HuB14-VA-PL2202, a novel antibody-drug conjugate targeting ASCT2, a novel ADC target over-expressed in both solid and hematological cancers

Danilo Cucchi[1], Elizabeth Horsley[1], Alina Chrzastek[1], Ben Leatherdale[1], Simone Sims[1], Nicole Bledowski[1], Banusha Rajeenthira[1], Narinder Janghra[1], Lara Montolio[1], Abigail Kimpalu[1], Christopher Gallagher[1], Charlie Britten[1], Lolke de Haan[1], Paul W. Hogg[1], Chris Pickford[1], Patrick H. van Berkel[1]

[1], ADC Therapeutics UK (Ltd), London, UK

Introduction

SLC1A5 (Solute Carrier family 1 member 5), also known as ASCT2 (Alanine, Serine, Cysteine Transporter 2), is a Na^+-dependent antiporter with its preferred substrate being glutamine[1]. ASCT2 expression is reported in normal tissues, however, it has been shown to be dramatically increased in several solid tumors (lung, colorectal) and hematological malignancies (acute myeloid leukemia (AML), multiple myeloma)[2,3]. Moreover, high level ASCT2 expression was associated with a worse prognosis in liver and lung cancers[3,4]. Because of this profile of expression, ASCT2 is an attractive membrane protein for pharmacological targeting.

We have developed a new antibody-drug conjugate (ADC) targeting ASCT2 (HuB14-VA-PL2202), composed of a humanized IgG1 directed against human ASCT2, to which PL2202, consisting of a proprietary cleavable linker and exatecan, a topoisomerase I inhibitor, has been site-specifically conjugated with a drug to antibody ratio ~6. FcγR-mediated effector function was abrogated via mutation in the Fc portion of the antibody.

Figure 1. ASCT2 (SLC1A5) structure

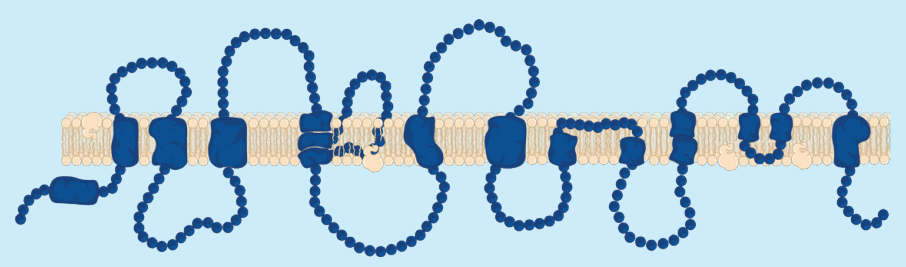

Figure 2. HuB14-VA-PL2202

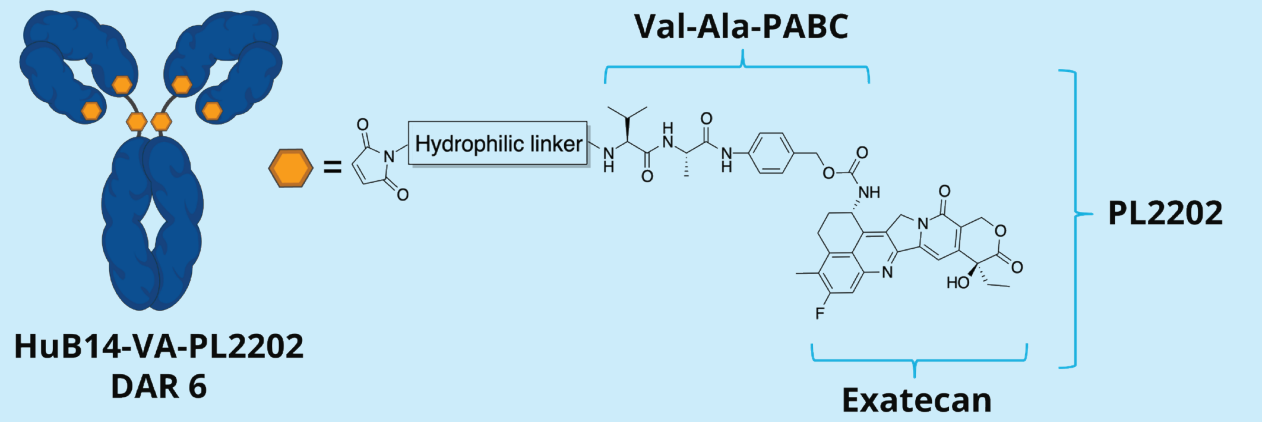
Val-Ala-PABC
Hydrophilic linker
PL2202
HuB14-VA-PL2202
DAR 6
Exatecan

Figure 2. HuB14-VA-PL2202 is a novel ADC, composed of a humanized IgG1, Fc silenced, against ASCT2 to which the exatecan-containing payload PL2202 has been site-specifically conjugated, with a DAR of 6. This was achieved by introducing the Cys228Val mutation in the hinge of the heavy chain. The FcγR-mediated effector function was abrogated via mutations in the Fc portion of the antibody.

Aim of the study

The aim of this study was to characterize the *in vitro* cytotoxicity of HuB14-VA-PL2202 in human cancer cell lines and determine its *in vivo* efficacy. The tolerability of HuB14-VA-PL2202 was also evaluated in cynomolgus monkeys. Membrane expression of ASCT2 was confirmed by IHC in sections from xenograft biopsies and human tissues.

