Reused with permission from the American Society of Hematology. © 2024/2023 The Authors. All rights reserved. Officially licensed by ASH for distribution via https://www.adctmedical.com/ and reactively by ADC Therapeutics.

LIMITED DURATION LONCASTUXIMAB TESIRINE INDUCES A HIGH RATE OF COMPLETE RESPONSES IN PATIENTS WITH RELAPSED/REFRACTORY MARGINAL ZONE LYMPHOMA—REPORT OF FIRST PLANNED INTERIM FUTILITY ANALYSIS OF A MULTICENTER PHASE II STUDY

I.S. LOSSOS[1], G. SHOUSE[2], A.J. ALENCAR[1], S. SAWLEH[1], D. LESSEN[1], C.H. MOSKOWITZ[1], J.R. CHAPMAN[1], I.M. REIS[1], R.A. KUKER[1], and J.P. ALDERUCCIO[1]

1. Sylvester Comprehensive Cancer Center, University of Miami Miller School of Medicine, Miami, FL

2. City of Hope National Medical Center, Duarte, CA

INTRODUCTION

- Marginal zone lymphoma (MZL) is a heterogeneous disease representing 7% of non-Hodgkin lymphomas[1]
- Complete response (CR) rate is one of the most important prognostic factors in MZL and was recently validated as the best surrogate marker in patients with extranodal MZL treated with systemic therapy[2]
- In the relapse setting, there is no standardized therapeutic approach, and CR is infrequently achieved
- We report a multicenter clinical trial examining loncastuximab tesirine (Lonca) efficacy in inducing CR and improving survival in patients with relapsed/refractory (R/R) MZL needing treatment

AIM

- This is an ongoing, open-label, multi-institutional, investigator-initiated study evaluating the safety and efficacy of Lonca in R/R MZL (NCT05296070)

METHODS

- Adult patients previously treated with ≥1 line of systemic therapy were eligible if they required treatment based on symptoms, met predefined criteria for an MZL subtype, or experienced disease progression within 24 months (POD24)
- Lonca was given intravenously at 0.15 mg/kg every 3 weeks (Q3W) for 2 cycles followed by 0.075 mg/kg Q3W for 4 cycles
 - Premedication with dexamethasone (4 mg twice daily for 3 days) and prophylaxis to prevent fluid overload with spironolactone (100 mg) was required
- The primary endpoint was CR after 6 cycles based on Lugano 2014 criteria[3] using PET-CT (if FDG avid at presentation) or MRI or CT(for nonavid disease)
- The study implements a Bayesian optimal phase 2 design, with 2 sequential interim analyses of the CR rate, at 20 and then at 40 evaluable patients, with a final analysis at 50 evaluable patients

RESULTS

- A total of 23 patients were enrolled from July 2022 to October 2024; patient demographics and disease characteristics are given in **Table 1**

Table 1. Patient demographics and disease characteristics

Characteristic	Patients (N = 23)
Age, median (range)	65 (45-82)
Gender (M:F)	8:15
Race, n (%)	
Asian	2 (9)
Black non-Hispanic	1 (4)
White Hispanic	9 (39)
White non-Hispanic	11 (48)
MZL type, n (%)	
EMZL	14 (61)
NMZL	7 (30)
SMZL	2 (9)
Stage, n (%)	
I	4 (17)
III	2 (9)
IV	17 (74)

Characteristic	Patients (N = 23)
ECOG PS 0-1, n (%)	23 (100)
POD24, n (%)	11 (48)
Median previous lines of treatment (range)	2 (1-4)
Relapsed, n (%)	14 (61)
Refractory, n (%)	9 (39)
Previous treatments,[a] n (%)	
Rituximab	8 (35)
XRT	7 (30)
R-CHOP	7 (30)
BR	6 (26)
BTKi	4 (17)
R^2	3 (13)
RICE	2 (9)

[a]Other previous treatments, each by 1 patient include CAR-T, R-CVP, R-GemOx, R-MPV, splenectomy, and zevalin.

BR, bendamustine/rituximab; BTKi, Bruton tyrosine kinase inhibitor; CAR-T, chimeric antigen receptor T-cell therapy; ECOG PS, Eastern Cooperative Oncology Group performance status; EMZL, extranodal MZL; MZL, marginal zone lymphoma; NMZL, nodal MZL; POD24, disease progression within 24 months; SMZL, splenic MZL; R^2, rituximab/lenalidomide; R-CHOP, rituximab/cyclophosphamide/doxorubicin/vincristine/prednisone; R-CVP, rituximab/cyclophosphamide/vincristine/prednisone; R-GemOx, rituximab/gemcitabine/oxaliplatin; RICE, rituximab/ifosfamide/carboplatin/etoposide; R-MPV, rituximab/methotrexate/procarbazine/vincristine; XRT, radiotherapy.

