Phase 1b Open-Label Study of Loncastuximab Tesirine in Combination With Other Anticancer Agents in Patients With Relapsed or Refractory B-cell Non-Hodgkin Lymphoma (LOTIS-7)

Brian T. Hess,[1*] Melhem M. Solh,[2] Mitul Gandhi,[3] Ying Wang,[4] Yajuan Qin,[4] Pier Luigi Zinzani,[5] Graham P. Collins[6]

[1]Division of Hematology and Medical Oncology, Department of Medicine, Medical University of South Carolina, Charleston, SC, USA; [2]Blood and Marrow Transplant Group of Georgia, Atlanta, GA, USA; [3]Virginia Cancer Specialists, Fairfax, VA, USA; [4]ADC Therapeutics America, Inc., Murray Hill, NJ, USA; [5]Institute of Hematology "Seràgnoli," University of Bologna, Bologna, Italy; [6]Oxford University Hospitals NHS Trust, Oxford, UK

ABCL-134

Product

Loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]) is an ADC comprising a humanized **anti-CD19** antibody conjugated to a **PBD dimer cytotoxin** that is indicated for **R/R DLBCL**

Patients

Adults with **R/R B-NHL** with ≥2 prior systemic treatments (part 1)

Trial

LOTIS-7 (NCT04970901) is a **phase 1b** study evaluating the safety/tolerability and antitumor activity of **Lonca in combination with other anticancer agents**

Current Status

Dose escalation for the **Lonca + polatuzumab** arm is complete

Enrollment for the **Lonca + glofitamab** and **Lonca + mosunetuzumab** arms started enrolling in **July 2023**

ADC, antibody-drug conjugate; B-NHL, B-cell non-Hodgkin lymphoma; DLBCL, diffuse large B-cell lymphoma; PBD, pyrrolobenzodiazepine; R/R, relapsed/refractory.

KEY MESSAGE

- The safety, tolerability, and anticancer activity of loncastuximab tesirine (an FDA- and EMA-approved, CD19-directed antibody-drug conjugate) in combination with other anticancer agents are being assessed in patients with relapsed/refractory B-cell non-Hodgkin lymphoma in a phase 1b, 2-part, open-label study (NCT04970901)

INTRODUCTION

- Loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]), an antibody-drug conjugate comprising a humanized anti-CD19 antibody conjugated to a pyrrolobenzodiazepine dimer cytotoxin, received accelerated approval by the United States Food and Drug Administration and has received conditional marketing authorization by the European Commission and the United Kingdom's Medicines and Healthcare products Regulatory Agency to treat adult patients with R/R DLBCL after ≥2 lines of systemic therapy[1-3]
 - A phase 2 trial of Lonca monotherapy in patients with R/R DLBCL showed that an intravenous (IV) infusion over 30 minutes on day (D) 1 of each 3-week cycle produced durable responses with manageable toxicity using a dose of 150 µg/kg for 2 cycles and then 75 µg/kg for subsequent cycles[4]
- Combining agents with different mechanisms of action may enhance treatment efficacy in patients with R/R B-cell non-Hodgkin lymphoma (B-NHL)
 - In preclinical WSU-DLCL2 and Ramos xenograft models, Lonca in combination with polatuzumab vedotin (Pola) showed improved antitumor activity with better response rates compared with either monotherapy alone[5]
 - In addition, combining CD20×CD3 T-cell engaging antibodies (eg, glofitamab[6] or mosunetuzumab[7]) and Lonca is expected to increase antitumor activity

OBJECTIVE

- To evaluate the safety, tolerability, and anticancer activity of Lonca in combination with other anticancer agents in patients with R/R B-NHL (LOTIS-7; NCT04970901)

METHODS

Study Design

- This is a phase 1b, 2-part (including dose-escalation [part 1] and dose-expansion [part 2]), open-label, multicenter, multiarm study (NCT04970901) that will enroll ~200 pts with R/R B-NHL (part 1: 60 patients; part 2: 140 patients) (**Figure 1**)[8]
 - Patients will be enrolled if they have failed, or been intolerant to, any approved therapy and had received at least 2 systemic treatment regimens in part 1
- The study will include a screening period (up to 28 days), a treatment period (every 3 weeks [Q3W] with cycles for up to 1 year or until disease progression, unacceptable toxicity, or other discontinuation criteria), and a follow-up period (every 12 weeks for up to 2 years after treatment completion or discontinuation)
- The study period is defined as the date of obtaining written informed consent to the completion of the follow-up period, withdrawal of consent, loss to follow-up, or death, whichever occurs first