Materials & methods

- Bulk tissue gene expression data of ASCT2, HER2 and TROP2 in normal tissues was extracted from the GTEx Portal. Expression values are shown in TPM (transcript per million).
- Analysis of ASCT2 membranous expression in human TMAs from patients diagnosed with NSCLC, SCLC, CRC and HNSCC and on FFPE sections from CDX and PDX models was performed by IHC using a commercial human ASCT2 antibody from Abcam (clone CAL33).
- Cytotoxicity of HuB14-VA-PL2202 and isotype control ADC B12-VA-PL2202, was determined using CellTiter 96® AQ_{ueous} One Solution Cell Proliferation or CellTitreGlo® assays (Promega). Antibody cell binding was determined by flow cytometry.
- Internalization of HuB14-VA-PL2202 and isotype control ADC B12-VA-PL2202 was performed using an Incucyte® analyser. FabFluor-labelled ADCs (4 μg/mL) were used in HT-29, HCT-116 and OVCAR-3 cells. HD phase and red fluorescence images (20x) were captured. Images of cells treated with FabFluor-ADC display red, cytosolic fluorescence indicative of internalization (shown in blue in figure 3).
- In *in vivo* studies, HuB14-VA-PL2202 was administered intravenously (i.v.) as a single dose to athymic nude mice containing MOLM-13 or G-402 xenografts and to NMRI nude mice containing CXF 260 colon cancer patient-derived xenograft.
- PK was evaluated in human FcRn Tg32 SCID mice (n=4/group) following one IV dose of HuB14-VA. Plasma exposure to the Ab was determined by ELISA.
- Exoposure to HuB14-VA-PL2202 in cynomolgus monkeys (n=3/group) was evaluated following two doses of HuB14-VA-PL2202 given three weeks apart via IV infusion. Serum exposure to total (conjugated and unconjugated Ab) and exatecan-conjugated Ab were determined by ELISA/ECLIA, with exposure to free exatecan determined by LC-MS/MS. PK parameters were determined by non-compartmental analysis (NCA).

Acknowledgements

In vivo studies: Charles River Discovery Research Services (USA) (mouse xenograft studies); Charles River Laboratories Germany GmbH (Germany) (mouse patient-derived xenograft studies); The Jackson Laboratory (UK) (PK study in hFcRn Tg32 mice) and LabCorp (UK) (cynomolgus monkey toxicity study).

Conclusions

- RNAseq analysis confirmed expression of ASCT2 in healthy tissue, albeit at similar levels compared to well-validated ADC targets such as HER2 and TROP2.
- Membranous expression of ASCT2 was confirmed by IHC in a high proportion of NSCLC (squamous), SCLC and HNSCC tissue samples, where it was found to be significantly higher than in the corresponding normal tissue.
- HuB14-VA-PL2202 was rapidly and efficiently internalized in cancer cell lines expressing different levels of ASCT2.
- *In vitro*, HuB14-VA-PL2202 demonstrated potent and target-mediated cytotoxicity in a panel of ASCT2-positive solid and hematological cancer cell lines. Overall, the cytotoxicity of HuB14-VA-PL2202 was higher in cell lines expressing higher level of ASCT2.
- HuB14-VA is cross-reactive to human and cyno ASCT2 and binds to ASCT2 with lower affinity than ASCT17C10[5], the antibody component of MEDI7274, a PBD-based ADC targeting ASCT2. A lower affinity antibody may drive binding to tumors over-expressing ASCT2 while reducing binding to healthy tissue expressing lower levels of ASCT2.
- *In vivo*, HuB14-VA-PL2202 showed strong antitumor activity in MOLM-13 and G-402 cancer xenograft models, and in a patient-derived xenograft model of colon cancer (CXF 260), highlighting its target-mediated antitumor activity.
- HuB14-VA-PL2202 was well tolerated at 40 mg/kg IV given as two doses, three weeks apart to male cynomolgus monkeys.
- Exposure profiles for HuB14-VA-PL2202 ADC and total antibody were generally comparable, with ADC concentrations slightly lower than total Ab concentrations. AUC values were equivalent for both analytes, indicating that the ADC is stable in circulation. Linear TK with a $t_{½}$ of 6.0-6.2 days was observed (based on data from the ADC assay). Free exatecan was detectable throughout the dosing period with apparent $t_{½}$ of 2.8-2.9 days.

In conclusion, ASCT2 represents an interesting novel tumor target for an ADC approach, both in solid and hematological malignancies. HuB14-VA-PL2202 demonstrated potent and specific *in vitro* and *in vivo* anti-tumor activity while it was stable and well tolerated in cynomolgus monkeys, warranting its future investigation in clinical trials.

Results

Figure 3. ASCT2 membranous expression in a panel of tissues from NSCLC, SCLC, Colon cancer and HNSCC patients

