#5387

Discovery of plasma protein biomarkers associated with overall survival in R/R DLBCL patients treated with loncastuximab tesirine

Francesco Vallania*[1], Victoria Cheung*[1], Anupriya Tripathi[1], Maggie Louie[1], Thomas Snyder[1], Jimmy Lin[1], Karin Havenith[2], Yajuan Qin[2], Serafino Pantano[2], Jens Wuerthner[2‡], Patrick H. van Berkel[2]

[1]Freenome, 279 East Grand Avenue, 5th Floor, South San Francisco, CA, USA; [2]ADC Therapeutics SA, Biopole, Route de la Corniche 3B, 1066 Epalinges, Switzerland

*Authors contributed equally to the work ‡Affiliation at the time the work was conducted

INTRODUCTION AND OBJECTIVES

- Plasma proteomics is a non-invasive approach towards the discovery of biomarkers associated with cancer treatment outcomes, including disease progression and overall survival (OS)
- Loncastuximab tesirine (lonca) is an antibody-drug conjugate, composed of a humanized anti-CD19 antibody conjugated to a pyrrolobenzodiazepine dimer cytotoxin, currently approved for the treatment of relapsed/refractory (R/R) diffuse large B-cell lymphoma (DLBCL)
- Here, we investigated the association of plasma protein biomarkers measured at baseline with OS in the LOTIS-2 phase 2 trial (NCT3589469; data cut March 1st 2021)

Table 1. Cohort description and profiling

	Samples
Sex	
Male	40 (64.5%)
Female	22 (35.5%)

METHODS

- Abundances of 888 plasma proteins, including inflammation, cancer and DNA repair associated markers, were measured for 62 patients with R/R DLBCL from plasma samples collected at baseline
- Protein markers predictive of overall survival (OS) were identified using an L1-regularized Cox Proportional Hazard model that incorporates survival time and censoring status
- To better characterize underlying biology and identify set of proteins associated with OS, we applied an L2-regularized (Ridge) Cox Proportional Hazard model. Gene Set Enrichment Analysis (GSEA) with the MSigDB gene sets was performed on the coefficients of this model.

Figure 1. Cox Proportional Hazard model with L1-regularization identifies five protein markers associated with overall survival