- As of October 15, 2024, 23 patients were evaluable for response
- The overall response rate was 91% (21/23 patients), with a CR rate of 70% (16/23 patients; **Figure 1**) and with 2 patients still on treatment
- Lonca led to a CR in 7 of 11 patients (64%) with POD24 who were assessed for response; 1 patient who had progressed after CAR-T achieved a CR with Lonca

Figure 1. Swimmer plot of evaluable patients

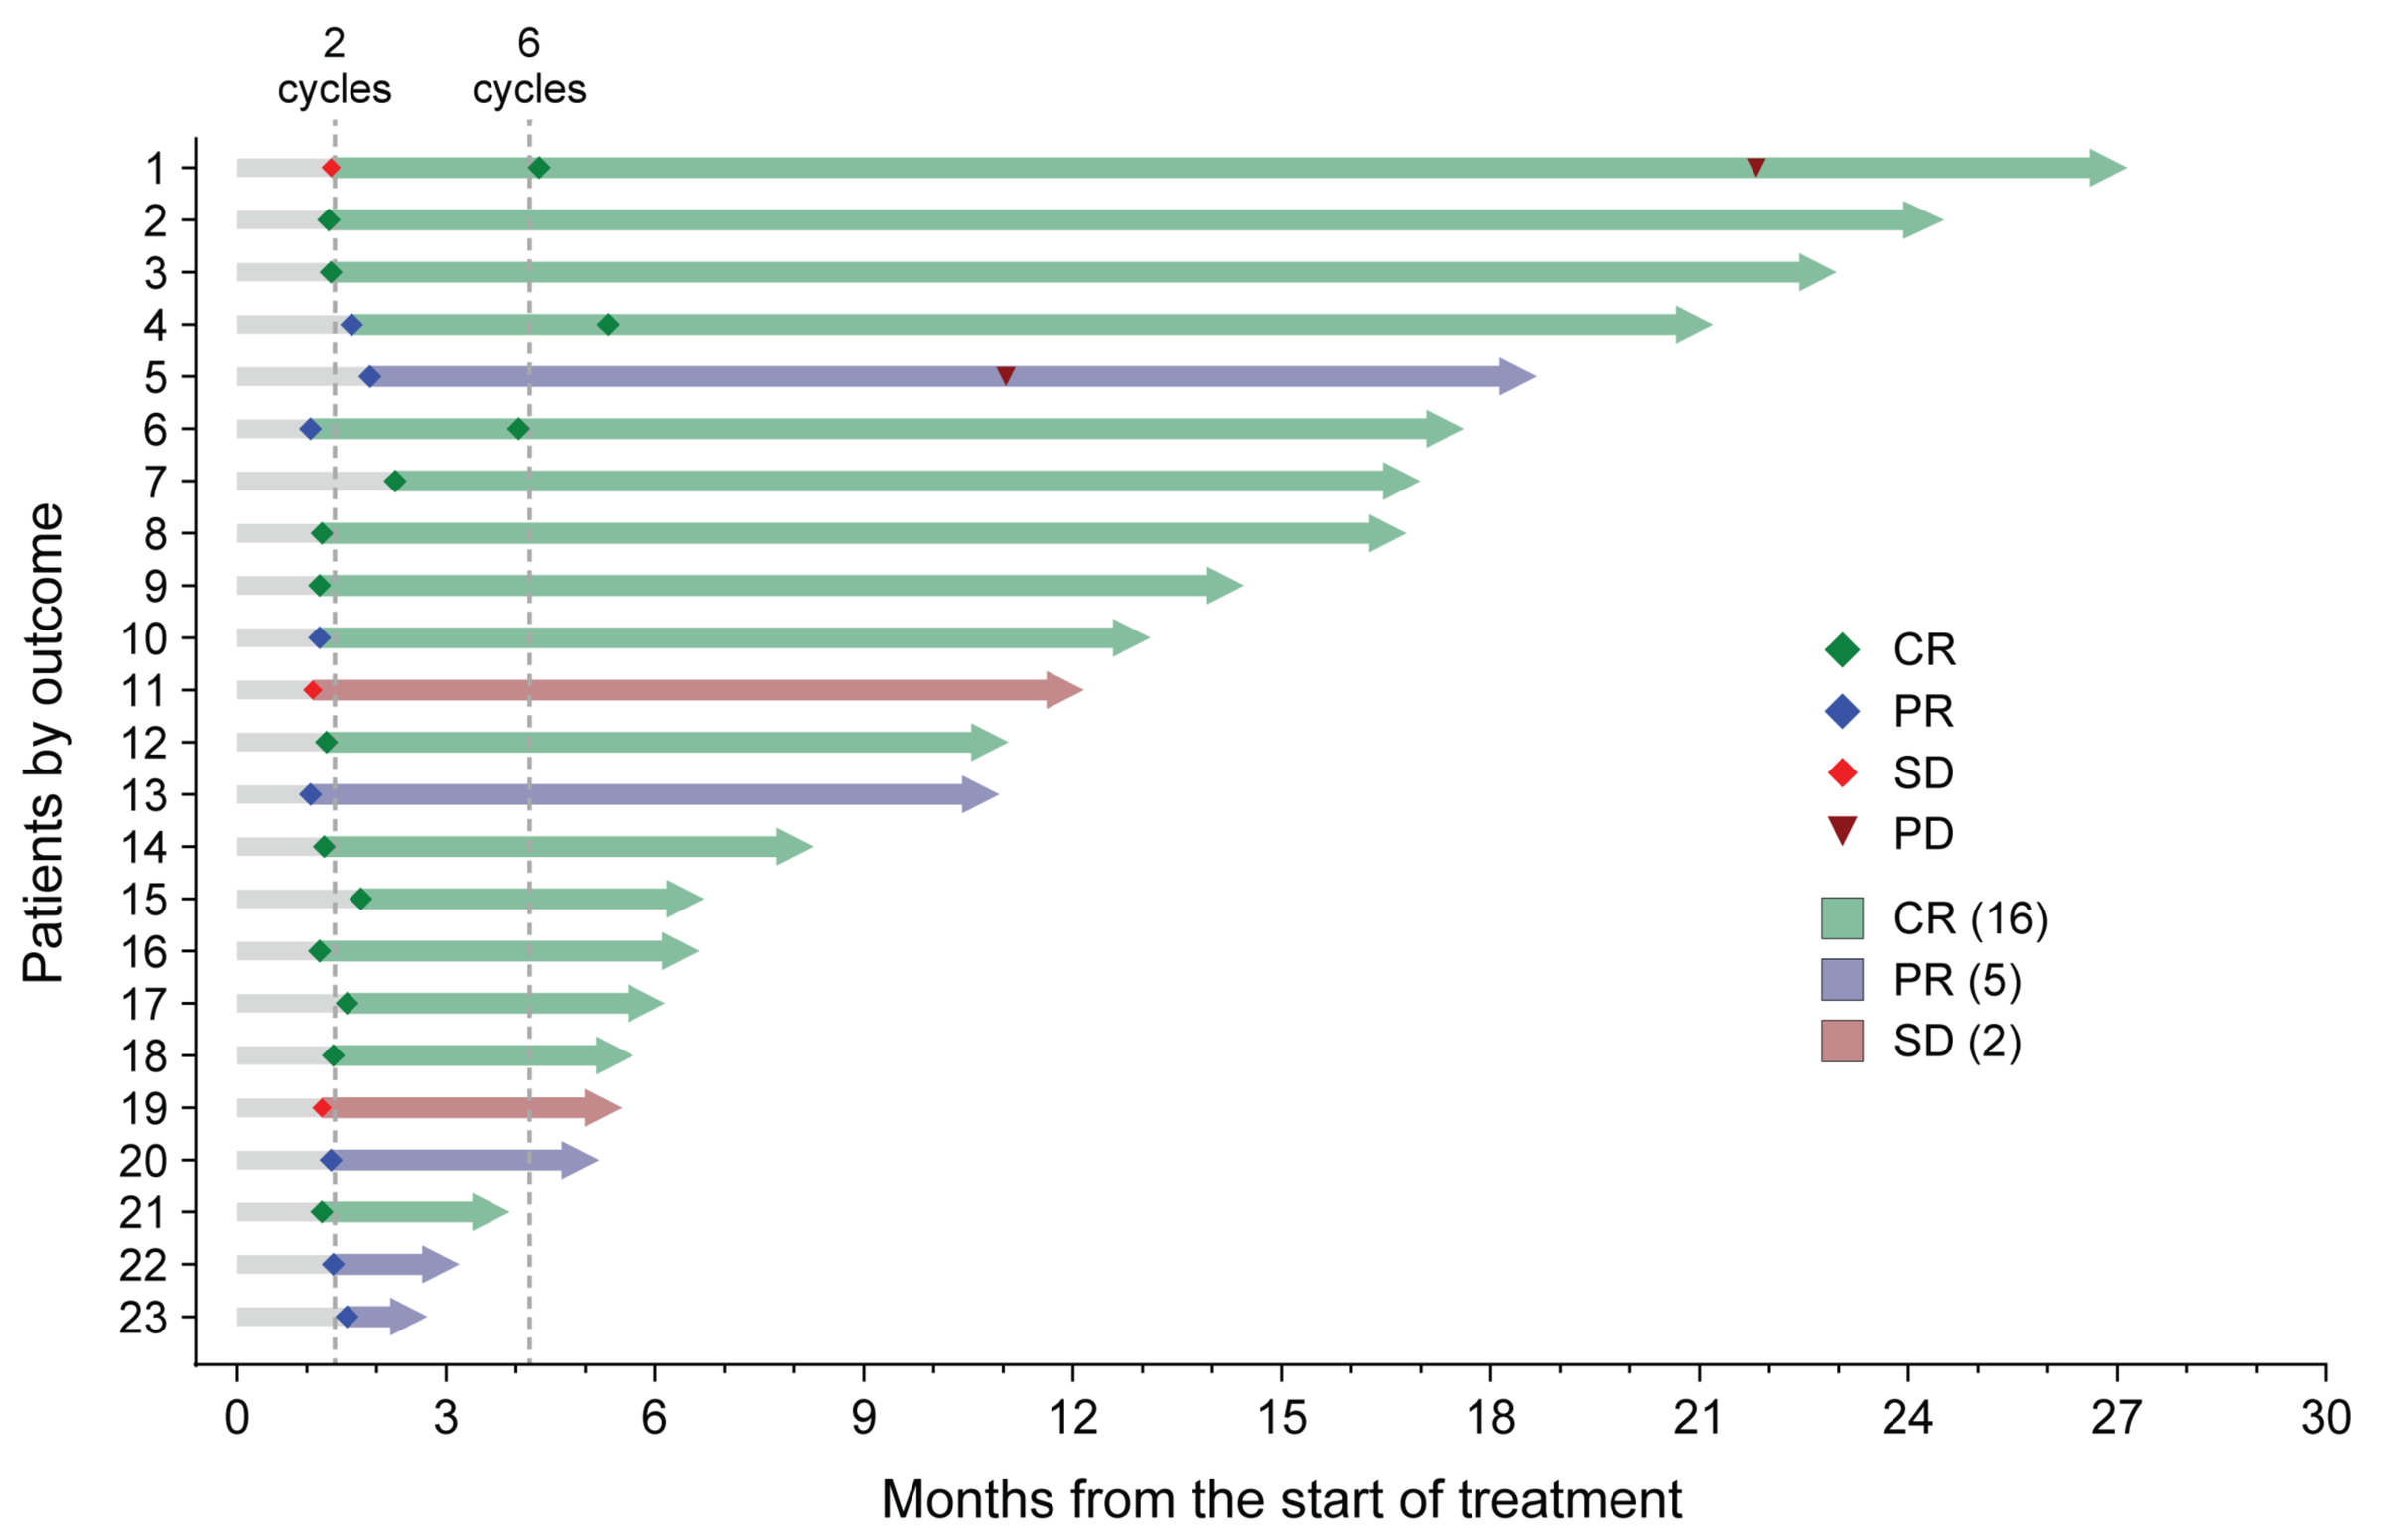
2 cycles
6 cycles
Patients by outcome
Months from the start of treatment
CR
PR
SD
PD
CR (16)
PR (5)
SD (2)

