POSTER 5085

Preclinical development of NaPi2b-PL2202, a novel camptothecin-based antibody-drug conjugate targeting solid tumors expressing NaPi2b

Elizabeth Horsley, Asma Jabeen, Nicolas Veillard, Karin Havenith, Narinder Janghra, Pedro Alves, Cecile Oblette, Ian Kirby, Paul W. Hogg, Francesca Zammarchi, Lolke de Haan, Patrick H. van Berkel

ADC Therapeutics UK (Ltd), London, UK

ACKNOWLEDGEMENTS

In vivo studies: Charles River Discovery Research Services (USA) (mouse xenograft studies); Syngene (India) (rat toxicity study) and LabCorp, (UK) (cynomolgus monkey toxicity study).

IHC study: Charles River Laboratories (Germany).

REFERENCES

1. Bodyak, N.D., et al., The Dolaflexin-based Antibody–Drug Conjugate XMT-1536 Targets the Solid Tumor Lineage Antigen SLC34A2/NaPi2b. Mol Cancer Ther, 2021. 20: p. 896–905.
2. Valsenkova, R., et al., Characterization of SLC34A2 as a Potential Prognostic Marker of Oncological Diseases. Biomolecules, 2021. 11(1878).
3. Hakim, S. A., et al., Evaluation of the Prognostic Value of Solute Carrier Family 34 Member 2 "SLC34A2" in Papillary Thyroid Carcinoma: An Immunohistochemical Study. Analytical Cellular Pathology, 2021.
4. Heynemann, S., et al., NaPi2b expression in a large surgical Non-Small Cell Lung Cancer (NSCLC) cohort. Clin Lung Cancer. 2022 Mar;23(2):e90-e98.
5. Lin, K. et al.,. Preclinical Development of an Anti-NaPi2b (SLC34A2) Antibody-Drug Conjugate as a Therapeutic for Non-Small Cell Lung and Ovarian Cancers. Clin Cancer Res. 2015 Nov 15;21(22):5139-50.

MATERIALS AND METHODS

Cytotoxicity of NaPi2b-PL2202, lifastuzumab-PL2202 and the isotype control ADC (B12-VA-PL2202) was determined by the CellTiterGlo® assays (Promega). Antibody cell binding data was analysed by flow cytometry.

Internalization of the NaPi2b antibody, lifastuzumab and IgG isotype control was performed by Incucyte®. FabFluor labelled α-NaPi2b or IgG1 isotype control (2 µg/mL) were used in OVCAR-3 cells. HD phase and red fluorescence images (10x) were captured. Images of cells treated with FabFluor-α-NaPi2b with red cytosolic fluorescence indicative of internalization.

Comparison of bystander activity was determined by a medium transfer method using the CellTiterGlo® assay (Promega).

In *in vivo* studies, NaPi2b-PL2202, lifastuzumab vedotin or B12-VA-PL2202 was administered intravenously (i.v.) as single dose to athymic nude or SCID mice containing G-402, OVCAR-3, or H441 s.c. xenografts.

PK was evaluated in rats (n=3/group) following a single IV bolus or infusion dose of NaPi2b-PL2202, and in cynomolgus monkeys (n=3/group) following one or two doses of NaPi2b-PL2202 three weeks apart via IV infusion. Serum exposure to total (conjugated and unconjugated Ab) and exatecan conjugated Ab were determined by ELISA/ECLIA and exposure to free exatecan was determined by LC-MS/MS. PK parameters determined by non-compartmental analysis (NCA).

Analysis of NaPi2b membranous expression on human TMAs from patients diagnosed with NSCLC, ovarian or endometrial cancer was performed by IHC using NaPi2b antibody, MERS67[4].

Disclosures

All authors are current or former employees of ADC Therapeutics that may hold stock or stock options.

CONCLUSIONS

- *In vitro*, NaPi2b-PL2202 demonstrated potent and target-mediated cytotoxicity in a panel of NaPi2b-positive solid tumor cell lines.
- NaPi2b-antibody was efficiently internalized in lysosomes by NaPi2b-expressing OVCAR-3 cells, showing increased intracellular uptake compared to lifastuzumab. Antibody-based ADC based on lifastuzumab was much less potent *in vitro* compared to NaPi2b-PL2202.
- NaPi2b-PL2202 showed indirect bystander killing activity of NaPi2b-negative KB cells incubated with conditioned medium from NaPi2b-PL2202-treated IGROV-1 cells. At equivalent doses, bystander activity of NaPi2b-PL2202 was higher than NaPi2b-DXd and lifastuzumab vedotin.
- *In vivo*, NaPi2b-PL2202 showed strong antitumor activity against G-402, OVCAR-3 and H441 cancer xenograft models, highlighting its target-mediated antitumor activity. NaPi2b-PL2202 showed higher tumor regression at a much lower dose compared to lifastuzumab vedotin.
- NaPi2b-PL2202 (binds to rat and cyno NaPi2b) was tolerated as a single, 300 mg/kg IV bolus dose in male rats and up to 40 mg/kg following two doses three weeks apart via IV infusion in male cynomolgus monkeys.
- TK profiles for NaPi2b-PL2202 ADC and total antibody were generally comparable in rats and cynomolgus monkeys, with ADC concentrations slightly higher than total Ab concentrations. Linear TK with a t½ of ~12.9-14 days in rats and ~7-10 days in monkeys (ADC assays); free exatecan was detectable throughout the dosing period with apparent t½ of 9.5 days in rats.
- Membranous expression of NaPi2b was confirmed by IHC in a high proportion of non-small cell lung cancer (NSCLC), ovarian and endometrial cancers.
- In conclusion, NaPi2b-PL2202 demonstrated potent and specific *in vitro* and *in vivo* anti-tumor activity and was stable and well tolerated in both rats and cynomolgus monkeys warranting progression of the ADC into clinical trials.

INTRODUCTION

- NaPi2b, encoded by the SLC34A2 gene, is a multi-transmembrane, sodium-dependent phosphate transporter expressed at high levels in tumors and low levels in normal tissue.
- Functionally, it is involved in trans-cellular flux of phosphate in small intestine and in the synthesis of surfactant in lung alveoli.
- NaPi2b is reported to be expressed at a high frequency in ovarian carcinoma as well as in NSCLC, bladder, endometrial and papillary thyroid carcinoma[1] and its expression is associated with poor prognosis[2, 3].

Figure 1.

