CHARACTERISTICS AND OUTCOMES IN PATIENTS WITH RELAPSED/REFRACTORY DIFFUSE LARGE B-CELL LYMPHOMA WHO RECEIVED ≥3 LINES OF THERAPIES

EUROPEAN HEMATOLOGY ASSOCIATION

EP528

M HAMADANI[1], L LIAO[2], T YANG[3], L CHEN[2], C MOSKOWITZ[4]

[1]Division of Hematology and Oncology, Medical College of Wisconsin, Milwaukee, WI USA; [2]ADC Therapeutics, New Providence, NJ USA; [3] TYStats, LLC, Marlboro, NJ USA; [4]Miller School of Medicine, University of Miami Health System, Miami, FL USA

INTRODUCTION

- Diffuse large B-cell lymphoma (DLBCL) is an aggressive B-cell malignancy and the most common form of non-Hodgkin lymphoma, accounting for approximately one-quarter of all new cases.[1]
- Approximately 60% of patients with DLBCL who receive first-line chemoimmunotherapy (CIT) or chemotherapy (CT) achieve a durable response.[2] For those patients who experience a relapse or are refractory to treatment (R/R DLBCL), multiple treatment regimens have been tried in the last decade with relatively little improvement seen in patient outcomes.[3]
- In the last few years, advancements have been made in the treatment of R/R DLBCL, especially chimeric antigen receptor T-cell therapies (CAR-T).[3] However, they cannot be offered to all patients due to both medical and other reasons.[4]
- With the rapidly evolving treatment landscape for R/R DLBCL, there is significant need for studies to examine the clinical outcomes of patients with R/R DLBCL using contemporary data in real-world settings.

OBJECTIVES

- In this study using de-identified electronic health records (EHRs) of patients with R/R DLBCL, we examined patient characteristics and the clinical outcomes of those who initiated either a third line of therapy (3L) or a fourth line of therapy (4L) during a recent time frame, i.e., January 2014 through September 2020.

METHODS

Study population

- Adult patients diagnosed with DLBCL who received ≥3L between January 1, 2014 and August 31, 2019 were selected from the COTA EHR database, a de-identified database of real-world data, derived from the EHRs of healthcare providers in the United States.
- Patients were grouped into cohorts by whether they initiated a 3L or 4L and further stratified by the type of treatment received: CT/CIT, Targeted therapy (TT), CAR-T, or salvage therapy consolidated with hematopoietic stem cell transplantation (HSCT).
 - A line of therapy for a patient was defined as initiation of a treatment until the earliest of one of the following events: clinical designation of disease progression, addition or substitution of a new drug >30 days from the treatment initiation, or a gap in therapy of >180 days.
- At patients' initiation of their 3L or 4L, demographics were examined and stratified by line of therapy (3L or 4L) and treatment type.
 - Eastern Cooperative Oncology Group performance status measurements were taken closest to patients' 3L or 4L initiation and were within 365 days prior to treatment initiation.

Outcome measurements

- The proportions of patients who achieved a complete response (CR) and partial response (PR) were determined and stratified by line of therapy (3L or 4L) and treatment type.
 - The proportions of patients with CR and PR were calculated according to the best response achieved after treatment initiation and prior to the start of the next line of therapy.
- Since HSCT is a consolidation therapy for patients achieving a CR or PR following salvage treatment, the response rates among patients who received HSCT were not reported.

Statistical analyses

- Descriptive statistics were utilized to describe demographics, clinical characteristics, and response rates of patient cohorts.
- Median overall survival time was estimated with Kaplan-Meier analyses for patient cohorts.

