Updated Results of the Safety Run-In of the Phase 3 LOTIS-5 Trial: Novel Combination of Loncastuximab Tesirine With Rituximab (Lonca-R) Versus Immunochemotherapy in Patients With R/R DLBCL

Michal Kwiatek,[1*] Sebastian Grosicki,[2] Javier Lopez Jimenez,[3] Sergio Felipe Pinzón Mariño,[4] Sylvia Snauwaert,[5] Edwin Kingsley,[6] Giovanna Zacchetti,[7] Ying Wang,[8] Luqiang Wang,[8] Julien Depaus[9]

[1]Centrum Medyczne Pratia Poznań, Skórzewo, Poland; [2]Medical University of Silesia, Katowice, Poland; [3]Hospital Universitario Ramón y Cajal, Madrid, Spain; [4]Department of Clinical Hematology, Hospital del Mar, Barcelona, Spain; [5]Algemeen Ziekenhuis Sint-Jan Brugge-Oostende – Campus Sint-Jan, Brugge, Belgium; [6]Comprehensive Cancer Centers of Nevada, Las Vegas, NV, USA; [7]ADC Therapeutics SA, Épalinges, Switzerland; [8]ADC Therapeutics America, Inc., Murray Hill, NJ, USA; [9]Centre Hospitalier Universitaire (CHU) Université Catholique de Louvain (UCL) Namur, Godinne Site, Yvoir, Belgium

ABCL-515

SUMMARY

OBJECTIVE

To characterize the safety and efficacy of Lonca + rituximab (Lonca-R) in patients with R/R DLBCL who have had ≥1 prior systemic therapy

METHODS

LOTIS-5

PART 1
Safety run-in (n = 20)
Lonca-R

PART 2
Lonca-R vs R-GemOx (n ≈ 330)
Lonca-R | R-GemOx

ORR 80%

CR 50%

FIXED DURATION TREATMENT OF LONCA + RITUXIMAB

There were no new safety signals

The most common grade ≥3 TEAEs were increased GGT (n = 5) and neutropenia (n = 3)

CONCLUSIONS

Lonca-R demonstrated no new safety signals and showed encouraging antitumor activity in patients with R/R DLBCL; initial signs of response durability with Lonca-R are promising

CR, complete response; DLBCL, diffuse large B-cell lymphoma; DoR, duration of response; GGT, gamma-glutamyltransferase; Lonca, loncastuximab tesirine; ORR, overall response rate; PFS, progression-free survival; R, rituximab; R-GemOx, rituximab + gemcitabine + oxaliplatin; R/R, relapsed/refractory; TEAE, treatment-emergent adverse event.

INTRODUCTION

- Patients with relapsed or refractory diffuse large B-cell lymphoma (R/R DLBCL) typically have poor outcomes following treatment[1]
- Loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]) is an antibody–drug conjugate (ADC) comprising an anti-CD19 monoclonal antibody conjugated to a pyrrolobenzodiazepine dimer toxin, indicated for patients with R/R DLBCL after ≥2 systemic treatments[2,3]
- Rituximab (R) is part of standard immunotherapy for DLBCL, both as frontline therapy and in subsequent treatments[4,5]
- Preclinical evidence suggests that the addition of R to anti-CD19 ADC therapy may result in prolonged tumor control[6]
- LOTIS-5 aims to evaluate Lonca + R (Lonca-R) versus standard immunochemotherapy of R + gemcitabine + oxaliplatin (R-GemOx) in patients with R/R DLBCL
 - In part 1, 20 patients were enrolled in a nonrandomized safety run-in period with Lonca-R to characterize the safety of Lonca-R combination therapy

OBJECTIVE

- To characterize the safety and efficacy of Lonca-R in patients with R/R DLBCL who have had ≥ 1 prior systemic therapy

METHODS

Study Design

- LOTIS-5 is a phase 3, randomized, open-label, 2-part, 2-arm, multicenter study of Lonca-R in patients with R/R DLBCL (NCT04384484)
- In part 1, 20 patients were enrolled in a nonrandomized safety run-in period with Lonca-R to characterize the safety of Lonca-R combination therapy
 - Preliminary results presented at SOHO 2022 were updated with new safety/efficacy data collected in part 1 of the study[7]
- In part 2, approximately 330 patients will be randomly assigned 1:1 to receive Lonca-R or R-GemOx
- Patients received 0.15 mg/kg Lonca + 375 mg/m² R every 3 weeks for 2 cycles, then 0.075 mg/kg Lonca + 375 mg/m² R every 3 weeks for up to 6 additional cycles (**Figure 1**)