Figure 1. Study design

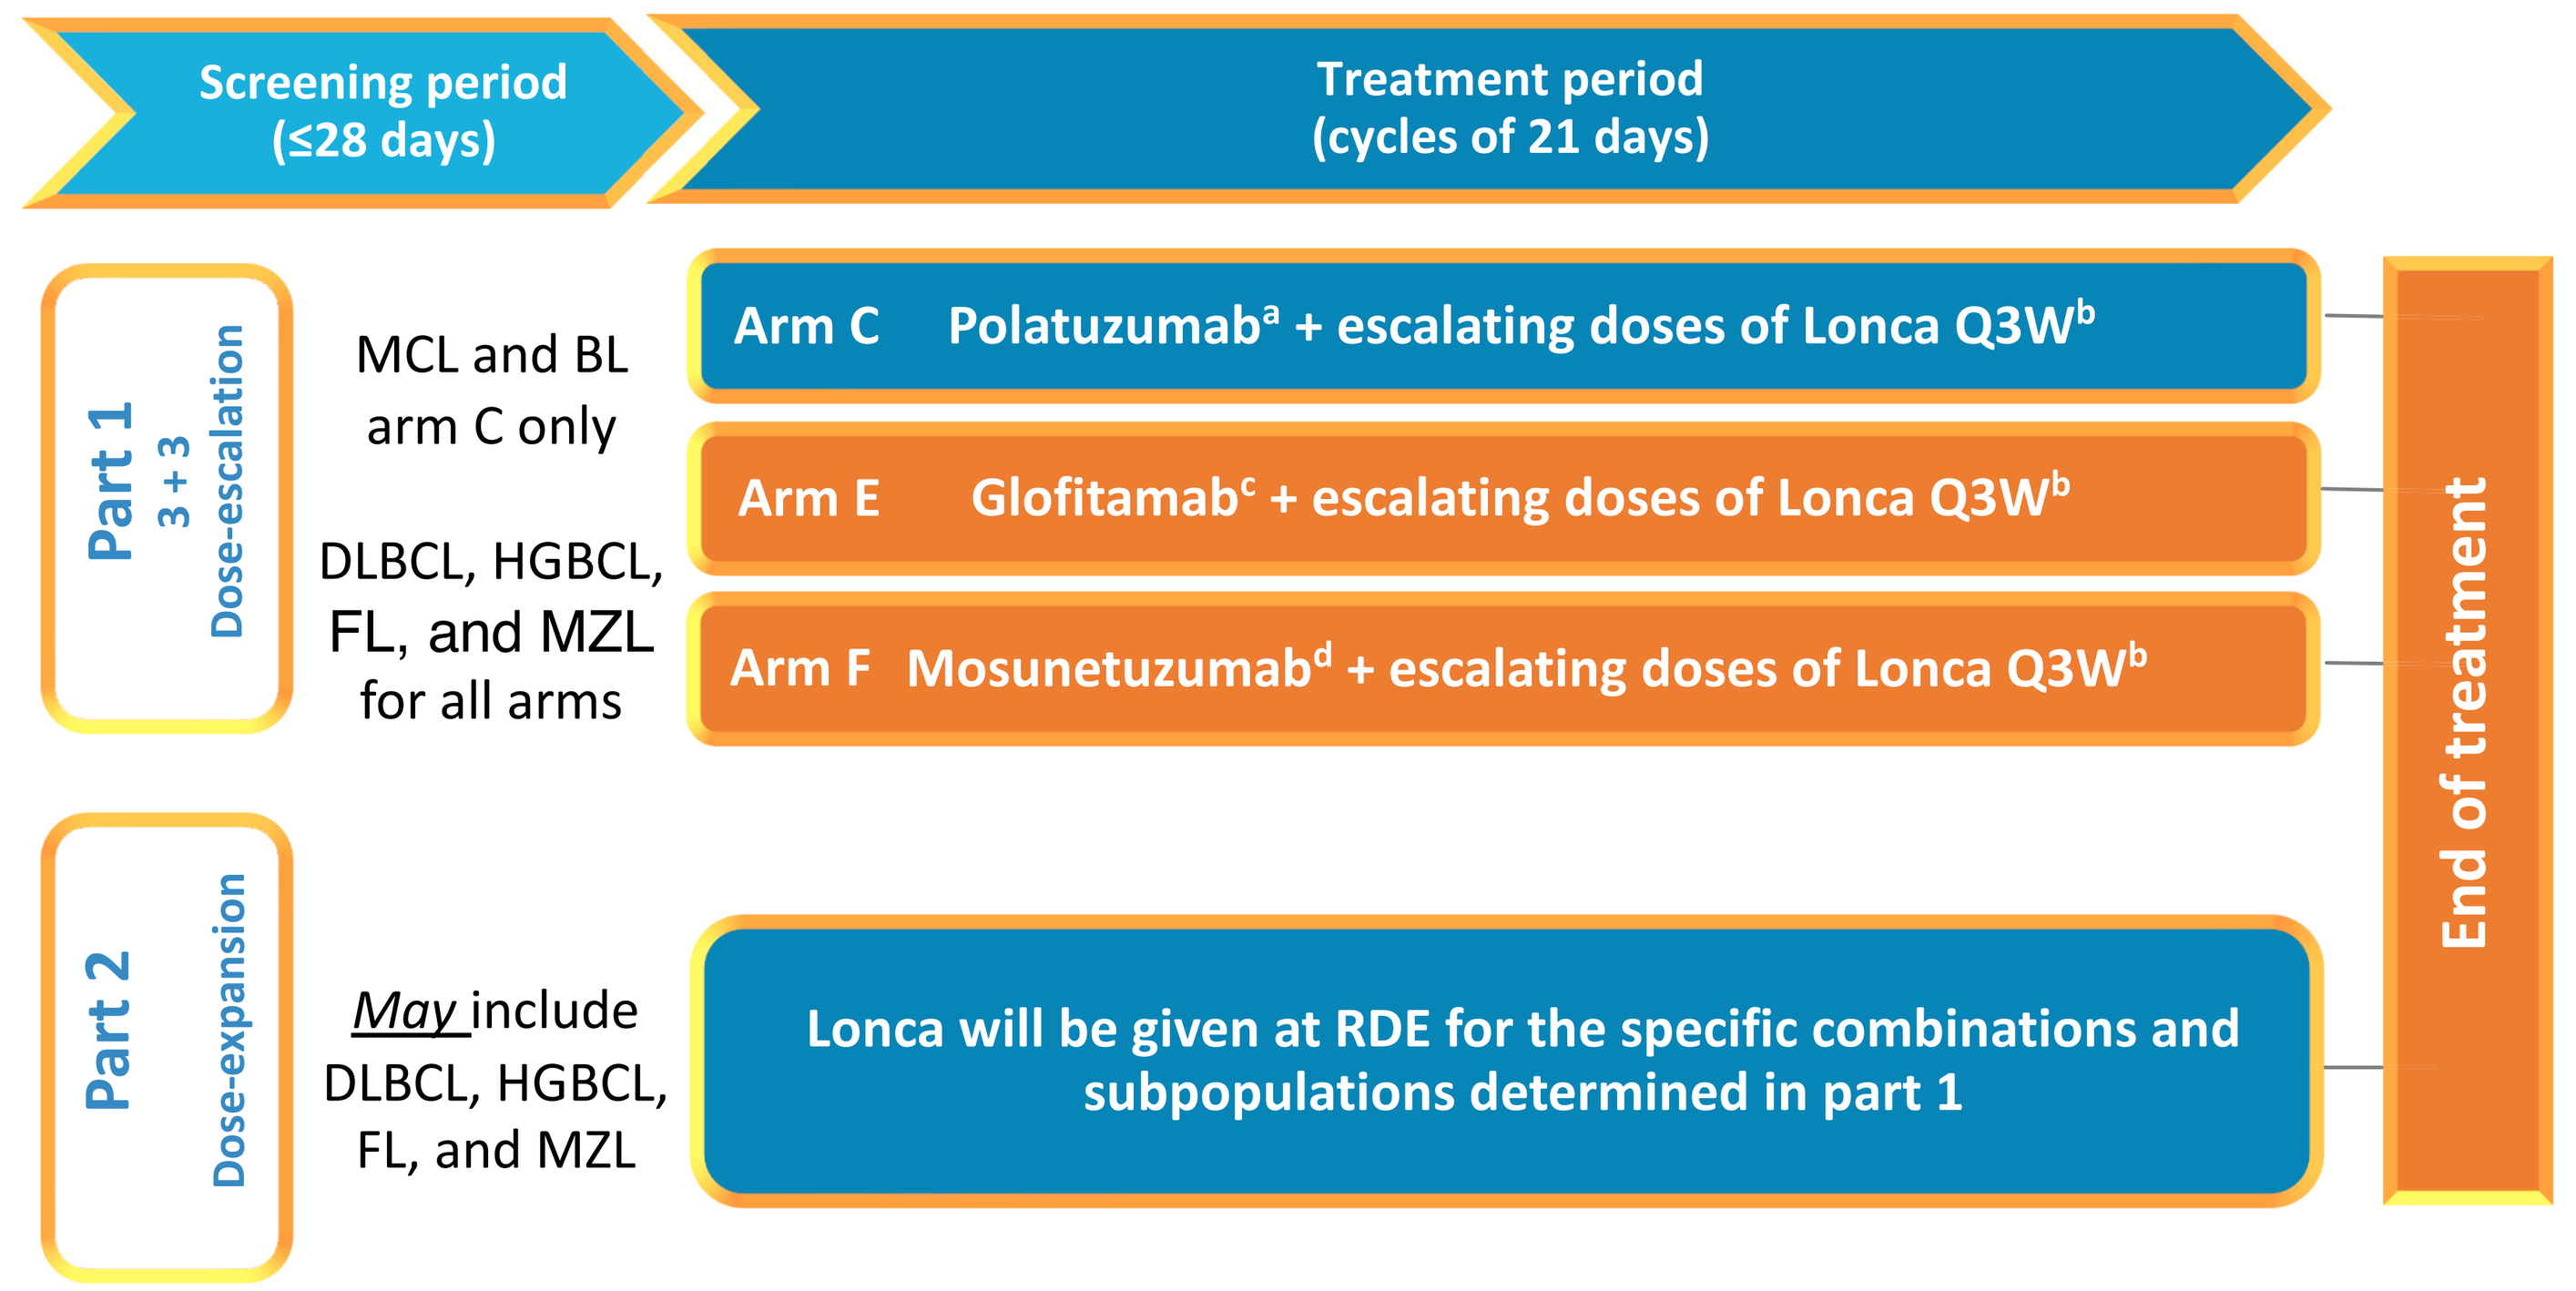
Screening period
(≤28 days)
Treatment period
(cycles of 21 days)
Part 1
3 + 3
Dose-escalation
MCL and BL
arm C only
DLBCL, HGBCL,
FL, and MZL
for all arms
Arm C Polatuzumab[a] + escalating doses of Lonca Q3W[b]
Arm E Glofitamab[c] + escalating doses of Lonca Q3W[b]
Arm F Mosunetuzumab[d] + escalating doses of Lonca Q3W[b]
Part 2
Dose-expansion
May include
DLBCL, HGBCL,
FL, and MZL
Lonca will be given at RDE for the specific combinations and subpopulations determined in part 1
End of treatment

