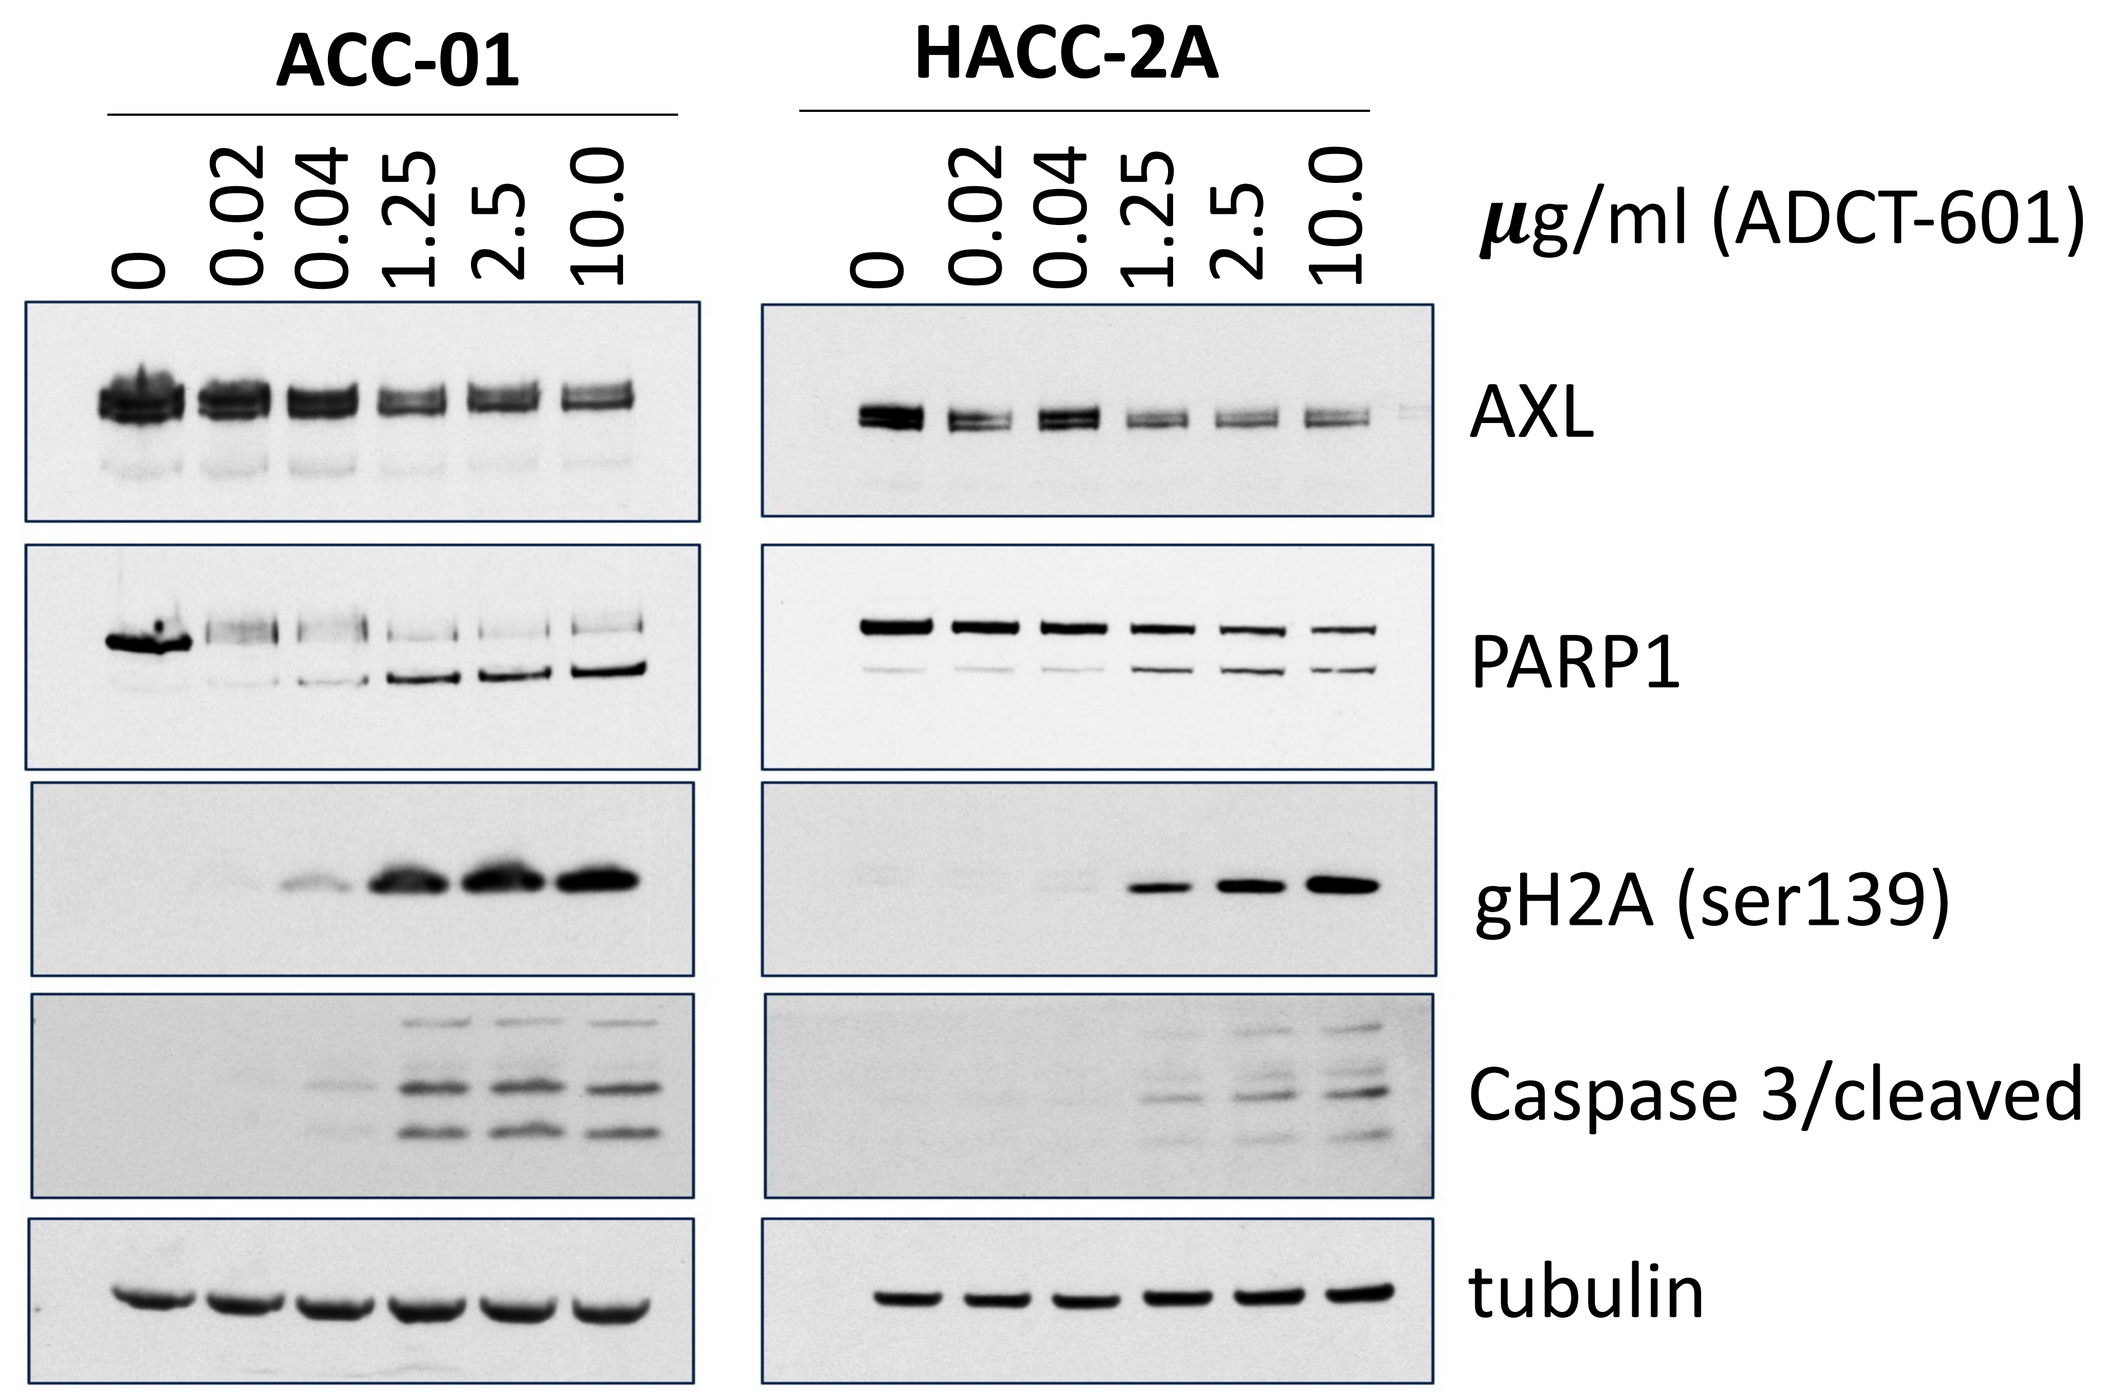
ACC-01
HACC-2A
0 0.02 0.04 1.25 2.5 10.0
µg/ml (ADCT-601)
AXL
PARP1
gH2A (ser139)
Caspase 3/cleaved
tubulin

