Preclinical Development of ADCT-241, a Novel Exatecan-based Antibody-Drug Conjugate Targeting PSMA for the Treatment of Prostate Cancer

Ben Leatherdale[1], Beccie Martin[1], Banusha Rajeenthira[1], Cecile Oblette[1], Abigail Kimpalu[1], Lara Montolio[1], Pedro L. Alves[1], Kristina Zaitseva[1], Hafsah Omar[1], Alison Faid[1], Elizabeth Horsley[1], Lina Baxter[1], Asma Jabeen[1], Valenti Gomez[1], Charlie Britten[1], Lolke de Haan[1], Paul W. Hogg[1], Chris Pickford[1], Patrick H. van Berkel[1]

[1], ADC Therapeutics UK (Ltd), London, UK

Introduction

Prostate specific membrane antigen (PSMA) is a 750 amino acid, homodimeric type II transmembrane glycoprotein expressed mainly in the prostate[1,2] and highly over-expressed in metastatic castration resistant prostate cancer (mCRPC) Whilst the role of PSMA in the prostate is not fully understood, in the small intestine it is involved in the removal of glutamate from dietary folates, and hydrolysis of the peptidic neurotransmitter N-acetylaspartyl glutamate[1,2]
PSMA is considered a clinically validated target for treatment of mCRPC with the PSMA radioligand Pluvicto® approved in 2022[3]

ADCT-241 is composed of a fully human IgG1 antibody directed against PSMA (anti-PSMA mAb), to which the PL2202 payload, containing the topoisomerase I inhibitor exatecan, has been stochastically conjugated via a cleavable valine-alanine linker with a drug:Ab ratio of 4 (DAR 4).

Figure 1.

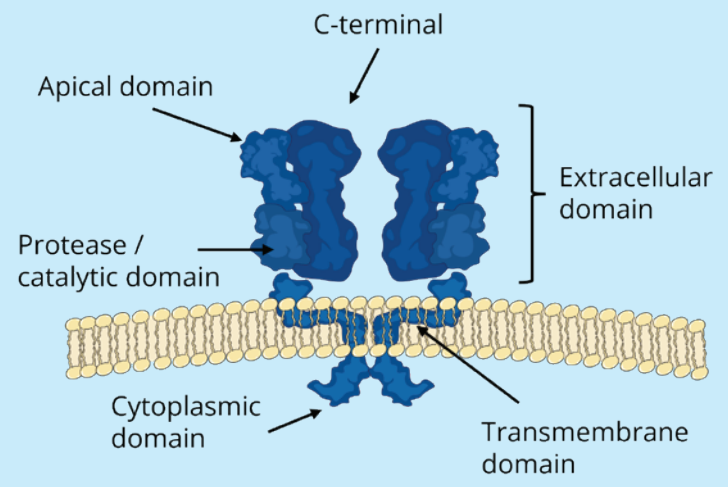
C-terminal
Apical domain
Extracellular domain
Protease / catalytic domain
Cytoplasmic domain
Transmembrane domain

Figure 2.

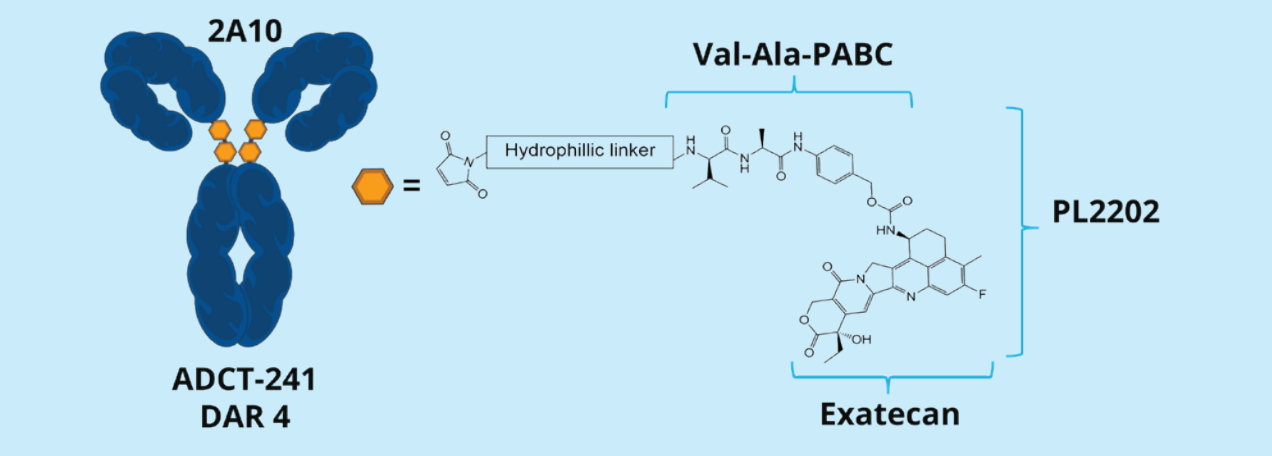
2A10
Val-Ala-PABC
Hydrophillic linker
PL2202
ADCT-241
DAR 4
Exatecan

Figure 2. ADCT-241 is a novel ADC, composed of a fully human IgG1, 2A10, directed against PSMA to which the exatecan-containing payload PL2202 has been stochastically conjugated to endogenous cysteine residues, via a cleavable valine-alanine linker, with a DAR of 4.

Aim of the study

- To investigate the *in vitro* cytotoxicity and *in vivo* anti-tumor activity of ADCT-241 in mouse xenograft models.
- To determine the potential bystander activity of ADCT-241 and compare this with a benchmark ADC, consisting of PSMA targeting huJ591 antibody conjugated via non-cleavable linker to an MMAF payload.
- Evaluation of the *in vitro* activity of the ADCT-241 and enzalutamide combination in tumor cell lines.
- To evaluate the toxicity and pharmacokinetic (PK) properties of ADCT-241 in toxicologically relevant species (cynomolgus monkey and rat).