Figure 1. LOTIS-5 study design

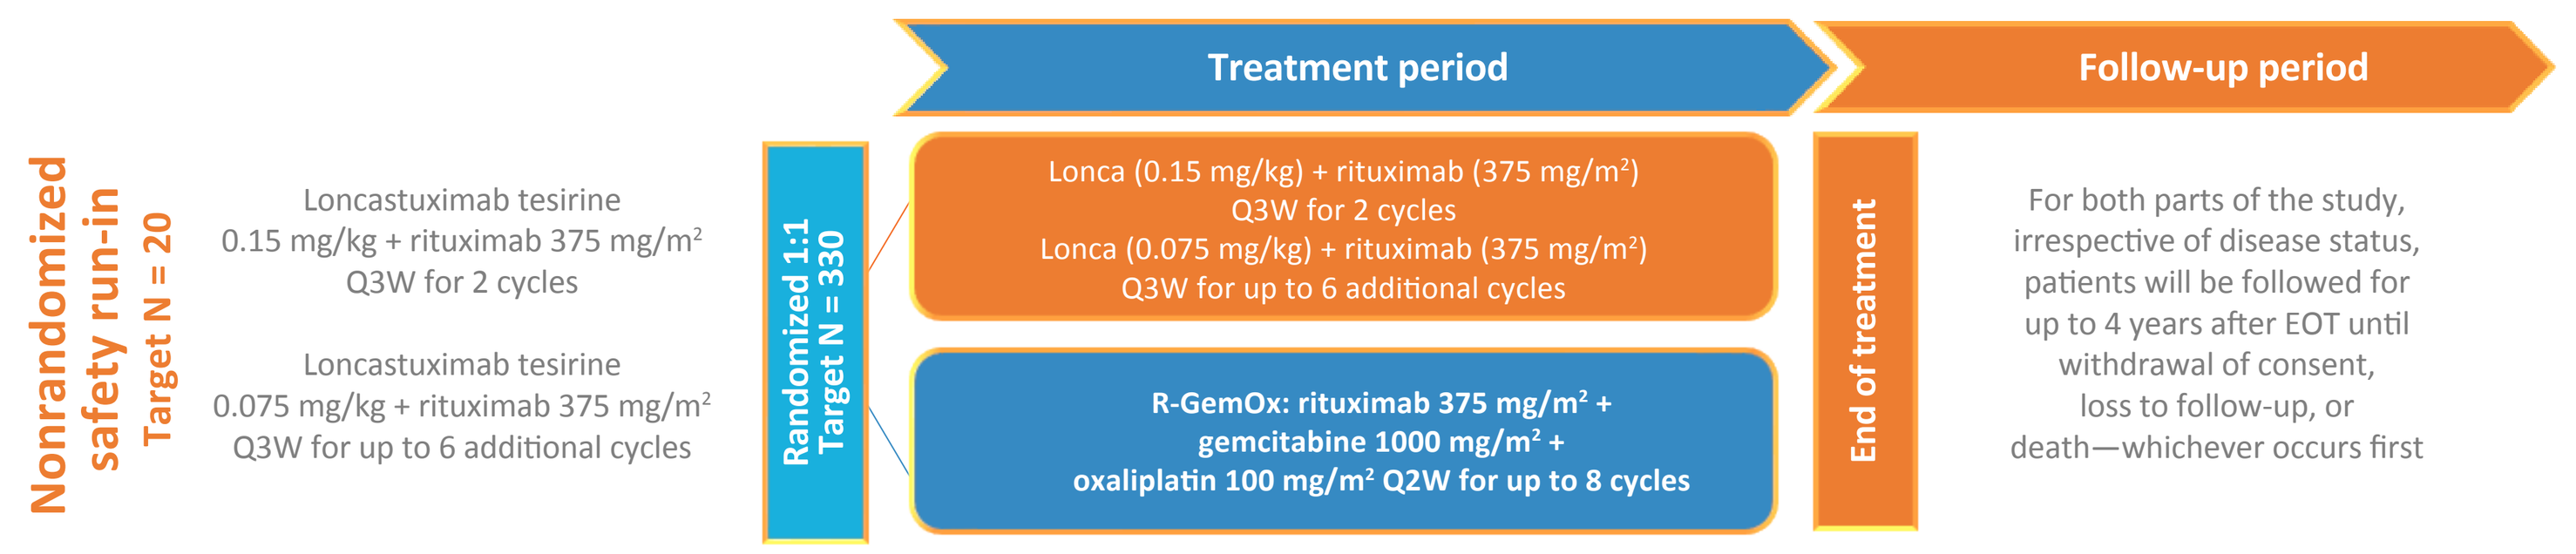

Nonrandomized safety run-in
Target N = 20
Loncastuximab tesirine
0.15 mg/kg + rituximab 375 mg/m²
Q3W for 2 cycles
Loncastuximab tesirine
0.075 mg/kg + rituximab 375 mg/m²
Q3W for up to 6 additional cycles
Randomized 1:1
Target N = 330
Treatment period
Lonca (0.15 mg/kg) + rituximab (375 mg/m²)
Q3W for 2 cycles
Lonca (0.075 mg/kg) + rituximab (375 mg/m²)
Q3W for up to 6 additional cycles
R-GemOx: rituximab 375 mg/m² +
gemcitabine 1000 mg/m² +
oxaliplatin 100 mg/m² Q2W for up to 8 cycles
End of treatment
Follow-up period
For both parts of the study, irrespective of disease status, patients will be followed for up to 4 years after EOT until withdrawal of consent, loss to follow-up, or death—whichever occurs first

EOT, end of treatment; Lonca, loncastuximab tesirine; QXW, every X weeks; R-GemOx, rituximab + gemcitabine + oxaliplatin.

