CD25, Soluble CD25, and CCL17 as Potential Predictors of Clinical Response to Camidanlumab Tesirine in Patients With Relapsed/Refractory Classical Hodgkin Lymphoma

Jerzy Dyczkowski, PhD[1], Alex F. Herrera, MD[2*], Carmelo Carlo-Stella, MD[3], Pier Luigi Zinzani, MD, PhD[4], Marie Toukam, PhD[5], Hans G. Cruz, PhD[1], Karin Havenith, PhD[6], Joseph Boni, PhD[5], Jens Wuerthner, MD, PhD[1‡], Serafino Pantano, PhD[1]

[1]ADC Therapeutics SA, Épalinges, Switzerland; [2]City of Hope Comprehensive Cancer Center, Department of Hematology and Hematopoietic Cell Transplantation, Duarte, CA, USA; [3]Humanitas University, IRCCS Humanitas Research Hospital, Department of Oncology and Hematology, Milan, Italy; [4]University of Bologna, Institute of Hematology "L. e A. Seràgnoli" Bologna, Italy; [5]ADC Therapeutics America, Inc., Murray Hill, NJ, USA; [6]ADC Therapeutics (UK) Ltd, London, UK. [‡]Affiliation at the time the work was conducted.

4192

INTRODUCTION

- CD25 expression on the surface of various hematologic tumor cell types is well established.[1]
- In classical Hodgkin lymphoma (cHL), CD25 (interleukin-2 receptor subunit alpha) is expressed on both Hodgkin/Reed-Sternberg cells and regulatory T cells (T_{regs}) present in the tumor microenvironment.[1-3]
 - Tumors with a higher ratio of CD4+ CD25+ FOXP3+ T_{regs} to CD8+ cytotoxic T cells are believed to have poorer prognosis than tumors with a lower ratio.[1]
- Soluble CD25 (sCD25) levels are proportional to cell surface expression of CD25, and increased sCD25 has been associated with poor prognosis in cHL.[1,4]
- CCL17 (C-C motif chemokine ligand 17) on Hodgkin/Reed-Sternberg cells is a marker of cHL disease activity and correlates with metabolic tumor volume. Additionally, circulating CCL17 has been correlated with response to therapies for cHL.[5]
- Camidanlumab tesirine (Cami), an antibody-drug conjugate targeting CD25, has shown single-agent antitumor activity and manageable toxicity in a phase 2 study of relapsed/refractory (R/R) cHL.[6]
- In the phase 1 study of R/R cHL, biopsy tumor cell CD25 H-scores were statistically significantly higher in responders versus nonresponders, and the low baseline sCD25 was possibly related to response.[7]

OBJECTIVE

To assess CD25, sCD25, and CCL17 as predictive biomarkers of clinical response to Cami (at baseline or during early cycles of treatment) in patients with R/R cHL.

METHODS

STUDY DESIGN AND PARTICIPANTS

- This analysis was based on an open-label, multicenter, phase 2 study of Cami monotherapy in patients with R/R cHL after ≥3 prior lines of therapy (or ≥2 in patients ineligible for stem cell transplantation [SCT]; NCT04052997).[8]
- Cami was administered as a 30-minute intravenous (IV) infusion on day 1 of each 3-week cycle at 45 µg/kg for 2 cycles, and then 30 µg/kg for subsequent cycles.

BIOMARKER ASSESSMENT

- CD25 expression was assessed by immunohistochemistry (IHC) and H-score in all archival tumor biopsies and in a subset of recent biopsies.
 - Recent biopsies were defined as archival biopsies taken *after* the last systemic anticancer treatment, including SCT if applicable.
 - H-score is reported as a composite score taking into account the percent of positive cells and the degree of positivity as follows: negative (0), weakly (1+), moderately (2+), and strongly (3+) stained membranes. H-score, with a potential range of 0-300, was calculated as follows: H-score = [(1 × % weakly stained cells) + (2 × % moderately stained cells) + (3 × % strongly stained cells)].[9]
- Biomarker analyses were conducted on serum sCD25 and CCL17 levels, as measured by immunoassays, which were collected prior to Cami infusion on day 1 of each cycle.
 - Cami may interfere with the sCD25 assay, underestimating postbaseline sCD25.

OUTCOMES

- Assessments of CD25 IHC, sCD25, and CCL17 were performed for 111 of 117 patients treated with Cami in the phase 2 study (6 patients were not evaluable for best overall response).
- CD25 IHC, baseline sCD25 and CCL17, and fold changes in sCD25 and CCL17 from baseline were compared between responders and nonresponders to Cami.
 - Responders were defined as patients with a best overall response of complete response (CR) or partial response (PR).
 - Nonresponders were defined as patients with stable disease (SD) or progressive disease (PD).
 - Patients with a best overall response of SD who were not on-study for a minimum of 35 days after the first dose of the study drug were considered nonevaluable for best overall response and therefore were excluded from analysis.
- Analyses were conducted on a dataset based on the data cutoff of March 16, 2022 (data transfer date: April 20, 2022).

