Mechanistic studies investigating the synergistic combination of Loncastuximab Tesirine and Ibrutinib in pre-clinical models of B-cell non-Hodgkin lymphoma

Danilo Cucchi, Nikoleta Sachini, Patrick H. van Berkel, Francesca Zammarchi

ADC Therapeutics UK (Ltd), London, UK

Abstract number: 1050

INTRODUCTION

- Loncastuximab tesirine-lpyl (formerly ADCT-402) is a pyrrolobenzodiazepine (PBD) dimer-based antibody-drug conjugate directed against human CD19 which has been approved by the United States Food and Drug Administration for the treatment of relapsed or refractory diffuse large B-cell lymphoma (DLBCL).
- Pre-clinically, loncastuximab tesirine has shown potent and specific anti-tumor activity in lymphoma models both as single agent and in combination with other approved drugs [1;2].
- Clinically, loncastuximab tesirine is being tested in multiple clinical trials, either as monotherapy or in combination with other anti-lymphoma drugs including ibrutinib.
- Ibrutinib is a small molecule inhibitor of Bruton's tyrosine kinase, approved for treatment of mantle cell lymphoma (MCL), chronic lymphocytic leukaemia and other haematological malignancies.

AIM OF THE STUDY

- Here, we investigated the combined effect of loncastuximab tesirine (lonca) and ibrutinib in pre-clinical models of B-cell non-Hodgkin lymphoma (NHL).

MATERIALS AND METHODS

- Cytotoxicity of lonca, ibrutinib and their combination was determined by the CellTiter 96® Aqueous One Solution Cell Proliferation Assay (MTS assay) (Promega). Chou-Talalay method was used to calculate median combination index (CI) (synergism CI<0.9, additive CI=0.9-1.1, antagonism/no benefit CI>1.1) [3].
- Apoptosis induction was measured using flow cytometry after staining with annexin-V/propidium iodide (PI).
- Whole cell lysates were analyzed by Western blot technique using standard protocol.
- Gene expression analysis was performed using the NanoString Tumour Signalling 360 panel and nSolver Analysis Software 4.0.
- Human IL-10 in cell culture supernatants was measured by solid phase sandwich ELISA (Quantikine™ Human IL-10 Immunoassay, R&D Systems).

RESULTS

Figure 1: Combination of lonca with ibrutinib is synergistic in NHL-derived cell lines

A

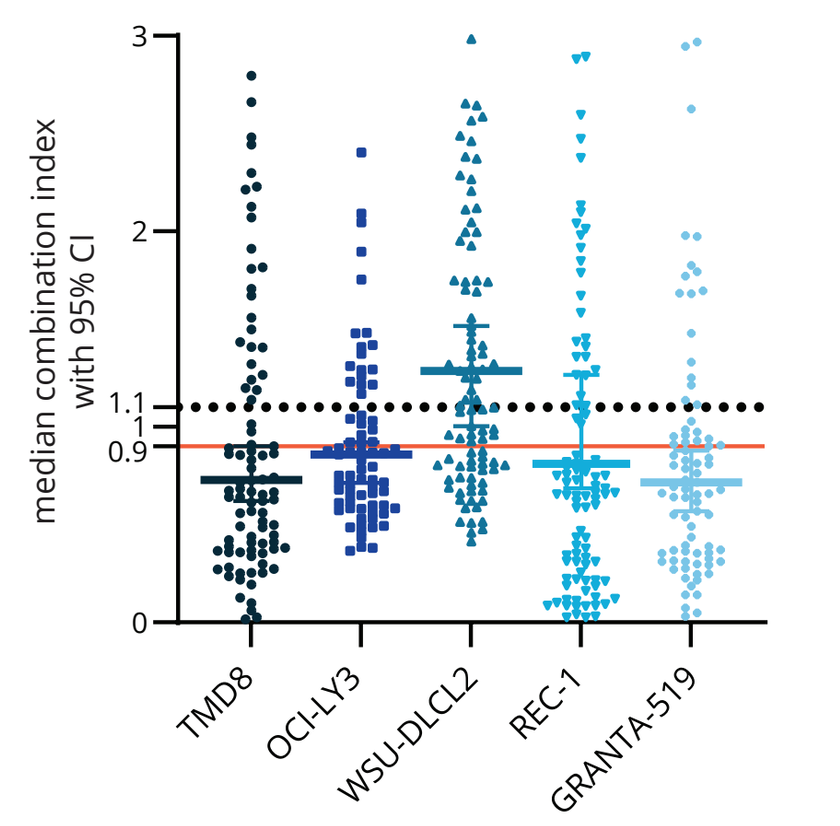
median combination index with 95% CI
TMD8
OCI-LY3
WSU-DLCL2
REC-1
GRANTA-519

B

	Lonca+Ibrutinib	
	Median Combination Index (CI)	95% Confidence interval
TMD8 (ABC)	0.72	0.62-0.90
OCI-LY3 (ABC)	0.85	0.71-0.92
WSU-DLCL2 (GBC)	1.28	1.00-1.51
REC-1 (MCL)	0.81	0.68-1.26
GRANTA-519 (MCL)	0.71	0.56-0.87

A. Distribution of Chou-Talalay Combination Index (CI) values obtained combining lonca with ibrutinib in TMD8, OCI-LY3, WSU-DLCL2, REC-1 and GRANTA-519. In each plot, the horizontal line indicates the median CI and the whiskers represent 95% confidence interval values. Orange horizontal line indicates threshold for synergy; dotted horizontal line indicates threshold for additivity. CI values > 3 have been omitted from the figure. **B.** Table summarizing median CI values with 95% confidence interval values (synergism CI<0.9, additive CI=0.9-1.1, antagonism/no benefit CI>1.1). ABC: activated B-cell-DLBCL; GBC: germinal center B-cell-DLBCL; MCL: mantle cell lymphoma.

