POSTER 3122

Preclinical development of a novel camptothecin-based antibody-drug conjugate targeting solid tumors expressing Claudin-6

Nicola Tsang, Nicolas Veillard, Elizabeth Horsley, Karin Havenith, Narinder Janghra, Kristina Zeitseva, Cecile Oblette, Ian Kirby, Paul W. Hogg, Francesca Zammarchi, Lolke de Haan, Patrick H. van Berkel

ADC Therapeutics UK (Ltd), London, UK

ACKNOWLEDGEMENTS

In vivo studies: Charles River Discovery Research Services (USA) (mouse xenograft studies); Crown Bioscience (China) (mouse xenograft study) and LabCorp (UK) (cynomolgus monkey toxicity study).

REFERENCES

1. Lin, D., Guo Y., & Li Y. et al., Bioinformatics analysis reveals potential properties of human Claudin-6 regulation and function. Oncology Reports 2017 38(2): 875-885.
2. Reinhard, K., et al., An RNA vaccine drives expansion and efficacy of claudin-CAR-T cells against solid tumours. Science 2020 367: 446-453.
3. Kohmoto, T., et al., Claudin-6 is a single prognostic marker and functions as a tumour-promoting gene in a subgroup of intestinal type gastric cancer. Gastric Cancer 2020 23: 403-417.
4. McDermott, M.S.J., et al., Preclinical Efficacy of the Antibody–Drug Conjugate CLDN6–23-ADC for the Treatment of CLDN6-Positive Solid Tumors. Clinical Cancer Research 2023 29:2131-43.

MATERIALS AND METHODS

Cytotoxicity of GB01-VA-PL2202, AB3-7-MMAE[4], AB3-7-VA-PL2202, and isotype control ADCs (B12-VA-PL2202 and B12-MMAE), was determined by the CellTiter 96® AQueous One Solution Cell Proliferation or CellTitreGlo® assays (Promega). Antibody cell binding data was analysed by flow cytometry. Internalization of the Claudin-6 antibody (GB01, AB3-7), and IgG isotype control was performed by Incucyte®. FabFluor labelled GB01, AB3-7 or IgG1 isotype control Ab (1 µg/mL) were used in PA-1 and KB cells. HD phase and red fluorescence images (10x) were captured. Images of cells treated with FabFluor- GB01 or FabFluor-AB3-7 display red, cytosolic fluorescence illustrating internalization. Comparison of bystander activity was determined by a medium transfer method using the CellTiterGlo® assays (Promega).

In *in vivo* studies, GB01-VA-PL2202 was administered intravenously (i.v.) as a single dose or AB3-7-MMAE[4] was administrated intravenously (i.v.) as three single doses weekly to athymic nude or SCID mice containing PA-1 or OV90 xenografts.

PK was evaluated in cynomolgus monkeys (n=3/group) following one or two doses of GB01-VA-PL2202 three weeks apart via IV infusion. Serum exposure to total (conjugated and unconjugated Ab) and exatecan conjugated Ab determined by ELISA/ECLIA and to free Exatecan determined by LC-MS/MS. PK parameters determined by non-compartmental analysis (NCA).

Analysis of Claudin-6 membranous expression on human TMAs from patients with testicular cancer, ovarian cancer and NSCLC was performed by IHC using a commercial human Claudin-6 antibody.

Disclosures

All authors are current or former employees of ADC Therapeutics that may hold stock or stock options.

CONCLUSIONS

- *In vitro*, GB01-VA-PL2202 demonstrated potent and target-mediated cytotoxicity in a panel of Claudin-6-positive solid tumor cell lines. A PL2202-based ADC based on AB3-7 was much less potent *in vitro* compared to GB01-VA-PL2202.
- The Claudin-6-specific mAb GB01 was efficiently internalized in lysosomes by Claudin-6-expressing PA-1 cells, showing increased intracellular uptake compared to AB3-7 Ab.
- GB01-VA-PL2202 showed bystander killing activity of Claudin-6-negative KB cells incubated with conditioned medium from GB01-VA-PL2202-treated PA-1 cells.
- *In vivo*, GB01-VA-PL2202 showed strong antitumor activity against PA-1 and OV-90 cancer xenograft models, highlighting its target-mediated antitumor activity. GB01-VA-PL2202 showed higher tumor regression and response compared to AB3-7-MMAE[4].
- GB01-VA-PL2202 was well tolerated up to 40 mg/kg IV following two doses three weeks apart via IV infusion in male cynomolgus monkeys.
- TK profiles for GB01-VA-PL2202 ADC and total antibody were generally comparable, with ADC concentrations slightly higher than total Ab concentrations. Linear TK with a t½ of 8.5-10 days (based on data from the ADC assay). Free exatecan was detectable throughout the dosing period with apparent t½ of 5.5-8 days.
- Membranous expression of Claudin-6 was confirmed by IHC in a high proportion of testicular, NSCLC and ovarian cancers.
- In conclusion, GB01-VA-PL2202 demonstrated potent and specific *in vitro* and *in vivo* anti-tumor activity while it was stable and well tolerated in cynomolgus monkeys, warranting progression of the ADC into clinical trials.

INTRODUCTION

- Claudin-6 is a member of the claudin family and consists of four transmembrane helices, two extracellular loops, and an amino- and carboxyl-terminal tail with a PDZ-binding motif in the cytoplasm (Figure 1)[1].
- It is involved in the formation of tight junctions and is expressed in developing human epithelial structures during early-to-mid gestation while expression in adult tissues is mostly absent[1].
- Claudin-6 expression is upregulated in ovarian cancer, testicular cancer, endometrial cancer, non-small cell lung cancer (NSCLC)[2] and gastric cancer[3].

