First-in-Human, Phase 1, Open-Label, Dose-Escalation Study of ADCT-901 as Monotherapy in Patients With Select Advanced Solid Tumors

R. Donald Harvey, PharmD, BCOP, FCCP, FHOPA[1*], Gerald Falchook, MD[2], Abdul Rafeh Naqash, MD[3], Joseph W. Kim, MD[4], Afshin Dowlati, MD[5], Yvan LeBruchec[6], Isabelle Coudert[6], Annette Ervin-Haynes, DO, MPA[7], David Sommerhalder, MD[8]

[1]Department of Hematology and Medical Oncology, Emory Winship Cancer Institute, Atlanta, GA, USA; [2]Sarah Cannon Research Institute at HealthOne, Denver, CO, USA; [3]The University of Oklahoma, College of Medicine, Oklahoma City, OK, USA; [4]Yale Cancer Center, Medical Oncology, Yale School of Medicine, New Haven, CT, USA; [5]Seidman Cancer Center and Case Western Reserve University, Cleveland, OH, USA; [6]ADC Therapeutics, Épalinges, Switzerland; [7]ADC Therapeutics, New Providence, NJ, USA; [8]NEXT Oncology, San Antonio, TX, USA

TPS3157

INTRODUCTION

- Kidney-associated antigen 1 (KAAG1) is expressed in a high percentage of ovarian tumors, prostate cancer, triple-negative breast cancer (TNBC), and renal cell carcinoma, while it has limited expression in normal tissue.[1,2]
- KAAG1 represents an attractive target for an antibody-drug conjugate (ADC) approach due to high and selective expression on the tumor cell surface, rapidly internalizing and colocalization with a lysosomal marker, and restricted expression in healthy tissues.[2]
- ADCT-901 is an ADC composed of a humanized monoclonal antibody, IgG1, directed against human KAAG1 and conjugated through a cathepsin-cleavable linker to SG3199, a pyrrolobenzodiazepine (PBD)-dimer cytotoxin (**Figure 1**).[1]
- In mouse xenograft models of human-derived TNBC and ovarian and renal cancers, significant tumor reduction was observed after a single dose of ADCT-901, providing the rationale for the clinical development of a PBD-based ADC to treat KAAG1-expressing tumors.[2]