Figure 2: Combination of lonca with ibrutinib results in increased apoptosis

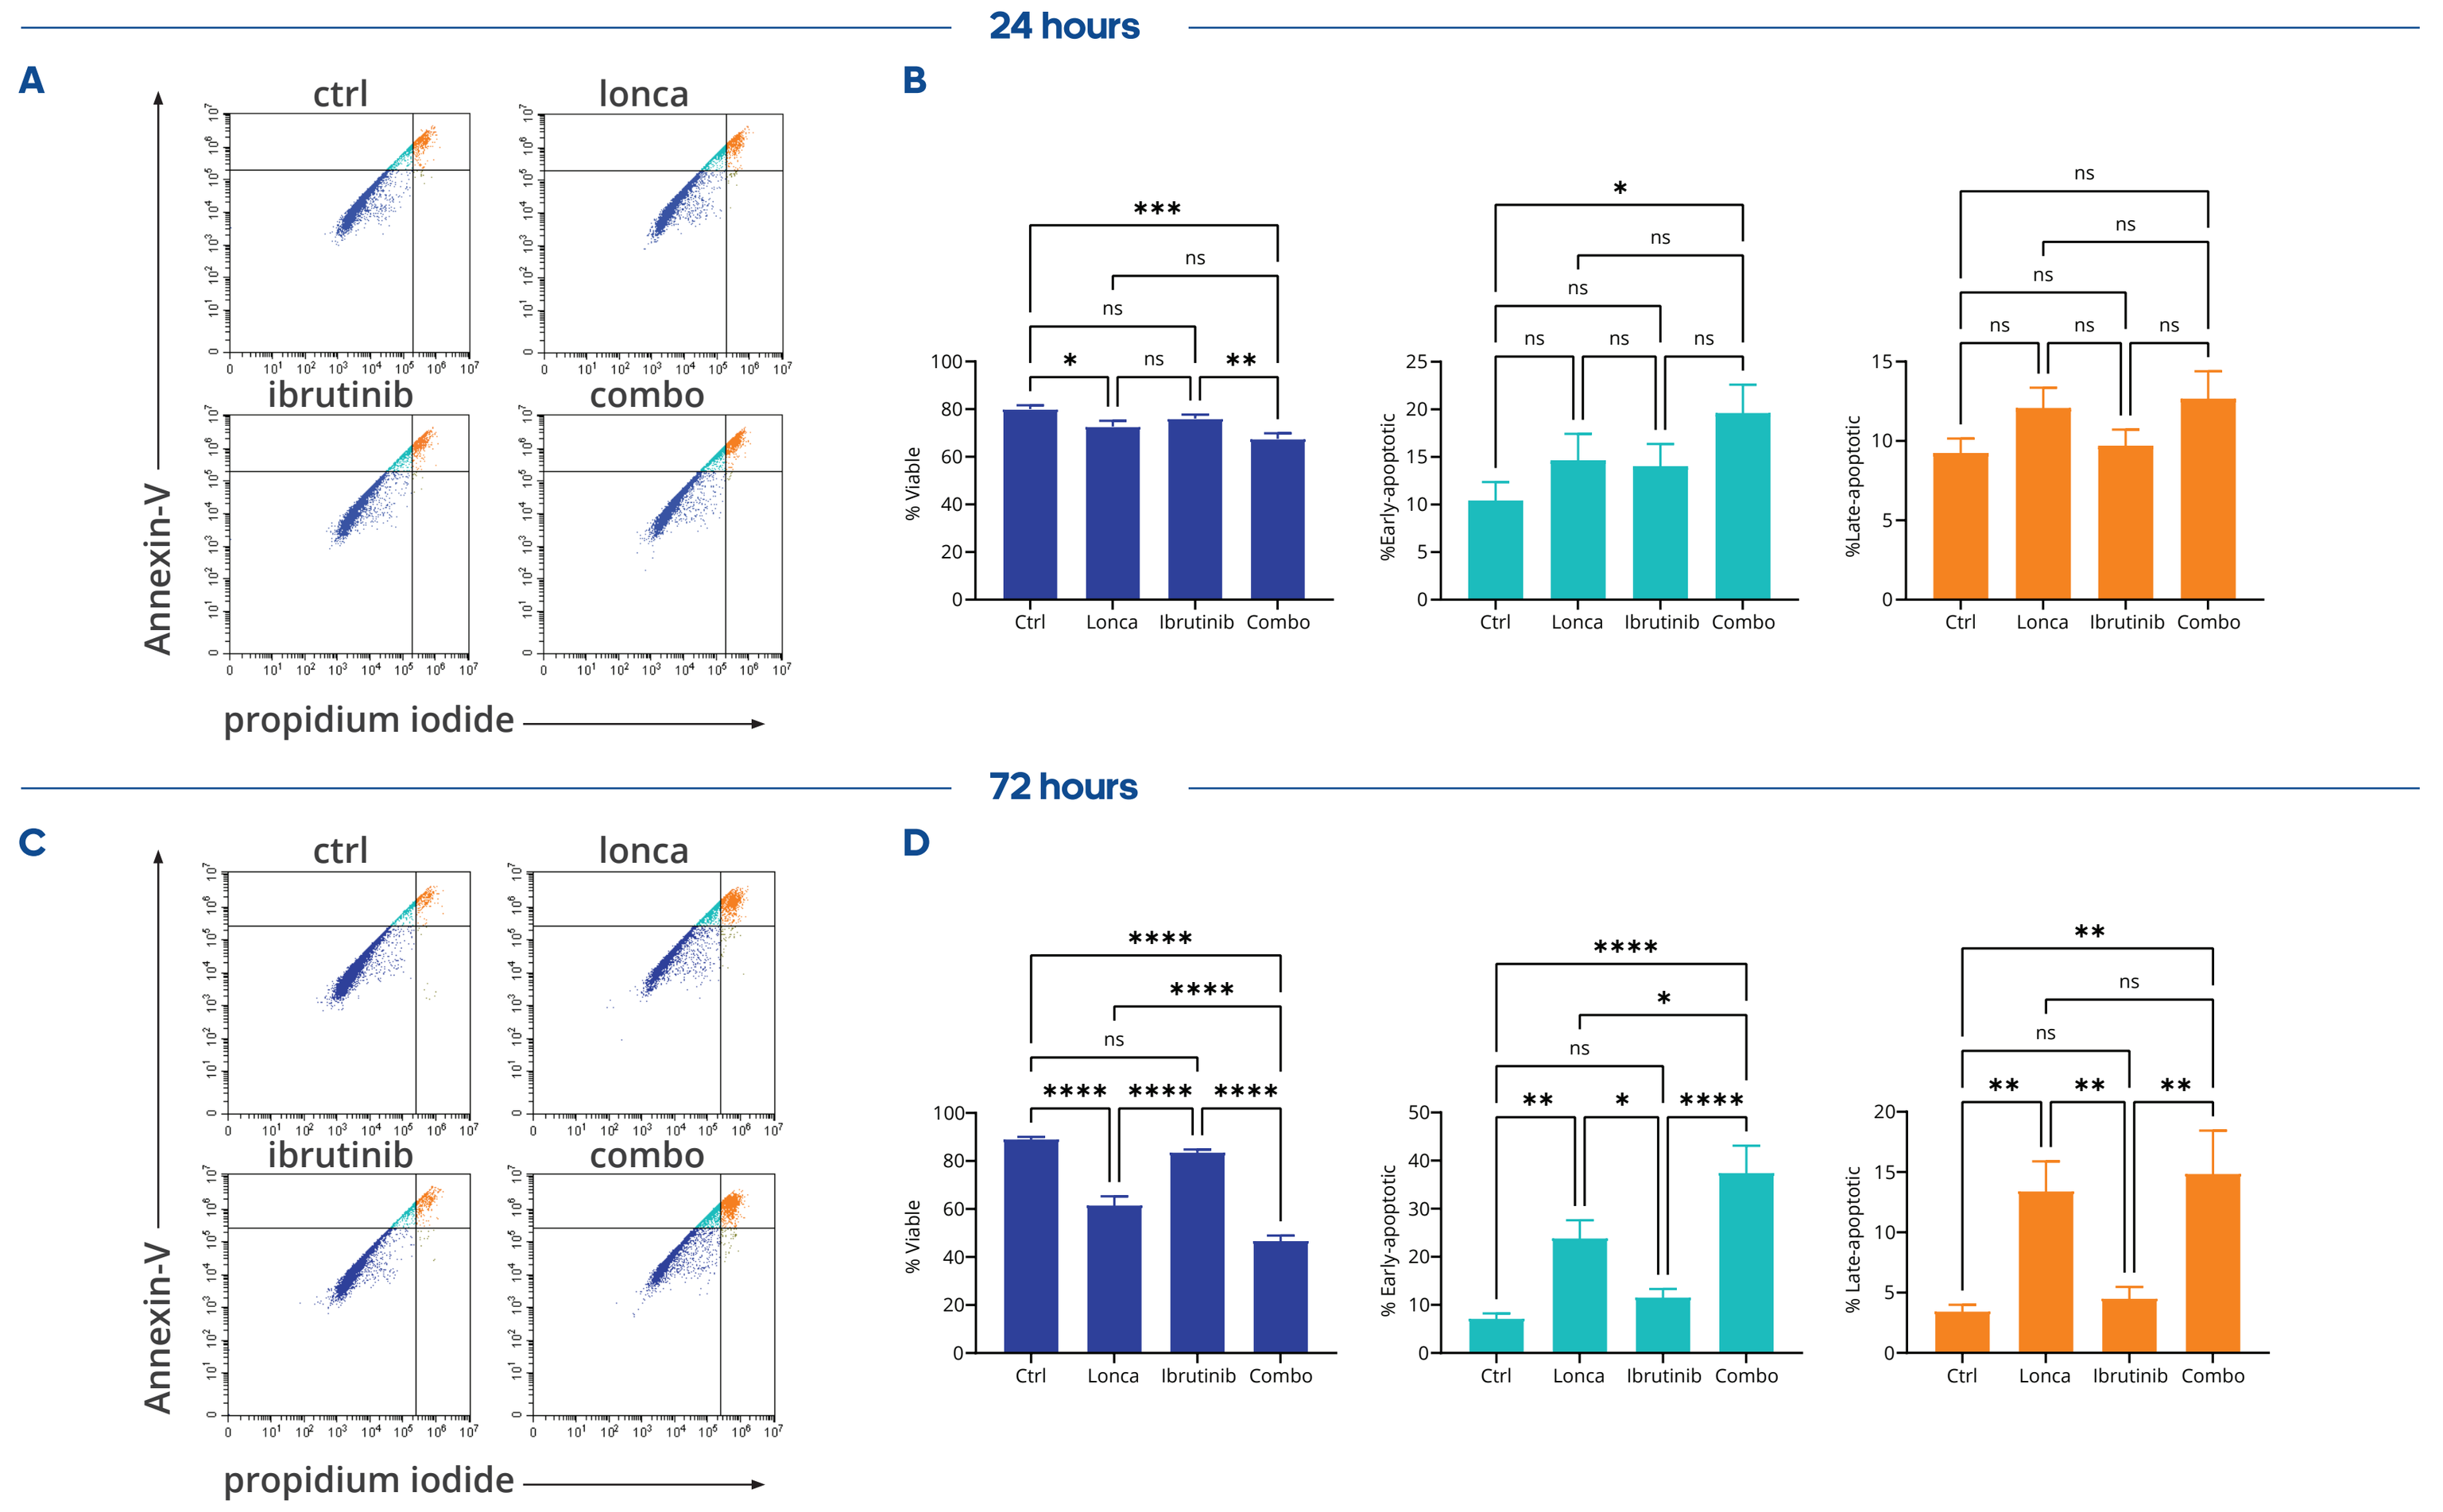
24 hours
A
B
ctrl
lonca
ibrutinib
combo
Annexin-V
propidium iodide
% Viable
%Early-apoptotic
%Late-apoptotic
Ctrl
Lonca
Ibrutinib
Combo
72 hours
C
D

