Identification of predictive biomarkers for response of R/R DLBCL patients treated with loncastuximab tesirine using low-pass whole-genome cell-free DNA sequencing (cfDNA-WGS)

Francesco Vallania[1], Victoria Cheung[1], Michael D Zamba[1], Joyce Liu[1], Amit Pasupathy[1], Hayley Donnella[1], Mitch Bailey[1], Maggie Louie[1], Jimmy Lin[1], Karin Havenith[2], Yajuan Qin[2], Serafino Pantano[2], Jens Wuerthner[2], Patrick H.van Berkel[2]

[1]Freenome, 279 East Grand Avenue, 5th Floor, South San Francisco, CA, USA; [2]ADC Therapeutics SA, Biopole, Route de la Corniche 3B, 1066 Epalinges, Switzerland

INTRODUCTION AND OBJECTIVES

- Plasma profiling can non-invasively identify biomarkers associated with treatment outcomes and reveal mechanisms underlying drug resistance.
- Loncastuximab tesirine (lonca), an antibody-drug conjugate comprising a humanized anti-CD19 antibody and a pyrrolobenzodiazepine dimer cytotoxin, is indicated for relapsed/refractory (R/R) diffuse large B-cell lymphoma (DLBCL) after ≥2 systemic treatments.
- The objective of this study was to investigate biological signatures associated with response to lonca using plasma-derived cell-free DNA (cfDNA) samples from 145 R/R DLBCL patients in a phase 2 trial (NCT3589469, LOTIS-2).

METHODS

- Cell-free DNA (cfDNA) was extracted from 100 patients at baseline and 80 patients on treatment (cycle 2 day 1; C2D1).
- Low-pass whole-genome sequencing was performed to characterize cfDNA fragments, which reflect nucleosome protection and chromatin state.
- Gene activation for protein-coding genes was inferred from fragment distribution around transcription start sites[1] to generate Transcription Start Site Gene Activation Probability scores (TSS-GAP; a schematic of the approach is shown in **Figure 1**).
- Binding activity of 504 cancer-associated transcription factors (TFs) was estimated by inferring the level of chromatin accessibility from ~1000 binding sites per factor across the genome to give Transcription Factor Binding Association (TFBA) scores[2].
- Class comparison statistics for TSS-GAP and TFBA features were performed using the Wilcoxon signed rank test for paired longitudinal comparisons.
- Gene Set Enrichment Analysis (GSEA) was performed using Molecular Signatures Database (MSigDB) sets. P-values were corrected for multiple hypothesis testing to control the False Discovery Rate (FDR).
- A multivariate classifier for response assessment according to 2014 Lugano classification was constructed by applying L1-regularized logistic regression on both TSS-GAP and TFBA features (as in **Figure 1**).
- Clinical lab data including Chem-20 panel results, complete blood count results, and additional immunophenotyping (CD5/19/20/21/23/43, BCL1/2/6, Cyclin D1, ZAP-70) were incorporated into the multivariate modeling by concatenating these values with the TSS-GAP scores.

Table 1. Cohort description and profiling

	CR/PR	SD/PD	p-value
Age	63 (23-83)	63 (24-82)	0.96[a]
Sex			
Male	31 (70.5%)	26 (63.4%)	0.65[b]
Female	13 (29.5%)	15 (36.6%)	0.65[b]
Prior Therapies			
Rituximab-CHOP	25 (61%)	32 (72.7%)	0.97[b]
Rituximab-GDP	6 (14.6%)	8 (18.2%)	0.97[b]
Rituximab-ICE	13 (31.7%)	15 (34.1%)	0.97[b]

