PS1911

Initial Results From LOTIS-7: A Phase 1b Study of Loncastuximab Tesirine Plus Glofitamab in Patients With Relapsed/Refractory (R/R) Diffuse Large B-Cell Lymphoma (DLBCL)

Juan Pablo Alderuccio,[1]* Craig Okada,[2] Gilles Crochet,[3] Emily Ayers,[4] Marie Hu,[5] Silvia Ferrari,[6] Paolo Caimi,[7] Mehdi Hamadani,[8] Julien Depaus,[9] Enrico Derenzini,[10] Jose Sandoval-Sus,[11] Ruben Reyes,[12] Andrzej Urban,[13] Erica Rave,[12] Adina Graf,[13] Eva Tiecke,[13] Marie Toukam,[12] Pier Luigi Zinzani[14]

[1]Sylvester Comprehensive Cancer Center, University of Miami Leonard M. Miller School of Medicine, Miami, FL, USA; [2]Oregon Health & Science University, Portland, OR, USA; [3]Hematology Department, Centre Hospitalier Universitaire (CHU) Université Catholique de Louvain (UCL) Namur, Yvoir, Belgium; [4]Division of Hematology & Oncology, University of Virginia, Charlottesville, VA, USA; [5]Division of Hematology, Oncology and Transplantation, Department of Medicine, University of Minnesota, Minneapolis, MN, USA; [6]Hematology and Bone Marrow Transplant Unit, Azienda Ospedaliera Papa Giovanni XXIII, Bergamo, Italy; [7]Cleveland Clinic Taussig Cancer Center, Cleveland, OH, USA; [8]Division of Hematology and Oncology, Medical College of Wisconsin, Milwaukee, WI, USA; [9]Department of Hematology, CHU UCL Namur Site Godinne, Yvoir, Belgium; [10]IEO Istituto Europeo di Oncologia, Milan, Italy; [11]Moffitt Malignant Hematology & Cellular Therapy at Memorial Healthcare System, Pembroke Pines, FL, USA; [12]ADC Therapeutics America, Inc., New Providence, NJ, USA; [13]ADC Therapeutics SA, Épalinges, Switzerland; [14]Institute of Hematology "Seràgnoli," University of Bologna, Bologna, Italy

OBJECTIVE

The primary objective is to characterize the safety, tolerability, and anticancer activity of Lonca in combination with other anticancer agents, including Glofit, and to identify the maximum tolerated dose or the recommended dose for expansion. Secondary objectives are to evaluate the antitumor effects of Lonca + Glofit, and to characterize the PK and immunogenicity of Lonca and Glofit, as measured by antidrug antibodies.

PHASE 1b LOTIS-7 TRIAL

Eligibility criteria

- **Part 1:** R/R DLBCL, HGBCL, trFL, FL (all grades), or MZL and ≥2 prior lines of therapy
- **Part 2:** R/R de novo DLBCL, HGBCL, trFL, or FL (grade 3b) and ≥1 prior line of therapy

Part 1: Dose escalation
Lonca + Glofit | Lonca + other anticancer agents

Part 2: Dose expansion
Lonca + Glofit

INITIAL SAFETY AND EFFICACY RESULTS

MANAGEABLE SAFETY PROFILE (N=41)

- The most common grade ≥3 TEAEs was neutropenia (n=10, 24.4%)
- Grade 3/4 TEAEs of interest included generalized edema (4.9%), rash (2.4%), and photosensitivity reaction (2.4%)
- One patient (2.4%) demonstrated grade 3 CRS, and no patients had grade 4 CRS; rates were numerically lower in the Lonca 150-µg/kg than the 120-µg/kg dose group

EFFICACY (N=30)

ORR: 93.3% (28/30)

CR: 86.7%

PR: 6.7%

Median DOR: not reached
Probability to maintain event free for 6 months: 95.2%

PK/BIOMARKER RESULTS

- Lonca + Glofit showed comparable exposure but lower maximum concentration vs Lonca monotherapy
- No post-dose Lonca antidrug antibodies were detected with Lonca + Glofit
- T-cell activation was appreciable, and circulating activated CD4+ and CD8+ T cells increased during treatment

CONCLUSIONS

To date, Lonca + Glofit in 2L+ B-NHL has shown a clinically meaningful CR rate (86.7%) in heavily pretreated patients with LBCL, with responses sustained beyond the end of treatment. Given the shorter time to CR and lower rates of CRS in the Lonca 150-µg/kg dose group than the 120-µg/kg dose group, further expansion at the Lonca 150-µg/kg dose is planned. These encouraging early findings suggest that the Lonca + Glofit combination has a manageable safety profile and robust early clinical benefit.

2L+, second line or later; B-NHL, B-cell non-Hodgkin lymphoma; CR, complete response; CRS, cytokine release syndrome; DLBCL, diffuse large B-cell lymphoma; DOR, duration of response; FL, follicular lymphoma; Glofit, glofitamab; HGBCL, high-grade B-cell lymphoma; LBCL, large B-cell lymphoma; Lonca, loncastuximab tesirine; MZL, marginal zone lymphoma; ORR, overall response rate; PK, pharmacokinetics; PR, partial response; R/R, relapsed/refractory; TEAE, treatment-emergent adverse event; trFL, transformed follicular lymphoma.

Copies of this poster obtained through the Quick Response (QR) Code are for personal use only and may not be reproduced without permission from the author of this poster.

Poster presented at the European Hematology Association 30th Annual Congress (EHA 2025).
June 12-15, 2025, Milan, Italy

CONCLUSIONS

- This multinational, phase 1b, 2-part, open-label trial (LOTIS-7; NCT04970901) evaluates the safety, tolerability, and anticancer activity of loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]) in combination with other anticancer agents, including glofitamab (Glofit), in patients with relapsed/refractory (R/R) B-cell non-Hodgkin lymphoma (B-NHL)
- Lonca + Glofit in second-line or later (2L+) large B-cell lymphoma (LBCL) demonstrated a manageable safety profile and robust efficacy (overall response rate [ORR], 93.3%; complete response [CR] rate, 86.7%), with the median time to first CR being shorter in the 150-µg/kg than the 120-µg/kg dose group (42.0 vs 80.0 days, respectively)
- No grade 5 treatment-emergent adverse events (TEAEs) occurred, and rates of cytokine-release syndrome (CRS) and immune effector cell-associated neurotoxicity syndrome (ICANS) were low, with no grade ≥3 ICANS events
 - Any-grade CRS was less frequent at the 150-µg/kg (23.8%) than at 120-µg/kg (55.0%) dose group and was lower than the rate previously reported for Glofit monotherapy (63%)[1]
- These encouraging early findings suggest that the Lonca + Glofit combination has a manageable safety profile and robust clinical benefit, which warrants further evaluation
- Enrollment is expanding at the Lonca 150-µg/kg dose to include additional patients

INTRODUCTION

- Loncastuximab tesirine (loncastuximab tesirine-lpyl [Lonca]), an antibody–drug conjugate comprising a humanized CD19-targeted antibody conjugated to a pyrrolobenzodiazepine dimer (PBD) cytotoxin, and glofitamab (Glofit), a CD20×CD3 T-cell–engaging, bispecific antibody, are both approved for third-line or later (3L+) R/R DLBCL by the United States Food and Drug Administration, the European Medicines Agency, and other national regulatory authorities.[2-9]
- The pivotal phase 2 LOTIS-2 trial of Lonca monotherapy in patients with R/R DLBCL showed that an intravenous (IV) infusion over 30 minutes on day 1 of each 3-week (Q3W) cycle produced durable responses with manageable toxicity using a dose of 150 µg/kg for 2 cycles and then 75 µg/kg for subsequent cycles[10]
- Combining agents with different mechanisms of action may enhance treatment efficacy in patients with R/R B-NHL
 - Combining CD20×CD3 T-cell–engaging antibodies, including Glofit, with the CD19-targeting antibody–drug conjugate Lonca is expected to increase antitumor activity[11]
- This LOTIS-7 analysis evaluates the safety and efficacy of Lonca (120 and 150 µg/kg) + Glofit in patients with 2L+ LBCL