ROC ANALYSES

- Receiver operating characteristic (ROC) analyses were performed to evaluate the predictive potential of baseline sCD25 alone, as well as baseline sCD25 plus tumor cell CD25 H-score in archival biopsies as response biomarkers.
- ROC analyses were also performed using the subsets of patients with sCD25 and CCL17 at baseline, sCD25 and CCL17 change from baseline to cycle 2, day 1, and sCD25 change from baseline to cycle 3, day 1.
- Area under the curve (AUC) and Youden's index (scale 0-1) were used to measure the sensitivity and specificity of the predictive responses.

RESULTS

BIOMARKER ANALYSIS

CD25

- No statistically significant difference in tumor cell CD25 H-score was observed between responders and nonresponders in n = 99 available biopsies or the subset of recent biopsies (n = 23; $P > 0.05$ for both), despite higher median levels in responders (**Figure 1**).
- Similar to what was observed in tumor cells, no statistically significant difference in tumor-associated nontumor cell CD25 expression was found between responders and nonresponders to Cami (data not shown).

Figure 1. Tumor cell CD25 H-score in responders vs nonresponders to Cami for (A) all biopsies and (B) recent biopsies

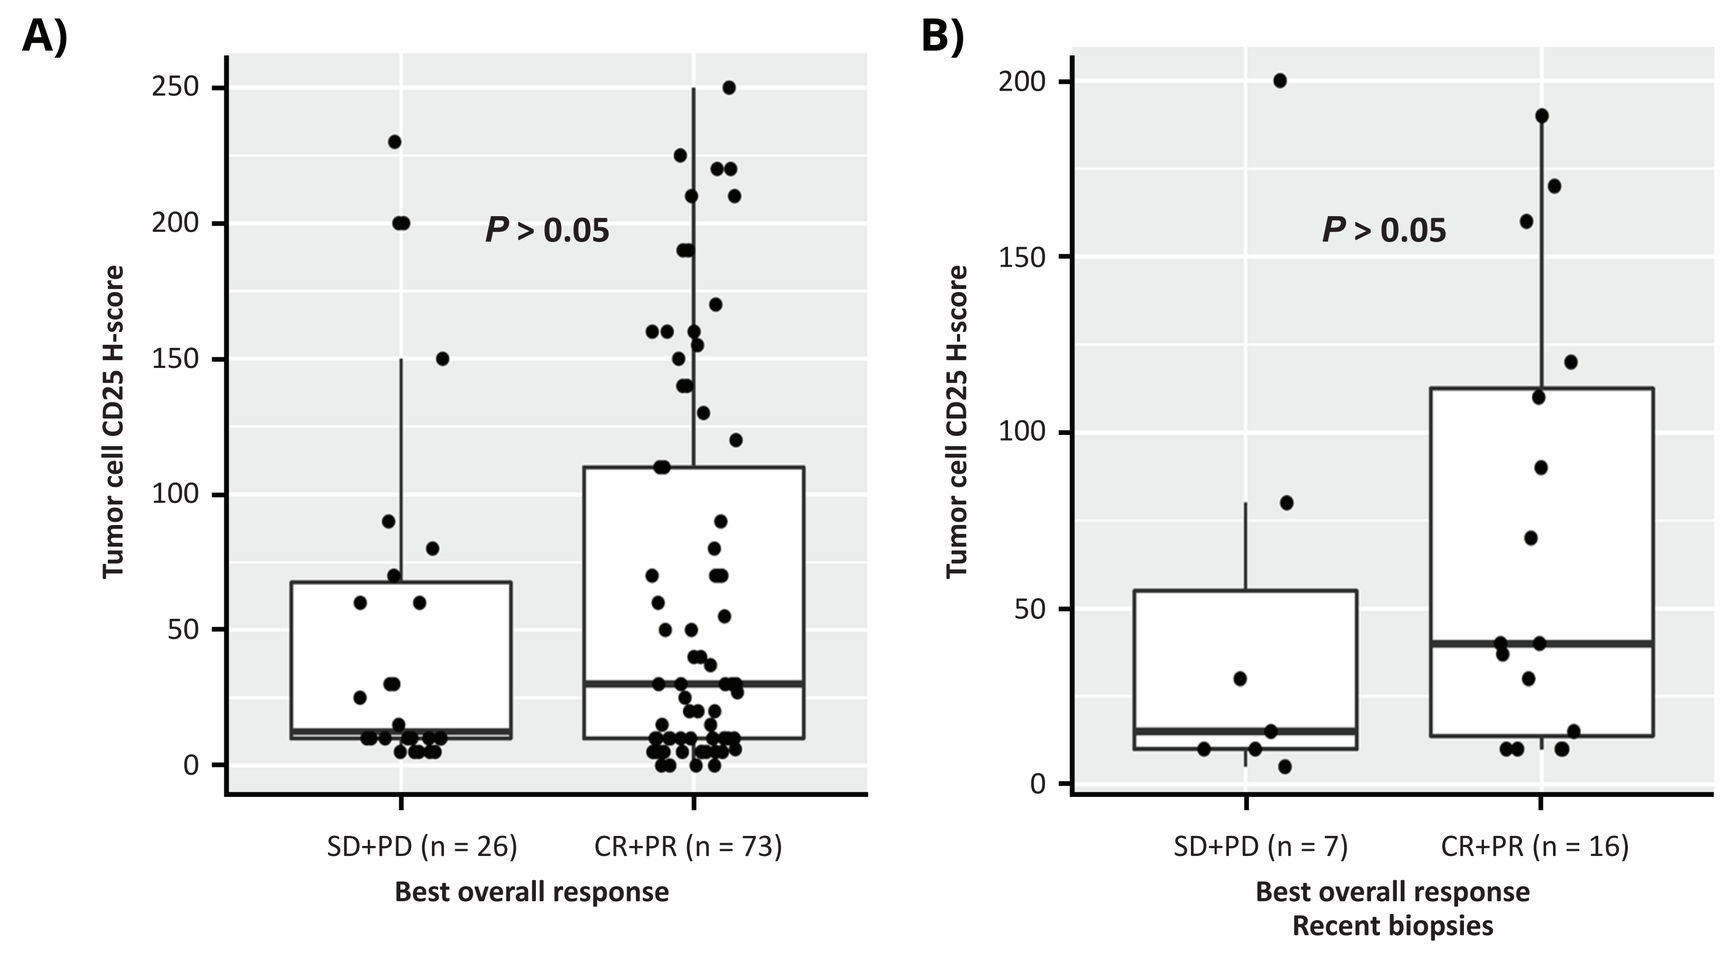

A)
P > 0.05
Tumor cell CD25 H-score
SD+PD (n = 26)
CR+PR (n = 73)
Best overall response
B)
P > 0.05
Tumor cell CD25 H-score
SD+PD (n = 7)
CR+PR (n = 16)
Best overall response
Recent biopsies

Best overall response evaluated by independent central review. Data cutoff: March 16, 2022.
CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease.

Figure 2. (A) Baseline sCD25 levels and (B) change in sCD25 from baseline to cycle 3, day 1 in responders vs nonresponders to Cami