Materials & methods

- Cross reactivity of ADCT-241 to mouse, rat, cynomolgus monkey and human PSMA was assessed by ELISA and flow cytometry using PSMA-transfected CHO cells. PSMA binding of the 2A10 antibody in prostate cancer cell lines was also assessed by flow cytometry.
- Primary and bystander cytotoxicity *in vitro* (ADCT-241, benchmark ADC & IgG1 isotype control ADC) was determined in a CellTiter 96® Aqueous One Solution Cell Proliferation assay (Promega). For the bystander assays a media transfer method was used.
- *In vivo* anti-tumor activity (ADCT-241, benchmark ADC & isotype control ADC) was evaluated following a single intravenous dose in SCID or athymic nude mice engrafted with either LNCaP or C4-2 xenografts. In addition, the anti-tumor activity of ADCT-241 was evaluated in a patient-derived xenograft (PDX) models in NSG & NOG mice following 4 weekly doses.
- Potential synergistic effects of ADCT-241 with enzalutamide were assessed in cytotoxicity CellTitreGlo® assays (Promega) using a combination matrix design. The Chou-Talalay method was used to calculate combination index values and determine synergy.
- Toxicity of ADCT-241 was evaluated in rats and cynomolgus monkeys following two intravenous doses given three weeks apart. Serum samples were taken as part of these studies to determine total antibody (conjugated and unconjugated) and ADC (conjugated Ab) concentrations using electrochemiluminescence assays (ECLIAs). In addition, serum free exatecan concentrations were determined by LC-MS/MS. Non-compartmental analysis (NCA) was performed using Phoenix WinNonlin to determine ADCT-241 PK parameters.

Acknowledgements
In vivo studies: Charles River Discovery Research Services (UK), Oncodesign Services (France), The Jackson laboratory (USA - mouse xenograft studies); Champions Oncology (USA); Labcorp (UK - cynomolgus monkey toxicity study); Syngene International (India - rat toxicity study). Lara Montolio (Graphic Design and Illustrations).

Disclosures
All authors are current employees of ADC Therapeutics and may hold stock or stock options.

Abstract number: 6736

Conclusions

- *In vitro*, ADCT-241 demonstrated specific binding and potent cytotoxicity in a panel of PSMA-expressing prostate cancer cell lines.
- Bystander killing of PSMA-negative PC-3 cells was observed when incubated with conditioned media from ADCT-241-treated cultures of PSMA-positive C4-2 and LNCaP cells, but not when incubated with conditioned media from PC-3 cells. The benchmark ADC showed no evidence of bystander killing.
- *In vivo*, ADCT-241 exhibited potent and specific anti-tumor activity after a single dose in LNCaP and C4-2 prostate cancer xenograft models. Comparable tumor suppression was observed with the benchmark ADC in the C4-2 model.
- Potent anti-tumor activity of ADCT-241 was observed when administered weekly for 4 weeks in TM00298 & CTG-2440, prostate cancer PDX models.
- *In vitro*, synergy of ADCT-241 and enzalutamide was observed in both LNCaP and C4-2 cells.
- ADCT-241 was well tolerated following two Q3W doses of up to 150 or 75 mg/kg in rats or cynomolgus monkeys.
- Total Ab and total ADC exposures were comparable, demonstrating good *in vivo* stability of ADCT-241 in both rats and cynomolgus monkeys. A $t_{½}$ of 7-10 and 8-13 days was determined for cynomolgus monkey and rat respectively.

In conclusion, the data generated are supportive of future clinical development of ADCT-241 for the treatment of prostate cancer patients.

Results

Figure 3. ADCT-241 Species Cross-reactivity

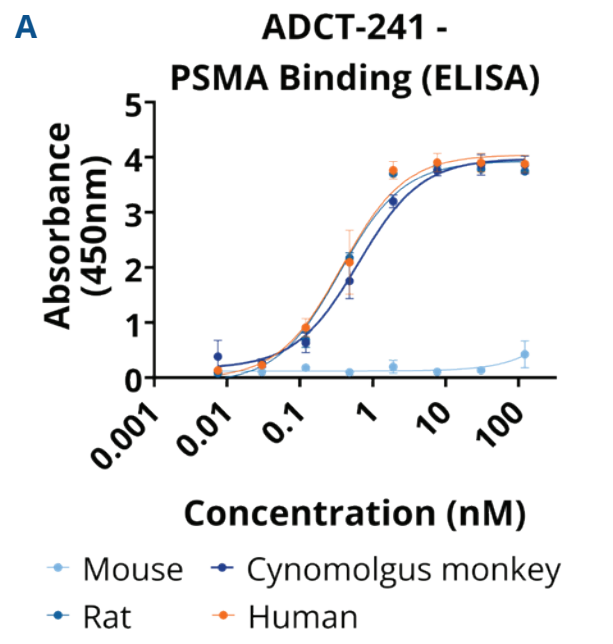
A
ADCT-241 -
PSMA Binding (ELISA)
Absorbance (450nm)
Concentration (nM)
Mouse
Cynomolgus monkey
Rat
Human

A. Graphical representation of the ADCT-241 binding capacity to different species of PSMA, mesured by ELISA. ADCT-241 shows cross reactivity with human, rat and cynomolgus monkey PSMA, but no binding was observed in mouse PSMA. **B.** Summary table of the Ec_{50} values obtained for each species.