OBJECTIVE

- To characterize the safety, efficacy, and pharmacokinetic (PK) profile of Lonca + Glofit; identify the maximum tolerated/recommended dose for expansion; and explore correlations between tumor tissue, biomarkers, PK parameters, and clinical activity

METHODS

Study Design

- This phase 1b, open-label, multicenter, multiarm trial (NCT04970901) is divided into 2 parts (part 1: dose escalation at 90, 120, and 150 µg/kg; part 2: dose expansion at 120 and 150 µg/kg) and is enrolling patients with third line or later (3L+) R/R B-NHL (part 1) and second line or later (2L+) R/R LBCL (part 2) (**Figure 1**)[12]
- Eligible patients will have either failed or are intolerant to any approved therapy and have received ≥2 (part 1) or ≥1 (part 2) prior systemic treatments
- The treated population received Lonca 120 or 150 µg/kg (part 1 and 2) Q3W for a fixed duration of up to 8 cycles; Lonca doses of 120 and 150 µg/kg were reduced to 75 µg/kg for cycles ≥3 per label
 - After an initial pretreatment with obinutuzumab at 1000 mg on cycle 1, day 1, Lonca was administered on day 2; then Glofit was administered on day 8 at 30 mg Q3W with step-up dosing for up to 12 cycles
- A total of 47 patients with R/R B-NHL were treated at the 90-, 120-, or 150-µg/kg dose level
- The treated population reported in this presentation (N=41) included all patients from parts 1 and 2 who received Lonca 120 µg/kg or 150 µg/kg and had LBCL histologies, including R/R de novo DLBCL, high-grade B-cell lymphoma (HGBCL), transformed follicular lymphoma (trFL), or FL (grade ≥3b)
 - The efficacy evaluable population (N=30), as defined in this presentation, included all patients who received ≥1 dose of study drug and had a baseline and ≥1 postbaseline disease assessment