Participants may continue treatment for up to 1 year or until disease progression, unacceptable toxicity, or other discontinuation criteria, whichever occurs first.
The follow-up period is for ≤2 years from the end of treatment.

BL, Burkitt lymphoma; DLBCL, diffuse large B-cell lymphoma; FL, follicular lymphoma; HGBCL, high-grade B-cell lymphoma; IV, intravenous; Lonca, loncastuximab tesirine; MCL, mantle cell lymphoma; MTD, maximum tolerated dose; MZL, marginal zone lymphoma; Q3W, every 3 weeks; RDE, recommended dose for expansion; R/R, relapsed/refractory.
[a]IV polatuzumab vedotin 1.8 mg/kg on day 1 of each 21-day cycle.
[b]Escalating doses of IV Lonca on day 1 of each cycle (C) at doses of 90, 120, and 150 µg/kg. Lonca will be administered on D1 of each cycle, 1 hour before combination drugs, with the exception of arm E, which will be administered on C1D2. If the starting dose of loncastuximab tesirine is 120 µg/kg or higher, the dose will be reduced to 75 µg/kg from C3.
[c]Obinutuzumab pretreatment on C1D1; IV glofitamab 2.5 mg on C1D8, 10 mg on C1D15, and then 30 mg for cycles 2-12 on D1.
[d]Subcutaneous mosunetuzumab 5 mg on C1D1 and then 45 mg for C1D8, C1D15, and cycles 2-8 on D1.

- For part 1 (**Figure 2**),
 - Patients in arm C will receive escalating doses of Lonca + Pola
 - Patients in arm E will receive escalating doses of Lonca + glofitamab after an initial pretreatment with obinutuzumab (1000 mg)
 - Patients in arm F will receive escalating doses of Lonca + mosunetuzumab