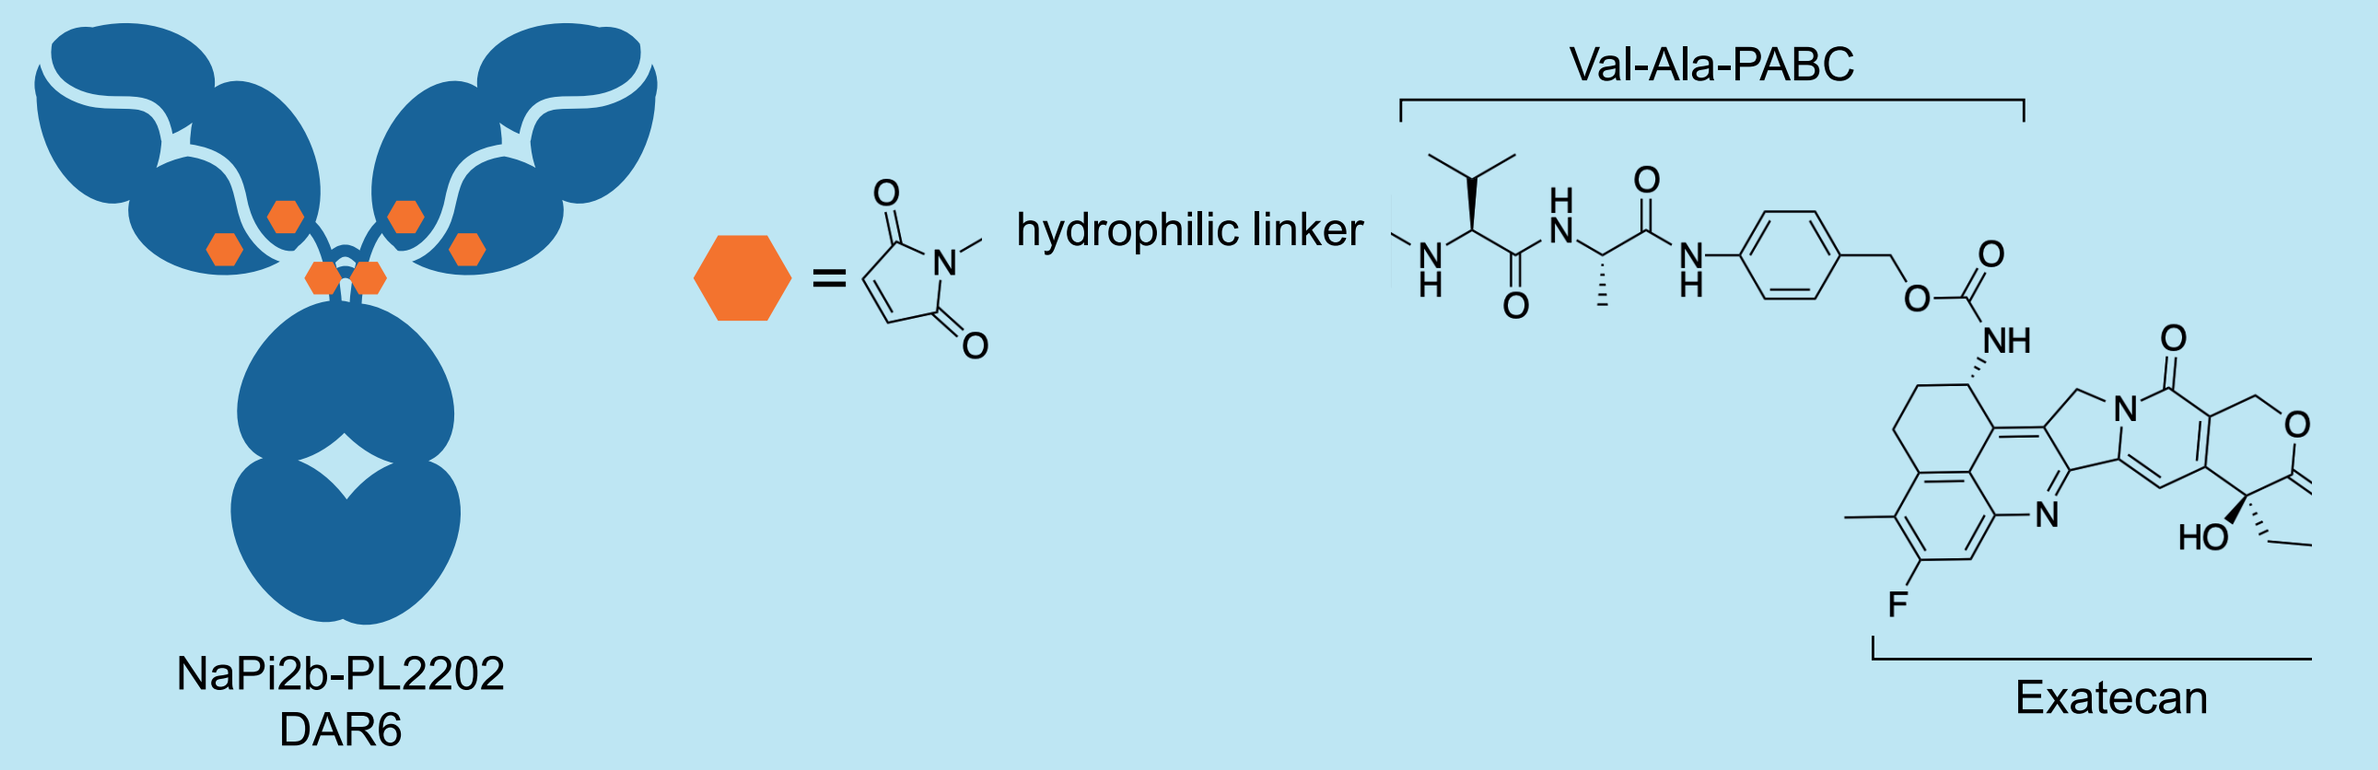

NaPi2b-PL2202
DAR6
hydrophilic linker
Val-Ala-PABC
Exatecan

- NaPi2b-PL2202 is a novel ADC, composed of a humanized IgG1, Fc silenced antibody against NaPi2b to which the exatecan-containing payload PL2202 has been site-specifically conjugated with a DAR of 6 (**Figure 1**). This was achieved by introduction of the Cys228Val mutation in the hinge of Heavy chain.

AIM OF THE STUDY

- The purpose of this study was to determine the *in vitro* cytotoxicity and *in vivo* efficacy of NaPi2b-PL2202 in xenograft models as well as its toxicity and pharmacokinetic (PK) profile in the rat and cynomolgus monkey.
- We also characterized the *in vitro* mechanism of action, such as internalization, bystander activity and measured NaPi2b expression by immunohistochemistry (IHC) in human tumor specimens of ovary, lung, and endometrium.

RESULTS

Table 1. NaPi2b Ab cell binding and NaPi2b-PL2202 *in vitro* cytotoxicity

	IGROV-1	G-402	OVCAR-3	HCC-78	NCI-H441	MC-IXC	EBC-1
NaPi2b status	+++	++	+++	+	++	+++	-
Tumor type	ovarian	renal leiomyoblastoma	ovarian	NSCLC	lung	brain	lung
NaPi2b-PL2202 IC50 (pM)	2,750	305	109	132	58,466	1,241	61,100
B12-VA-PL2202 IC50 (pM)	125400	34800	43180	26990	106000	11450	42,700

NaPi2b antibody binding to cells and *in vitro* cytotoxicity (IC50) of NaPi2b-PL2202 and isotype-control ADC (B12-VA-PL2202) on a panel of seven cancer cell lines.