RESULTS

- Of the DLBCL patients identified from the COTA EHR database, 212 initiated their 3L and 128 their 4L. Among those who initiated a 3L, 55% initiated CT/CIT, 27% TT, 9% CAR-T, and 9% salvage therapy consolidated with HSCT. Among those who initiated a 4L, 52% initiated CT/CIT, 34% TT, 9 % CAR-T, and 5% salvage therapy consolidated with HSCT.
- During a median follow-up of 22.5 months of patients who initiated 3L CT/CIT or TT, 14.9% subsequently received CAR-T. During a median follow-up of 19.3 months of patients who initiated 4L CT/CIT or TT, 10.9% subsequently received CAR-T.

Table 1. Characteristics of Patients who Initiated Their Third Line of Therapy (3L)

Characteristic	CT/CIT n=117 (55%)	TT n=57 (27%)	CAR-T n=20 (9%)	HSCT n=18 (9%)
Age in yrs, mean (SD)	62.6 (12.4)	65.6 (14.4)	54.2 (17.1)	53.2 (17.3)
≥65, n (%)	48 (41.0)	30 (52.6)	4 (20.0)	5 (27.8)
Sex, n (%)				
Female	46 (39.3)	25 (43.9)	8 (40.0)	8 (44.4)
Male	71 (60.7)	32 (56.1)	12 (60.0)	10 (55.6)
Race, n (%)				
White	94 (80.3)	43 (75.4)	18 (90.0)	16 (88.9)
Black/African American	6 (5.1)	4 (7.0)	0 (0)	0 (0)
Asian	5 (4.3)	3 (5.3)	0 (0)	1 (5.6)
Other	5 (4.3)	5 (8.8)	1 (5.0)	1 (5.6)
Missing	7 (6.0)	2 (3.5)	1 (5.0)	0 (0.0)
ECOG, n (%)				
0-1	45 (38.5)	24 (42.1)	14 (70.0)	14 (77.8)
2-3	24 (20.5)	19 (33.3)	5 (25.0)	1 (5.6)
4+	0 (0.0)	1 (1.8)	0 (0.0)	0 (0.0)
Not reported	48 (41.0)	13 (22.8)	1 (5.0)	3 (16.7)

ECOG: Eastern Cooperative Oncology Group performance status; SD: Standard deviation.

Table 2. Characteristics of Patients who Initiated Their Fourth Line of Therapy (4L)

Characteristic	CT/CIT n=66 (52%)	TT n=44 (34%)	CAR-T n=12 (9%)	HSCT n=6 (5%)
Age in yrs, mean (SD)	62.6 (15.3)	59.2 (13.1)	59.7 (10.7)	60.9 (8.0)
≥65, n (%)	34 (51.5)	12 (27.3)	4 (33.3)	1 (16.7)
Sex, n (%)				
Female	25 (37.9)	15 (34.1)	6 (50.0)	4 (66.7)
Male	41 (62.1)	29 (65.9)	6 (50.0)	2 (33.3)
Race, n (%)				
White	49 (74.2)	32 (72.7)	9 (75.0)	6 (100)
Black/African American	7 (10.6)	2 (4.6)	0 (0)	0 (0)
Asian	5 (7.6)	4 (9.1)	1 (8.3)	0 (0)
Other	3 (4.6)	4 (9.1)	1 (8.3)	0 (0)
Missing	2 (3.0)	2 (4.6)	1 (8.3)	0 (0)
ECOG, n (%)				
0-1	33 (50.0)	21 (47.7)	10 (83.3)	3 (50.0)
2-3	18 (27.3)	14 (31.8)	2 (16.7)	3 (50.0)
4+	0 (0.0)	2 (4.6)	0 (0)	0 (0)
Not reported	15 (22.7)	7 (15.9)	0 (0)	0 (0)

ECOG: Eastern Cooperative Oncology Group performance status; SD: Standard deviation.