Figure 1. Claudin-6 structure

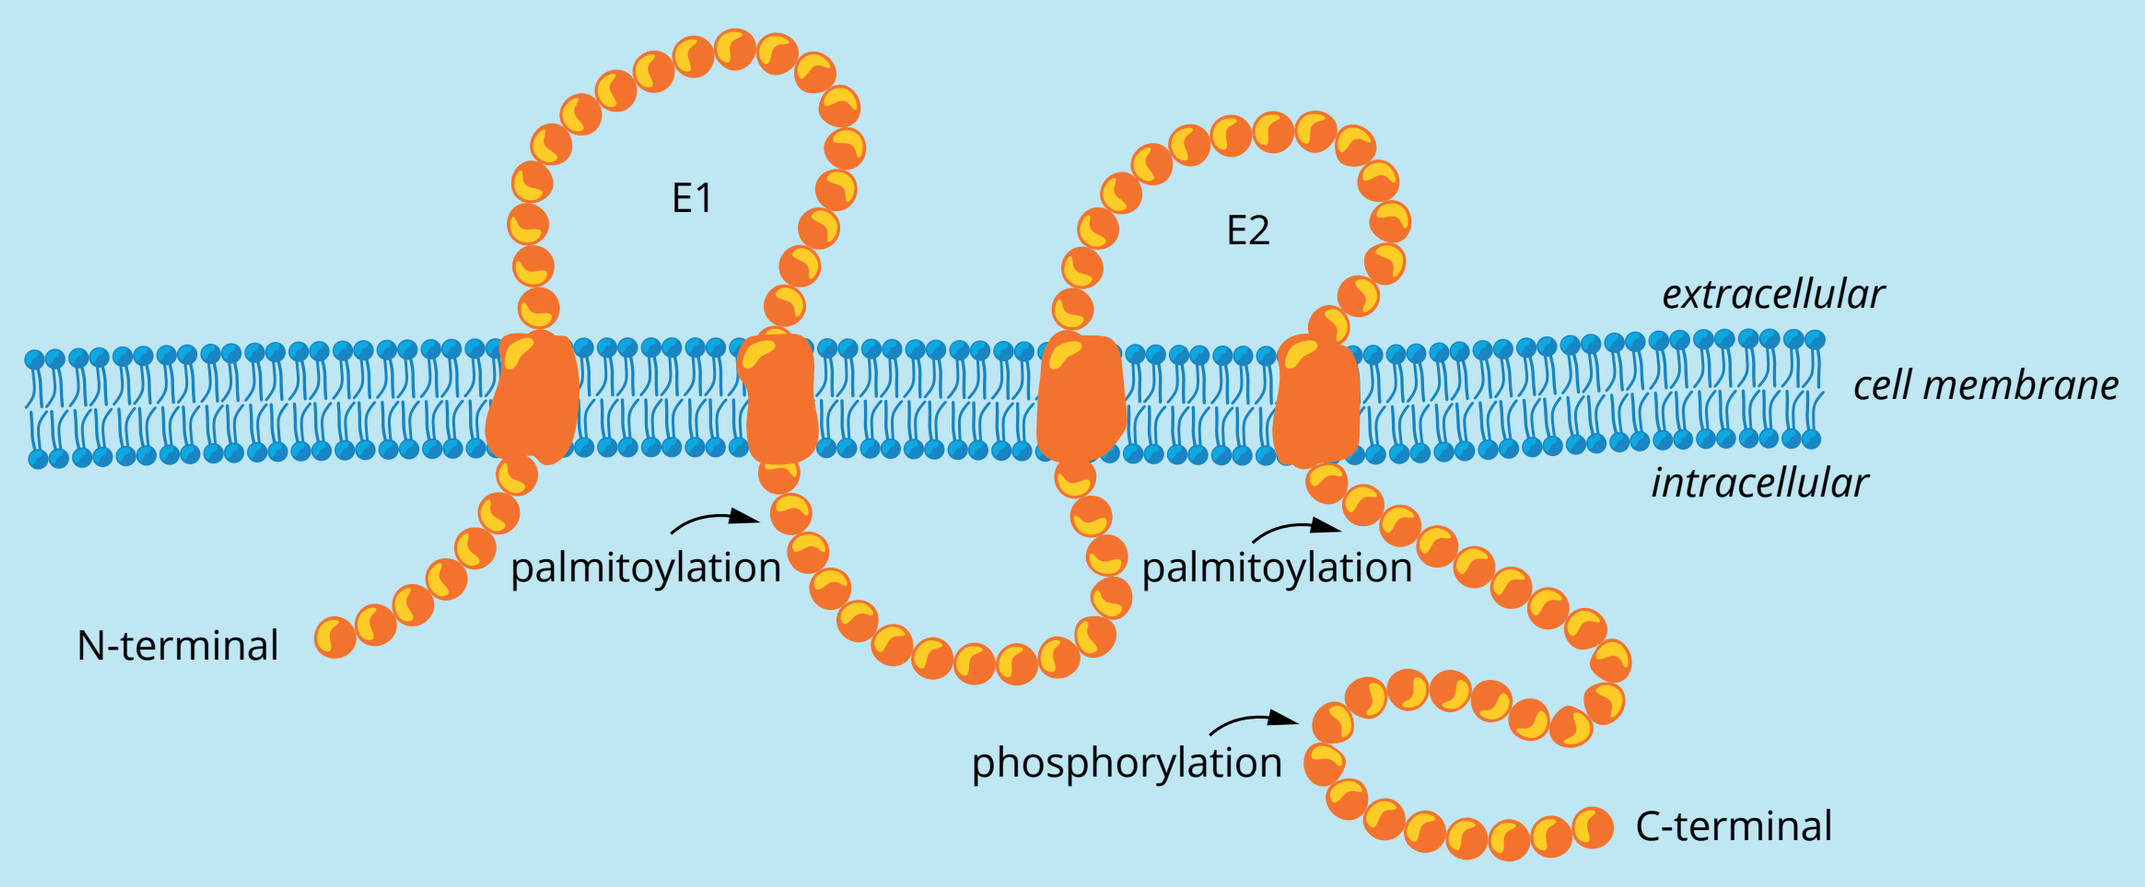
E1
E2
extracellular
cell membrane
intracellular
N-terminal
palmitoylation
palmitoylation
phosphorylation
C-terminal

- GB01-VA-PL2202 is a novel ADC, composed of a humanized IgG1, Fc-silenced antibody against Claudin-6 to which the exatecan-containing payload PL2202 has been site-specifically conjugated with a DAR of 6 (Figure 2).
- This was achieved by introduction of the Cys228Val mutation in the hinge of the heavy chain.
- FcγR-mediated effector function of the ADC was abrogated via mutations in the Fc portion of the antibody.

AIM OF THE STUDY

- The purpose of this study was to determine the *in vitro* cytotoxicity and *in vivo* efficacy of GB01-VA-PL2202 in xenograft models as well as its toxicity and pharmacokinetic (PK) profile in cynomolgus monkeys.
- We also characterized the *in vitro* mechanism of action, such as internalization, bystander activity and determined Claudin-6 expression by immunohistochemistry (IHC) in human tumor specimens of ovary, testicular and NSCLC.

RESULTS

Figure 2. GB01-VA-PL2202

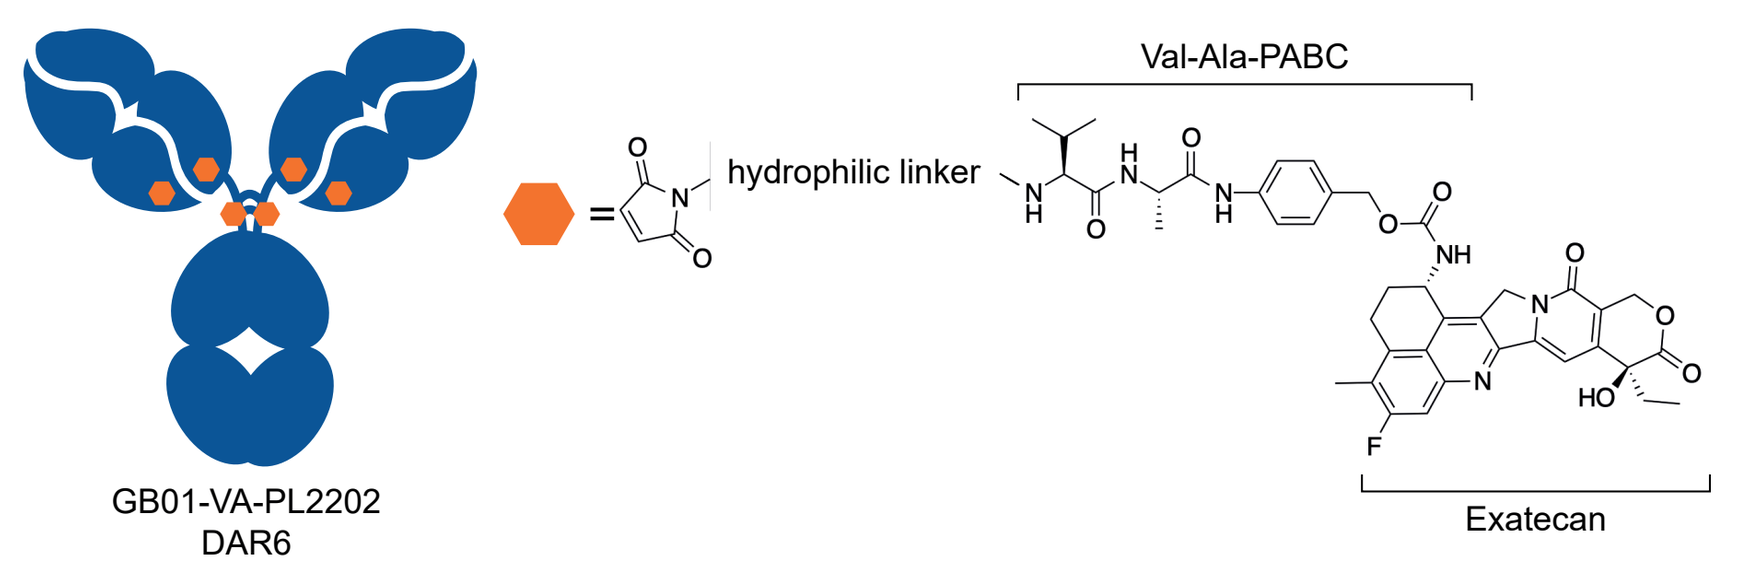
GB01-VA-PL2202
DAR6
hydrophilic linker
Val-Ala-PABC
Exatecan

Table 1. Claudin-6 Ab cell binding and *in vitro* cytotoxicity

IC_{50} for Cytotoxicity (pM) in Cell Lines with Differential Claudin-6 Expression							
ADC	PA-1++++	NTERA-2+++	NUGC-3+++	BeWo++	OVCAR3++	FU-97+	KB-
GB01-VA-PL2202	308	71.1	2,380	20,894	822	1,865	399,054
B12-VA-PL2202	11,027	4,578	26,400	49,046	14,790	66,503	364,040
AB3-7-MMAE	370.9	161.5	3,055	397.2	6,501	1,218	98,294
B12-MMAE	6,015	37,859	129,395	7,680	103,339	10,610	133,281

Claudin-6 antibody (GB01) binding to cells and *in vitro* cytotoxicity (IC_{50}) of GB01-VA-PL2202, AB3-7-MMAE[4] and isotype control ADCs (B12-VA-PL2202 and B12-MMAE) in a panel of 7 cancer cell lines.