Annexin-V/PI flow cytometry analysis of TMD8 cells treated with lonca, ibrutinib or combination of lonca with ibrutinib for 24 hours (A-B) and 72 hours (C-D). **A-C.** Representative flow cytometry plots showing viable (lower left quadrant), early apoptotic (upper left quadrant) and late apoptotic (upper right quadrant) cells. **B-D.** Percentage of viable, early and late apoptotic cells. Data represent mean± S.E.M. of three independent experiments (*p ≤ 0.05, **p ≤ 0.01, ****p ≤ 0.0001 calculated by ANOVA using the uncorrected Fisher's LSD multiple comparisons test with 95% confidence). Ctrl, control (untreated TMD8).

Figure 3: Combination of lonca with ibrutinib results in increased γH2AX expression

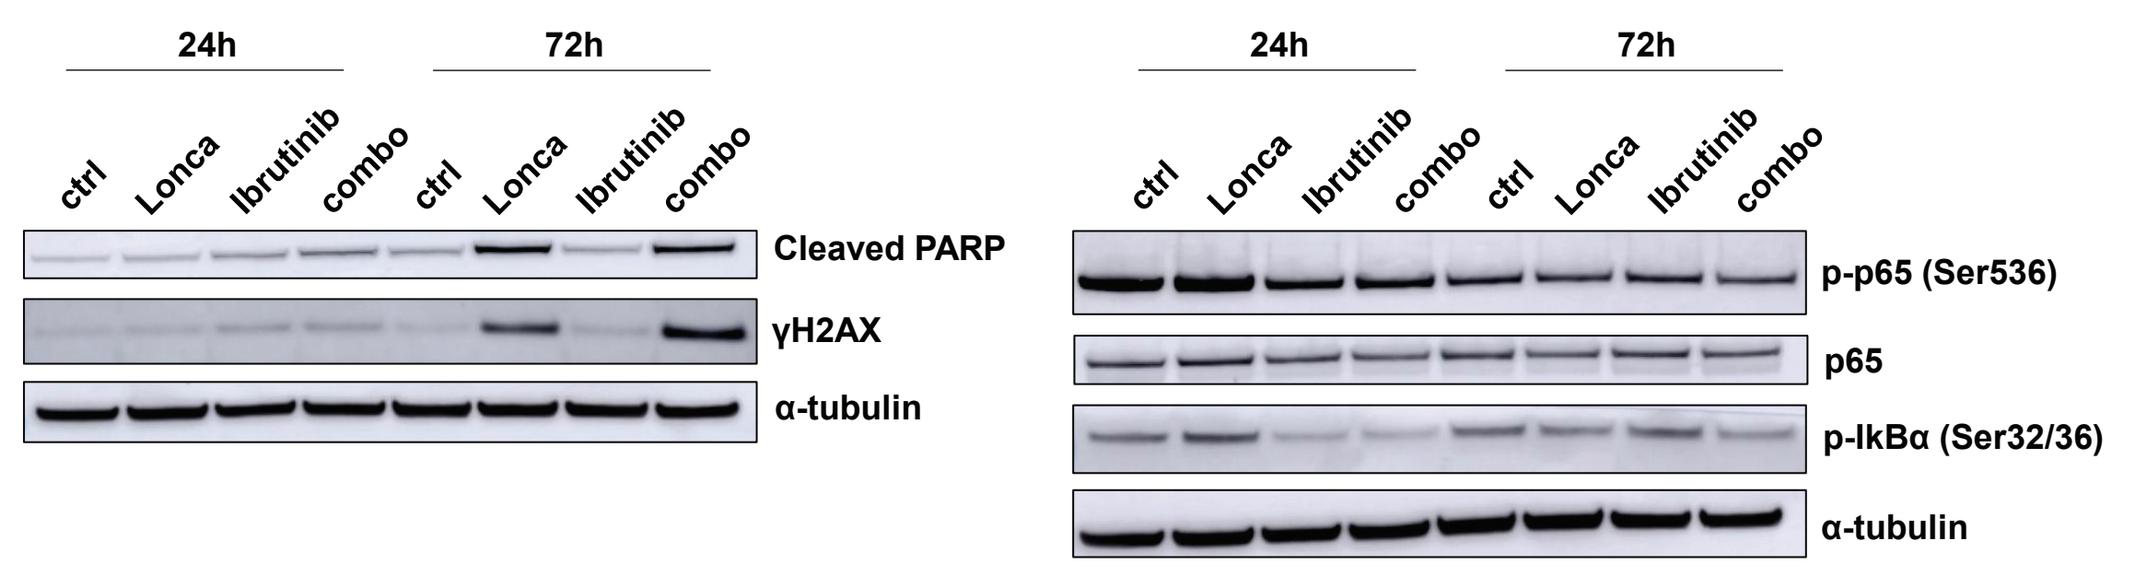
24h
72h
ctrl
Lonca
Ibrutinib
combo
Cleaved PARP
γH2AX
α-tubulin
p-p65 (Ser536)
p65
p-IkBα (Ser32/36)

Representative western blot showing expression of cleaved PARP, γH2AX, p65, p-p65, p-Ikbα and α-tubulin in TMD8 cells treated with lonca, ibrutinib or combination of lonca with ibrutinib for 24 and 72 hours. Ctrl, control (untreated TMD8).