[a]1-way ANOVA
[b]Chi-square frequency test

Figure 1. Cohort description and profiling

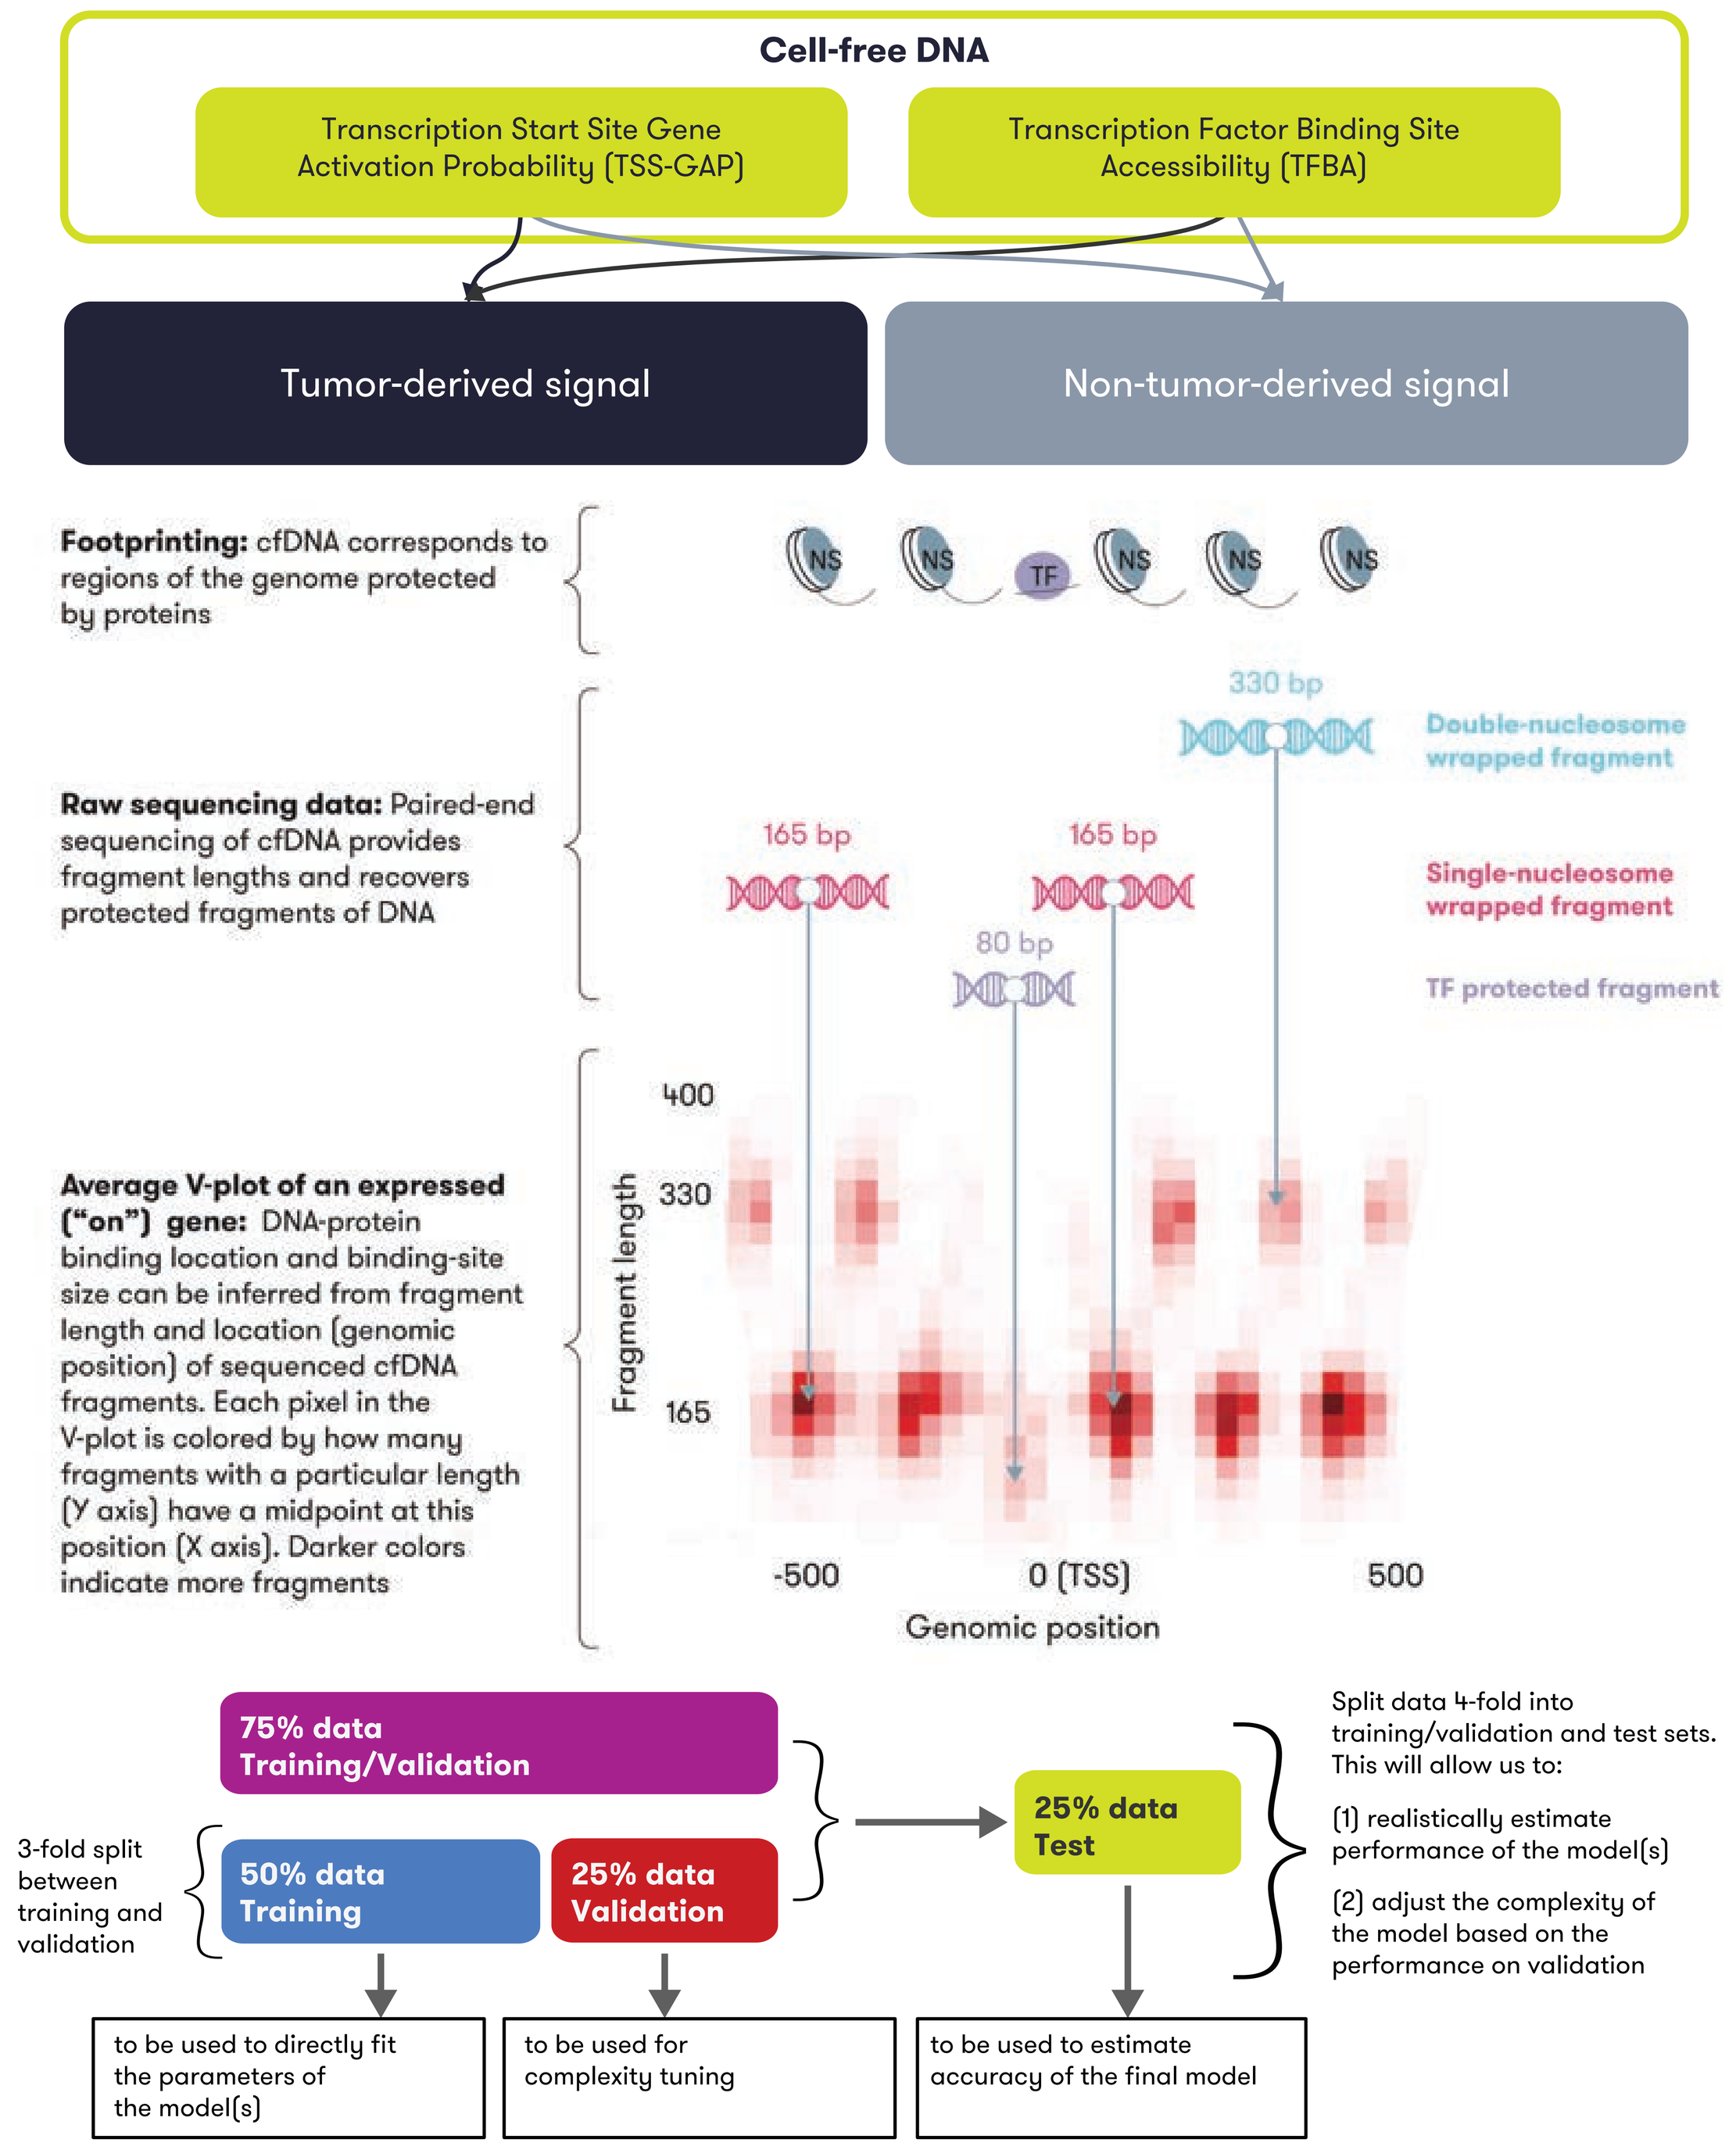
Cell-free DNA
Transcription Start Site Gene Activation Probability (TSS-GAP)
Transcription Factor Binding Site Accessibility (TFBA)
Tumor-derived signal
Non-tumor-derived signal
Footprinting: cfDNA corresponds to regions of the genome protected by proteins
NS
TF
Raw sequencing data: Paired-end sequencing of cfDNA provides fragment lengths and recovers protected fragments of DNA
330 bp
165 bp
80 bp
Double-nucleosome wrapped fragment
Single-nucleosome wrapped fragment
TF protected fragment
Average V-plot of an expressed ("on") gene: DNA-protein binding location and binding-site size can be inferred from fragment length and location (genomic position) of sequenced cfDNA fragments. Each pixel in the V-plot is colored by how many fragments with a particular length (Y axis) have a midpoint at this position (X axis). Darker colors indicate more fragments
Fragment length
400
330
165
-500
0 (TSS)
500
Genomic position
75% data Training/Validation
3-fold split between training and validation
50% data Training
25% data Validation
25% data Test
Split data 4-fold into training/validation and test sets. This will allow us to:
(1) realistically estimate performance of the model(s)
(2) adjust the complexity of the model based on the performance on validation
to be used to directly fit the parameters of the model(s)
to be used for complexity tuning
to be used to estimate accuracy of the final model

TF = Transcription Factor (small region protected)
NS = Nucleosome (large region protected)

- Regression modeling strategy including training/validation folds
- Logistic regression using L1 regularization