Figure 1. Study design and dosing schedule

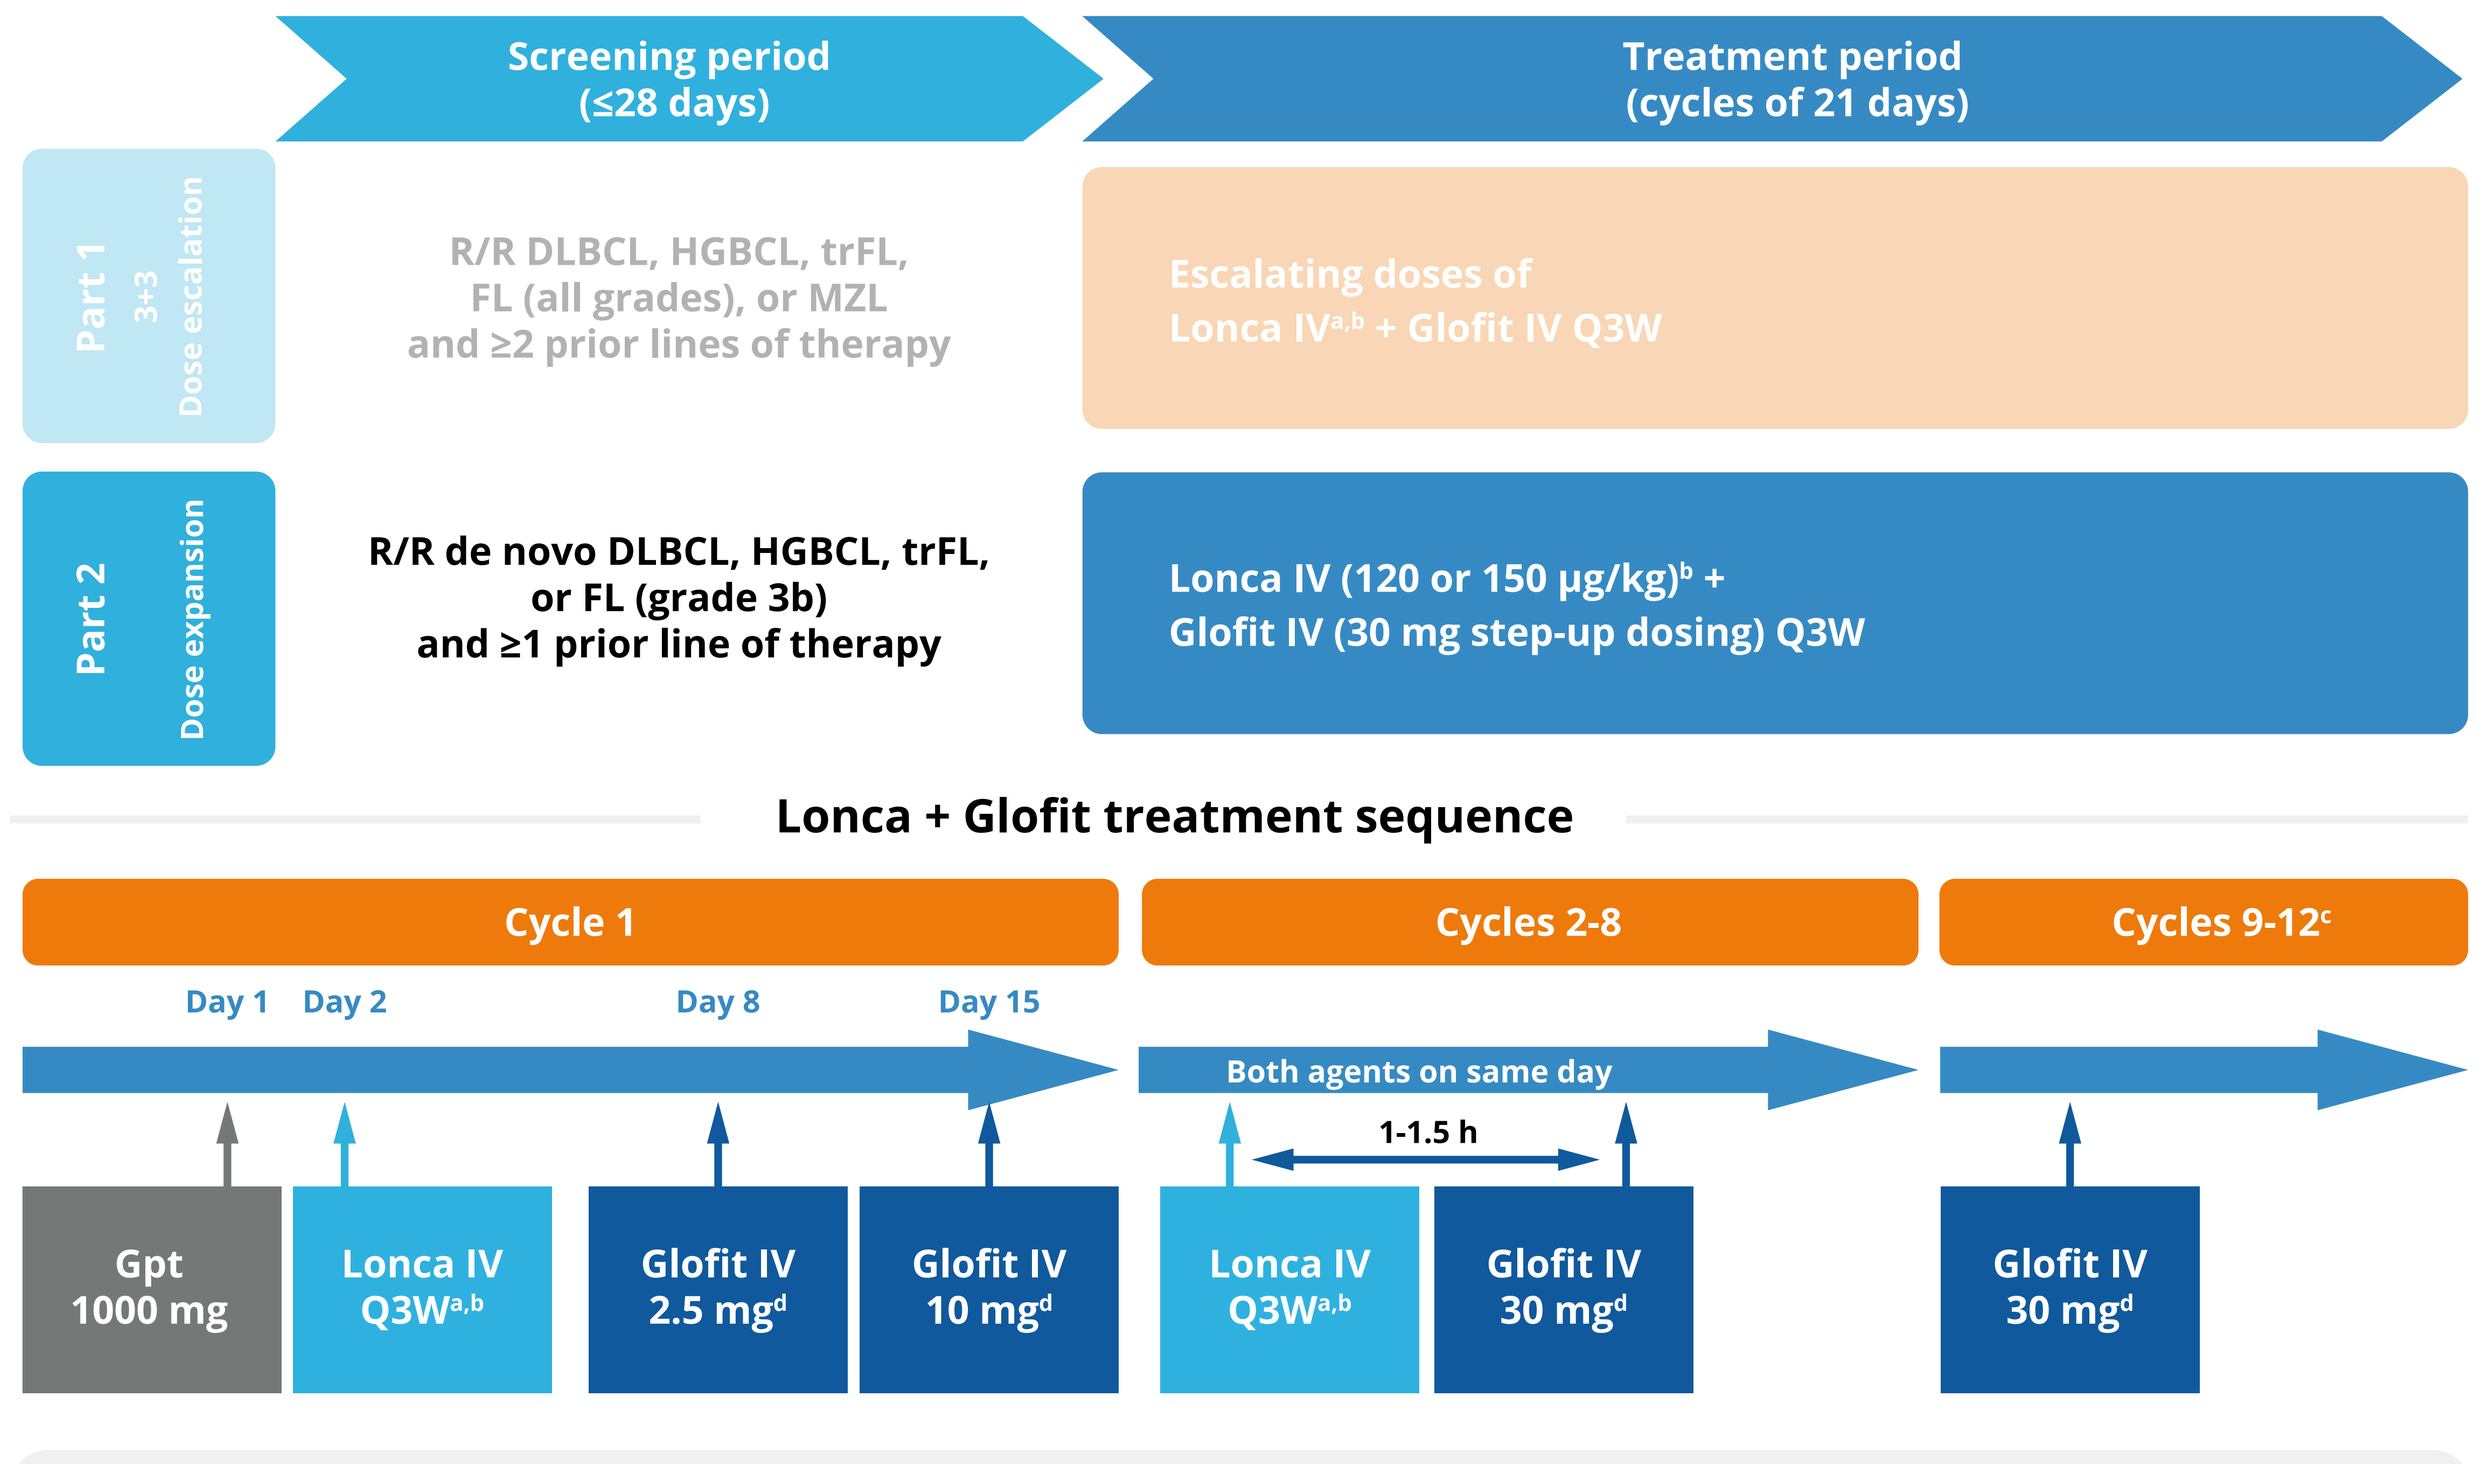
Screening period
(≤28 days)
Treatment period
(cycles of 21 days)
Part 1
3+3
Dose escalation
R/R DLBCL, HGBCL, trFL,
FL (all grades), or MZL
and ≥2 prior lines of therapy
Escalating doses of
Lonca IVa,b + Glofit IV Q3W
Part 2
Dose expansion
R/R de novo DLBCL, HGBCL, trFL,
or FL (grade 3b)
and ≥1 prior line of therapy
Lonca IV (120 or 150 µg/kg)b +
Glofit IV (30 mg step-up dosing) Q3W
Lonca + Glofit treatment sequence
Cycle 1
Cycles 2-8
Cycles 9-12c
Day 1
Day 2
Day 8
Day 15
Both agents on same day
1-1.5 h
Gpt
1000 mg
Lonca IV
Q3Wa,b
Glofit IV
2.5 mgd
Glofit IV
10 mgd
Lonca IV
Q3Wa,b
Glofit IV
30 mgd
Glofit IV
30 mgd

Study population

- Patients with 3L+ R/R B-NHL (part 1) and 2L+ R/R LBCL (part 2)
- ECOG PS 0-2
- ≥2 (part 1) or ≥1 (part 2) systemic treatments
- Prior autologous SCT (>100 days) or CAR-T therapy (>100 days) is allowed
- Measurable disease (per 2014 Lugano Classification[8])
- Excludes patients with clinically significant third-space fluid accumulation

Endpoints

- Primary: safety and tolerability; MTD and/or RDE
- Secondary: ORR, DOR, CR rate, PFS, RFS, and OS; PK and immunogenicity
- Exploratory: concentration of Glofit in circulation; association between tumor tissue and/or blood biomarkers and PK measures with clinical endpoints