Figure 4: Gene expression analysis of TMD8 cells treated with lonca, ibrutinib or their combination

A: Lonca has a gene expression pattern closer to the control, while ibrutinib and the combination cluster together

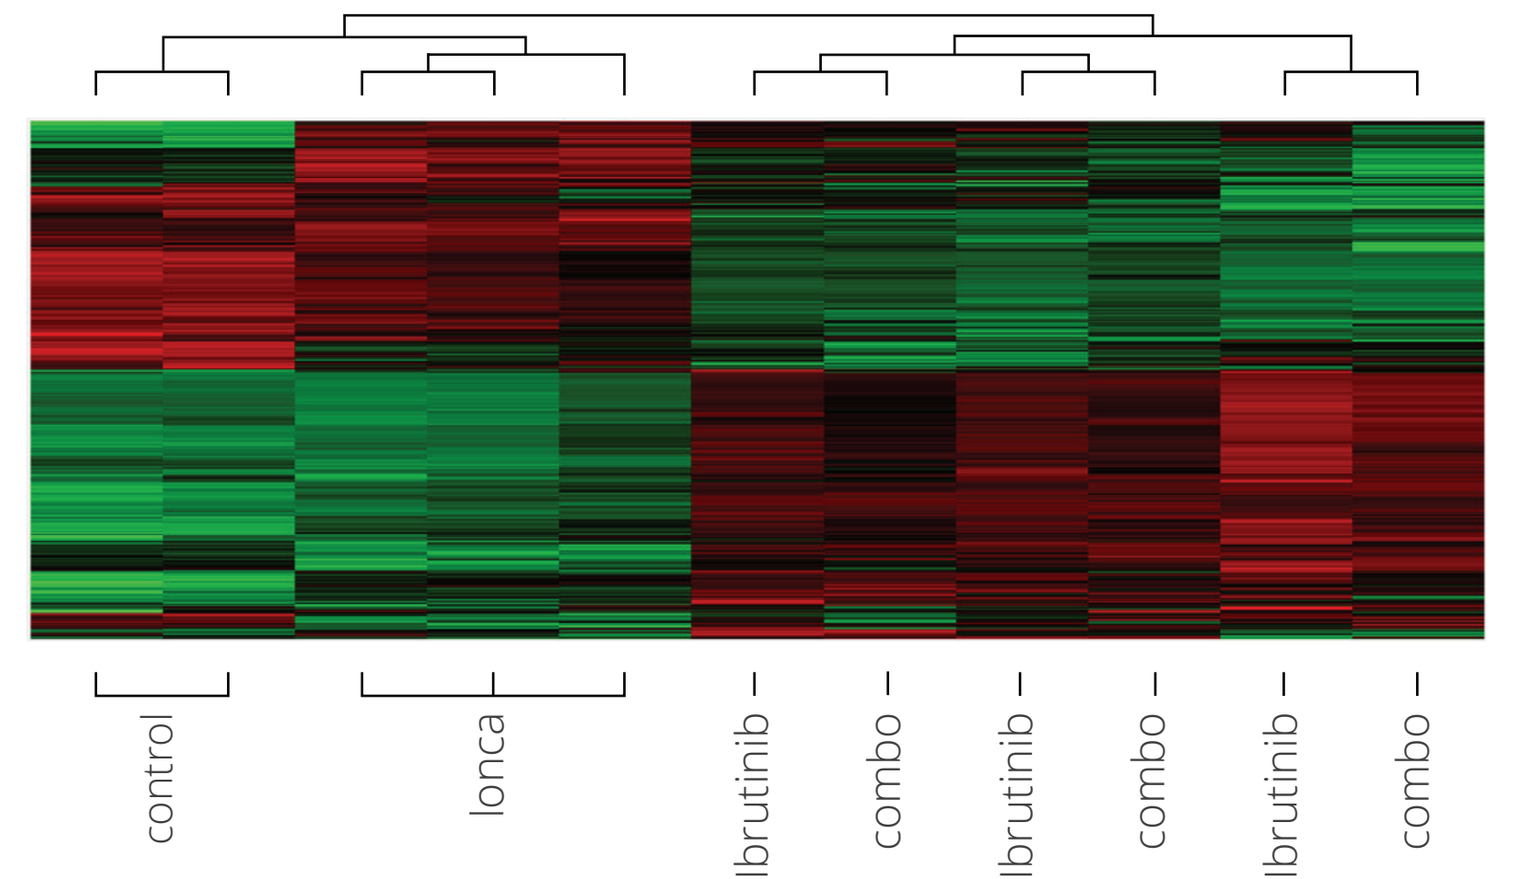
control
lonca
Ibrutinib
combo

B: NF-kB signalling and cytokine-cytokine receptor interaction are among the most affected pathways

