Pre-clinical characterization of the mechanism of action of a CD25-targeted pyrrolobenzodiazepine dimer-based antibody-drug conjugate targeting regulatory T cells in solid cancers

Francesca Zammarchi, Simon Chivers, Patrick H. van Berkel

ADC Therapeutics SA, London, United Kingdom

ADC THERAPEUTICS

P697
abstract poster number

Introduction

1. Regulatory T cells (T_{regs}) play an important role in the establishment and progression of tumors and are considered a major obstacle to tumor eradication by immunotherapies [1]. Moreover, the intra-tumoral balance between Tregs and effector T cells (T_{effs}) appears to influence the outcome of immunotherapies [2], and poor prognosis in solid tumors is often associated with high tumor infiltration by T_{regs} and a low ratio of T_{effs}/T_{regs} [3].
2. Sur301 is an antibody-drug conjugate (ADC) composed of PC61, a rat monoclonal antibody directed against mouse CD25, stochastically conjugated to tesirine, a protease-cleavable, pyrrolobenzodiazepine (PBD) dimer-based payload, with a drug-to-antibody ratio of 2 [4] (Figure 1).

Figure 1

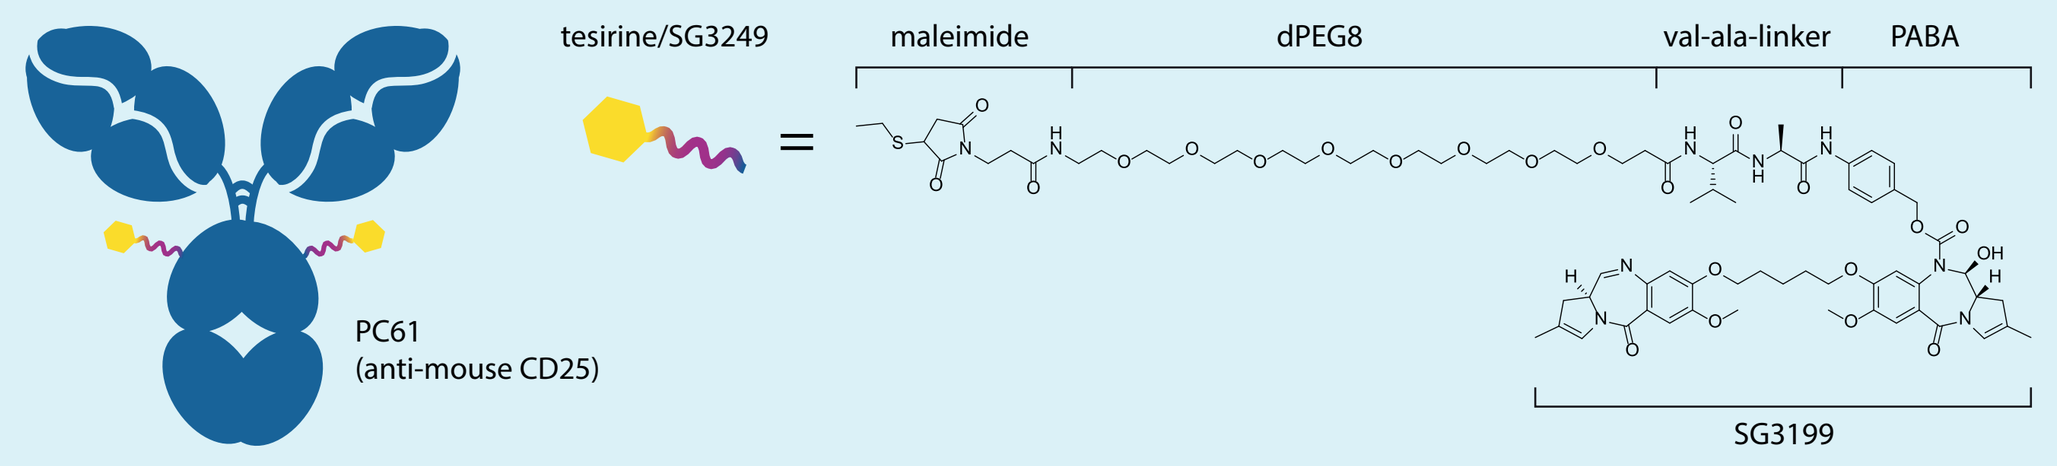

tesirine/SG3249
maleimide
dPEG8
val-ala-linker
PABA
PC61
(anti-mouse CD25)
SG3199

3. We have previously shown that sur301 has potent and durable anti-tumor activity *in vivo* against CD25-negative immunogenic solid tumors with infiltrating CD25-positive T_{regs} and, when used at a sub-optimal dose, its activity is further enhanced in combination with PD-1 blockade [4]. Sur301 anti-tumor activity, either alone or combined with an anti-PD-1 antibody, was significantly reduced in the absence of $CD8^+$ T cells, and when tumor-free survivors were re-challenged, they did not develop new tumors indicating that sur301 was able to induce tumor-specific protective immunity [4].

Aim of the study

The purpose of this study was to characterize the mechanism of action of sur301 by performing T-cell immunophenotyping analysis in MC38 tumor-bearing or non-tumor-bearing mice following a single dose of sur301, alone or in combination (MC38 tumor-bearing only) with an anti-PD-1 antibody.

Material & methods

- *In vivo* efficacy study: sur301 was administered intraperitoneally (i.p.) as single dose to female C57BL/6 mice containing established MC38 tumors (group mean tumor volumes 89 mm³) on Day 1. The other compounds used i.p were the isotype control ADC and the anti-PD-1 antibody (clone RMP1-14).
- For the T-cell dynamic study, female C57BL/6 mice were injected intravenously with vehicle, sur301 (0.5 mg/kg) or isotype control ADC (0.5 mg/kg) on Day 0. Spleen, lymph node and thymus were collected at 4 hours post dose, and on Days 6, 13, and 20 post dose for T-cell immune profiling.
- The Coefficient of Drug Interaction (CDI) was assessed for sub-additive, additive, or supra-additive (synergism) properties on the last day at least 50% of the animals remained on study, as previously described [5].