Figure 1. Structure of ADCT-901

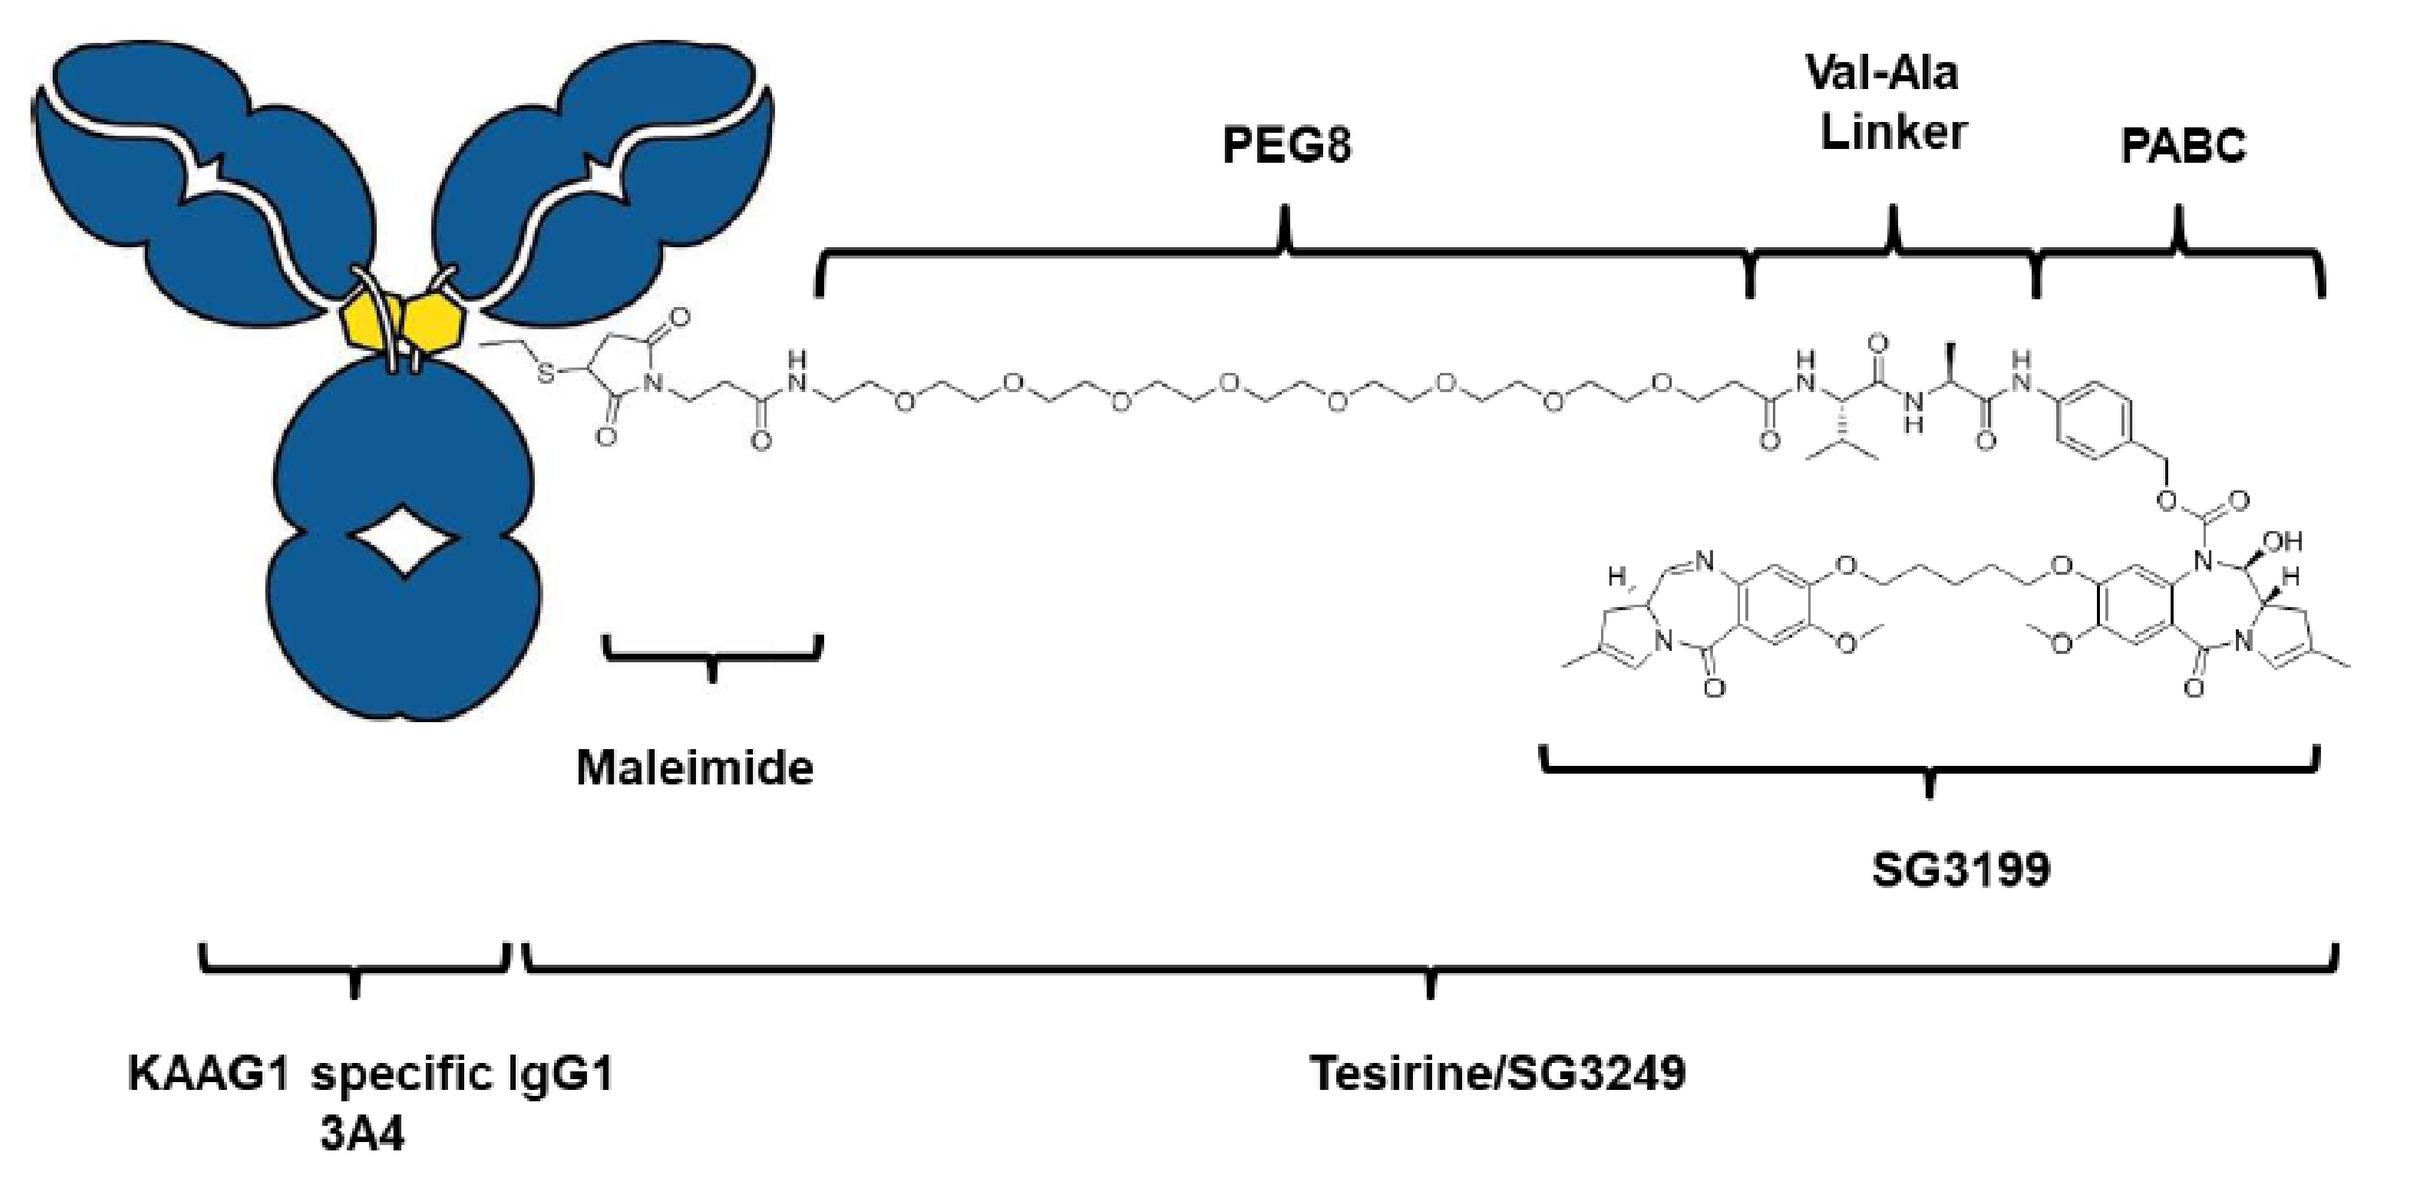
PEG8
Val-Ala Linker
PABC
Maleimide
SG3199
KAAG1 specific IgG1 3A4
Tesirine/SG3249