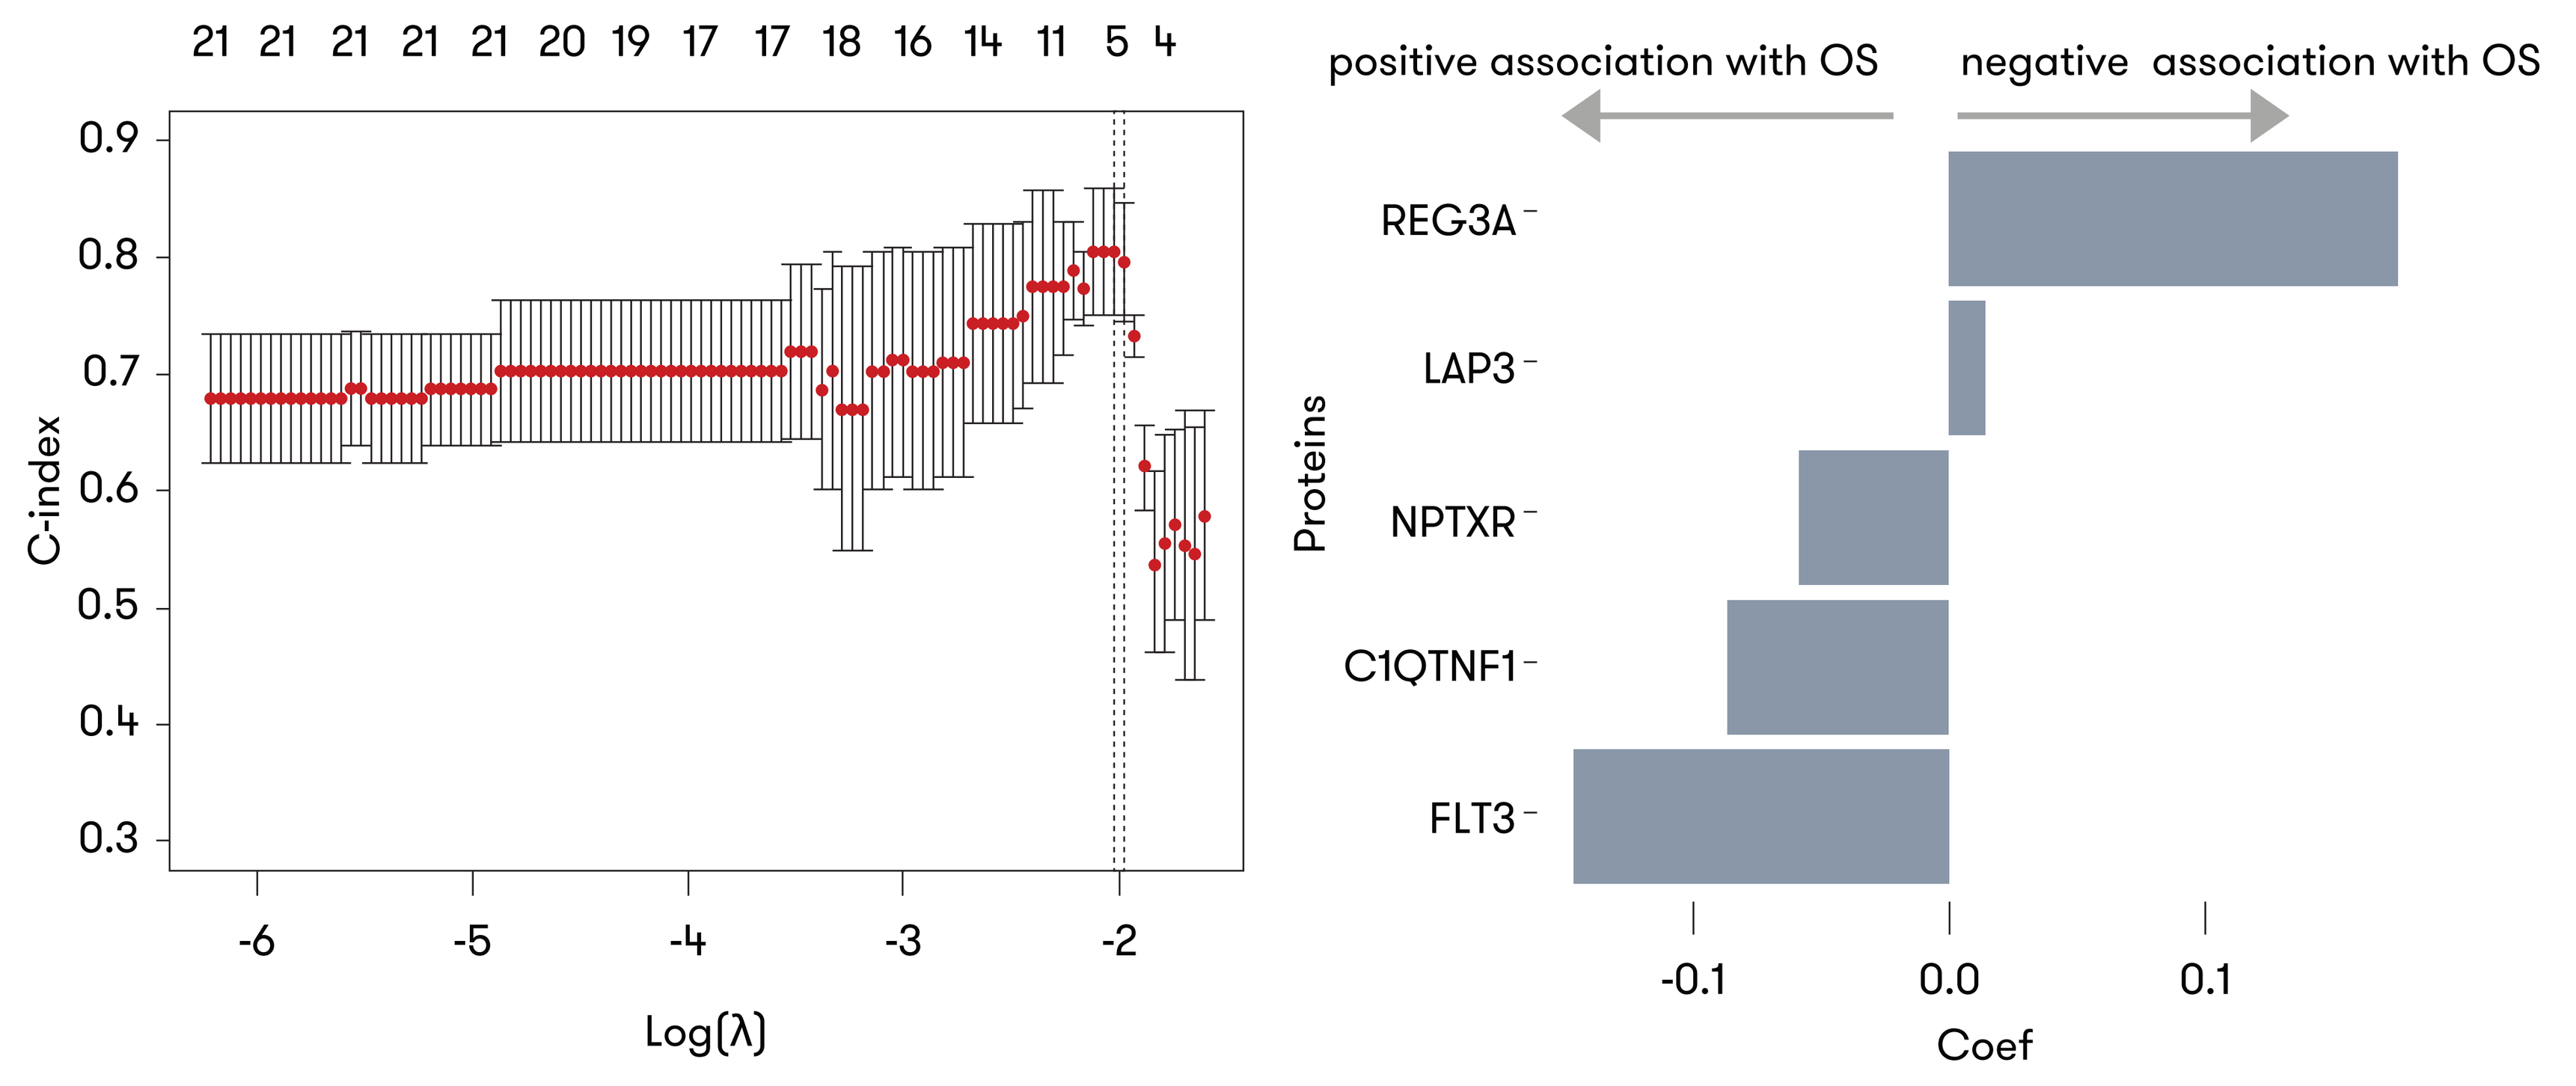
21 21 21 21 21 20 19 17 17 18 16 14 11 5 4
C-index
0.9
0.8
0.7
0.6
0.5
0.4
0.3
-6
-5
-4
-3
-2
Log(λ)
positive association with OS
negative association with OS
Proteins
REG3A
LAP3
NPTXR
C1QTNF1
FLT3
-0.1
0.0
0.1
Coef

