Clinical activity of loncastuximab tesirine plus ibrutinib in non-Hodgkin lymphoma: Updated LOTIS-3 Phase 1 results

Julien Depaus[1], Nina Wagner-Johnston[2], Pier Luigi Zinzani[3], Tycel J. Phillips[4], Joseph Maly[5], Silvia Ferrari[6], Emmanuel Bachy[7], Locke J. Bryan[8], Vincent Delwail[9], Murali Janakiram[10], Sophie de Guibert[11], Monica Tani[12], Vivian Dai[13], Karin Havenith[14], Joseph Boni[13], Xiaomin He[13], Annette Ervin-Haynes[13], Carmelo Carlo-Stella[15]

[1]Department of Hematology, CHU UCL Namur site Godinne, Yvoir, Belgium; [2]Division of Oncology, The Sidney Kimmel Comprehensive Cancer Center, Johns Hopkins University School of Medicine, Baltimore, MD, USA; [3]IRCCS Azienda Ospedaliero-Universitaria di Bologna Istituto di Ematologia "Seràgnoli", and Dipartimento di Medicina Specialistica, Diagnostica e Sperimentale Università di Bologna, Bologna, Italy; [4]University of Michigan Comprehensive Cancer Center, Ann Arbor, MI, USA; [5]Norton Cancer Institute, Louisville, KY, USA; [6]Hematology and Bone Marrow Transplant Unit, Azienda Ospedaliera Papa Giovanni XXIII, Bergamo, Italy; [7]Department of Hematology, Hôpital Lyon Sud, Pierre-Bénite, France; [8]Department of Medicine, Division of Hematology/Oncology, Georgia Cancer Center at Augusta University, Augusta, GA, USA; [9]Department of Hematology and Cell Therapy, Centre Hospitalier Universitaire de Poitiers, Poitiers, France; [10]Division of Hematology, Oncology and Transplantation, University of Minnesota, Minneapolis, MN, USA; [11]Department of Clinical Hematology, Centre Hospitalier Universitaire de Rennes Hôpital Pontchaillou, Rennes, France; [12]Unit of Hematology, Santa Maria delle Crioci Hospital, Ravenna, Italy; [13]Clinical Development, ADC Therapeutics America, Inc., Murray Hill, NJ, USA; [14]ADC Therapeutics (UK) Ltd, London, UK; [15]Department of Biomedical Sciences, Humanitas University and IRCCS Humanitas Research Hospital, Milan, Italy

BACKGROUND

- Prognosis is poor for patients with relapsed/refractory (R/R) diffuse large B-cell lymphoma (DLBCL) and mantle cell lymphoma (MCL) and effective, less toxic treatment options are needed[1,2]
- Combination therapy using agents with different mechanisms of action may improve therapeutic outcomes
- We investigated the combination of loncastuximab tesirine (Lonca; an antibody-drug conjugate composed of a humanized anti-CD19 monoclonal antibody conjugated to a pyrrolobenzodiazepine dimer toxin) with ibrutinib (a small-molecule inhibitor of Bruton's tyrosine kinase) in patients with R/R DLBCL or R/R MCL (LOTIS-3)
 - Initial Phase 1 results identified the maximum tolerated dose (MTD) as Lonca 60 µg/kg intravenous every 3 weeks and oral ibrutinib 560 mg/day taken orally[3,4]
- Interim safety, efficacy and pharmacokinetic (PK) data from the Phase 1 portion of the study were previously presented at EHA 2020 and ASH 2020[3,4]
- Enrollment for the Phase 1 portion of the study is now complete
 - Here we present safety and efficacy data (data cut-off March 01, 2021) and PK data (data cut-off August 20, 2020) from the Phase 1 portion of a Phase 1/2 study (NCT03684694)