Results

Figure 2A: *In vivo* anti-tumor activity in the MC38 syngeneic model

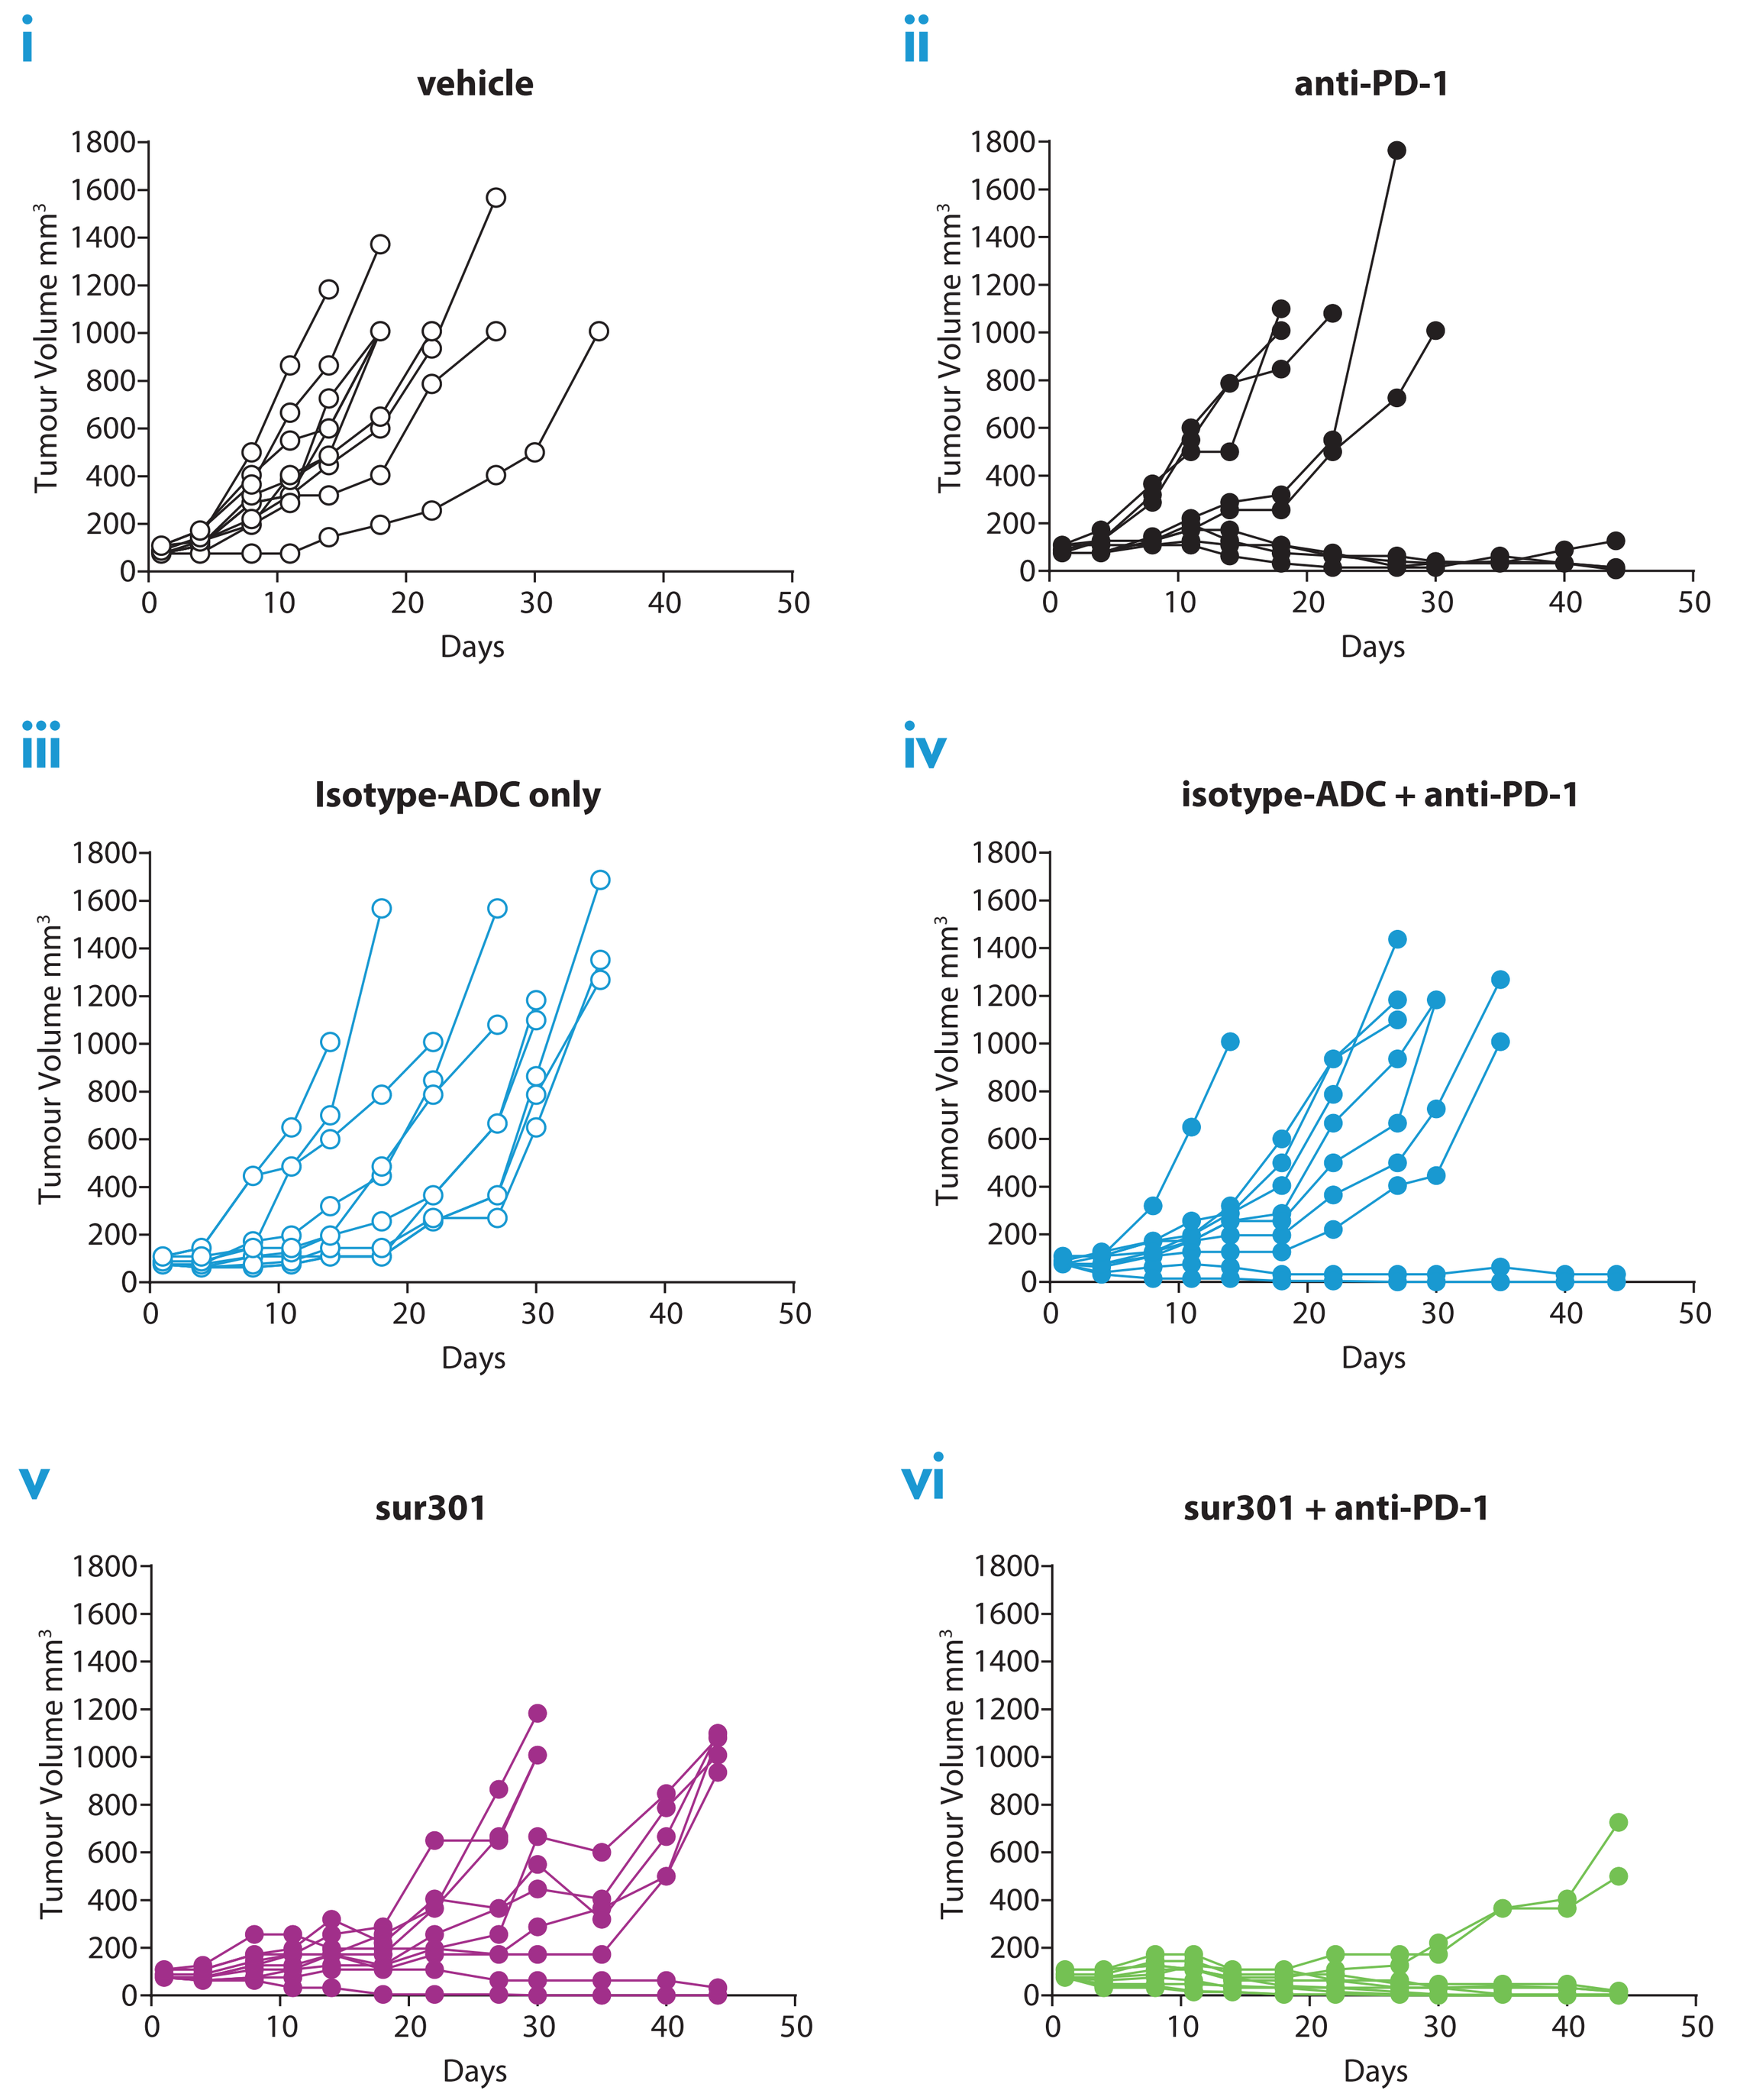

i
vehicle
ii
anti-PD-1
iii
Isotype-ADC only
iv
isotype-ADC + anti-PD-1
v
sur301
vi
sur301 + anti-PD-1
Tumour Volume mm³
Days