Using an L1-regularized Cox proportional hazard model with 3-fold cross-validation, we identified 5 proteins markers associated with survival using a final model with L1 tuning parameter λ selected per the 1SD rule.

- CV accuracy was measured by the Harrel C-index (y-axis) as a function of the log of λ (x-axis) with max (left) and 1SD (right) values indicated by dashed lines. Corresponding number of identified protein markers is shown above.
- Coefficients of identified proteins are shown, with positive sign indicating increased hazard (reduced survival time)

Figure 2. Cox Proportional Hazard model with L1-regularization identifies protein markers associated with overall survival

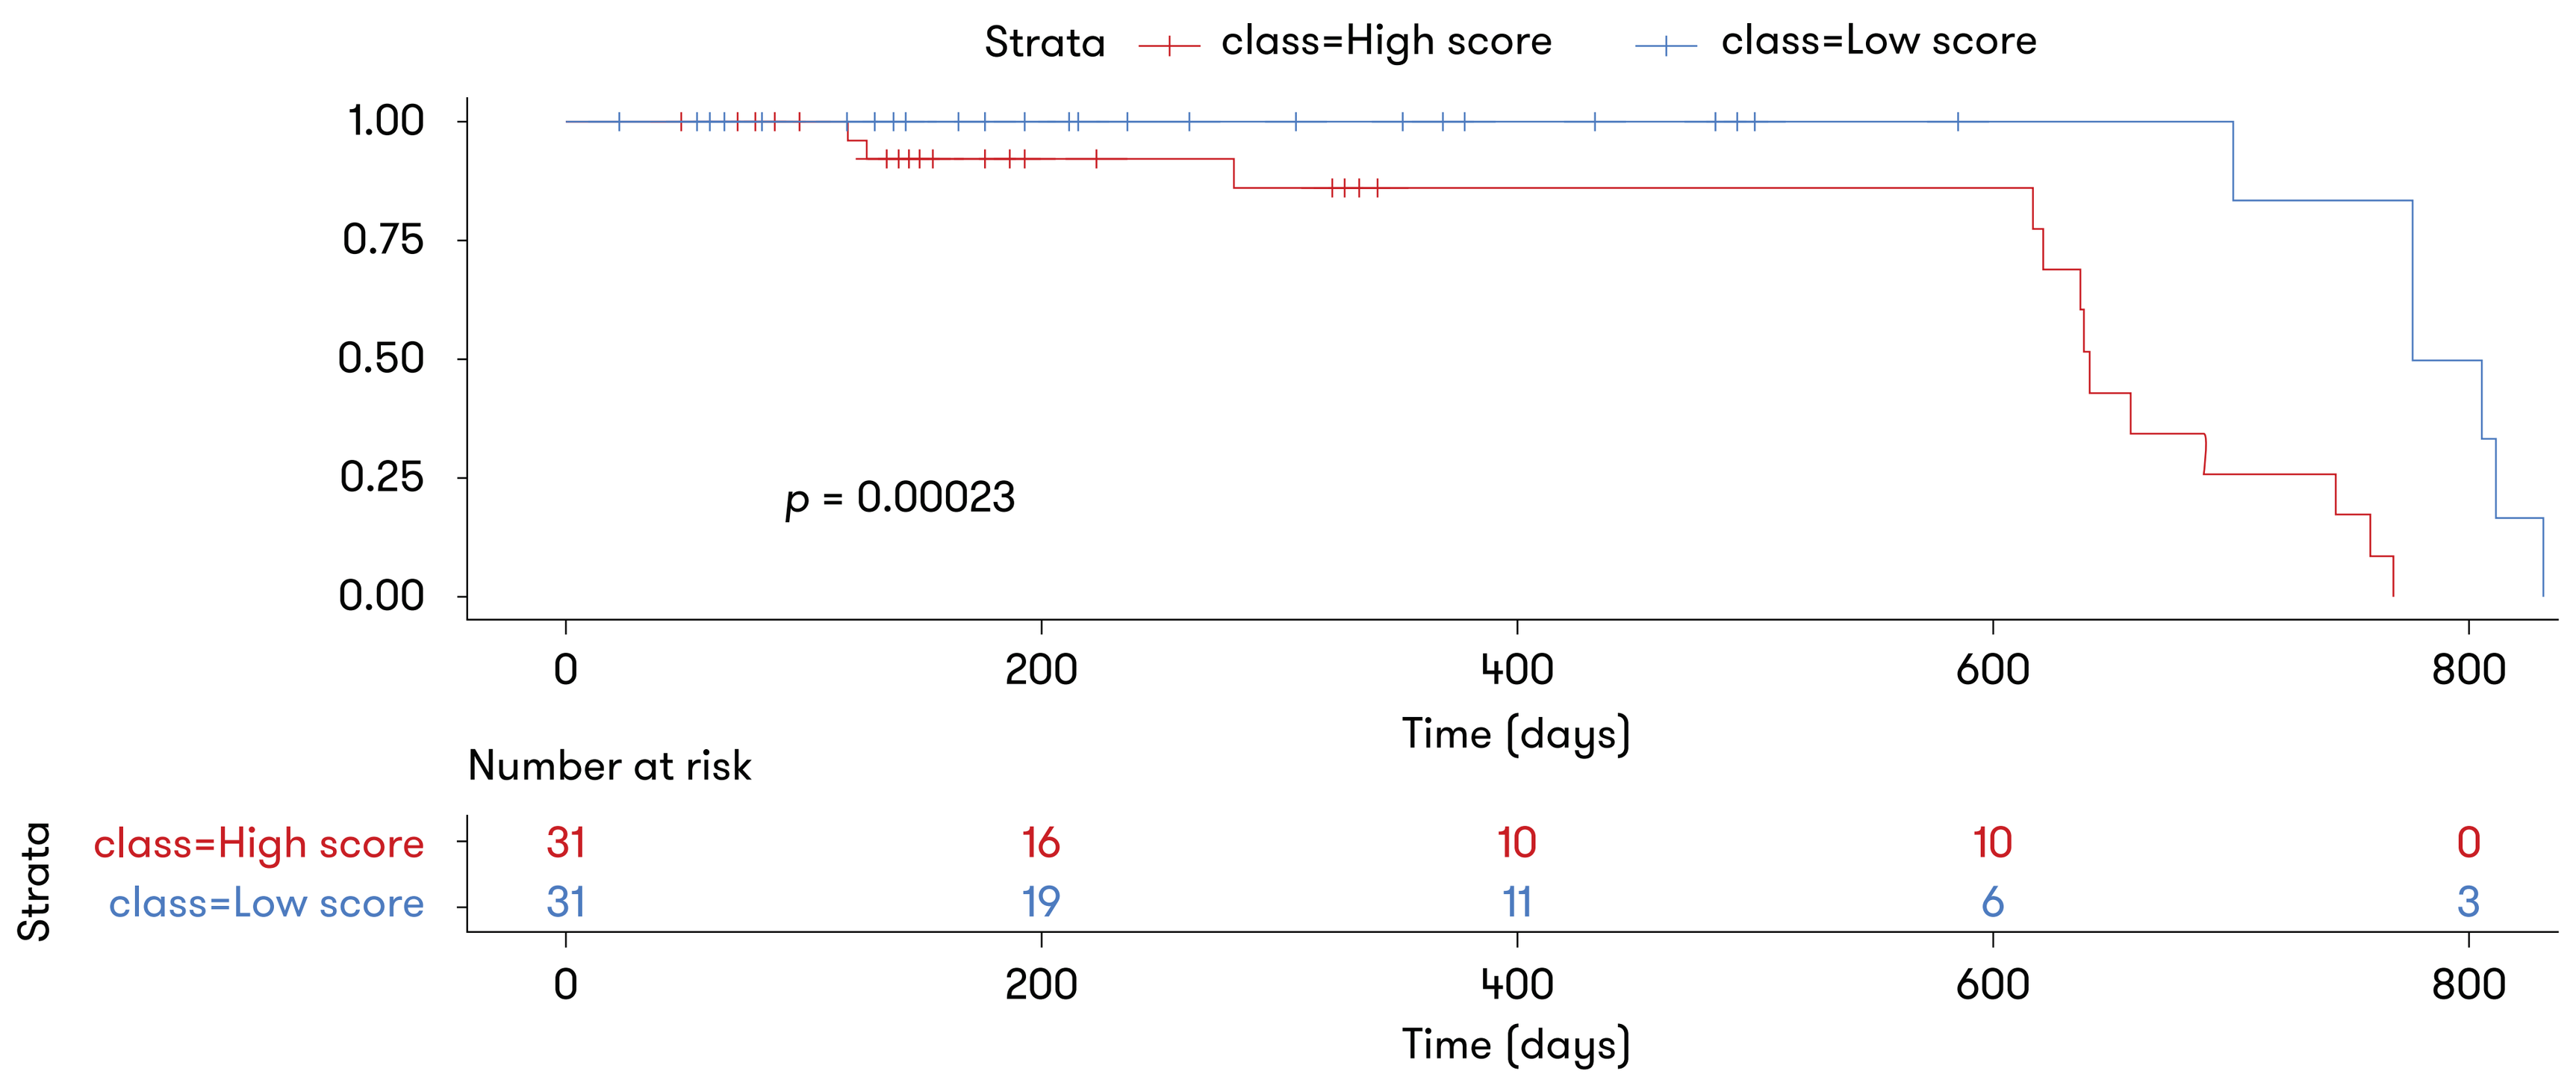
Strata
class=High score
class=Low score
1.00
0.75
0.50
0.25
0.00
p = 0.00023
0
200
400
600
800
Time (days)
Number at risk
Strata
class=High score 31 16 10 10 0
class=Low score 31 19 11 6 3
0
200
400
600
800
Time (days)