Figure 2. NaPi2b Ab cell binding and NaPi2b-PL2202 *in vitro* cytotoxicity

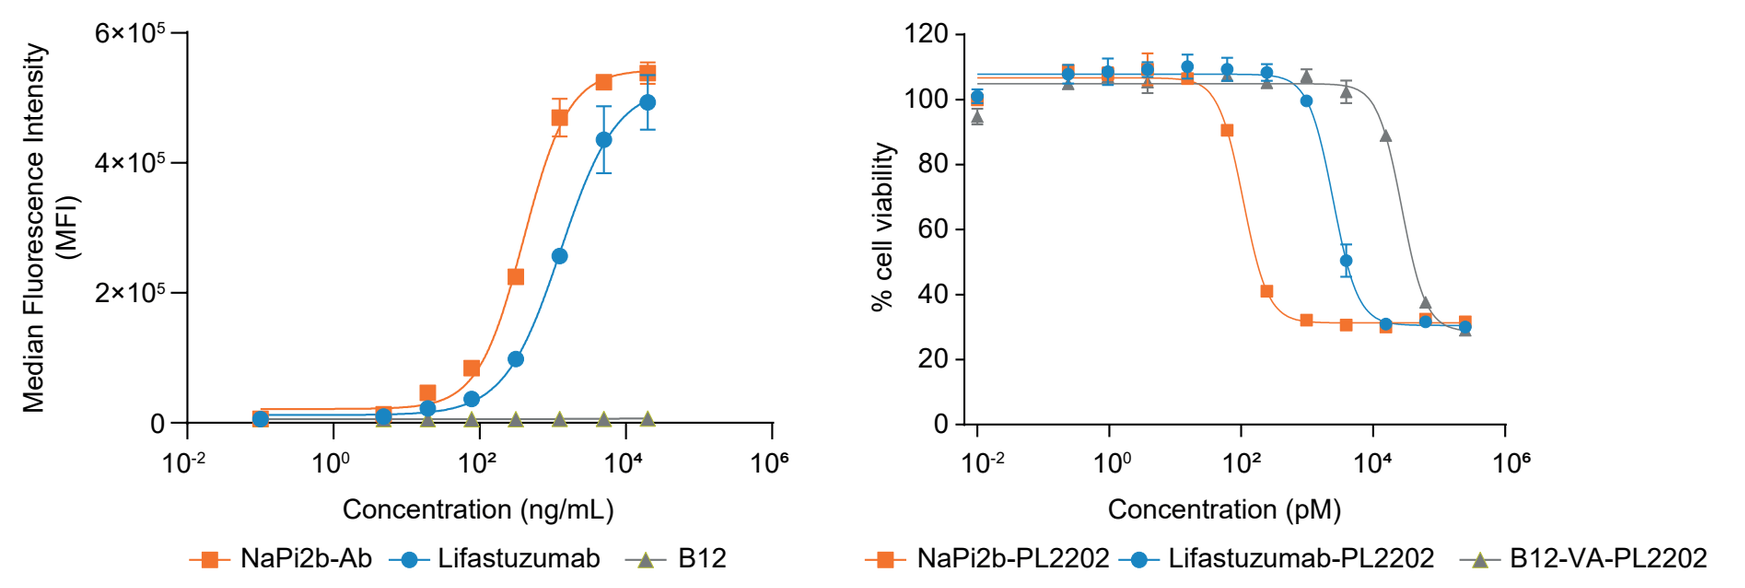

A
B
Median Fluorescence Intensity (MFI)
Concentration (ng/mL)
NaPi2b-Ab
Lifastuzumab
B12
% cell viability
Concentration (pM)
NaPi2b-PL2202
Lifastuzumab-PL2202
B12-VA-PL2202

antibodies	EC_{50} pM
NaPi2b	398.7
lifastuzumab	1281
B12 isotype IgG	12079

ADCs	IC_{50} pM
NaPi2b-PL2202	107.3
lifastuzumab-PL2202	2473
B12-VA-PL2022	27168

A. NaPi2b Ab has higher binding to OVCAR-3 cells compared to lifastuzumab[5]. **B.** NaPi2b-PL2202 exhibits higher *in vitro* cytotoxicity compared to lifastuzumab conjugated to PL2202 (Both DAR 6). Side-by-side comparison of IC50 curves for OVCAR-3 cells treated with NaPi2b-PL2202, Lifastuzumab-PL2202 and B12-VA-PL2202 (isotype control ADC) by CellTiter-Glo luminescent assay.

Figure 3. Internalization of NaPi2b antibodies

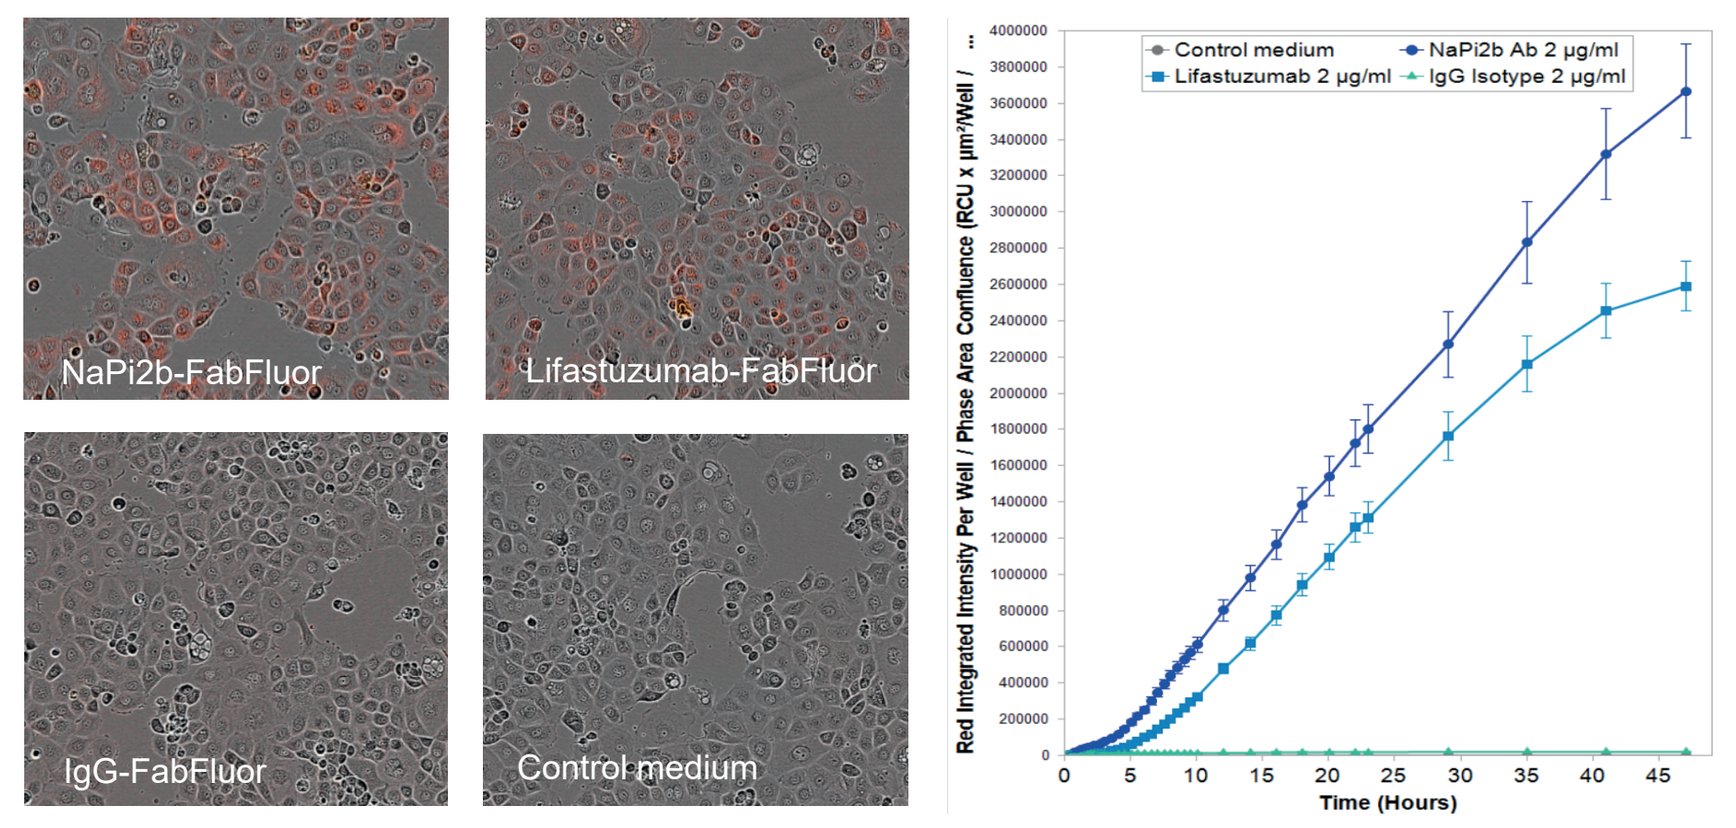

A
B
NaPi2b-FabFluor
Lifastuzumab-FabFluor
IgG-FabFluor
Control medium
Control medium
NaPi2b Ab 2 µg/ml
Lifastuzumab 2 µg/ml
IgG Isotype 2 µg/ml
Time (Hours)

A. Internalization of Incucyte FabFluor labelled NaPi2b, lifastuzumab and IgG isotype control antibodies and control medium in OVCAR-3 cells. **B.** Time course data shows a rapid increase in red fluorescence intensity over time in cells incubated with labelled NaPi2b and lifastuzumab but not with isotype control. Side-by-side comparison shows increased uptake of NaPi2b Ab compared to lifastuzumab in OVCAR-3 cells.