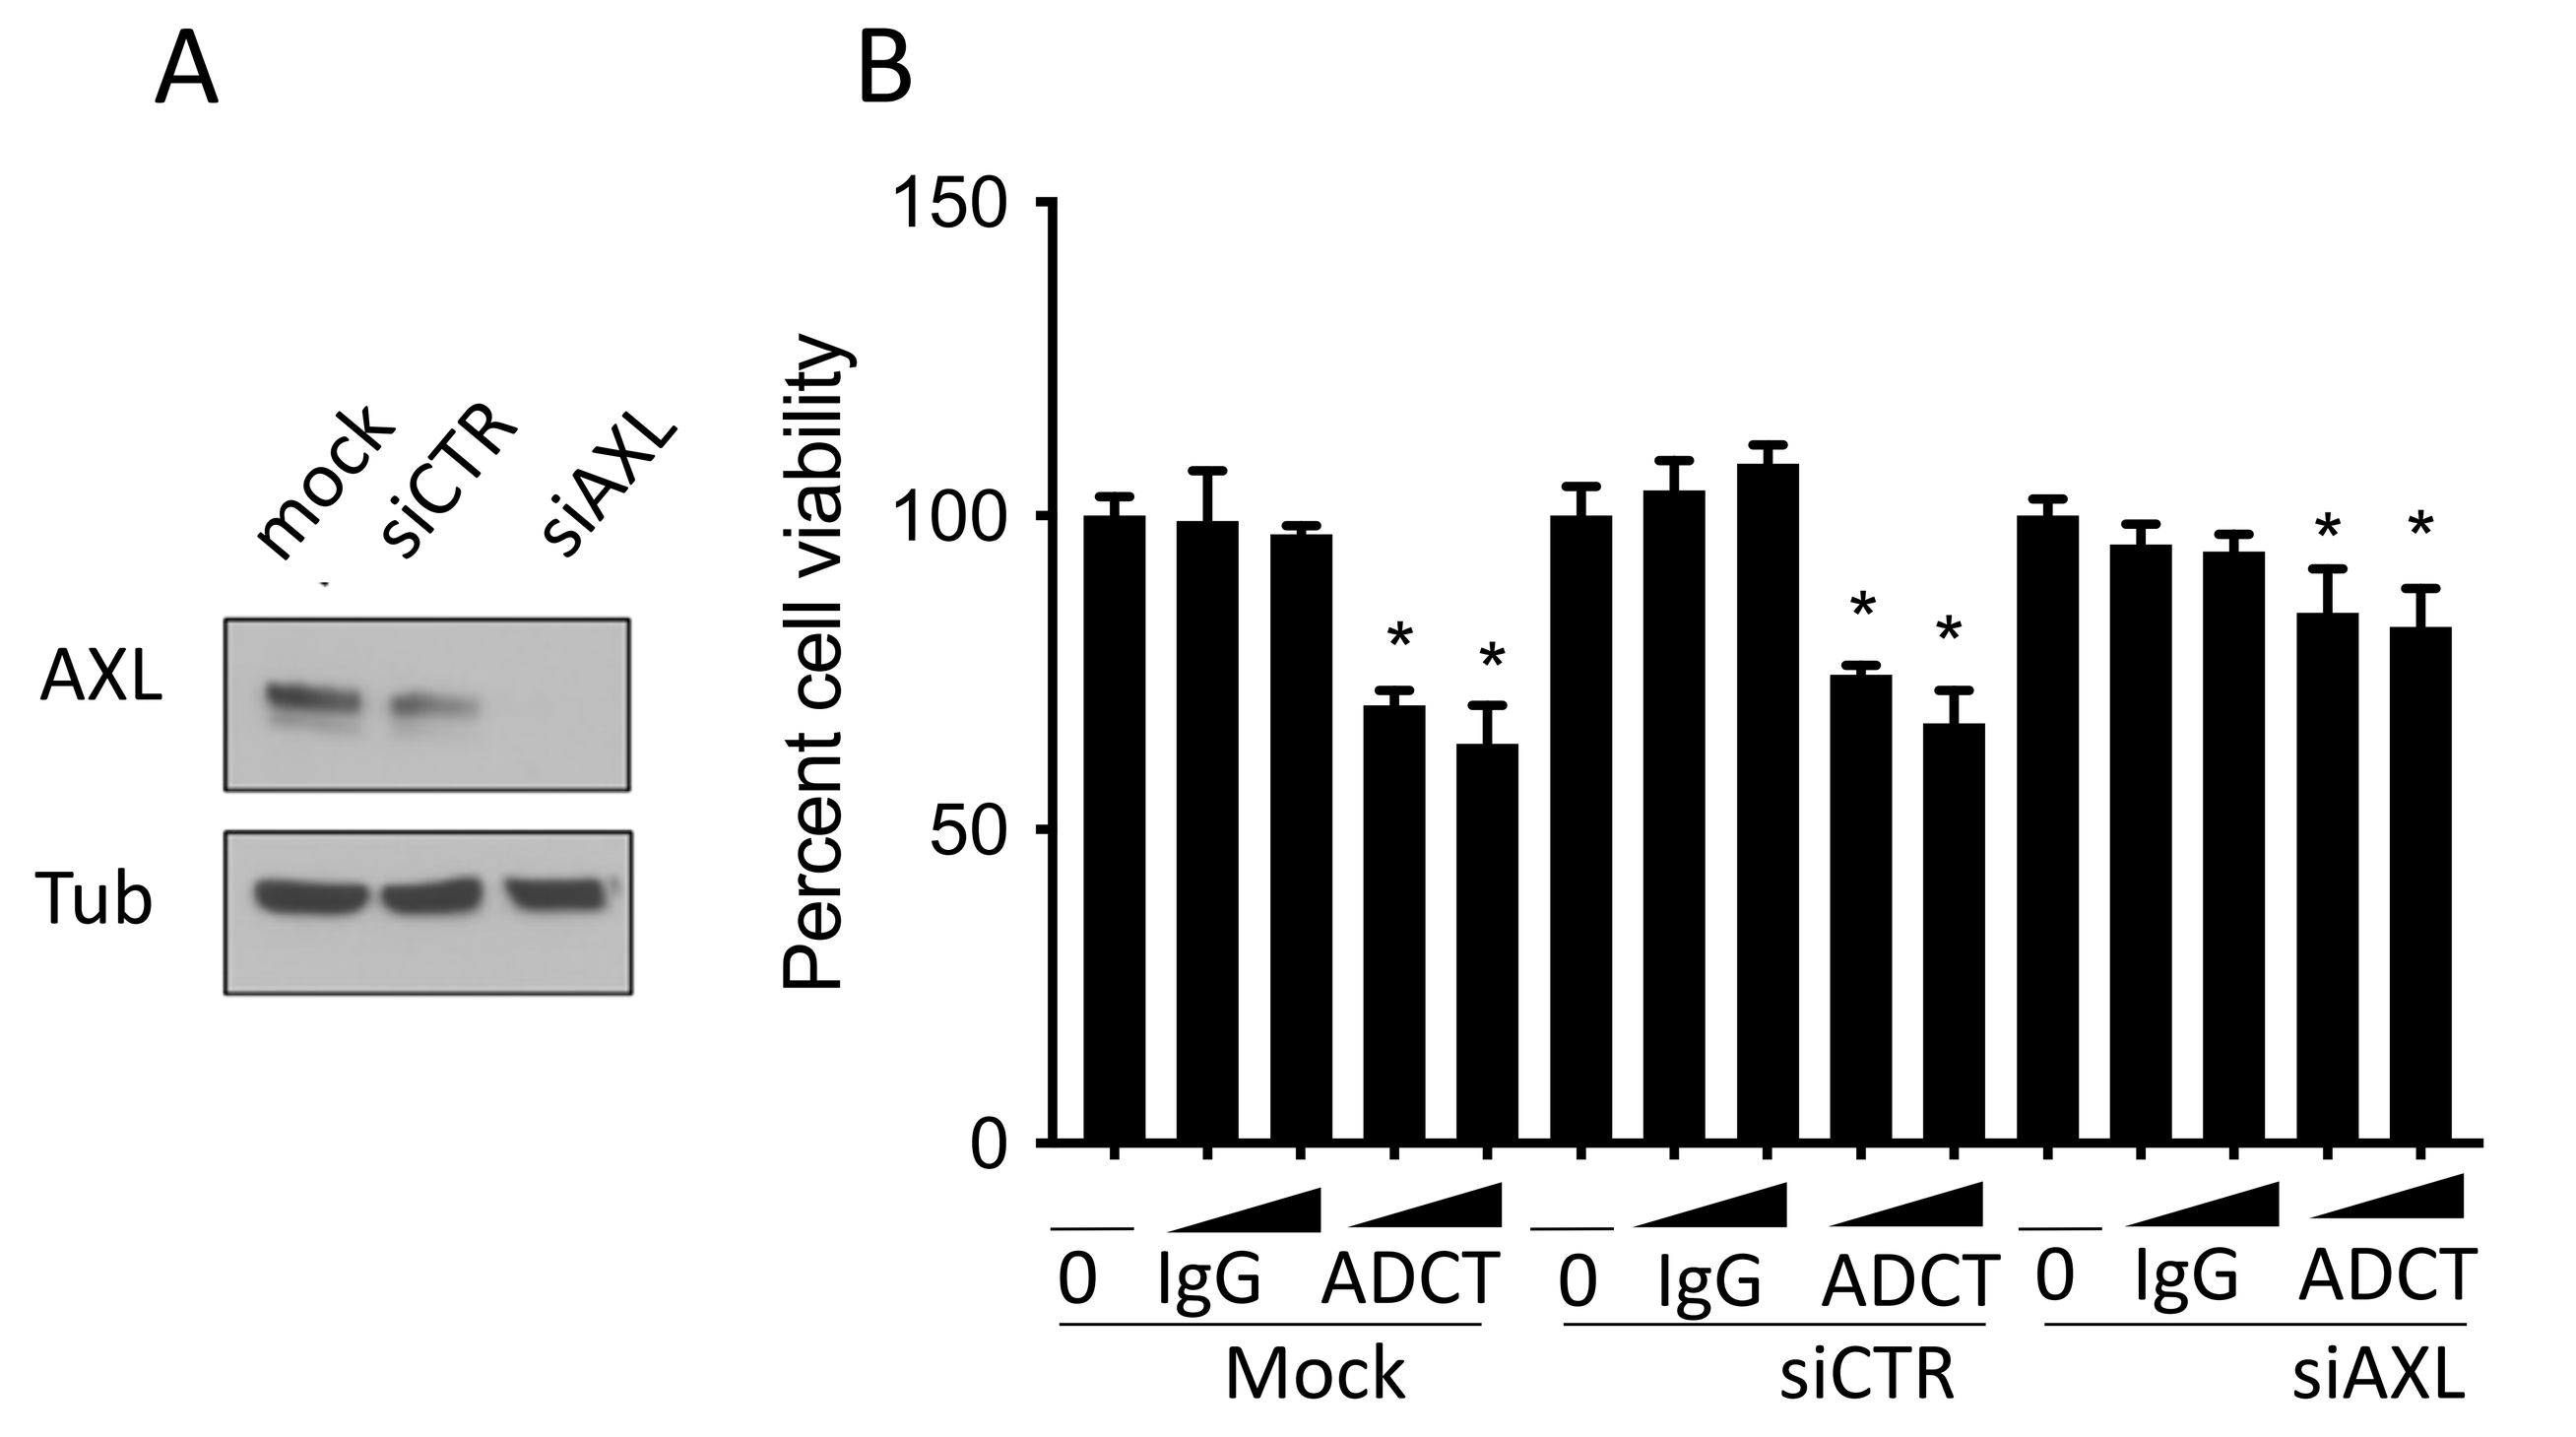
A
mock
siCTR
siAXL
AXL
Tub
B
Percent cell viability
0 IgG ADCT
Mock
siCTR
siAXL