METHODS

Study design

- The study is an open-label, single-arm, combination study with a dose-escalation phase (Phase 1) and a dose-expansion phase (Phase 2) (**Figure 1**)
- Eligible patients were male or female; aged ≥18 years; had a pathologic diagnosis of R/R DLBCL for whom standard treatment was unsuccessful or who were intolerant to standard therapy, or had a pathologic diagnosis of R/R MCL with ≥1 prior therapy; and an Eastern Cooperative Oncology Group performance status of 0–2
 - Patients previously treated with Lonca or ibrutinib or other Bruton's tyrosine kinase inhibitors were excluded from the study

Figure 1. Study design

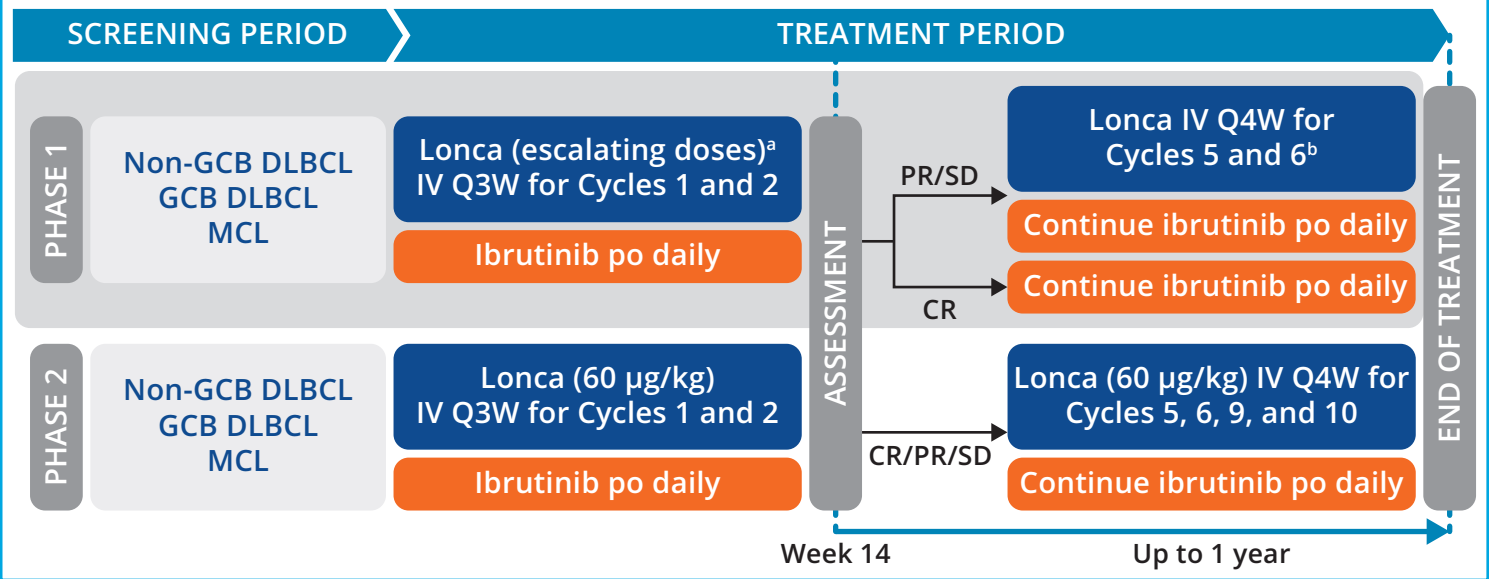

SCREENING PERIOD
TREATMENT PERIOD
PHASE 1
Non-GCB DLBCL
GCB DLBCL
MCL
Lonca (escalating doses)a IV Q3W for Cycles 1 and 2
Ibrutinib po daily
ASSESSMENT
PR/SD
CR
Lonca IV Q4W for Cycles 5 and 6b
Continue ibrutinib po daily
Continue ibrutinib po daily
PHASE 2
Non-GCB DLBCL
GCB DLBCL
MCL
Lonca (60 µg/kg) IV Q3W for Cycles 1 and 2
Ibrutinib po daily
CR/PR/SD
Lonca (60 µg/kg) IV Q4W for Cycles 5, 6, 9, and 10
Continue ibrutinib po daily
END OF TREATMENT
Week 14
Up to 1 year

[a]Doses of 60 µg/kg and 90 µg/kg. [b]At the discretion of the investigator.