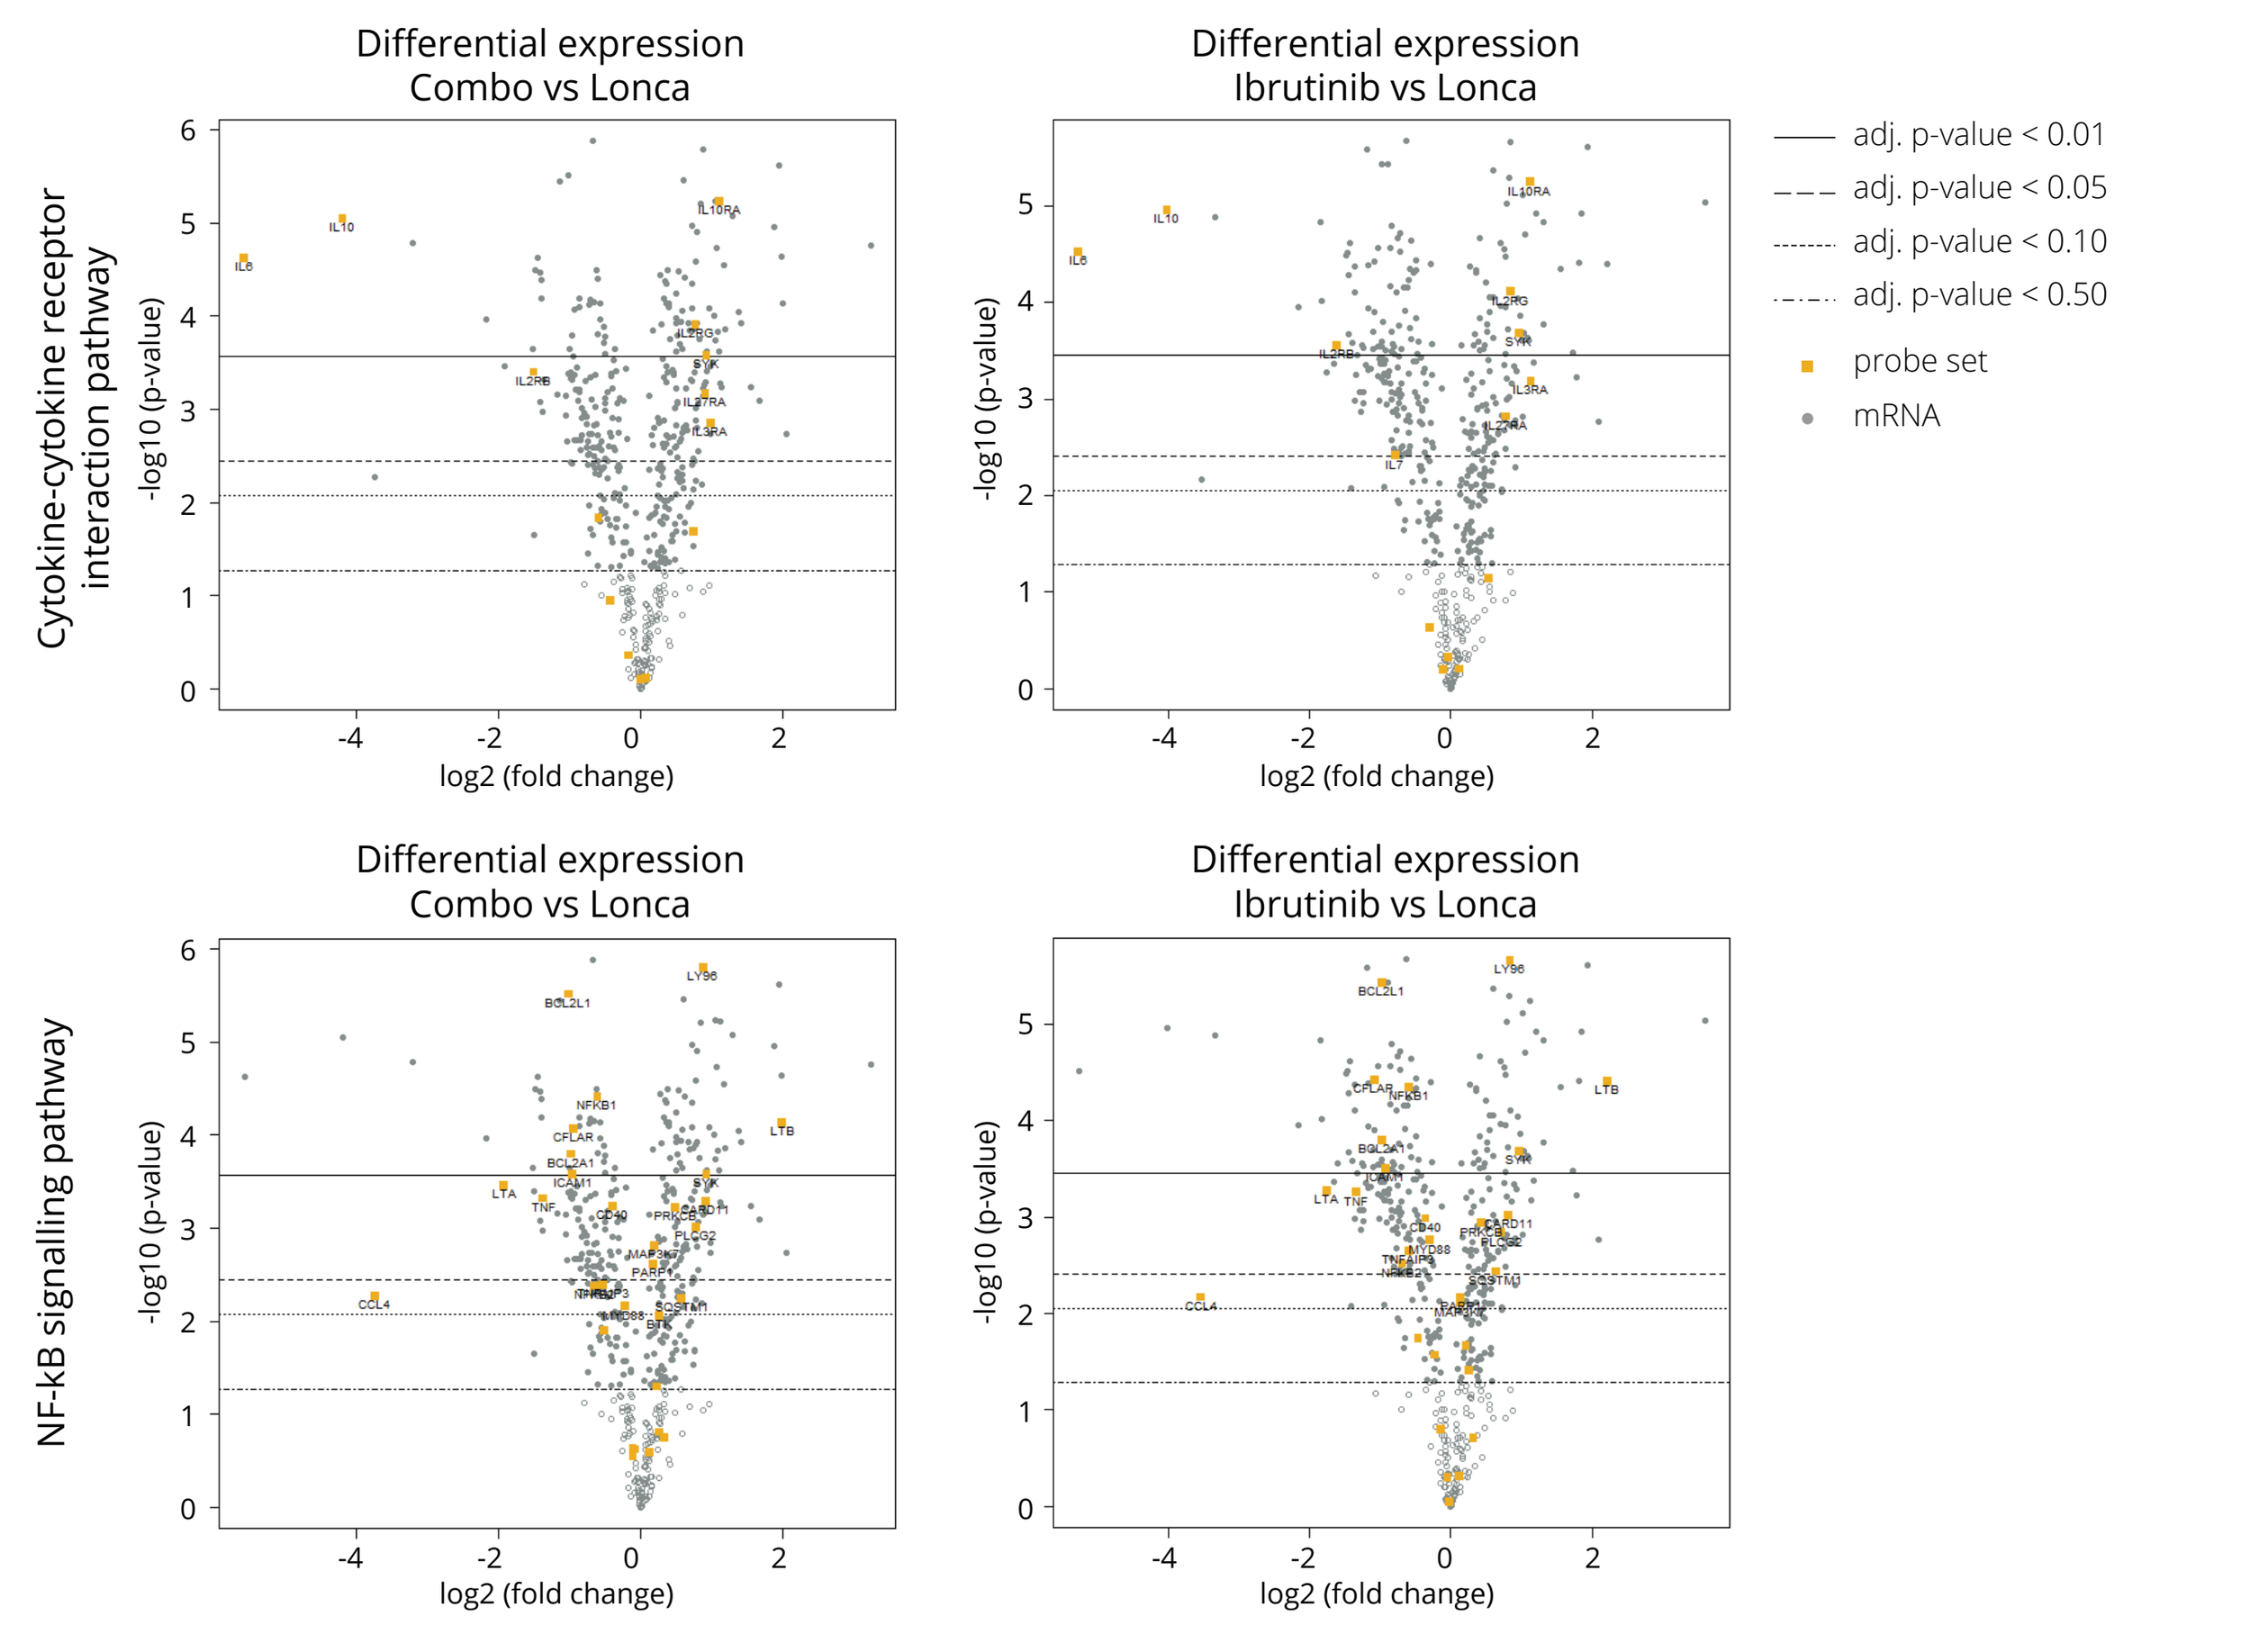
Differential expression
Combo vs Lonca
Ibrutinib vs Lonca
Cytokine-cytokine receptor interaction pathway
NF-kB signalling pathway
-log10 (p-value)
log2 (fold change)
adj. p-value < 0.01
adj. p-value < 0.05
adj. p-value < 0.10
adj. p-value < 0.50
probe set
mRNA

