EUROPEAN HEMATOLOGY ASSOCIATION

Health-Related Quality of Life and Tolerability of Loncastuximab Tesirine in High-Risk Patients With Relapsed/Refractory Diffuse Large B-Cell Lymphoma Treated in a Phase 2 Clinical Trial (LOTIS-2)

Alexander Spira,[1] Laura Liao,[2] Xiaolei Zhou,[3] Ari Gnanasakthy,[3] Lei Chen,[2] David Ungar,[2] Eric Yu,[2] Turk Kilavuz,[2] John Radford,[4] Mehdi Hamadani[5]

[1]Virginia Cancer Specialists Research Institute, Fairfax, VA, United States; [2]ADC Therapeutics America Inc., New Providence, NJ, United States; [3]RTI Health Solutions, Research Triangle Park, NC, United States; [4]NIHR Clinical Research Facility, The Christie NHS Foundation Trust and University of Manchester, Manchester Academic Health Centre, Manchester, United Kingdom; [5]Medical College of Wisconsin, Division of Hematology & Oncology, Milwaukee, WI, United States

INTRODUCTION

- Loncastuximab tesirine (loncastuximab tesirine-lpyl; Lonca), a CD19-directed antibody-drug, was granted approval by the US Food and Drug Administration for the treatment of adult patients with relapsed or refractory large B-cell lymphoma after two or more lines of systemic therapy, including diffuse large B-cell lymphoma (DLBCL) not otherwise specified, DLBCL arising from low-grade lymphoma, and high-grade B-cell lymphoma.
- Lonca has shown antitumor activity with an acceptable toxicity profile and provides stable or improved health-related quality of life (HRQOL) in adult patients with relapsed/refractory diffuse large B-cell lymphoma (R/R DLBCL) after ≥ 2 prior therapies.[1,2]
- Lonca has also shown promising results for patients who are at high risk of a poor prognosis (i.e., with double-/triple-hit, primary refractory, or transformed disease).

OBJECTIVE

- This analysis evaluates the impact of Lonca on HRQOL and treatment tolerability stratified by high-risk group.

METHODS

- The LOTIS-2 study (NCT03589469) in a single-arm, open-label, phase 2 study of adult patients with R/R DLBCL after ≥ 2 prior treatments who had measurable disease and Eastern Cooperative Oncology Group performance status 0-2.
- Eligible patients received Lonca as an intravenous infusion on day 1 of each 3-week treatment cycle at 150 μg/kg for two cycles then at 75 μg/kg thereafter for up to 1 year or until disease relapse or progression; unacceptable toxicity; death; or patient or investigator decision (Figure 1).
- The high-risk group included patients with double-/triple-hit, primary refractory, or transformed disease at baseline. Primary refractory disease was defined as no response to first-line therapy.

Patient-Reported Outcome Assessments

- The EuroQol EQ-5D-5L and Functional Assessment of Cancer Treatment–Lymphoma (FACT-Lym) were assessed at baseline (cycle 1, day 1 predose) and day 1 of each subsequent treatment cycle until end of treatment.
 - The EQ-5D visual analog scale (VAS) measures overall health (current health state). A score of 100 indicates “the best health you can imagine,” and a score of 0 indicates “the worst health you can imagine.”
 - The FACT-Lym total score (range, 0-168) is the sum of Physical Well-Being (range, 0-28), Social/Family Well-Being (range, 0-28), Emotional Well-Being (range, 0-24), and Functional Well-Being (range, 0-28) subscales plus the Lymphoma Subscale for lymphoma-specific symptoms and concerns (range, 0-60).
 - Treatment tolerability was measured by FACT-Lym item GP5 (“I am bothered by side effects of treatment”). This single item has been used to measure overall side effect impact on patients.[3,4]

Analysis Method

- Changes in HRQOL from baseline were summarized descriptively for EQ-5D VAS and FACT-Lym total by visit and risk group.
- Analysis of covariance models were conducted for EQ-5D VAS and FACT-Lym total to estimate the least squares mean changes from baseline in high-risk and non-high-risk groups, as well as the differences between risk groups, after adjusting for age, sex, race, and baseline score.
- Percentages of responses to GP5 (tolerability) were summarized by visits and risk group.
- Analysis was conducted using data collected from study initiation (August 2018) through March 2021 in the LOTIS-2 study.
- Data were analyzed as observed without imputation on missing data.