Figure 2. Lonca treatment reduces cfDNA-estimated levels of CD19 but not of other immune markers

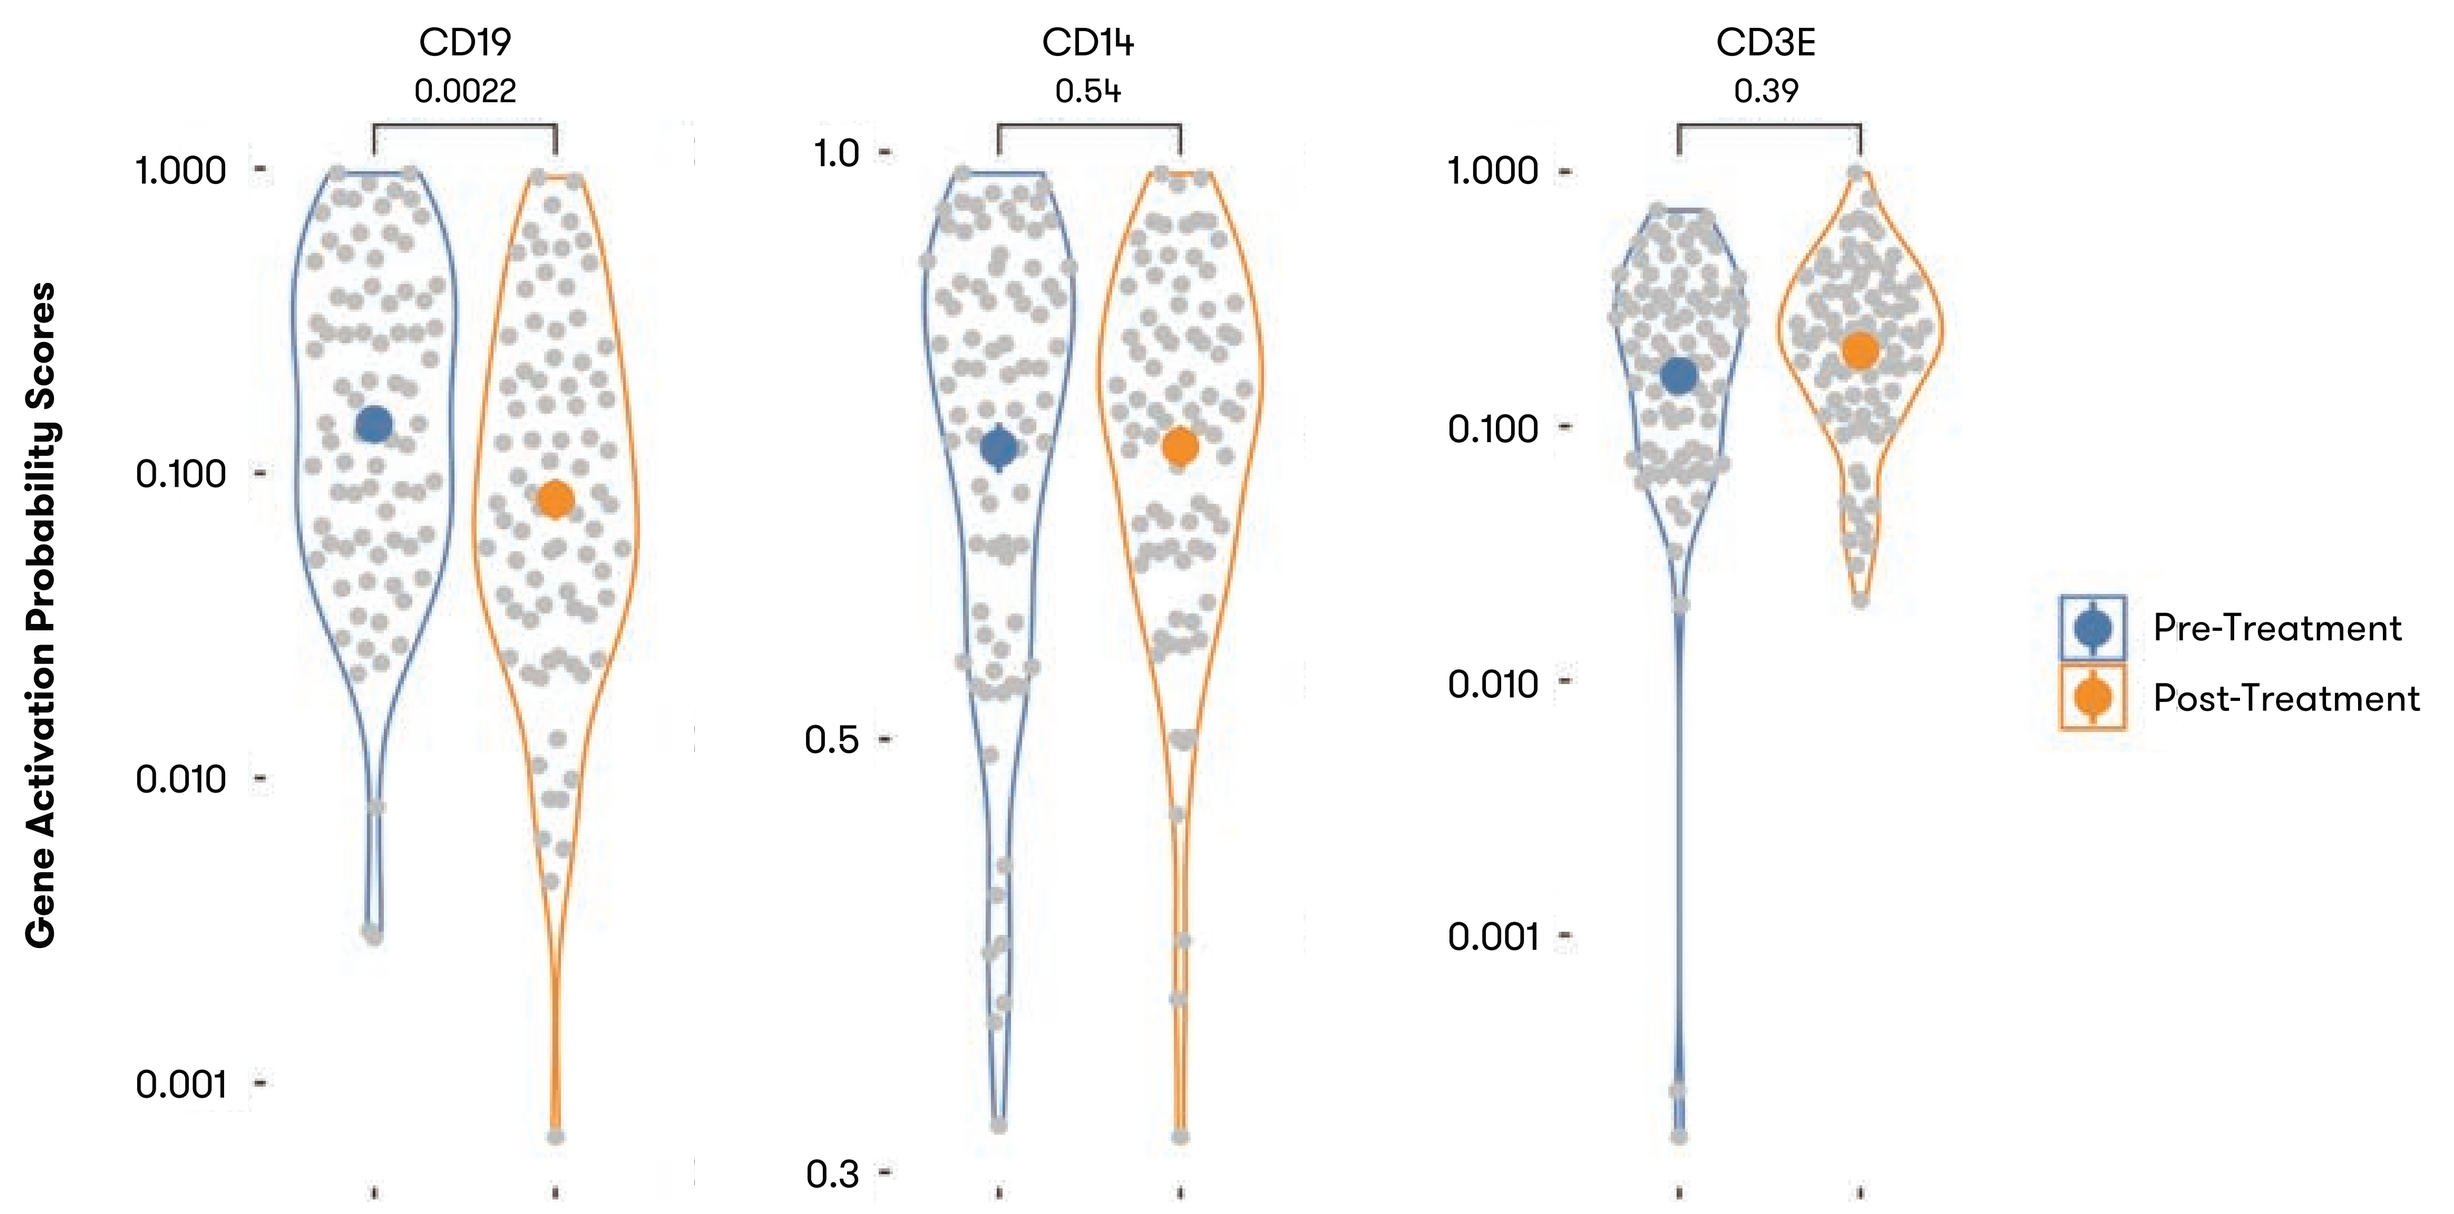
CD19
0.0022
CD14
0.54
CD3E
0.39
Gene Activation Probability Scores
1.000
0.100
0.010
0.001
1.0
0.5
0.3
Pre-Treatment
Post-Treatment

