Long-Term Responses With Loncastuximab Tesirine: Updated Results From LOTIS-2, the Pivotal Phase 2 Study of Patients With Relapsed/Refractory Diffuse Large B-cell Lymphoma

Paolo F Caimi[1*], Weiyun Z Ai[2], Juan Pablo Alderuccio[3], Kirit M Ardeshna[4], Mehdi Hamadani[5], Brian Hess[6], Brad S Kahl[7], John Radford[8], Melhem Solh[9], Anastasios Stathis[10], Pier Luigi Zinzani[11,12], Ying Wang[13], Yajuan Qin[13], Luqiang Wang[13], Zhiying Cindy Xu[13], Carmelo Carlo-Stella[14]

[1]Cleveland Clinic Taussig Cancer Center, Cleveland, OH, USA; [2]Helen Diller Family Comprehensive Cancer Center, University of California, San Francisco, CA, USA; [3]Sylvester Comprehensive Cancer Center, University of Miami Miller School of Medicine, Miami, FL, USA; [4]University College London Hospitals NHS Foundation Trust, London, UK; [5]Division of Hematology and Oncology, Medical College of Wisconsin, Milwaukee, WI, USA; [6]Medical University of South Carolina, Charleston, SC, USA; [7]Washington University, St. Louis, MO, USA; [8]NIHR Clinical Research Facility, University of Manchester and the Christie NHS Foundation Trust, Manchester Academic Health Science Centre, Manchester, UK; [9]Blood and Marrow Transplant Program at Northside Hospital, Atlanta, GA, USA; [10]Oncology Institute of Southern Switzerland, EOC, Bellinzona, Switzerland; [11]IRCCS Azienda Ospedaliero-Universitaria di Bologna, Istituto di Ematologia "Seràgnoli", Bologna, Italy; [12]Dipartimento di Scienze Mediche