Figure 3. GB01 Ab cell binding and GB01-VA-PL2202 *in vitro* cytotoxicity

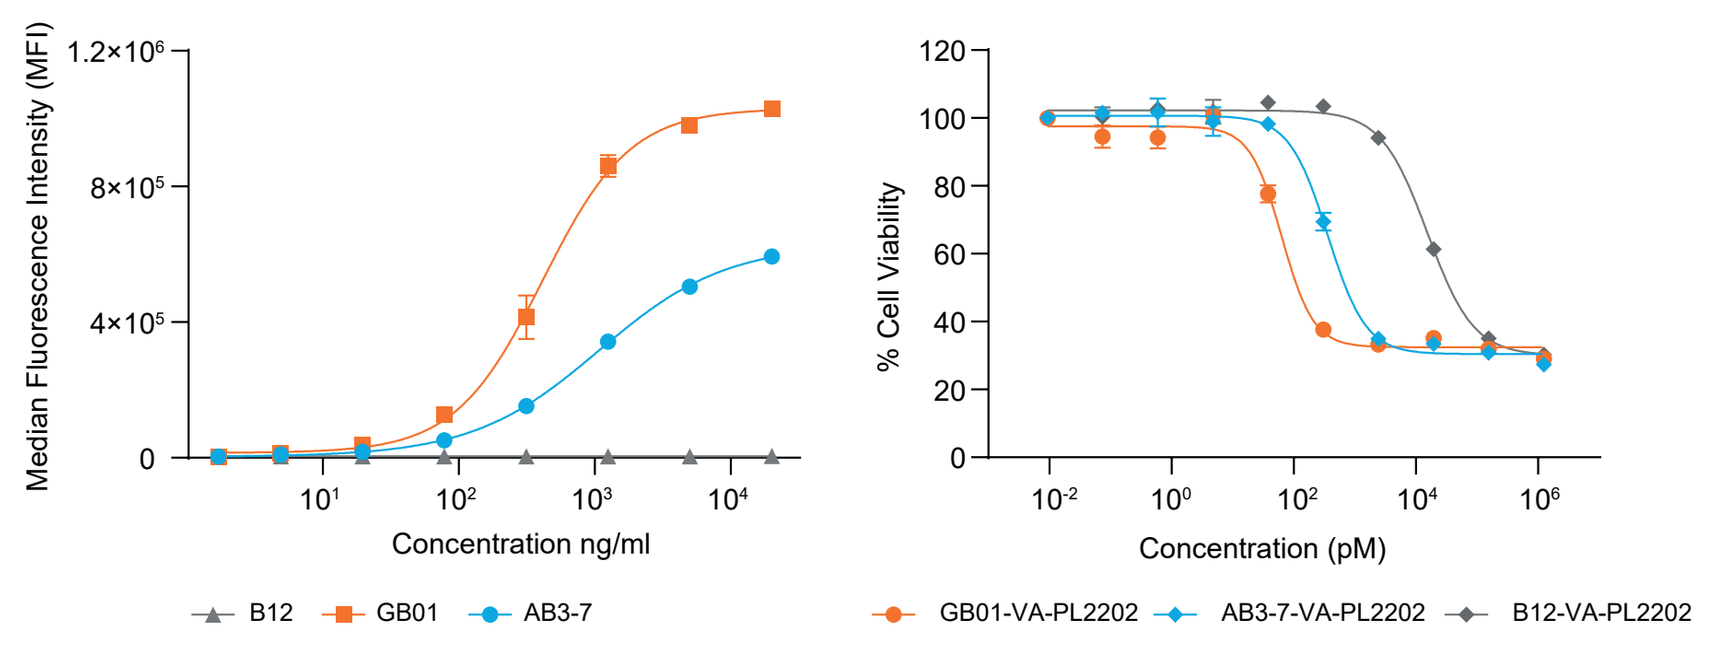
A
B
Median Fluorescence Intensity (MFI)
Concentration ng/ml
B12
GB01
AB3-7
% Cell Viability
Concentration (pM)
GB01-VA-PL2202
AB3-7-VA-PL2202
B12-VA-PL2202

Abs	EC_{50} pM
GB01	412
AB3-7	1070
B12 isotype IgG	no binding

ADCs	EC_{50} pM
GB01-VA-PL2022	63.67
AB3-7-VA-PL2022	359.9
B12-VA-PL2022	15496

A. Claudin-6-specific Ab GB01 shows increased binding to NTERA-2 cells compared to AB3-7 Ab[4]. **B.** Comparison of the cytotoxicity of GB01-VA-PL2202, AB3-7-VA-PL2202 and B12-VA-PL2202 (isotype control ADC) on NTERA-2 cells. Cytotoxicity was determined using a Cell Titer Aqueous One assay. GB01-VA-PL2202 exhibits increased *in vitro* cytotoxicity compared to AB3-7-VA-PL2202 (both DAR 6).