CR, complete response; PD, progressive disease; PR, partial response; SD, stable disease.

- A total of 14 patients were in CR at the end of treatment; all but 1 CR are currently maintained, with the longest follow-up being 27 months from the start of treatment (median duration of CR, 11.5 months); 1 patient with dural extranodal MZL relapsed 17.5 months after achieving a CR (**Figure 2**)

Figure 2. (A) Duration of response[a] and (B) duration of complete response

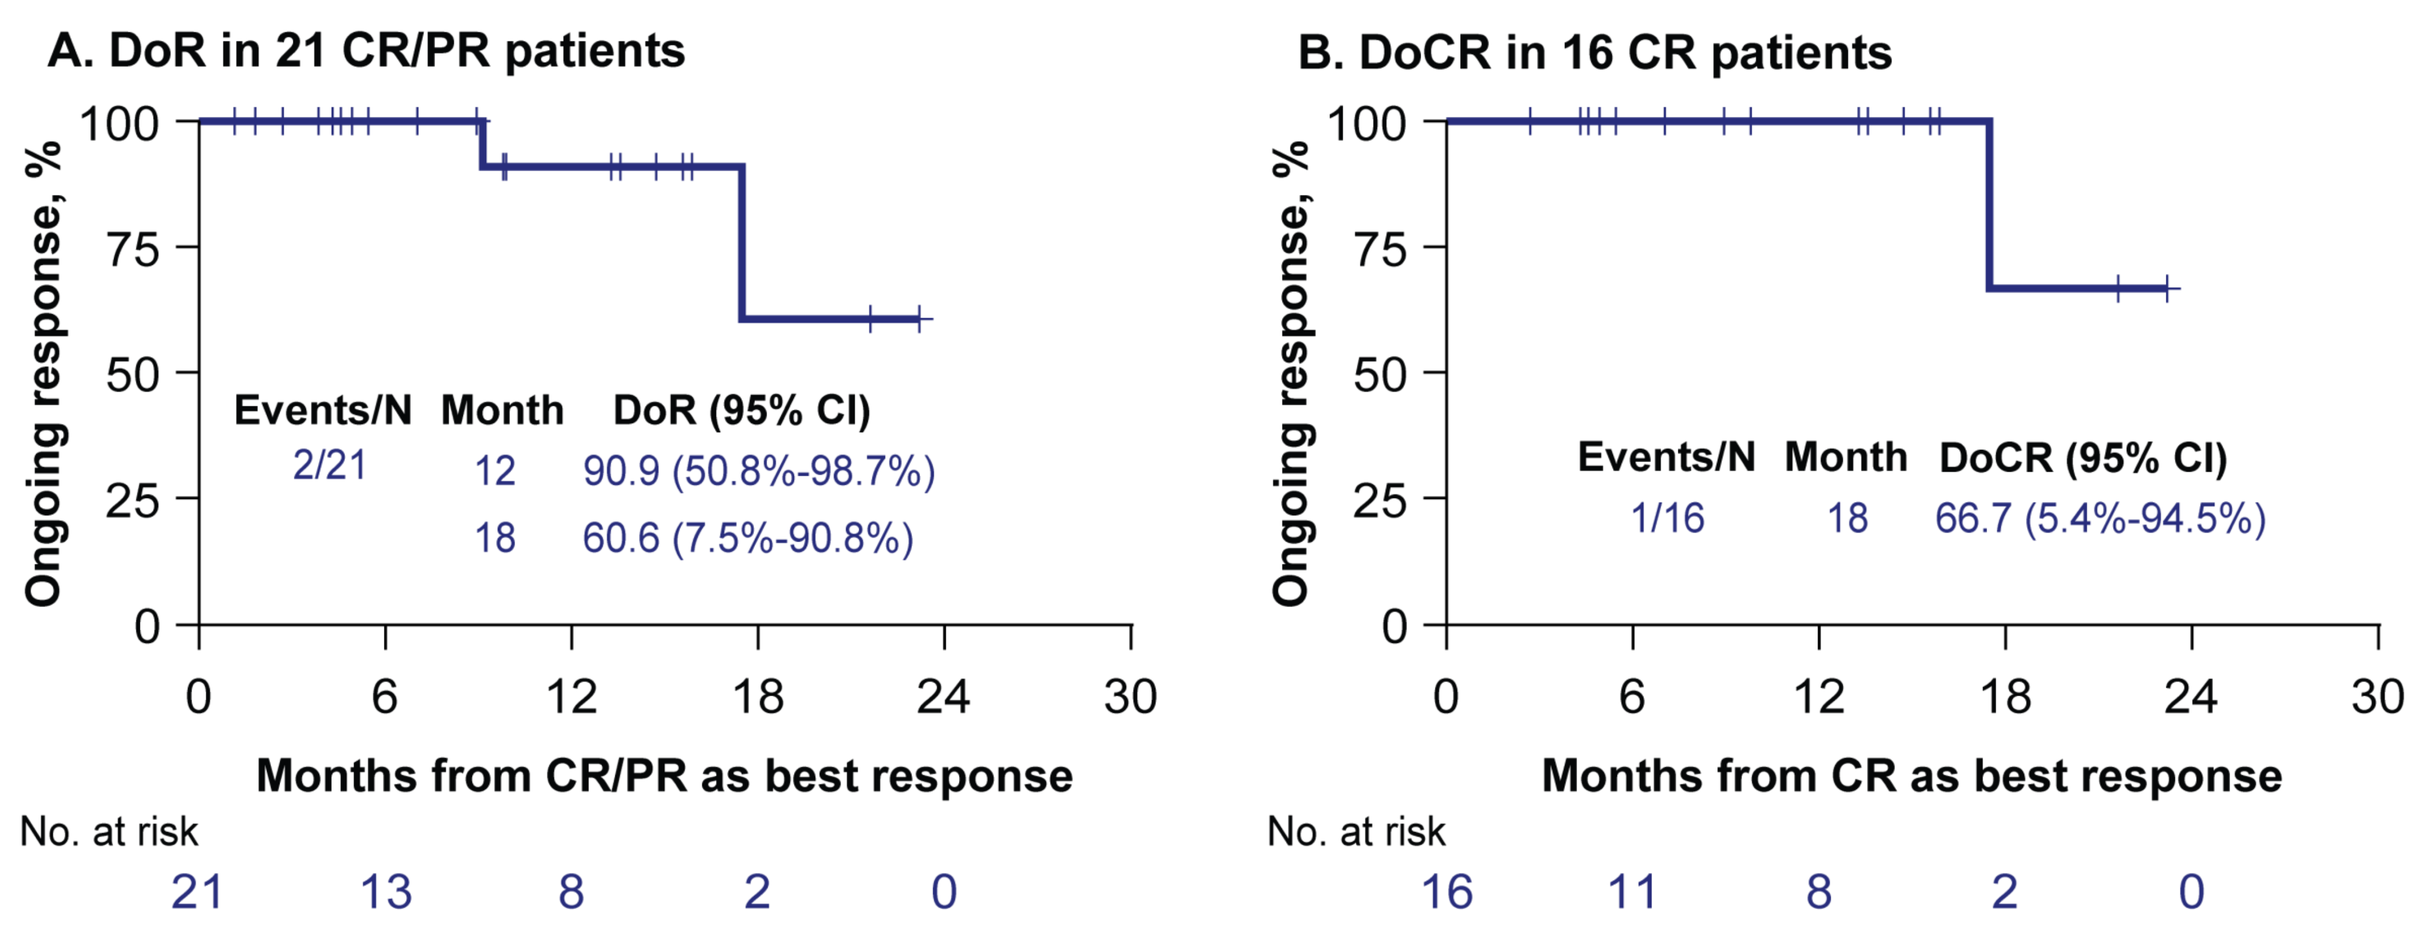
A. DoR in 21 CR/PR patients
Ongoing response, %
Events/N Month DoR (95% CI)
2/21 12 90.9 (50.8%-98.7%)
18 60.6 (7.5%-90.8%)
Months from CR/PR as best response
No. at risk 21 13 8 2 0
B. DoCR in 16 CR patients
Ongoing response, %
Events/N Month DoCR (95% CI)
1/16 18 66.7 (5.4%-94.5%)
Months from CR as best response
No. at risk 16 11 8 2 0