Table 3. Response Rates of Patients who Received a 3L or 4L

3L	CT/CIT n=117	TT n=57	CAR-T n=20
Complete response, n (%)	11 (9.4)	5 (8.8)	12 (60.0)
Partial response, n (%)	31 (26.5)	18 (31.6)	2 (10.0)
4L	**CT/CIT n=66**	**TT n=44**	**CAR-T n=12**
Complete response, n (%)	5 (7.6)	7 (15.9)	6 (50.0)
Partial response, n (%)	22 (33.3)	10 (22.7)	3 (25.0)

- CR rates of patients who initiated non-cell 3L (CT/CIT: 9.4%; TT: 8.8%) were markedly lower than among those who received 3L CAR-T (60.0%); similar findings were observed among patients who initiated a 4L (CT/CIT: 7.6%; TT: 15.9%; CAR-T: 50.0%; Table 3).

RESULTS CONT.

- Among the R/R DLBCL patients who received non-cell therapy as their 3L, median OS was 7.7 months (CT/CIT: 7.7 months; TT: 7.9 months) and among those who received these therapies as their 4L, median OS was 4.5 months (CT/CIT: 4.5 months; TT: 5.1 months).
- Among the R/R DLBCL patients who received cell-based therapy (CAR-T or HSCT) median OS time was not reached in either the 3L or 4L setting.

Figure 1. Median Overall Survival (OS) Time of Patients who Received a) 3L and b) 4L