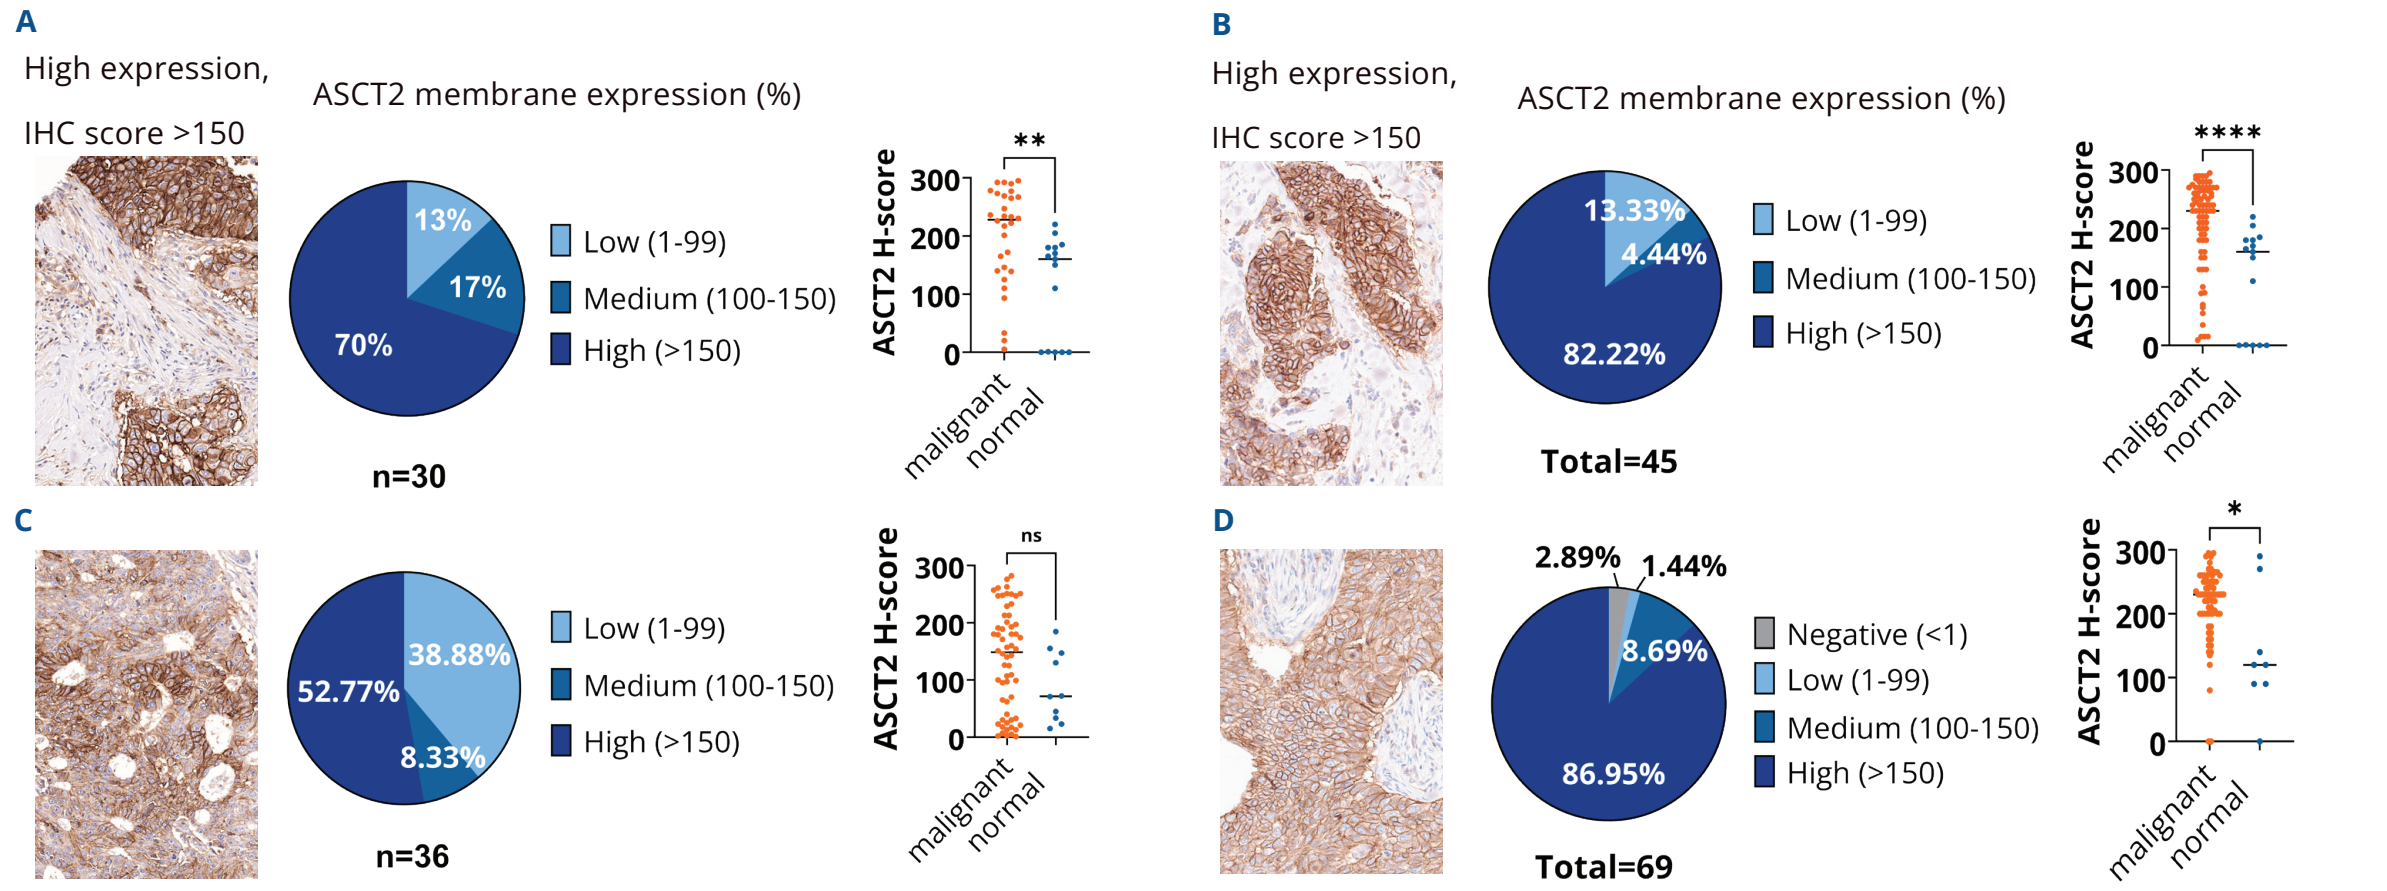
A
High expression,
IHC score >150
ASCT2 membrane expression (%)
13%
17%
70%
Low (1-99)
Medium (100-150)
High (>150)
n=30
ASCT2 H-score
**
malignant
normal
B
High expression,
IHC score >150
ASCT2 membrane expression (%)
13.33%
4.44%
82.22%
Low (1-99)
Medium (100-150)
High (>150)
Total=45