Figure 4. Antibody internalisation

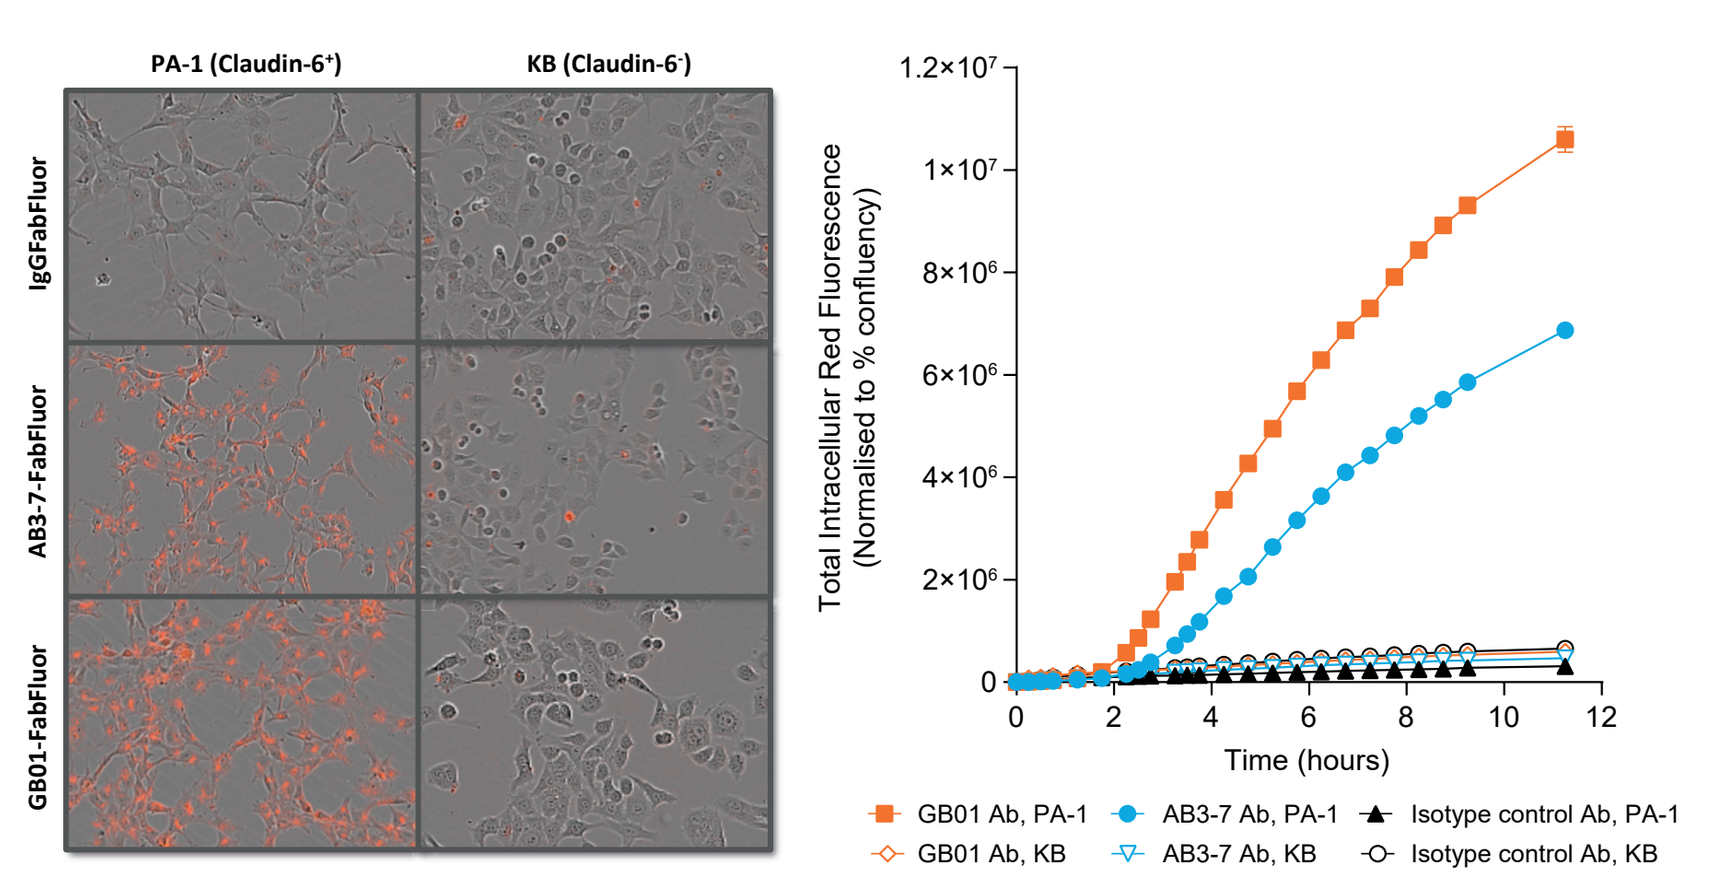
A
B
PA-1 (Claudin-6+)
KB (Claudin-6-)
IgGFabFluor
AB3-7-FabFluor
GB01-FabFluor
Total Intracellular Red Fluorescence (Normalised to % confluency)
Time (hours)
GB01 Ab, PA-1
GB01 Ab, KB
AB3-7 Ab, PA-1
AB3-7 Ab, KB
Isotype control Ab, PA-1
Isotype control Ab, KB

A. Internalisation of IncuCyte FabFluor-labelled GB01, AB3-7 and IgG isotype control antibodies in PA-1 (Claudin-6 +) and KB (Claudin-6 -) cells. **B.** Time course data shows a rapid increase in intracellular red fluorescence over time in cells treated with labelled GB01 antibody and AB3-7 but not with isotype control antibody. Internalisation of GB01 antibody in PA-1 cells is more rapid and increased relative to internalisation of AB3-7.

Figure 5. Bystander killing

A. Cytotoxicity in primary culture

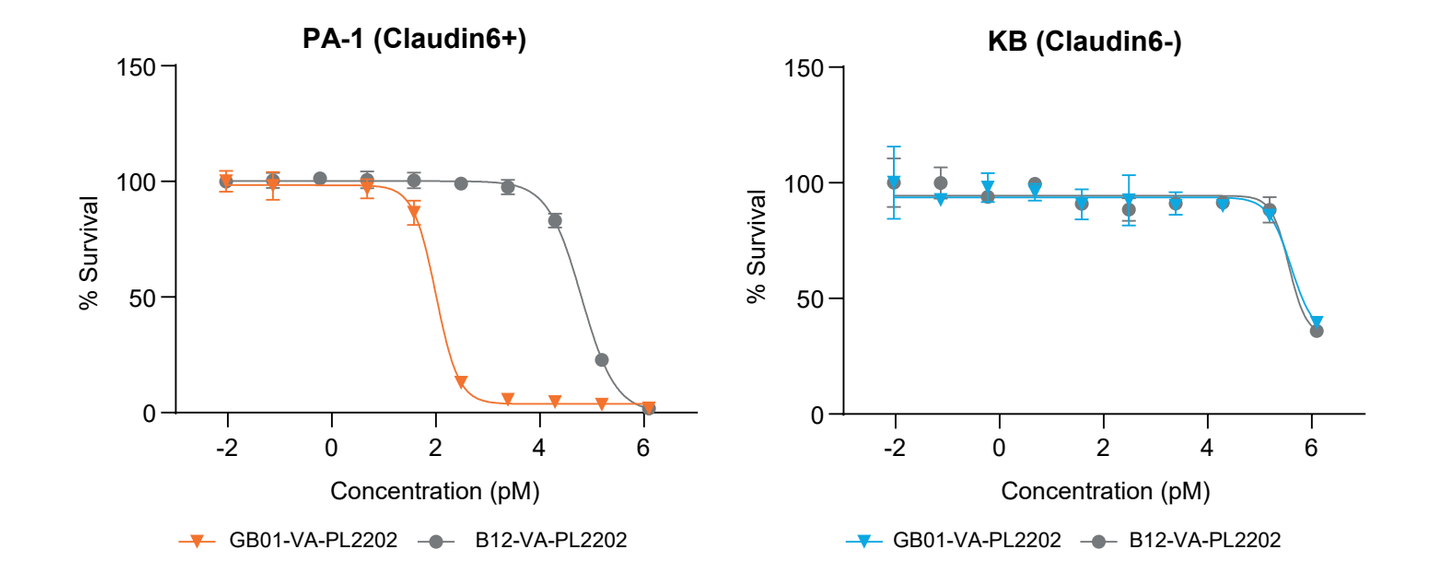
PA-1 (Claudin6+)
KB (Claudin6-)
% Survival
Concentration (pM)
GB01-VA-PL2202
B12-VA-PL2202

ADCs	PA-1 primay culture, IC_{50} (pM)	KB primary culture, IC_{50} (pM)
GB01-VA-PL2022	65	319,771
B12-VA-PL2022	70,318	518,196