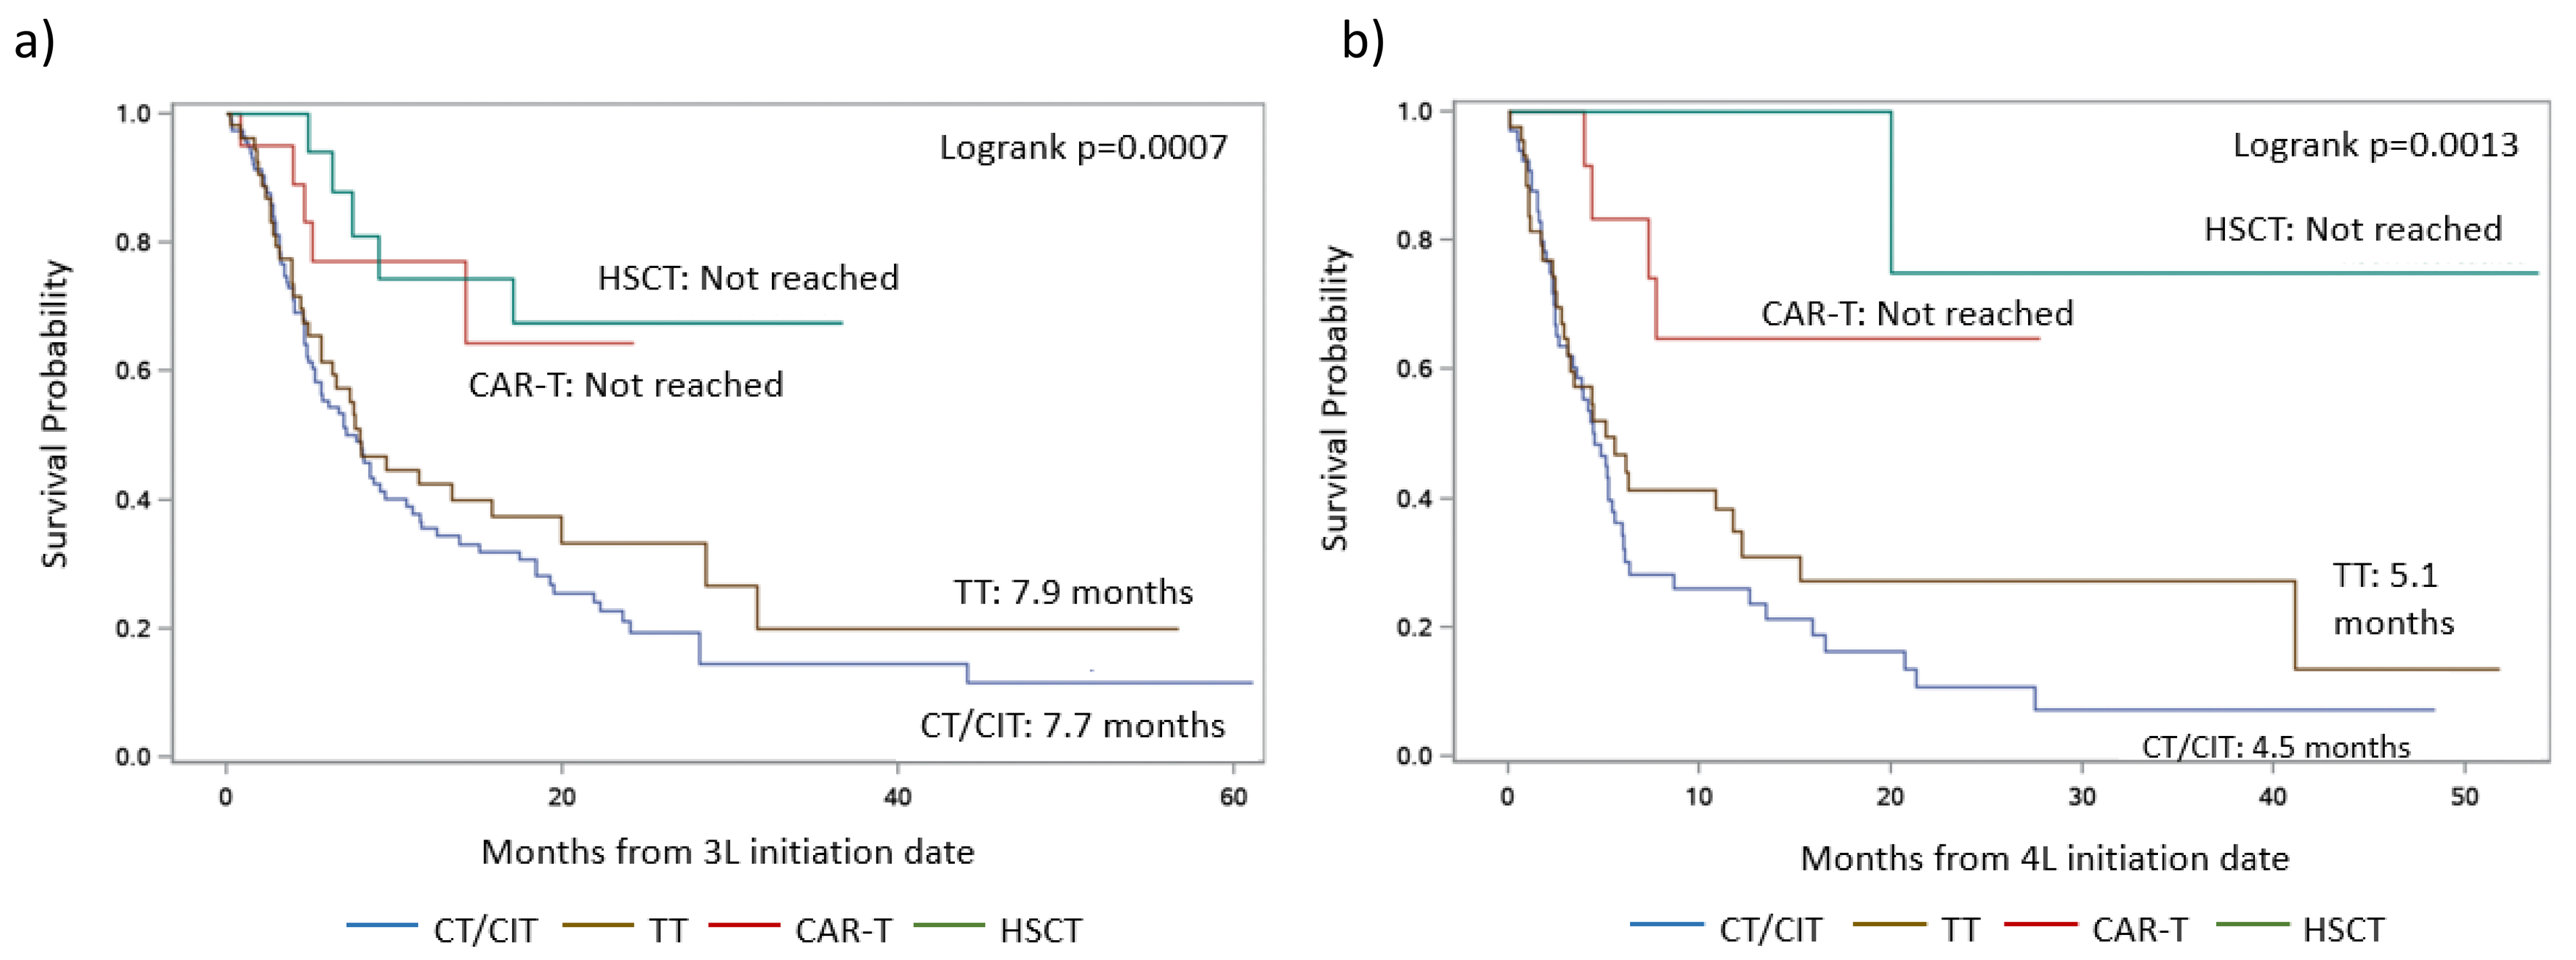
a)
Logrank p=0.0007
HSCT: Not reached
CAR-T: Not reached
TT: 7.9 months
CT/CIT: 7.7 months
Survival Probability
Months from 3L initiation date
CT/CIT
TT
CAR-T
HSCT
b)
Logrank p=0.0013
HSCT: Not reached
CAR-T: Not reached
TT: 5.1 months
CT/CIT: 4.5 months
Survival Probability
Months from 4L initiation date
CT/CIT
TT
CAR-T
HSCT