OBJECTIVE

- To identify the recommended dose and schedule for expansion of ADCT-901 and to characterize safety and tolerability in patients with selected advanced solid tumors that potentially express KAAG1.

METHODS

Study Design

- This is a phase 1, multicenter, two-part, open-label study (NCT04972981) that will enroll ~76 patients (**Figure 2**).[3]

Figure 2. Study design

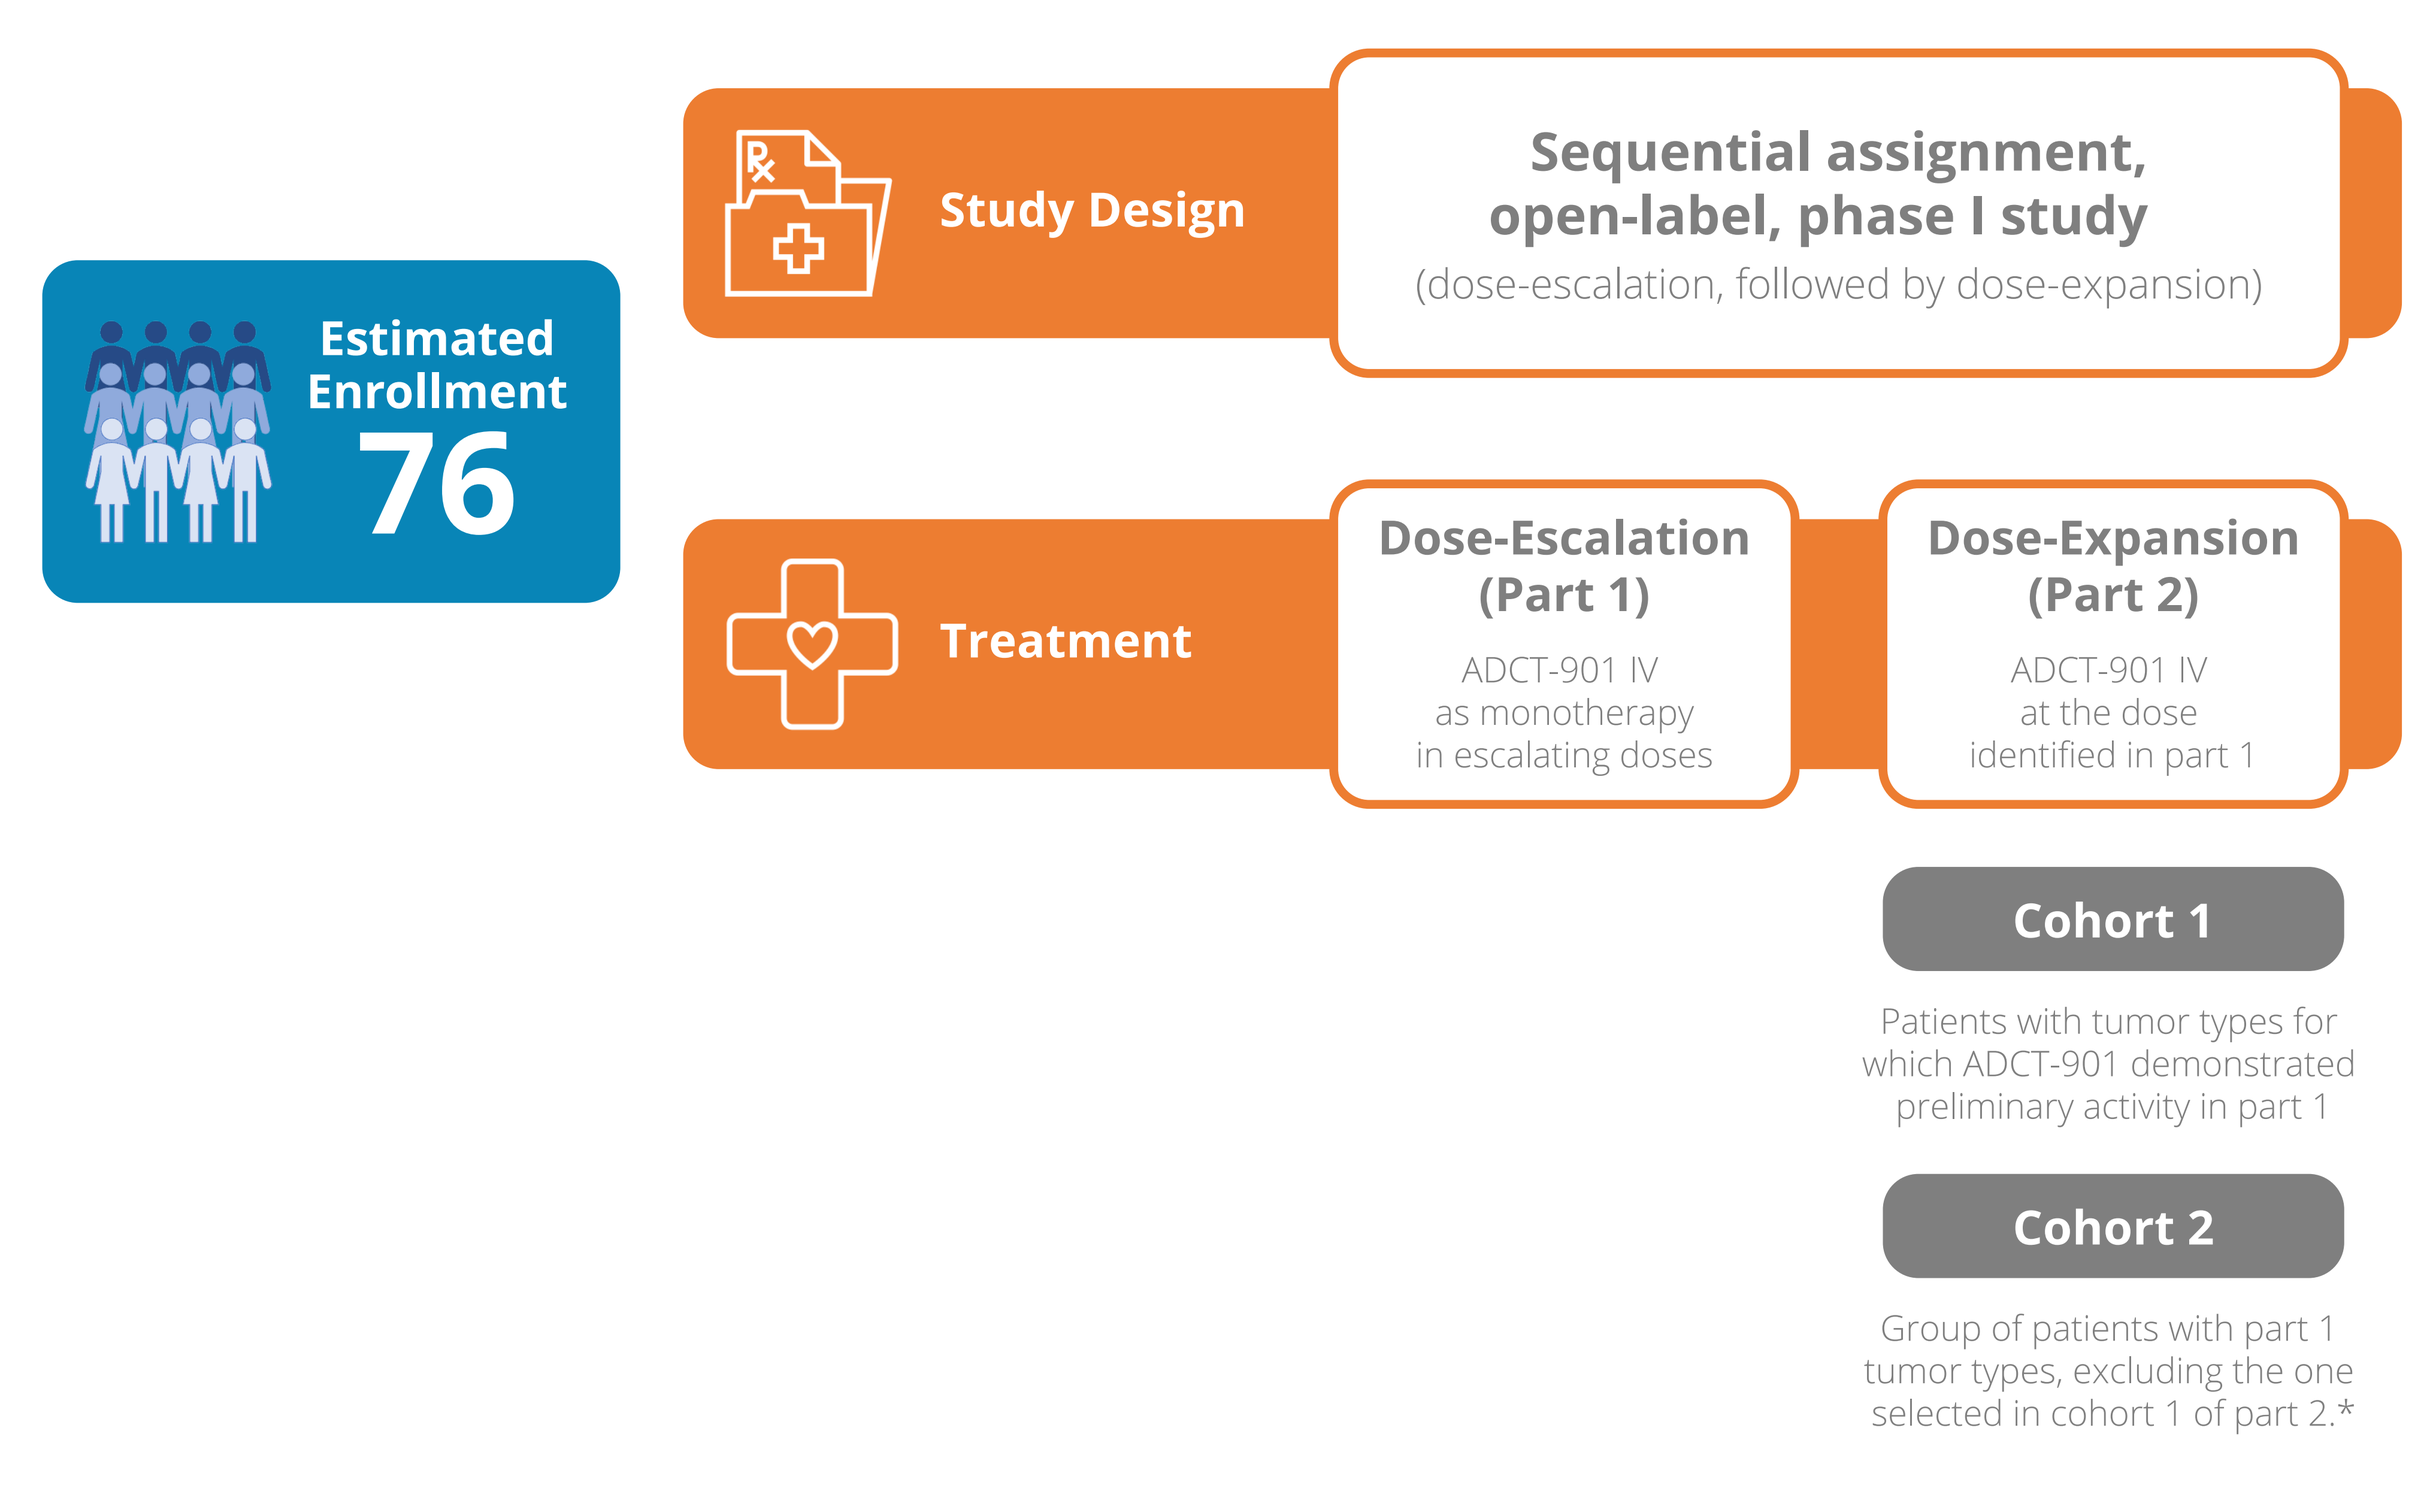
Estimated Enrollment
76
Study Design
Sequential assignment, open-label, phase I study
(dose-escalation, followed by dose-expansion)
Treatment
Dose-Escalation (Part 1)
ADCT-901 IV as monotherapy in escalating doses
Dose-Expansion (Part 2)
ADCT-901 IV at the dose identified in part 1
Cohort 1
Patients with tumor types for which ADCT-901 demonstrated preliminary activity in part 1
Cohort 2
Group of patients with part 1 tumor types, excluding the one selected in cohort 1 of part 2.*