Response summary

	PD	CR	TFS
vehicle	0	0	0
anti-PD-1	2	0	0
isotype-ADC	0	0	0
isotype-AD + anti-PD-1	1	1	1
sur301	0	1	1
sur301 + anti-PD-1	2	5	5

Coefficient of drug interaction

		DAY 18
CDI	- anti-PD-1 - sur301 - sur301 + anti-PD-1	0.672 (synergism)

Sur301 or an isotype control-ADC were administered i.p. at a group mean tumor volume of 89 mm³ as a single dose at 0.5 mg/kg either alone (on Day 1) or in combination with anti-PD-1 antibody (5 mg/kg, on Days 2, 5 and 8). Each graph represents tumor volumes over time for each individual mouse (10 mice/group).

PR, partial responders; CR, complete responders; TFS, tumor-free survivors; CDI, coefficient of drug interaction.

Figure 2B: T-cells Immunophenotyping from MC38 efficacy study

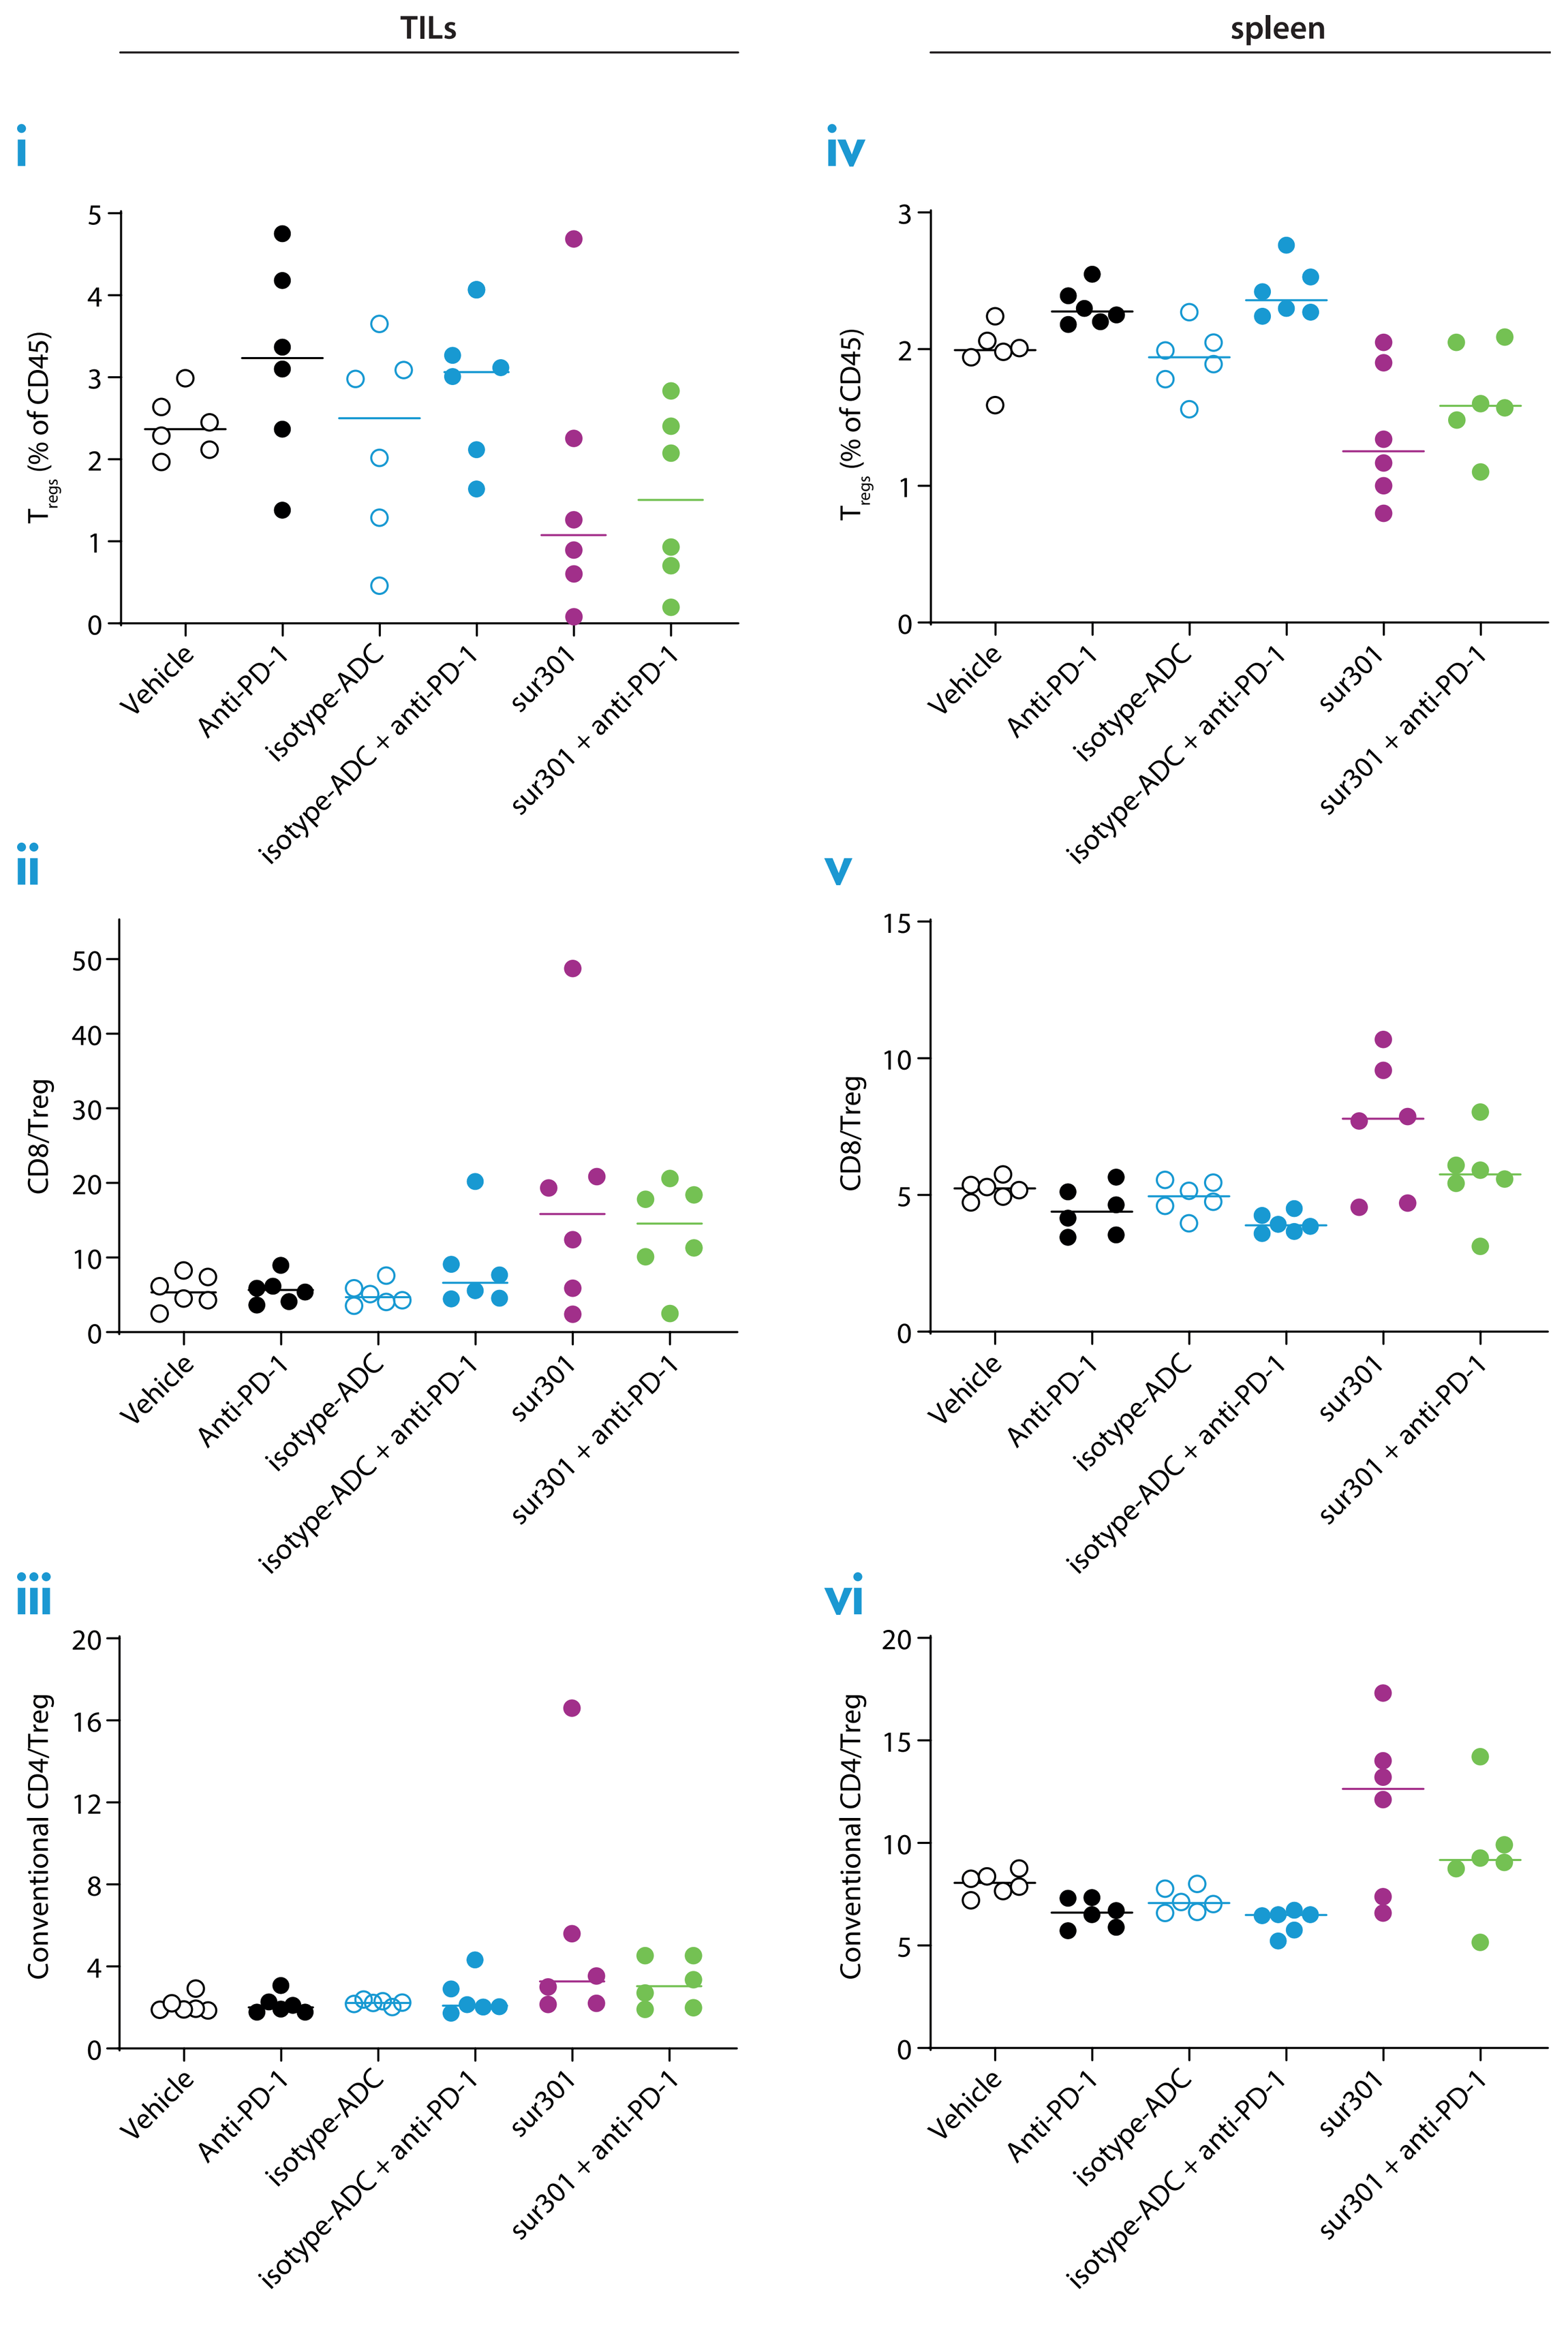

TILs
spleen
i
ii
iii
iv
v
vi
Tregs (% of CD45)
CD8/Treg
Conventional CD4/Treg
Vehicle
Anti-PD-1
isotype-ADC
isotype-ADC + anti-PD-1
sur301
sur301 + anti-PD-1

Quantification of T_{reg}, $CD8^+/T_{reg}$ and conventional $CD4^+$ T cells/T_{reg} ratios from MC38-bearing mice following i.p. treatment with sur301 (0.5 mg/kg) either alone on Day 1 or in combination with anti-PD-1 antibody (5 mg/kg) on Days 2, 5 and 8. Tumors and spleens were collected on Day 9 for T-cell immunophenotyping. (**i–iii**) Levels of intratumoral T_{regs} and T_{eff}/T_{reg} ratios. (**iv–vi**) Levels of T_{regs} and T_{eff}/T_{reg} ratios in spleen. Each panel shows individual values for each of the 6 mice/group analysed. Horizontal bar represents median value.

TIL, tumor-infiltrating lymphocytes; T_{eff}: effector T cell; T_{reg}: regulatory T cell.