B

ADC	EC_{50} nM			
	Mouse	Rat	Cynomolgus Monkey	Human
ADCT-241	N/A	0.35	0.63	0.37
Isotype Ctrl ADC	N/A	N/A	N/A	N/A

Figure 4. 2A10 Binding and *In Vitro* Cytotoxicity

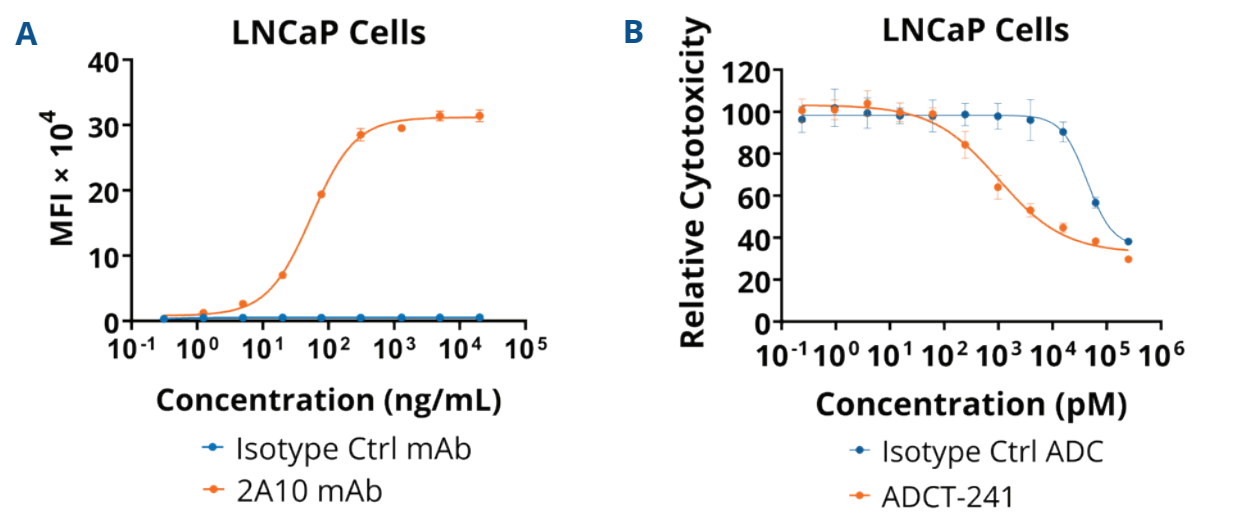
A
LNCaP Cells
MFI × 10^4
Concentration (ng/mL)
Isotype Ctrl mAb
2A10 mAb
B
LNCaP Cells
Relative Cytotoxicity
Concentration (pM)
Isotype Ctrl ADC
ADCT-241

mAb	EC_{50} ng/mL
2A10	55.65
Isotype Ctrl	no binding

mAb	EC_{50} ng/mL
ADCT-241	996.9
Isotype ADC	43155

A. The 2A10 antibody shows specific binding to LNCaP cells compared to an isotype control antibody. **B.** ADCT-241 shows specific cytotoxicity in LNCaP prostate cancer cell line when compared to an isotype ADC that uses the same exatecan-containing payload (DAR4). Cytotoxicity was assessed in a CellTitre AQ_{ueous} One-based proliferation assay.

Table 1. ADCT-241 Cytotoxicity in Panel of Prostate Cancer Cell Lines with Differential PSMA Expression

	LNCaP	C4-2	CWR22Rv1	PC3
PSMA status	****	***	*	-
ADCT-241 EC_{50} (nM)	0.495	1.03	37.0	186
Isotype Ctrl ADC EC_{50} (nM)	54.3	27.3	113.0	N/A

Table 1. PSMA surface expression in a panel of cancer cell lines, measured by flow cytometry (- neg; * low; ** medium; *** high; **** very high) and EC_{50} of ADCT-241 and Isotype Ctrl ADC in the same cell lines, measured via cytotoxicity assay.