LIMITATIONS

- In this study, response rates may be under reported since unlike in clinical trials, the decision of whether or when to evaluate these outcomes may be associated with patient response to treatment. Additionally, response rates may be inaccurate or delayed in documentation in real-world historical EHR data.
- The sample sizes of patients who initiated CAR-T and HSCT were relatively small in this study.

CONCLUSIONS

- In this real-world US study, a majority of patients with R/R DLBCL were treated with a third or fourth line of chemotherapy/chemoimmunotherapy or targeted therapy and had poor clinical outcomes.
- Though patients who received CAR-T had better clinical outcomes, the number of patients who received this therapy was low and the characteristics of these patients (eg, younger age) differed from those who received non-cell therapies.
- In the treatment landscape during years 2014 through 2020, the prognosis of most patients with R/R DLBCL remained poor, underscoring the need for the development of more efficacious treatment options, especially for patients who cannot be offered CAR-T.

ACKNOWLEDGEMENTS

Funding sources: This research and preparation of this poster was supported by ADC Therapeutics.
Acknowledgement: Melissa Lingohr-Smith and Jay Lin from Novosys Health provided writing support for the preparation of this poster. Jackson Tang from Asclepius Analytics provided consultation service for the research.

REFERENCES

1. Liu Y, Barta SK. *Am J Hematol*. 2019;94:604-616.
2. Coiffier B, Thieblemont C, Van Den Neste E, et al. *Blood*. 2010;116:2040-2045.
3. Wang L, Li L-R, Young KH. *J Hematol Oncol*. 2020;13:175.
4. Thornton Snider J, Brauer M, Kee R, et al. *Am J Manag Care*. 2019;25:379-386.

CONTACT INFORMATION

Laura Liao email: Laura.Liao@adctherapeutics.com

| Connecting Hematology For Clinical and Research Excellence

EHA2021 VIRTUAL JUNE 9 - 17, 2021