C
38.88%
52.77%
8.33%
Low (1-99)
Medium (100-150)
High (>150)
n=36
ns
D
2.89%
1.44%
8.69%
86.95%
Negative (<1)
Low (1-99)
Medium (100-150)
High (>150)
Total=69
*

Representative images of ASCT2 staining showing high membrane expression (IHC score >150). Pie charts indicate % positivity for each IHC-score across cancer patients; scatter plots show mean H score malignant vs normal for each indication. **A.** NSCLC squamous (n=30), **B.** SCLC (n=45), **C.** Colon cancer (n=36), **D.** HNSCC (n=69). Statistical analysis performed comparing H-scores (malignant vs normal) using un-paired t-test (Mann-Whitney); *p<0.05; **p<0.01; ****p<0.0001

Figure 4. ASCT2 gene expression levels across normal tissues, compared to HER2 and Trop2

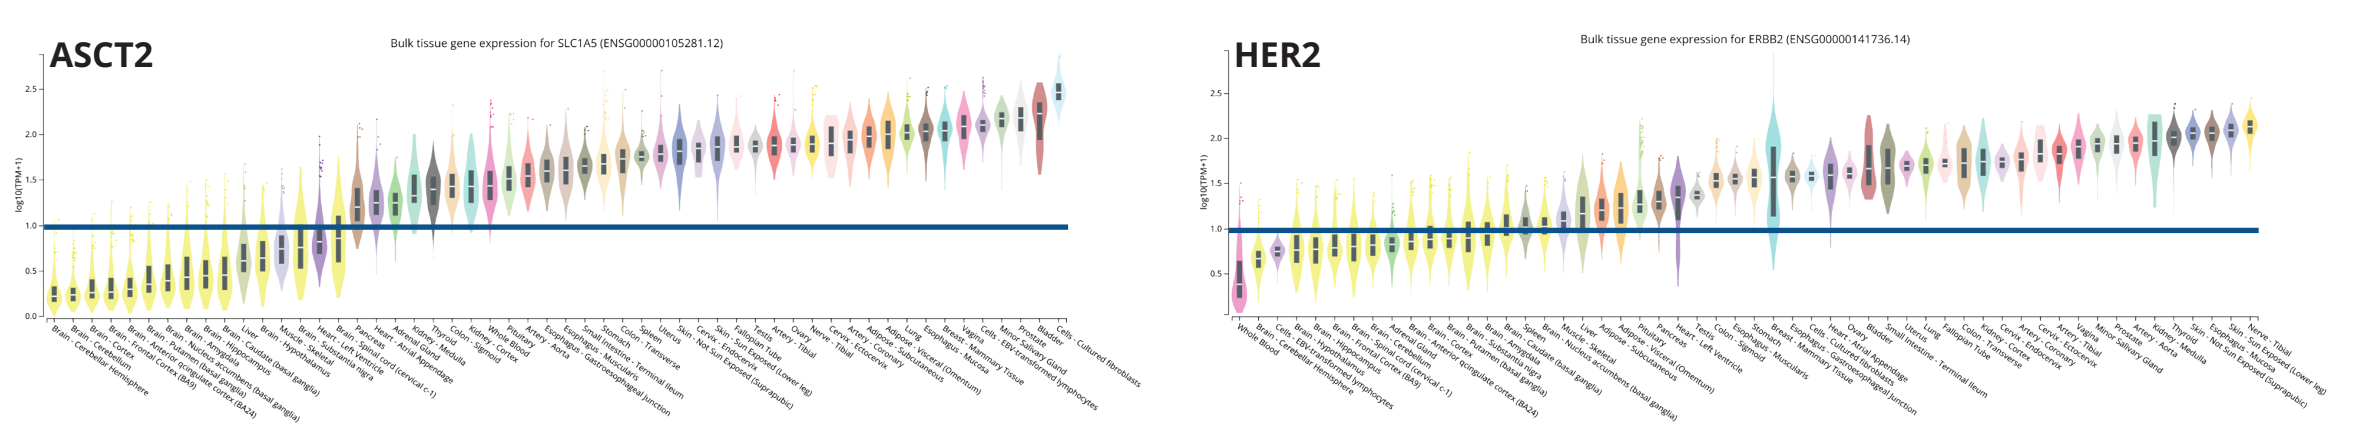
ASCT2
Bulk tissue gene expression for SLC1A5 (ENSG00000105281.12)
HER2
Bulk tissue gene expression for ERBB2 (ENSG00000141736.14)