Figure 5. ADCT-241 Bystander Activity vs Benchmark ADC

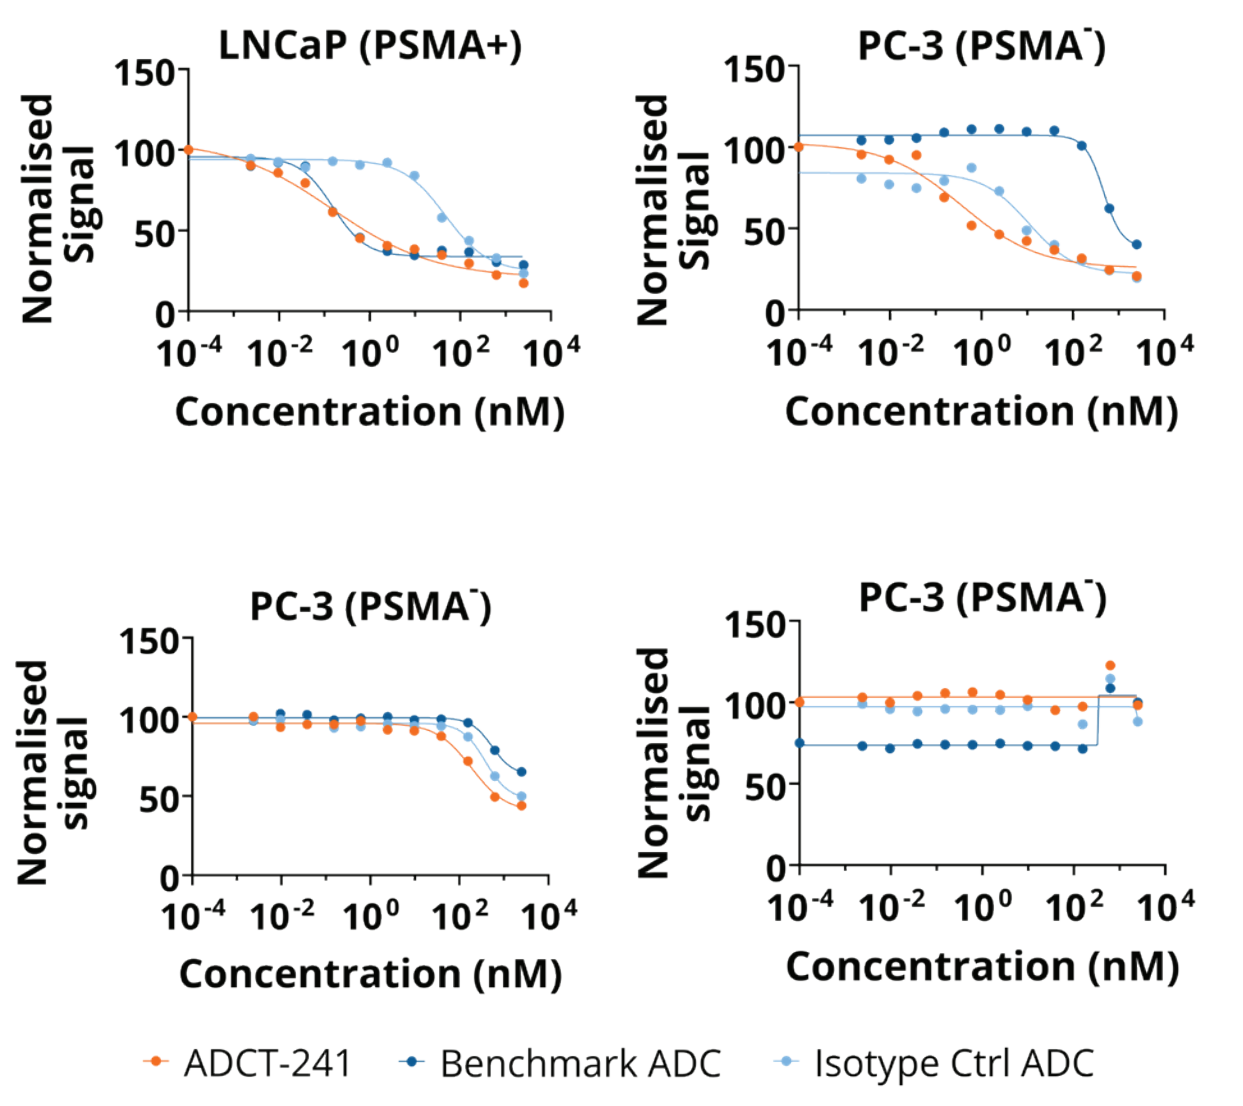
A. Primary Cytotoxicity
B. Bystander Cytotoxicity
LNCaP (PSMA+)
PC-3 (PSMA-)
PC-3 (PSMA-)
PC-3 (PSMA-)
Normalised Signal
Concentration (nM)
ADCT-241
Benchmark ADC
Isotype Ctrl ADC

A. Representative dose-response curve of the primary cytotoxic activity of ADCT-241, benchmark ADC and Isotype Ctrl ADC in PSMA positive cancer cell line (LnCaP) and PSMA negative cancer cell line (PC-3). **B.** Representative dose-response curve of the Bystander cytotoxic activity of ADCT-241, Benchmark ADC and Isotype Ctrl ADC in negative PSMA cancer cell lines (PC-3).

Figure 6. ADCT-241 anti-tumor activity in LNCaP prostate xenograft model

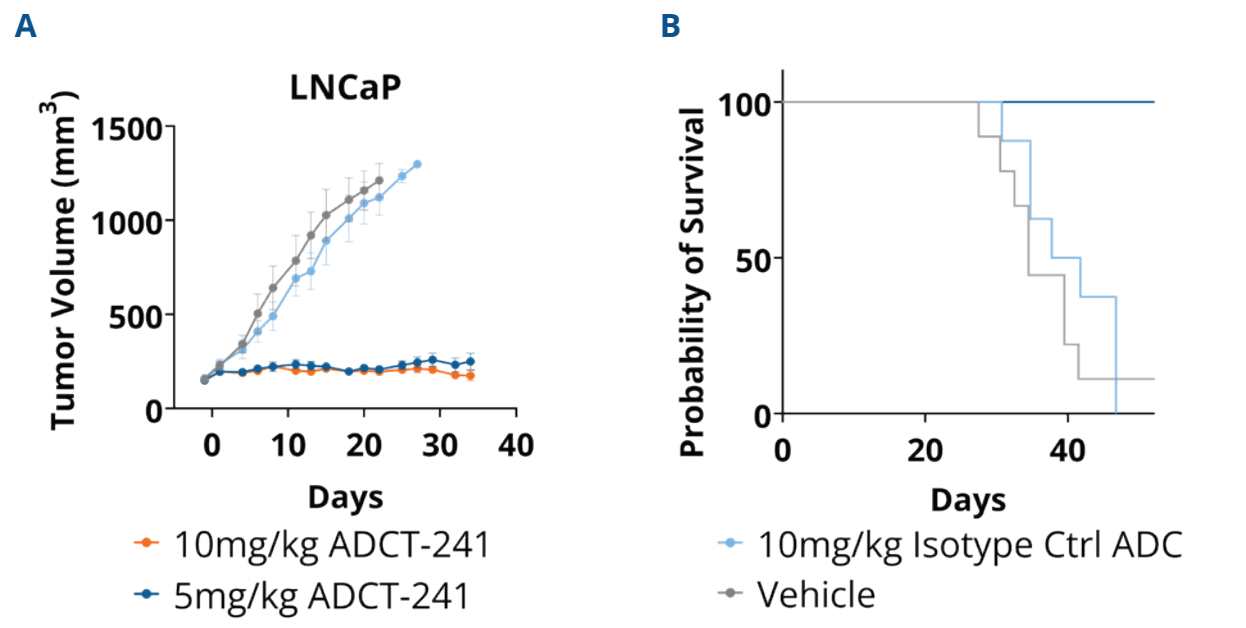
A
LNCaP
Tumor Volume (mm^3)
Days
10mg/kg ADCT-241
5mg/kg ADCT-241
B
Probability of Survival
Days
10mg/kg Isotype Ctrl ADC
Vehicle