- Using the markers and coefficients selected by the Cox Proportional Hazard model, we computed a hazard score for each patient
- Samples were stratified by score, splitting at the median
- Patients with higher scores showed significantly shorter survival (log-rank *p*-value = 0.00023) compared to patients with lower scores

Figure 3. GSEA of Ridge regression coefficients identifies KRAS signaling gene set to be associated with lower OS

- Gene set enrichment results using MSigDB Hallmark gene sets are plotted for visualization; one statistically significant Hallmark pathway is highlighted.
- Upregulation of KRAS signaling gene set is significantly associated with lower OS
 - Increased KRAS signaling is known to promote cancer stemness and metastasis (Najumudeen et al. *Oncogene* 2016, Vendramini et al. *Cancers* 2022), therefore downregulation of KRAS signaling is in concordance with increased overall survival.
 - Notable leading edge genes in the KRAS signaling set include IL-7R and CCL20
 - IL-7R is associated with non-germinal center B-cell like (non-GCB) DLBCL (Zhao et al. *Oncol Lett.* 2020; Nyman et al. *Blood* 2007)
 - CCL20 is involved in promotion of DLBCL cell stemness and aggressiveness (Chang et al. *Sci Rep* 2020)

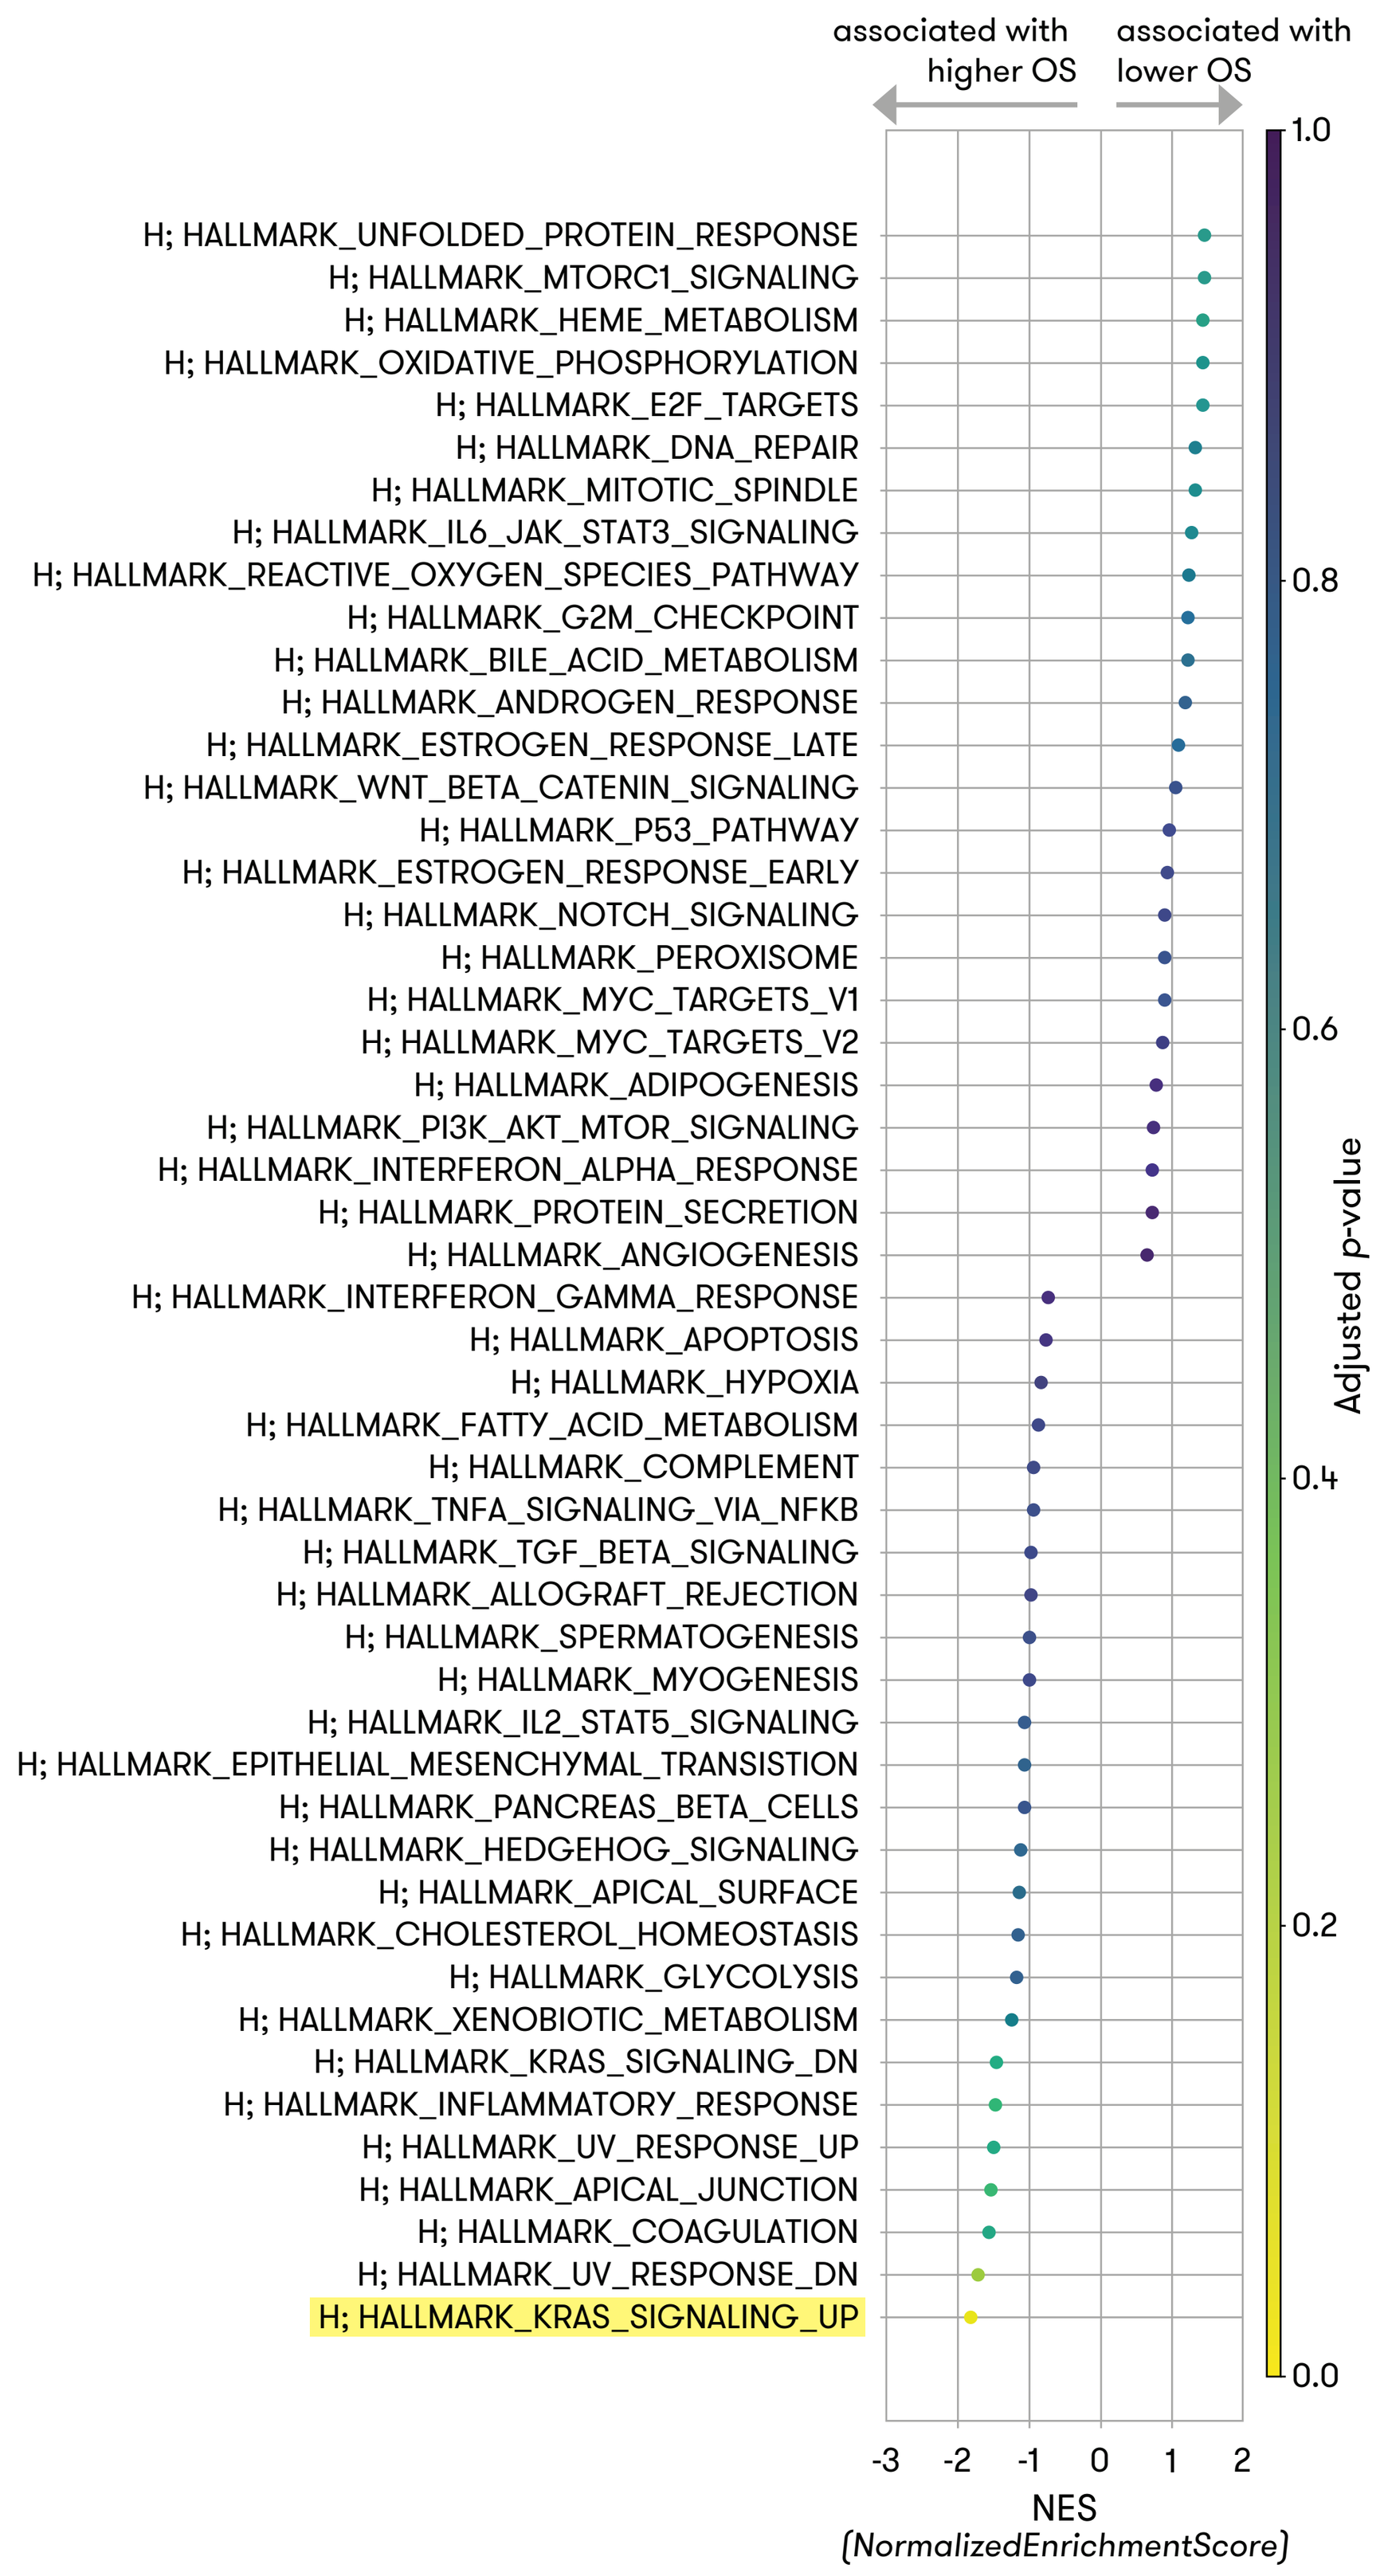
associated with higher OS
associated with lower OS
H; HALLMARK_UNFOLDED_PROTEIN_RESPONSE
H; HALLMARK_MTORC1_SIGNALING
H; HALLMARK_HEME_METABOLISM
H; HALLMARK_OXIDATIVE_PHOSPHORYLATION
H; HALLMARK_E2F_TARGETS
H; HALLMARK_DNA_REPAIR
H; HALLMARK_MITOTIC_SPINDLE
H; HALLMARK_IL6_JAK_STAT3_SIGNALING
H; HALLMARK_REACTIVE_OXYGEN_SPECIES_PATHWAY
H; HALLMARK_G2M_CHECKPOINT
H; HALLMARK_BILE_ACID_METABOLISM
H; HALLMARK_ANDROGEN_RESPONSE
H; HALLMARK_ESTROGEN_RESPONSE_LATE
H; HALLMARK_WNT_BETA_CATENIN_SIGNALING
H; HALLMARK_P53_PATHWAY
H; HALLMARK_ESTROGEN_RESPONSE_EARLY
H; HALLMARK_NOTCH_SIGNALING
H; HALLMARK_PEROXISOME
H; HALLMARK_MYC_TARGETS_V1
H; HALLMARK_MYC_TARGETS_V2
H; HALLMARK_ADIPOGENESIS
H; HALLMARK_PI3K_AKT_MTOR_SIGNALING
H; HALLMARK_INTERFERON_ALPHA_RESPONSE
H; HALLMARK_PROTEIN_SECRETION
H; HALLMARK_ANGIOGENESIS
H; HALLMARK_INTERFERON_GAMMA_RESPONSE
H; HALLMARK_APOPTOSIS
H; HALLMARK_HYPOXIA
H; HALLMARK_FATTY_ACID_METABOLISM
H; HALLMARK_COMPLEMENT
H; HALLMARK_TNFA_SIGNALING_VIA_NFKB
H; HALLMARK_TGF_BETA_SIGNALING
H; HALLMARK_ALLOGRAFT_REJECTION
H; HALLMARK_SPERMATOGENESIS
H; HALLMARK_MYOGENESIS
H; HALLMARK_IL2_STAT5_SIGNALING
H; HALLMARK_EPITHELIAL_MESENCHYMAL_TRANSITION
H; HALLMARK_PANCREAS_BETA_CELLS