CR, complete response; DLBCL, diffuse large B-cell lymphoma; GCB, germinal center B-cell; IV, intravenous; Lonca, loncastuximab tesirine; MCL, mantle cell lymphoma; po, taken orally; PR, partial response; SD, stable disease; Q3W, every 3 weeks; Q4W, every 4 weeks.

Objectives

- The primary objectives of Phase 1 were to characterize the safety and tolerability of Lonca plus ibrutinib and identify the MTD/recommended Phase 2 dose and schedule for future studies
- Secondary objectives included evaluation of antitumor effects of Lonca plus ibrutinib and characterization of PK profile of Lonca when combined with ibrutinib

Endpoints

- Primary endpoints included frequency and severity of adverse events
- Secondary endpoints included investigator-determined overall response rate (ORR [complete response or partial response]; according to the 2014 Lugano Classification[5]) and concentrations and PK parameters

RESULTS

Patient demographics and baseline characteristics

- At data cut-off (March 01, 2021), 30 patients with DLBCL (24 with non-germinal center B-cell [non-GCB] DLBCL and 6 with GCB DLBCL) and 7 patients with MCL were included in the study
- Overall, the median patient age was 72 years (range 40–91) and 27 (73.0%) had Stage IV disease (**Table 1**)
- Patients received a median of 2 (range 1–6) prior therapies
 - Eight (21.6%) patients were primary refractory and 18 (48.6%) were refractory to their last line of systemic therapy; 24 (64.9%) and 17 (45.9%) had relapsed with first-line and last-line systemic therapy, respectively

Table 1. Baseline characteristics

Characteristic	DLBCL (n=30)	MCL (n=7)	All patients (n=37)
Sex, n (%)			
Male	21 (70.0)	6 (85.7)	27 (73.0)
Age, years, median (range)	72 (40–91)	69 (54–89)	72 (40–91)
ECOG status, n (%)			
0	16 (53.3)	4 (57.1)	20 (54.1)
1	11 (36.7)	3 (42.9)	14 (37.8)
2	3 (10.0)	0	3 (8.1)
NHL subtype, n (%)			
Non-GCB DLBCL	24 (80.0)	–	24 (64.9)
GCB DLBCL	6 (20.0)	–	6 (16.2)
MCL	–	7 (100)	7 (18.9)
Disease stage[a], n (%)			
Stage I	1 (3.3)	0	1 (2.7)
Stage II	3 (10.0)	1 (14.3)	4 (10.8)
Stage III	4 (13.3)	1 (14.3)	5 (13.5)
Stage IV	22 (73.3)	5 (71.4)	27 (73.0)
Number of prior therapies[b]			
Median (range)	2 (1–6)	2 (1–4)	2 (1–6)
First-line prior systemic therapy response, n (%)[c]			
Relapsed	20 (66.7)	4 (57.1)	24 (64.9)
Refractory	7 (23.3)	1 (14.3)	8 (21.6)
Other	3 (10.0)	2 (28.6)	5 (13.5)
Last-line prior systemic therapy response, n (%)[c,d]			
Relapsed	13 (43.3)	4 (57.1)	17 (45.9)
Refractory	17 (56.7)	1 (14.3)	18 (48.6)
Other	0	2 (28.6)	2 (5.4)
Prior SCT, n (%)			
Autologous	2 (6.7)	1 (14.3)	3 (8.1)
Allogeneic	0	1 (14.3)	1 (2.7)

[a]Ann Arbor Criteria; [b]Prior SCT is included. For patients who received an autologous transplant, the mobilization regimen was considered a line of therapy if it was chemotherapy based and distinct from the other previous lines of treatment. [c]Systemic therapy; Relapsed: complete or partial response, followed by relapse; Refractory: stable disease or progressive disease; Other: missing data or not evaluable. [d]If SCT is most recent line, the variable is defined as response to the therapy immediately preceding SCT.