B. Bystander cytotoxicity

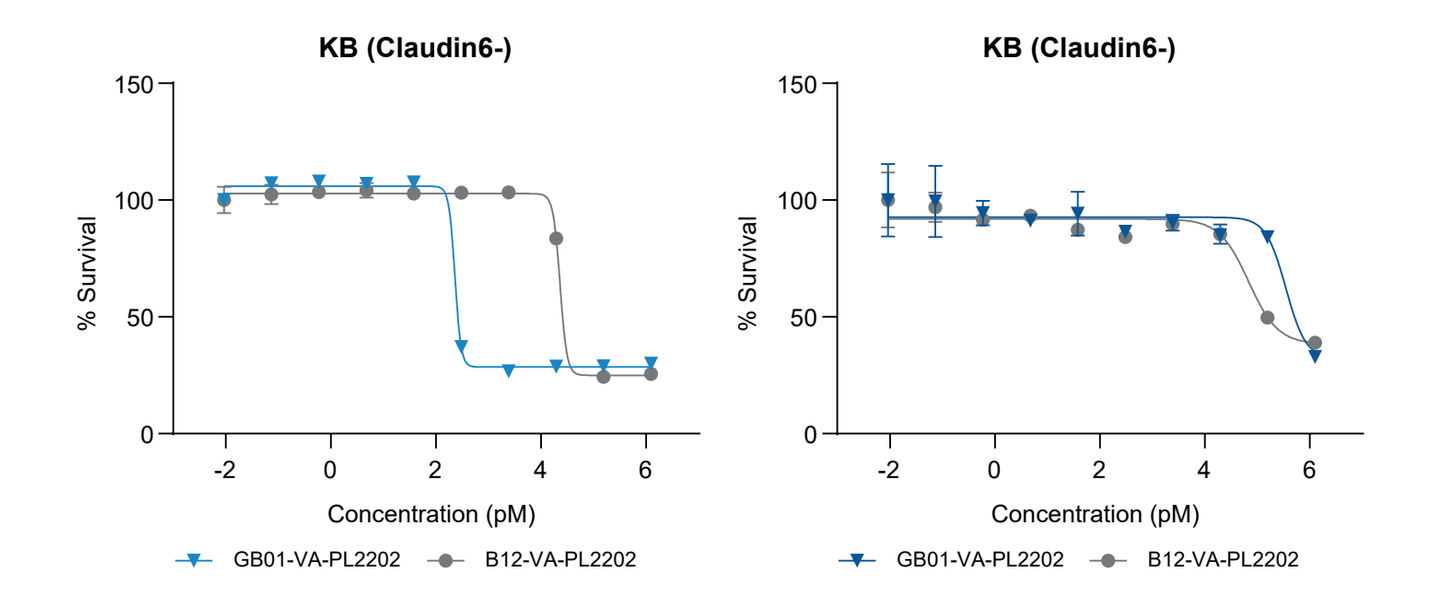
KB (Claudin6-)
KB (Claudin6-)
% Survival
Concentration (pM)
GB01-VA-PL2202
B12-VA-PL2202

ADCs	Bystander cytotox from PA-1, IC_{50} (pM)	Bystander cytotox from KB, IC_{50} (pM)
GB01-VA-PL2022	142	124,938
B12-VA-PL2022	25,337	38,699

Figures show a representative experiment of IC_{50} values for primary (**A**) and bystander (**B**) GB01-VA-PL2202 cytotoxicity in PA-1 and KB cell cultures.

Figure 6. *In vivo* anti-tumor activity in the PA-1 teratocarcinoma xenograft model

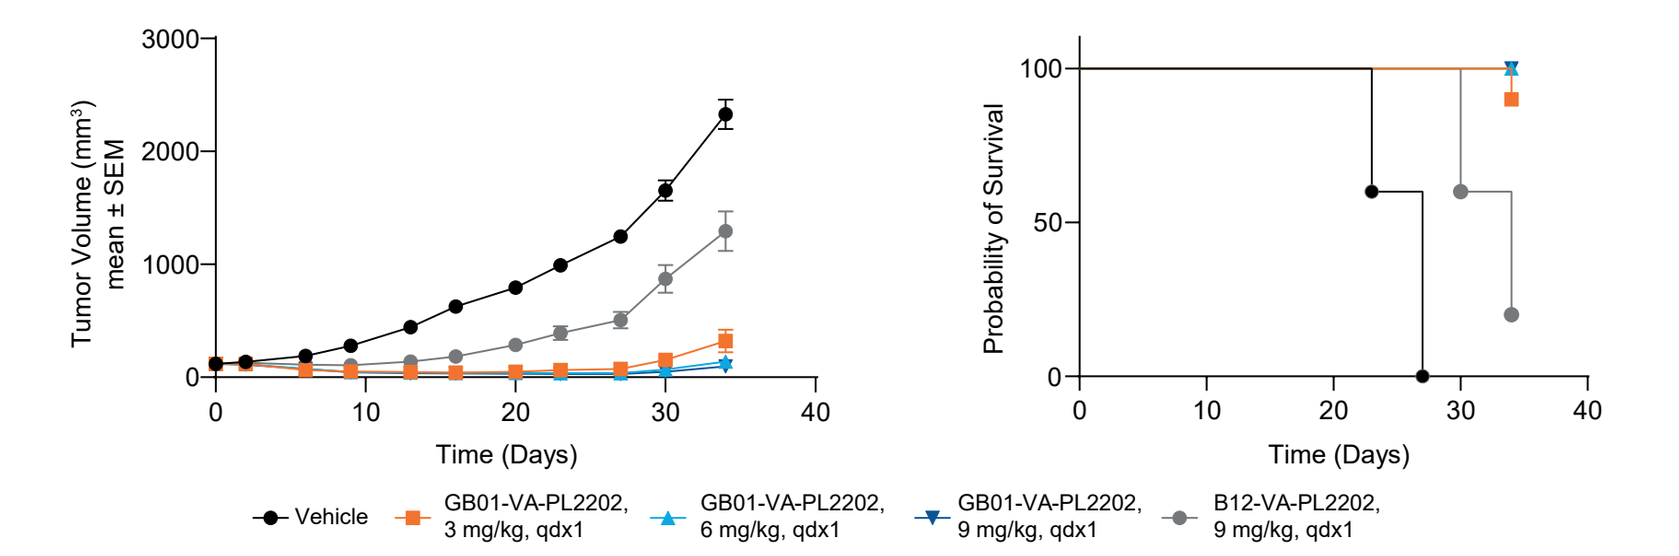
A
B
Tumor Volume (mm³) mean ± SEM
Time (Days)
Probability of Survival
Vehicle
GB01-VA-PL2202, 3 mg/kg, qdx1
GB01-VA-PL2202, 6 mg/kg, qdx1
GB01-VA-PL2202, 9 mg/kg, qdx1
B12-VA-PL2202, 9 mg/kg, qdx1

C

ADCs	PR	CR	TFS
3 mg/kg GB01-VA-PL2202	9/10	0/10	0/10
6 mg/kg GB01-VA-PL2202	10/10	0/10	0/10
9 mg/kg GB01-VA-PL2202	10/10	0/10	0/10
9 mg/kg B12-VA-PL2202	0/10	0/10	0/10

D

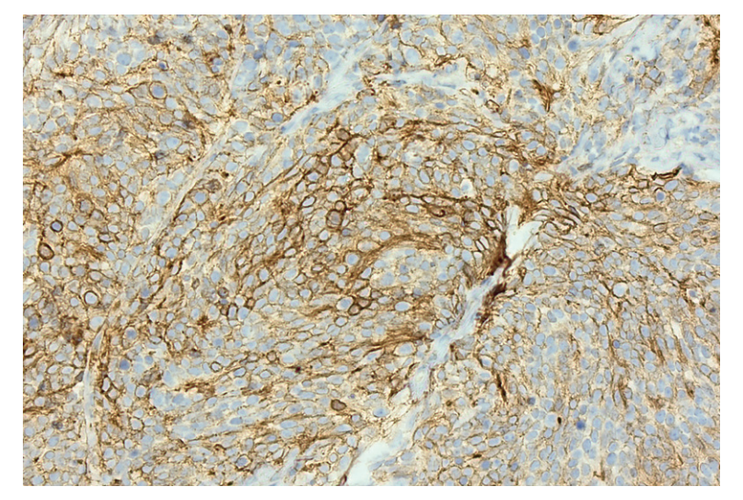

A. GB01-VA-PL2202 and B12 isotype-control ADC (B12-VA-PL2202) were administered i.v. (Day 0) to treatment groups of 10 mice. A vehicle-treated group was used as control. **B.** Kaplan-Meier analysis of survival. **C.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor. **D.** Representative scan of FFPE PA-1 tumor section stained for Claudin-6 by IHC.