Figure 1. LOTIS-2 Study Design

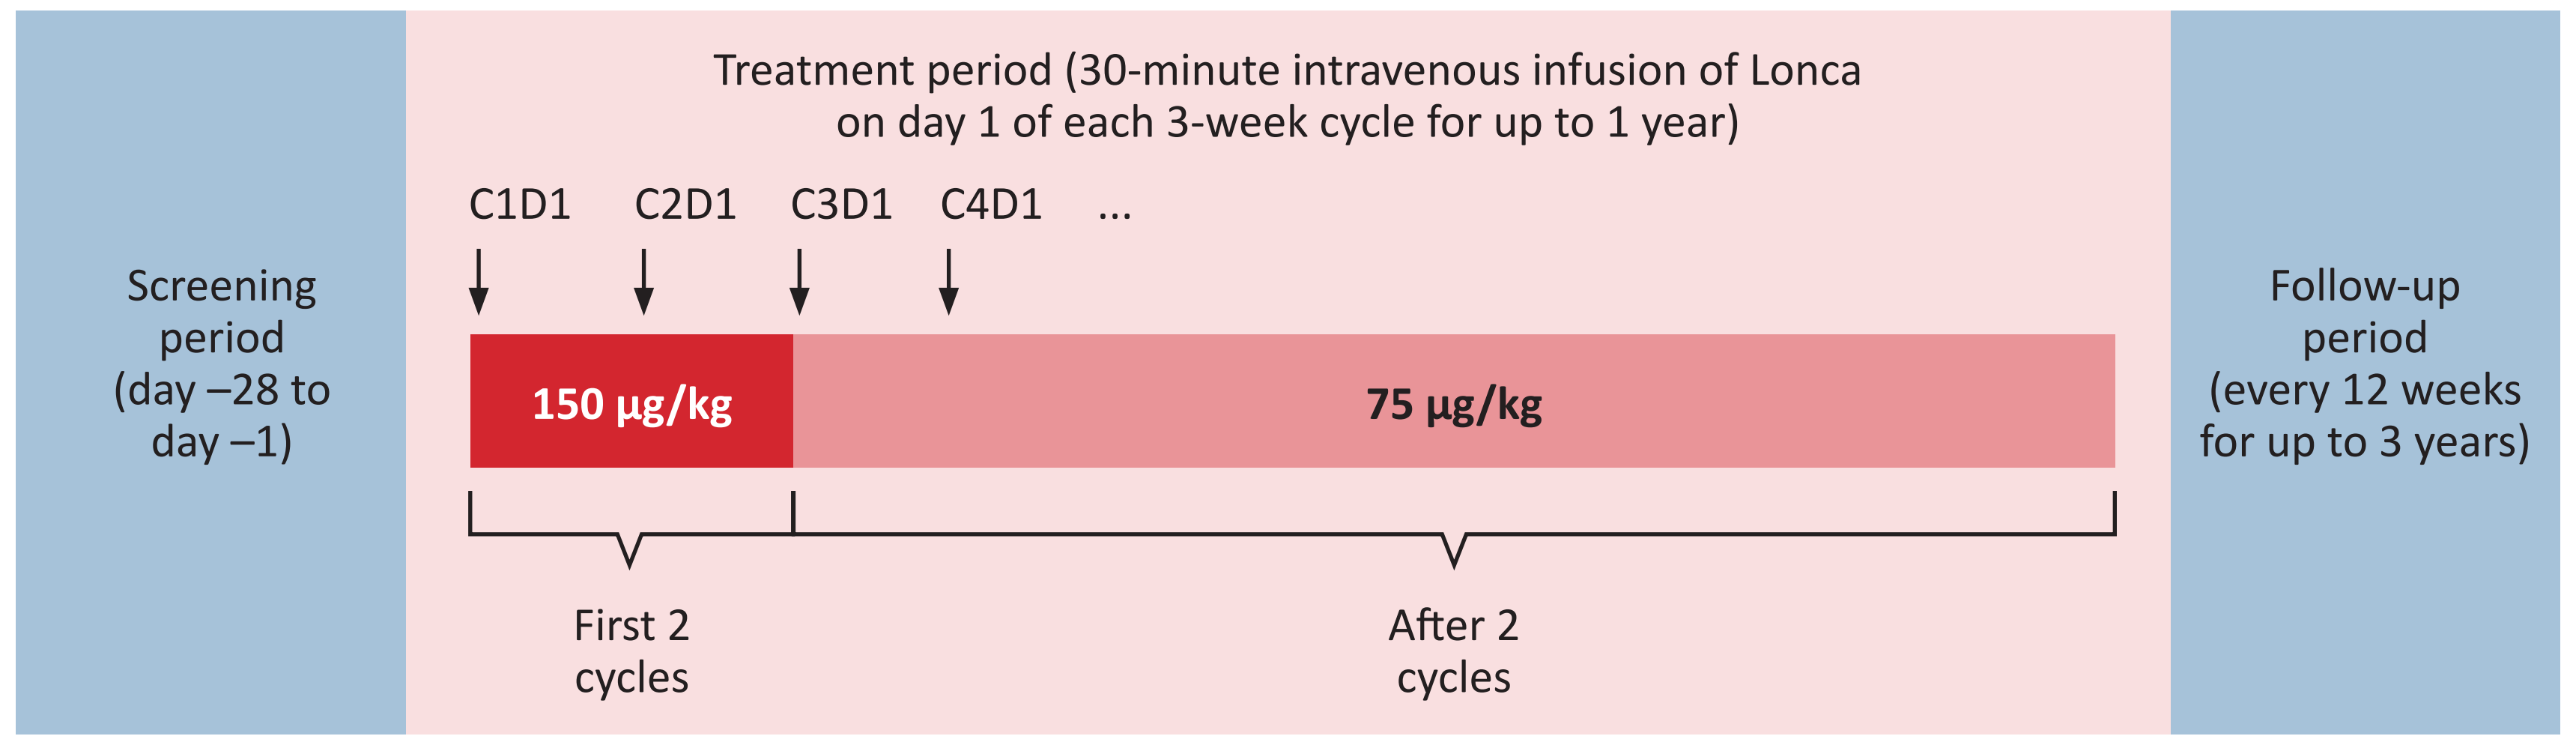
Screening period (day −28 to day −1)
Treatment period (30-minute intravenous infusion of Lonca on day 1 of each 3-week cycle for up to 1 year)
C1D1 C2D1 C3D1 C4D1 ...
150 μg/kg
75 μg/kg
First 2 cycles
After 2 cycles
Follow-up period (every 12 weeks for up to 3 years)