C

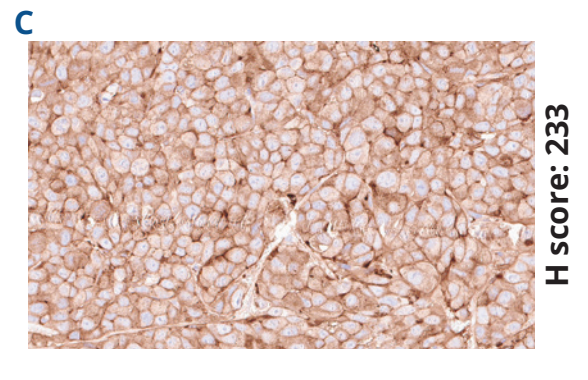
H score: 233

A. ADCT-241 and isotype control ADC were administered as single i.v. dose (day 0) in treatment groups of 10 mice. Vehicle control group was also included. **B.** Kaplan-Meier analysis of survival. **C.** Representative scan of FFPE LNCaP tumor section stained for PSMA by IHC.

Figure 7. ADCT-241 *in vivo* anti-tumor activity in C4-2 prostate xenograft model

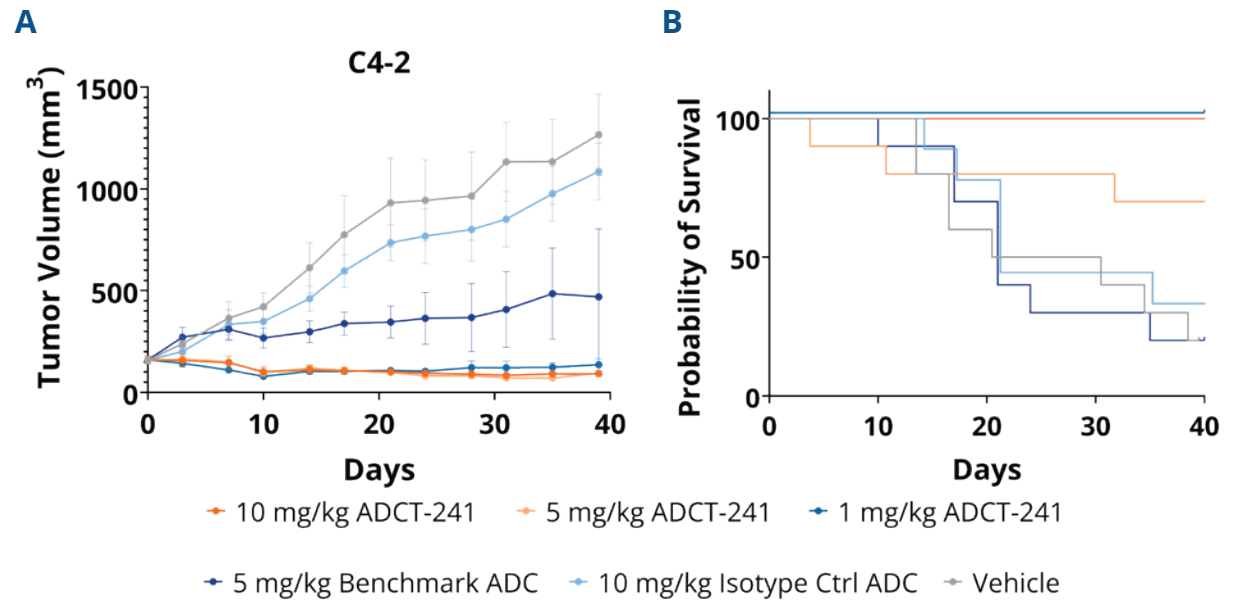
A
C4-2
Tumor Volume (mm^3)
Days
B
Probability of Survival
Days
10 mg/kg ADCT-241
5 mg/kg ADCT-241
1 mg/kg ADCT-241
5 mg/kg Benchmark ADC
10 mg/kg Isotype Ctrl ADC
Vehicle

C

ADC	PR	CR	TFS
Vehicle	0/10	0/10	0/10
10 mg/kg Isotype Ctrl ADC	0/9	0/9	0/9
10 mg/kg ADCT-241	4/10	0/10	0/10
5 mg/kg ADCT-241	3/7	0/7	0/7
5 mg/kg benchmark ADC	2/10	0/10	0/10
1 mg/kg ADCT-241	0/10	0/10	0/10

D

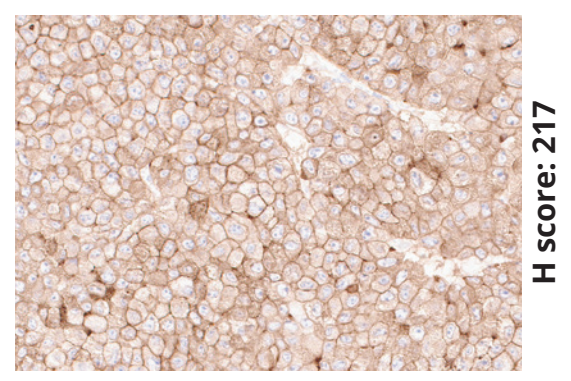
H score: 217

A. ADCT-241 and isotype control ADC were administered as single i.v. dose (day 0) in treatment groups of 10 mice. Vehicle control group was also included. **B.** Kaplan-Meier analysis of survival. **C.** Representative scan of FFPE C4-2 tumor section stained for PSMA by IHC. **D.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor.