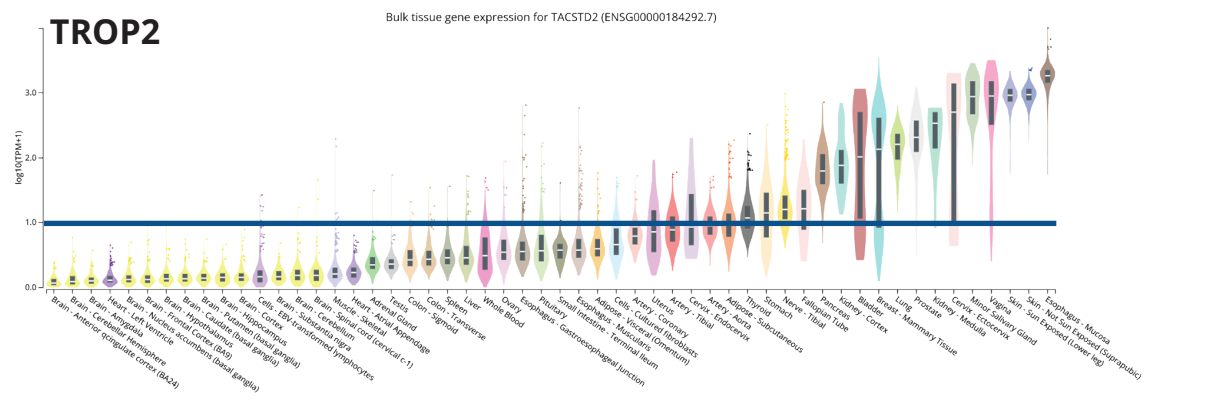
TROP2
Bulk tissue gene expression for TACSTD2 (ENSG00000184292.7)

Gene expression levels of ASCT2, HER2 and Trop2 across a wide range of normal tissues (source: GTEx Portal). Although present in a number of normal tissues, the profile of expression of ASCT2 follows a similar pattern of that of HER2 and Trop2, two well validated ADC targets.

Figure 5. HuB14-VA cross-reactivity and affinity comparison vs ASCT17C10

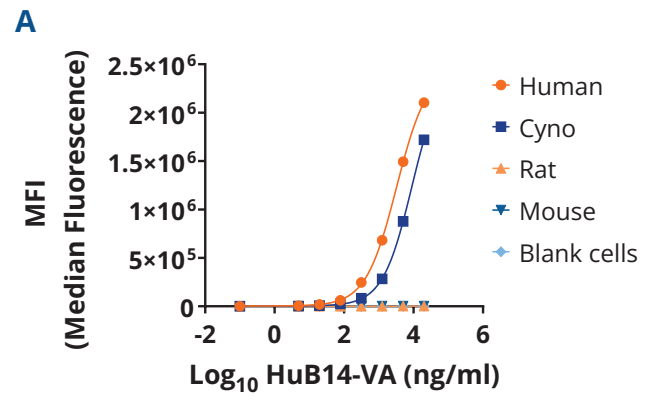
A
MFI
(Median Fluorescence)
Log10 HuB14-VA (ng/ml)
Human
Cyno
Rat
Mouse
Blank cells

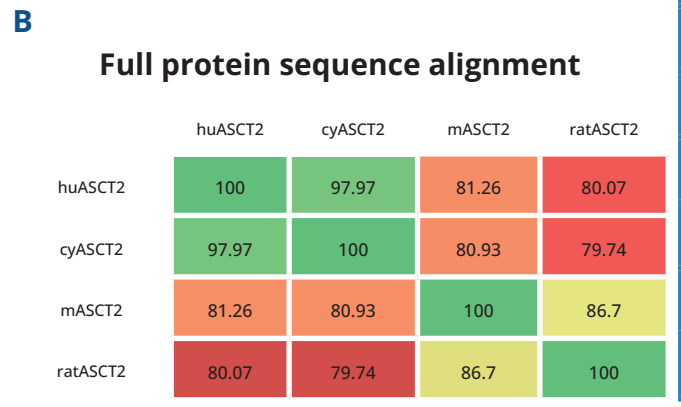
B
Full protein sequence alignment
huASCT2
cyASCT2
mASCT2
ratASCT2
100
97.97
81.26
80.07
97.97
100
80.93
79.74
81.26
80.93
100
86.7
80.07
79.74
86.7
100

C

Cells	EC_{50} ng/ml	
	HuB14-VA	ASCT17C10
CHO-Human	1845	314.6
CHO-Cyno	5537	759.4
OVCAR-3	18339	1027
MOLM-13	25220	2242
Raji	21842	642.7
G-402	838.1	98.44
SW480	3893	1066
KB	7954	652.2
NCI-H292	4267	419.8
NCI-HCT-116	3177	122.6

A. HuB14-VA cross-reacts only to human and cynomolgus ASCT2 (whilst not recognising mouse and rat ASCT2), recombinantly expressed in CHO cells. **B.** % of shared amino acid sequence identity of ASCT2 across human, cynomolgus monkey, mouse and rat **C.** HuB14-VA binds ASCT2 with lower affinity than ASCT17C10 (the antibody component of MEDI7247, a PBD-based ADC targeting ASCT2[5]), both in CHO cells expressing recombinant human or cyno ASCT2 and in a range of cancer cell lines.

Figure 6. HuB14-VA-PL2202 internalisation

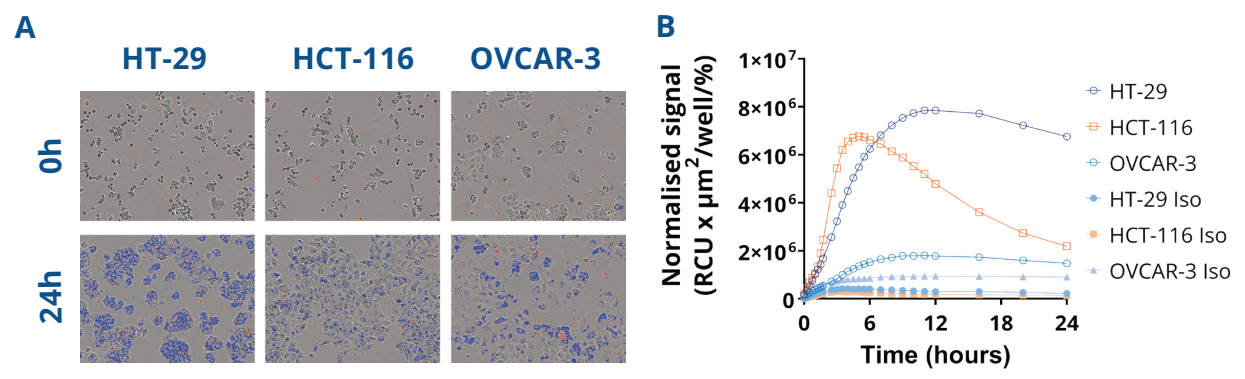
A
HT-29
HCT-116
OVCAR-3
0h
24h
B
Normalised signal
(RCU x μm²/well/%)
Time (hours)
HT-29
HCT-116
OVCAR-3
HT-29 Iso
HCT-116 Iso
OVCAR-3 Iso