- We evaluated longitudinal changes in TSS-GAP scores occurring during treatment with lonca between C2D1 and baseline
- Estimated levels of CD19, a proxy for B-cell abundance, significantly decreased after treatment (Wilcoxon signed-rank paired test: p = 2.2E-3), which supports the hypothesis that lonca is actively reducing B-cells
- Markers of two other immune populations showed no significant change (CD14, p = 0.54; CD3E, p = 0.39).

Figure 3. Gene set enrichment analysis reveals significantly decreased signatures for B-cell gene sets in treated cases compared to baseline

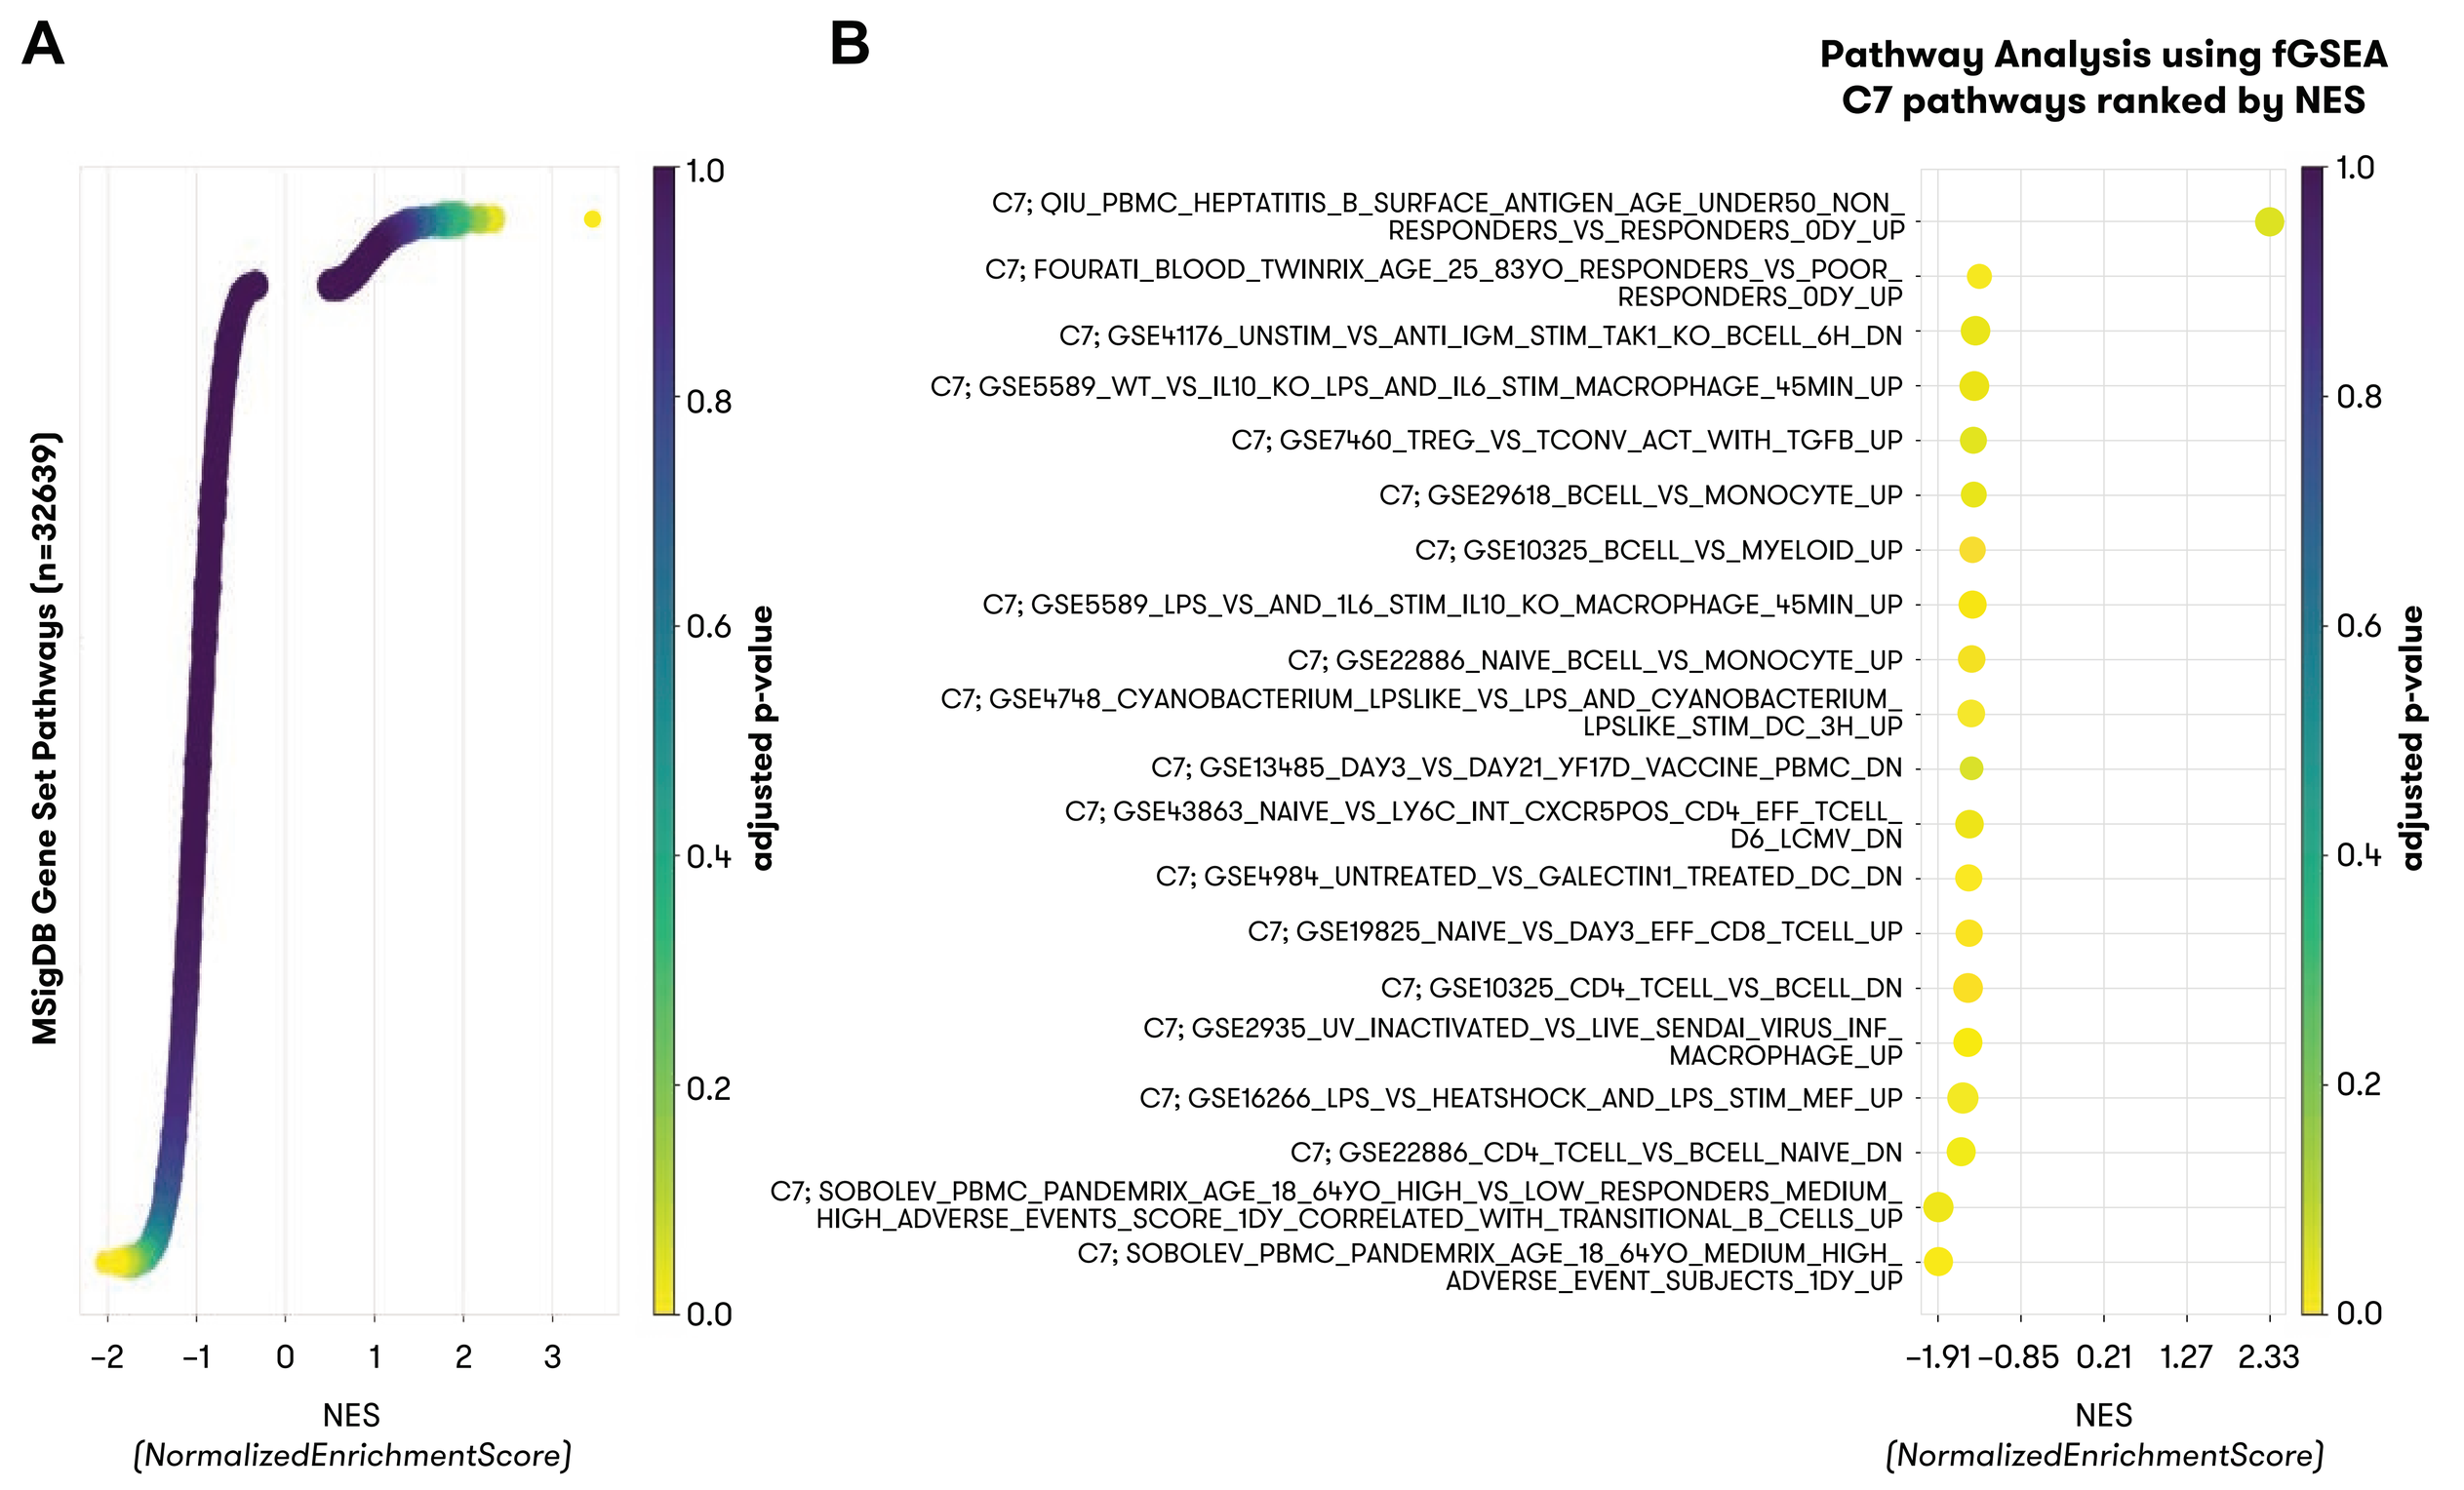
A
B
MSigDB Gene Set Pathways (n=32639)
NES
(NormalizedEnrichmentScore)
adjusted p-value
Pathway Analysis using fGSEA
C7 pathways ranked by NES
C7; QIU_PBMC_HEPTATITIS_B_SURFACE_ANTIGEN_AGE_UNDER50_NON_RESPONDERS_VS_RESPONDERS_0DY_UP
C7; FOURATI_BLOOD_TWINRIX_AGE_25_83YO_RESPONDERS_VS_POOR_RESPONDERS_0DY_UP
C7; GSE41176_UNSTIM_VS_ANTI_IGM_STIM_TAK1_KO_BCELL_6H_DN
C7; GSE5589_WT_VS_IL10_KO_LPS_AND_IL6_STIM_MACROPHAGE_45MIN_UP
C7; GSE7460_TREG_VS_TCONV_ACT_WITH_TGFB_UP
C7; GSE29618_BCELL_VS_MONOCYTE_UP
C7; GSE10325_BCELL_VS_MYELOID_UP