Outcomes

- The primary end point is progression-free survival by independent central review
- Key secondary end points include overall survival, overall response rate, duration of response, safety, pharmacokinetic parameters, and changes in patient-reported outcomes

Eligibility Criteria

- Key inclusion criteria and exclusion criteria are shown in **Table 1**

Table 1. Key inclusion and exclusion criteria

Key inclusion criteria	Key exclusion criteria
• Adults with a pathologic diagnosis of R/R DLBCL (including DLBCL transformed from indolent lymphoma) or HGBCL, with *MYC* and *BCL2* and/or *BCL6* rearrangements • R/R disease following ≥1 multiagent systemic treatment regimen • Measurable disease (2014 Lugano Classification[8]) • Not a candidate for SCT based on performance status, advanced age, and/or significant medical comorbidities (as considered by the investigator) • Patients who have received previous CD19-directed therapy must have a biopsy that shows CD19 expression after completion of the CD19-directed therapy • ECOG performance status score of 0-2 • Adequate organ function	• Previous treatment with Lonca or R-GemOx • Autologous SCT within 30 days before the start of the study drug • Allogeneic SCT within 60 days before the start of the study drug • Lymphoma with active CNS involvement, including leptomeningeal disease • Serologic evidence of chronic HBV infection and inability or unwillingness to receive standard prophylactic antiviral therapy or with detectable HBV viral load • Serologic evidence of HCV infection without completion of curative treatment or with detectable HCV viral load • Clinically significant third-space fluid accumulation (ie, ascites requiring drainage or pleural effusion either requiring drainage or associated with shortness of breath) • Major surgery within 4 weeks before the start of the study drug, unless approved by the sponsor • Radiotherapy, chemotherapy, or other antineoplastic therapy within 14 days before the start of the study drug, unless approved by the sponsor

CNS, central nervous system; DLBCL, diffuse large B-cell lymphoma; ECOG, Eastern Cooperative Oncology Group; HBV, hepatitis B virus; HCV, hepatitis C virus; HGBCL, high-grade B-cell lymphoma; Lonca, loncastuximab tesirine; R-GemOx, rituximab + gemcitabine + oxaliplatin; R/R, relapsed/refractory; SCT, stem cell transplant.

RESULTS

Patient Population

- The 20 patients in the safety run-in were a median age of 74.5 years (range, 35-93 years) (**Table 2**)
 - Patients received a median of 1 previous therapy (range, 1-7)
 - Seven patients (35%) received ≥2 previous therapies
- As of the April 10, 2023, data cutoff date:
 - The median number of doses administered was 5 (range, 1-8), and the median duration of follow-up was 10.8 months (range, 1.9-21.9 months)

Safety Outcomes

- All patients had ≥1 treatment-emergent adverse event (TEAE), and 11 (55%) patients had grade ≥3 TEAEs (**Table 3**)
- The most common all-grade TEAEs, regardless of the relationship to the study treatment, were increased gamma-glutamyltransferase (5 [25%]), rash (5 [25%]), decreased appetite (4 [20%]), and fatigue (4 [20%])
- The most common grade ≥3 TEAEs were increased gamma-glutamyltransferase (5 patients [25%]) and neutropenia (3 patients [15%])
- Serious adverse events (SAE) were observed in 9 patients (45%). The most common SAE was infection in 6 patients (30%)
- Overall, including survival follow-up, 8 of 20 patients died (4 due to disease progression, 2 due to COVID-19 infection, 1 due to pancreatic neoplasia, and 1 due to stroke), 2 of which occurred within 30 days of last treatment (1 due to disease progression and 1 due to COVID-19 infection).