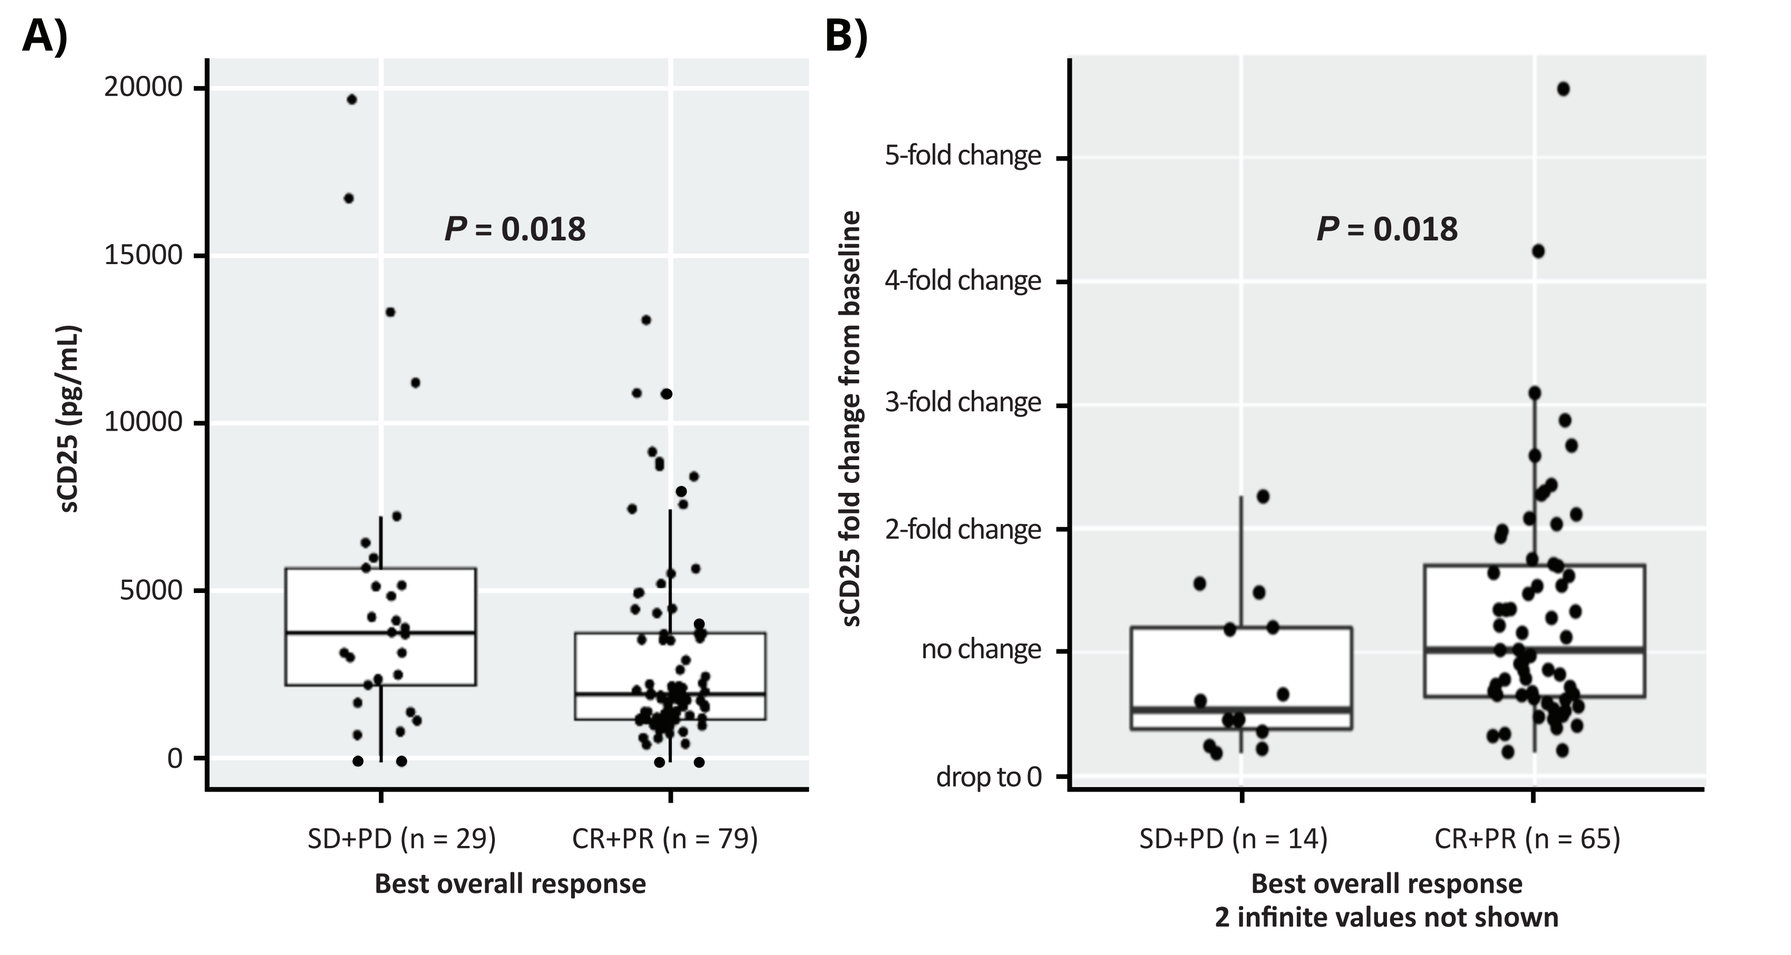

A)
P = 0.018
sCD25 (pg/mL)
SD+PD (n = 29)
CR+PR (n = 79)
Best overall response
B)
P = 0.018
sCD25 fold change from baseline
5-fold change
4-fold change
3-fold change
2-fold change
no change
drop to 0
SD+PD (n = 14)
CR+PR (n = 65)
Best overall response
2 infinite values not shown

Baseline sCD25 levels measured at cycle 1, day 1 prior to infusion of Cami. Best overall response evaluated by independent central review. Data cutoff: March 16, 2022.
CR, complete response; PD, progressive disease; PR, partial response; sCD25, soluble CD25; SD, stable disease.

sCD25

- Baseline sCD25 levels (n = 108) were statistically significantly higher in nonresponders than responders ($P = 0.018$); although, overlapping values were observed in a substantial number of patients, potentially rendering sCD25, alone, suboptimal as a potential predictive marker (**Figure 2**).
- From baseline to cycle 3, day 1, there was a statistically significantly greater decrease in sCD25 in nonresponders versus responders (n = 79; median 0.53 vs 1.02; $P = 0.018$).
 - This trend was also seen at cycle 2, day 1, but the difference was not statistically significant (n = 99; $P = 0.067$).

CCL17