*No more than 30% of patients with the same indication will be permitted in this basket group. IV, intravenous.

- In part 1, patients will receive escalating doses of ADCT-901 guided by a 3+3 design (**Table 1**).
 - 1st dose: 15 µg/kg every 3 weeks (Q3W); highest dose: 290 ug/kg Q3W (or equivalent).
- Oral dexamethasone (4 mg), or equivalent, will be administered the day before, the day of, and the day after each ADCT-901 dose.
- Dose-limiting toxicities (DLTs) include febrile neutropenia or neutropenic infection (≥grade 3), neutropenia lasting >7 days (≥grade 4), thrombocytopenia (≥grade 4), thrombocytopenia with clinically significant bleeding (≥grade 3), thrombocytopenia requiring a platelet transfusion (≥grade 3), anemia (≥grade 4), anemia requiring transfusion (≥grade 3), Hy's law case, hypersensitivity/infusion-related reactions (≥grade 3), ocular disorders (≥grade 2), and all other nonhematologic toxicities, including mucositis (≥grade 3).
- The recommended dose for expansion (RDE, dose and schedule of ADCT-901 identified in part 1) will be tested in part 2 to further characterize the safety, tolerability, and preliminary efficacy of ADCT-901.
- The treatment period is defined as the date when a patient receives the first dose of the study drug until the end of the treatment visit.
- Patients will be followed every 9 weeks for up to 1 year and after the end of treatment for survival.

Table 1. Provisional dose levels in the dose-escalation (part 1)

Dose level*	Dose of ADCT-901, Q3W (µg/kg)
1 (starting dose)	15
2	30
3	60
4	90
5	120
6	150
7	190
8	240
9	290

Q3W, every 3 weeks.
*Additional or intermediate dose levels, or different dosing schedules, may be implemented during the study; however, the dose will not be higher than 290 ug/kg Q3W (or equivalent).

Outcomes

- Primary endpoints
 - Incidence of DLTs (part 1 only).
 - Frequency/severity of adverse events (AE) and serious AEs.
 - Changes from baseline of safety laboratory values, vital signs, Eastern Cooperative Oncology Group performance status, and 12-lead electrocardiograms.
 - Frequency of dose interruptions and reductions.
- Secondary endpoints
 - Overall response rate, duration of response, progression-free survival, and overall survival.
 - Pharmacokinetic parameters of ADCT-901 total antibody, PBD-conjugated antibody, and unconjugated SG3199 in serum.
 - Frequency of confirmed positive antidrug antibody responses.
- Exploratory endpoints
 - Relationship between exposure and selected safety and efficacy endpoints.
 - Relationship between tumor and/or blood biomarkers, and/or cell-free DNA, and selected efficacy and safety endpoints.
 - Presence of KAAG1 expression (by immunohistochemistry) at baseline in tumor tissue.
 - Changes in tissue biomarkers between baseline and treatment biopsies.
 - Changes in values of blood biomarkers.

Eligibility Criteria

- Key inclusion and exclusion criteria are shown in **Table 2**.