Figure 8. ADCT-241 efficacy in TM00298 PDX model

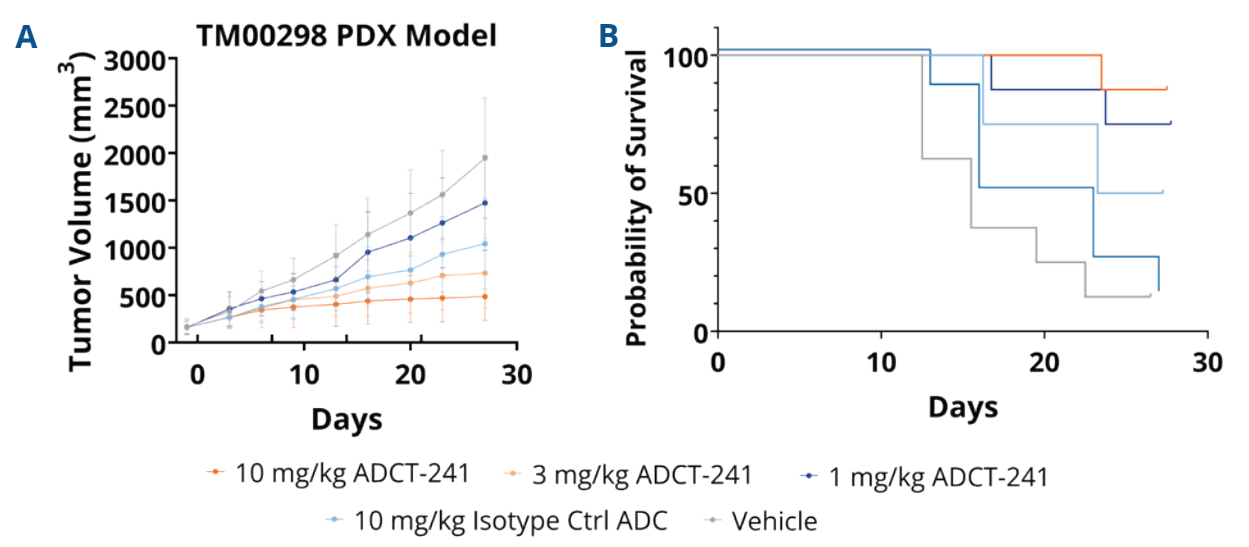
A
TM00298 PDX Model
Tumor Volume (mm^3)
Days
B
Probability of Survival
Days
10 mg/kg ADCT-241
3 mg/kg ADCT-241
1 mg/kg ADCT-241
10 mg/kg Isotype Ctrl ADC
Vehicle

C

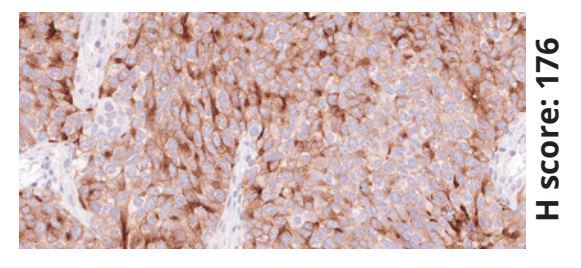
H score: 176

A. ADCT-241 and isotype control ADC were administered weekly over 4 weeks in treatment groups of 8 mice. Vehicle control group was also included.
B. Kaplan-Meier analysis of survival.
C. Representative scan of FFPE TM00298 tumor section stained for PSMA by IHC.

Figure 9. ADCT-241 efficacy in CTG-2440 PDX model

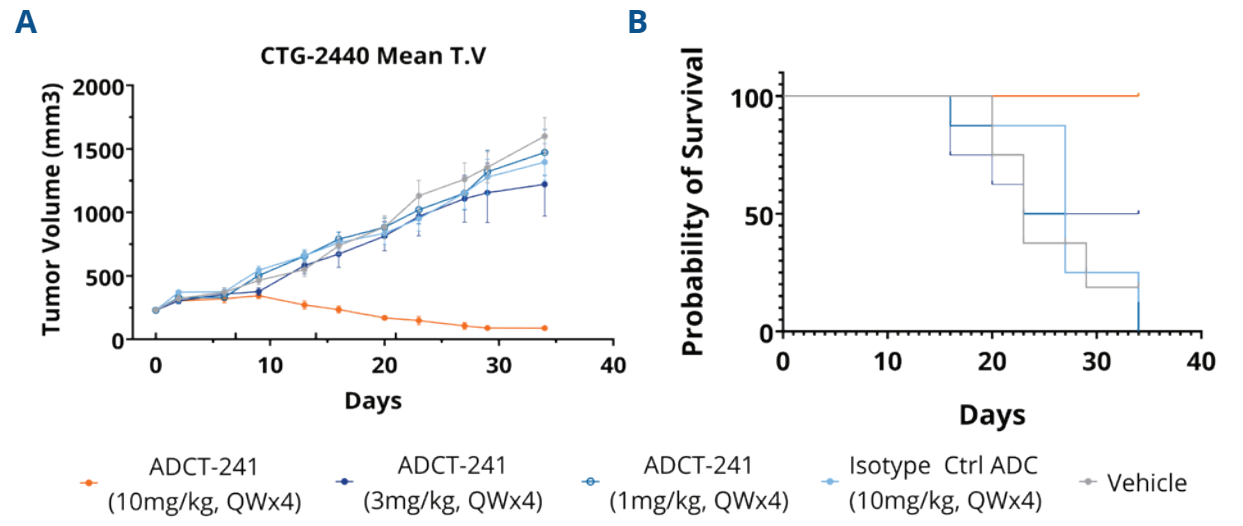
A
CTG-2440 Mean T.V
Tumor Volume (mm3)
Days
B
Probability of Survival
Days
ADCT-241 (10mg/kg, QWx4)
ADCT-241 (3mg/kg, QWx4)
ADCT-241 (1mg/kg, QWx4)
Isotype Ctrl ADC (10mg/kg, QWx4)
Vehicle

C

ADC	PR	CR	TFS
Vehicle	0/8	0/8	0/8
10 mg/kg Isotype Ctrl ADC	0/8	0/8	0/8
10 mg/kg ADCT-241	4/8	1/8	1/8
3 mg/kg ADCT-241	0/8	0/8	0/8
1 mg/kg ADCT-241	0/8	0/8	0/8

D

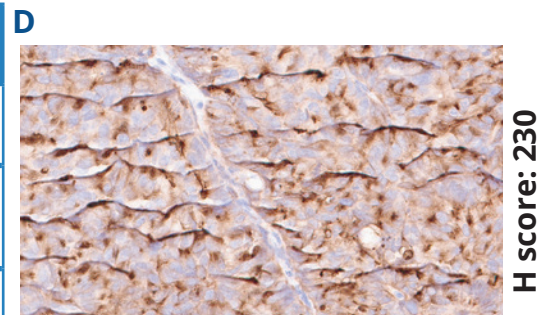
H score: 230

A. ADCT-241 and isotype control ADC were administered weekly over 4 weeks in treatment groups of 8 mice. Vehicle control group was also included. **B.** Kaplan-Meier analysis of survival. **C.** Response summary PR, partial response; CR, complete response; TFS, tumor-free survivor. **D.** Representative scan of FFPE CTG-2440 tumor section stained for PSMA by IHC.