DLBCL, diffuse large B-cell lymphoma; ECOG, Eastern Cooperative Oncology Group; GCB, germinal center B-cell; MCL, mantle cell lymphoma; NHL, non-Hodgkin lymphoma; SCT, stell cell transplant.

Treatment

- Patients received a median of 2 cycles (range 1–4) of Lonca and 4 cycles (range 1–15) of ibrutinib
 - Median (range) Lonca treatment duration was 22 (1–127) days
 - Median (range) ibrutinib treatment duration was 105 (18–379) days

Safety

- Treatment-emergent adverse events (TEAEs) were reported in 37/37 (100%) patients. The most common TEAEs (≥20%) were thrombocytopenia (12 [32.4%]); anemia (9 [24.3%]); diarrhea (9 [24.3%]); and fatigue, nausea, and rash (all 8 [21.6%])
- Grade ≥3 TEAEs were reported in 24/37 (64.9%) patients. The most common (≥5%) were anemia (4 [10.8%]); neutropenia (4 [10.8%]); and thrombocytopenia, fatigue, and acute kidney injury (all 2 [5.4%])
- TEAEs that led to dose delay, reduction, or interruption were reported in 19 (51.4%) patients
- TEAEs that led to treatment discontinuation were reported in 5 (13.5%) patients

Efficacy

- ORR by investigator was 62.2% (35.1% and 27.0% for complete and partial response, respectively)
- ORR for patients with non-GCB DLBCL, GCB DLBCL, all DLBCL, and MCL was 66.7%, 16.7%, 56.7%, and 85.7%, respectively (**Figure 2**)