Figure 4. AXL knockdown using siRNA attenuates cytotoxic effect of ADCT-601 in ACC cells. (**A**). Western blot analysis to verify knockdown of AXL. (**B**). Transfected cells were treated with 100 and 200 ng/ml of control Ig-ADC or ADCT-601, and cell viability was measured after 72 hr. * p<0.001.

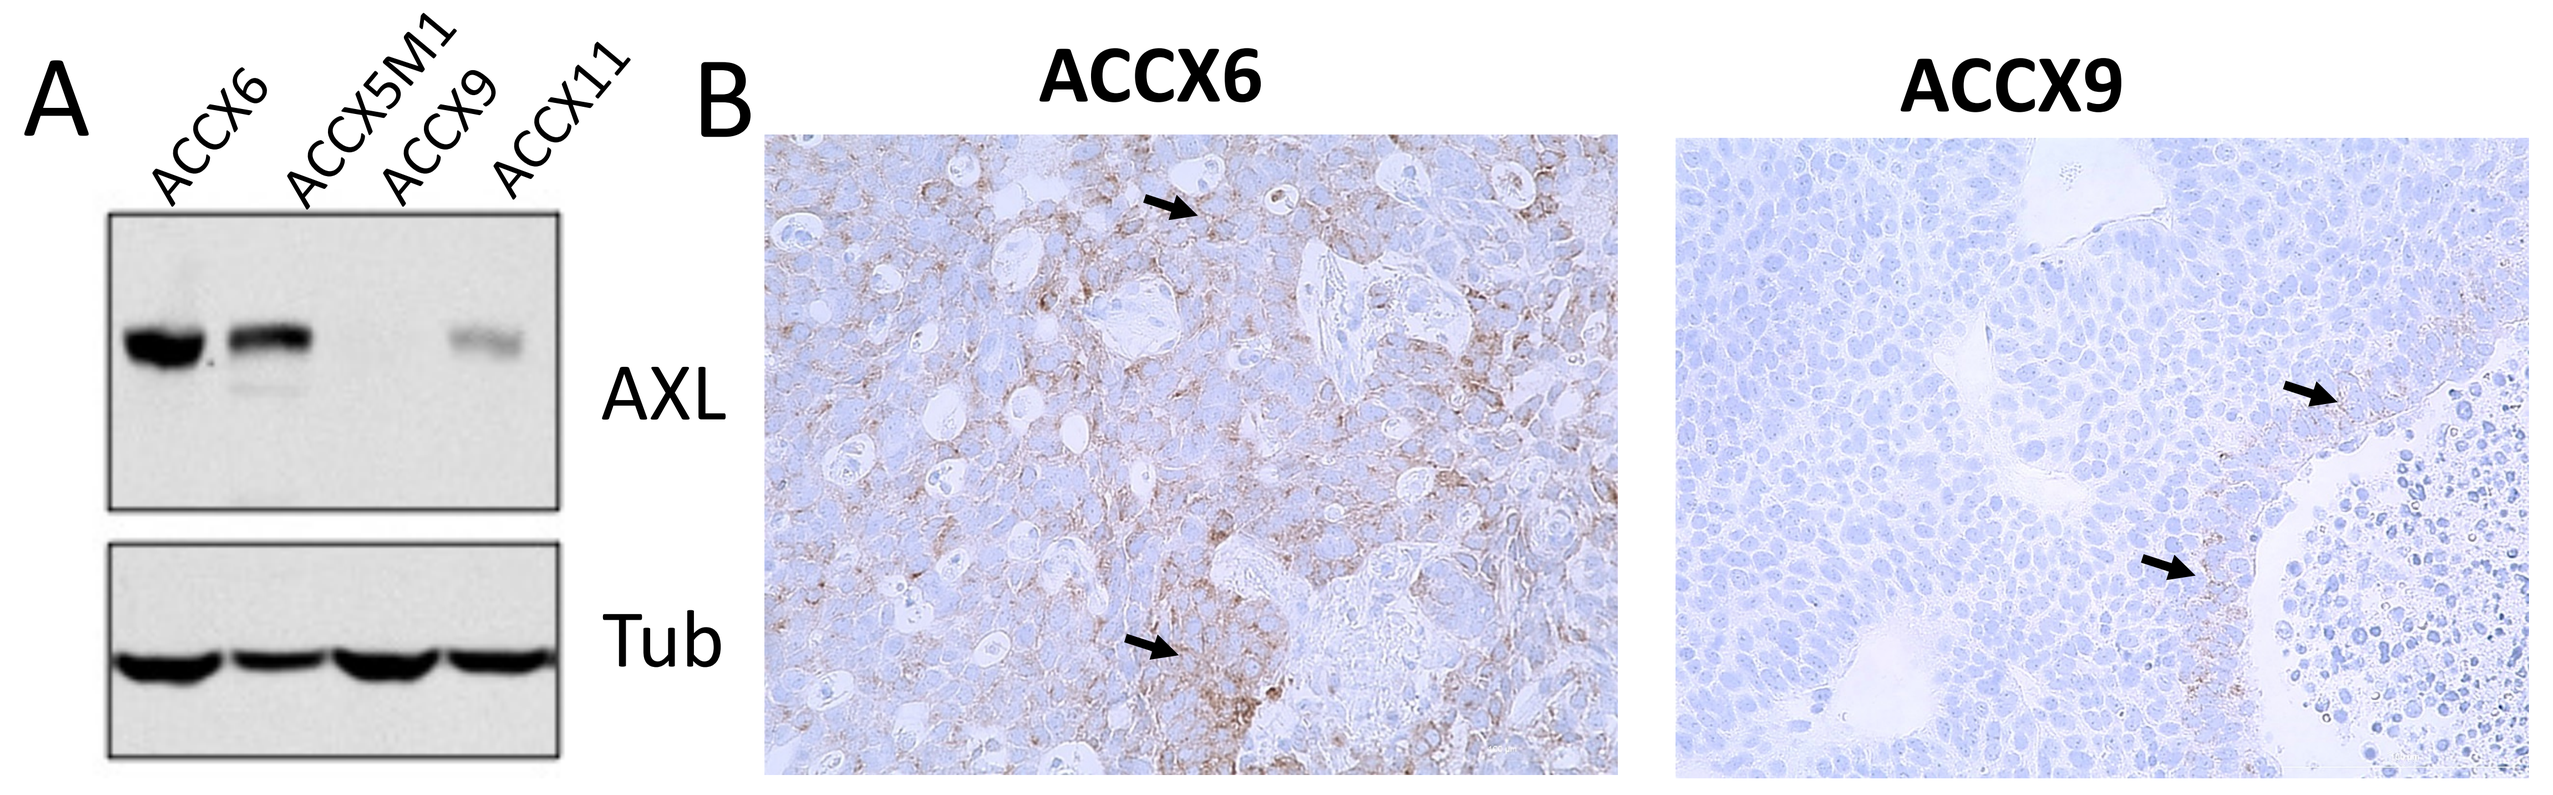
A
ACCX6
ACCX5M1
ACCX9
ACCX11
AXL
Tub
B
ACCX6
ACCX9