- There was no statistically significant difference in baseline CCL17 levels between responders and nonresponders (n = 106; $P = 0.32$; **Figure 3**).
- From baseline to cycle 2, day 1, there was a statistically significantly greater decrease in CCL17 in responders versus nonresponders (n = 97; median 0.50 vs 0.81; $P < 0.01$).
 - Although this trend remained at cycle 3, day 1, it was not statistically significant (n = 75; $P = 0.3044$).

PREDICTIVE POTENTIAL AS RESPONSE BIOMARKERS

- ROC analyses in the subset of patients with results available for CD25 IHC and baseline sCD25 (n = 96) confirmed that, as expected from the sCD25 box plot, sCD25 alone had a modest predictive potential (AUC = 0.65) that was not substantially improved by the addition of tumor cell CD25 H-score (AUC = 0.67; **Table 1**).
- In the subset of patients with results available for sCD25 and CCL17 at baseline and cycle 2, day 1 and sCD25 at cycle 3, day 1 (n = 76), ROC curves of baseline sCD25 (AUC = 0.67), baseline sCD25 plus baseline CCL17 (AUC = 0.67), sCD25 change from baseline to cycle 2, day 1 (AUC = 0.67), and CCL17 change from baseline to cycle 2, day 1 (AUC = 0.67) showed modest predictive potential (**Table 1**).