Figure 7. *In vivo* anti-tumour activity in the OV-90 ovarian cancer xenograft model

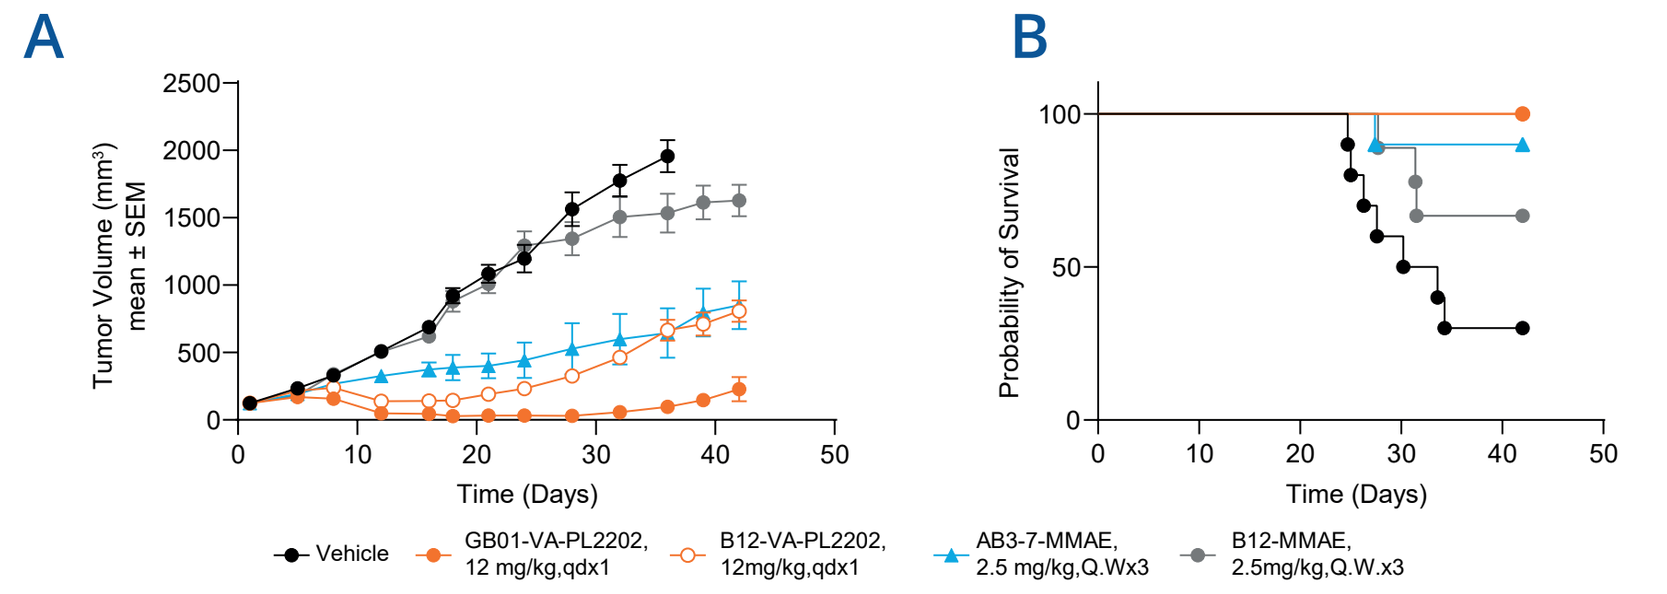
A
B
Tumor Volume (mm³) mean ± SEM
Time (Days)
Probability of Survival
Vehicle
GB01-VA-PL2202, 12 mg/kg,qdx1
B12-VA-PL2202, 12mg/kg,qdx1
AB3-7-MMAE, 2.5 mg/kg,Q.Wx3
B12-MMAE, 2.5mg/kg,Q.W.x3

C

ADCs	PR	CR	TFS
12 mg/kg GB01-VA-PL2202	4/10	5/10	0/10
12 mg/kg B12-VA-PL2202	0/10	0/10	0/10
2.5 mg/kg AB3-7-MMAE	0/10	1/10	1/10
2.5 mg/kg B12-MMAE	0/10	0/10	0/10

D

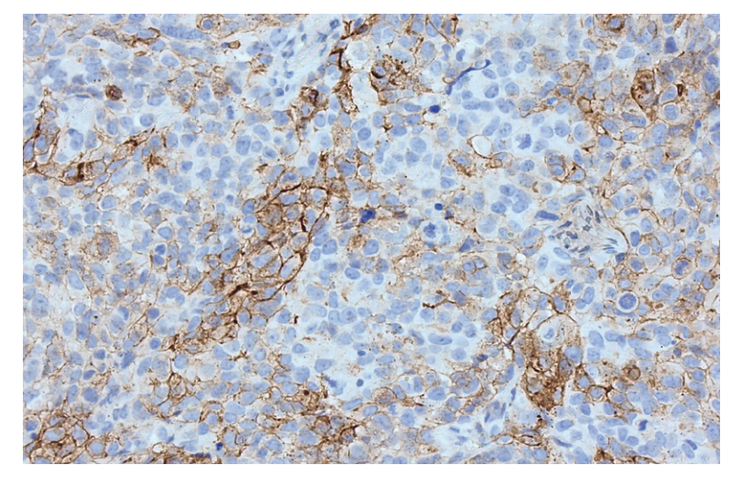

A. GB01-VA-PL2202, AB3-7-MMAE and B12 isotype-control ADCs (B12-MMAE and B12-VA-PL2202) were administered i.v. (Day 1) as indicated to treatment groups of 10 mice. A vehicle-treated group was used as control. **B.** Kaplan-Meier analysis of survival. **C.** Response summary: PR, partial response; CR, complete response; TFS, tumor-free survivor. **D.** Representative scan of FFPE OV-90 tumor section stained for Claudin-6 by IHC.