A. Internalisation of FabFluor-labelled HuB14-VA-PL2202 and B12-VA-PL2202 ADCs in HT-29, HCT-116 and OVCAR-3 cells, 0h and 24h after incubation. The blue mask indicates the fluorescent signal emitted after internalisation and trafficking of the labelled ADC to the lysosomes **B.** Normalised fluorescent signal measured over time in treated cells; empty symbols indicate treatment with HuB14-VA-PL2202, whilst solid symbols indicate treatment with the non-targeting isotype control B12-VA-PL2202.

Table 1 / Figure 7. HuB14-VA cell surface binding and HuB14-VA-PL2202 *in vitro* cytotoxicity

Table 1

	SW480	HT-29	H292	HCT-116	H82	OV-CAR-3	MOLM-13	RAJI	HCC827	H69
ASCT2 status	++++	+++	++	++	++	+	+	+	+/-	+/-
HuB14-VA-PL2202 IC_{50} (pM)	275	20960	4967	6420	7800	1126	710	1791	71400	346900
B12-VA-PL2202 IC_{50} (pM)	179000	431666	95040	420600	7030	29195	22442	13671	222666	578700

Table 1. ASCT2 surface expression in a panel of cancer cell lines, measured by flow cytometry (+/- very low/neg; + low; ++ medium; +++ high; ++++ very high) and IC_{50} of HuB14-VA-PL2202 and B12-VA-PL2202 in the same cell lines, measured via cytotoxicity assay.

Figure 7

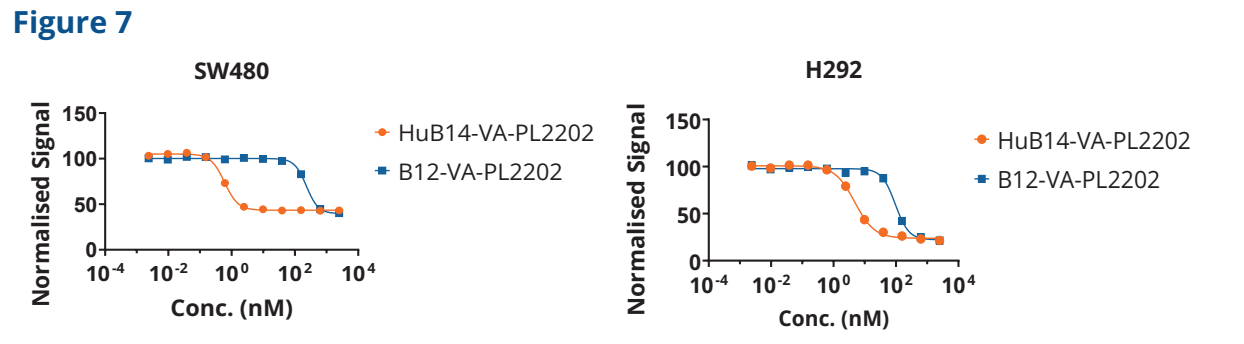
SW480
H292
Normalised Signal
Conc. (nM)
HuB14-VA-PL2202
B12-VA-PL2202

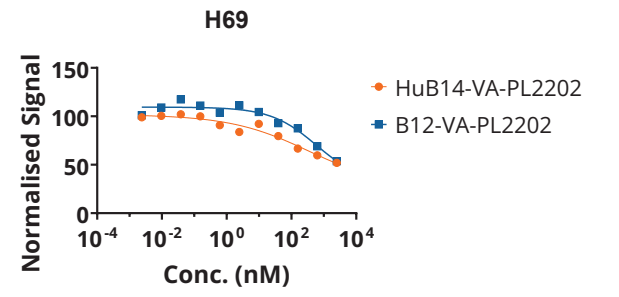
H69
Normalised Signal
Conc. (nM)
HuB14-VA-PL2202
B12-VA-PL2202

Figure 7. Representative dose-response curve of the cytotoxic activity of HuB14-VA-PL2202 and B12-VA-PL2202 in SW480, H292 and H69 cancer cell lines.

HuB14-VA-PL2202 is tolerated in cynomolgus monkeys up to 40 mg/kg

Cynomolgus monkeys are pharmacologically relevant species for HuB14-VA-PL2202 (see Fig 5 for cross-reactivity to cyno ASCT2). In a dose-range finding toxicity study in male cynomolgus monkey, HuB14-VA-PL2202 was well tolerated at the dose of 40 mg/kg IV when administered on Day 1 and 22 with minimal clinical pathology and no histopathology findings.