Figure 2. Part 1 dosing

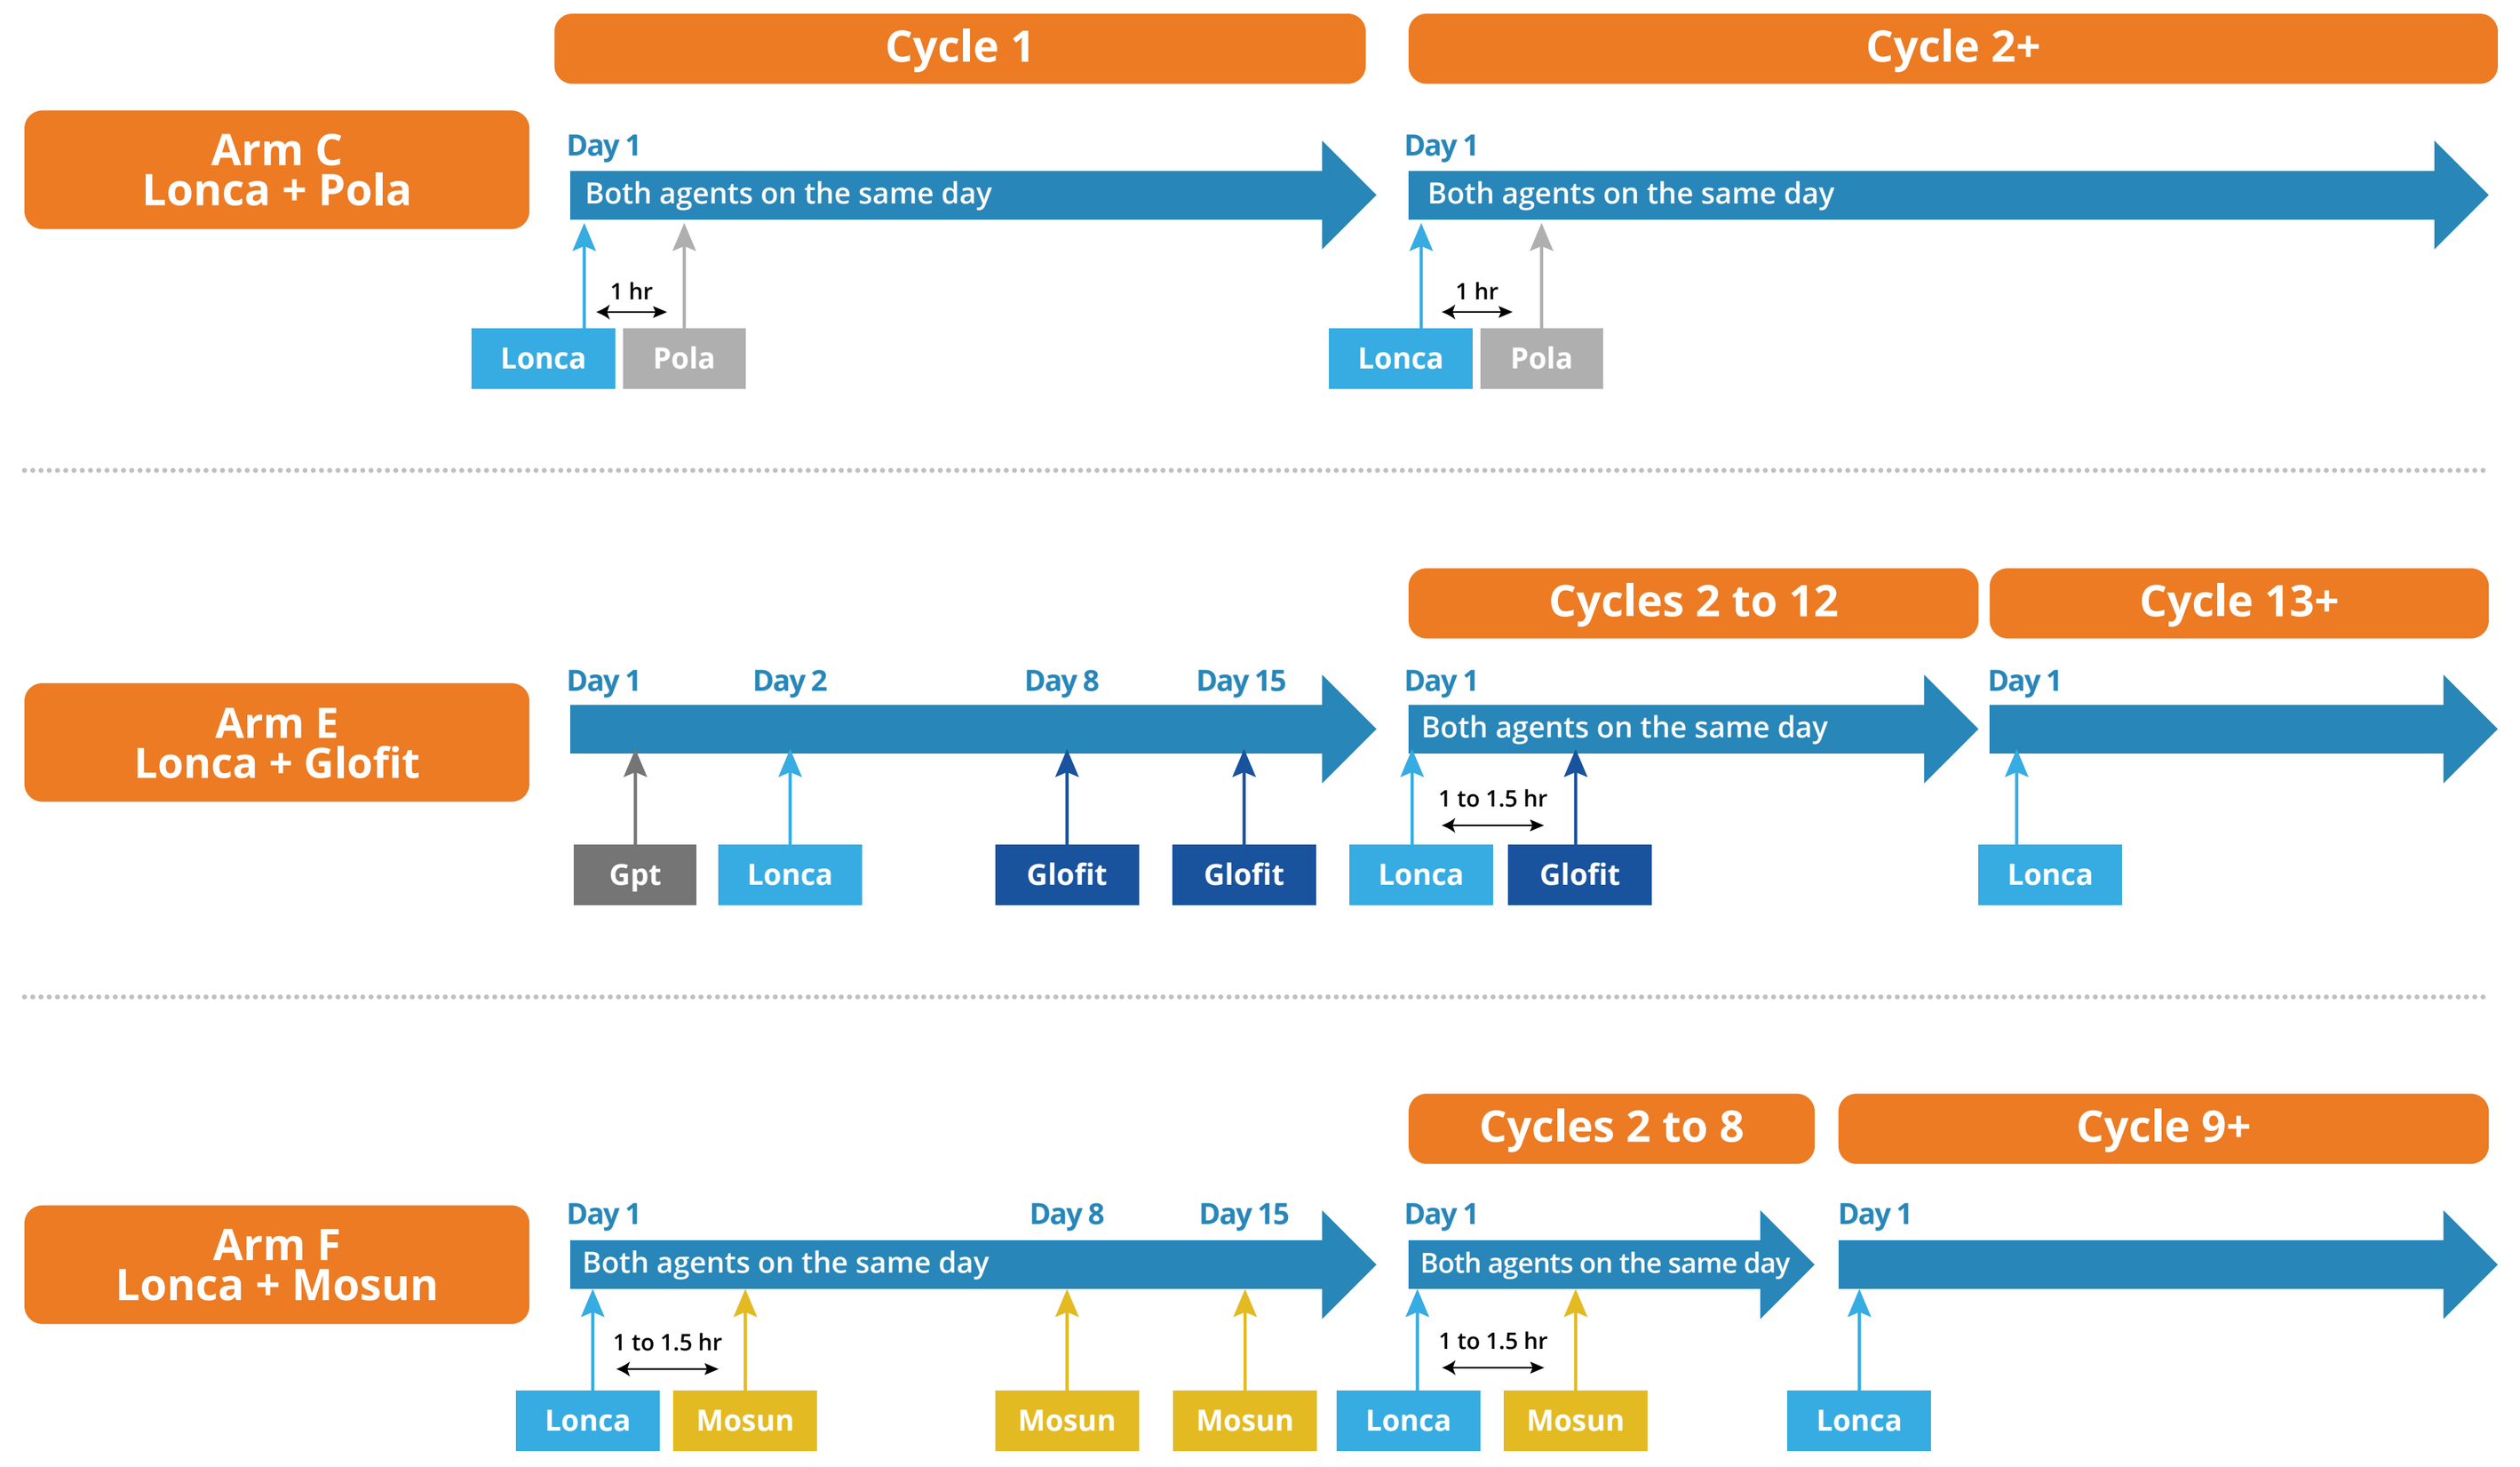
Cycle 1
Cycle 2+
Arm C
Lonca + Pola
Day 1
Both agents on the same day
1 hr
Lonca
Pola
Day 1
Both agents on the same day
1 hr
Lonca
Pola
Cycles 2 to 12
Cycle 13+
Arm E
Lonca + Glofit
Day 1
Day 2
Day 8
Day 15
Day 1
Both agents on the same day
1 to 1.5 hr
Day 1
Gpt
Lonca
Glofit
Glofit
Lonca
Glofit
Lonca
Cycles 2 to 8
Cycle 9+
Arm F
Lonca + Mosun
Day 1
Both agents on the same day
Day 8
Day 15
1 to 1.5 hr
Day 1
Both agents on the same day
1 to 1.5 hr
Day 1
Lonca
Mosun
Mosun
Mosun
Lonca
Mosun
Lonca