Figure 2. ORR by histology

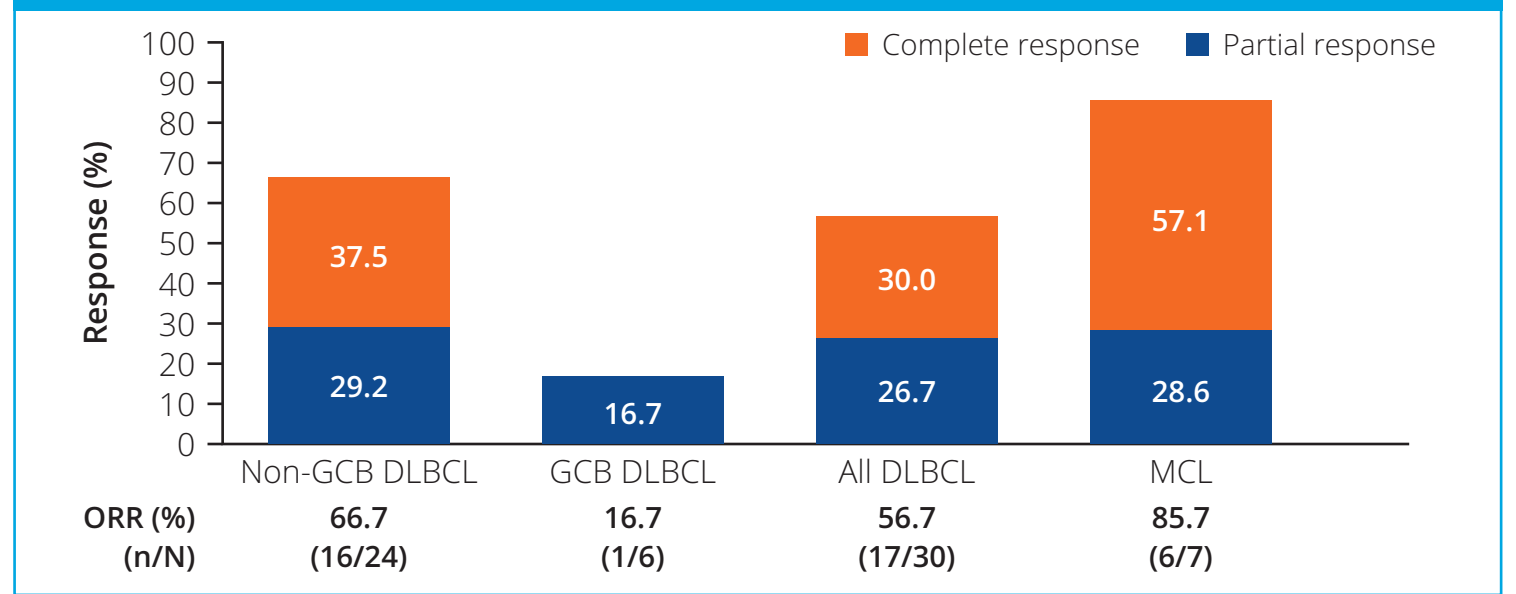

Complete response
Partial response
Response (%)
100
90
80
70
60
50
40
30
20
10
0
37.5
29.2
16.7
30.0
26.7
57.1
28.6
Non-GCB DLBCL
GCB DLBCL
All DLBCL
MCL
ORR (%)
(n/N)
66.7
(16/24)
16.7
(1/6)
56.7
(17/30)
85.7
(6/7)

DLBCL, diffuse large B-cell lymphoma; GCB, germinal center B-cell; MCL, mantle cell lymphoma; ORR, overall response rate.

- Median (interquartile range) duration of response was 5.55 months (2.07–not reached)
 - Median duration of response for patients with non-GCB DLBCL, GCB DLBCL, all DLBCL, and MCL was 4.65 months (1.92–not reached), not reached (not reached–not reached), 5.55 months (2.07–not reached), and not reached (2.17–not reached)
- Response is ongoing in 4/24 patients with non-GCB DLBCL, and 3/7 patients with MCL (**Figure 3**)