Notes: Patients could continue treatment for up to 1 year or until disease progression, unacceptable toxicity, or other discontinuation criteria, whichever occurred first. Patients benefiting clinically at 1 year could have continued treatment after a case-by-case review.

RESULTS

- The study enrolled 145 patients. Through cycle 9, the completion rate among patients treated at each cycle was ≥ 92% for EQ-5D and ≥ 88% for FACT-Lym. At cycle 9, 20 patients were still on treatment with Lonca, and only 5 were still on treatment at cycle 15.
- Patients with a baseline score and at least one postbaseline score were included in the analysis (Table 1).
 - Of the 130 patients included, the median age was 66.0 years, 59% were male, and 88% were White.
 - Among the 51 patients in the high-risk group, 27% were double/triple hit, 43% were primary refractory, and 47% had transformed DLBCL.
 - At baseline, the mean EQ-5D VAS score was 71.4 (standard deviation [SD] = 19.1), and the mean FACT-Lym total score was 118.4 (SD = 23.8), similar in both groups.
 - The median number of Lonca cycles administered was 4 (range, 1-26) in the high-risk group and 3 (range, 1-22) in the non–high-risk group.
- In the high-risk group, the mean changes from baseline in VAS overall health score improved at almost all visits from cycle 3 day 1 to cycle 15 day 1 (Figure 2); the mean changes from baseline in FACT-Lym total score also improved across most visits, especially between cycle 6 day 1 and cycle 8 day 1.
- In the non–high-risk group, the mean changes from baseline in VAS and FACT-Lym total remained stable or improved except for later cycles with small sample sizes (n ≤ 5).
- In the analysis adjusting for age, sex, race, and baseline score (Table 2), the least squares mean differences between high risk and non-high risk were positive for all visits (for FACT-Lym) or nearly all visits (for EQ-5D VAS) between cycle 3 day 1 and cycle 9 day 1. The *P* value is < 0.10 for FACT-Lym total at cycle 7 day 1 and cycle 8 day 1, suggesting better improvement in the high-risk group. The least squares means were not estimated after cycle 9 because of the small sample sizes (n < 20).
- When asked how much they were bothered by treatment side effects, a majority of patients reported “a little bit” or “not at all” at most cycles (71%-89% in the high-risk group and 46%-86% in the non–high-risk group) (Figure 3).

Table 1. Baseline Characteristics

	High-Risk Group (N = 51)	Non–High-Risk Group (N = 79)	Overall (N = 130)
Age, years, median (range)	65.0 (25-85)	67.0 (24-94)	66.0 (24-94)
Sex, male, n (%)	27 (53)	50 (63)	77 (59)
Race, White, n (%)	46 (90)	69 (87)	115 (88)
ECOG score, n (%)			
0	23 (45)	31 (39)	54 (42)
1	24 (47)	44 (56)	68 (52)
2	4 (8)	4 (5)	8 (6)
Prior systemic therapies, n (%)			
2 prior lines	26 (51)	33 (42)	59 (45)
3 prior lines	12 (24)	20 (25)	32 (25)
> 3 prior lines	13 (25)	26 (33)	39 (30)
Disease stage (Ann Arbor criteria),[a] n (%)			
Stage I	5 (10)	5 (6)	10 (8)
Stage II	6 (12)	15 (19)	21 (16)
Stage III	5 (10)	12 (15)	17 (13)
Stage IV	35 (69)	47 (60)	82 (63)
EQ-5D VAS, mean (SD) (score range, 0-100)	72.6 (20.0)	70.5 (18.5)	71.4 (19.1)
FACT-Lym total, mean (SD) (score range, 0-168)	119.0 (23.7)	118.0 (24.1)	118.4 (23.8)

[a] The sum of the percentages may not be 100 due to rounding.