e Chirurgiche, Università di Bologna, Bologna, Italy; [13]ADC Therapeutics America, Inc., Murray Hill, NJ, USA; [14]Department of Biomedical Sciences, Humanitas University, and Department of Oncology and Hematology, Humanitas Research Hospital–IRCCS, Milano, Italy

P1132

SUMMARY

AIM Present long-term efficacy and safety data in patients with R/R DLBCL treated with Lonca in the LOTIS-2 study

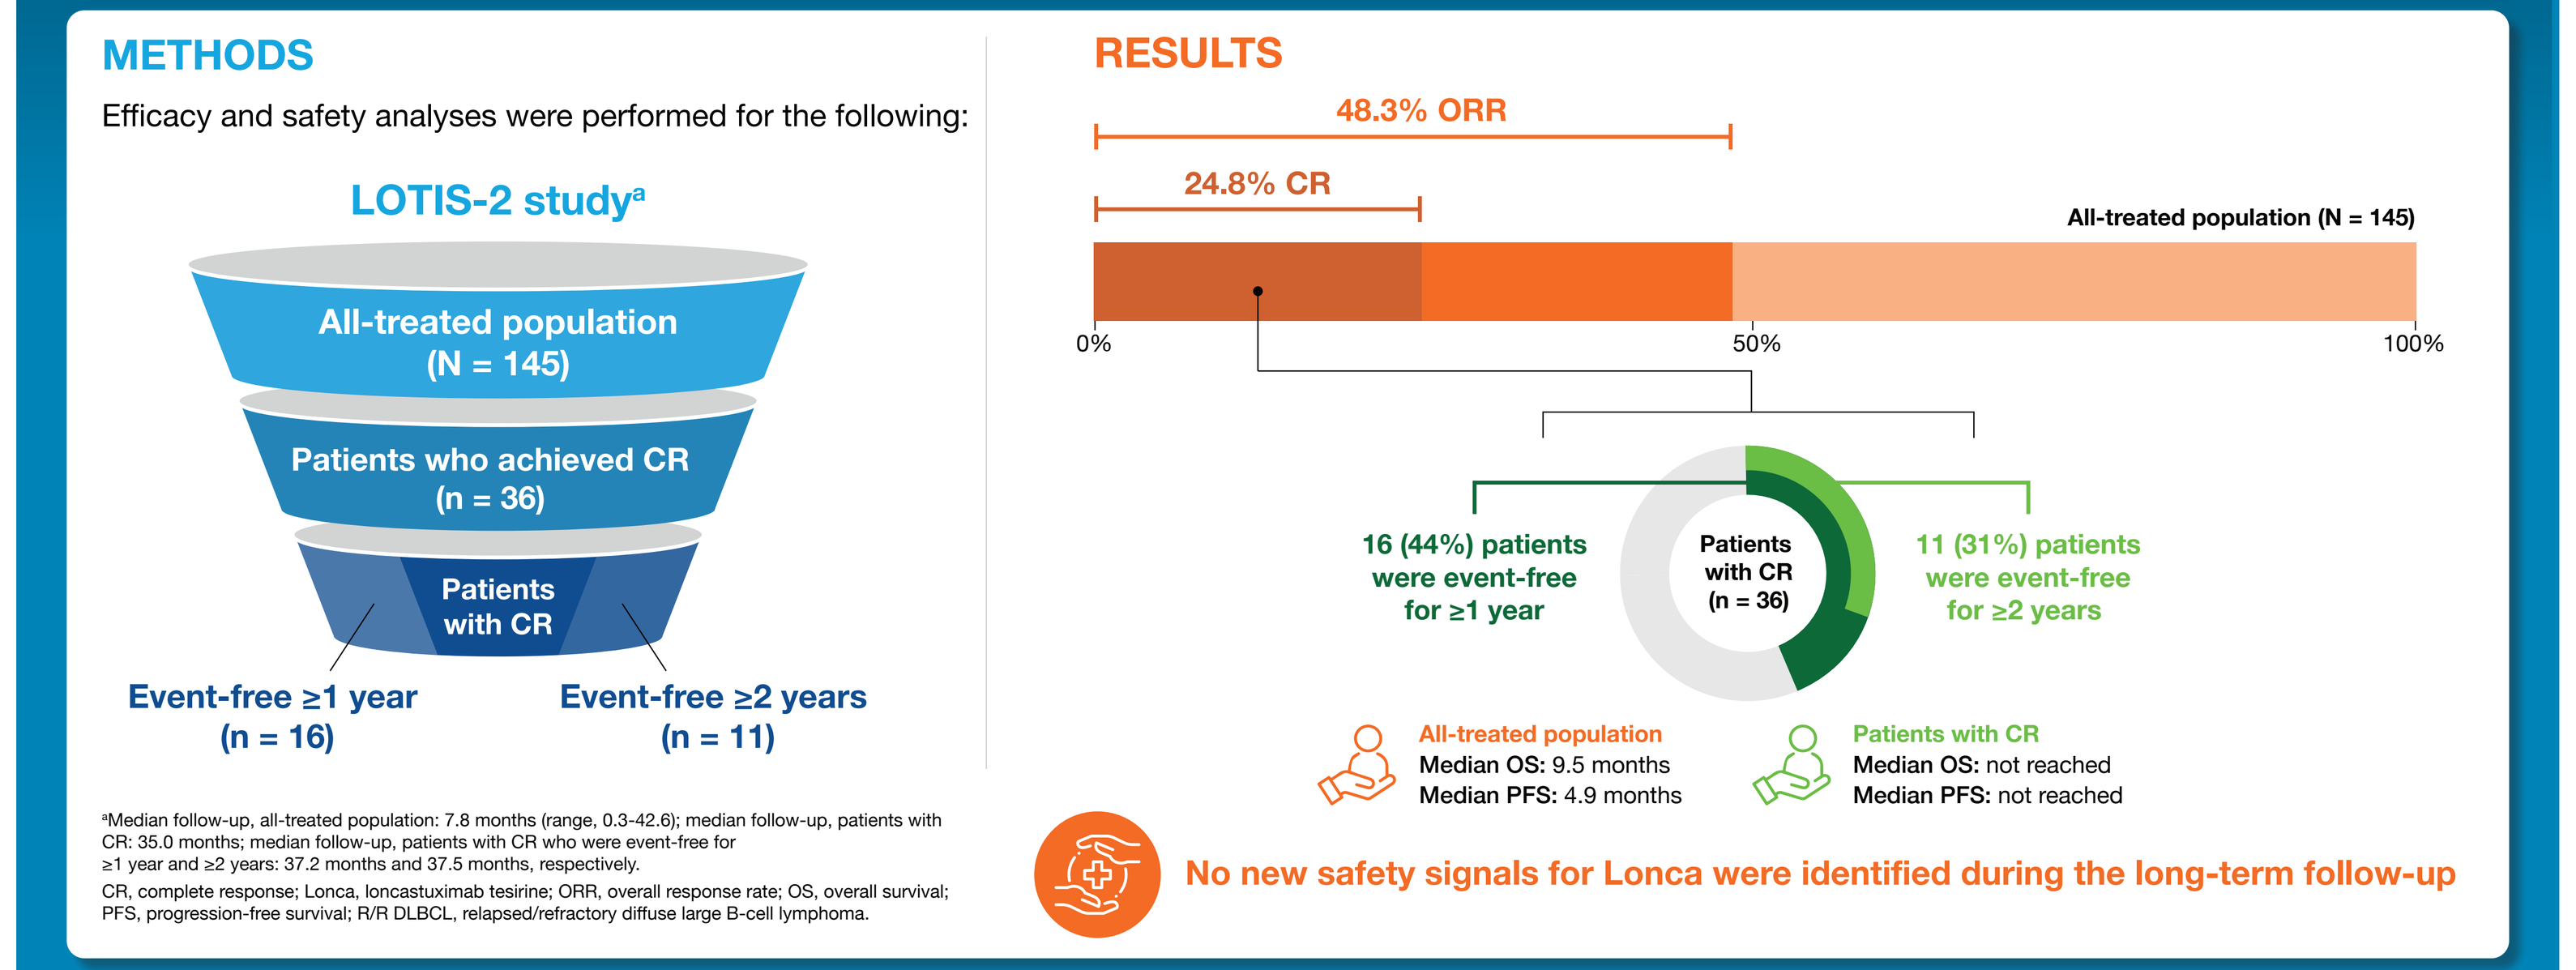
METHODS
Efficacy and safety analyses were performed for the following:
LOTIS-2 study[a]
All-treated population (N = 145)
Patients who achieved CR (n = 36)
Patients with CR
Event-free ≥1 year (n = 16)
Event-free ≥2 years (n = 11)
[a]Median follow-up, all-treated population: 7.8 months (range, 0.3-42.6); median follow-up, patients with CR: 35.0 months; median follow-up, patients with CR who were event-free for ≥1 year and ≥2 years: 37.2 months and 37.5 months, respectively.
CR, complete response; Lonca, loncastuximab tesirine; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; R/R DLBCL, relapsed/refractory diffuse large B-cell lymphoma.
RESULTS
48.3% ORR
24.8% CR
All-treated population (N = 145)
0%
50%
100%
16 (44%) patients were event-free for ≥1 year
Patients with CR (n = 36)
11 (31%) patients were event-free for ≥2 years
All-treated population
Median OS: 9.5 months
Median PFS: 4.9 months
Patients with CR
Median OS: not reached
Median PFS: not reached
No new safety signals for Lonca were identified during the long-term follow-up