Figure 5. AXL expression in ACC PDXs (ACCXs). (**A**), Western blot analysis showing AXL expression in a panel of four ACCXs. (**B**), IHC showing AXL expression status in ACCX6 vs ACCX9. Note that in ACCX6, AXL level is higher and dispersed throughout tumor region, while AXL levels are lower and spatially localized at tumor peripheral edges in ACCX9 (arrows).

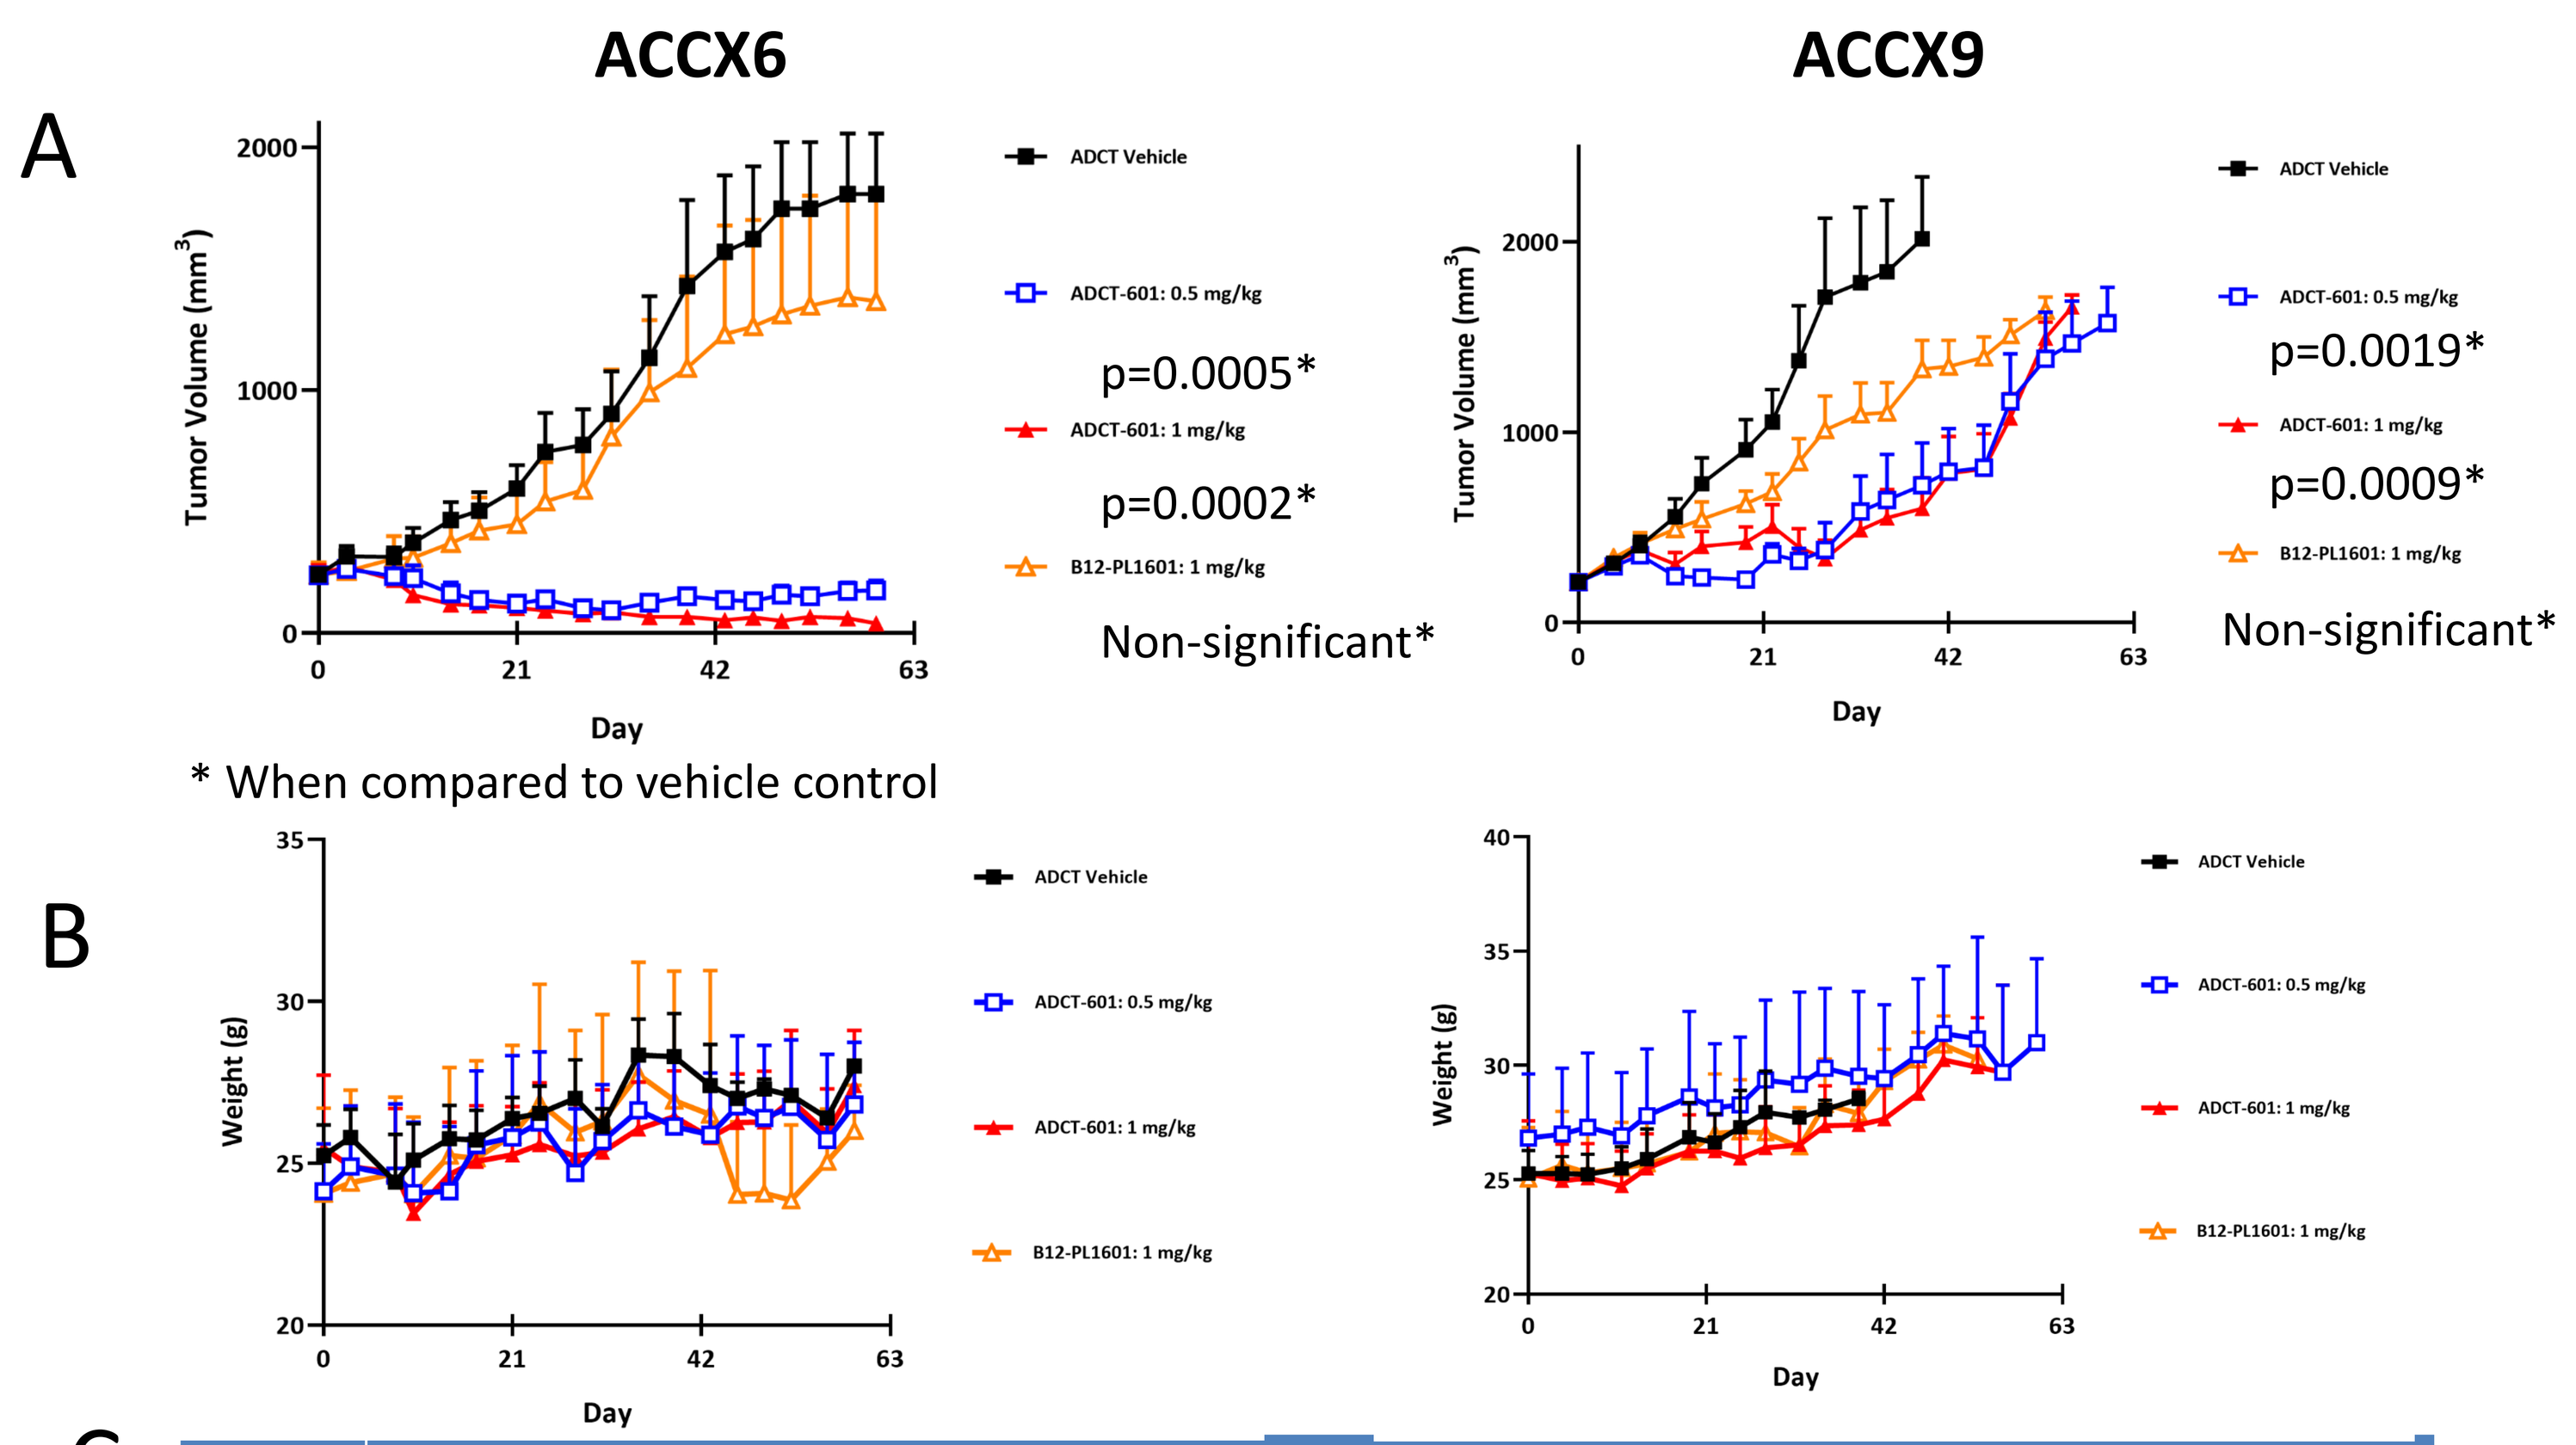
ACCX6
ACCX9
A
Tumor Volume (mm3)
Day
ADCT Vehicle
ADCT-601: 0.5 mg/kg
p=0.0005*
ADCT-601: 1 mg/kg
p=0.0002*
B12-PL1601: 1 mg/kg
Non-significant*
p=0.0019*
p=0.0009*
* When compared to vehicle control
B
Weight (g)