Efficacy Outcomes

- The overall response rate by central review was 16/20 (80%) patients (**Table 3**)
- Complete responses were observed in 10/20 (50%) patients
- Partial responses were observed in 6/20 (30%) patients
- The median duration of response was 8.0 months (95% CI, 3.2-NR) (**Figure 2**)
- The median progression-free survival was 8.3 months (95% CI, 4.5-NR) (**Figure 3**)

Table 2. Baseline characteristics for patients enrolled in LOTIS-5 safety run-in (n = 20)

Age, median (min, max), years	74.5 (35, 93)
Prior systemic therapies, median (min, max)	1 (1, 7)
Sex, n (%)	
Female	11 (55)
Male	9 (45)
ECOG PS	
Grade 0	7 (35)
Grade 1	10 (50)
Grade 2	3 (15)
Disease stage (Ann Arbor criteria), n (%)	
Stage I	1 (5)
Stage II	6 (30)
Stage III	6 (30)
Stage IV	7 (35)
Primary category, n (%)	
DLBCL, NOS	18 (90)
HGBCL with *MYC* and *BCL2* and/or *BCL6* rearrangements	2 (10)
Prior stem cell transplant, n (%)	
Yes	1 (5)
No	19 (95)
First-line prior systemic response, n (%)	
Relapsed	18 (90)
Refractory	2 (10)
Last-line prior systemic response, n (%)	
Relapsed	15 (75)
Refractory	5 (25)

DLBCL, diffuse large B-cell lymphoma; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HGBCL, high-grade B-cell lymphoma; NOS, not otherwise specified.

Table 3. Results for LOTIS-5 safety run-in (n = 20)

	n (%)	95% CI
Efficacy in patients with R/R DLBCL		
Overall response rate	16 (80)	56.3-94.3
Complete response	10 (50)	27.2-72.8
Partial response	6 (30)	11.9-54.3
Safety end points		
All TEAEs	20 (100)	
Increased gamma-glutamyltransferase	5 (25)	
Rash	5 (25)	
Decreased appetite	4 (20)	
Fatigue	4 (20)	
Grade ≥3 TEAEs	11 (55)	
Increased gamma-glutamyltransferase	5 (25)	
Neutropenia	3 (15)	
Serious adverse events	9 (45)	
Infection	6 (30)	
Hyponatremia	1 (5)	
Anaphylactic reaction	1 (5)	
Pleural effusion	1 (5)	
Malaise	1 (5)	
Neurological decompensation	1 (5)	
Deaths		
During treatment	1 (5)	
During follow up	7 (35)	

DLBCL, diffuse large B-cell lymphoma; R/R, relapsed or refractory; TEAE, treatment-emergent adverse event.