CONCLUSIONS After the long-term follow-up, Lonca continued to demonstrate durable responses and an acceptable safety and tolerability profile in R/R DLBCL

INTRODUCTION

- Patients with diffuse large B-cell lymphoma (DLBCL) who relapse after stem cell transplant (SCT) or chimeric antigen receptor T-cell therapy or are refractory to second-line therapy have a poor prognosis and few treatment options[1,2]
- There is an unmet need for accessible therapies with manageable toxicity profiles that have demonstrated long-term disease control in patients with relapsed/refractory (R/R) DLBCL
- Loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]), an anti-CD19 antibody conjugated to a pyrrolobenzodiazepine dimer, demonstrated single-agent antitumor activity in LOTIS-2, the pivotal phase 2 study, in heavily pretreated patients with R/R DLBCL[3,4]
 - Lonca monotherapy was approved in the US in 2021 and in Europe in 2022[5,6]
- A follow-up analysis (data cutoff: March 1, 2021), at a median (range) of 7.8 (0.3 to 31.0) months, showed a similar overall response rate (ORR) to the primary analysis, with a complete response (CR) rate of 24.8% and a median duration of response (DOR) of 13.4 months[7]

OBJECTIVE

- To present updated long-term efficacy and safety results from patients with R/R DLBCL treated with Lonca in the phase 2 LOTIS-2 study (NCT03589469), including for subgroups of patients with durable CR

METHODS

Study Design

- LOTIS-2 was a multicenter, open-label, single-arm, phase 2 study of Lonca monotherapy in patients with R/R DLBCL after ≥2 prior systemic therapies, with measurable disease (2014 Lugano criteria[8]) and an Eastern Cooperative Oncology Group (ECOG) performance status of 0-2[4]
- Intravenous Lonca was administered every 3 weeks on day 1 of each 21-day cycle at a dose of 0.15 mg/kg for 2 cycles followed by 0.075 mg/kg for subsequent cycles
- Follow-up was every 12 weeks for up to 3 years after the end of treatment

Efficacy and Safety Outcomes

- The primary endpoint was the ORR (2014 Lugano criteria[8])
- Secondary endpoints included the CR rate, DOR, progression-free survival (PFS), and overall survival (OS)
- Safety endpoints included the frequency and severity of treatment-emergent adverse events (TEAEs)

Statistical Analysis

- The data cutoff for this analysis was September 15, 2022
- Efficacy and safety analyses were performed for the following:
 - All-treated patients
 - Patients with a CR
 - Patients with a CR who were event-free (defined as no progressive disease or death) for ≥1 year from day 1 of cycle 1
 - Patients with a CR who were event-free for ≥2 years from day 1 of cycle 1

RESULTS

Patient Population

- The median (range) follow-up was 7.8 (0.3 to 42.6) months, and 145 patients had received at least 1 dose of Lonca
- In patients with a CR, the median (range) duration of follow-up was 35.0 (4.4 to 42.6) months
- The baseline patient demographics and disease characteristics, both in the all-treated population and in subgroups of patients with a long-term response, are shown in **Table 1**
- The median (range) number of treatment cycles was 3.0 (1 to 26) in the all-treated population, 8.0 (1 to 26) in patients with a CR, 12.5 (1 to 26) in patients with a CR who were event free for ≥1 year, and 13.0 (1 to 22) in patients with a CR who were event free for ≥2 years

Efficacy Outcomes

- The ORR was 48.3% (70/145), with a CR rate of 24.8% (36/145)
- Among patients with a CR, 44% (16/36) and 31% (11/36) were event-free for ≥1 and ≥2 years, respectively
 - All 11 patients with a CR who were event-free for ≥2 years were censored due to patient discontinuation of the study
- The median (range) time to response was 41.0 (35 to 247) days for all responders and 42.0 (36 to 247) days for patients with a CR
- The median (95% CI) DOR was 13.37 (6.87, –) months in the all-treated population and was not reached among patients with a CR (**Figure 1**)
- The median (95% CI) PFS (4.93 months [2.89 to 8.3]) and OS (9.53 months [6.7 to 11.5]) in the all-treated population and in patients with a CR (not reached for both PFS and OS) are shown in **Figures 2A** and **2B**
- Additional efficacy outcomes for the all-treated population and the subset of patients with CR are summarized in **Table 2**

Table 1: Baseline patient demographics and disease characteristics

	All-treated N = 145	Best response of CR at any time n = 36	Patients with CR who were event-free ≥1 year n = 16	Patients with CR who were event-free ≥2 years n = 11
Sex, n (%)				
Female	60 (41.4)	22 (61.1)	13 (81.3)	9 (81.8)
Age				
Median, years (range)	66.0 (23, 94)	67.5 (45, 94)	71.0 (53, 84)	70.0 (53, 82)
ECOG score, n (%)				
0	58 (40.0)	19 (52.8)	9 (56.3)	7 (63.6)
1	78 (53.8)	14 (38.9)	6 (37.5)	3 (27.3)
2	9 (6.2)	3 (8.2)	1 (6.3)	1 (9.1)
Histology,[a] n (%)				
DLBCL, NOS	128 (88.3)	31 (86.1)	11 (68.8)	8 (72.7)
HGBCL[b]	10 (6.9)	5 (13.9)	5 (31.3)	3 (27.3)
Primary mediastinal DLBCL	7 (4.8)	0	0	0
Transformed DLBCL, n (%)	30 (20.7)	7 (19.4)	4 (25.0)	2 (18.2)
Double/triple hit, n (%)				
Double hit	12 (8.3)	5 (13.9)	5 (31.3)	3 (27.3)
Triple hit	3 (2.1)	0	0	0
Stage, n (%)				
I-II	33 (22.8)	9 (25.0)	3 (18.8)	2 (18.2)
III-IV	112 (77.2)	27 (75.0)	13 (81.3)	9 (81.8)
Prior systemic therapies				
Median (range)	3.0 (2, 7)	3.0 (2, 7)	2.0 (2, 7)	2.0 (2, 7)
Primary refractory, n (%)	29 (20.0)	5 (13.9)	2 (12.5)	0
Refractory to last line of therapy	89 (61.4)	11 (30.6)	5 (31.3)	4 (36.4)
Prior stem cell transplant, n (%)	24 (16.6)	8 (22.2)	1 (6.3)	1 (9.1)
Prior CAR-T therapy, n (%)	14 (9.7)	3 (8.3)	2 (12.5)	0