Figure 3. (A) Baseline CCL17 levels and (B) change in CCL17 from baseline to cycle 2, day 1 in responders vs nonresponders to Cami

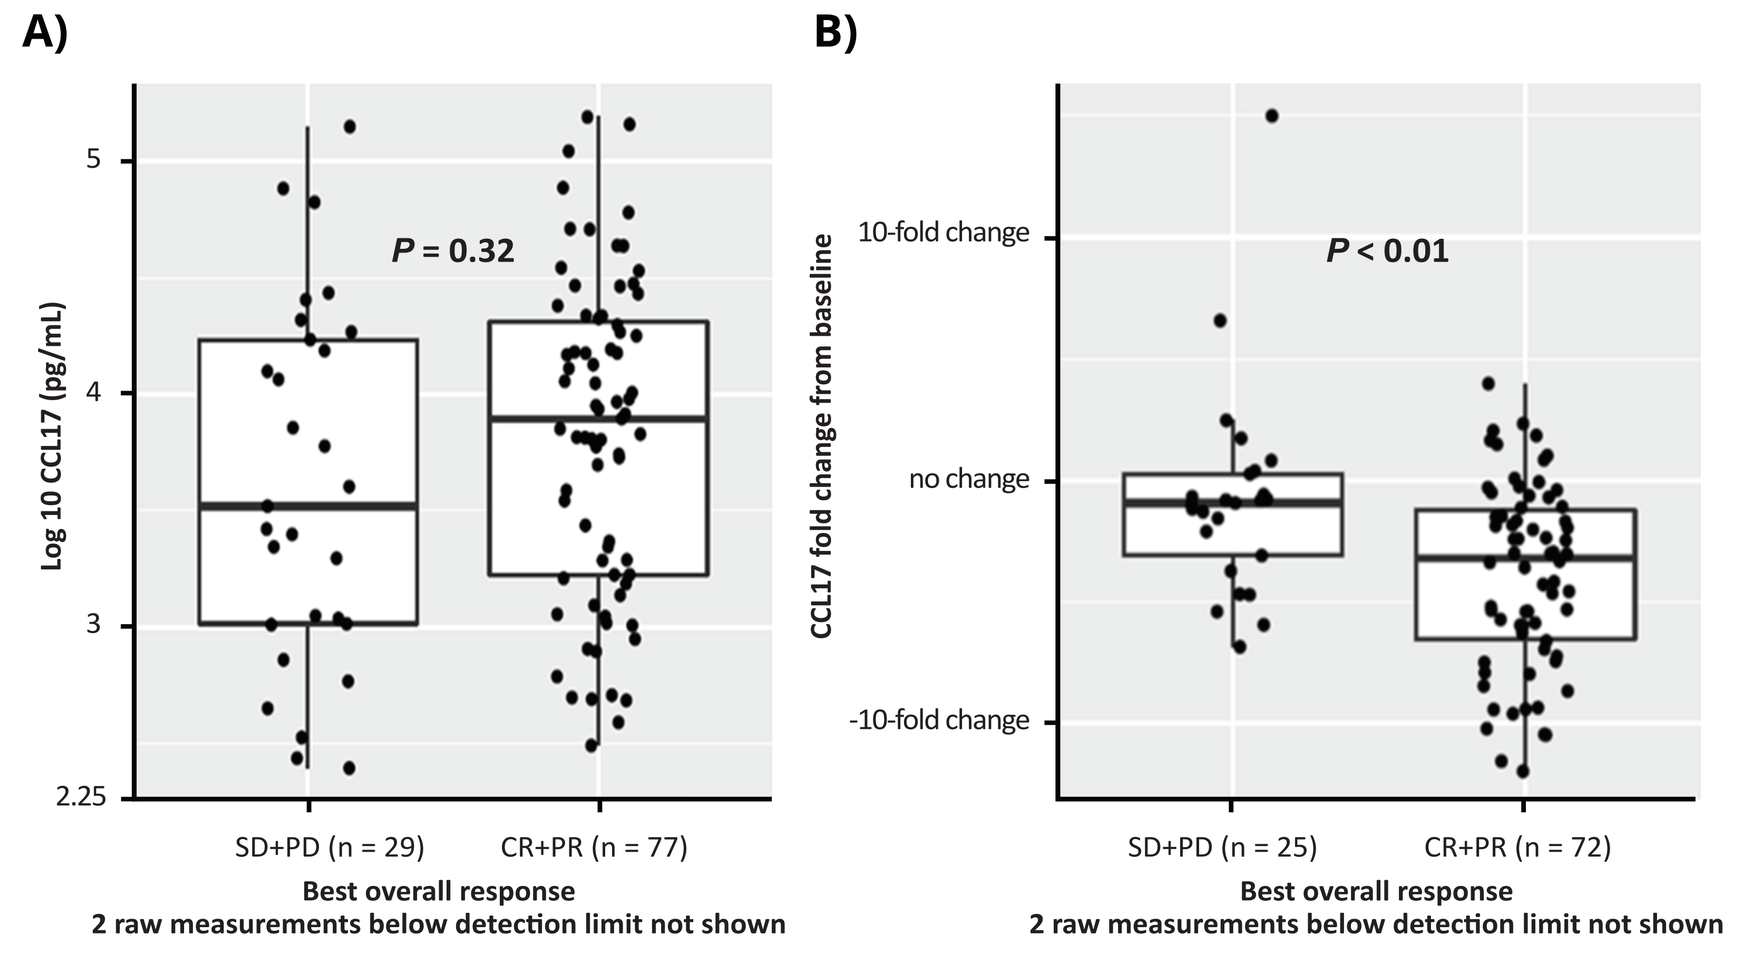

A)
P = 0.32
Log 10 CCL17 (pg/mL)
SD+PD (n = 29)
CR+PR (n = 77)
Best overall response
2 raw measurements below detection limit not shown
B)
P < 0.01
CCL17 fold change from baseline
10-fold change
no change
-10-fold change
SD+PD (n = 25)
CR+PR (n = 72)
Best overall response
2 raw measurements below detection limit not shown

CCL17 levels measured at cycle 1, day 1 prior to infusion of Cami. Best overall response evaluated by independent central review. Data cutoff: March 16, 2022.
CCL17, C-C motif chemokine ligand 17; CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease.

Table 1. Predictive potential of biomarkers using ROC analyses

Biomarkers included	n	AUC
• BL sCD25	96	0.65
• BL sCD25 • Tumor cell CD25 H-score	96	0.67
• BL sCD25	76	0.67
• BL sCD25 • BL CCL17	76	0.67
• Change from BL to C2D1 in sCD25	76	0.67
• Change from BL to C2D1 in CCL17	76	0.67
• Change from BL to C2D1 in sCD25 • Change from BL to C2D1 in CCL17	76	0.74
• BL sCD25 • BL CCL17 • Change from BL to C2D1 in sCD25 • Change from BL to C2D1 in CCL17	76	0.76
• BL sCD25 • Change from BL to C2D1 in sCD25 • Change from BL to C2D1 in CCL17	76	0.76
• BL sCD25 • Change from BL to C2D1 in sCD25 • Change from BL to C3D1 in sCD25 • Change from BL to C2D1 in CCL17	76	0.77