Figure 2. Mean Change From Baseline in EQ-5D VAS and FACT-Lym Total by Visit and Risk Group

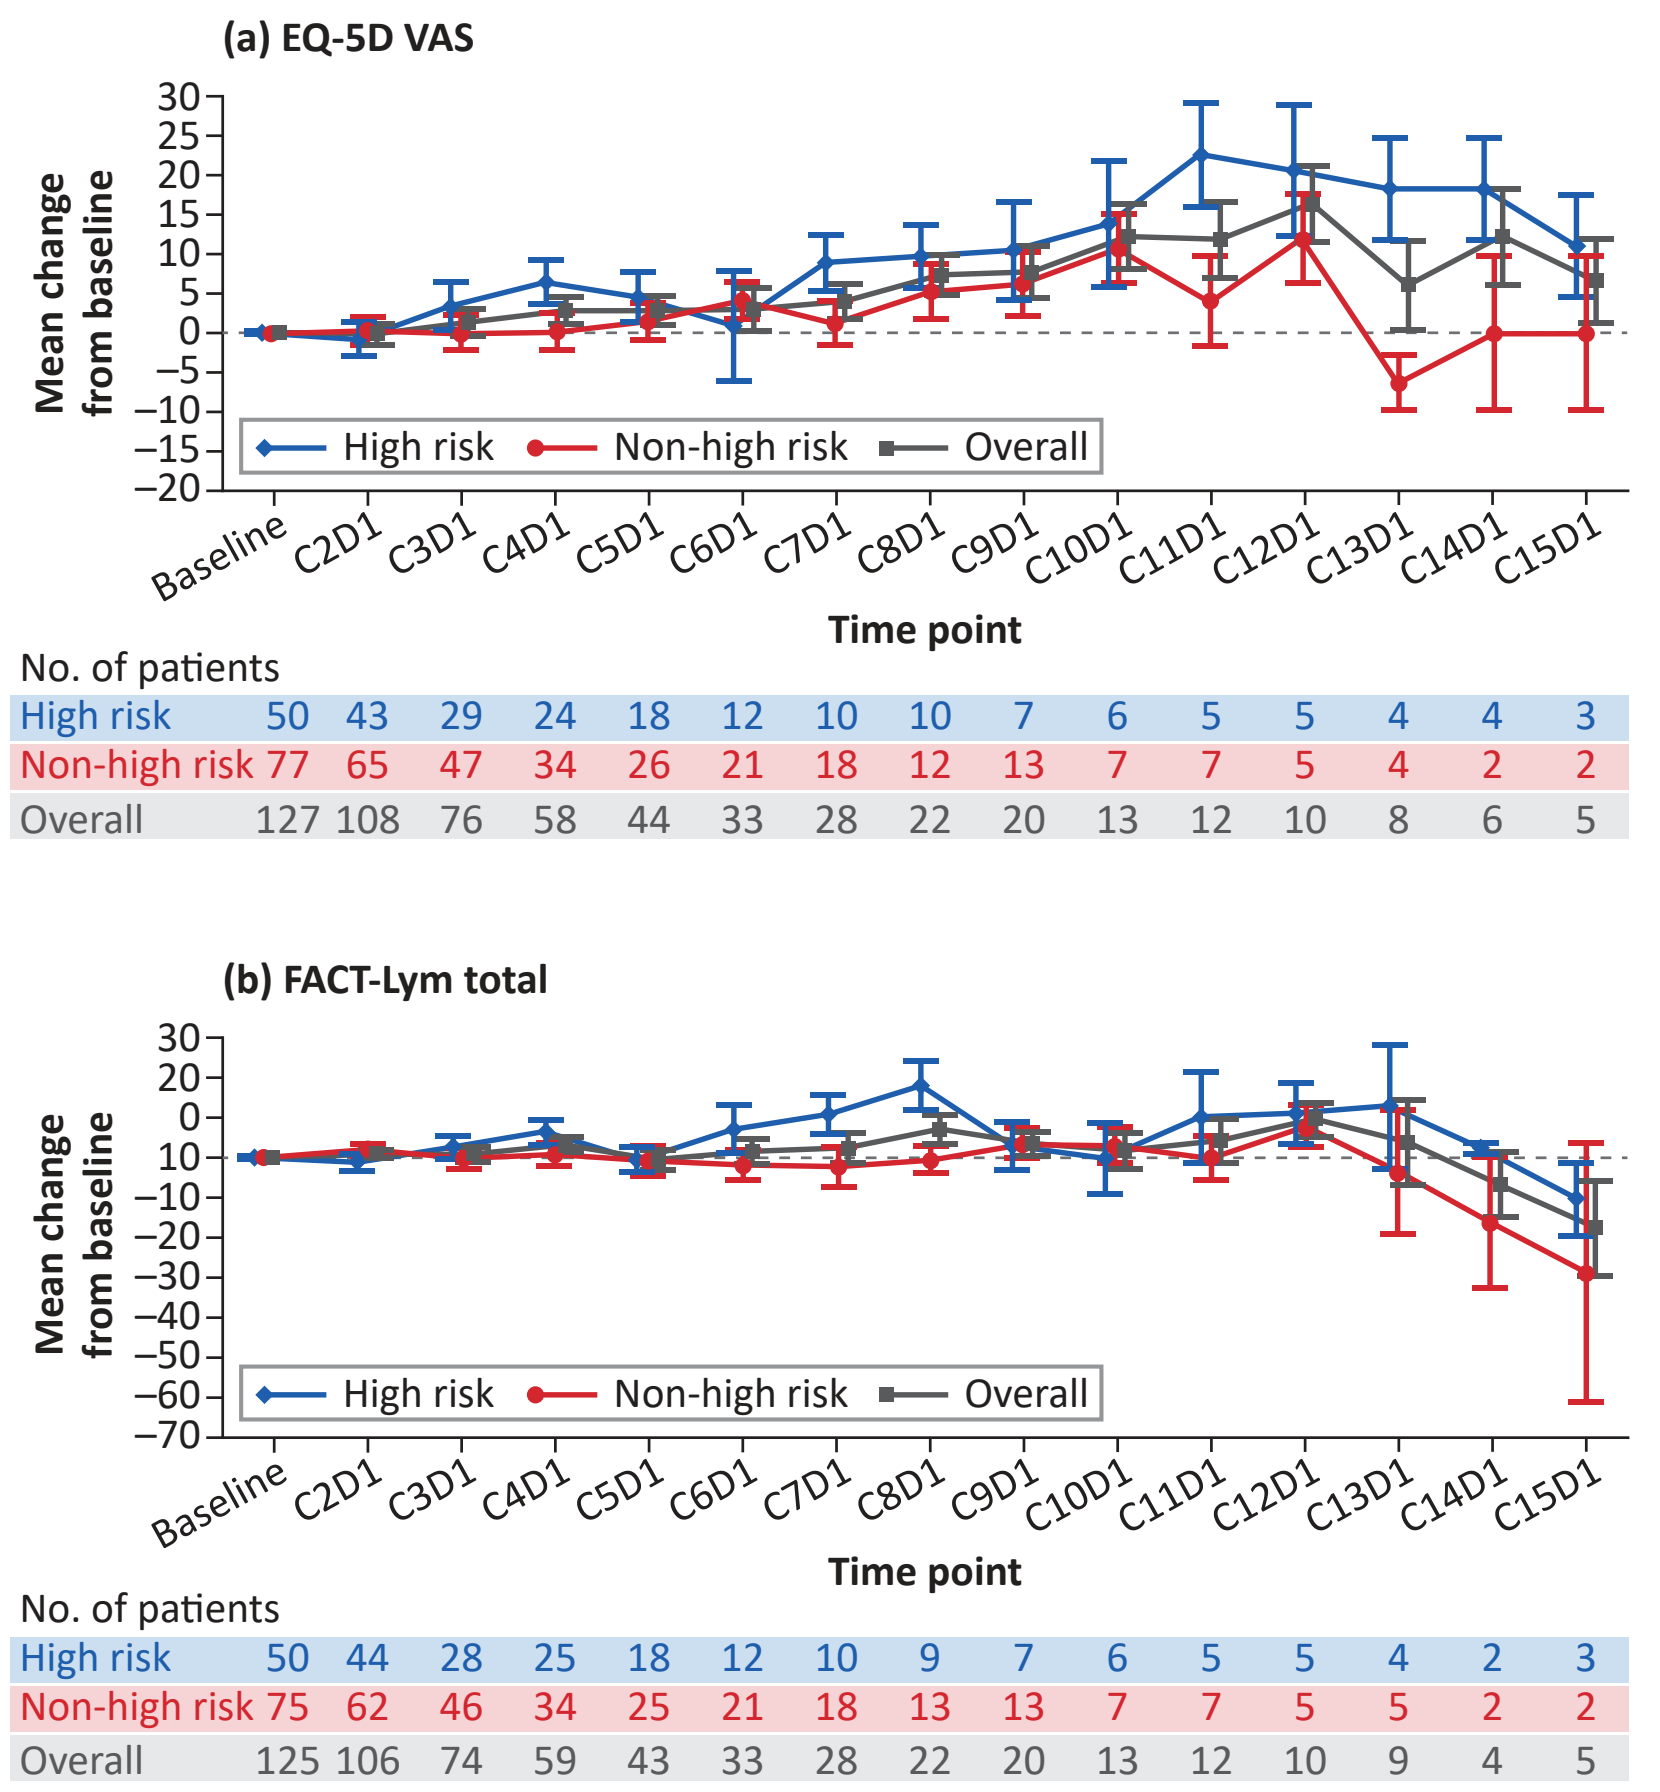
(a) EQ-5D VAS
Mean change from baseline
High risk
Non-high risk
Overall
Baseline C2D1 C3D1 C4D1 C5D1 C6D1 C7D1 C8D1 C9D1 C10D1 C11D1 C12D1 C13D1 C14D1 C15D1
Time point
No. of patients
High risk 50 43 29 24 18 12 10 10 7 6 5 5 4 4 3
Non-high risk 77 65 47 34 26 21 18 12 13 7 7 5 4 2 2
Overall 127 108 76 58 44 33 28 22 20 13 12 10 8 6 5
(b) FACT-Lym total
Mean change from baseline
High risk
Non-high risk
Overall
Baseline C2D1 C3D1 C4D1 C5D1 C6D1 C7D1 C8D1 C9D1 C10D1 C11D1 C12D1 C13D1 C14D1 C15D1
Time point
No. of patients
High risk 50 44 28 25 18 12 10 9 7 6 5 5 4 2 3
Non-high risk 75 62 46 34 25 21 18 13 13 7 7 5 5 2 2
Overall 125 106 74 59 43 33 28 22 20 13 12 10 9 4 5