C7; GSE5589_LPS_VS_AND_1L6_STIM_IL10_KO_MACROPHAGE_45MIN_UP
C7; GSE22886_NAIVE_BCELL_VS_MONOCYTE_UP
C7; GSE4748_CYANOBACTERIUM_LPSLIKE_VS_LPS_AND_CYANOBACTERIUM_LPSLIKE_STIM_DC_3H_UP
C7; GSE13485_DAY3_VS_DAY21_YF17D_VACCINE_PBMC_DN
C7; GSE43863_NAIVE_VS_LY6C_INT_CXCR5POS_CD4_EFF_TCELL_D6_LCMV_DN
C7; GSE4984_UNTREATED_VS_GALECTIN1_TREATED_DC_DN
C7; GSE19825_NAIVE_VS_DAY3_EFF_CD8_TCELL_UP
C7; GSE10325_CD4_TCELL_VS_BCELL_DN
C7; GSE2935_UV_INACTIVATED_VS_LIVE_SENDAI_VIRUS_INF_MACROPHAGE_UP
C7; GSE16266_LPS_VS_HEATSHOCK_AND_LPS_STIM_MEF_UP
C7; GSE22886_CD4_TCELL_VS_BCELL_NAIVE_DN
C7; SOBOLEV_PBMC_PANDEMRIX_AGE_18_64YO_HIGH_VS_LOW_RESPONDERS_MEDIUM_HIGH_ADVERSE_EVENTS_SCORE_1DY_CORRELATED_WITH_TRANSITIONAL_B_CELLS_UP
C7; SOBOLEV_PBMC_PANDEMRIX_AGE_18_64YO_MEDIUM_HIGH_ADVERSE_EVENT_SUBJECTS_1DY_UP
-1.91 -0.85 0.21 1.27 2.33

- GSEA identified 157 significantly enriched sets (152 reduced after treatment; False Discovery Rate (FDR) < 5%), including B-cell gene set signatures decreasing from baseline
- The 20 significantly enriched immunologic signatures (C7) gene sets (FDR < 5%) reveal downregulation of B-cell-related sets amongst other signatures of immune suppression compared to baseline.