Dose-escalation

Arm C	Arm E	Arm F
Lonca[ab]: IV Q3W on D1 of all cycles	Lonca[ab]: IV Q3W on D2 of cycle 1 and D1 of cycles 2+	Lonca[ab]: IV Q3W on D1 of all cycles
Pola[c]: 1.8 mg/kg IV Q3W on D1 of all cycles	Glofit: 2.5 mg on cycle 1 D8, 10 mg on cycle 1 D15, and then 30 mg on D1 of cycles 2-12	Mosun: 5 mg on cycle 1 D1, 45 mg on cycle 1 D8 and D15, and then 45 mg on D1 of cycles 2-8

D, day; Glofit, glofitamab; Gpt, obinutuzumab; IV, intravenous; Lonca, loncastuximab tesirine; Mosun, mosunetuzumab; Pola, polatuzumab vedotin; Q3W, every 3 weeks.
[a]Dose level 1, 90 µg/kg; dose level 2, 120 µg/kg; and dose level 3, 150 µg/kg.
[b]If the starting dose of Lonca is ≥120 µg/kg, the dose will be reduced to 75 µg/kg from cycle 3.
[c]If a dose-limiting toxicity (DLT) is clearly related to Pola, the DLT does not recur after a dose reduction of Pola, and Lonca has not been escalated to the 150-µg/kg level, dose-escalation of Lonca will be continued with a reduced Pola dose of 1.4 mg/kg.

- For part 2, patients will receive the maximum tolerated dose or recommended dose for expansion determined in part 1

Outcomes

- Primary endpoints include the following:
 - Frequency and severity of AEs and serious AEs
 - Dose-limiting toxicities (part 1 only)
 - Frequency of dose delays, dose interruptions, and dose reductions due to AEs
 - Changes from baseline of safety laboratory variables, vital signs, Eastern Cooperative Oncology Group (ECOG) performance status, and 12-lead electrocardiograms
- Secondary endpoints include the following:
 - Overall response rate and complete response rate (2014 Lugano criteria[9]); duration of response; and progression-free, relapse-free, and overall survival
 - Concentrations and pharmacokinetic (PK) parameters of Lonca total antibody, pyrrolobenzodiazepine (PBD)-conjugated antibody, and unconjugated cytotoxin dimer in combination with Pola, glofitamab, or mosunetuzumab
 - Antidrug antibody titers
- Exploratory endpoints include the following:
 - Glofitamab and mosunetuzumab concentrations in circulation
 - Relation between tumor tissue and/or blood biomarkers and selected PK with clinical endpoints

Disclosures

B.T. Hess: consultant or advisory role: ADC Therapeutics and Bristol-Myers Squibb/Celgene. **M.M. Solh:** research funding: Partner Therapeutics; other remuneration: Bristol-Myers Squibb, Amgen, AbbVie, and Seattle Genetics. **M. Gandhi:** honoraria: GlaxoSmithKline, TG Therapeutics, Karyopharm Therapeutics, and Janssen Oncology. **Y. Wang:** employment or leadership position in a company: ADC Therapeutics; stock ownership: Johnson and Johnson (family member). **Y. Qin:** employment or leadership position in a company: ADC Therapeutics; stock ownership: ADC Therapeutics; research funding: ADC Therapeutics. **P.L. Zinzani:** consultant or advisory role: Celltrion, Gilead Sciences, Janssen-Cilag, Bristol-Myers Squibb, Servier, Sandoz, MSD Oncology, Roche, EUSA Pharma, Kyowa Kirin, Takeda, Secura Bio, TG Therapeutics, Novartis, ADC Therapeutics, Incyte, and BeiGene; other remuneration: MSD Oncology, EUSA Pharma, and Novartis. **G.P. Collins:** consultant or advisory role: Roche, Takeda, Incyte, Pfizer, MSD Oncology, Celgene, BeiGene, Daiichi Sankyo, Celleron Therapeutics, and ADC Therapeutics; honoraria: Roche, Takeda, Gilead Sciences, Pfizer, Novartis, Daiichi Sankyo, Incyte, Celleron Therapeutics, MSD Oncology, BeiGene, ADC Therapeutics, and AstraZeneca; research funding: MSD Oncology, Celgene, Bristol-Myers Squibb, Amgen, and Pfizer; travel grants: Roche and Takeda; other remuneration: Roche, Takeda, Novartis, and Gilead Sciences.

Eligibility Criteria

- Key inclusion and exclusion criteria are shown in **Table 1**

Table 1. LOTIS-7 key eligibility criteria

Key inclusion criteria (all arms)	Key exclusion criteria (all arms)
Age ≥18 years	Previously received study medication (applied to relevant arm only)[a]
Pathologic diagnosis of R/R B-NHL (2016 WHO classification) with treatment failures/intolerance • DLBCL (including transformed diseases, but for arms E and F, including transformed FL only) • HGBCL • FL • MZL • For arm C only – MCL – BL	Lymphoma with active CNS involvement Clinically significant third space fluid accumulation (ascites or pleural effusion requiring drainage or associated with breath) Active acute Graft-versus-host disease Post-transplant lymphoproliferative disorder
≥2 prior systemic treatment regimens for part 1 and ≥1 for part 2	Known history of hypersensitivity resulting in treatment discontinuation or positive serum human ADA to a CD19 antibody
Measurable disease (2014 Lugano classification)	
ECOG performance status of 0-2	History of confirmed progressive multifocal leukoencephalopathy
Adequate organ function based on laboratory parameters: • Absolute neutrophil count ≥1.5 × 10³/µL • Platelet count ≥75 × 10³/µL without transfusion in the past 7 days • Hemoglobin ≥9 g/dL; transfusion allowed • ALT, AST, or GGT ≤2.5 × ULN • Total bilirubin ≤1.5 × ULN • Calculated CrCl ≥60 mL/min (Cockcroft–Gault)	History of Stevens–Johnson syndrome, toxic epidermal necrolysis, or macrophage activation syndrome/hemophagocytic lymphohistiocytosis Significant medical comorbidities
Additional key exclusion criteria (arm C)	
Received a stem-cell transplant within 60 days before study treatment	
Additional key exclusion criteria (arms E and F)	
Received autologous stem-cell transplant within 100 days before study treatment	
Received allogenic stem cell or solid organ transplant	
History of CNS lymphoma or leptomeningeal infiltration	
Current or history of CNS disease	
Known active infection; reactivation of a latent infection, whether bacterial, viral, fungal, mycobacterial, or other pathogens; or any major episode of infection requiring hospitalization or treatment with IV antibiotics within four weeks prior to C1D1	
Active or history of autoimmune disease or immune deficiency	
Prior treatment with CAR T-cell therapy within 30 days prior to C1D1	