Figure 8. *In vivo* anti-tumor activity in the AML MOLM-13 xenograft model

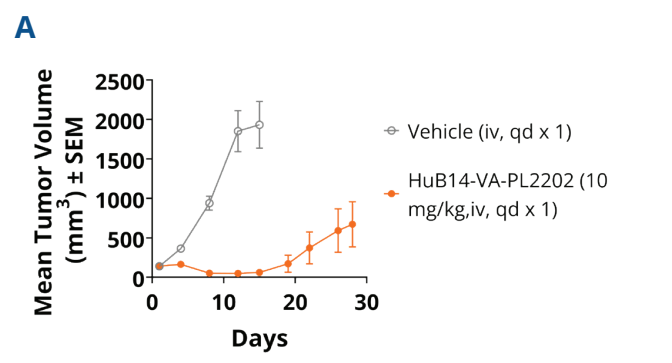
A
Mean Tumor Volume
(mm³) ± SEM
Days
Vehicle (iv, qd x 1)
HuB14-VA-PL2202 (10 mg/kg,iv, qd x 1)

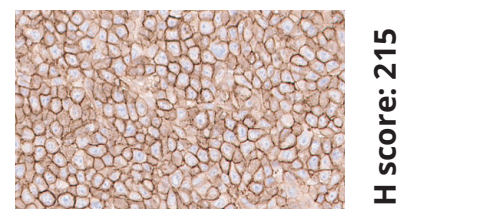
B
H score: 215

C

ADC	PR	CR	TFS
HuB14-VA-PL2202	1	6	5

A. HuB14-VA-PL2202 was administered i.v. (Day 0) to treatment group of 10 mice. A vehicle-treated group was used as control. The non-targeting control ADC B12-VA-PL2202 was dosed in previous studies without displaying any effect on tumour growth (data non shown) **B.** Representative scan of FFPE MOLM-13 tumor section stained for ASCT2 by IHC. **C.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor.

Figure 9. *In vivo* anti-tumour activity in the G-402 leiomyoblastoma xenograft model

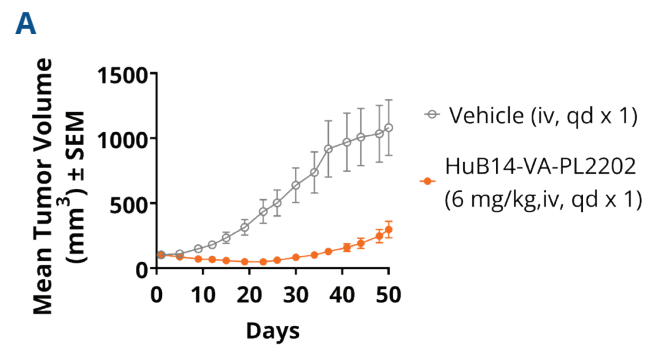
A
Mean Tumor Volume
(mm³) ± SEM
Days
Vehicle (iv, qd x 1)
HuB14-VA-PL2202
(6 mg/kg,iv, qd x 1)

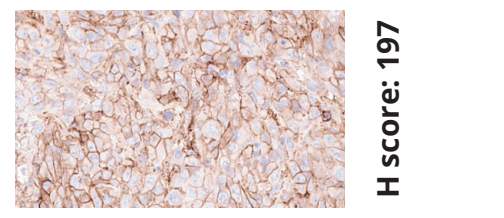
B
H score: 197

C

ADC	PR	CR	TFS
HuB14-VA-PL2202	5	1	0

A. HuB14-VA-PL2202 was administered i.v. (Day 0) to treatment group of 10 mice. A vehicle-treated group was used as control. The non-targeting control ADC B12-VA-PL2202 was dosed in previous studies without displaying any effect on tumour growth (data non shown) **B.** Representative scan of FFPE G-402 tumor section stained for ASCT2 by IHC. **C.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor.

Figure 10. *In vivo* anti-tumor activity in the colon cancer CXF 260 patient-derived xenograft model

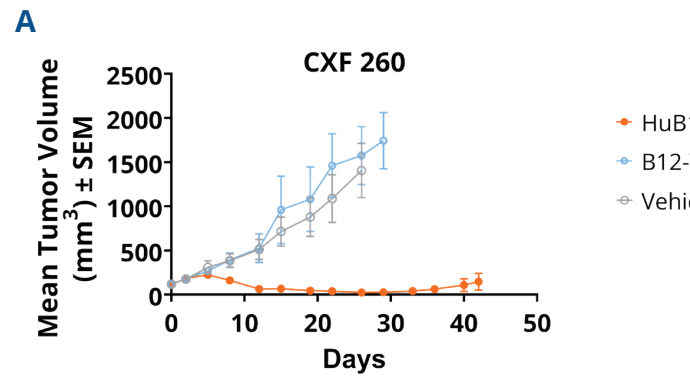
A
CXF 260
Mean Tumor Volume
(mm³) ± SEM
Days
HuB14-VA-PL2202 (10 mg/kg, iv, qdx1)
B12-VA-PL2202 (10 mg/kg, iv, qdx1)
Vehicle (iv, qdx1)

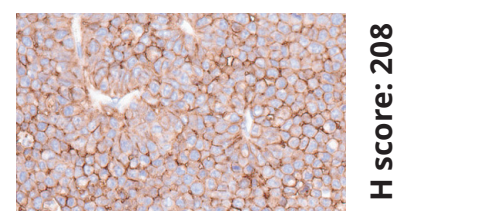
B
H score: 208

C

ADC	PR	CR	TFS
HuB14-VA-PL2202	5/5	0/5	0/5
B12-VA-PL2202	0/5	0/5	0/5

A. HuB14-VA-PL2202 and the non-targeting control B12-VA-PL2202 were administered i.v. (Day 0) to treatment group of 5 mice. A vehicle-treated group was used as control.

B. Representative scan of FFPE CXF 260 tumor section stained for ASCT2 by IHC. **C.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor.