Figure 4. Multivariate classifier distinguishes responders from non-responders at baseline in cross-validation

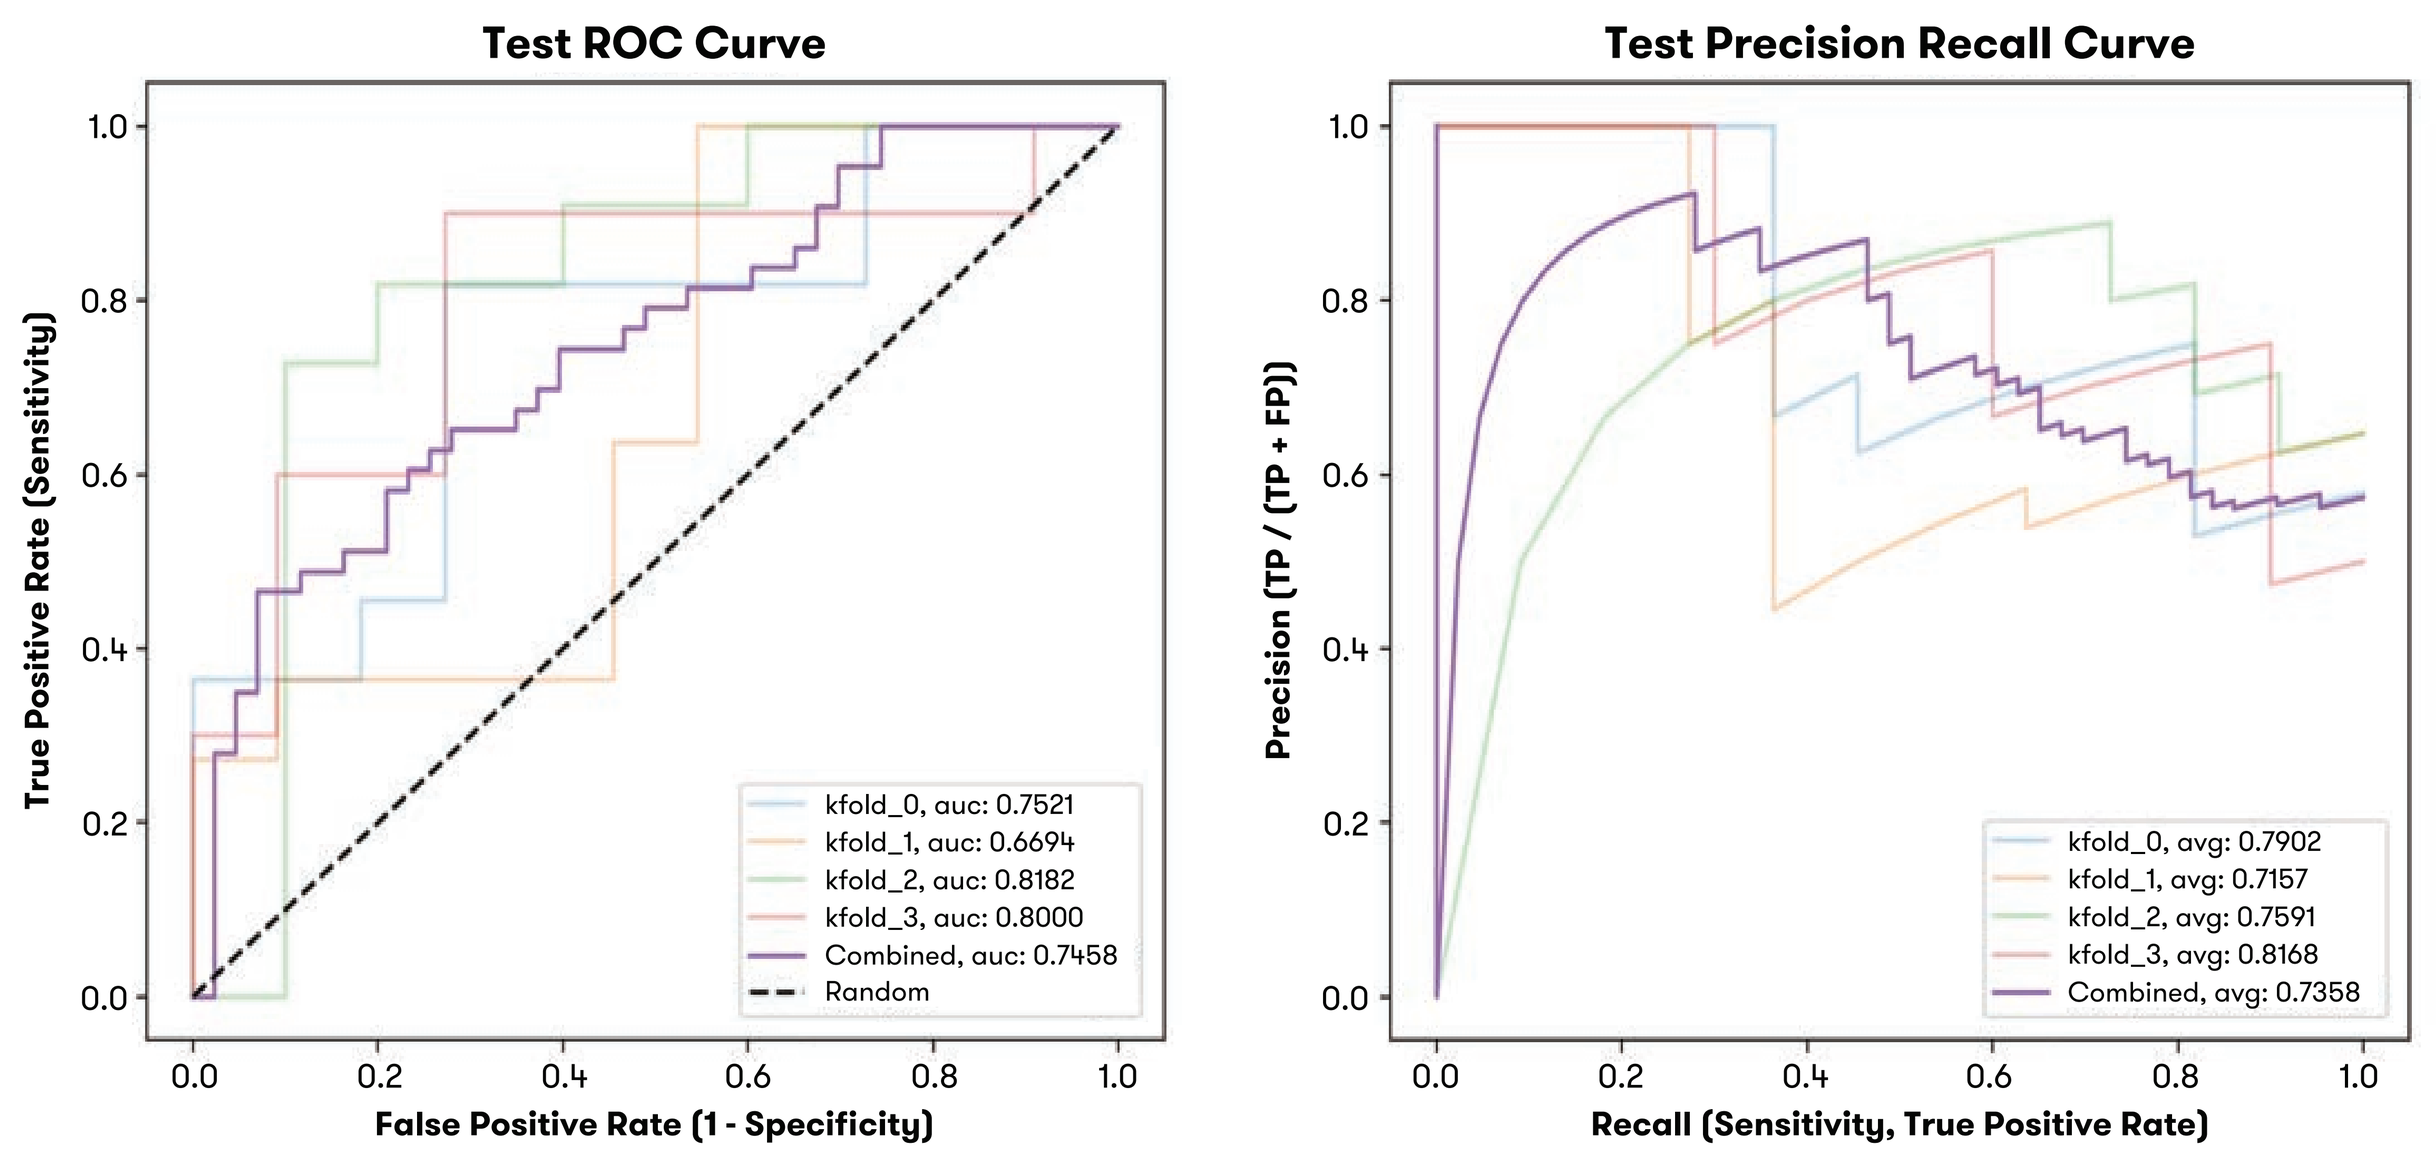
Test ROC Curve
True Positive Rate (Sensitivity)
False Positive Rate (1 - Specificity)
kfold_0, auc: 0.7521
kfold_1, auc: 0.6694
kfold_2, auc: 0.8182
kfold_3, auc: 0.8000
Combined, auc: 0.7458
Random
Test Precision Recall Curve
Precision (TP / (TP + FP))
Recall (Sensitivity, True Positive Rate)
kfold_0, avg: 0.7902
kfold_1, avg: 0.7157
kfold_2, avg: 0.7591
kfold_3, avg: 0.8168
Combined, avg: 0.7358