Table 2. Key eligibility criteria

Inclusion	Exclusion
Aged ≥18 years	Prior solid organ transplant
Pathologic diagnosis of selected solid tumor locally advanced or metastatic at the time of screening: cholangiocarcinoma, renal cell carcinoma, ovarian/fallopian tube and prostate cancers, triple-negative breast cancer	History of recent infection
	Symptomatic CNS metastases or leptomeningeal disease
	Clinically significant third space fluid accumulation
Refractory or intolerant to existing therapies known to provide clinical benefit	Active diarrhea ≥ CTCAE grade 2 or a medical condition associated with chronic diarrhea
Measurable disease as determined by RECIST v1.1	Active or clinically significant ocular disease
ECOG performance status 0-2	HIV seropositive, serologic evidence of hepatitis B or C virus infection, recent SARS-CoV-2 infection
Adequate organ function based on laboratory parameters: • Absolute neutrophil count ≥1.5 × 10^3/µL • Platelet count ≥100 × 10^3/µL without transfusion in the past 10 days • Hemoglobin ≥9 g/dL • ALT, AST, or GGT ≤2 × ULN if there is no liver involvement or ≤5 × ULN if there is liver involvement • Total bilirubin ≤1.5 × ULN • Calculated CrCl ≥60 mL/min (Cockcroft-Gault) • INR <2 × ULN	Significant medical comorbidities, including uncontrolled hypertension (BP ≥160 mmHg systolic and/or ≥110 mmHg diastolic repeatedly with or without antihypertensive medication), unstable angina, congestive heart failure (greater than NYHA class II), electrocardiographic evidence of acute ischemia, coronary angioplasty or myocardial infarction within 6 months prior to screening, severe uncontrolled atrial or ventricular cardiac arrhythmia, poorly controlled diabetes, active ulceration of the upper GI tract or GI bleeding, or severe chronic pulmonary disease
	Major surgery, radiotherapy, chemotherapy, or other antineoplastic therapy within 14 days prior to the start of the study drug
	Active second primary malignancy (other than NMSC), nonmetastatic prostate cancer, in situ cervical cancer, ductal or lobular carcinoma in situ of the breast, or other malignancy medically monitored by the sponsor

ALT, alanine aminotransferase; AST, aspartate aminotransferase; BP, blood pressure; CNS, central nervous system; CrCl, creatinine clearance; CTCAE, Common Terminology Criteria for Adverse Events; ECOG, Eastern Cooperative Oncology Group; GGT, gamma-glutamyl transferase; GI, gastrointestinal; HIV, human immunodeficiency virus; INR, international normalized ratio; NMSC, nonmelanoma skin cancer; NYHA, New York Heart Association; RECIST, Response Evaluation Criteria in Solid Tumors; SARS-CoV-2, severe acute respiratory syndrome coronavirus 2; ULN, upper limit of normal.

Study Assessments

- Study assessments are shown in **Table 3**.
- Screening visits occur from 21 days to 1 day prior to day 1 of cycle 1.
- During the treatment period, imaging will be performed at 6 weeks and 12 weeks (±1 week) after day 1 of cycle 1 and then every 9 weeks (±2 weeks) thereafter until disease progression or withdrawal of consent.
- All safety assessments on dosing days will be performed prior to study drug administration. Additional assessments may be performed as clinically indicated.

Table 3. Study assessments

Efficacy	Safety
Disease assessment • Imaging • Clinical examination **PK, PD, and immunogenicity** • PK of ADCT-901–conjugated antibody, total antibody, and unconjugated SG3199 warhead in serum • ADA in whole blood • Blood biomarkers, cfDNA, gDNA • Tumor tissue biomarkers	• AEs • SAEs • Physical examination • ECOG performance status • Vital signs • Height and weight • Laboratory tests • Pregnancy test • ECGs • Ophthalmology testing

ADA, antidrug antibody; AE, adverse events; cfDNA, circulating free DNA; ECG, electrocardiogram; ECOG, Eastern Cooperative Oncology Group; gDNA, genomic DNA; PK, pharmacokinetic; SAEs, serious adverse events.

STUDY STATUS

- The study opened for recruitment in September 2021; as of April 26, 2022, 8 patients have been treated across the first three dose levels (or equivalent).
- Enrollment in the dose-escalation part is ongoing.