Figure 3: T cells dynamic study in non-tumor-bearing mice

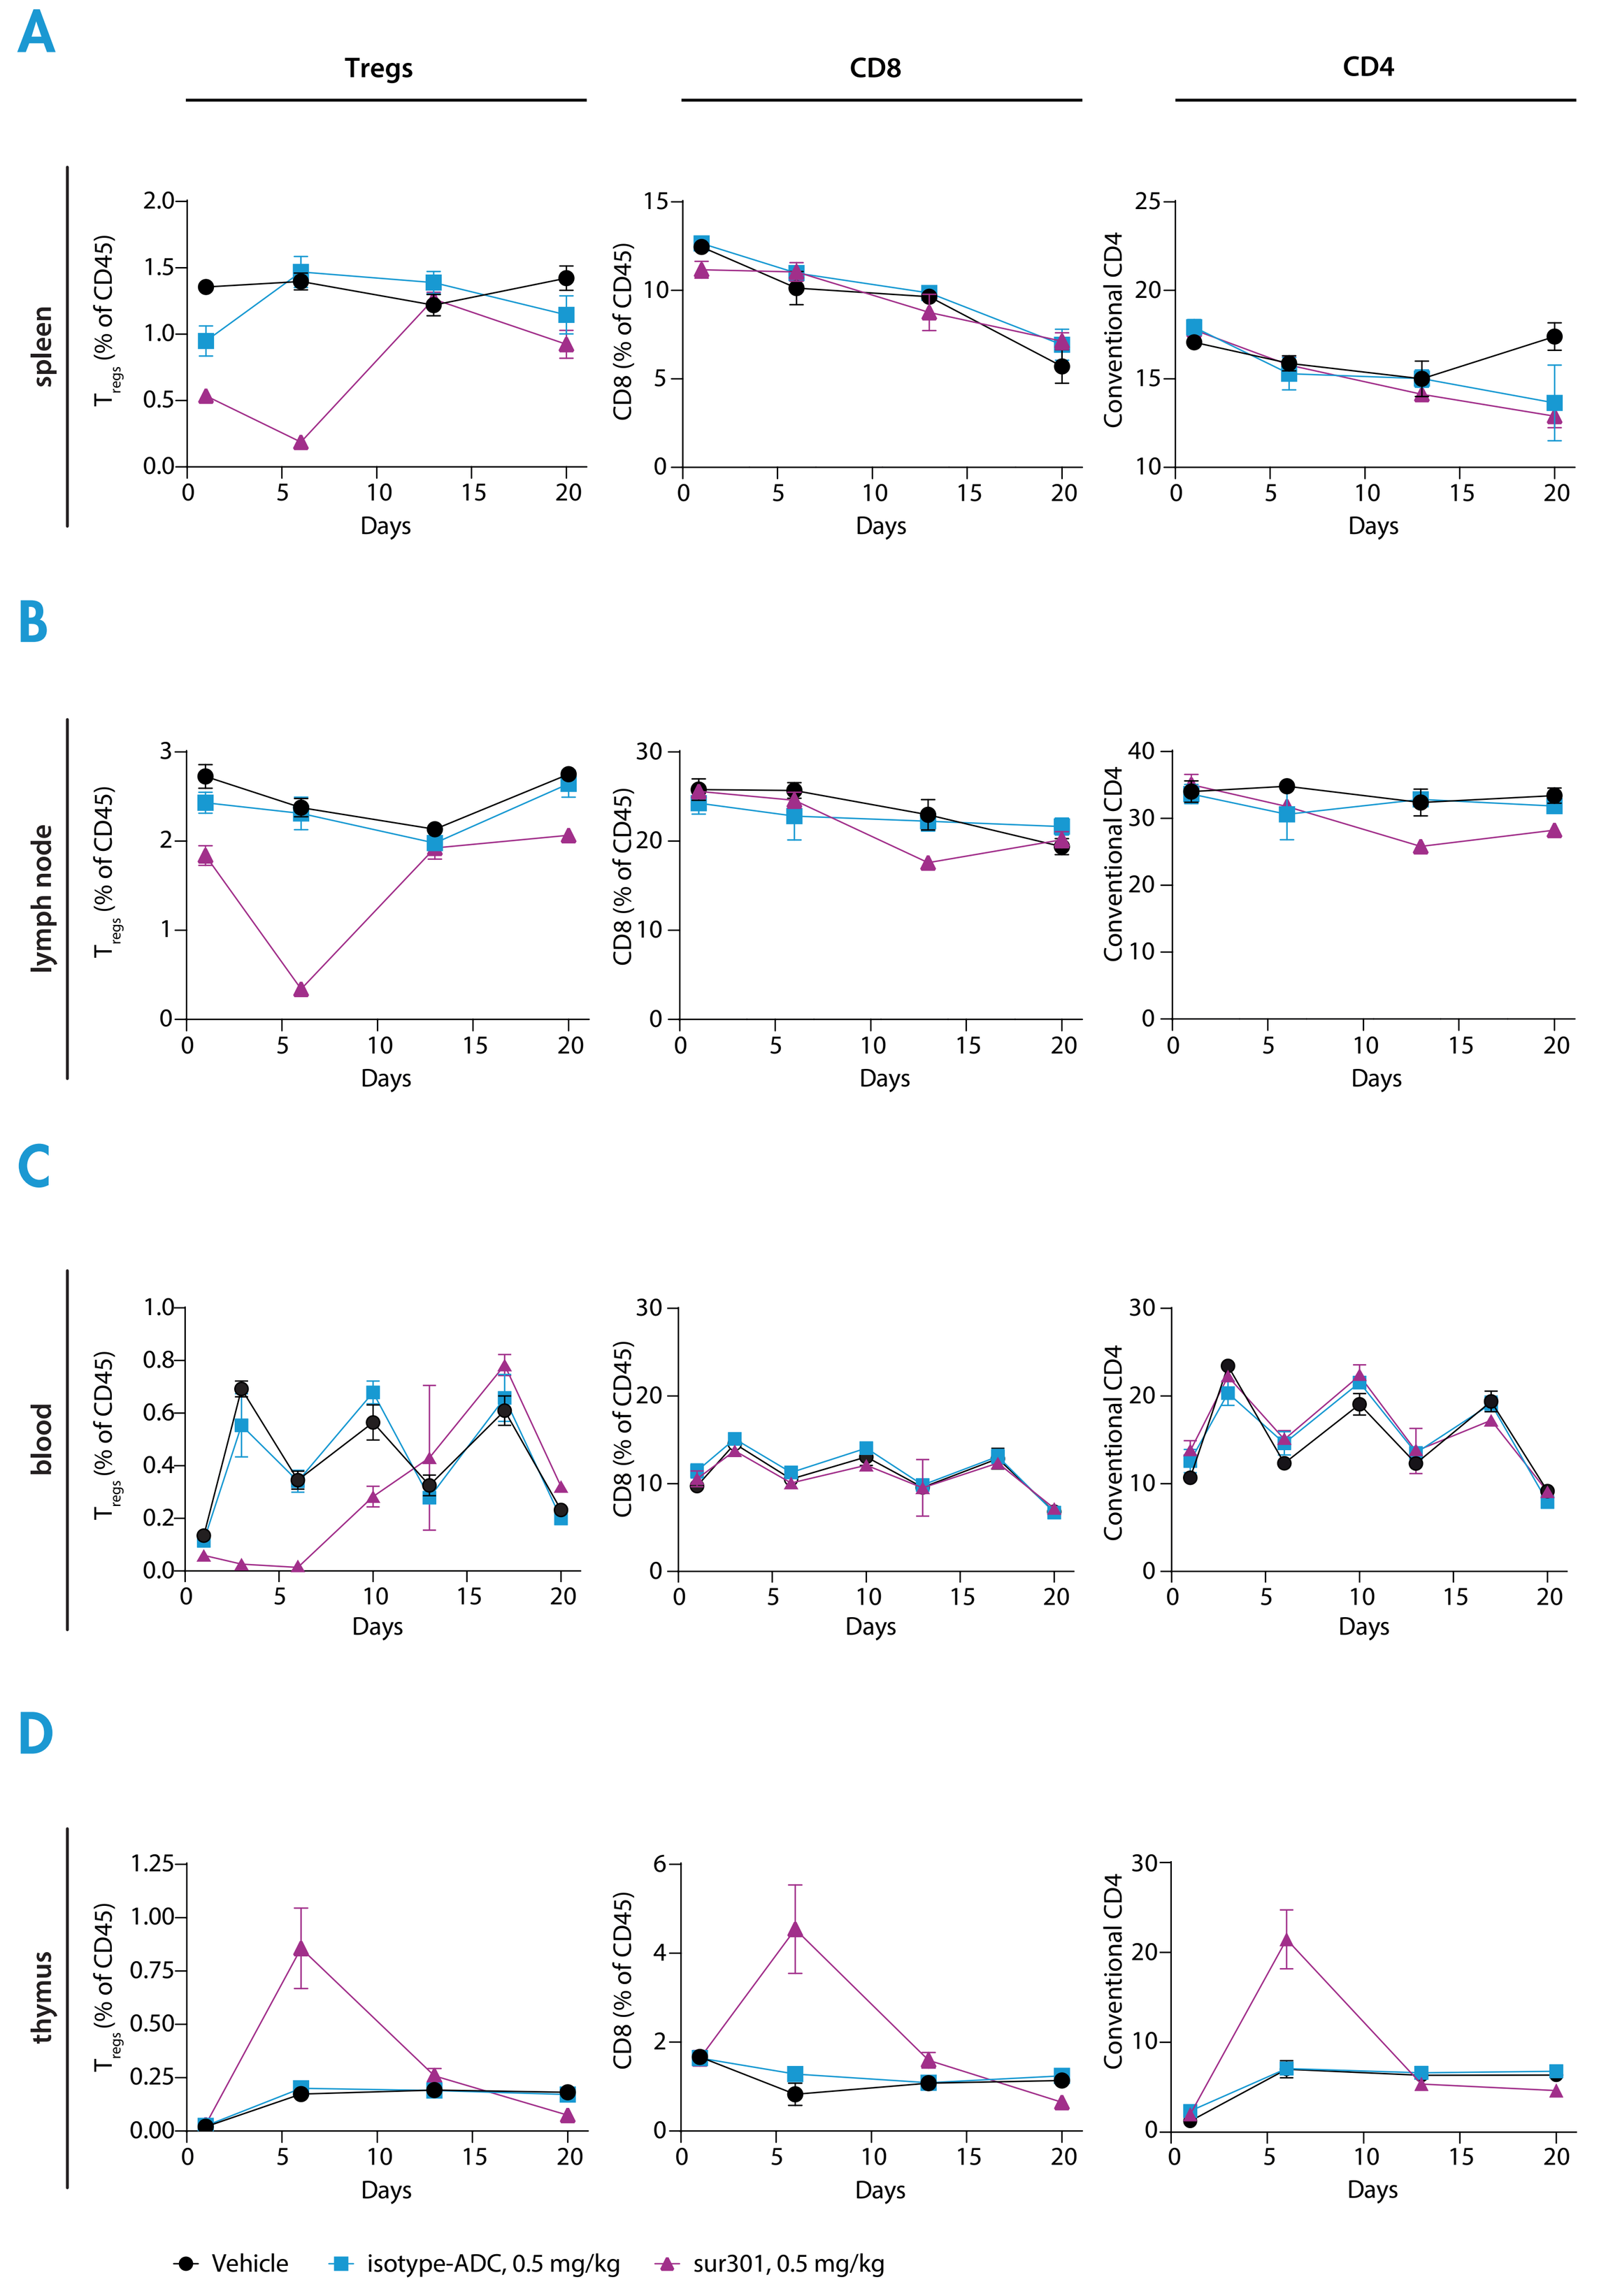

A
B
C
D
Tregs
CD8
CD4
spleen
lymph node
blood
thymus
Tregs (% of CD45)
CD8 (% of CD45)
Conventional CD4
Days
Vehicle
isotype-ADC, 0.5 mg/kg
sur301, 0.5 mg/kg

Levels of T_{regs}, CD8 and conventional CD4 levels in (**A**), spleen, (**B**), lymph nodes, (**C**), thymus and blood (**D**) are presented as % of CD45 cells ± SEM over time (6 mice for each time point). T-cell immune profiling analysis was carried out at 4 hours post dose, and on Days 6, 13, and 20 post dose.

T_{effs}: effector T cells; T_{regs}: regulatory T cells.

Conclusions

1. *In vivo*, a single dose of sur301 induced potent and durable anti-tumor activity against CD25-negative established MC38 solid tumors, and its activity was further increased when combined with an anti-PD-1 antibody (synergistic interaction).
2. T-cell immunophenotyping analysis from MC38 tumor-bearing mice showed that sur301 mediates specific T_{regs} depletion without affecting T_{effs} cells, improving the CD8/T_{regs} ratio.