Figure 4. Comparison of bystander activity in medium-transfer assay

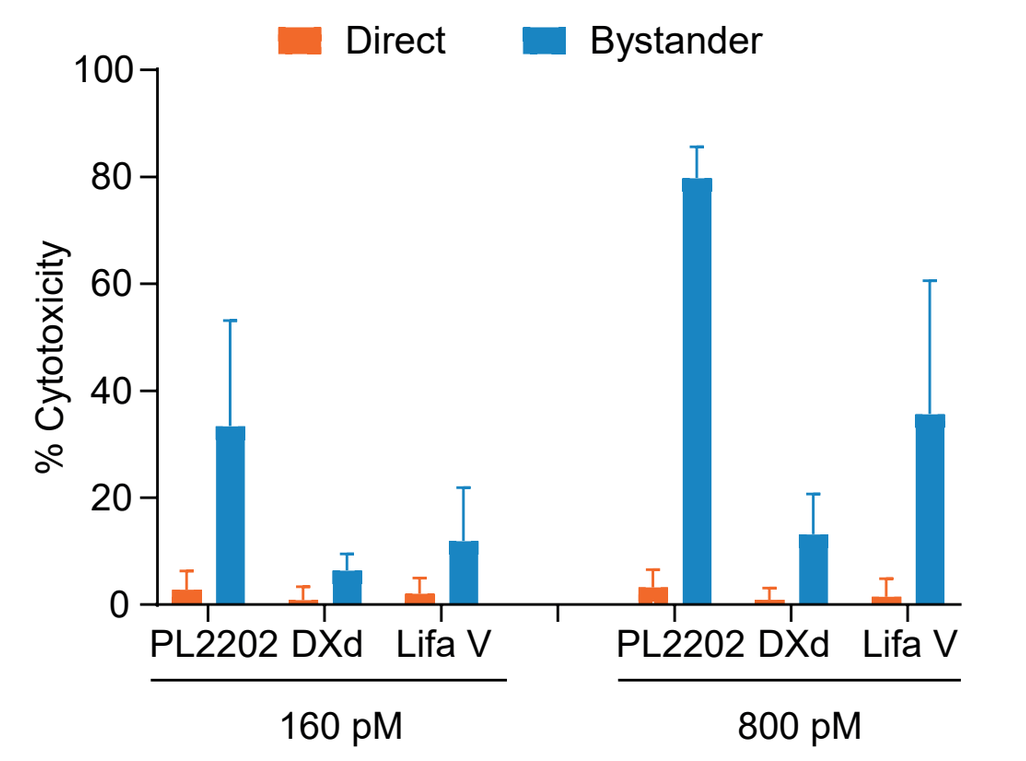

Direct
Bystander
% Cytotoxicity
PL2202 DXd Lifa V
PL2202 DXd Lifa V
160 pM
800 pM

At equivalent treatment concentrations, NaPi2b-PL2202 exhibits stronger bystander activity than NaPi2b-DXd and lifastuzumab vedotin. Bystander activity, as shown by increased cytotoxicity to NaPi2b– (KB) cells when conditioned medium from NaPi2b+ (IGROV-1) cells is transferred onto NaPi2b– (KB) cells, was assessed in a medium transfer cytotoxicity assay, 5 days incubation with ADCs, by CellTiter-Glo luminescent assay. Direct= NaPi2b- (KB) directly incubated with ADCs, Bystander= NaPi2b- (KB) incubated with conditioned medium from NaPi2b+ (IGROV-1) cells.

Figure 5. *In vivo* anti-tumor activity in the G402 renal leiomyoblastoma xenograft model

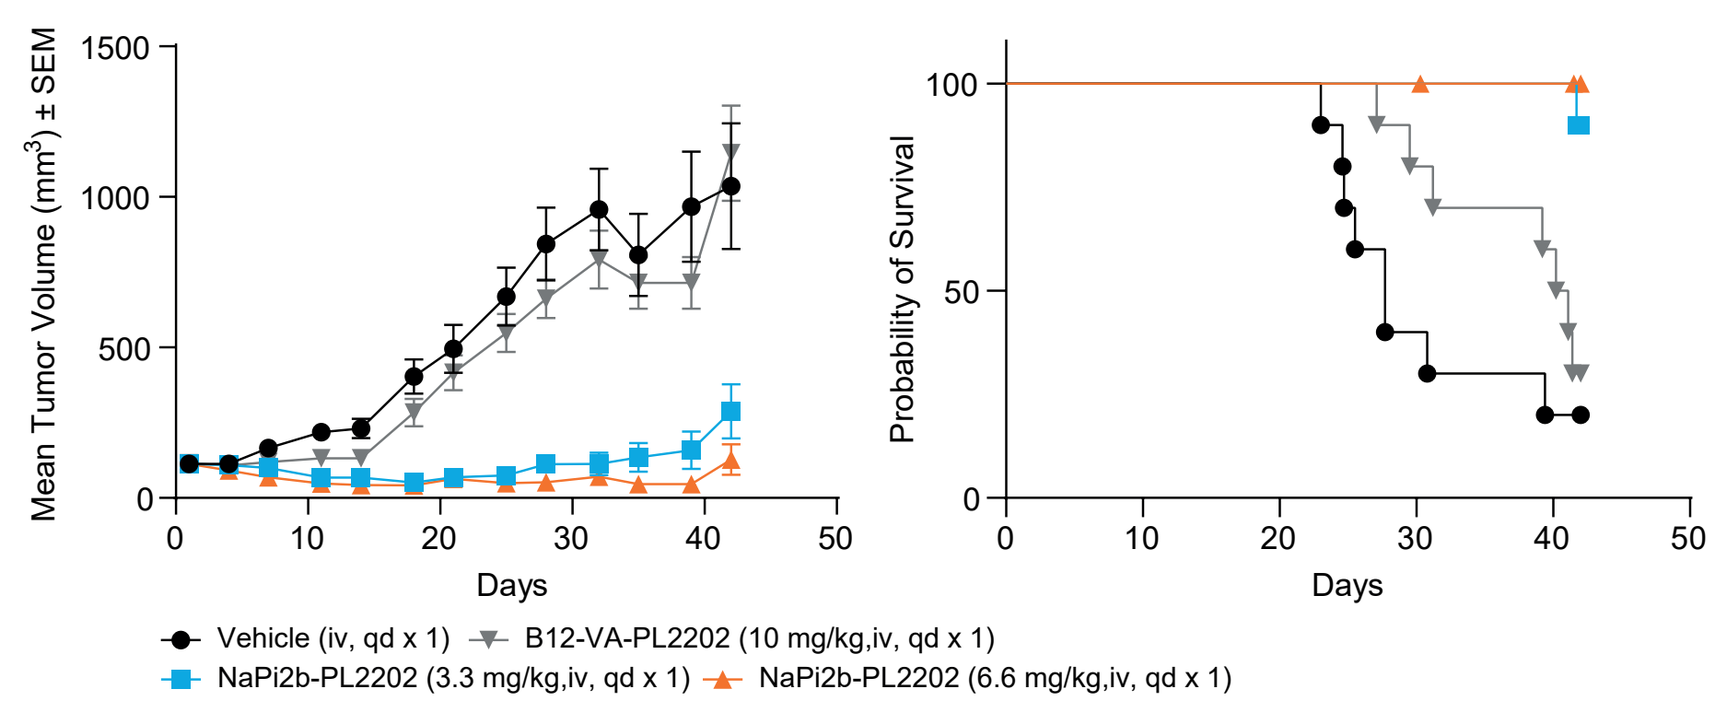

A
B
Mean Tumor Volume (mm³) ± SEM
Probability of Survival
Days
Vehicle (iv, qd x 1)
B12-VA-PL2202 (10 mg/kg,iv, qd x 1)
NaPi2b-PL2202 (3.3 mg/kg,iv, qd x 1)
NaPi2b-PL2202 (6.6 mg/kg,iv, qd x 1)