Figure 2. Kaplan–Meier curve of the duration of response by independent review

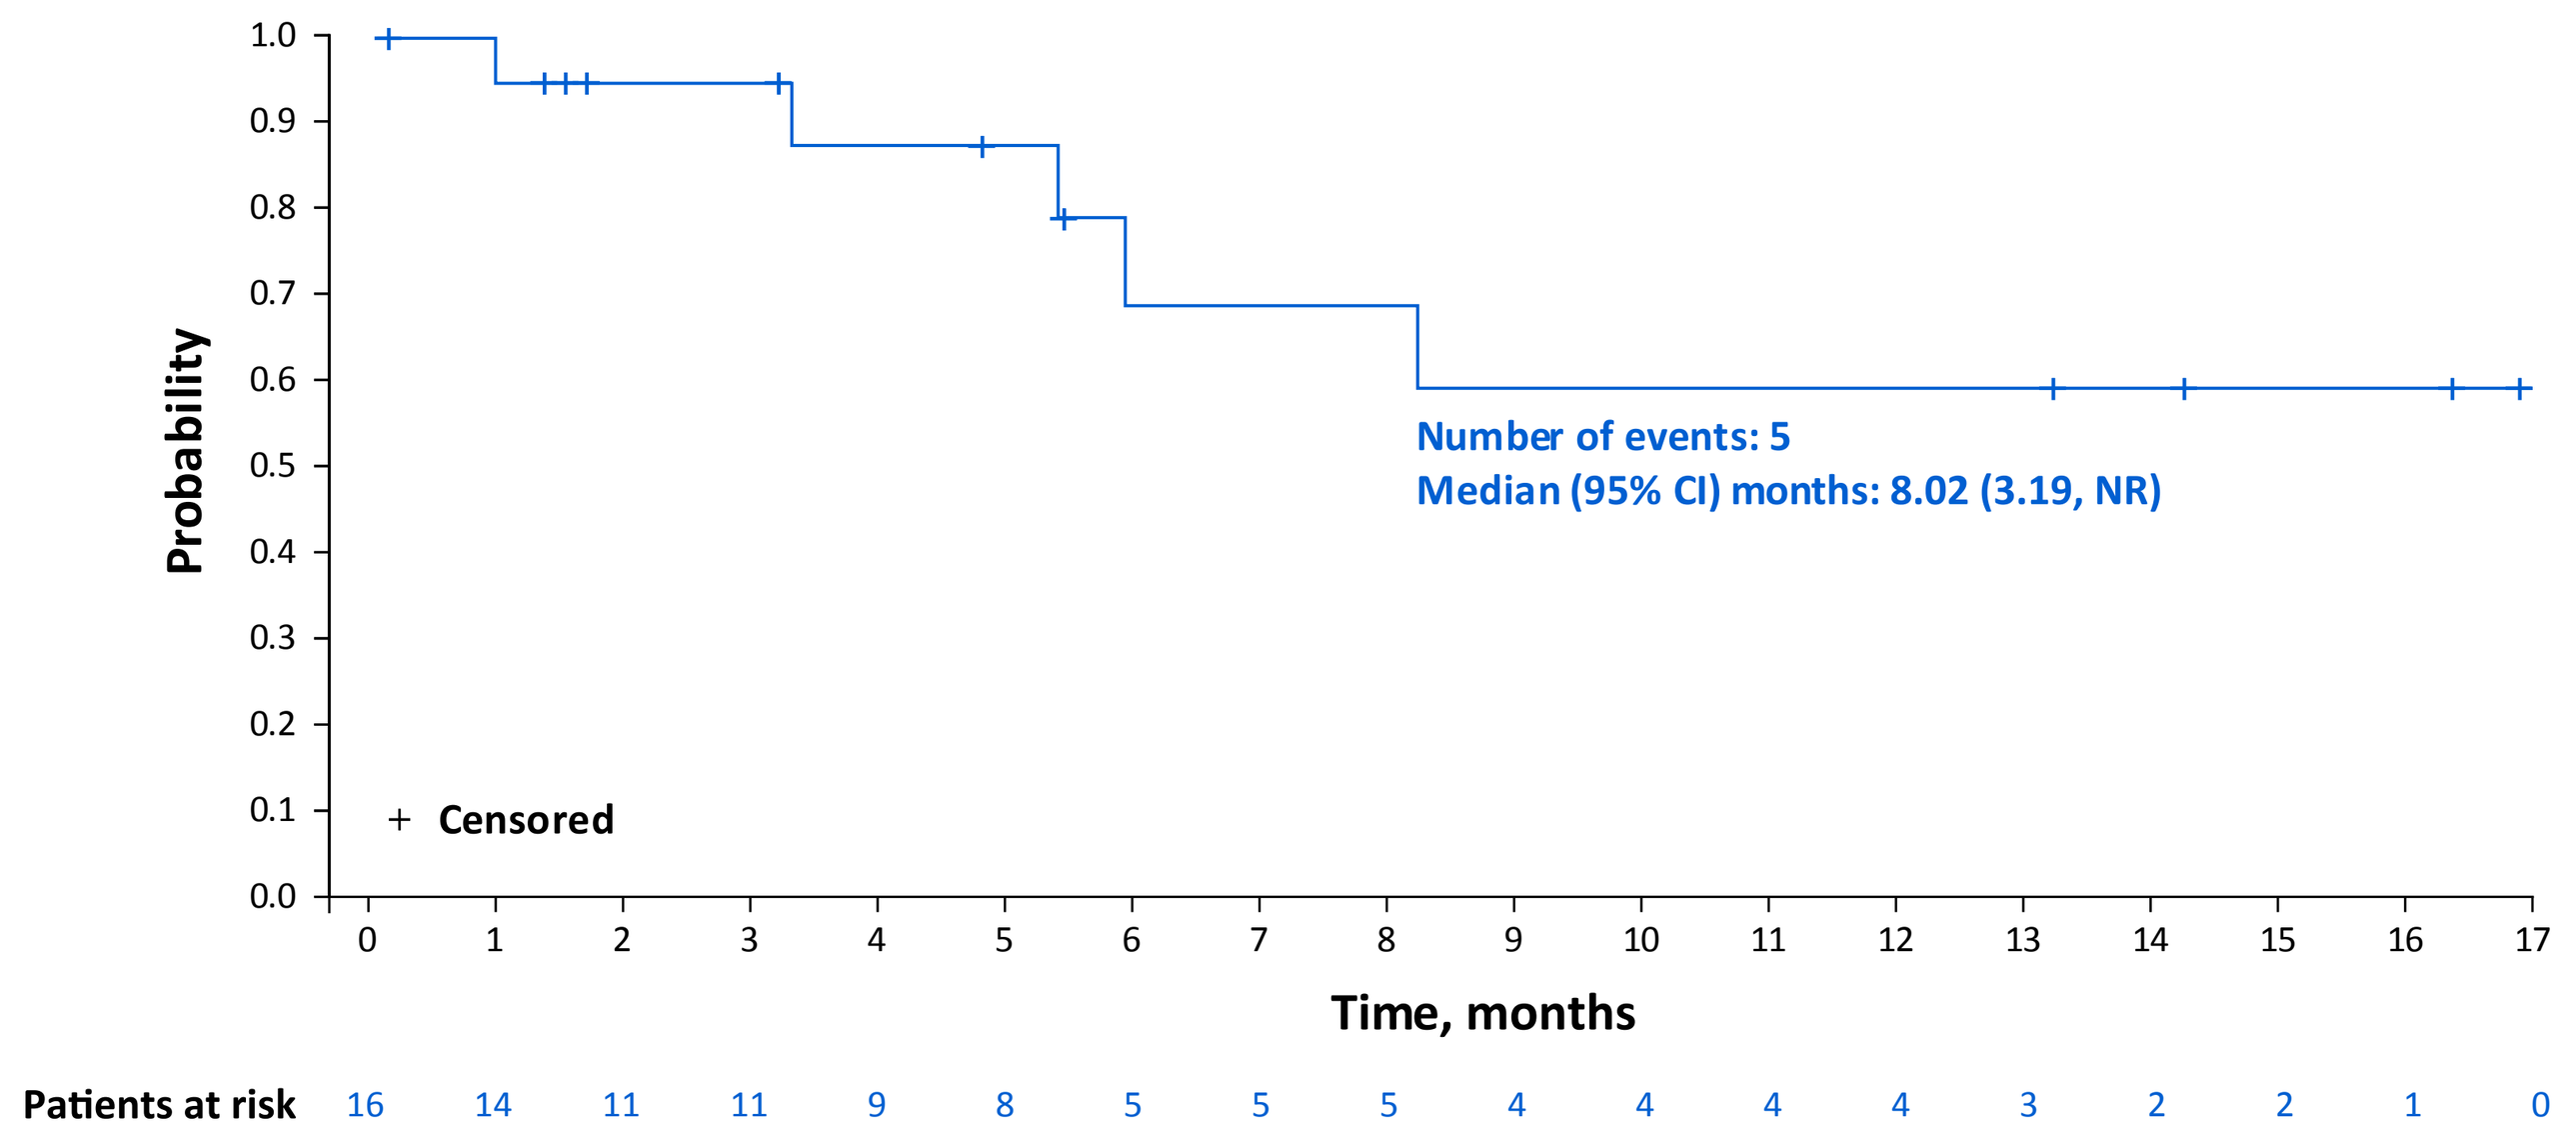

Probability
Time, months
+ Censored
Number of events: 5
Median (95% CI) months: 8.02 (3.19, NR)
Patients at risk 16 14 11 11 9 8 5 5 5 4 4 4 4 3 2 2 1 0