CAR-T, chimeric antigen receptor T-cell; CR, complete response; DLBCL, diffuse large B-cell lymphoma; ECOG, Eastern Cooperative Oncology Group; HGBCL, high-grade B-cell lymphoma; NOS, not otherwise specified.
[a]Relapsed/refractory DLBCL was classified according to the 2016 WHO classification.
[b]The primary analyses reported HGBCL in 11 patients.

Figure 1. Kaplan–Meier curve of the DOR in the all-treated population and the subset of patients with a CR

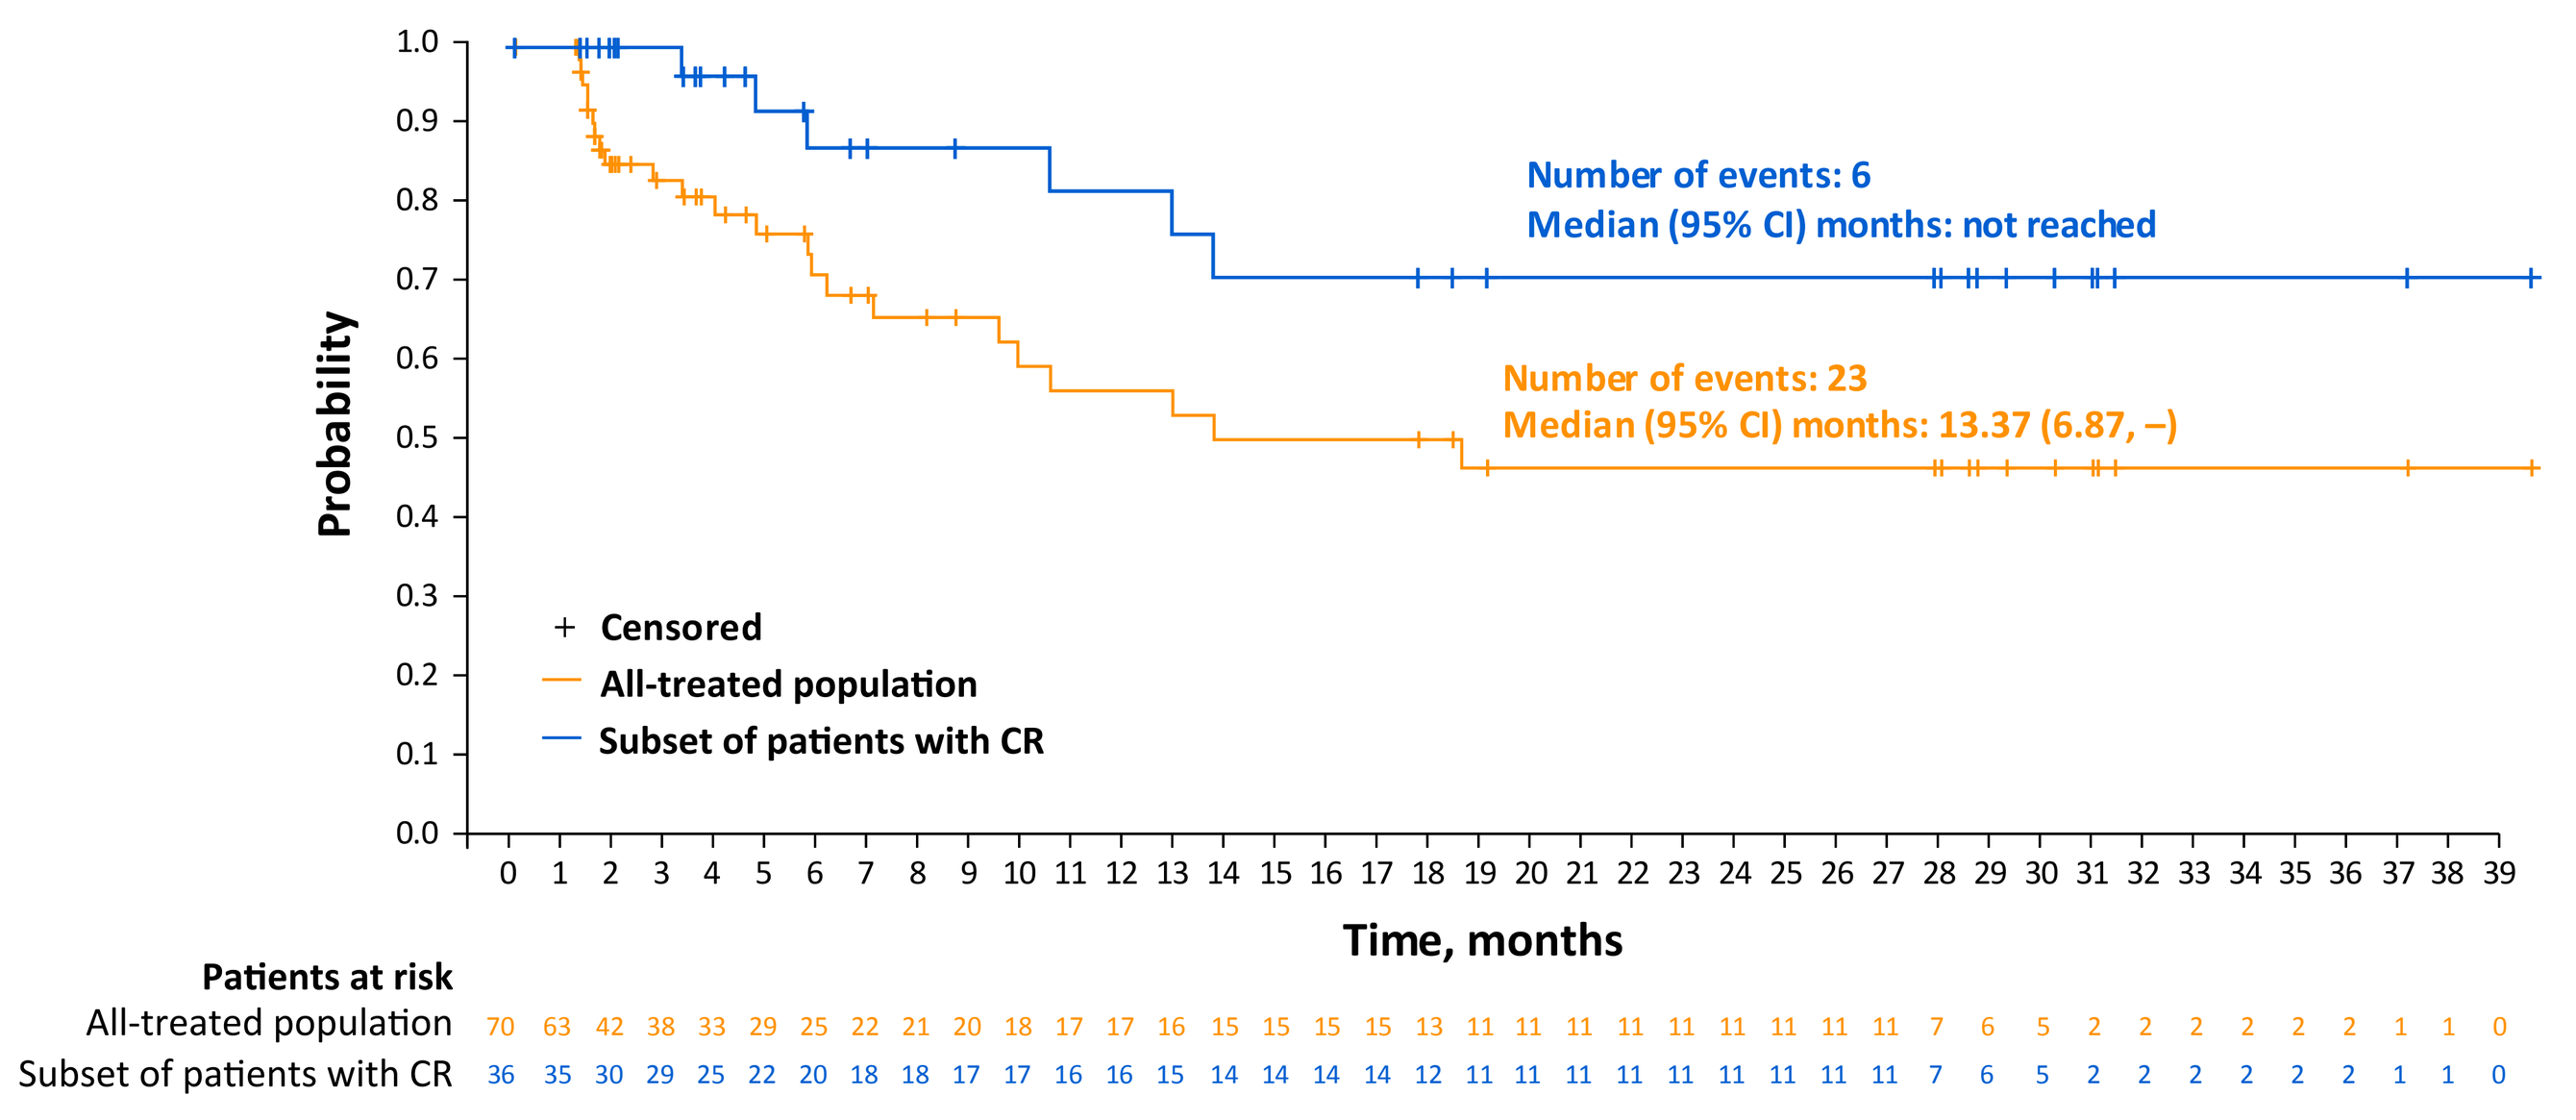
Probability
Number of events: 6
Median (95% CI) months: not reached
Number of events: 23
Median (95% CI) months: 13.37 (6.87, –)
+ Censored
All-treated population
Subset of patients with CR
Time, months
Patients at risk
All-treated population 70 63 42 38 33 29 25 22 21 20 18 17 17 16 15 15 15 15 13 11 11 11 11 11 11 11 11 11 7 6 5 2 2 2 2 2 2 1 1 0
Subset of patients with CR 36 35 30 29 25 22 20 18 18 17 17 16 16 15 14 14 14 14 12 11 11 11 11 11 11 11 11 11 7 6 5 2 2 2 2 2 2 1 1 0