2L+, second line or later; 3L+, third line or later; B-NHL, B-cell non-Hodgkin lymphoma; CAR-T, chimeric antigen receptor T cell; CR, complete response; CRS, cytokine release syndrome; DLBCL, diffuse large B-cell lymphoma; DOR, duration of response; ECOG PS, Eastern Cooperative Oncology Group Performance Status; FL, follicular lymphoma; Glofit, glofitamab; Gpt, obinutuzumab; HGBCL, high-grade B-cell lymphoma; IV, intravenous; LBCL, large B-cell lymphoma; Lonca, loncastuximab tesirine; MTD, maximum tolerated dose; MZL, marginal zone lymphoma; NOS, not otherwise specified; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PK, pharmacokinetic; Q3W, every 3 weeks; RDE, recommended dose for expansion; RFS, relapse-free survival; R/R, relapsed or refractory; SCT, stem cell transplant; trFL, transformed follicular lymphoma.
[a]Dose level 1, 90 µg/kg; dose level 2, 120 µg/kg; and dose level 3, 150 µg/kg.
[b]If the starting dose of Lonca is 120 µg/kg or 150 µg/kg, the dose will be reduced to 75 µg/kg for cycles ≥3 per label.
[c]Participants may continue Glofit for up to 12 cycles or until disease progression, unacceptable toxicity, or other discontinuation criteria, whichever occurs first. The follow-up period is for ≤2 years from the end of treatment.
[d]The first dose of Glofit on cycle 1, day 8 has a 24-hour mandatory hospitalization; subsequent doses require hospitalization if grade ≥2 CRS occurs.

RESULTS

Patient Population

- As of 14 April 2025, 47 patients with R/R B-NHL had received ≥1 dose of treatment at the Lonca 90-, 120-, or 150-µg/kg dose level
 - Of these, 41 patients with LBCL were treated at Lonca 120 or 150 µg/kg and comprised the treated population for this presentation; 30 were evaluable for efficacy
 - Six patients who received Lonca 90 µg/kg or had another B-NHL histology (ie, FL grades 1-3a or marginal zone lymphoma) were excluded from the present analysis
- Among the treated population, the median age was 71 years, 73.2% had de novo DLBCL, 51.2% received ≥2 prior therapies, and 19.5% had received prior CAR-T therapy (**Table 1**)

Table 1. Baseline characteristics, treated population (N=41)

Characteristics	Glofit + Lonca, 120 µg/kg (n=20)	Glofit + Lonca, 150 µg/kg (n=21)	All dose levels (N=41)
Age, years, median (range)	70 (50-82)	74 (26-85)	71 (26-85)
Sex, male, n (%)	11 (55.0)	12 (57.1)	23 (56.1)
ECOG PS score, n (%)			
0	9 (45.0)	14 (66.7)	23 (56.1)
1	10 (50.0)	7 (33.3)	17 (41.5)
2	1 (5.0)	0	1 (2.4)
Ann Arbor Disease stage, n (%)			
Stage I/II	3 (15.0)	3 (14.3)	6 (14.6)
Stage III/IV	17 (85.0)	18 (85.7)	35 (85.4)
IPI score, n (%)			
0-2	9 (45.0)	10 (47.6)	19 (46.3)
3-5	11 (55.0)	11 (52.4)	22 (53.7)
Bulky disease, n (%)	2 (10.0)	2 (9.5)	4 (9.8)
LDH levels high, n (%)	11 (55.0)	10 (47.6)	21 (51.2)
LBCL histology, n (%)			
de novo DLBCL	13 (65.0)	17 (81.0)	30 (73.2)
HGBCL	4 (20.0)	2 (9.5)	6 (14.6)
trFL	2 (10.0)	2 (9.5)	4 (9.8)
FL grade 3b	1 (5.0)	0	1 (2.4)
DLBCL subtype, n (%)			
GCB	10 (50.0)	11 (52.4)	21 (51.2)
non-GCB	5 (25.0)	8 (38.1)	13 (31.7)
Double hit/triple hit, n (%)	3 (15.0)	5 (23.8)	8 (19.5)
Number of prior lines of therapy			
Median (range)	2 (1-4)	2 (1-5)	2 (1-5)
1, n (%)	10 (50.0)	10 (47.6)	20 (48.8)
≥2, n (%)	10 (50.0)	11 (52.4)	21 (51.2)
Refractory status, n (%)			
Refractory to primary therapy	8 (40.0)	13 (61.9)	21 (51.2)
Refractory to last prior therapy	7 (35.0)	13 (61.9)	20 (48.8)
Prior stem cell transplant, n (%)	3 (15.0)	1 (4.8)	4 (9.8)
Prior CAR-T therapy, n (%)	4 (20.0)	4 (19.0)	8 (19.5)

CAR-T, chimeric antigen receptor T cell; DLBCL, diffuse large B-cell lymphoma; ECOG PS, Eastern Cooperative Oncology Group Performance Status; FL, follicular lymphoma; Glofit, glofitamab; HGBCL, high-grade B-cell lymphoma; IPI, International Prognostic Index; LBCL, large B-cell lymphoma; LDH, lactate dehydrogenase; Lonca, loncastuximab tesirine; trFL, transformed follicular lymphoma.

Safety Outcomes

- A total of 38 (92.7%) patients presented with all-grade treatment-emergent adverse events (TEAEs), and 23 (56.1%) presented with grade ≥3 TEAEs (**Table 2**)
- The most common grade ≥3 TEAE was neutropenia (24.4%)
- Grade 3 CRS occurred in 1 patient (120-µg/kg dose level); no grade 4 instances of CRS were observed at either dose
 - The rate of any-grade CRS was less frequent at the Lonca 150-µg/kg dose (23.8%) than at 120-µg/kg dose (55.0%)
- Grade 1 ICANS occurred in 1 patient (2.4%), and grade 2 ICANS, in 2 patients (4.9%); no grade ≥3 ICANS were observed at either dose
 - The rate of any-grade ICANS was 4.8% in the Lonca 150-µg/kg dose group and 10.0% in the Lonca 120-µg/kg dose group
- Neutropenia was the most common TEAE leading to dose delay of Lonca (12.2%) and Glofit (17.1%)
- There was 1 patient who had a dose reduction of Lonca

Table 2. Safety outcomes, treated population (N=41)

Characteristic, n (%)	Glofit + Lonca, 120 µg/kg (n=20)	Glofit + Lonca, 150 µg/kg (n=21)	All dose levels (N=41)
Any-grade TEAEs	20 (100)	18 (85.7)	38 (92.7)
Grade ≥3 TEAEs	11 (55.0)	12 (57.1)	23 (56.1)
Grade ≥3 TEAEs occurring in >5% of patients			
Neutropenia	4 (20.0)	6 (28.6)	10 (24.4)
Anemia	1 (5.0)	3 (14.3)	4 (9.8)
Aspartate aminotransferase increased	2 (10.0)	1 (4.8)	3 (7.3)
Gamma-glutamyltransferase increased	1 (5.0)	2 (9.5)	3 (7.3)
Thrombocytopenia	2 (10.0)	1 (4.8)	3 (7.3)
Grade ≥3 treatment-related TEAEs	9 (45.0)	11 (52.4)	20 (48.8)
Grade ≥3 treatment-related TEAEs occurring in >5% of patients			
Neutropenia	4 (20.0)	6 (28.6)	10 (24.4)
Anemia	0	3 (14.3)	3 (7.3)
Aspartate aminotransferase increased	2 (10.0)	1 (4.8)	3 (7.3)
Thrombocytopenia	2 (10.0)	1 (4.8)	3 (7.3)
Rates of CRS and ICANS			
CRS, grade 1	7 (35.0)	5 (23.8)	12 (29.3)
CRS, grade 2	3 (15.0)	0	3 (7.3)
CRS, grade 3	1 (5.0)	0	1 (2.4)
CRS, grade ≥4	0	0	0
ICANS, grade 1	1 (5.0)	0	1 (2.4)
ICANS, grade 2	1 (5.0)	1 (4.8)	2 (4.9)
ICANS, grade ≥3	0	0	0
TEAEs leading to study drug discontinuation			
TEAEs leading to Lonca discontinuation only	1 (5.0)	2 (9.5)	3 (7.3)
TEAEs leading to Glofit discontinuation only	0	3 (14.3)	3 (7.3)

A TEAE is defined as an adverse event that occurs or worsens in the period extending from the first dose of the study drug to 15 weeks after the last dose of the study drug or the start of a new anticancer therapy, whichever is earlier.
CRS, cytokine release syndrome; ICANS, immune effector cell-associated neurotoxicity syndrome; TEAE, treatment-emergent adverse event.