Figure 11. HuB14-VA-PL2202 Mean Serum Concentration-Time Profiles

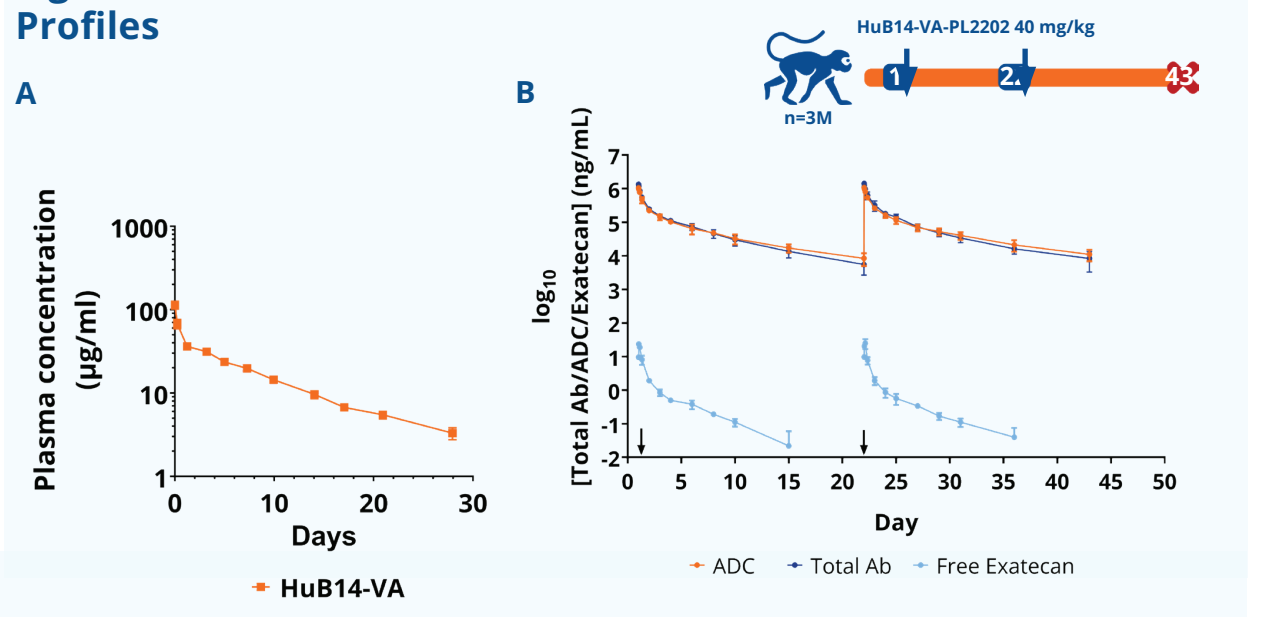
A
Plasma concentration
(μg/ml)
Days
HuB14-VA
B
HuB14-VA-PL2202 40 mg/kg
n=3M
log10
[Total Ab/ADC/Exatecan] (ng/mL)
Day
ADC
Total Ab
Free Exatecan

A. Concentration-time profile in human FcRn transgenic mice (n=4/group) following IV administration of HuB14-VA on Day 1. HuB14-VA exhibited linear PK with a $t_{½}$ of 7.4 days. **B.** Concentration-time profile in cynomolgus monkeys (n=3 males) following IV administration of HuB14-VA-PL2202 on Day 1 and 22. HuB14-VA-PL2202 exhibited linear PK with a $t_{½}$ of 6.0-6.2 days (based on data from the ADC assay); free exatecan was detectable throughout the dosing period with an apparent $t_{½}$ of 2.8-2.9 days.

References

1. Scalise M, Pochini L, Console L, Losso MA, Indiveri C. The Human SLC1A5 (ASCT2) Amino Acid Transporter: From Function to Structure and Role in Cell Biology. Front Cell Dev Biol. 2018 Sep 4;6:96. doi: 10.3389/fcell.2018.00096. PMID: 30234109; PMCID: PMC6131531.
2. Ni J, Dai W, Liu C, Ling Y, Mou H. A pan-cancer analysis of SLC1A5 in human cancers. Heliyon. 2023 Jun 23;9(6):e17598. doi: 10.1016/j.heliyon.2023.e17598. PMID: 37408893; PMCID: PMC10319225.
3. Hassanein M, Qian J, Hoeksema MD, Wang J, Jacobovitz M, Ji X, Harris FT, Harris BK, Boyd KL, Chen H, Eisenberg R, Massion PP. Targeting SLC1a5-mediated glutamine dependence in non-small cell lung cancer. Int J Cancer. 2015 Oct 1;137(7):1587-97. doi: 10.1002/ijc.29535. Epub 2015 May 6. PMID: 25821004; PMCID: PMC4640891.
4. Sun HW, Yu XJ, Wu WC, Chen J, Shi M, Zheng L, Xu J. GLUT1 and ASCT2 as Predictors for Prognosis of Hepatocellular Carcinoma. PLoS One. 2016 Dec 30;11(12):e0168907. doi: 10.1371/journal.pone.0168907. PMID: 28036362; PMCID: PMC5201247.
5. Maris M, Salles G, Kim WS, Kim TM, Lyons RM, Arellano M, Karmali R, Schiller G, Cull E, Abboud CN, Batlevi C, Kagiampakis I, Rebelatto MC, Lee Y, Kirby LC, Wang F, Bothos J, Townsley DM, Fathi AT, Ribrag V. ASCT2-Targeting Antibody-Drug Conjugate MEDI7247 in Adult Patients with Relapsed/Refractory Hematological Malignancies: A First-in-Human, Phase 1 Study. Target Oncol. 2024 May;19(3):321-332. doi: 10.1007/s11523-024-01054-z. Epub 2024 Apr 29. PMID: 38683495; PMCID: PMC11111564.

Abstract number: 1580