H; HALLMARK_HEDGEHOG_SIGNALING
H; HALLMARK_APICAL_SURFACE
H; HALLMARK_CHOLESTEROL_HOMEOSTASIS
H; HALLMARK_GLYCOLYSIS
H; HALLMARK_XENOBIOTIC_METABOLISM
H; HALLMARK_KRAS_SIGNALING_DN
H; HALLMARK_INFLAMMATORY_RESPONSE
H; HALLMARK_UV_RESPONSE_UP
H; HALLMARK_APICAL_JUNCTION
H; HALLMARK_COAGULATION
H; HALLMARK_UV_RESPONSE_DN
H; HALLMARK_KRAS_SIGNALING_UP
1.0
0.8
0.6
0.4
0.2
0.0
Adjusted p-value
-3 -2 -1 0 1 2
NES
(NormalizedEnrichmentScore)

CONCLUSIONS

- Using our platform, we characterized a cohort of R/R DLBCL patients at baseline with lonca using plasma proteomics
- Our results identified plasma protein markers associated with overall survival of patients with R/R DLBCL treated with lonca, highlighting the potential of plasma proteins as a source of relevant biomarkers
- Our analysis revealed a protein set reflective of changes in KRAS signaling to be negatively associated with OS. This set include CCL20 and IL7-R, two genes whose expression has been previously associated with DLBCL outcomes.
- Future studies with our platform may enable targeted precision medicine applications and support therapeutic decisions

Presented at the American Association for Cancer Research (AACR) Annual Meeting | April 14-19, 2023 | Orlando, Florida, USA