[a]DoR was defined as the time from achievement of CR or PR until progression, relapse, death, or initiation of new treatment.
CR, complete response; DoCR, duration of complete response; DoR, duration of response; OS, overall survival; PR, partial response.

- The estimated progression-free survival rate at 12 months was 91.7%. No patients have died during treatment (**Figure 3**)

Figure 3. (A) PFS and (B) OS in evaluable patients

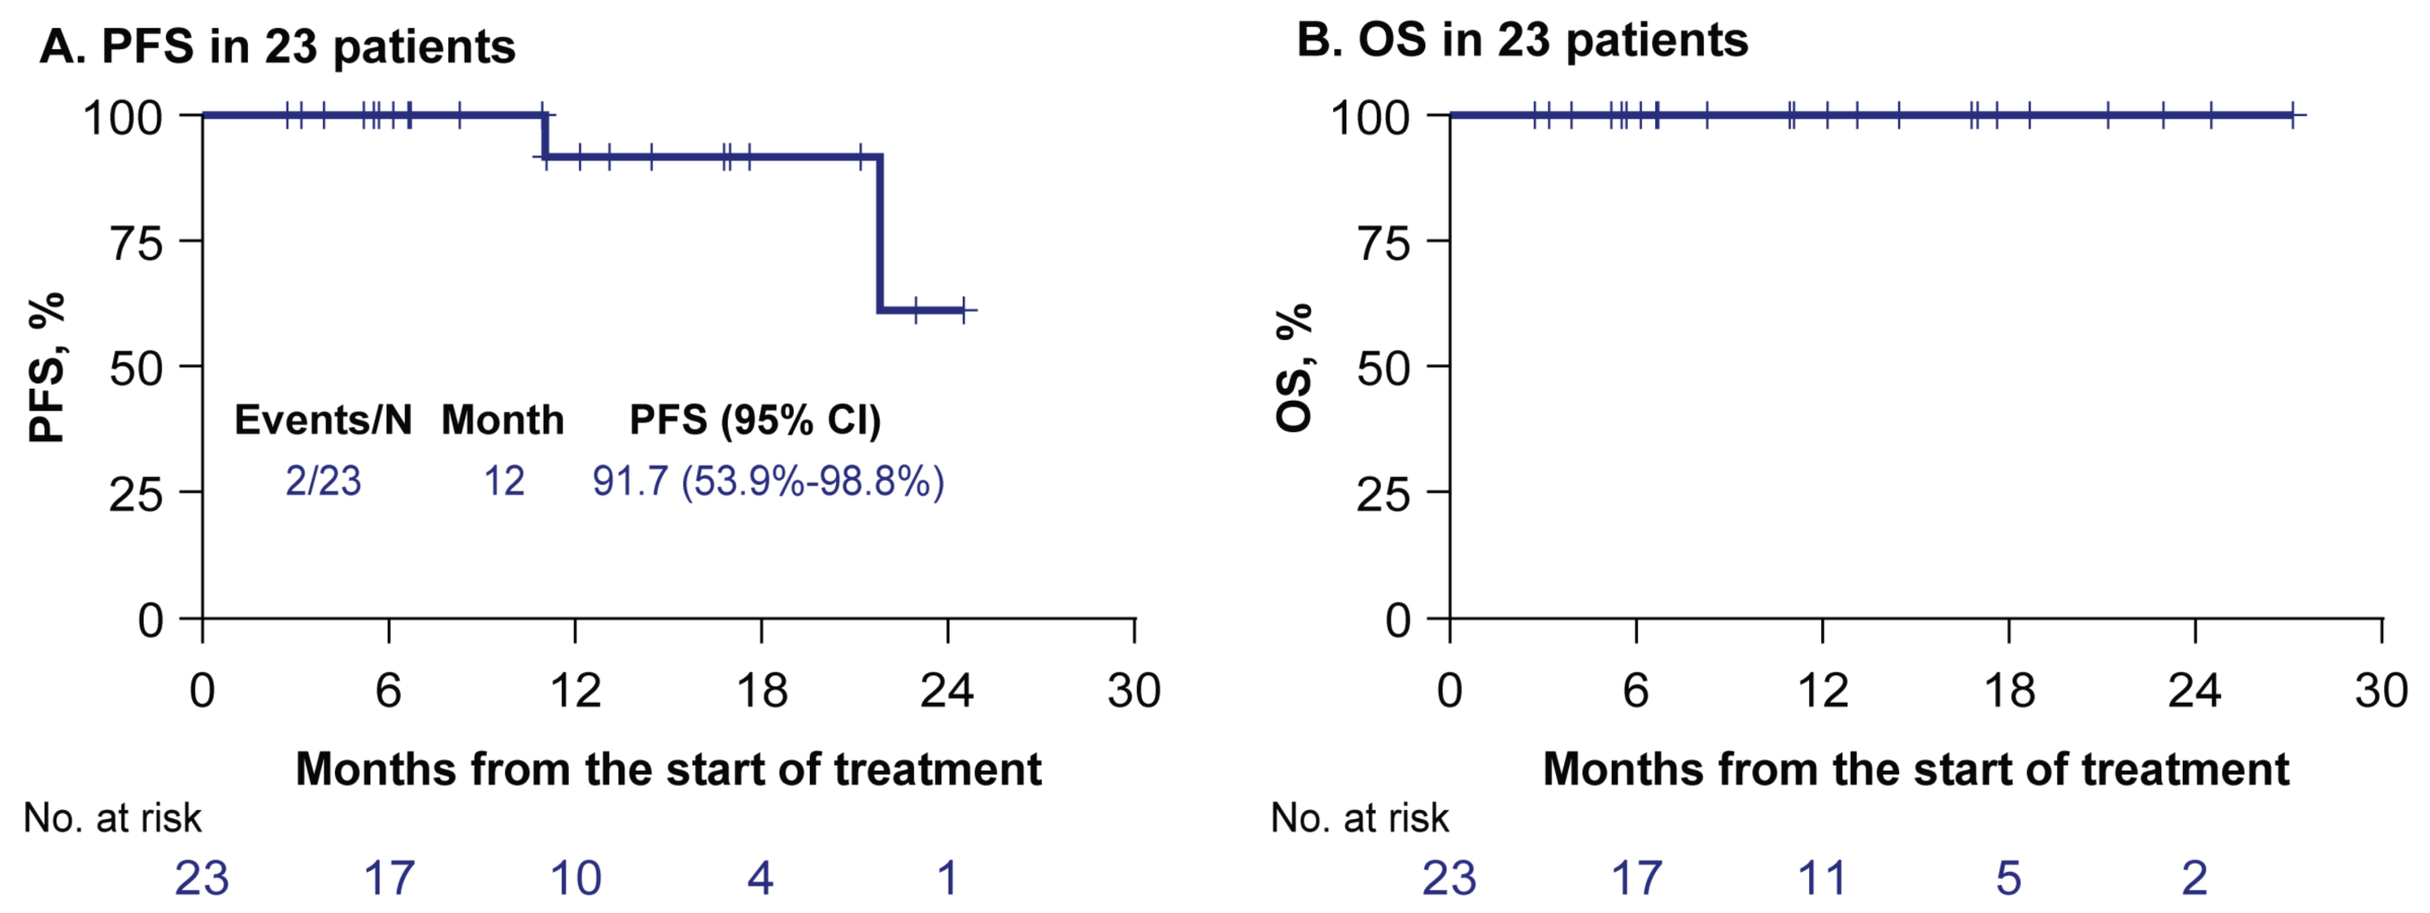
A. PFS in 23 patients
PFS, %
Events/N Month PFS (95% CI)
2/23 12 91.7 (53.9%-98.8%)
Months from the start of treatment
No. at risk 23 17 10 4 1
B. OS in 23 patients
OS, %
Months from the start of treatment
No. at risk 23 17 11 5 2