C

ADCs	PR	CR	TFS
NaPi2b-PL2022, 3.3 mg/kg	0/10	1/10	1/10
NaPi2b-PL2022, 6.6 mg/kg	0/10	1/10	1/10
B12-VA-PL2202, 10 mg/kg	0/10	0/10	0/10

D

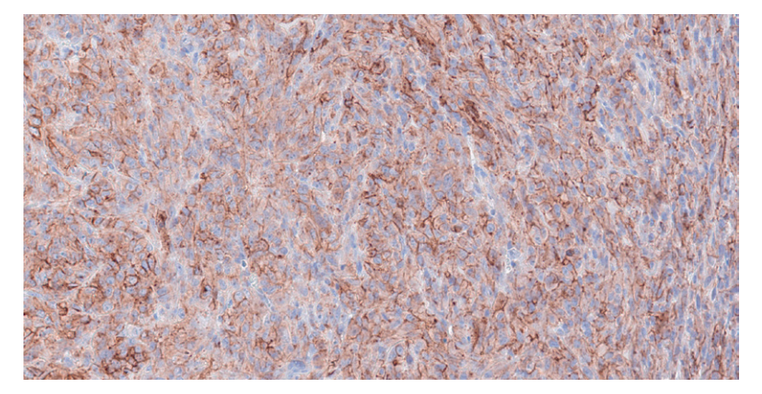

A. NaPi2b-PL2202 and B12-VA-PL2202 isotype-control ADC were administered i.v. (day 1) to treatment groups of 10 mice. A vehicle-treated group served as control. **B.** Kaplan-Meier analysis of survival. **C.** Response summary. PR, partial response; CR, complete response; TFS, tumor-free survivors. **D.** Representative scan of FFPE G402 tumor section stained for NaPi2b by IHC.

Figure 6. *In vivo* anti-tumor activity in the OVCAR-3 ovarian xenograft model

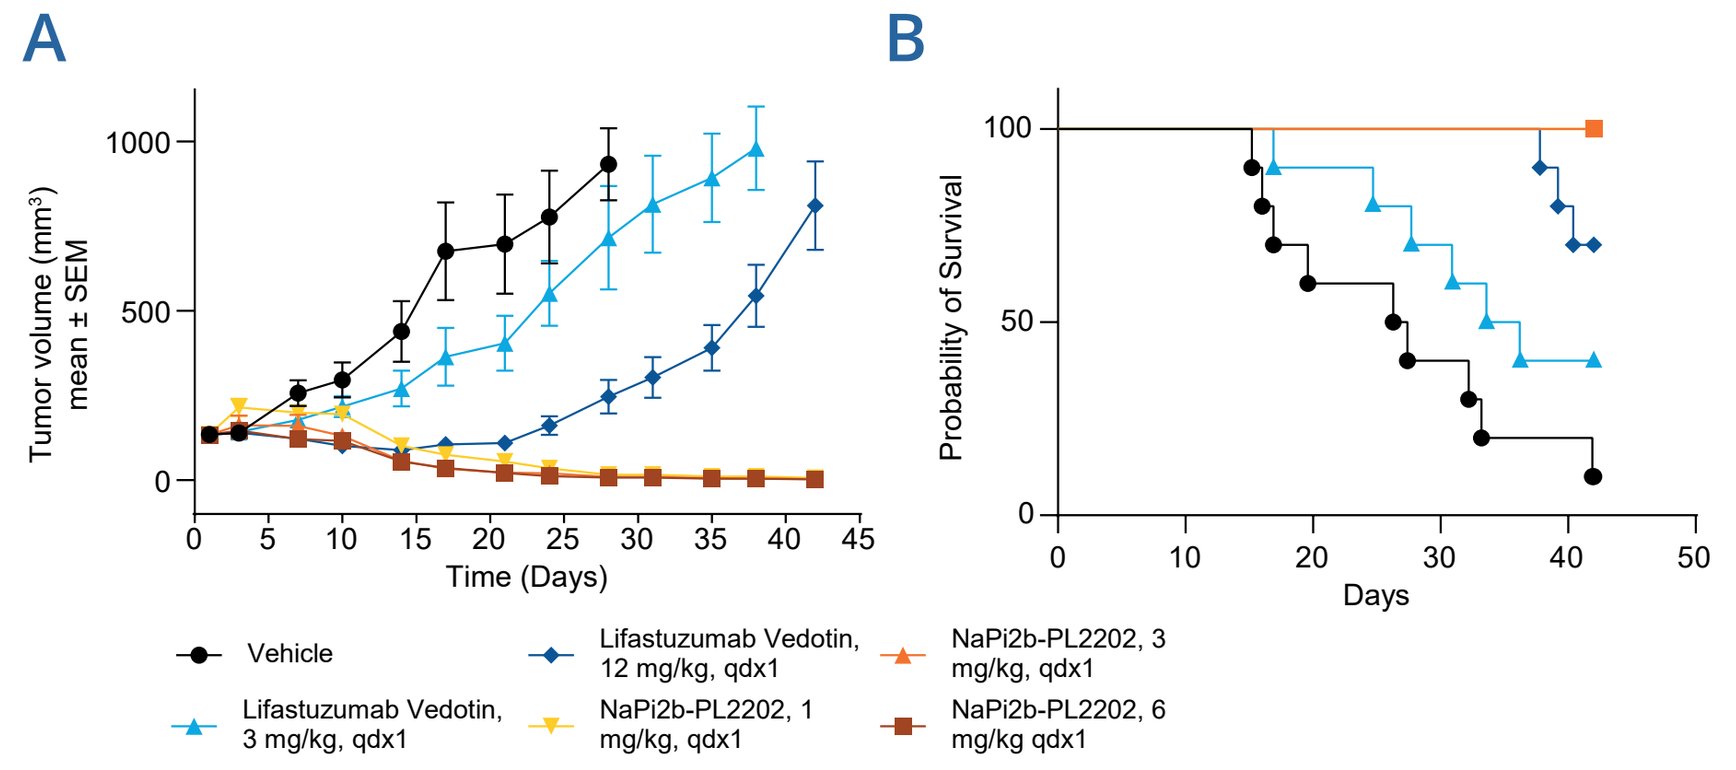

A
B
Tumor volume (mm³) mean ± SEM
Time (Days)
Probability of Survival
Days
Vehicle
Lifastuzumab Vedotin, 3 mg/kg, qdx1
Lifastuzumab Vedotin, 12 mg/kg, qdx1
NaPi2b-PL2202, 1 mg/kg, qdx1
NaPi2b-PL2202, 3 mg/kg, qdx1
NaPi2b-PL2202, 6 mg/kg qdx1

C

ADCs	PR	CR	TFS
NaPi2b-PL2022, 6 mg/kg	0/10	10/10	10/10
NaPi2b-PL2022, 3 mg/kg	1/10	9/10	9/10
NaPi2b-PL2022, 1 mg/kg	1/10	9/10	9/10
lifastuzumab vedo, 12 mg/kg	3/10	0/10	0/10
lifastuzumab vedo, 3 mg/kg	0/10	0/10	0/10