C

	Best Response Observed											
	Vehicle N=5			IgG-ADC (1mg/kg) N=5			ADCT-601 (0.5mg/kg) N=5			ADCT-601 (1mg/kg) N=5		
	CR	PR	SD/PD	CR	PR	SD/PD	CR	PR	SD/PD	CR	PR	SD/PD
ACCX6	0	0	5	0	0	5	0	4	1	1	4	0
ACCX9	0	0	5	0	0	5	0	0	5	0	0	5

Figure 6. ADCT-601 inhibits tumor growth in high AXL expressing ACCX. (**A**),anti-tumor effect of control IgG-ADC or ADCT-601 in high AXL expressing ACCX6 vs low AXL expressing ACCX9. (**B**), Body weight changes of tumor-bearing mice treated with the control IgG-ADC or ADCT-601. (C), Summary of drug response. CR: complete response, PR: partial response (≧50% tumor shrinkage), SD/PD: stable disease/progression of disease

Summary

- ADCT-601 induced a potent dose-dependent cytotoxic effect in ACC cell lines; AXL knockdown attenuates cytotoxic activity of ADCT-601.
- Biochemical data indicates that ADCT-601 treatment led to DNA damage and induction of apoptotic factors in ACC.
- In high AXL expressing ACCX6 model, a single dose of ADCT-601 induced significant tumor regression. Even in low AXL expressing ACCX9 model, a single dose of ADCT-601 significantly inhibited tumor growth.

Conclusion

This study demonstrates that ADCT-601 induced a potent and specific in vitro and in-vivo anti-tumor activity in AXL-expressing ACC models and suggests further development of ADCT-601 in biomarker driven clinical trials.

References

1. Thyparambil SP, Kim YJ, Chambers A et al (2018). Comprehensive proteomic and genomic profiling to identify therapeutic targets in adenoid cystic carcinoma. J Clin Oncol. 36(15_suppl):6053
2. Ferrarotto R, Mitani Y, McGrail DJ et al. (2021). Proteogenomic analysis of salivary adenoid cystic carcinomas defines molecular subtypes and identifies therapeutic targets. Clin Cancer Res 27(3):852-864
3. Humtsoe, JO. Humtsoe JO, Kim HS, et al (2021). Newly identified members of FGFR1 splice variants engage in cross-talk with AXL/AKT axis in salivary adenoid cystic carcinoma. Cancer Res. 15;81(4):1001-1013
4. Zammarchi F, Havenith KE, et al (2022). Preclinical Development of ADCT-601, a Novel Pyrrolobenzodiazepine Dimer-based Antibody-drug Conjugate Targeting AXL-expressing Cancers. Mol Cancer Ther. Jan 27: doi: 10.1158/1535-7163.MCT-21-0715. PMID: 35086955.

© Poster Template by Genigraphics® 1.800.790.4001 www.genigraphics.com