A. Nanostring heatmap of individual samples normalised data. Expression level of each gene is plotted by z-score with Euclidean distance metric and average linkage. Colour scale bar ranges from higher (green) to lower (red) expression. **B.** Nanostring volcano plots of the cytokine-cytokine receptor interaction pathway (top) and NF-kB signaling pathway (bottom). Data are presented as fold change in gene expression (log2(fold change)) against significance of change (–log10(P value)). The P-value adjustment for differential expression analysis has been performed with the Benjamini-Yekutieli method. Genes with adjusted p-value <0.01 are considered highly statistically significant. Highly statistically significant genes fall at the top of the plot above the horizontal lines, and highly differentially expressed genes fall to either side. Highlighted in orange are the probes from the gene set relative to the analysed pathways. The data shown represent three biological replicates of TMD8 cells treated with lonca, ibrutinib or their combination for 24 hours. Control, TMD8 untreated.

Figure 5: Combination of lonca with ibrutinib reduces IL-10 production

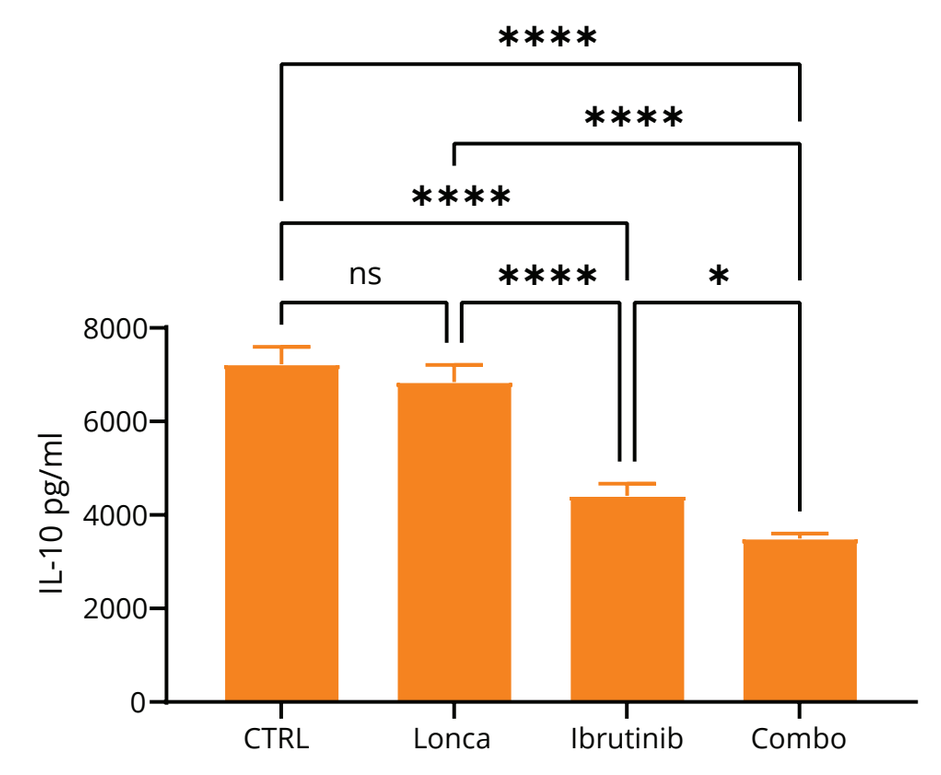
IL-10 pg/ml
CTRL
Lonca
Ibrutinib
Combo

Amount of IL-10 detected in the supernatant of TMD8 cells treated with lonca, ibrutinib or combination of lonca with ibrutinib for 5 days. Data represent mean± S.E.M. of four independent experiments (*p ≤ 0.05, ****p ≤ 0.0001 calculated by ANOVA using the uncorrected Fisher's LSD multiple comparisons test with 95% confidence).

CONCLUSIONS

- *In vitro*, combination of lonca with ibrutinib resulted in synergistic effect in four out of five NHL-derived cell lines and in increased apoptosis (both early and late apoptosis) in TMD8 cells (Figures 1 and 2).
- Protein expression analysis revealed that ibrutinib drives a slight increase in γH2AX and cleaved PARP after 24 hours of treatment, while lonca is largely responsible for the upregulation of both markers after 72 hours of treatment. Interestingly, combination of lonca with ibrutinib resulted in increased γH2AX expression compared to the two single agents at the 72-hour time-point (Figure 3).
- Gene expression analysis of TMD8 cells treated with lonca, ibrutinib and their combination revealed that lonca has a gene expression profile closer to the control (in line with the targeted mode of action of lonca and its PBD dimer component SG3199), while ibrutinib and the combination have similar profiles. Cytokine-cytokine receptor interaction and the NF-kB signaling were among the pathways most affected by ibrutinib and the combination. IL-10 was found to be amongst the most affected genes.
- Production of IL-10 was more effectively downregulated in the combination setting compared to the two single agents.
- In conclusion, these preclinical data support the ongoing investigation into the mechanism of action of the combination of lonca with ibrutinib in B-cell NHL models.

REFERENCES

1. Zammarchi, F., et al., ADCT-402, a PBD dimer-containing antibody drug conjugate targeting CD19-expressing malignancies. Blood, 2018. 131(10): p. 1094-1105.
2. Tarantelli, C., et al., The antibody-drug conjugate (ADC) loncastuximab tesirine (ADCT-402) targeting CD19 shows strong in vitro anti-lymphoma activity both as single agents and in combination Hematological Oncology, 2019. 37(S2).
3. Chou, T.C., Drug combination studies and their synergy quantification using the Chou-Talalay method. Cancer Res, 2010. 70(2): p. 440-6.