• BL sCD25 • BL CCL17 • Tumor cell CD25 H-score • Change from BL to C2D1 in sCD25 • Change from BL to C2D1 in CCL17	67	0.75

AUC, area under curve; BL, baseline; C, cycle; CCL17, C-C motif chemokine ligand 17; D, day; H-score, HistoScore; ROC, receiver operating characteristic; sCD25, soluble CD25.

- A reasonable model was obtained using change from baseline at cycle 2, day 1 of sCD25 and CCL17 (AUC = 0.74).
- This AUC was further increased in a model using baseline sCD25 and CCL17 plus their changes from baseline to cycle 2, day 1 (AUC = 0.76; Youden index = 0.48) and a model of baseline sCD25 plus the changes from baseline in sCD25 and CCL17 at cycle 2, day 1 (AUC = 0.76; Youden index = 0.52) (**Figure 4**).
 - The addition of tumor cell CD25 H-score did not add value to the model based on baseline sCD25 and CCL17 plus their changes from baseline to cycle 2, day 1 (AUC = 0.75).
- A model including, among other parameters, sCD25 change from baseline to cycle 3, day 1 had an AUC similar to the best models, including change from baseline to cycle 2, day 1 (AUC = 0.77; **Table 1**).
 - However, this model was considered of limited value because most responders could be identified by conventional imaging at cycle 3, day 1.

Figure 4. Model ROC analysis of (A) baseline sCD25 and CCL17 plus their change from baseline to cycle 2, day 1 and (B) baseline sCD25 plus changes from baseline to cycle 2, day 1 in sCD25 and CCL17