Notes: Error bars indicate ± standard error. Note that sample sizes are very small in subgroups at later visits. Visits with fewer than 5 assessments in total are not displayed. A change of 7 points for the EQ-5D VAS or FACT-Lym total is considered a minimally important difference. A positive change indicates improvement.

Figure 3. Percentages of Responses to Item GP5 (“I am bothered by side effects of treatment”) by Visit and Risk Group

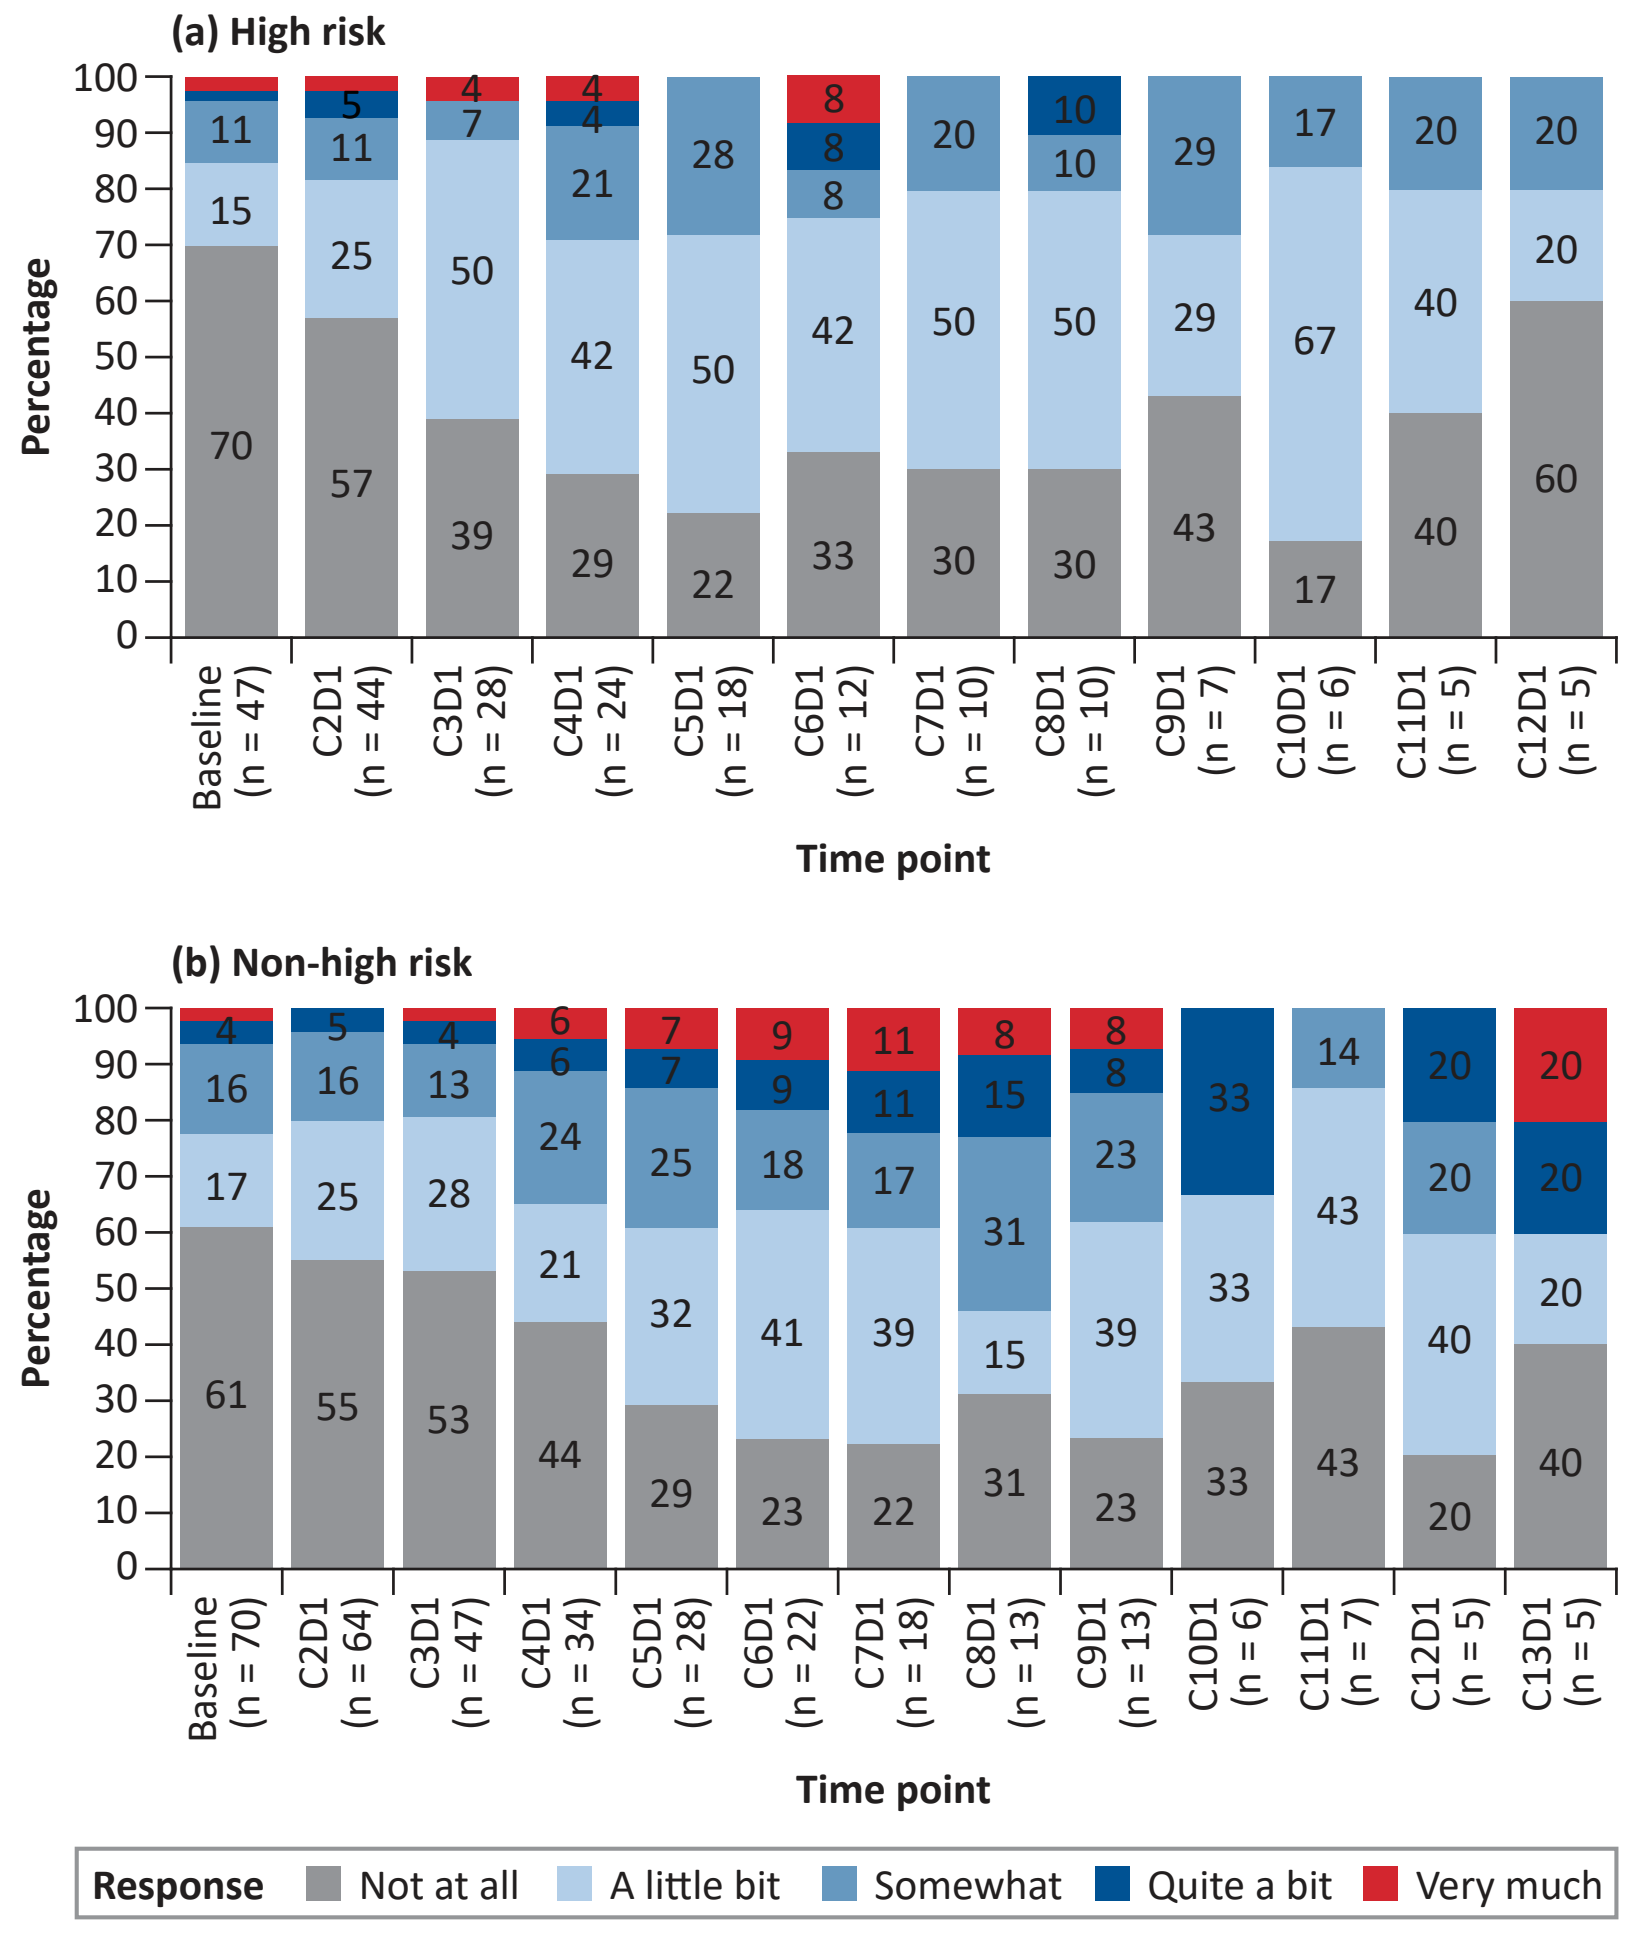
(a) High risk
Percentage
Baseline (n = 47)
C2D1 (n = 44)
C3D1 (n = 28)
C4D1 (n = 24)
C5D1 (n = 18)
C6D1 (n = 12)
C7D1 (n = 10)
C8D1 (n = 10)
C9D1 (n = 7)
C10D1 (n = 6)
C11D1 (n = 5)
C12D1 (n = 5)
Time point
(b) Non-high risk
Percentage
Baseline (n = 70)
C2D1 (n = 64)
C3D1 (n = 47)
C4D1 (n = 34)
C5D1 (n = 28)
C6D1 (n = 22)
C7D1 (n = 18)
C8D1 (n = 13)
C9D1 (n = 13)
C10D1 (n = 6)
C11D1 (n = 7)
C12D1 (n = 5)
C13D1 (n = 5)
Time point
Response
Not at all
A little bit
Somewhat
Quite a bit
Very much