Figure 3. Swimmer plot of progression-free survival by NHL subtype

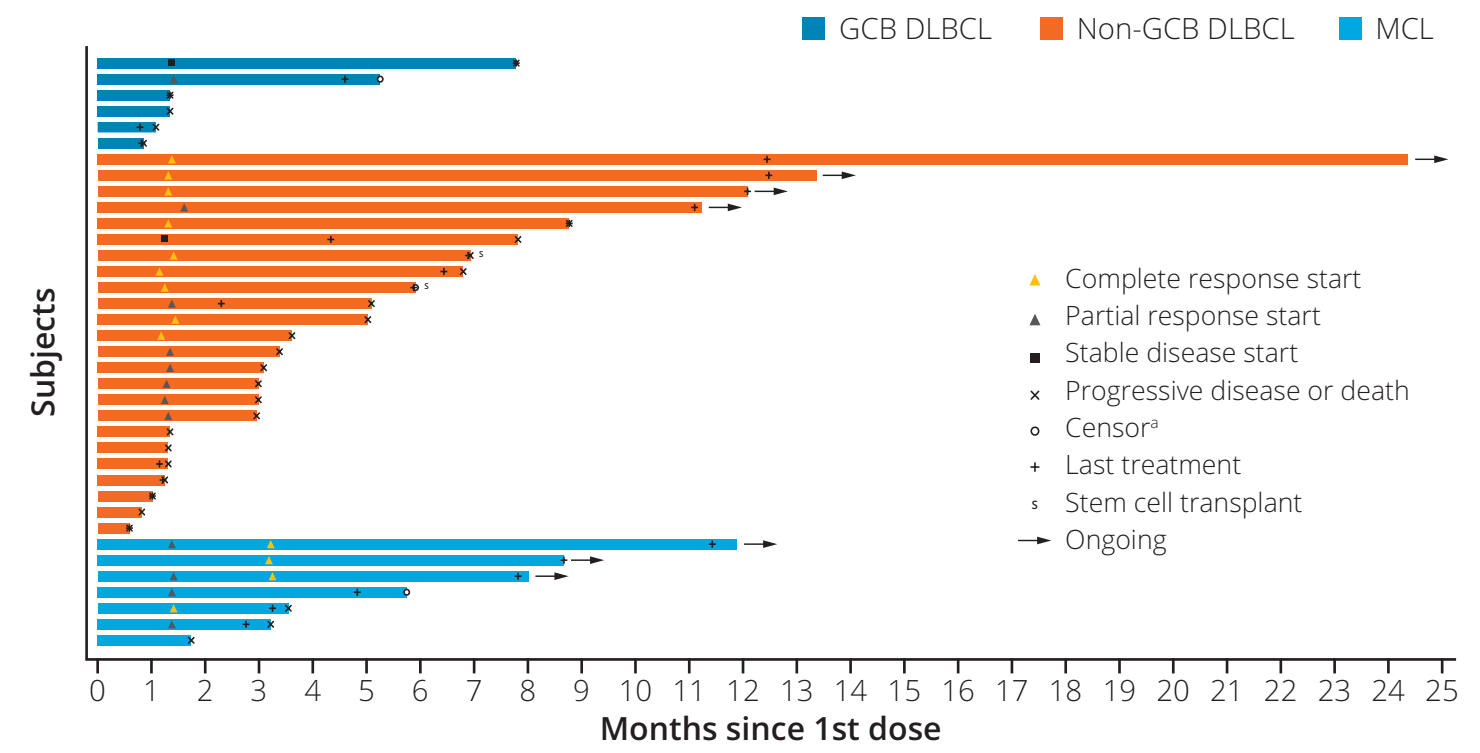

GCB DLBCL
Non-GCB DLBCL
MCL
Subjects
Complete response start
Partial response start
Stable disease start
Progressive disease or death
Censora
Last treatment
Stem cell transplant
Ongoing
0 1 2 3 4 5 6 7 8 9 10 11 12 13 14 15 16 17 18 19 20 21 22 23 24 25
Months since 1st dose

[a]Only for censored patients who discontinued the trial due to reasons other than progression or who went onto a different anticancer treatment.

DLBCL, diffuse large B-cell lymphoma; GCB, germinal center B-cell; MCL, mantle cell lymphoma; NHL, non-Hodgkin lymphoma.

Pharmacokinetics

- At data cut-off (August 20, 2020), cycle-related increase in PK exposure was observed and inter-patient exposure variability was moderate (**Table 2**)
- Sustained exposure and modest accumulation were seen by Cycle 2 (**Table 2, Figure 4**)

Table 2. Summary of PK parameters of conjugated and total antibody in Cycles 1 and 2

PK parameters	Conjugated Ab	Total Ab
Cycle 1: Lonca 60 µg/kg and ibrutinib 560 mg		
C_{max} (ng/mL)	659 (45.3) [26]	1280 (41.4) [26]
AUC_{inf} (ng·day/mL)	4364 (61.9) [8]	7449 (54.8) [9]
T_{half} (day)	6.31 (46.7) [8]	5.65 (38.2) [9]
CL (L/day)	0.893 (60.4) [8]	0.590 (50.1) [9]
V_{ss} (L)	5.52 (47.2) [8]	3.43 (43.6) [9]
Cycle 2: Lonca 60 µg/kg and ibrutinib 560 mg		
C_{max} (ng/mL)	761 (91.7) [21]	1461 (80.8) [21]
AUC_{tau} (ng·day/mL)	5582 (65.0) [15]	10,423 (60.1) [13]
T_{half} (day)	7.57 (43.0) [11]	7.79 (33.0) [6]
CL (L/day)	0.705 (63.3) [15]	0.451 (58.8) [13]
V_{ss} (L)	7.85 (64.0) [11]	6.01 (50.0) [6]
AI	1.21 (15.9) [11]	1.20 (10.4) [6]

Blood samples for PK analysis were drawn on Day 1 (pre-dose), Day 8 and Day 15 of treatment Cycles 1 and 2. Data shown as geometric mean (geometric % coefficient of variation) [n]. Ab, antibody; AI, accumulation index; AUC_{inf}, area under the curve vs time curve from 0 to infinity; AUC_{tau}, area under the curve from 0–21 days; CL, apparent clearance; C_{max}, maximum observed concentration; Lonca, Loncastuximab tesirine; PK, pharmacokinetics; T_{half}, apparent terminal half-life; V_{ss}, apparent steady-state volume of distribution.