ADA, antidrug antibody; ALT, alanine aminotransferase; AST, aspartate aminotransferase; BL, Burkitt Lymphoma; C, cycle; CAR, chimeric antigen receptor; CNS, central nervous system; CrCl, creatinine clearance; D, day; DLBCL, diffuse large B-cell lymphoma; ECOG, Eastern Cooperative Oncology Group; FL, follicular lymphoma; GGT, gamma-glutamyl transferase; HGBCL, high-grade B-cell lymphoma; IV, intravenous; MCL, mantle cell lymphoma; MZL, marginal zone lymphoma; R/R B-NHL, relapsed/refractory B-cell non-Hodgkin Lymphoma; ULN, upper limit of normal; WHO, World Health Organization.

[a]Patients who received previous polatuzumab treatment were excluded from arm C; patients with previous glofitamab treatment were excluded from arm E; and patients with previous mosunetuzumab treatment were excluded from arm F.

Study Assessments

- Study assessments are shown in **Table 2**

Table 2. Study assessments

Efficacy	Safety
Disease assessment • Imaging • Clinical examination	• AEs • SAEs • Physical examination • ECOG performance status • Height and weight • Vital signs • Laboratory tests • Pregnancy test • ECGs
PK, PD, and immunogenicity	
• PK of Lonca, PBD-conjugated antibody, total antibody, and unconjugated PBD-dimer cytotoxin in serum • ADA in whole blood • Blood cfDNA, gDNA, mRNA, flow cytometry, and cytokines • Tumor tissue biomarkers	

ADA, antidrug antibody; AE, adverse events; cfDNA, cell-free DNA; gDNA, genomic DNA; ECG, electrocardiogram; ECOG, Eastern Cooperative Oncology Group; Lonca, loncastuximab tesirine; PBD, pyrrolobenzodiazepine; PD, pharmacodynamic; PK, pharmacokinetic; SAE, serious adverse events.

STUDY STATUS

- The study opened for recruitment in June 2022, and as of March 2023, 17 patients have been screened, and 12 patients have been treated in arm C (Lonca + Pola)
- Enrollment in part 1 (dose-escalation) started for arms E and F in July 2023

Acknowledgments

Funding: ADC Therapeutics SA; Medical writing support was provided by CiTRUS Health Group (United States) in accordance with Good Publication Practice (GPP 2022) guidelines.

***Contact Information**

Brian T. Hess, MD. Email: hessbr@musc.edu

References

1. ZYNLONTA® (loncastuximab tesirine-lpyl) prescribing information. Available at: https://www.adctherapeutics.com/wp-content/uploads/2022/10/ZYLONTA-PI_October-2022_LOCKED.pdf. Accessed July 12, 2023.
2. ZYNLONTA® (loncastuximab tesirine-lpyl) product information. Available at: https://www.ema.europa.eu/en/medicines/human/EPAR/zynlonta#product-information-section. Accessed July 12, 2023.
3. 3. ADC Therapeutics. ADC Therapeutics Reports Fourth Quarter and Full Year 2022 Financial Results and Provides Business Updates. Press release; 2023.
4. Caimi PF, et al. *Lancet Oncol*. 2021;22:790-800
5. Sachini N, et al. *Blood*. 2021;138 (Supplement 1):2273
6. Hutchings M, et al. *J Clin Oncol*. 2021;39:1959-1970
7. Budde LE, et al. *J Clin Oncol*. 2022;40:481-491
8. ADC Therapeutics S.A. ClinicalTrials.gov registration number: NCT04970901. Updated April 26, 2023. Accessed July 12, 2023. https://clinicaltrials.gov/ct2/show/NCT04970901
9. Cheson BD, et al. *J Clin Oncol*. 2014;32:3059-3068

Copies of this poster obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

Poster presented at the Eleventh Annual Meeting of the Society of Hematologic Oncology (SOHO 2023). September 6-9, 2023, Houston, TX, USA | © 2023 American Society of Clinical Oncology, Inc. Reused with permission. This poster was accepted and previously presented at the 2023 ASCO Annual Meeting. All rights reserved. Annual Meeting. June 2-6, 2023, Chicago, IL USA