3. In non-tumor-bearing immunocompetent mice, a single dose of sur301 transiently and specifically depleted T_{regs} in spleen, lymph nodes and blood while neither $CD8^+$ nor conventional $CD4^+$ were affected.
4. Interestingly, the strong T_{regs} depletion observed in spleen, lymph node and blood was accompanied by a temporary but significant elevation of thymic T_{regs}, possibly a homeostatic mechanism in response to the transient but significant T_{regs} depletion.
5. Together with our previous data [4], these results suggest that sur301's mode of action is at least in part mediated by T_{regs} depletion while T_{effs} are not affected.
6. Translation of these pre-clinical data in the clinic is currently being investigated in a phase I trial evaluating the efficacy of camidanlumab tesirine (ADCT-301), a PBD-based ADC targeting human CD25, in patients with selected advanced solid tumors (NCT03621982).

Acknowledgements

In vivo studies: Charles River Discovery Research Services (USA).

References

1. *Sasidharan Nair, V. and E. Elkord, Immune checkpoint inhibitors in cancer therapy: a focus on T-regulatory cells. Immunol Cell Biol, 2018. 96(1): p. 21-33.*
2. *Menetrier-Caux, C., et al., Targeting regulatory T cells. Target Oncol, 2012. 7(1): p. 15-28.*
3. *Arce Vargas, F., et al., Fc-Optimized Anti-CD25 Depletes Tumor-Infiltrating Regulatory T Cells and Synergizes with PD-1 Blockade to Eradicate Established Tumors. Immunity, 2017. 46(4): p. 577-586.*
4. *Zammarchi, F., et al., A CD25-targeted pyrrolobenzodiazepine dimer-based antibody-drug conjugate shows potent anti-tumor activity in pre-clinical models of solid tumors either alone or in combination with a PD-1 inhibitor. Journal for ImmunoTherapy of Cancer, 2018. 6(Supplement 1).*
5. *Wu, J., L. Tracey, and A.M. Davidoff, Assessing interactions for fixed-dose drug combinations in tumor xenograft studies. J Biopharm Stat, 2012. 22(3): p. 535-43.*

SITC 34th Annual Meeting, November 7-10 2019, National Harbor, MD, USA