Efficacy Outcomes

- The efficacy evaluable population (N=30) treated at 120- and 150-µg/kg doses of Lonca consisted of patients who had LBCL histologies, including R/R de novo DLBCL (n=19; 63.3%), HGBCL (n=6; 20.0%), transformed FL (n=4; 13.3%), and FL grade 3b (n=1; 3.3%)
- The ORR was 93.3% (28/30), CR rate was 86.7% (26/30), and the median duration of response was not reached; responses were maintained after the end of treatment (**Table 3**)
 - The ORRs for the 120- and 150-µg/kg dose levels were both 93.3% (14/15)
- The median time to first response (CR or partial response [PR]) was 42.0 days, and the median time to CR was 70.5 days
 - The median time to first CR was numerically shorter for the 150-µg/kg (42.0 days) versus the 120-µg/kg (80.0 days) dose group

Table 3. Best overall response and DOR, efficacy evaluable population (N=30)

Efficacy measures	Glofit + Lonca, 120 µg/kg	Glofit + Lonca, 150 µg/kg	Total
Best overall response[a]	(n=15)	(n=15)	(N=30)
ORR (CR+PR), n (%) [95% CI]	14 (93.3) [68.1-99.8]	14 (93.3) [68.1-99.8]	28 (93.3) [77.9-99.2]
CR, n (%) [95% CI]	13 (86.7) [59.5-98.3]	13 (86.7) [59.5-98.3]	26 (86.7) [69.3-96.2]
PR, n (%)	1 (6.7)	1 (6.7)	2 (6.7)
SD, n (%)	1 (6.7)	0	1 (3.3)
PD, n (%)	0	1 (6.7)	1 (3.3)
DOR[b]	(n=14)	(n=14)	(n=28)
Event, n (%)	0	1 (7.1)	1 (3.6)
Median	NE	NE	NE
Probability to remain event-free for 6 months (95% CI)	100 (100-100)	90.9 (50.8-98.7)	95.2 (70.7-99.3)
Time to first response (CR or PR)	(n=14)	(n=14)	(n=28)
Median, days	42.0	42.0	42.0
Time to first CR	(n=13)	(n=13)	(n=26)
Median, days	80.0	42.0	70.5

CR, complete response; DOR, duration of response; NE, not estimable; ORR, overall response rate; PD, progressive disease; PR, partial response; SD, stable disease.
[a]The efficacy evaluable population (N=30) included all patients who received ≥1 dose of the study drug with a valid baseline and ≥1 valid postbaseline disease assessment. Patients who did not have a postbaseline assessment owing to early clinical progression or death were also included.
[b]In the efficacy evaluable population, the DOR and probability of maintaining an event-free response were evaluated in responders (n=28), including all patients who had a best response of CR or PR.

- Of the 30 efficacy evaluable patients, 28 were responders with a best response of CR or PR, including 26 CRs
- Of the 28 responders, 27 remained in response, with 25 of 26 maintaining their CR at the data cutoff (**Figure 2**)
- A total of 12 patients converted to a CR from a PR (11) or SD (1)
 - Most responses were observed at the initial assessment at 6 weeks for both doses
- For most patients who have ended treatment, response was sustained beyond the end of treatment

Figure 2. Efficacy over time (N=41)