D

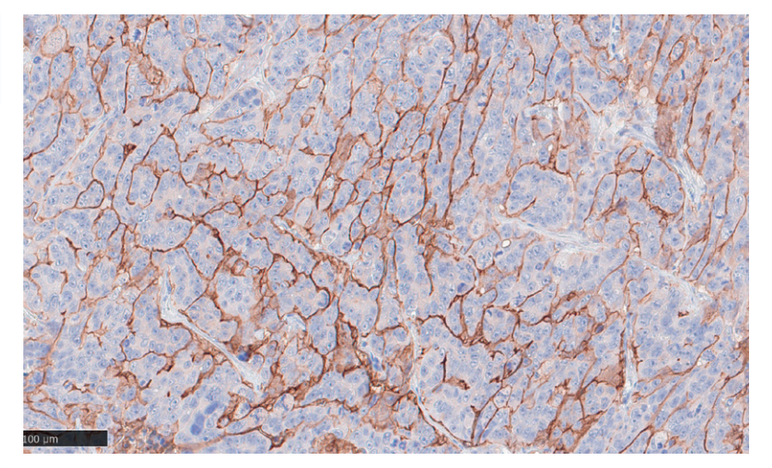

A. NaPi2b ADCs were administered i.v. (day 1) to treatment groups of 10 mice. A vehicle-treated group served as control. **B.** Kaplan-Meier analysis of survival. **C.** Response summary. PR, partial response; CR, complete response; TFS, tumor-free survivors. **D.** Representative scan of FFPE OVCAR-3 tumor section stained for NaPi2b by IHC.

Figure 7. *In vivo* anti-tumor activity in the H441 lung-papillary adenocarcinoma xenograft model

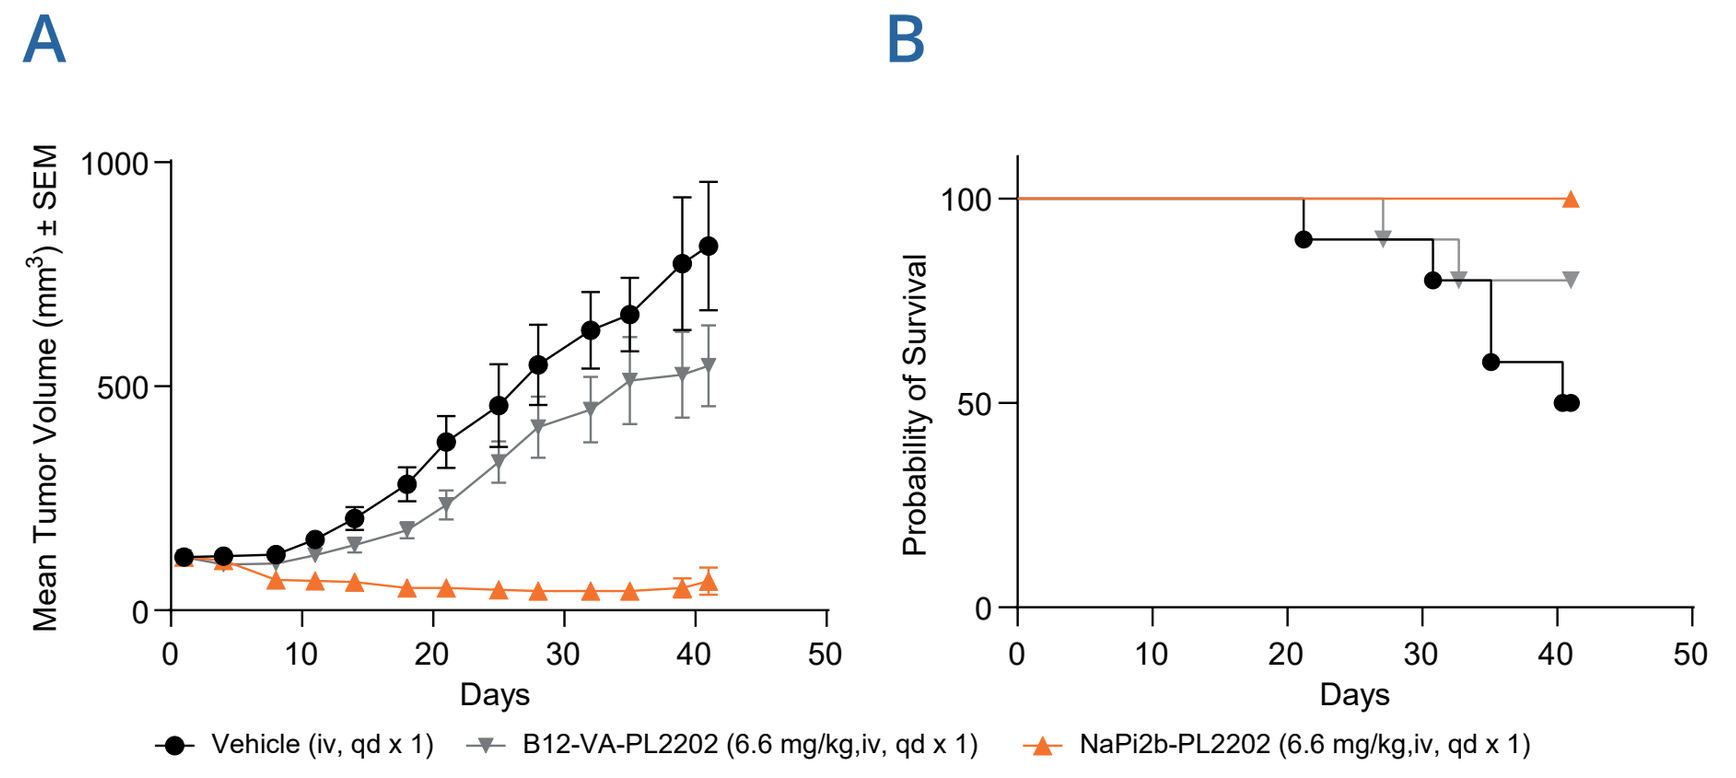

A
B
Mean Tumor Volume (mm³) ± SEM
Probability of Survival
Days
Vehicle (iv, qd x 1)
B12-VA-PL2202 (6.6 mg/kg,iv, qd x 1)
NaPi2b-PL2202 (6.6 mg/kg,iv, qd x 1)

C

ADCs	PR	CR	TFS
NaPi2b-PL2022, 6.6 mg/kg	9/10	0/10	0/10
B12-VA-PL2022, 10 mg/kg	0/10	0/10	0/10

A. NaPi2b-PL2202 and B12-VA-PL2202 isotype-control ADC were administered i.v. (day 1) to treatment groups of 10 mice. A vehicle-treated group served as control. **B.** Kaplan-Meier analysis of survival. **C.** Response summary. PR, partial response; CR, complete response; TFS, tumor-free survivors.