- To construct a multivariate classifier for response using baseline data, we applied a L1-logistic regression model to both the computational TFBA and TSS-GAP features to identify the best features.
- Early results with TFBA scores suggested transcription factor features to be the most informative (AUC = 0.65), so we selected the 2,751 TSS-GAP scores for all transcription factors across the genome to include in the multivariate classifier.
- Clinical lab data, from standard blood chemistry tests and immunophenotyping as detailed in the methods, was also incorporated into the modeling by concatenating it with the TSS-GAP TF scores.
- Shown here is the final model combining TSS-GAP TF scores and clinical lab data to distinguish patients based on predicted response groups in cross-validation (AUC = 0.75; AUPRC = 0.74).

Figure 5. Gene set enrichment analysis reveals higher activation levels of proliferative and immune gene sets in non-responders at baseline

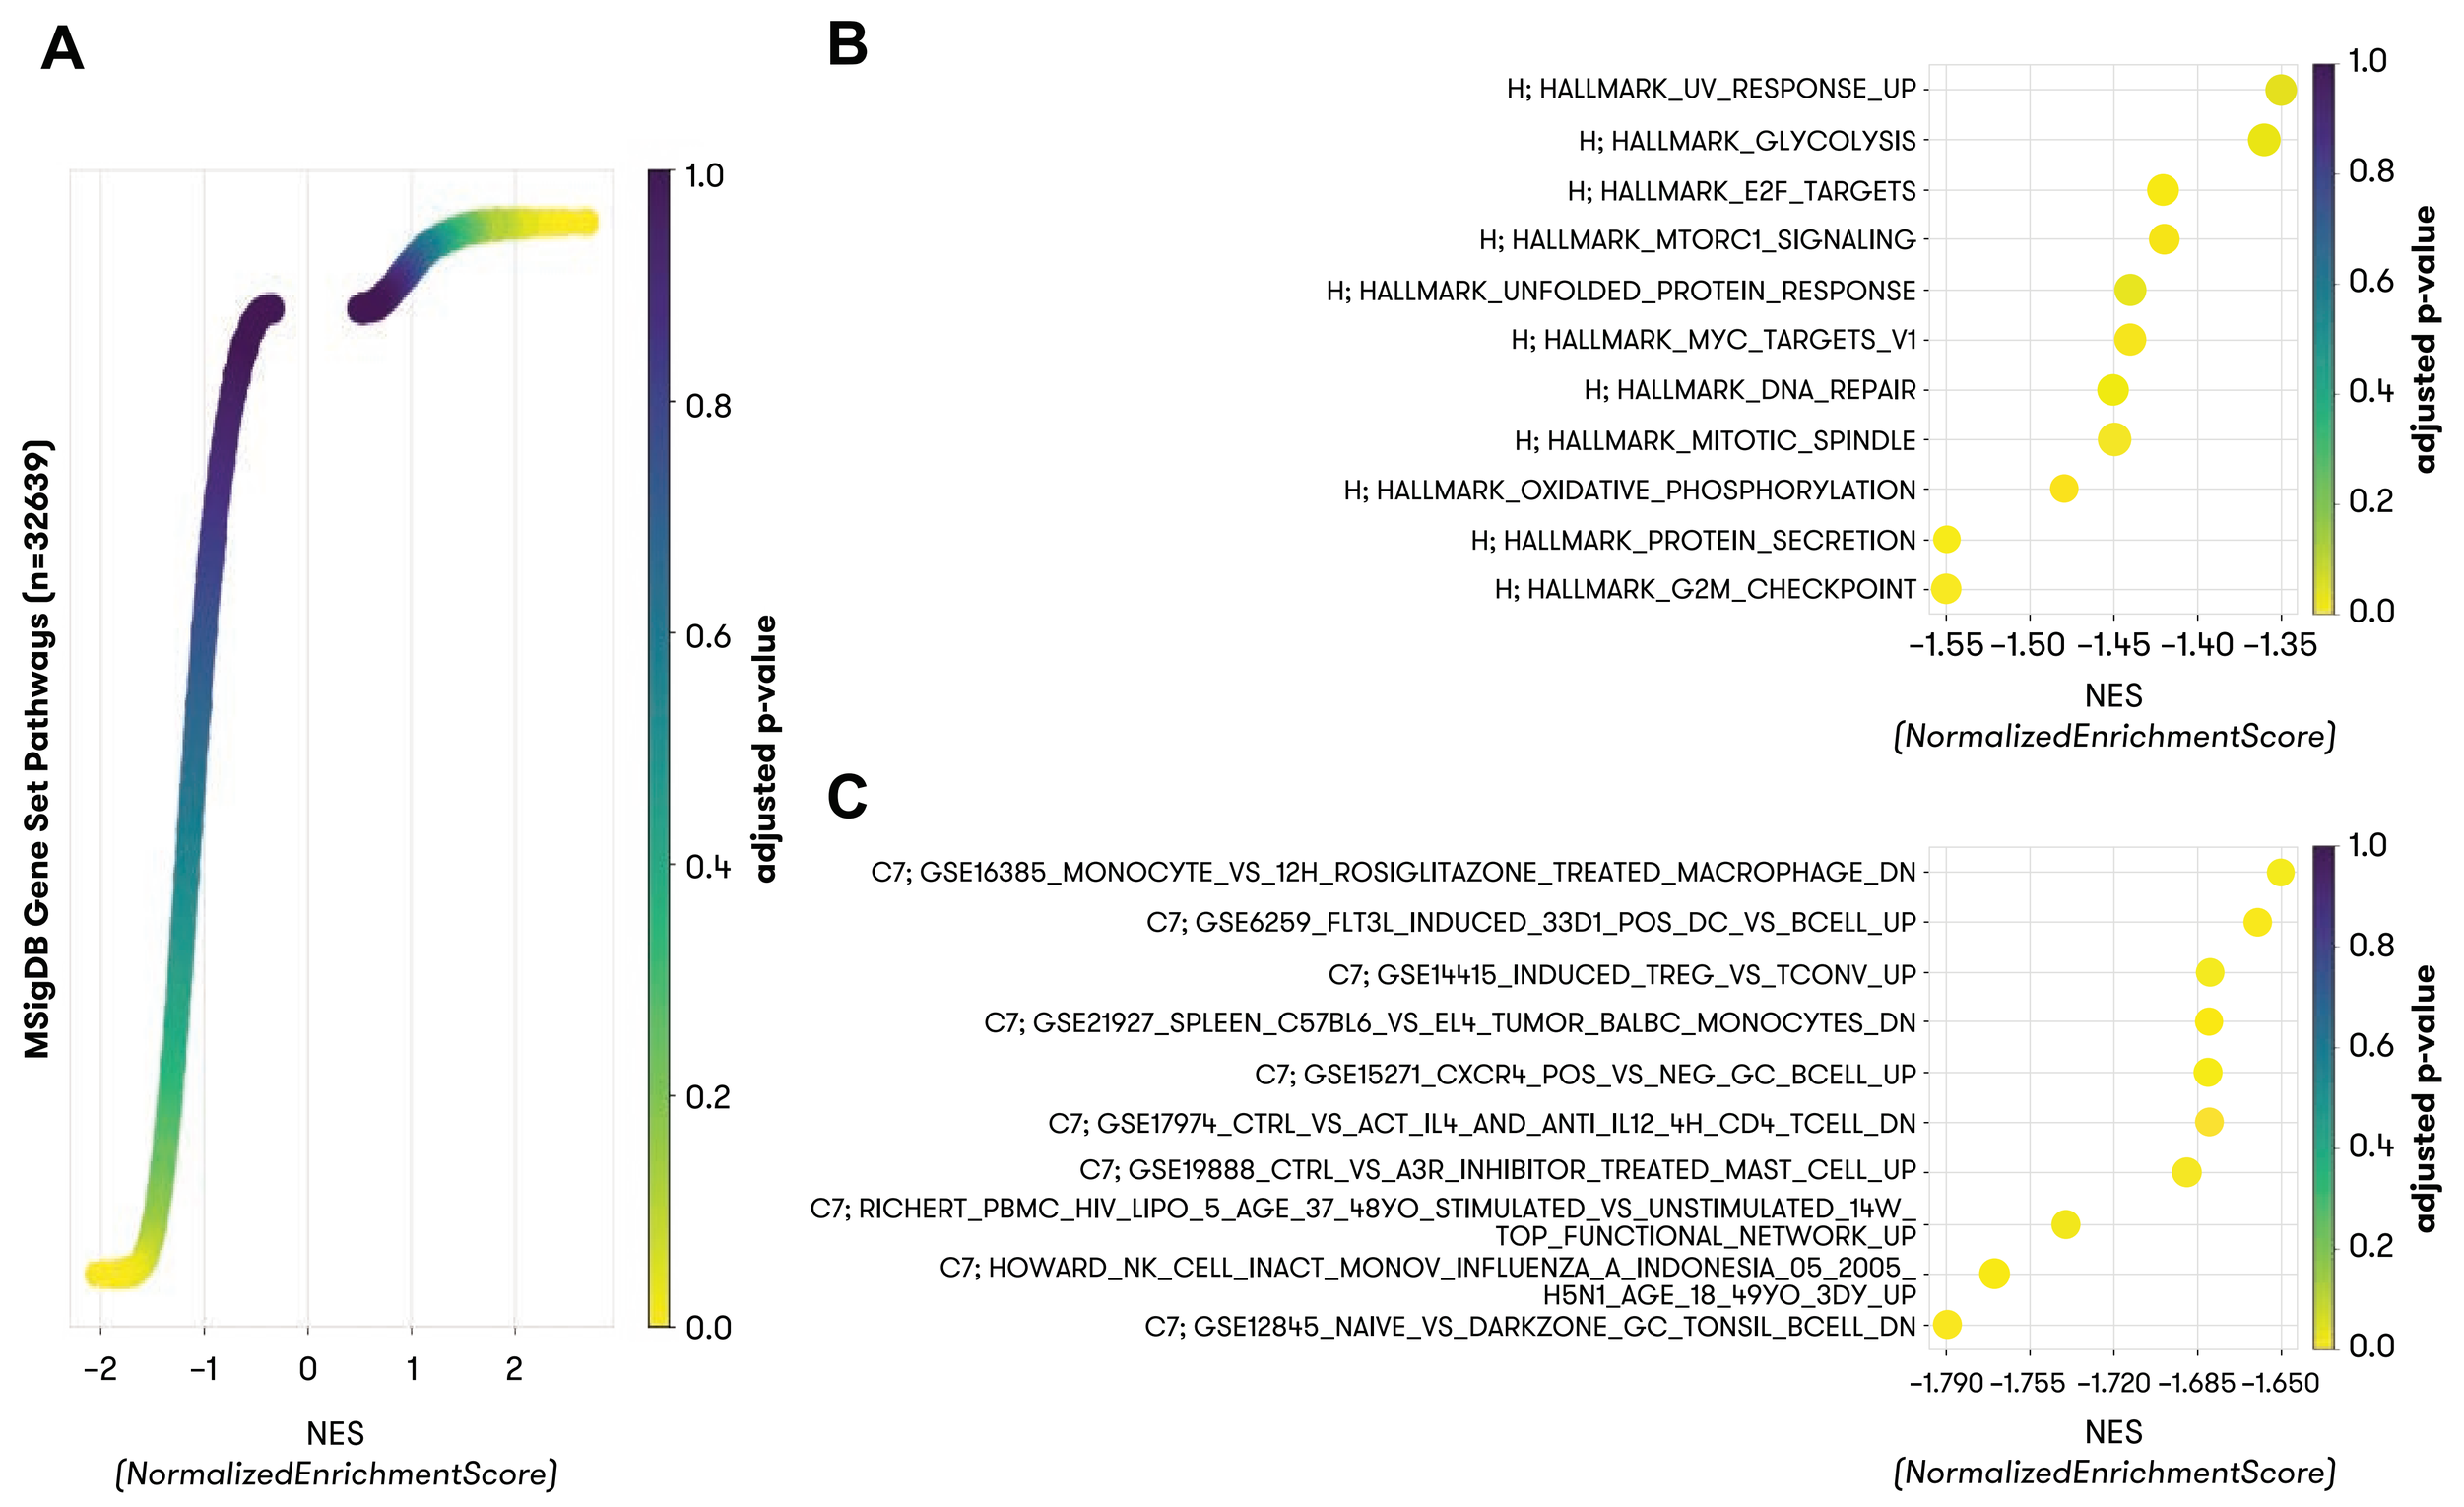
A
B
C
MSigDB Gene Set Pathways (n=32639)
NES
(NormalizedEnrichmentScore)
adjusted p-value
H; HALLMARK_UV_RESPONSE_UP
H; HALLMARK_GLYCOLYSIS
H; HALLMARK_E2F_TARGETS
H; HALLMARK_MTORC1_SIGNALING
H; HALLMARK_UNFOLDED_PROTEIN_RESPONSE
H; HALLMARK_MYC_TARGETS_V1
H; HALLMARK_DNA_REPAIR
H; HALLMARK_MITOTIC_SPINDLE
H; HALLMARK_OXIDATIVE_PHOSPHORYLATION
H; HALLMARK_PROTEIN_SECRETION
H; HALLMARK_G2M_CHECKPOINT
C7; GSE16385_MONOCYTE_VS_12H_ROSIGLITAZONE_TREATED_MACROPHAGE_DN
C7; GSE6259_FLT3L_INDUCED_33D1_POS_DC_VS_BCELL_UP
C7; GSE14415_INDUCED_TREG_VS_TCONV_UP
C7; GSE21927_SPLEEN_C57BL6_VS_EL4_TUMOR_BALBC_MONOCYTES_DN
C7; GSE15271_CXCR4_POS_VS_NEG_GC_BCELL_UP
C7; GSE17974_CTRL_VS_ACT_IL4_AND_ANTI_IL12_4H_CD4_TCELL_DN