CR, complete response; DOR, duration of response.

Figure 2. Kaplan–Meier curves of the (A) PFS in the all-treated population (N = 145) and the subset of patients with a best response of a CR (n = 36), and (B) OS in the all-treated population and the subset of patients with a best response of a CR (n = 36)

A.

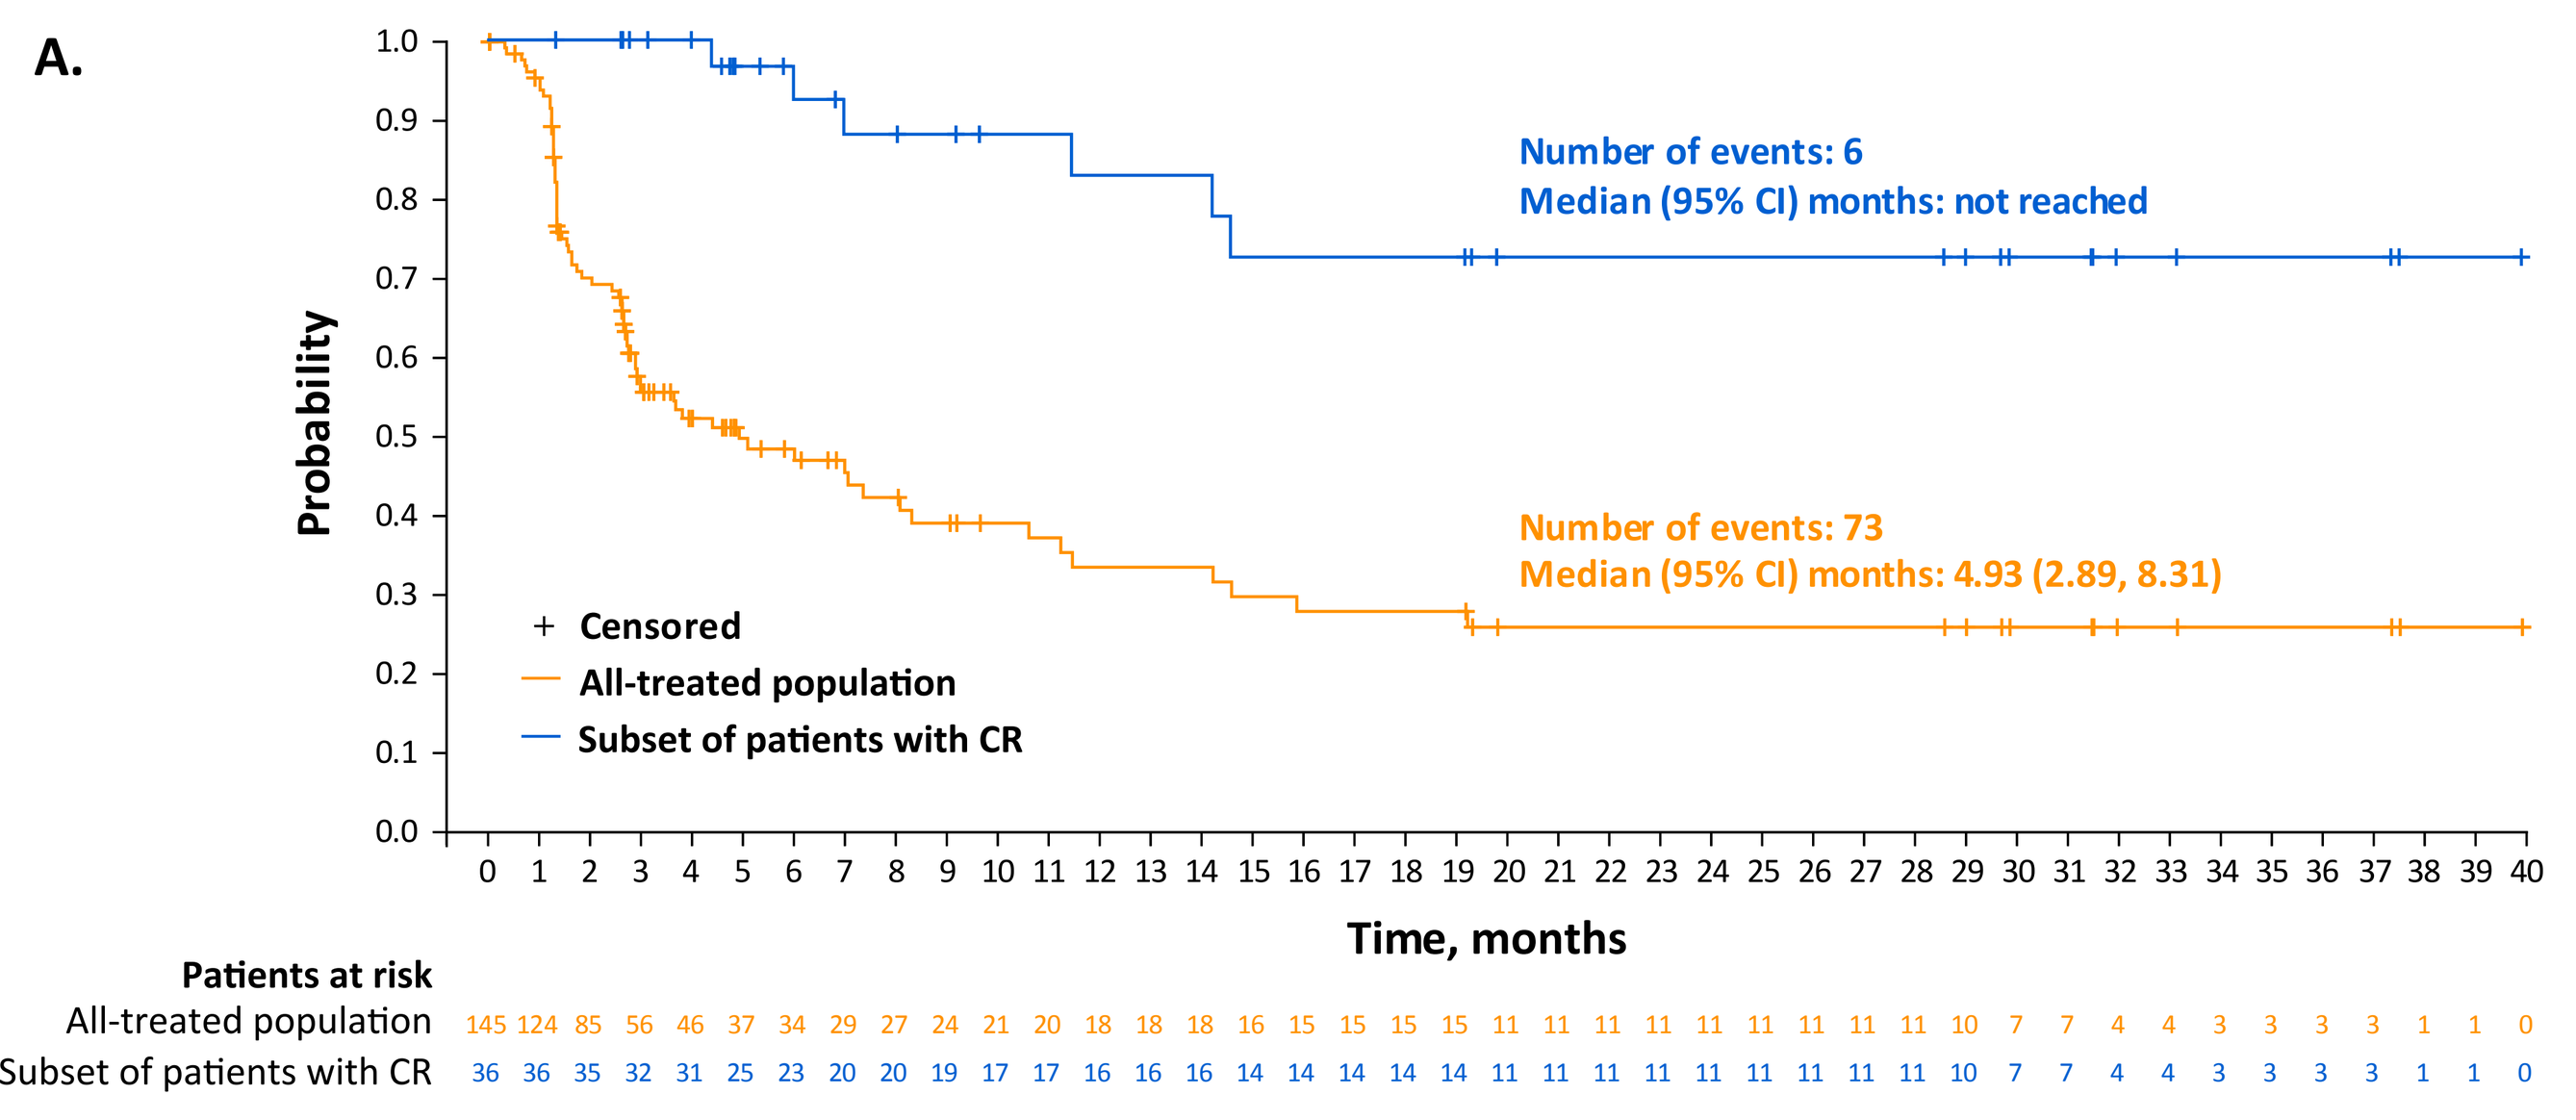
Probability
Number of events: 6
Median (95% CI) months: not reached
Number of events: 73
Median (95% CI) months: 4.93 (2.89, 8.31)
+ Censored
All-treated population
Subset of patients with CR
Time, months
Patients at risk
All-treated population 145 124 85 56 46 37 34 29 27 24 21 20 18 18 18 16 15 15 15 15 11 11 11 11 11 11 11 11 11 10 7 7 4 4 3 3 3 3 1 1 0
Subset of patients with CR 36 36 35 32 31 25 23 20 20 19 17 17 16 16 16 14 14 14 14 14 11 11 11 11 11 11 11 11 11 10 7 7 4 4 3 3 3 3 1 1 0