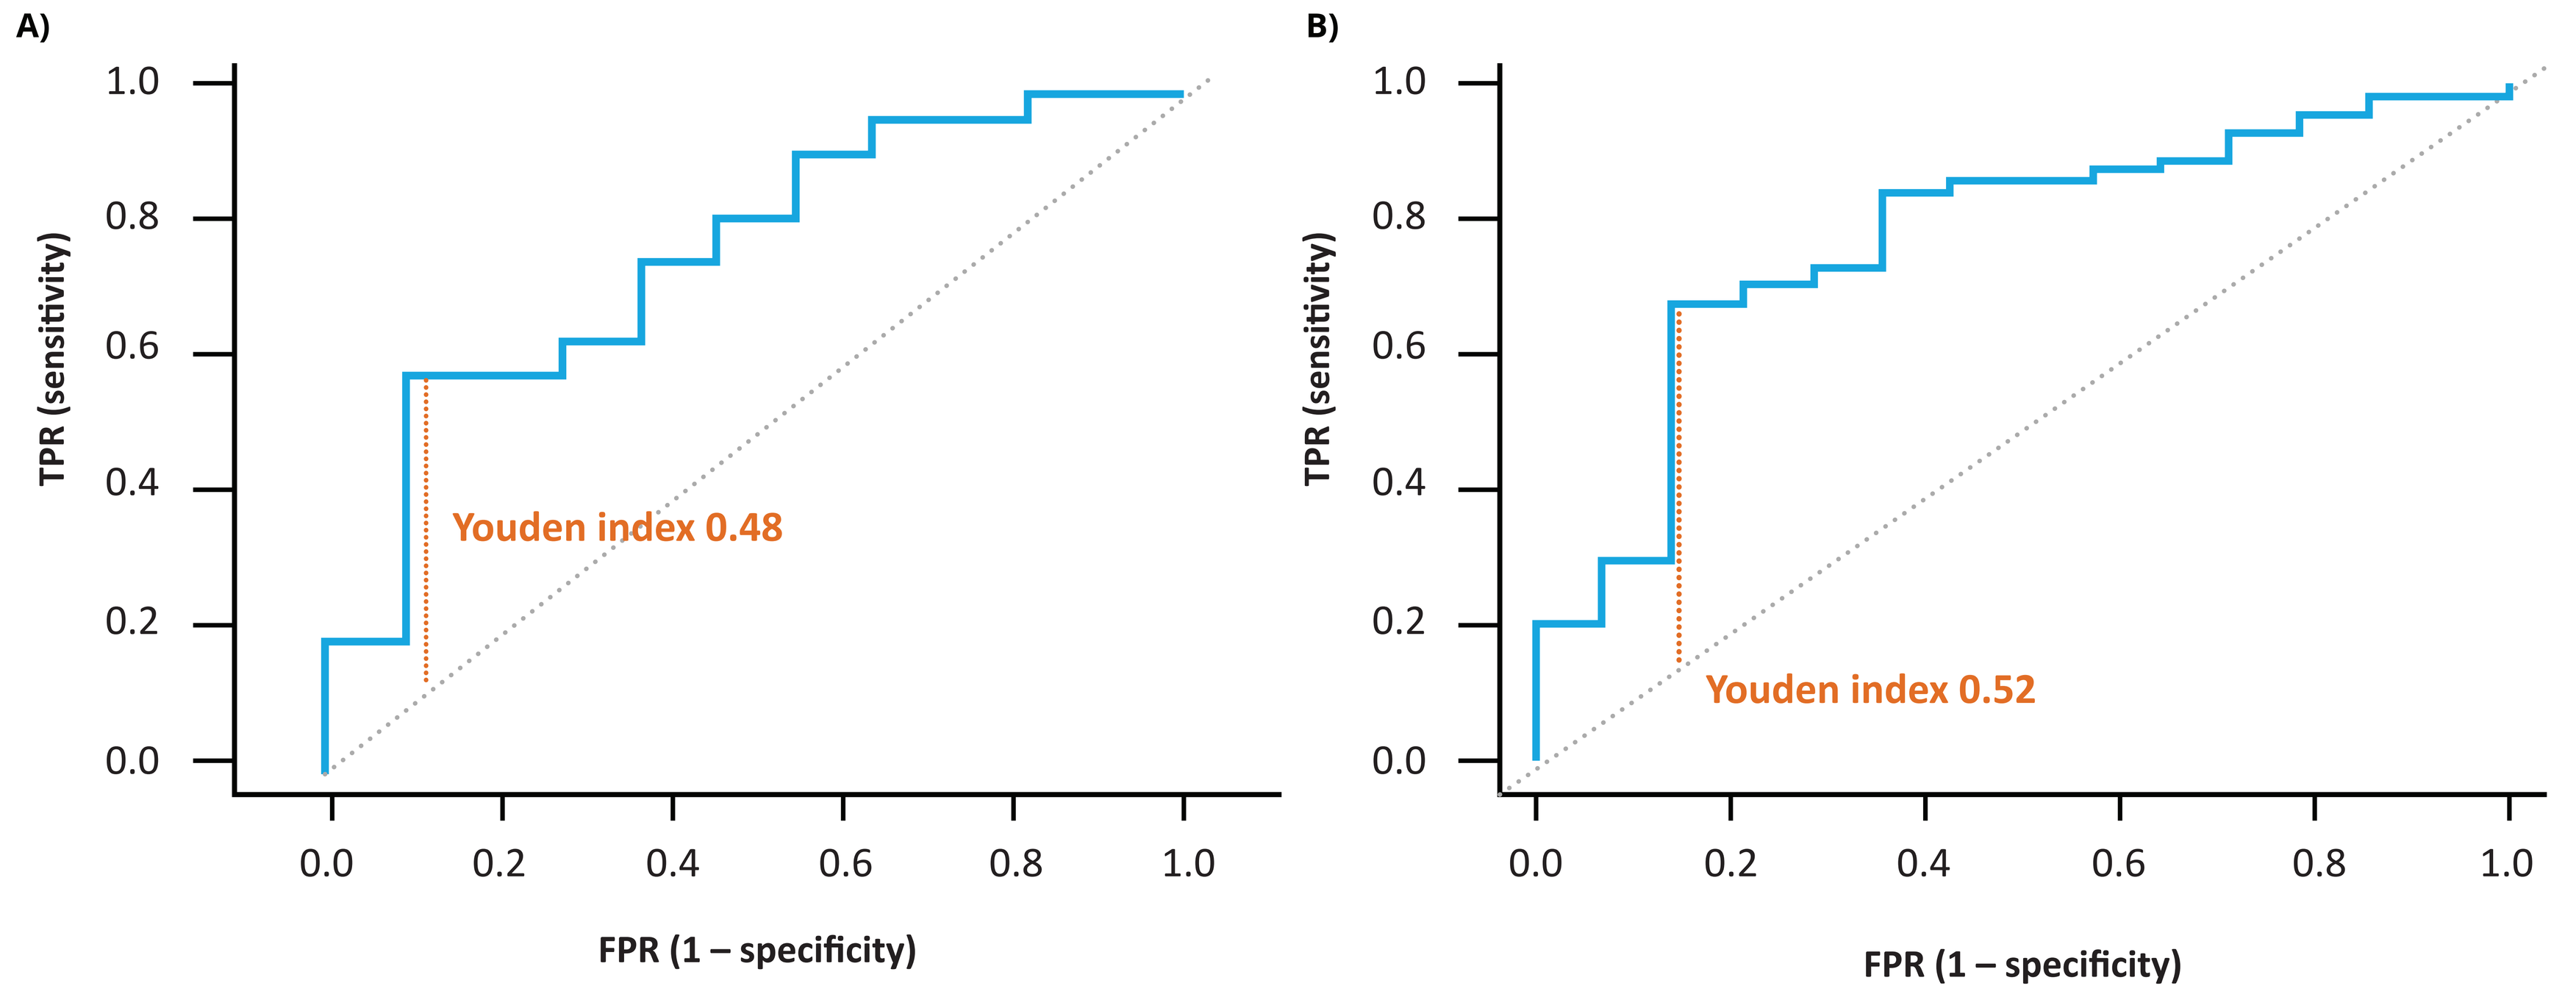

A)
TPR (sensitivity)
FPR (1 – specificity)
Youden index 0.48
B)
TPR (sensitivity)
FPR (1 – specificity)
Youden index 0.52

ROC AUC = 0.76 for both (n = 76). Data cutoff: March 16, 2022.
AUC, area under curve; CCL17, C-C motif chemokine ligand 17; FPR, false-positive rate; ROC, receiver operating characteristic; sCD25, soluble CD25; TPR, true positive rate.

CONCLUSIONS

- Higher median tumor cell CD25 H-scores were observed in responders versus nonresponders in all biopsies and the subgroup of recent biopsies, but the differences did not reach statistical significance.
 - These data warrant further investigations as phase 1 data showed statistically significantly higher tumor CD25 expression in responders.[4]
- Potential predictive models including baseline sCD25, with or without baseline CCL17, plus sCD25 and CCL17 change from baseline at cycle 2, day 1 showed the best ROC AUCs.
 - These models need further improvement to be clinically relevant. Given the high overall and early responses in most patients treated with Cami, biomarkers predicting response at cycle 2, day 1 are suboptimal unless they reach a higher sensitivity and specificity.
 - Furthermore, the predictive values of these models (or refined versions) would need further validation in an independent validation cohort as present results are based on a training cohort alone.
- Nonresponders had statistically significantly higher baseline sCD25; however, this parameter alone has an insufficient predictive potential for Cami's efficacy.