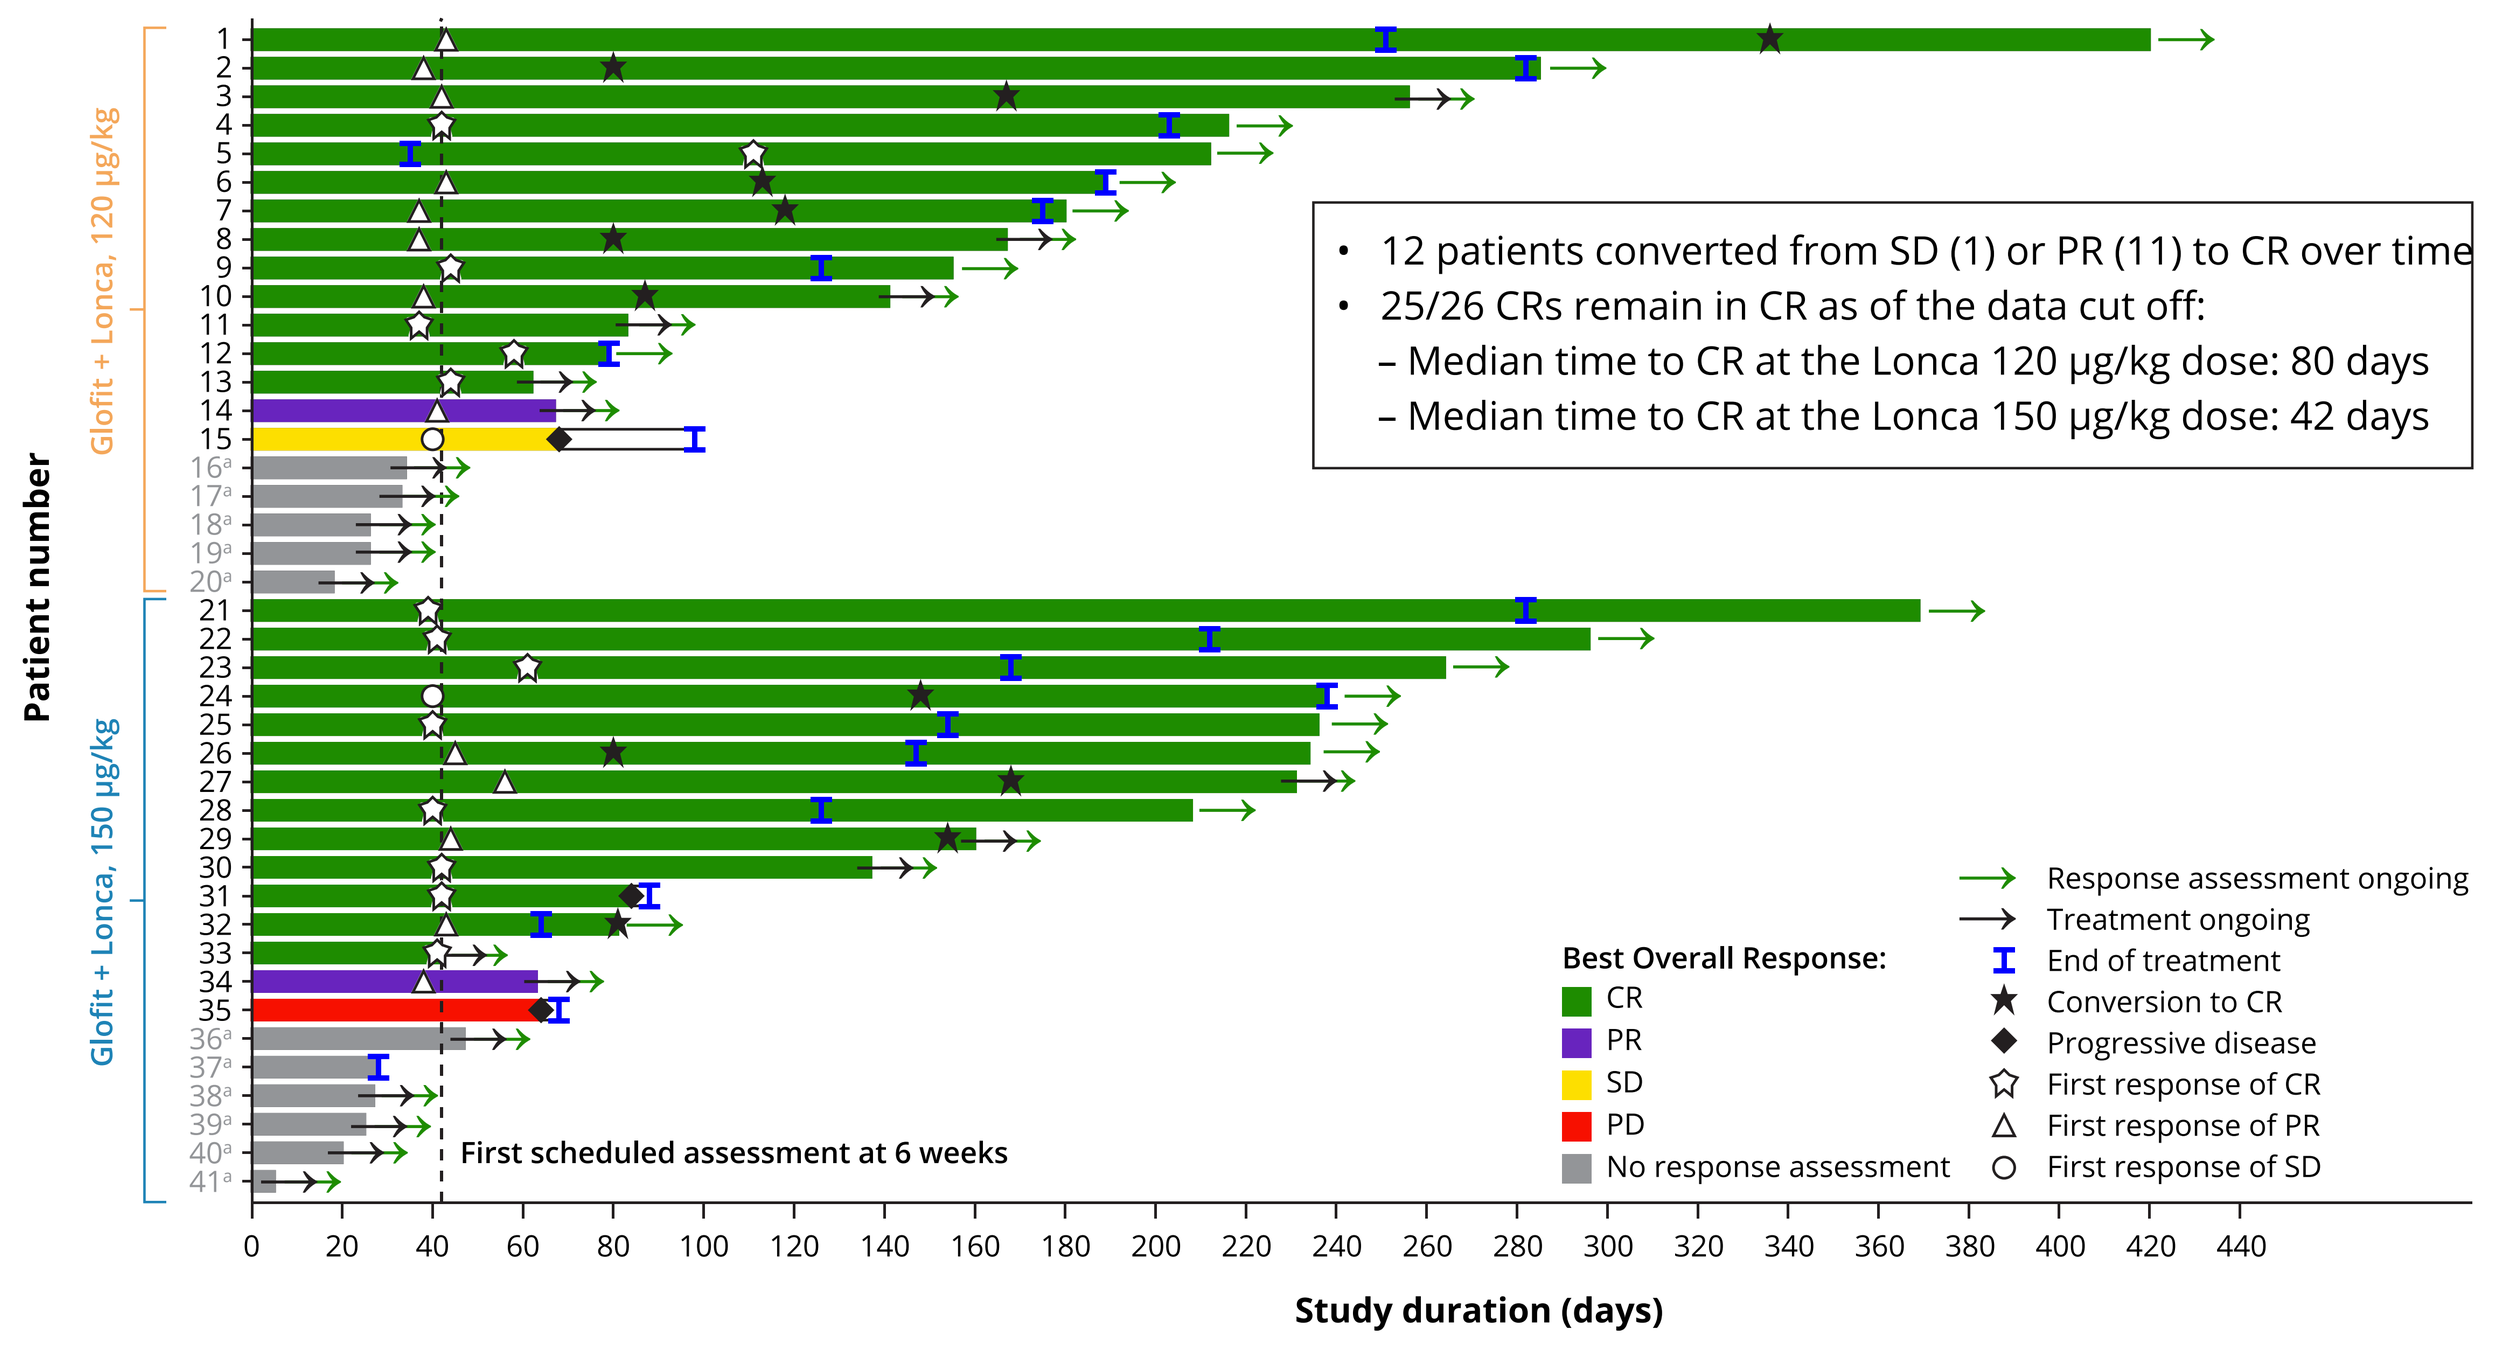
Patient number
Glofit + Lonca, 120 µg/kg
Glofit + Lonca, 150 µg/kg
12 patients converted from SD (1) or PR (11) to CR over time
25/26 CRs remain in CR as of the data cut off:
– Median time to CR at the Lonca 120 µg/kg dose: 80 days
– Median time to CR at the Lonca 150 µg/kg dose: 42 days
First scheduled assessment at 6 weeks
Best Overall Response:
CR
PR
SD
PD
No response assessment
Response assessment ongoing
Treatment ongoing
End of treatment
Conversion to CR
Progressive disease
First response of CR
First response of PR
First response of SD
Study duration (days)

Each bar represents one patient in the study. Response is determined by independent reviewer.
CR, complete response; Glofit, glofitamab; Lonca, loncastuximab tesirine; PR, partial response; SD, stable disease.
[a]Patients were not yet efficacy evaluable because they had not reached the 6-week assessment (n=10) or had withdrawn before any assessment (patient 39).