B.

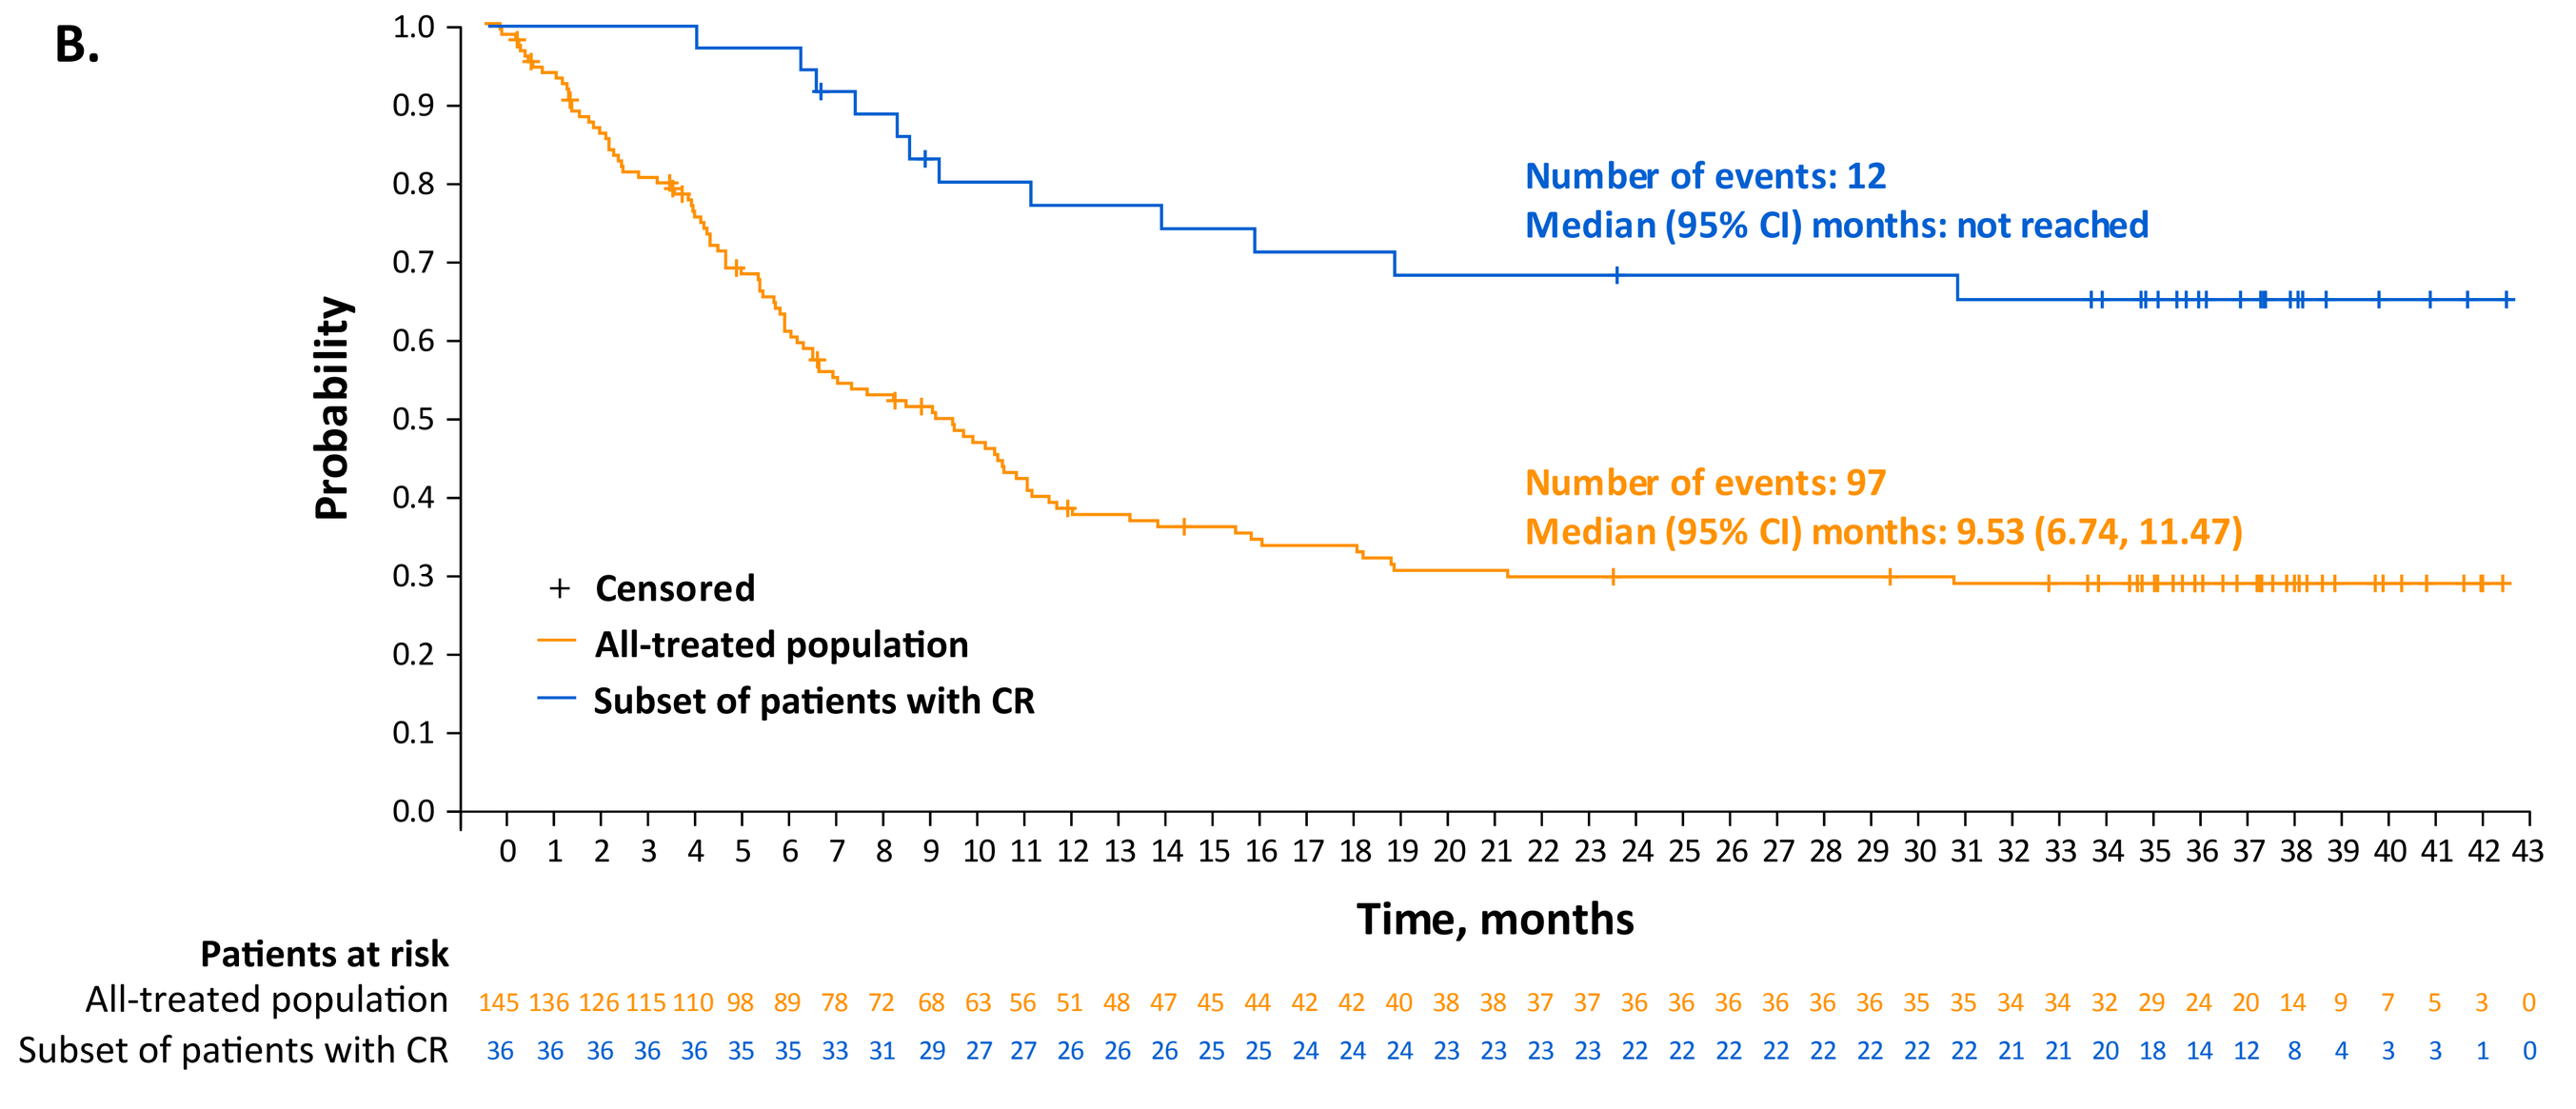
Probability
Number of events: 12
Median (95% CI) months: not reached
Number of events: 97
Median (95% CI) months: 9.53 (6.74, 11.47)
+ Censored
All-treated population
Subset of patients with CR
Time, months
Patients at risk
All-treated population 145 136 126 115 110 98 89 78 72 68 63 56 51 48 47 45 44 42 42 40 38 38 37 37 36 36 36 36 36 36 35 35 34 34 32 29 24 20 14 9 7 5 3 0
Subset of patients with CR 36 36 36 36 36 35 35 33 31 29 27 27 26 26 26 25 25 24 24 24 23 23 23 23 22 22 22 22 22 22 22 21 21 20 18 14 12 8 4 3 3 1 0

CR, complete response; OS, overall survival; PFS, progression-free survival.

Table 2: Summary of efficacy outcomes

	All-treated N = 145	Best response of CR n = 36
Median DOR, months (95% CI)	13.4 (6.9, –)	NR
Probability of maintaining response at 1 year	54.7% (37.9, 68.8)	82.8% (59.9, 93.3)
Probability of maintaining response at 2 years	44.6% (27.9, 60.0)	72.4% (48.1, 86.8)
Median PFS, months (95% CI)	4.9 (2.9, 8.3)	NR
Probability of maintaining PFS at 1 year	33.5% (23.3, 44.0)	82.9% (60.0, 93.3)
Probability of maintaining PFS at 2 years	25.9% (16.2, 36.7)	72.5% (48.2, 86.8)
Median OS, months (95% CI)	9.5 (6.7, 11.5)	NR
Probability of maintaining OS at 1 year	39.0% (30.7, 47.1)	77.1% (59.4, 87.9)
Probability of maintaining OS at 2 years	29.5% (22.0, 37.4)	68.2% (50.0, 81.0)

CR, complete response; DOR, duration of response; NR, not reached; OS, overall survival; PFS, progression-free survival.