Figure 4. Mean (±SE) concentration of conjugated antibody and total antibody vs time for Cycles 1 and 2

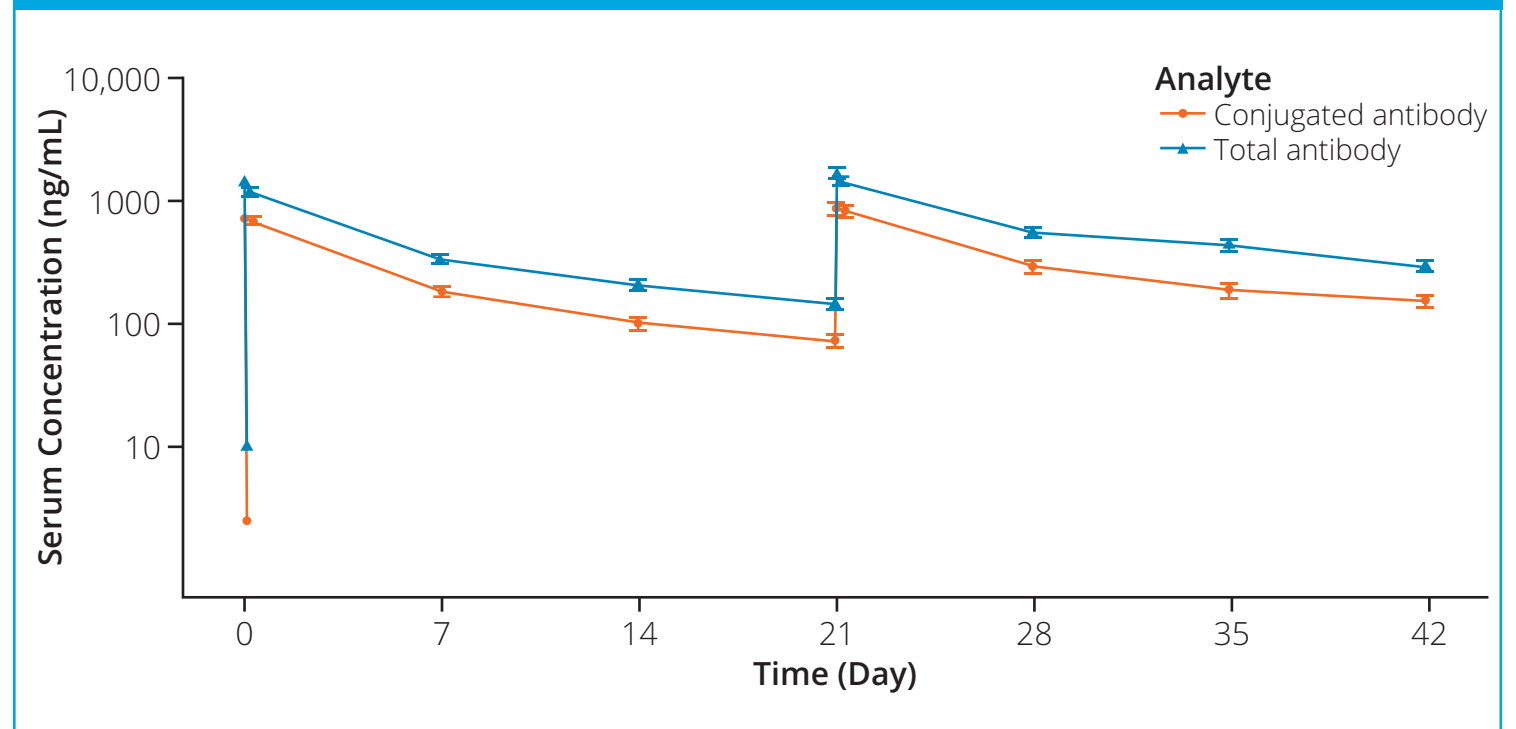

Analyte
Conjugated antibody
Total antibody
Serum Concentration (ng/mL)
10,000
1000
100
10
0
7
14
21
28
35
42
Time (Day)

Dose: Lonca 60 µg/kg + ibrutinib 560 mg. Concentrations below the LLOQ of 20 ng/mL for conjugated antibody and 5.06 for total antibody are imputed as 1/2 LLOQ.