Figure 8. NaPi2b-PL2202 mean serum concentration-time profiles in rats and cynomolgus monkeys

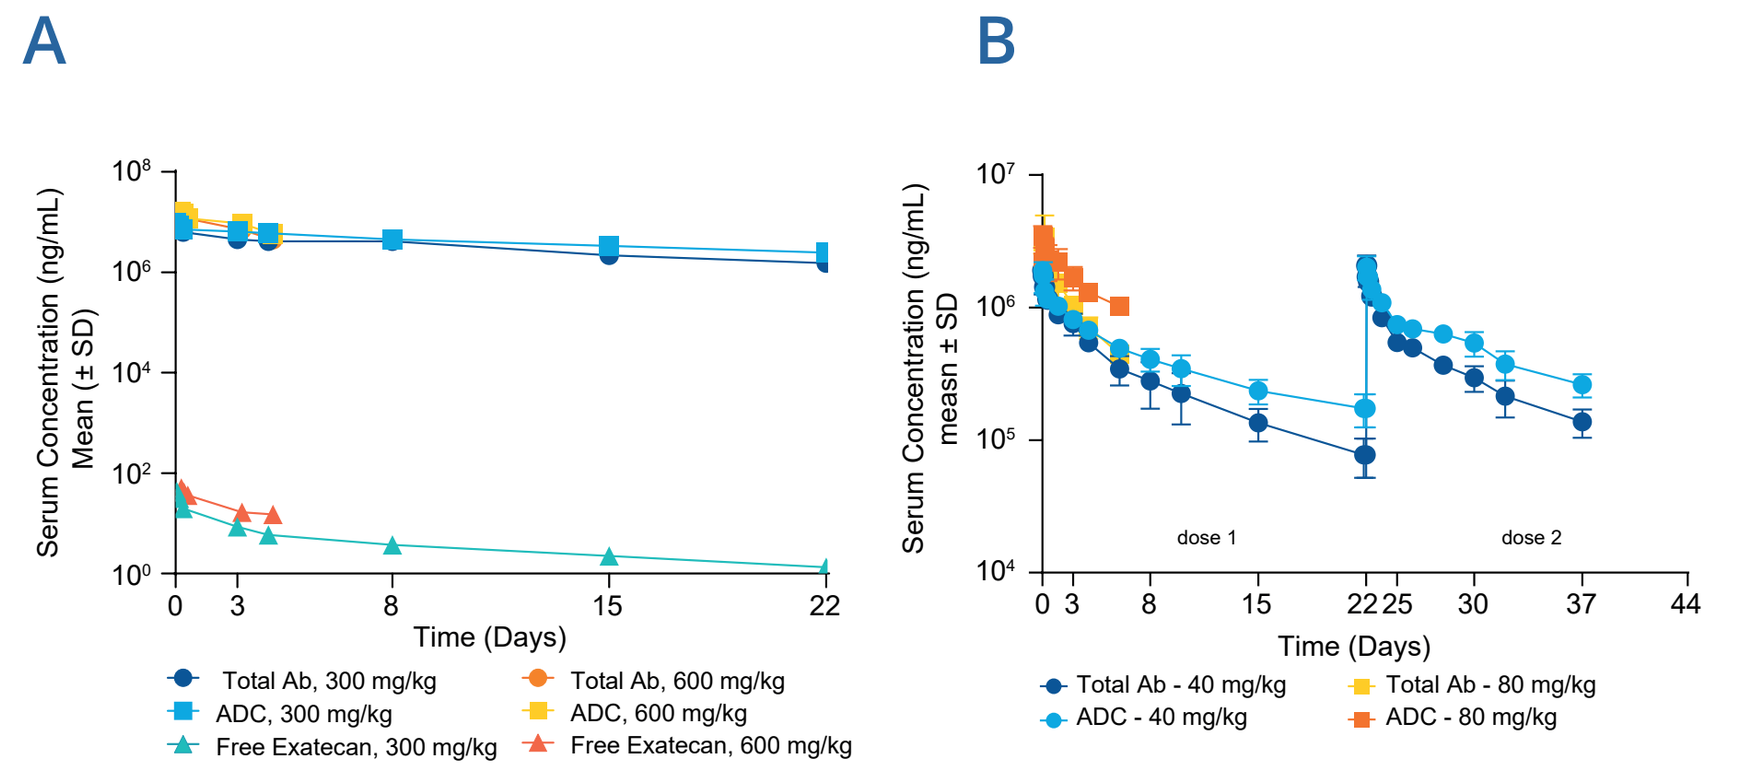

A
B
Serum Concentration (ng/mL) Mean (± SD)
Time (Days)
Total Ab, 300 mg/kg
ADC, 300 mg/kg
Free Exatecan, 300 mg/kg
Total Ab, 600 mg/kg
ADC, 600 mg/kg
Free Exatecan, 600 mg/kg
Serum Concentration (ng/mL) mean ± SD
dose 1
dose 2
Total Ab - 40 mg/kg
ADC - 40 mg/kg
Total Ab - 80 mg/kg
ADC - 80 mg/kg

A. PK following a single IV dose of NaPi2b-PL2202 to rats (n=3/group). Linear TK with a t½ of 12.9-15 days (based on data from the ADC assay); free exatecan detectable throughout the dosing period with apparent t½ of 9.5 days. **B.** PK following one or two doses of NaPi2b-PL2202 three weeks apart via IV infusion (n=3/group) in cynomolgus monkeys. Linear TK with a t½ of ~7-10 days (based on data from the ADC assay).

NaPi2b-PL2202 is tolerated in both rats and cynomolgus monkeys

- Both rats and cynomolgus monkeys are pharmacologically relevant species for NaPi2b-PL2202. In a single dose study in male rats, NaPi2b-PL2202 was tolerated up to 300 mg/kg IV with minimal clinical pathology and histopathology findings.
- In a toxicity study in male cynomolgus monkeys, NaPi2b-PL2202 was well tolerated up to 40 mg/kg IV given on Day 1 and 22 with minimal clinical pathology and histopathology findings.
- At a higher dose level of 80 mg/kg the dose limiting toxicity was degeneration/regeneration in the gastrointestinal tract, a known class effect of topoisomerase inhibitors such as exatecan.

Figure 9. NaPi2b membranous expression in a panel of NSCLC, ovarian and endometrial cancer patient samples