Figure 10. ADCT-241 Mean Serum Concentration-Time Profiles from Rat Toxicology Study (Combined Sexes)

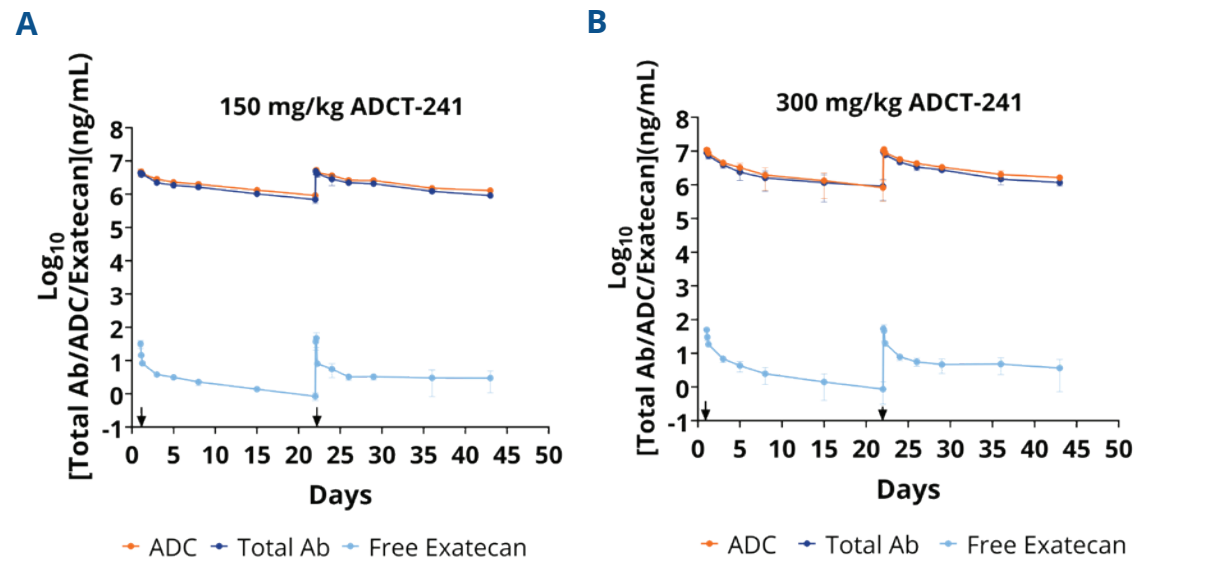
A
150 mg/kg ADCT-241
B
300 mg/kg ADCT-241
Log10 [Total Ab/ADC/Exatecan](ng/mL)
Days
ADC
Total Ab
Free Exatecan

PK in rat following IV administration of ADCT-241 on Day 1 and 22 (n=3/group).
A. ADCT-241 given at 150 mg/kg Q3W2 was well tolerated. **B.** Administration at 300 mg/kg exceeded the maximum tolerated dose and was associated with adverse clinical signs and 1 unscheduled termination. **A&B.** Show Linear PK with $t_{½}$ ~of 8-13 days based on data from the ADC assay. Free exatecan detectable across the dosing interval with an apparent $t_{½}$ of 6-10 days.

Figure 11. ADCT-241 Mean Serum Concentration-Time Profiles from Cynomolgus Monkey Study (Combined Sexes)

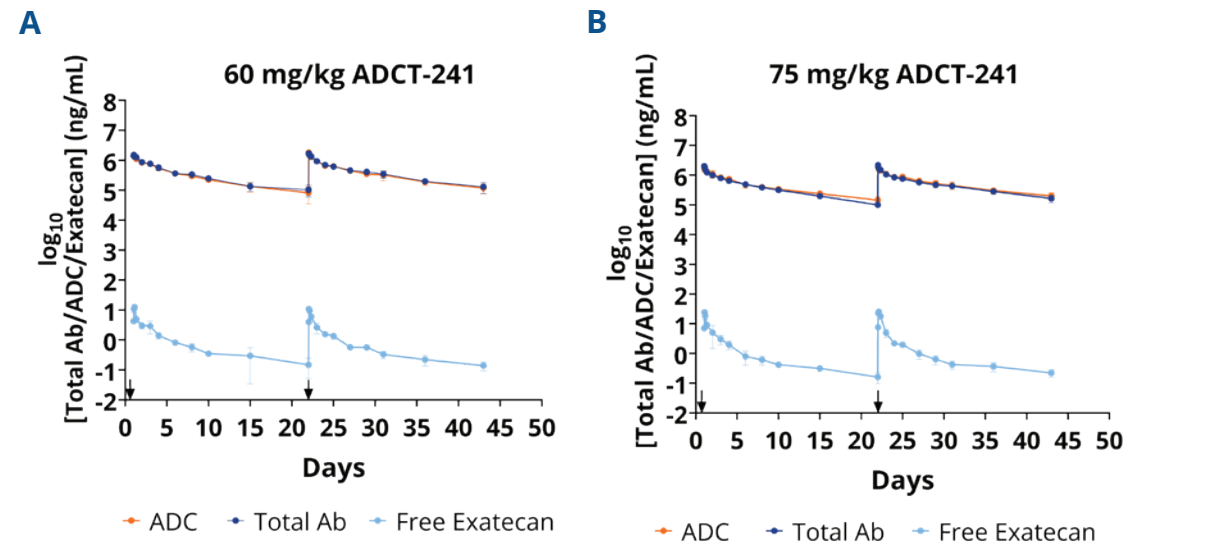
A
60 mg/kg ADCT-241
B
75 mg/kg ADCT-241
log10 [Total Ab/ADC/Exatecan] (ng/mL)
Days
ADC
Total Ab
Free Exatecan