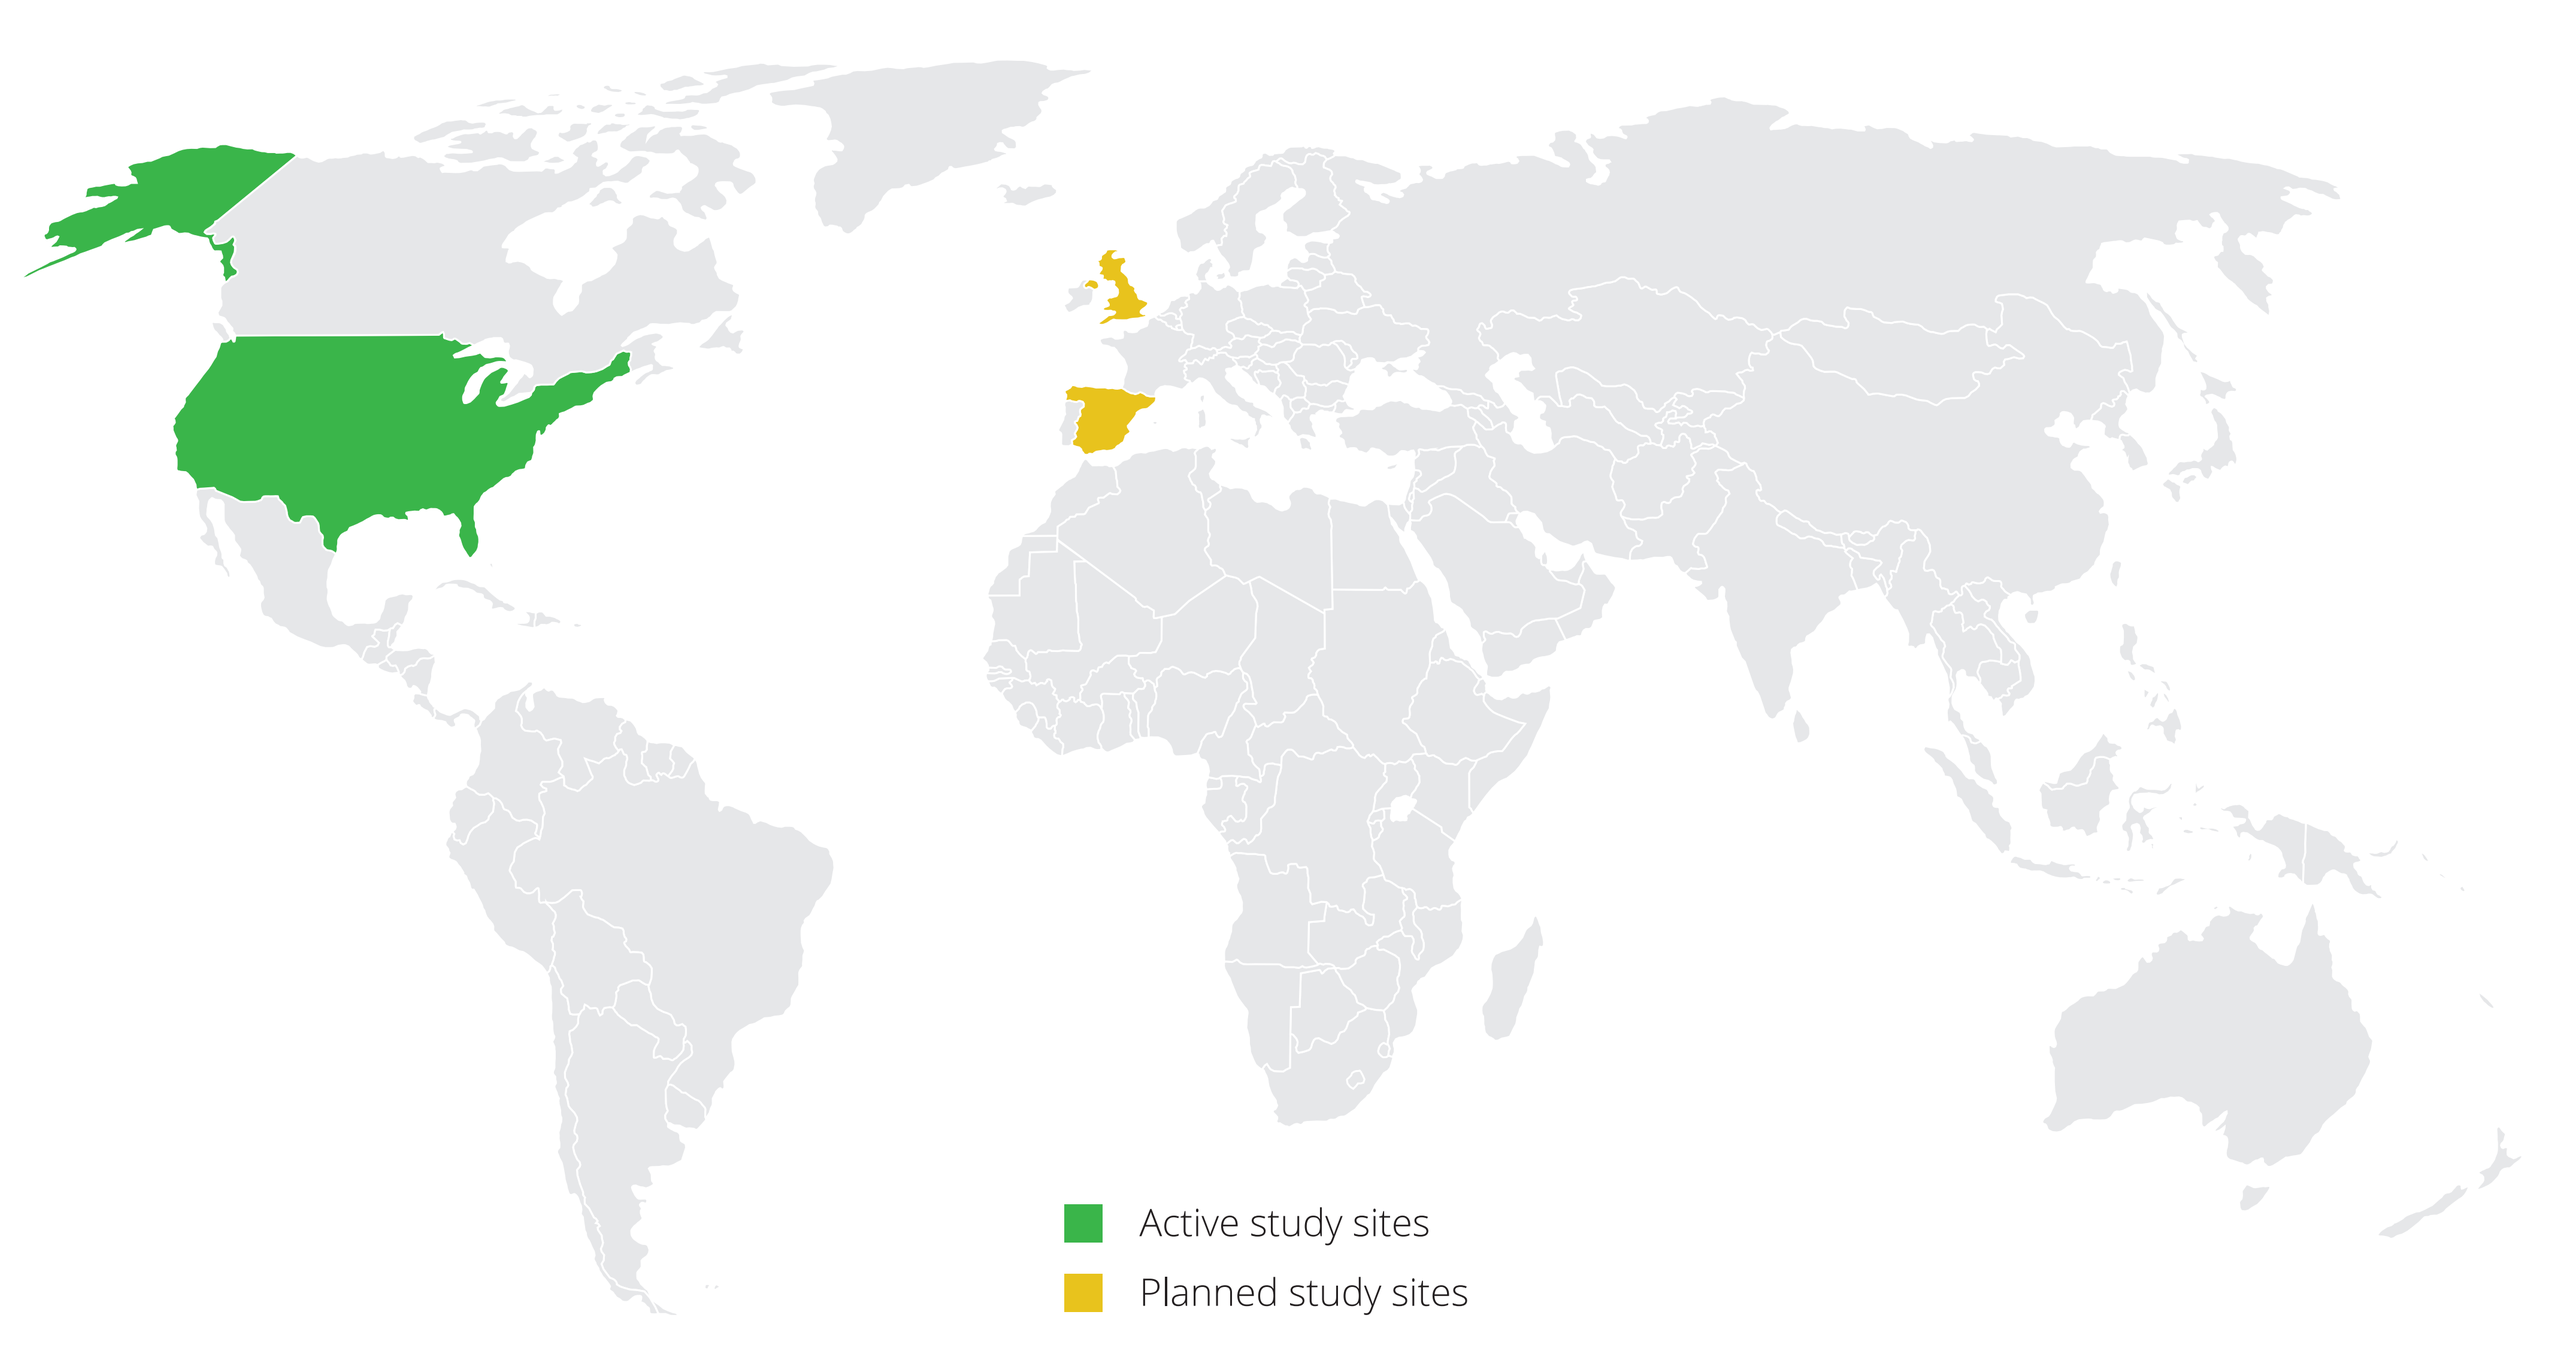
Active study sites
Planned study sites

KEY MESSAGE

- The recommended dose for expansion, safety, and clinical activity of ADCT-901, a KAAG1-directed antibody-drug conjugate, are being assessed in patients with selected advanced solid tumors that potentially express KAAG1 in a phase 1, two-part, open-label study (NCT04972981).

Acknowledgments

- ADC Therapeutics SA; medical writing support: CiTRUS Health Group.

*Contact Information

R. Donald Harvey, PharmD, BCOP, FCCP, FHOPA. Email: rdharve@emory.edu

References

1. ADC Therapeutics. ADCT-901. https://adctherapeutics.com/adct-901/. Accessed April 1, 2022.
2. Zammarchi F, et al. *AACR Cancer Res*. 2019;79(13 Suppl):Abstract 234.
3. Clinicaltrials.gov. A study to evaluate the safety, tolerability, pharmacokinetics, and antitumor activity of ADCT-901 in selected advanced solid tumors. Updated April 1, 2022. https://clinicaltrials.gov/ct2/show/NCT04972981. Accessed April 27, 2022.

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Poster presented at the American Society of Clinical Oncology (ASCO) Annual Meeting. June 3–7, 2022, Chicago, IL USA