Table 2. Adjusted Mean Change From Baseline Scores of EQ-5D VAS and FACT-Lym Total

		Least Squares Mean by Visit							
		C2D1	C3D1	C4D1	C5D1	C6D1	C7D1	C8D1	C9D1
EQ-5D VAS	High risk	−0.6	6.8*	7.5**	5.8	0.3	8.7	13.0*	9.9
	Non-high risk	0.1	2.1	3.3	1.5	2.5	1.6	7.7	8.5
	Difference	**−0.7**	**4.7**	**4.2**	**4.4**	**−2.2**	**7.1**	**5.3**	**1.4**
FACT-Lym total	High risk	−0.7	6.7	6.8	2.5	14.1**	24.5**	29.1**	20.6**
	Non-high risk	2.6	4.7	3.9	2.0	4.2	9.9	8.8	17.9**
	Difference	**−3.3**	**2.0**	**2.9**	**0.6**	**9.9**	**14.6***	**20.3****	**2.7**

* $0.05 < P < 0.10$; ** $P < 0.05$.

Note: Age, sex, race, and baseline score were adjusted in the analysis of covariance model. Visits after C9D1 are not displayed due to small sample sizes. A change of 7 points for the EQ-5D VAS or FACT-Lym total is considered a minimally important difference. A positive change indicates improvement.

CONCLUSIONS

- The overall health state and HRQOL were stable or improved in high-risk patients during the Lonca treatment period.
- Patients in the high-risk group tolerated the treatment just as well as, if not better than, other patients.
- The findings further support the clinical use of Lonca for the treatment of patients with high-risk R/R DLBCL.

REFERENCES

1. Caimi PF, et al. Lancet Oncol. 2021 Jun 1;22(6):790-800.
2. Spira A, et al. Clin Lymphoma Myeloma Leuk. 2021 Sep 22.
3. Pearman TP, et al. Cancer. 2018 Mar 1;124(5):991-997.
4. Food and Drug Administration. 2020. https://www.fda.gov/drugs/news-events-human-drugs/fda-asco-public-workshop-2020-clinical-outcome-assessments-cancer-clinical-trials-fifth-annual.

ACKNOWLEDGMENTS

Funding source: This research and preparation of this poster was supported by ADC Therapeutics America Inc.

CONTACT INFORMATION

Laura Liao
Director HEOR

ADC Therapeutics America Inc.
430 Mountain Avenue, Suite #404
New Providence, NJ 07974
United States

Phone: +1.908.731.5556
Email: Laura.Liao@adctherapeutics.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the presenter of this poster

EHA2022

HYBRID JUNE 9-17 VIENNA