Figure 8. GB01-VA-PL2202 mean serum concentration-time profiles

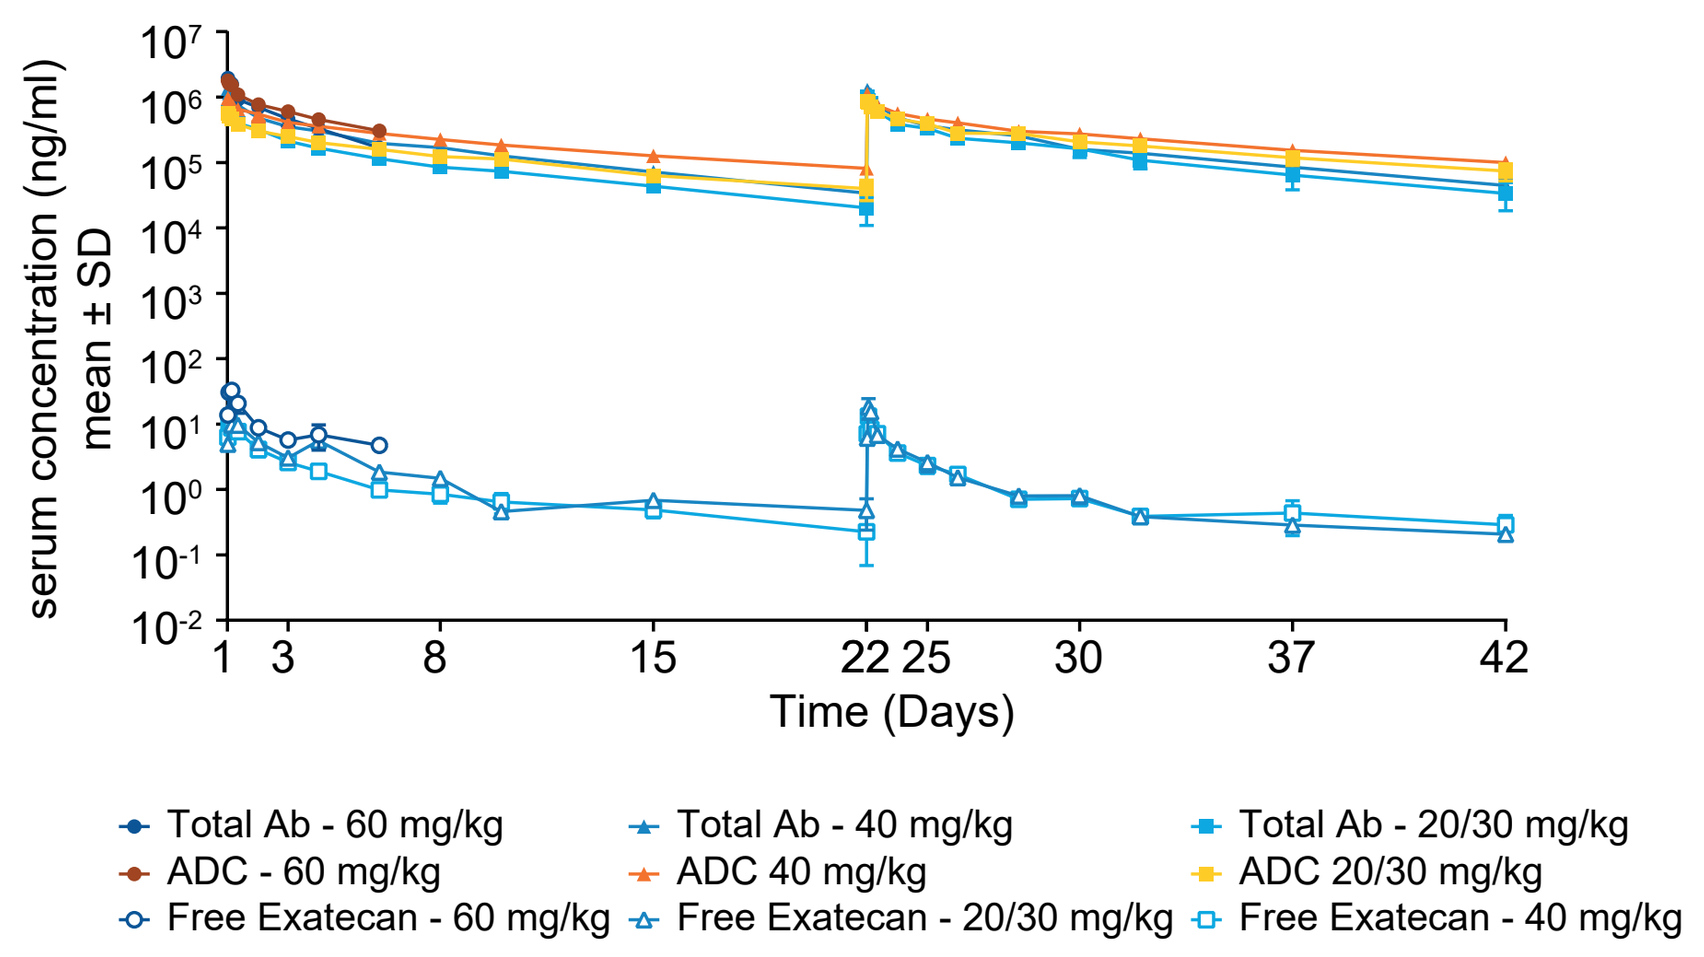
serum concentration (ng/ml) mean ± SD
Time (Days)
Total Ab - 60 mg/kg
ADC - 60 mg/kg
Free Exatecan - 60 mg/kg
Total Ab - 40 mg/kg
ADC 40 mg/kg
Free Exatecan - 20/30 mg/kg
Total Ab - 20/30 mg/kg
ADC 20/30 mg/kg
Free Exatecan - 40 mg/kg

PK in cynomolgus monkeys following IV administration of GB01-VA-PL2202 on Day 1 and 22 (n=3/group). GB01-VA-PL2202 exhibited linear PK with a t½ of ~8.5–10 days (based on data from the ADC assay); free exatecan was detectable throughout the dosing period with an apparent t½ of 5.5–8 days.

GB01-VA-PL2202 is tolerated in cynomolgus monkeys up to 40 mg/kg

- Cynomolgus monkeys are pharmacologically relevant species for GB01-VA-PL2202.
- In a dose-range finding toxicity study in male cynomolgus monkeys, GB01-VA-PL2202 was well tolerated at doses up to 40 mg/kg IV when administered on Day 1 and 22 with minimal clinical pathology and no histopathology findings.
- At the higher dose level of 60 mg/kg, the dose limiting toxicity was degeneration/regeneration in the gastrointestinal tract, consistent with a known class effect of topoisomerase inhibitors such as exatecan.

Figure 9. Claudin-6 membranous expression in a panel of tissues from testicular cancer, ovarian cancer and NSCLC patients

A

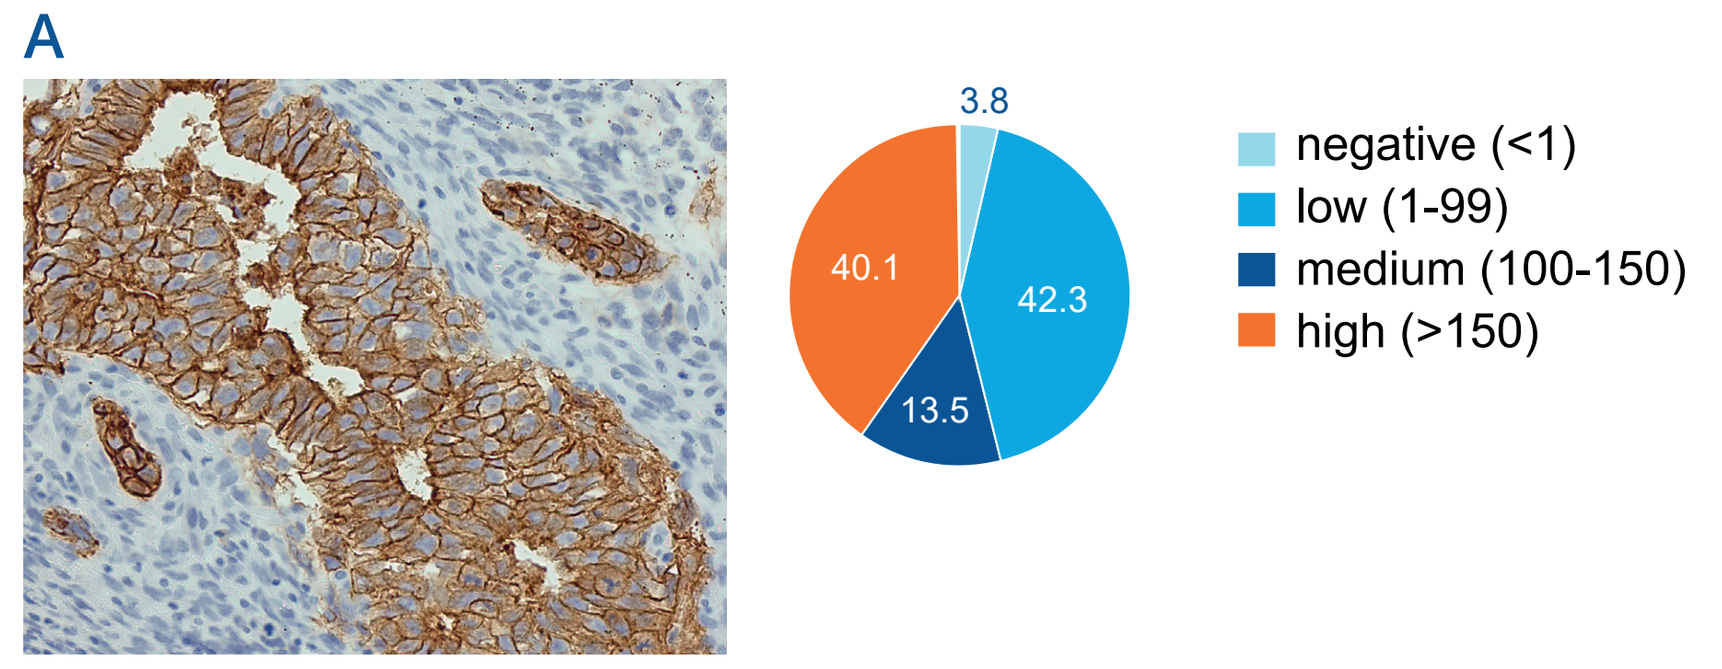
3.8
40.1
42.3
13.5
negative (<1)
low (1-99)
medium (100-150)
high (>150)