- Among patients with a CR (n = 36), the median (range) duration of time patients remained treatment-free post-Lonca was 6.1 (1.0 to 37.5) months
 - In the subsets of patients who were event-free for ≥1 year and ≥2 years, the median (range) duration of time patients remained treatment-free post-Lonca was 24.8 (3.4 to 37.5) months and 27.7 (20.7 to 37.5) months, respectively
- Ten patients with a CR proceeded to SCT (**Figure 3**)
 - As assessed by principal investigators, 4 of the 5 patients with a record of response achieved a CR after SCT; the remaining one patient had disease progression

Figure 3. Swimmer plot for patients with CR

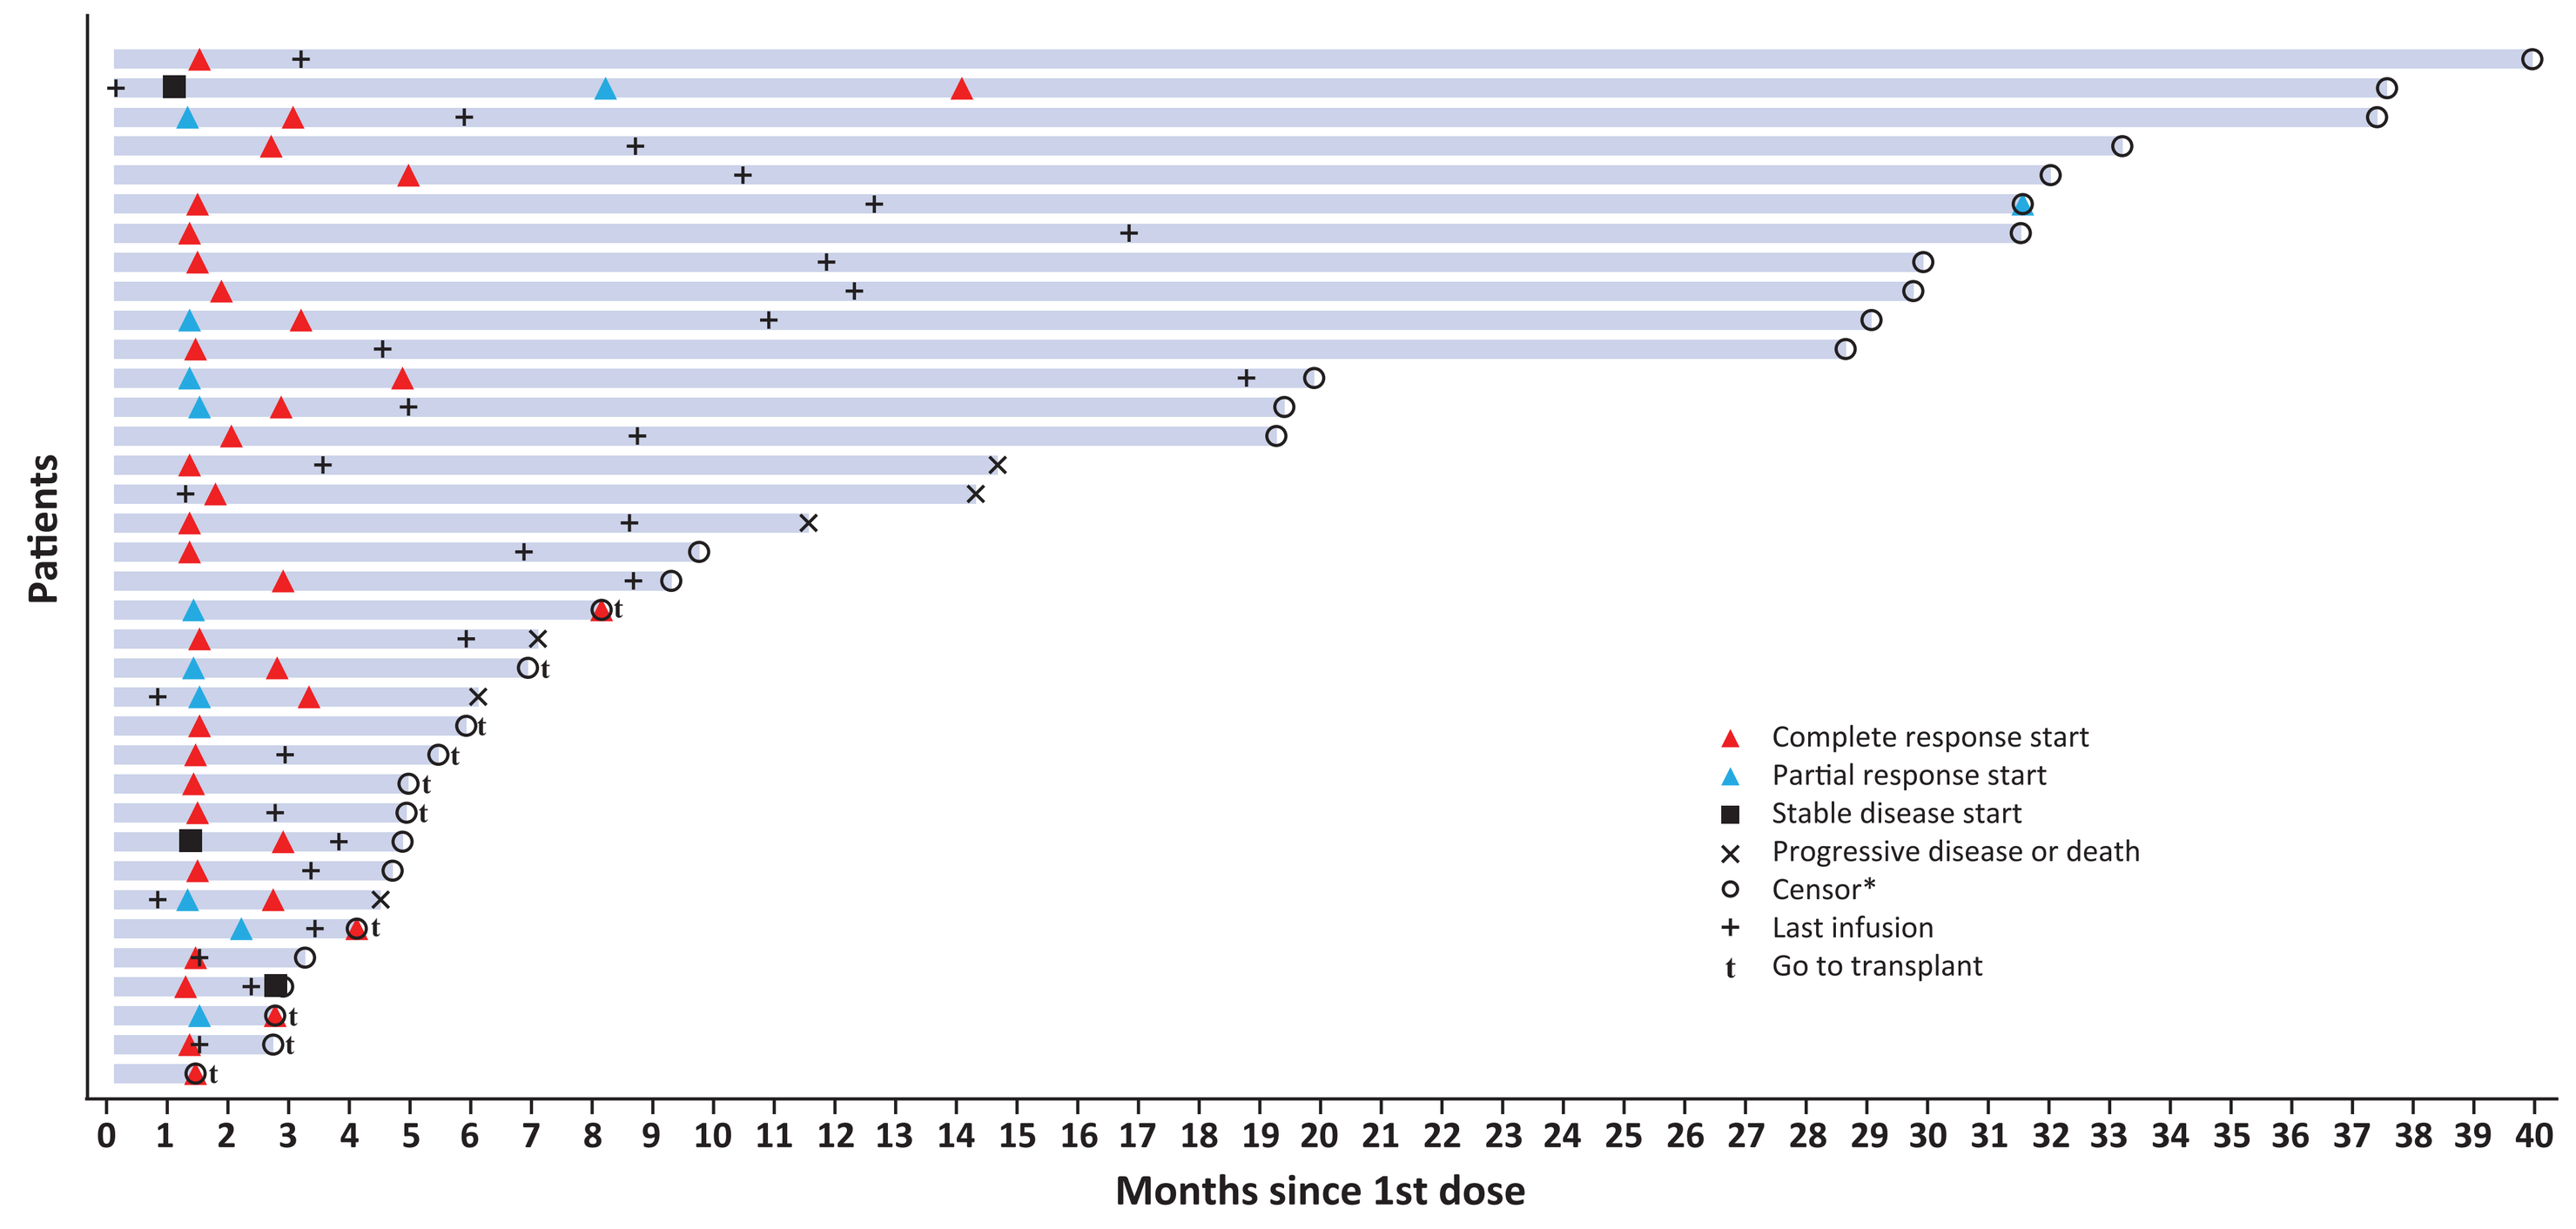
Patients
Complete response start
Partial response start
Stable disease start
Progressive disease or death
Censor*
Last infusion
Go to transplant
Months since 1st dose