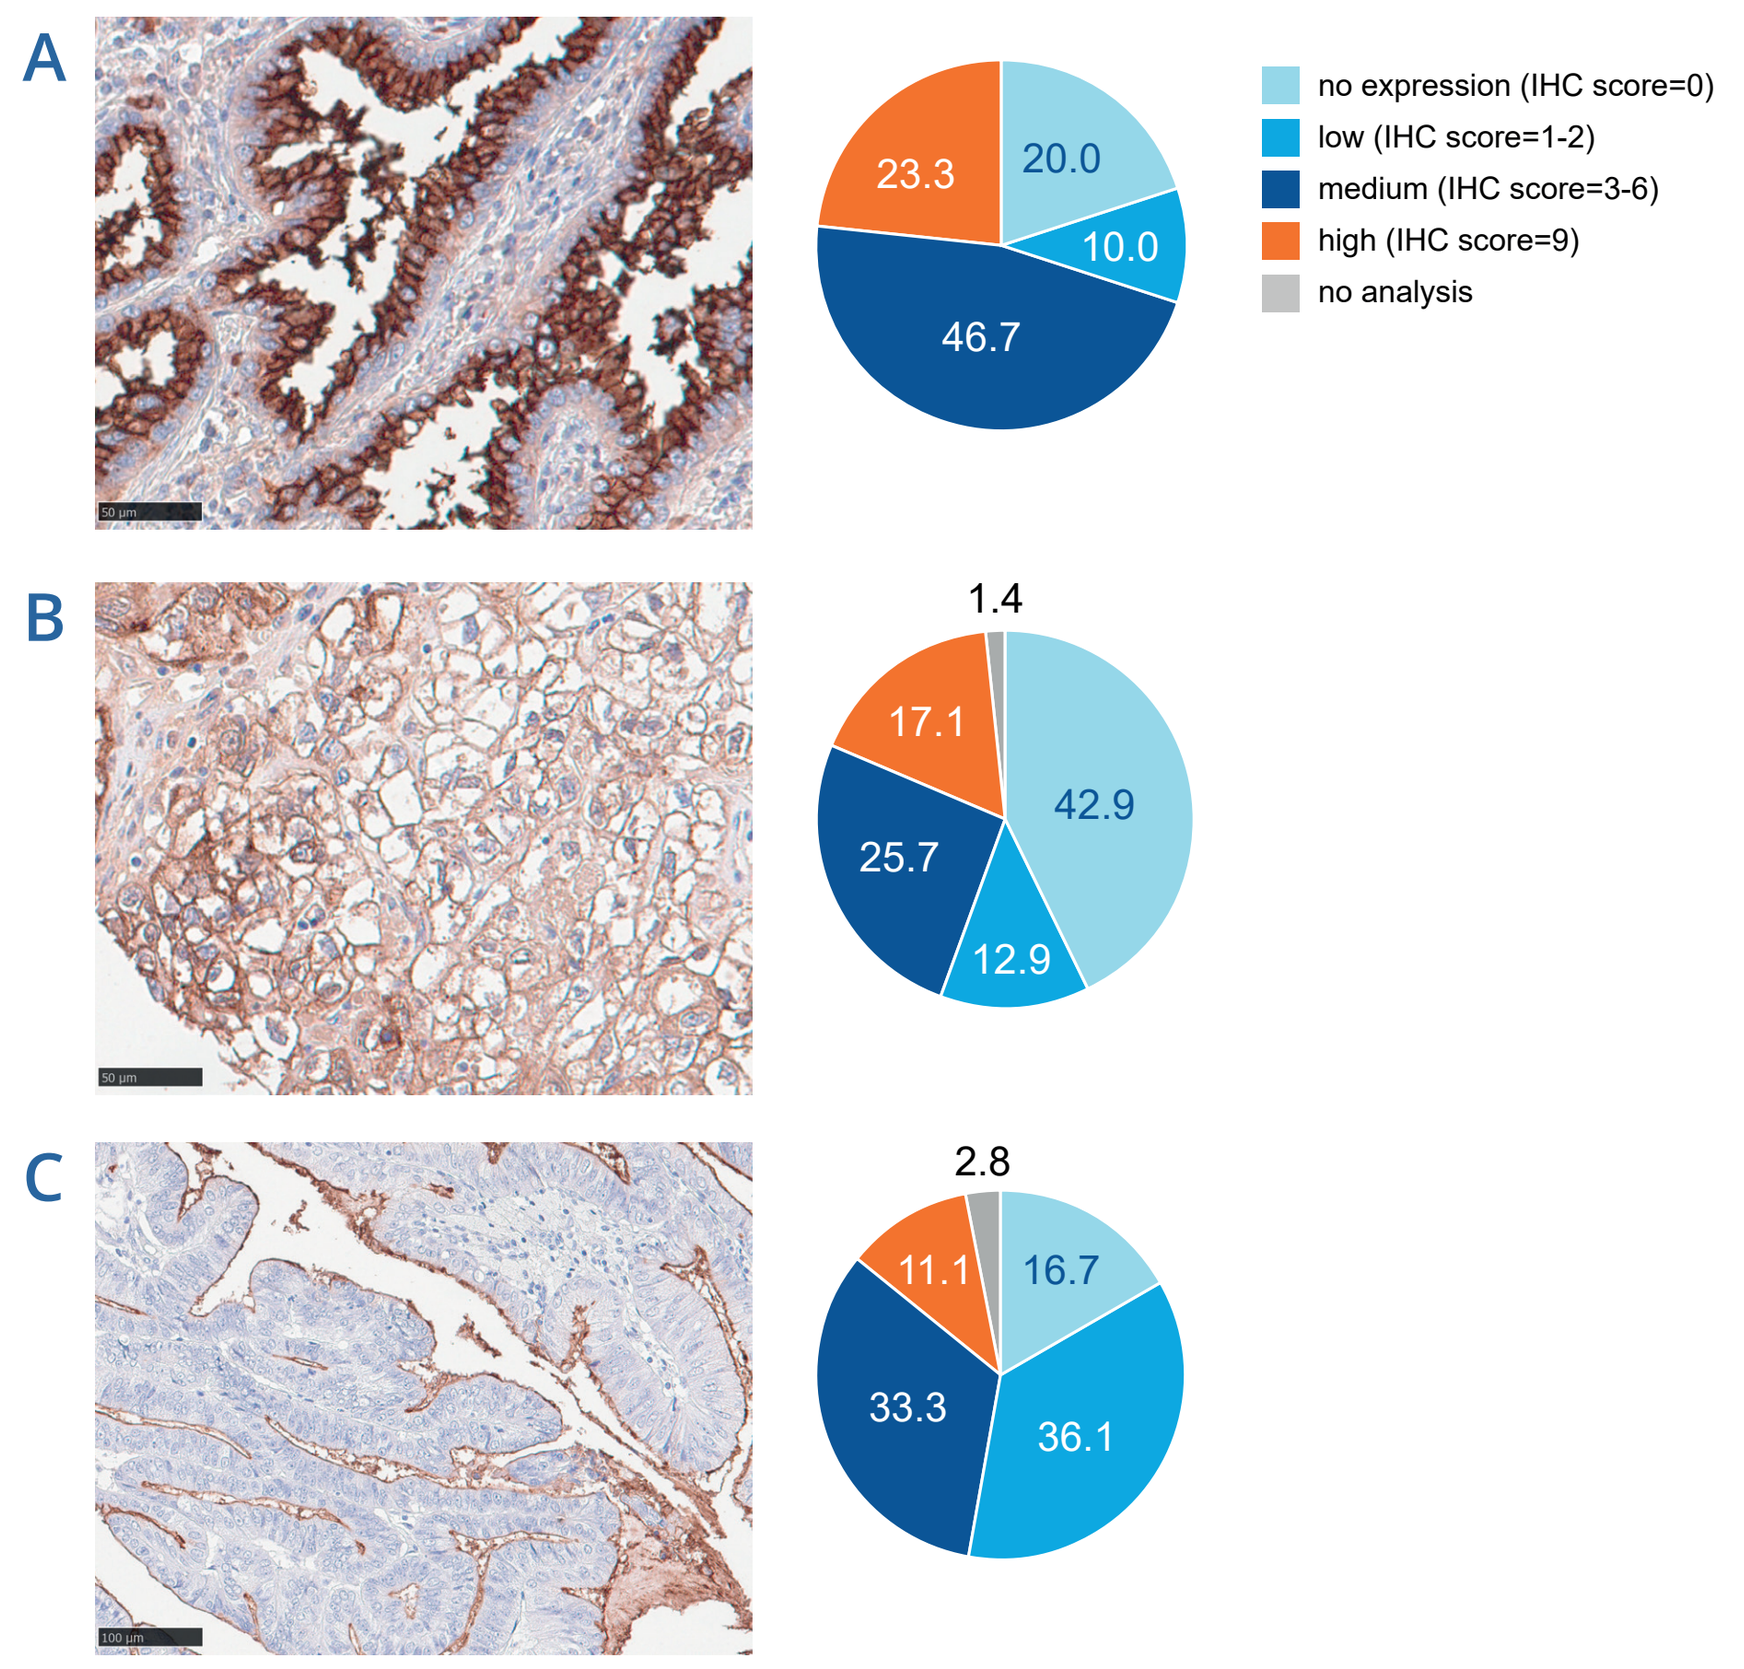

high expression IHC score=9
NaPi2b membrane expression (%)
A
B
C
no expression (IHC score=0)
low (IHC score=1-2)
medium (IHC score=3-6)
high (IHC score=9)
no analysis
23.3
20.0
10.0
46.7
1.4
17.1
42.9
25.7
12.9
2.8
11.1
16.7
33.3
36.1

Representative images of Napi2b staining showing high membrane expression (IHC score=9). Pie charts indicate % positivity for each IHC-score across cancer patients, **A.** NSCLC (n=30), **B.** Ovarian (n=70), **C.** Endometrial (n=45).