B

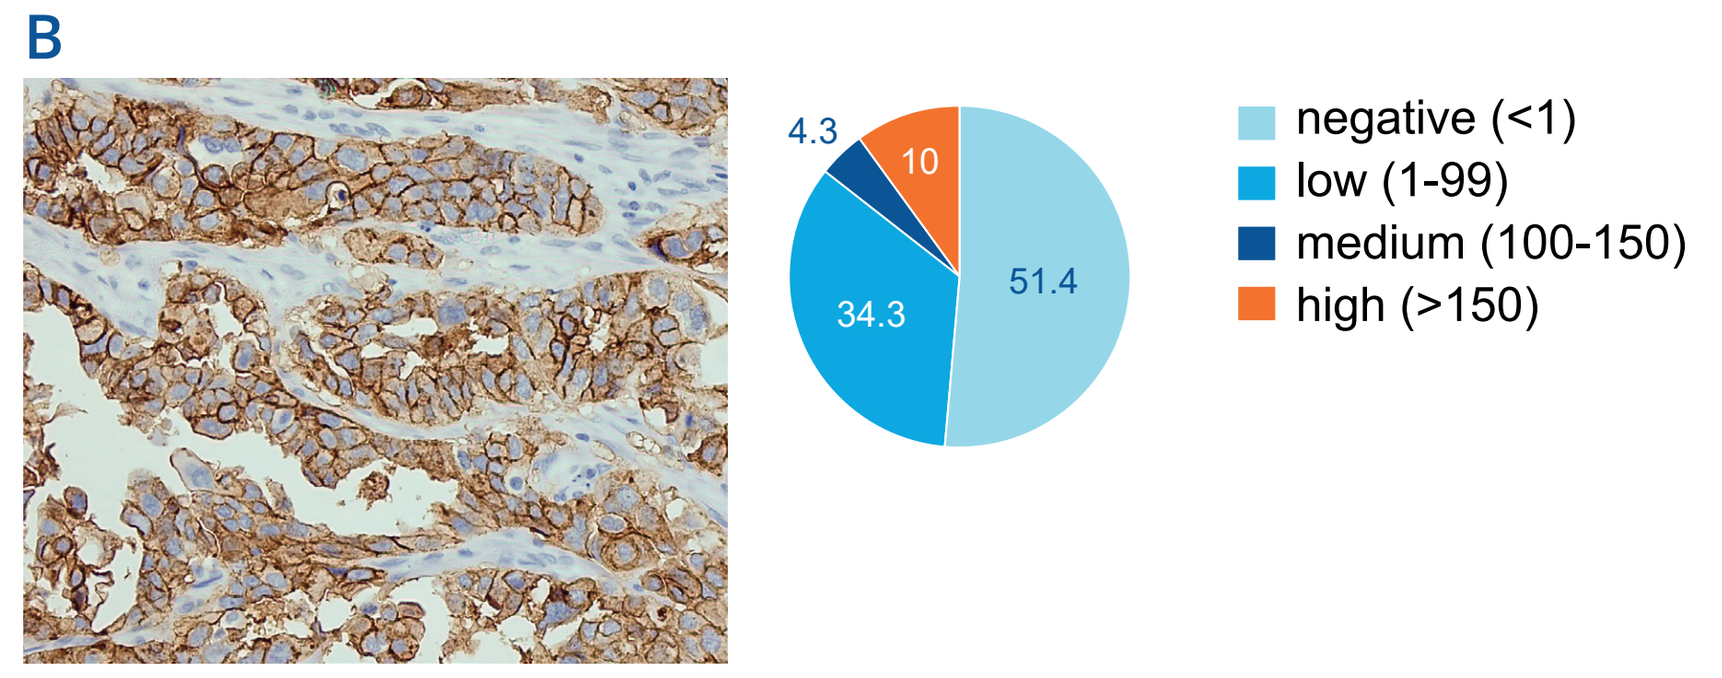
4.3
10
34.3
51.4
negative (<1)
low (1-99)
medium (100-150)
high (>150)

C

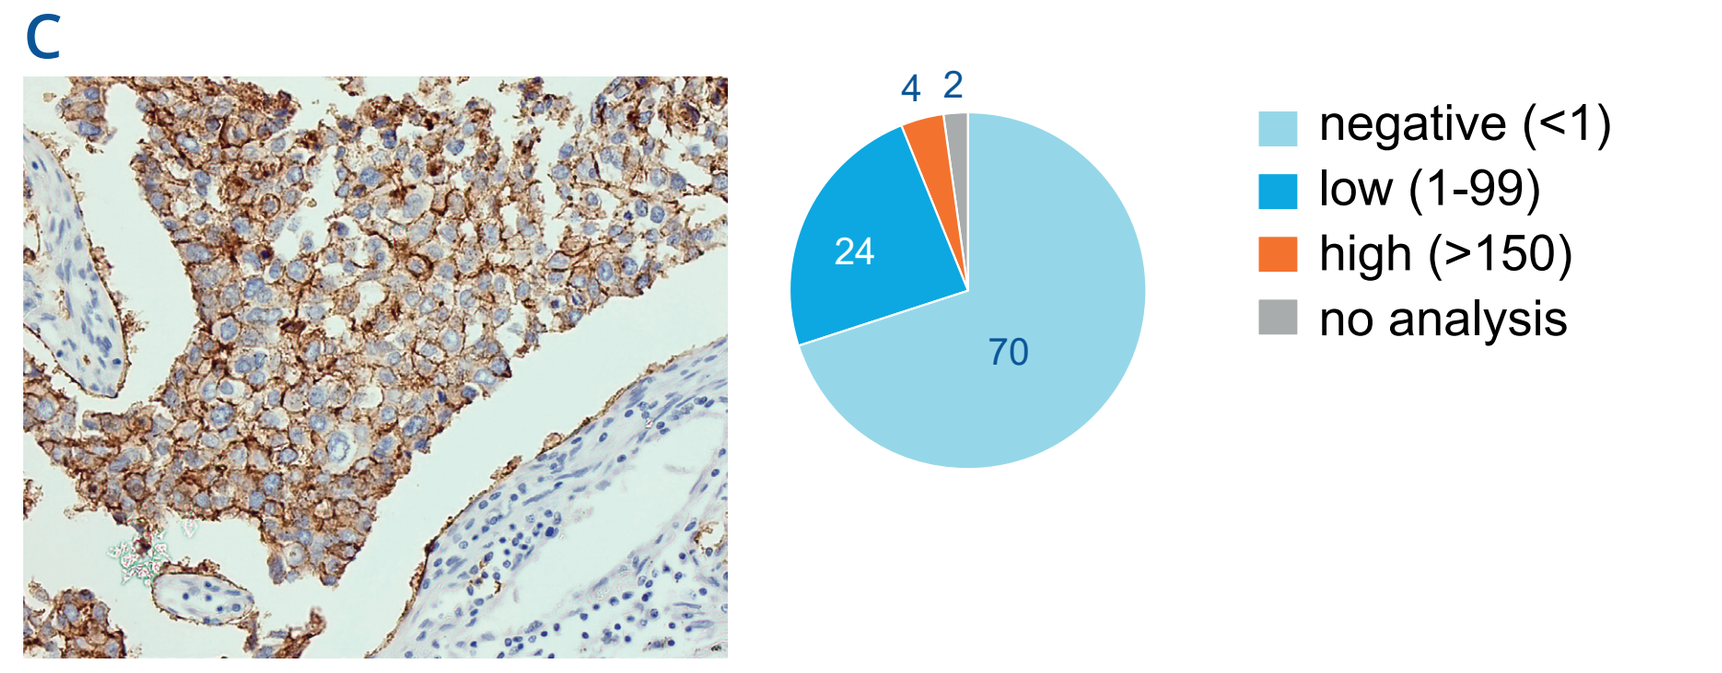
4
2
24
70
negative (<1)
low (1-99)
high (>150)
no analysis

Representative images of Claudin-6 staining showing high membrane expression (IHC score >150). Pie charts indicate % positivity for each IHC-score across cancer patients, **A.** testicular cancer (n=52), **B.** ovarian cancer (n=70), **C.** NSCLC (n=50).