Each bar represents one patient in the study. Response was determined by an independent reviewer.
*Reasons for censoring included study discontinuation, new anticancer treatment started (excluding SCT), no valid post-baseline assessment, or transplant. CR, complete response; SCT, stem cell transplant.

Safety Outcomes

- No new safety signals were identified during the long-term follow-up
- All-grade TEAEs were reported in 98.6% of the all-treated population and 100% of patients with a CR
 - All-grade TEAEs occurring in ≥30% of all patients were increased gamma-glutamyltransferase (GGT; 42%), neutropenia (40%), and thrombocytopenia (33%)
 - All-grade TEAEs occurring in ≥30% of patients with a CR were increased GGT (50%), neutropenia (42%), anemia (36%), thrombocytopenia (36%), peripheral edema (33%), and nausea (31%)
- Grade ≥3 TEAEs were reported in 73.8% of patients and in 75% of patients with a CR
 - Grade ≥3 TEAEs occurring in ≥10% of the all-treated population were neutropenia (26%), thrombocytopenia (18%), increased GGT (17%), and anemia (10%)
 - Grade ≥3 TEAEs occurring in ≥10% of patients with a CR were neutropenia (28%), increased GGT (19%), thrombocytopenia (19%), leukopenia (14%), and hypophosphatemia (11%)

CONCLUSIONS

- Among heavily pretreated patients in the LOTIS-2 study, Lonca continued to demonstrate durable, long-term responses with a manageable safety profile
 - Eleven of the 36 patients with a CR were event-free for ≥2 years with no evidence of disease and no new anticancer therapy post-Lonca
 - Patients with a CR maintained a median treatment-free period of 6.1 months from the last Lonca dose
- Further study is needed to identify factors predictive of long-term response to Lonca

Acknowledgments

Funding was provided by ADC Therapeutics SA. Medical writing support was provided by CiTRUS Health Group (United States) in accordance with Good Publication Practice (GPP 2022) guidelines.

Disclosures

PF Caimi: Consultant/advisor for Genmab, Bristol-Myers Squibb/Celgene, Genentech, Takeda, MEI Pharma, ADC Therapeutics, Novartis, and Kite Pharma. **WZ Ai:** Advisory boards for Acrotech Biopharma, ADC Therapeutics, BeiGene, Kymera Therapeutics, and Nurix Therapeutics; research funding from Nurix Therapeutics. **JP Alderuccio:** Honoraria from OncLive and OncInfo; consultant and research funding with ADC Therapeutics; consultant for Genentech; immediate family member has served on the advisory boards of Puma Biotechnology, Inovio Pharmaceuticals, Agios Pharmaceuticals, Forma Therapeutics, and Foundation Medicine. **KM Ardeshna:** Honoraria from Bristol Myers Squibb, Gilead, and Novartis. **M Hamadani:** Consultant for AbGenomics, ADC Therapeutics, Celgene Corporation, Incyte Corporation, Janssen R&D, Omeros, Pharmacyclics, TeneoBio, and Verastem; speakers' bureau for AstraZeneca, BeiGene, and Sanofi Genzyme; research support for Astellas Pharma, Spectrum Pharmaceuticals, and Takeda. **B Hess:** Consultant for AstraZeneca, Bristol Myers Squibb, and ADC Therapeutics; speakers' bureau for Bristol Myers Squibb. **BS Kahl:** Consultant for Genentech, ADC Therapeutics, AbbVie, AstraZeneca, BeiGene, Pharmacyclics, Celgene/BMS, TG Therapeutics, HUTCHMED, MEI, MTEM, Kite, Epizyme, Takeda, Genmab, Incyte, and Eli Lilly; research funding from Genentech, ADC Therapeutics, AbbVie, AstraZeneca, and BeiGene. **J Radford**: Consultant/advisor for ADC Therapeutics, BMS, Kite Pharma, Novartis, and Takeda; speaker for ADC Therapeutics, Seattle Genetics, and Takeda; stock ownership of ADC Therapeutics and AstraZeneca (spouse); honoraria/expert testimony with ADC Therapeutics and Takeda; research funding from Takeda. **M Solh:** Speakers' bureau for Bristol Myers Squibb, Sanofi, and GSK; consultant/advisor for ADC Therapeutics and Genentech. **A Stathis:** Consultant/honoraria with Eli Lilly, Bayer, Roche, Novartis, Janssen Oncology, and AstraZeneca; research funding from ADC Therapeutics, Pfizer, Roche, Novartis, Bayer, Eli Lilly, MEI Pharma, Cellestia, Debiopharm Group, Merck/Merck Sharp & Dohme, AbbVie, Amgen, AstraZeneca, Incyte, Loxo, and Philogen. **PL Zinzani:** Consultant for EUSA Pharma, Merck Sharp & Dohme, Sanofi, and Verastem; advisory committee with ADC Therapeutics and Sandoz; speakers' bureau or advisory committee with Bristol-Myers Squibb, Celltrion, EUSA Pharma, Gilead, Janssen-Cilag, Kyowa Kirin, Merck Sharp & Dohme, Roche, Servier, Takeda, TG Therapeutics, and Verastem. **Y Wang:** Employee with stock options at ADC Therapeutics; immediate family member employed by/stock ownership with Johnson & Johnson. **Y Qin:** Employee with stock options at ADC Therapeutics. **L Wang:** Employee with stock options at ADC Therapeutics. **C Xu:** Employee with stock options at ADC Therapeutics. **C Carlo-Stella:** Consultant/advisor for ADC Therapeutics, Celgene/Bristol Myers Squibb, Karyopharm, Novartis, Sanofi, Roche, Merck Sharp & Dohme, and Scenic Biotech; honoraria from Merck Sharp & Dohme, Janssen Oncology, AstraZeneca, Celgene, Incyte, Gilead Sciences, and Roche.

References

1. Crump, M, et al. *Blood*. 2017;130:1800-1808.
2. Chow, VA, et al. *Am J Hematol*. 2019;94:e209-213.
3. Zammarchi, F, et al. *Blood*. 2018;131:1094-1105.
4. Caimi, PF, et al. *Lancet Oncol*. 2021;22:790-800.
5. ZYNLONTA® US prescribing information. Available at: https://www.accessdata.fda.gov/drugsatfda_docs/label/2021/761196s000lbl.pdf. Accessed April 12, 2023.
6. ZYNLONTA® summary of product characteristics. Available at: https://www.ema.europa.eu/en/medicines/human/EPAR/zynlonta. Accessed April 12, 2023.
7. Zinzani, PL, et al. Poster presented at: International Conference on Malignant Lymphoma Virtual Congress, June 18-22, 2021.
8. Cheson, BD, et al. *J Clin Oncol*. 2014;32:3059-3068.

Contact information

Paolo F Caimi, MD: caimip@ccf.org

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at the European Hematology Association 2023 Hybrid Congress. June 8-11, 2023, Frankfurt, Germany, and Virtual