PK in Cynomolgus Monkey following IV administration of ADCT-241 on day 1 and 22 (was detectable across the dosing interval=3/group). **A.** ADCT-241 given at 60 mg/kg Q3W2 was well tolerated. **B.** Administration at 75 mg/kg was well tolerated representing a No Observed Adverse Effect Level. **A&B.** Linear PK with no accumulation after repeat dosing, and a $t_{½}$ of ~7-10 days based on data from the ADC assay.

Figure 12. ADCT-241 Shows Synergistic Activity in Combination with Enzalutamide

A

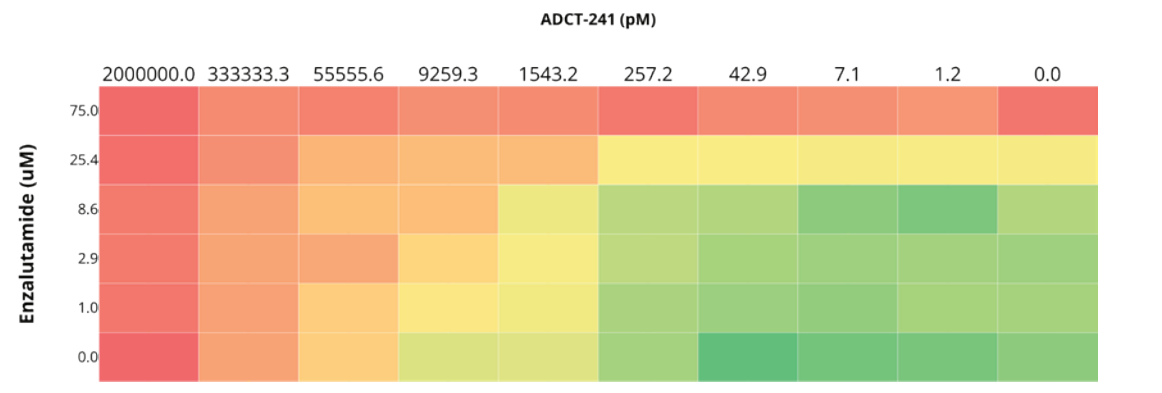
ADCT-241 (pM)
2000000.0
333333.3
55555.6
9259.3
1543.2
257.2
42.9
7.1
1.2
0.0
Enzalutamide (uM)
75.0
25.4
8.6
2.9
1.0
0.0

B

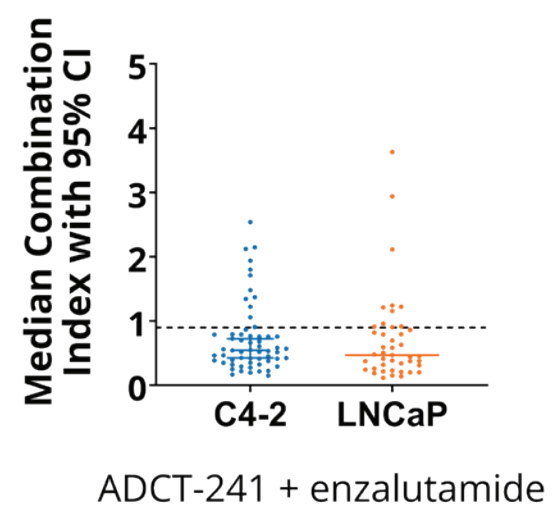
Median Combination Index with 95% CI
C4-2
LNCaP
ADCT-241 + enzalutamide

C

Cells	ADCT-241 + enzalutamide	
	Median Combination Index	95% Confidence Interval
C4-2	0.54	0.42-0.72
LNCaP	0.47	0.37-0.79

A. ADCT-241 and enzalutamide combination matrix design. Individual drugs and 45 dose combinations were tested on each cell line. **B.** Distribution of Chou-Talalay Combination Index (CI) values obtained following incubation of C4-2 and LNCaP cell lines with ADCT-241 and enzalutamide. For each cell line, the horizontal line indicates the median CI, with the whiskers representing the 95% confidence interval values. The dotted horizontal line indicates the threshold for synergy. **C.** Table summarizing median CI values with 95% confidence interval values.

References
1. Ristau BT, O'Keefe DS, Bacich DJ. The prostate-specific membrane antigen: lessons and current clinical implications from 20 years of research. Urol Oncol. 2014 Apr;32(3):272-9. doi: 10.1016/j.urolonc.2013.09.003. Epub 2013 Dec 8. PMID: 24321253; PMCID: PMC3972351.
2. Cho et al. Antitumor Activity of MEDI3726 (ADCT-401), a Pyrrolobenzodiazepine Antibody-Drug Conjugate Targeting PSMA, in Preclinical Models of Prostate Cancer. Mol Cancer Ther. 2018 Oct;17(10):2176-2186. doi: 10.1158/1535-7163.MCT-17-0982. Epub 2018 Jul 31. PMID: 30065100.
3. Fallah et al. FDA Approval Summary: Lutetium Lu 177 Vipivotide Tetraxetan for Patients with Metastatic Castration-Resistant Prostate Cancer. Clin Cancer Res. 2023 May 1;29(9):1651-1657. doi: 10.1158/1078-0432.CCR-22-2875. PMID: 36469000; PMCID: PMC10159870.