PK and Biomarker Outcomes

- Lonca exposure (AUC_{last} and C_{max}) showed a dose-dependent increase in the first 2 cycles
 - Coadministration of Lonca + Glofit showed lower Lonca C_{max}, especially in cycle 2, compared with Lonca monotherapy, while AUC_{last} was within the range of values observed with Lonca monotherapy
- No post-dose Lonca antidrug antibodies were detected with Lonca + Glofit, indicating low immunogenicity with the combination
- Flow cytometry assessment showed similar patterns of T-cell ($CD3^+/CD4^+$ and $CD3^+/CD8^+$) margination with Lonca + Glofit compared to that previously reported with Glofit monotherapy (data not shown)[13]
- The number of circulating activated T cells (HLA^-DR^+) also increased during treatment (orange and yellow lines, **Figure 3**)
- Monocytes ($CD14^+$) and natural killer cells ($CD3^-/CD16^+/CD56^+$) were similarly modulated and showed a trend of increase over time (data not shown)
- Cytokine profiles, assessed by multiplex immunoassay, indicated immune activation as exemplified by IFN-γ (blue line, **Figure 3**)
 - Patterns of IFN-γ were consistent with previous reports of Glofit monotherapy; there was a transient increase of IFN-γ after the first Glofit infusion, which decreased by the second infusion and then normalized; a similar pattern was seen for IL-6 (data not shown)

Figure 3. Patterns of helper T cells, cytotoxic T cells, and IFN-γ

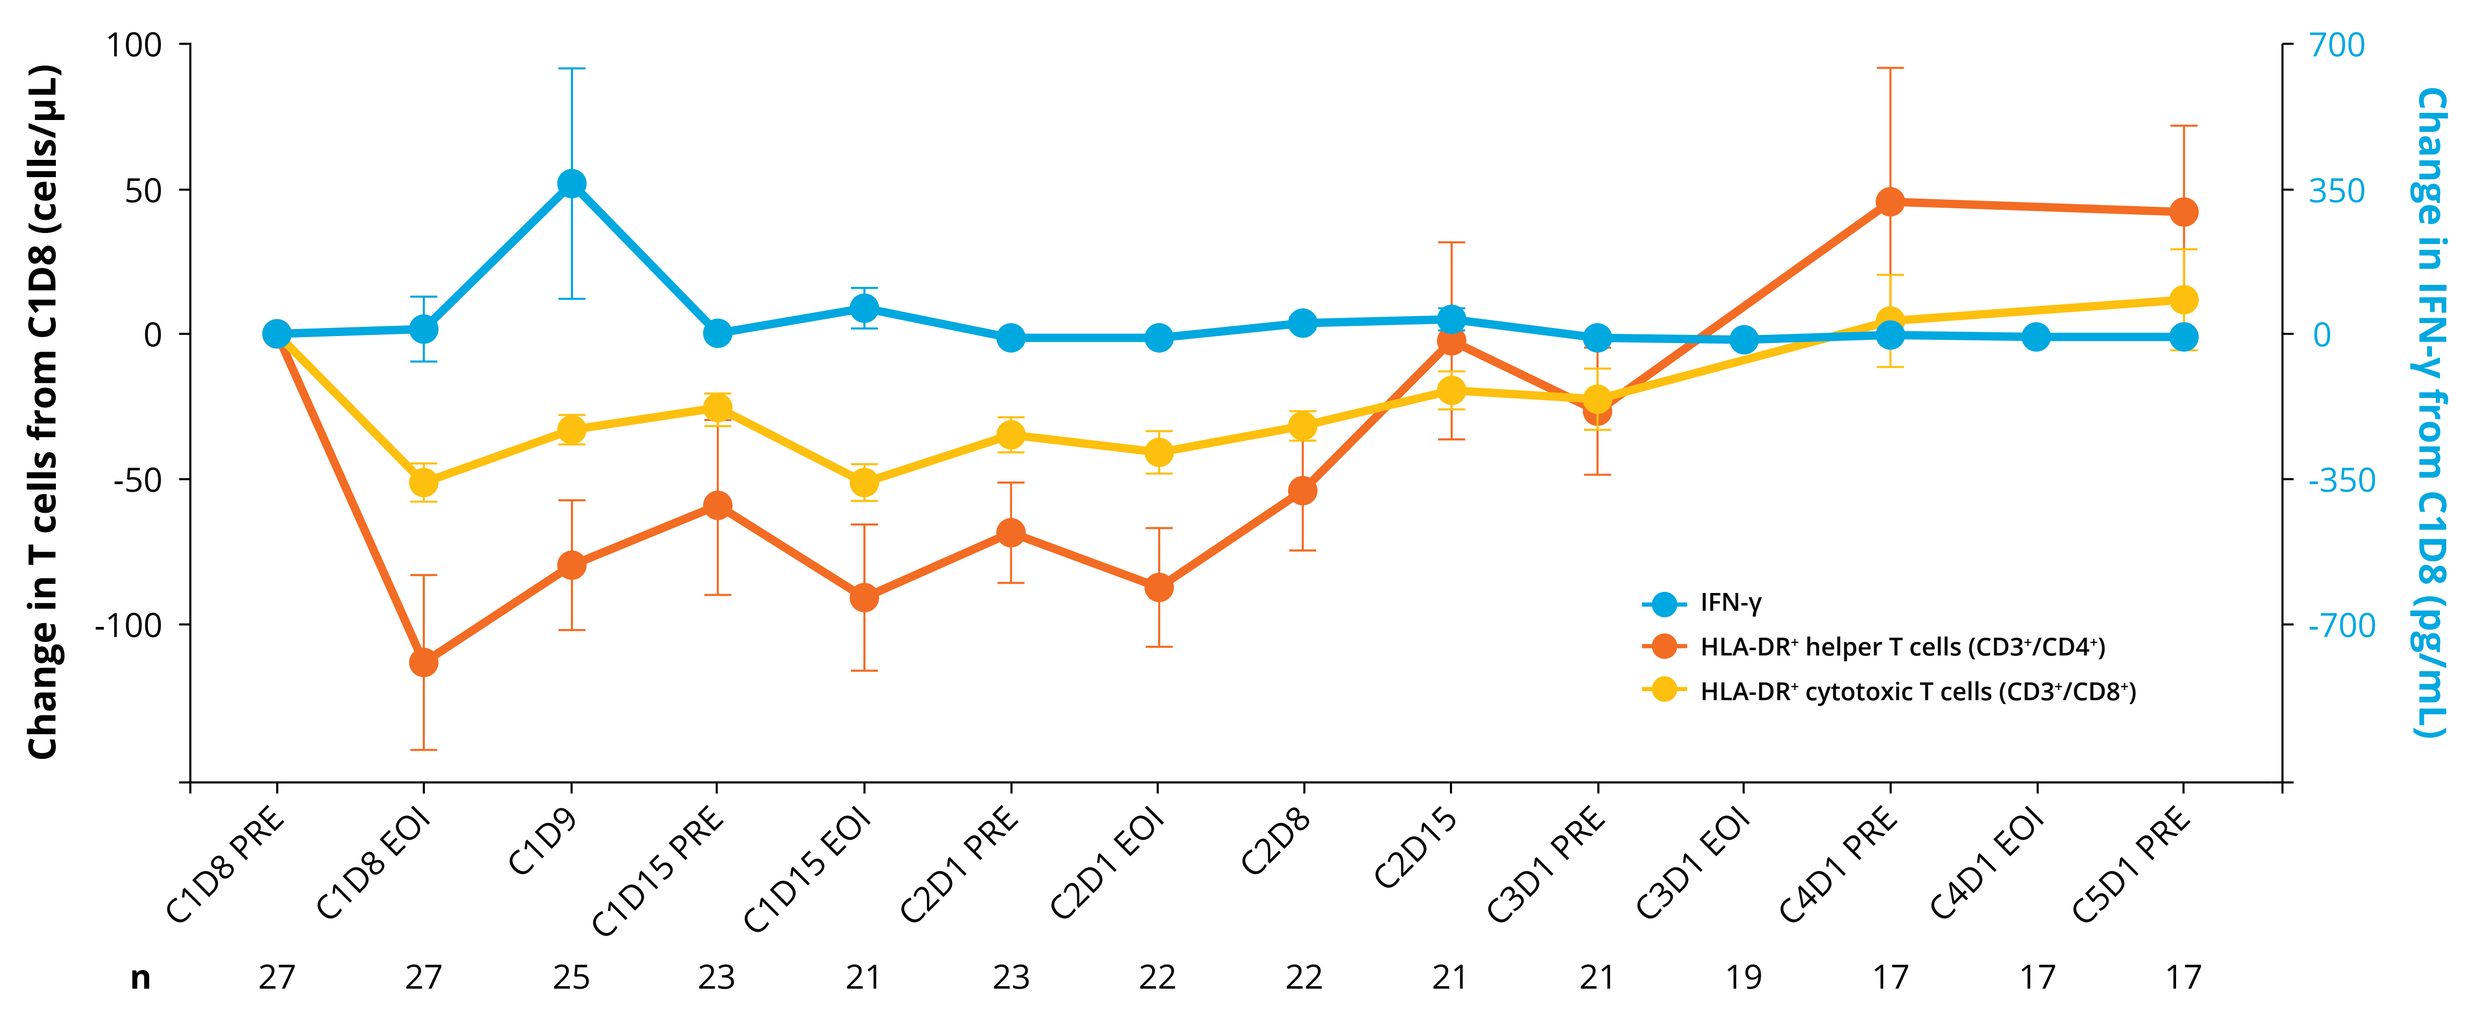
Change in T cells from C1D8 (cells/µL)
Change in IFN-γ from C1D8 (pg/mL)
IFN-γ
HLA-DR+ helper T cells (CD3+/CD4+)
HLA-DR+ cytotoxic T cells (CD3+/CD8+)
C1D8 PRE
C1D8 EOI
C1D9
C1D15 PRE
C1D15 EOI
C2D1 PRE
C2D1 EOI
C2D8
C2D15
C3D1 PRE
C3D1 EOI
C4D1 PRE
C4D1 EOI
C5D1 PRE
n
27
27
25
23
21
23
22
22
21
21
19
17
17
17