Figure 3. Kaplan–Meier curve of progression-free survival by independent review

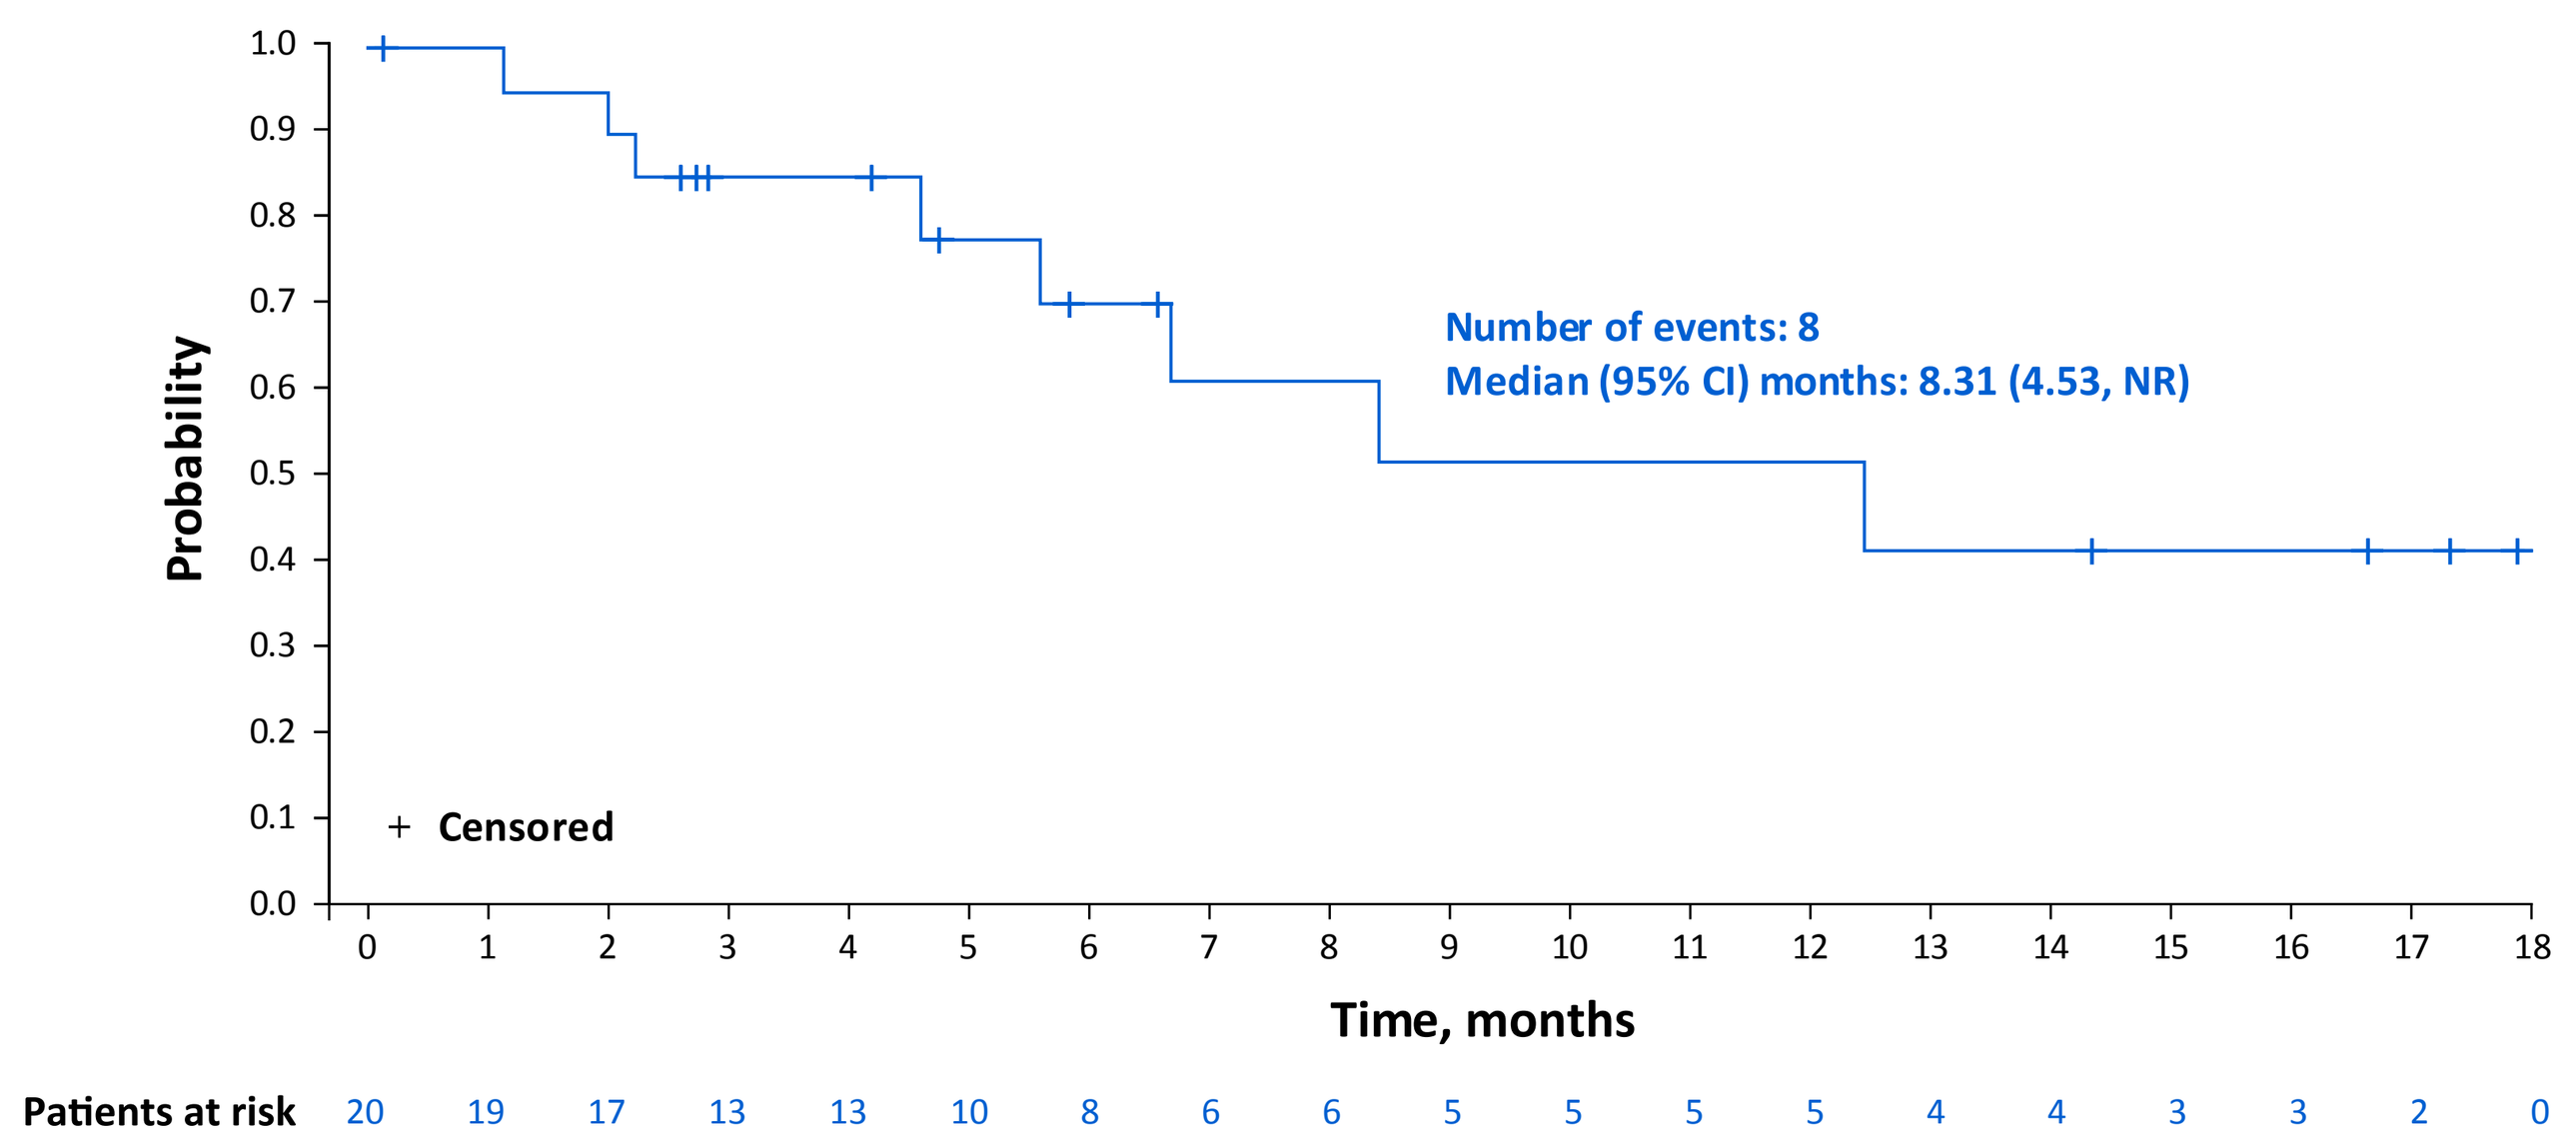

Probability
Time, months
+ Censored
Number of events: 8
Median (95% CI) months: 8.31 (4.53, NR)
Patients at risk 20 19 17 13 13 10 8 6 6 5 5 5 5 4 4 3 3 2 0

CONCLUSIONS

- Lonca-R demonstrated no new safety signals and showed encouraging antitumor activity in patients with R/R DLBCL
- Initial signs of response durability (8.02 months) with Lonca-R are promising
- Lonca-R has a fixed treatment duration, potentially making it an appealing alternative to continuous therapies
- Part 2 of LOTIS-5 commenced in January 2022; recruitment is ongoing

Acknowledgments

This study is funded by ADC Therapeutics SA. Medical writing support was provided by CiTRUS Health Group in accordance with Good Publication Practices (GPP 2022), funded by ADC Therapeutics SA. The authors would like to thank all participating patients and their families, all study coinvestigators, and the research coordinators.

Disclosures

M Kwiatek: consultant/advisor for ADC Therapeutics; presenting author for MorphoSys. **S Grosicki:** no disclosures. **J Lopez Jimenez:** consultant/advisor for AbbVie, BMS, Gilead, Janssen, and MSD; speaker bureau for AbbVie and Janssen; travel/accommodations/expenses