OS, overall survival; PFS, progression-free survival.

- Generally, Lonca was well tolerated, and the observed safety was consistent with the known profile (**Table 2**)

Table 2. Safety profile of Lonca

TEAE, n (%)	Patients (N = 23)		
	Any grade	Grade 3	Grade 4
Maculopapular rash	15 (65.2)	1 (4.3)	0
Increased AST	15 (65.2)	0	0
Increased ALT	14 (60.8)	2 (8.7)	0
Increased alkaline phosphatase	11 (47.8)	3 (13.0)	0
Neutropenia	10 (43.4)	3 (13.0)	1 (4.3)
Local edema	10 (43.4)	0	0
Photosensitivity	7 (30.4)	1 (4.3)	0
Anemia	7 (30.4)	1 (4.3)	0
Urinary infection	3 (13.0)	1 (4.3)	0
Lung infection	2 (8.7)	0	0
Pleural effusion	2 (8.7)	0	0
Anorexia	1 (4.3)	1 (4.3)	0
COVID-19	1 (4.3)	0	0
Weight loss	1 (4.3)	1 (4.3)	0

ALT, alanine aminotransferase; AST, aspartate aminotransferase; Lonca, loncastuximab tesirine; TEAE, treatment-emergent adverse event.

- All 23 patients experienced expected adverse events (AEs), most commonly grade 1 or 2. Grade 3 and 4 AEs were observed in 15 and 1 (neutropenia) patients, respectively
- Three patients needed a dose reduction. One patient discontinued treatment after cycle 4 because of cholestatic hepatitis; the patient clinically fully recovered, with normalization in liver function test abnormalities

CONCLUSIONS

- Lonca demonstrates clinically meaningful activity with a robust CR rate in patients with R/R MZL in our ongoing phase 2 study, and passed first and second interim futility analysis thresholds for early stopping
- The treatment is well tolerated, and the safety profile is consistent with known adverse effects with no new unexpected toxicities
- The patients demonstrated durable CRs, but longer follow-up is needed

REFERENCES

1. Cerhan JR and Habermann TM. *Ann Lymphoma*. 2021;5:1.
2. Bommier C, et al. *Blood*. 2024;143(5):422-428.
3. Cheson BD, et al. *J Clin Oncol*. 2014;32(27):3059-3068.

CONTACT INFORMATION

Presenting author: Izidore S. Lossos (ilossos@med.miami.edu)

Copies of this poster obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

ACKNOWLEDGMENTS

The authors thank the patients who volunteered to participate in this trial, their families, and the staff members at the trial sites who cared for them.

Medical writing and editorial support, provided by Citrus Scientific, a Citrus Health Group, Inc., company (Chicago, Illinois), was provided in accordance with Good Publication Practices (GPP 2022) and funded by ADC Therapeutics SA and Sobi.