LLOQ, lower limit of quantification; SE, standard error.

Funding

This study (NCT03684694) is sponsored by ADC Therapeutics SA

Acknowledgments

The authors would like to thank and acknowledge the participating patients and their families, and all study co-investigators and research coordinators. Ibrutinib provided by Pharmacyclics LLC, an AbbVie Company. The authors received editorial/writing support in the preparation of this poster provided by Sarah Meadows of Fishawack Communications Ltd, part of Fishawack Health, funded by ADC Therapeutics SA.

Disclosures

J Depaus reports consultancy/advisory roles for Takeda, Novartis, and Janssen. **N Wagner-Johnston** reports consultancy/advisory roles for ADC Therapeutics, Regeneron, CALIB-R, Verastem, Karyopharm, Sanofi, SeaGen, Epizyme, and Grunenthal. **PL Zinzani** reports consultancy/advisory roles for Verastem, MSD, Eusapharma, Sanofi, ADC Therapeutics (advisory board agreement), Celltrion, Gilead, Janssen-Cilag, BMS, Servier, Sandoz, TG Therapeutics, Takeda, Roche, and Kyowa Kirin. **TJ Phillips** reports consultancy/advisory roles for Celgene/BMS, Kite/Gilead, Seattle Genetics, AbbVie/Pharmacyclics, Incyte, and Genentech. **J Maly, S Ferrari, V Delwail, and M Tani** report no conflicts of interest. **E Bachy** reports employment/leadership position for Université Claude Bernard Lyon 1, Lyon, France, consultancy/advisory roles for Roche, and Gilead, and receipt of honoraria from Roche, Celgene, Amgen, Janssen, Gilead, Novartis, and Sanofi. **LJ Bryan** reports employment/leadership position for Augusta University, Augusta, GA. **M Janakiram** reports receipt of research funding from ADC Therapeutics, FATE Therapeutics, and Takeda Pharmaceuticals. **S Guibert** reports employment/leadership position for CHU Pontchaillou, Rennes, France and receipt of honoraria from Janssen, AbbVie, and Gilead. **V Dai** is an employee of ADC Therapeutics and SUNY Research Foundation and has stock and other ownership interests from ADC Therapeutics. **K Havenith, J Boni, X He, and A Ervin-Haynes** are employees of ADC Therapeutics and hold stock and other ownership interests with the company. **C Carlo-Stella** reports consultancy/advisory roles for Sanofi, ADC Therapeutics, Roche, Karyopharm Therapeutics, Celgene/Bristol-Myers Squibb, and Incyte, receipt of honoraria from Bristol-Myers Squibb, Janssen Oncology, and AstraZeneca, receipt of research funding from ADC Therapeutics, Sanofi, and Roche, and receipt of travel grants from Roche, Janssen, Takeda, and ADC Therapeutics.

Contact information

Dr Julien Depaus: Julien.depaus@chuuclnamur.be

References

1. Levin A, Shah, NN. *Am J Hematol* 2019;94(S1):S18–S23
2. Maddocks K, *Blood* 2018;132(16):1647–1656
3. Depaus J, et al. *ASH 2020*; abstract 2099
4. Depaus J, et al. *EHA 2020*; abstract EP1284
5. Cheson DB, et al. *J Clin Oncol* 2014;32(27):3059–3068

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster

CONCLUSIONS

- Lonca 60 µg/kg plus ibrutinib 560 mg had encouraging antitumor activity in R/R DLBCL or R/R MCL, with an investigator-assessed ORR of 62.2%
 - ORR for non-GCB DLBCL was 66.7%, GCB DLBCL was 16.7%, and MCL was 85.7%
- Toxicity was manageable at the MTD, with safety data comparable to that previously reported[3,4]
- PK profiles exhibit sustained exposure and modest accumulation with the Q3W regimen by Cycle 2

Presented at the International Conference on Malignant Lymphoma (ICML) Virtual Congress, June 18–22, 2021