C, cycle; D, day; EOI, end of infusion; HLA-DR, human leukocyte antigen – DR isotype; IFN-γ, interferon gamma; PRE, pretreatment.

***Contact Information:**
Juan Pablo Alderuccio, MD: jalderuccio@med.miami.edu

Acknowledgments

The LOTIS-7 trial (NCT04970901) was funded by ADC Therapeutics SA and partially funded by Sobi. Medical writing and editorial support were provided by Nathan Hutcheson, PhD, CMPP, from Citrus Health Group (Chicago, Illinois, USA), and funded by ADC Therapeutics SA and Sobi.

Disclosures

JP Alderuccio: consultant for AbbVie, ADC Therapeutics SA, Genentech, Genmab, Lilly, Novartis, and Regeneron; recipient of research funding from AbbVie, ADC Therapeutics SA, BeiGene, and Genmab; immediate family member has served on the advisory boards of Anheart, Fore, Rigel, and Servier; and speaker for Medscape. **C Okada:** recipient of research funding from ADC Therapeutics SA, Genentech, Genmab A/S, Ono Pharmaceutical, and Pfizer. **G Crochet:** recipient of honorary, consultancy, or travel grant from AbbVie, Gilead, Roche, and Sobi. **E Ayers:** consultant or advisor for AbbVie, ADC Therapeutics SA, and Genentech; and recipient of research funding from AbbVie and Loxo Oncology/Lilly. **M Hu:** no disclosures. **S Ferrari:** no disclosures. **P Caimi:** member of board of directors or advisory committees for ADC Therapeutics SA, Autolus, BMS, Genentech, Genmab, Novartis, and Synthekine. **M Hamadani:** consultant for AbGenomics, ADC Therapeutics SA, Celgene Corporation, Incyte Corporation, Janssen R&D, Omeros, Pharmacyclics, TeneoBio, and Verastem; member of speakers bureau for AstraZeneca, BeiGene, and Sanofi Genzyme; and recipient of research support from Astellas Pharma, Spectrum Pharmaceuticals, and Takeda. **J Depaus:** consultant or advisor for Janssen/J&J Innovative Medicine, Novartis AG, and Takeda Pharmaceutical Company Limited. **E Derenzini:** recipient of research funding from ADC Therapeutics SA and Takeda and advisory board for AbbVie, AstraZeneca, BeiGene, and Roche. **J Sandoval-Sus:** consultant for AbbVie, Acrotech, ADC Therapeutics SA, BeiGene, BMS, Genmab, and MassiveBio and recipient of speakers bureau fees from Pfizer. **R Reyes:** was an employee of ADC Therapeutics SA at the time of the study and current equity holder at ADC Therapeutics SA. **A Urban:** employee and stock options at ADC Therapeutics SA. **E Rave:** employee and stock options at ADC Therapeutics SA. **A Graf:** employee and stock options at ADC Therapeutics SA. **E Tiecke:** employee at ADC Therapeutics SA. **M Toukam:** employee and stock options at ADC Therapeutics SA. **PL Zinzani:** consultant for Merck Sharp & Dohme, Novartis, Recordati, and Takeda and member of speakers bureau or advisory board for ADC Therapeutics SA, AstraZeneca, BeiGene, BMS, Incyte, Janssen-Cilag, Kite-Gilead, Kyowa Kirin, Merck Sharp & Dohme, Novartis, Recordati, Roche, Sobi, and Takeda.

References

1. Dickinson MJ et al., *N Engl J Med* 2022;387:2220-2231.
2. ZYNLONTA® [Prescribing information]. US Food and Drug Administration. October 2022.
3. ZYNLONTA® [Product information]. European Medicines Agency. October 2024.
4. ZYNLONTA® [Approval notice]. National Medical Products Administration (China). December 2024.
5. ZYNLONTA® [Summary Basis of Decision]. Health Canada. March 2025.
6. COLUMVI® [Prescribing information]. US Food and Drug Administration. June 2023.
7. COLUMVI® [Product information]. European Medicines Agency. March 2025.
8. COLUMVI® [Summary Basis of Decision]. Health Canada. 24 Mar 2023.
9. COLUMVI® (glofitamab) – Conditional marketing approval. NMPA. 8 Nov 2023.
10. Caimi PF, et al. *Lancet Oncol*. 2021;22(6):790-800.
11. Hutchings M, et al. *J Clin Oncol*. 2021;39(18):1959-1970.
12. ClinicalTrials.gov. A study to evaluate the safety and anti-cancer activity of loncastuximab tesirine in combination with other anti-cancer agents in participants with relapsed or refractory B-cell non-Hodgkin lymphoma (LOTIS-7) (NCT04970901). 2021. Accessed March 14, 2025. https://clinicaltrials.gov/ct2/show/NCT04970901.
13. Broske A-M, et al. *Blood Adv*. 2022;6(3):1025-1037.