from AbbVie, AstraZeneca, BMS, Gilead, MSD, and Roche. **SF Pinzón Mariño:** no disclosures. **S Snauwaert:** consultant/advisor for AbbVie, AstraZeneca, Janssen, Lilly, and Takeda. **E Kingsley:** employee of US Oncology; research funding from ADC Therapeutics, BeiGene, Daiichi Sankyo, and Seagen. **G Zacchetti:** employee of ADC Therapeutics with stock ownership. **Y Wang:** employee of ADC Therapeutics with stock ownership. **L Wang:** employee of ADC Therapeutics with stock ownership. **J Depaus:** consultant for Janssen, Novartis, and Takeda.

*Contact information

Michal Kwiatek, MD: michal.kwiatek@pratia.com

References

1. Crump, M, et al. *Blood*. 2017;130:1800-1808.
2. Caimi, PF, et al. *Lancet Oncol*. 2021;22:790-800.
3. ADC Therapeutics SA. ZYNLONTA® prescribing information. October 2022.
4. Sehn, LH, et al. *Blood*. 2015;125:22-32.
5. Ngu, H, et al. *Am Soc Clin Oncol Educ Book*. 2022;42:1-14.
6. Ryan, MC, et al. *Blood*. 2017;130:2018-2026.
7. Kingsley, E, et al. *Clin Lymphoma Myeloma Leuk*. 2022;22:S372.
8. Cheson, BD, et al. *J Clin Oncol*. 2014;32:3059-3068.

Copies of this poster obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at the Eleventh Annual Meeting of the Society of Hematologic Oncology (SOHO 2023). September 6-9, 2023, Houston, TX, USA