C7; GSE19888_CTRL_VS_A3R_INHIBITOR_TREATED_MAST_CELL_UP
C7; RICHERT_PBMC_HIV_LIPO_5_AGE_37_48YO_STIMULATED_VS_UNSTIMULATED_14W_TOP_FUNCTIONAL_NETWORK_UP
C7; HOWARD_NK_CELL_INACT_MONOV_INFLUENZA_A_INDONESIA_05_2005_H5N1_AGE_18_49YO_3DY_UP
C7; GSE12845_NAIVE_VS_DARKZONE_GC_TONSIL_BCELL_DN

- GSEA of TSS-GAP scores between responders vs non-responders revealed 3145 significant gene sets (FDR<5%) out of 32639 tested.
- 11 significant Hallmark (H) gene set signatures (FDR<5%) reveal higher activation levels of proliferative activity sets in non-responders at baseline.
- 1257 significant immunologic signatures (C7) gene set signatures (FDR<5%) reveal changes in immune activation; notably evidence of increased immune activity is seen in non-responders relative to responders at baseline. Top 10 gene sets ranked by score are plotted for visualization.

CONCLUSIONS

- Using our platform, we characterized a cohort of R/R DLBCL patients at baseline and on treatment (cycle 2 day 1; C2D1) with loncastuximab tesirine.
- The results showcase the potential of our approach to identify markers associated with response to lonca and suggest mechanisms of resistance, potentially informing rational drug combination research.
- Our modeling efforts integrating TSS-GAP and clinical data indicate the potential of this approach to predict lonca efficacy, pending additional validation.
- Future studies with our platform may enable targeted precision medicine applications and therapeutic decisions.

REFERENCES

1. Saint John J *et al* AACR 2019
2. Ulz P *et al* *Nature Communications* 2019

Presented at the The 64th ASH Annual Meeting in New Orleans, Louisiana, December 10-13, 2022