- Additional investigations are ongoing to better understand the following:
 - The effect and significance of different levels of sCD25 in the context of Cami's mode of action and exposure-response;
 - The potential value of the changes in sCD25 and CCL17 as early indicators of Cami's effect and as biomarkers for disease monitoring and response.

Acknowledgments

ADC Therapeutics SA; medical writing support: CiTRUS Health Group.

References

1. Flynn MJ, et al. *Br J Haematol*. 2017;179:20-35.
2. Connors JM, et al. *Nat Rev Dis Primers*. 2020;6:61.
3. Strauchen JA, et al. *Am J Pathol*. 1987;126:506-512.
4. Ambrosetti A, et al. *Cancer*. 1993;72:201-206.
5. Plattel WJ, et al. *Br J Haematol*. 2016;175:868-875.
6. Carlo-Stella C, et al. *Hemasphere*. 2022;6:102-103.
7. Hamadani M, et al. *Lancet Haematol*. 2021;8:e433-e445.
8. Clinicaltrials.gov. Study to evaluate the efficacy and safety of camidanlumab tesirine (ADCT-301) in patients with relapsed or refractory Hodgkin lymphoma. Updated March 8, 2022. https://clinicaltrials.gov/ct2/show/NCT04052997. Accessed October 12, 2022.
9. Jensen K, et al. *Mod Pathol*. 2017;30:180-193.

*Contact Information:

Alex F. Herrera, MD: aherrera@coh.org

Copies of this poster obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the presenter of this poster.

Poster presented at the 64th Annual American Society of Hematology (ASH) Meeting. December 10-13, 2022, New Orleans, LA USA